Economic burden and healthcare resource utilization in patients with prurigo nodularis in the United States

Amy H. Huang MPH[1], Rajeev Chavda MD[2], Sylvie Gabriel[2], Joseph K. Canner MHS[3], Shawn G. Kwatra MD[1]

[1]Department of Dermatology, Johns Hopkins University School of Medicine, Baltimore, MD, USA
[2]Rx Strategy and Innovation Group, Galderma S.A., La Tour-de-Peilz, Switzerland
[3]Johns Hopkins Center for Outcomes Research (JSCOR), Johns Hopkins University School of Medicine, Baltimore, MD, USA

Objectives

Prurigo nodularis (PN) is a chronic dermatosis characterized by severely pruritic nodules. In addition to PN's significant impact on quality of life, many patients are recalcitrant to therapy with no FDA-approved therapies to date. Despite this, little is known about real-world healthcare resource utilization and economic burden in prurigo nodularis in the US.

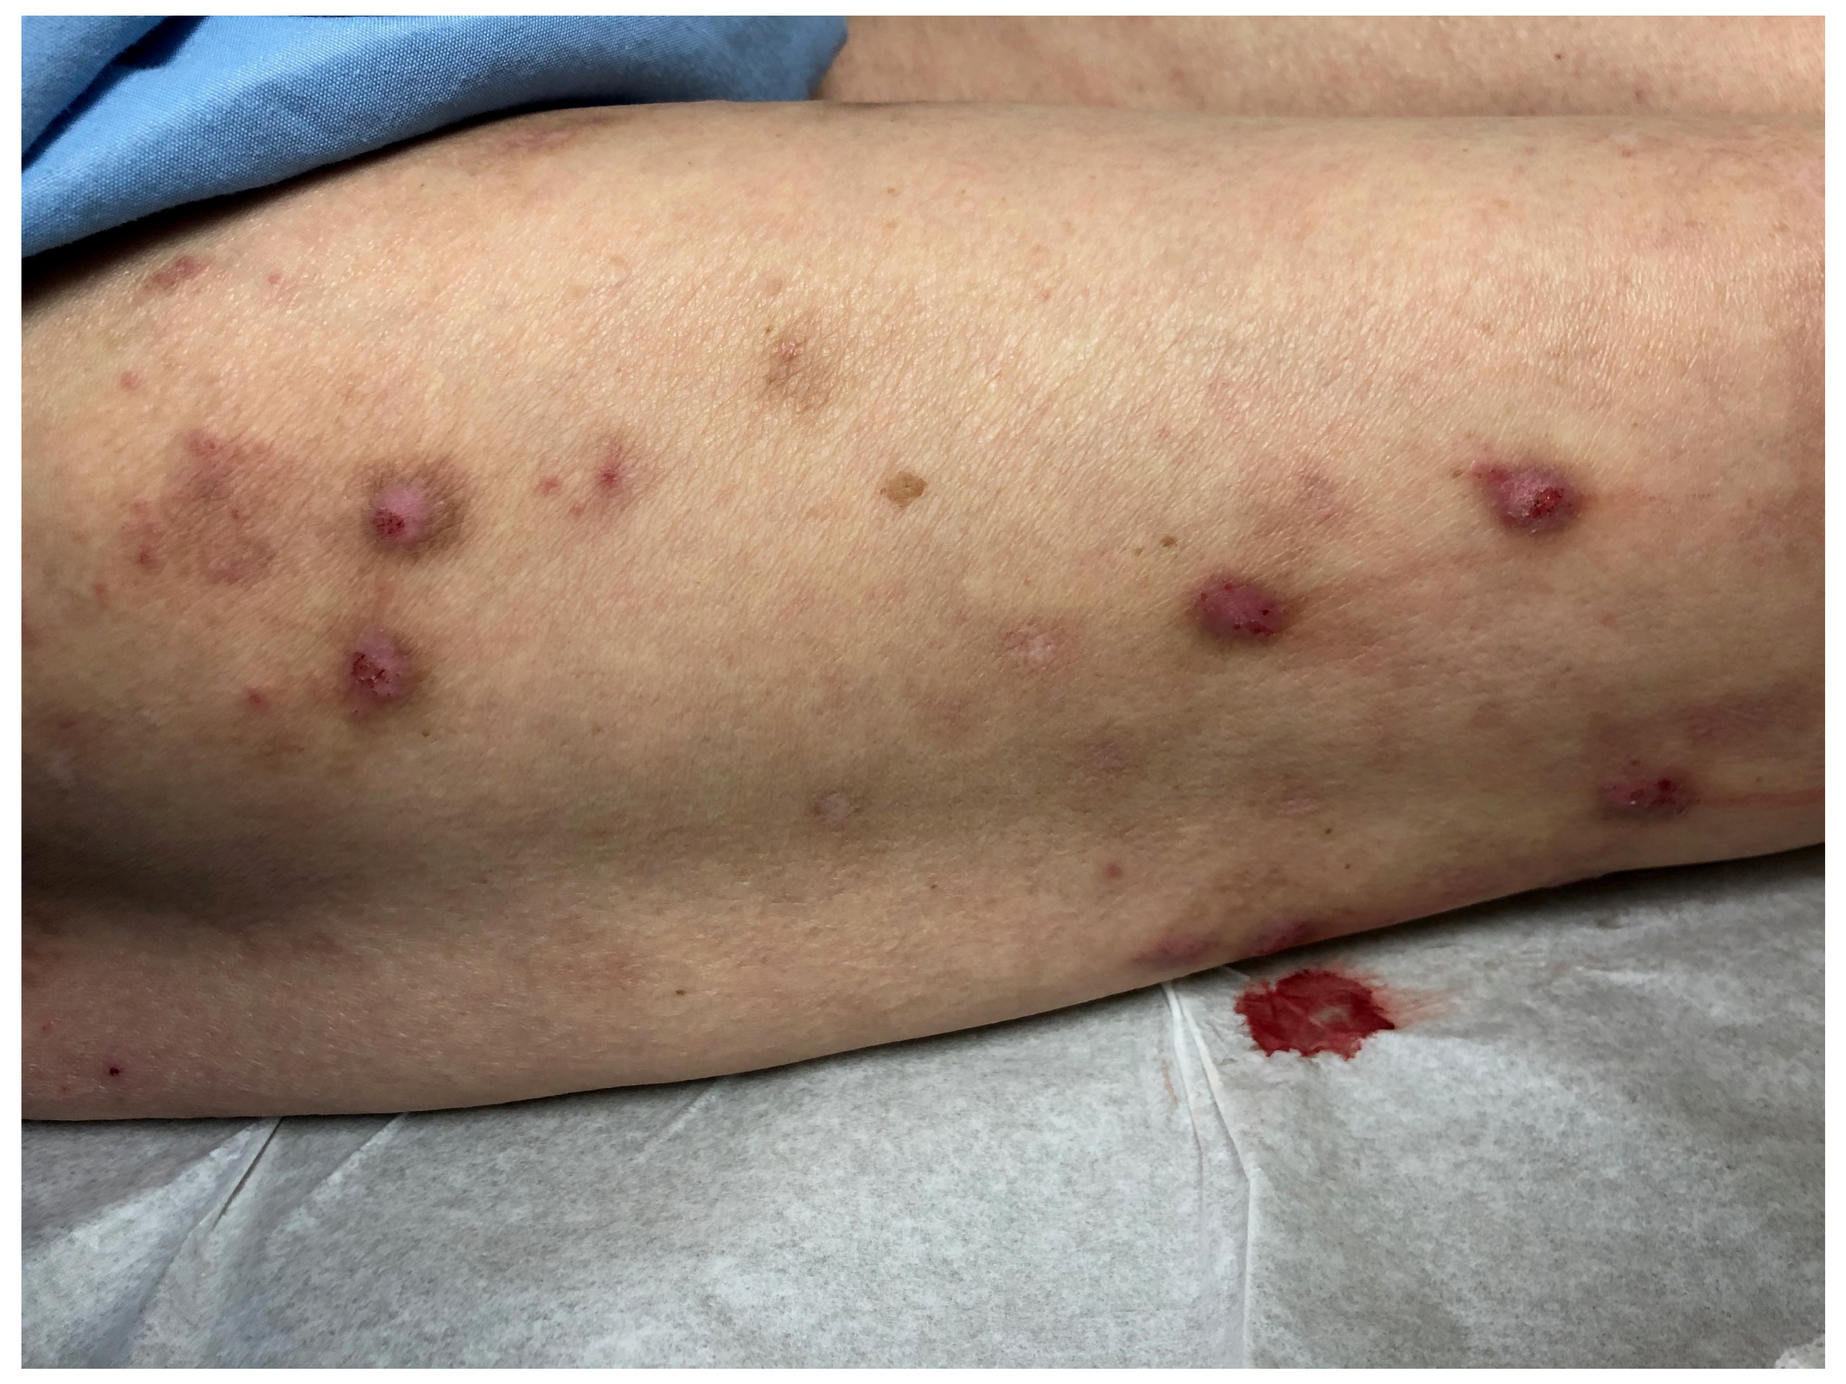

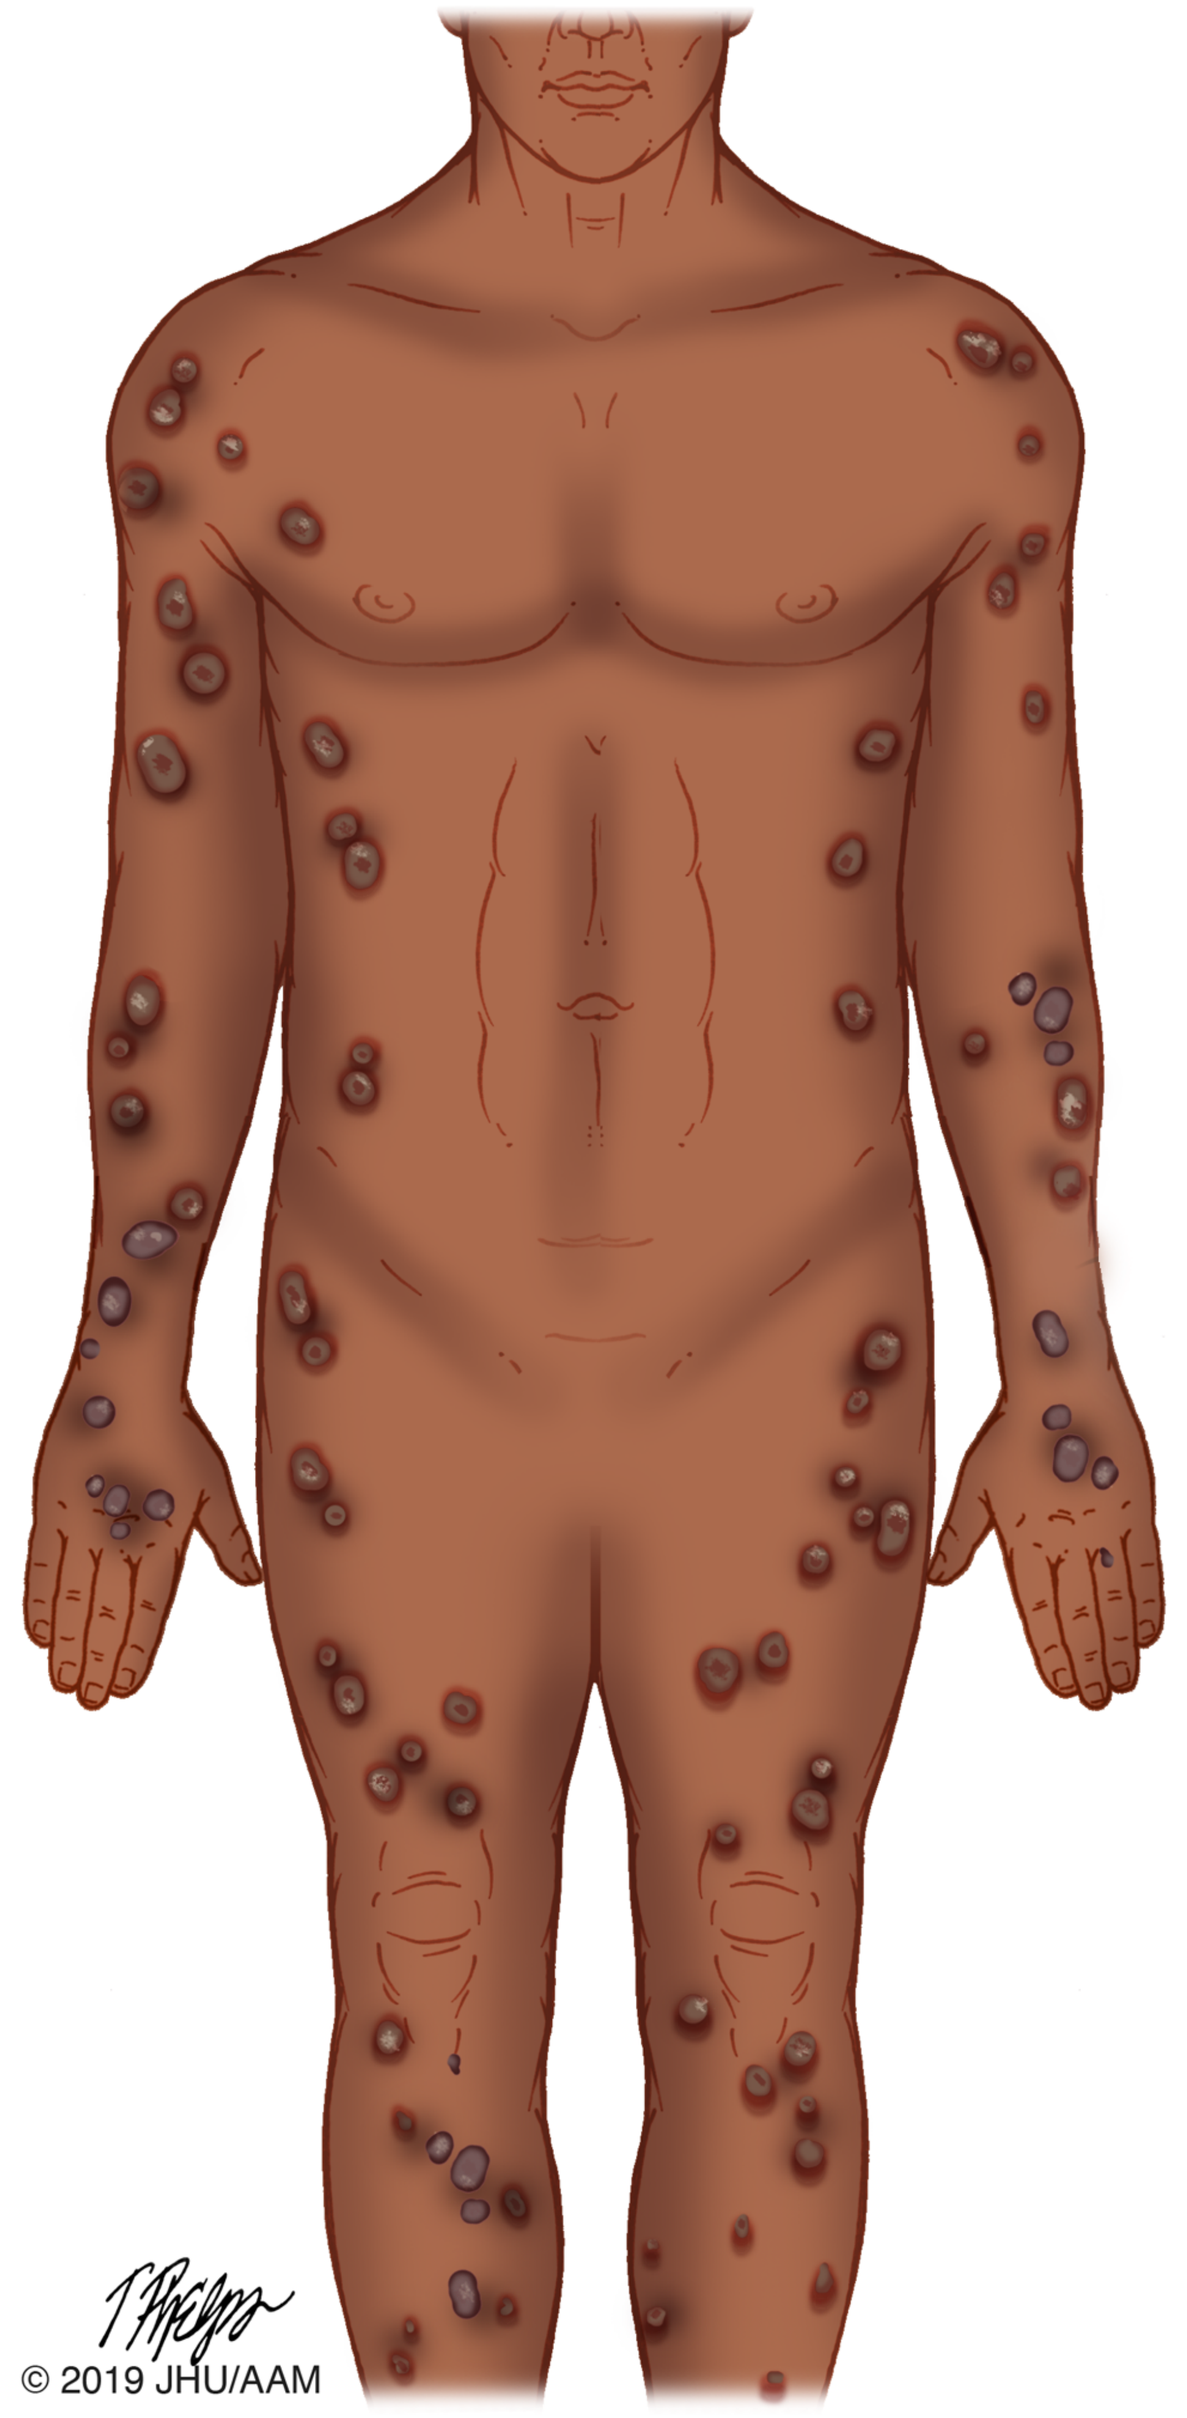
© 2019 JHU/AAM

Methods

IBM's MarketScan Commercial Claims database was analyzed between October 2015 and December 2016. ICD-10-CM codes were used to identify patients age 18-64 years with PN and 3 additional cohorts without PN: age and sex-matched controls, atopic dermatitis, and psoriasis. Relative cost ratios were calculated generalized linear models with log link and gamma distribution. Incidence rate ratios were calculated using negative binomial regression.

Results

Table 1. Relative cost ratios of healthcare utilization of prurigo nodularis relative to comparison groups

	Comparison group									
Setting	Reference: Matched controls (N=16,595)		Reference: Atopic dermatitis (N= 23,915)				Reference: Psoriasis (N=39,346)			
	Cost ratio	p-value	Unadjusted cost ratio	p-value	Adjusted cost ratio	p-value	Unadjusted cost ratio	p-value	Adjusted cost ratio	p-value
Inpatient	1.28	0.026	1.35	0.009	1.05	0.703	0.75	0.008	0.69	0.001
ED	0.94	0.427	0.90	0.111	0.95	0.426	0.78	<0.001	0.79	<0.001
Outpatient	1.67	<0.001	1.39	<0.001	1.17	0.001	0.85	<0.001	0.76	<0.001
Pharmacy/Lab	2.70	<0.001	1.05	0.515	0.96	0.558	0.70	0.001	0.65	<0.001
Total	1.52	<0.001	1.32	<0.001	1.10	0.030	0.81	<0.001	0.74	0.001

Table 2. Relative incidence rates of being seen by provider type

	Incidence rate ratio (IRR) in patients with prurigo nodularis (N=7,095), (95% CI)				
Provider type	Reference: Matched controls (N=16,595)	Reference: Atopic dermatitis (N= 23,882)		Reference: Psoriasis (N=39,283)	
	IRR (95% CI)	Unadjusted IRR (95% CI)	Adjusted IRR (95% CI)	Unadjusted IRR (95% CI)	Adjusted IRR (95% CI)
Dermatology	31.9 (29.3, 34.7)	1.89 (1.81, 1.99)	5.78 (5.38, 6.22)	1.67 (1.59, 1.75)	5.69 (5.30, 6.10)
Internal/family medicine	1.09 (1.02, 1.16)	0.97 (0.93, 1.01)	0.76 (0.72, 0.81)	0.86 (0.83, 0.89)	0.62 (0.58, 0.65)
Physical therapy/chiropractor	1.10 (1.01, 1.20)	1.19 (1.02, 1.39)	1.10 (1.01, 1.19)	1.19 (1.03, 1.38)	1.06 (0.98, 1.14)
Cardiology	1.18 (1.07, 1.29)	1.79 (1.58, 2.04)	1.18 (1.08, 1.28)	1.19 (1.06, 1.34)	0.97 (0.90, 1.05)
Gastroenterology	1.08 (0.98, 1.19)	1.45 (1.28, 1.63)	1.12 (1.03, 1.22)	0.98 (0.87, 1.10)	0.97 (0.89, 1.05)
Psychiatry	2.03 (1.79, 2.32)	1.18 (0.95, 1.45)	1.73 (1.55, 1.92)	1.21 (0.99, 1.48)	1.45 (1.31, 1.60)
Endocrine	1.64 (1.42, 1.89)	1.62 (1.30, 2.03)	1.49 (1.32, 1.69)	1.28 (1.04, 1.59)	1.28 (1.15, 1.44)
Allergy and immunology	2.07 (1.73, 2.48)	0.22 (0.18, 0.26)	0.23 (0.20, 0.26)	1.65 (1.39, 1.96)	1.56 (1.36, 1.78)
Rheumatology	1.82 (1.51, 2.18)	1.44 (1.25, 1.66)	1.36 (1.17, 1.57)	0.14 (0.12, 0.16)	0.15 (0.13, 0.17)
Oncology	1.24 (1.00, 1.53)	1.47 (1.00, 2.14)	1.20 (1.00, 1.45)	0.78 (0.54, 1.12)	0.98 (0.83, 1.16)
Pain	1.38 (0.91, 2.07)	2.35 (1.54, 3.60)	2.05 (1.67, 2.52)	1.58 (1.05, 2.37)	1.36 (1.14, 1.61)
Nephrology	1.62 (1.23, 2.12)	3.13 (1.98, 4.97)	1.64 (1.29, 2.09)	2.61 (1.68, 4.04)	1.24 (1.01, 1.52)
Hematology	1.26 (0.98, 1.63)	1.26 (0.82, 1.95)	1.20 (0.96, 1.51)	0.85 (0.56, 1.28)	0.94 (0.77, 1.15)
Infectious diseases	1.85 (1.35, 2.55)	1.90 (1.23, 2.95)	1.17 (0.90, 1.51)	0.71 (0.47, 1.07)	0.83 (0.65, 1.04)
Critical care medicine	1.26 (0.71, 2.25)	1.67 (0.80, 3.47)	1.26 (0.76, 2.08)	1.10 (0.55, 2.20)	0.94 (0.60, 1.46)

Conclusion

Prurigo nodularis is a chronic dermatosis with significant economic burden, incurring significantly higher overall healthcare costs compared to age and sex-matched controls. Furthermore, patients with PN more often require specialist care by dermatology and psychiatry. Optimizing clinical management of these patients may help mitigate the economic burden of PN and its impact on quality of life.

References

1. Huang AH, Canner JK, Khanna R, Kang S, Kwatra SG. Real-world prevalence of prurigo nodularis and burden of associated diseases. *J Invest Dermatol*. 2019.
2. Boozalis E, Tang O, Patel S, et al. Ethnic differences and comorbidities of 909 prurigo nodularis patients. *J Am Acad Dermatol*. 2018;79:714-719.
3. Larson VA, Tang O, Stander S, Miller LS, Kang S, Kwatra SG. Association between prurigo nodularis and malignancy in middle-aged adults. *J Am Acad Dermatol*. 2019.
4. Huang AH, Canner JK, Kang S, Kwatra SG. Analysis of real-world treatment patterns in patients with prurigo nodularis. *J Am Acad Dermatol*. 2019;Epub ahead. doi:10.1016/j.jaad.2019.09.007
5. Whang KA, Kang S, Kwatra SG. Inpatient Burden of Prurigo Nodularis in the United States. *Medicines*. 2019;6(98). doi:doi:10.3390/medicines6030088
6. Jørgensen KM, Egeberg A, Gislason GH, Skov L, Thyssen JP. Anxiety, depression and suicide in patients with prurigo nodularis. *JEADV*. 2017;31(2):e106-e107.

Financial Disclosures
Dr. Shawn G. Kwatra is on the advisory board and serves as a consultant for Menlo Therapeutics, Pfizer Inc., and Regeneron Pharmaceuticals, and has received grant funding from Kiniksa Pharmaceuticals. Dr. Chavda and Dr. Gabriel are employees of Galderma. The other authors have no disclosures.