EE49

Economic Burden of HPV-related Cancers in Bulgaria

Elina Petrova[1]; Hristina Lebanova[2]; Svetoslav Stoev[2]

[1]MSD, Sofia, Bulgaria; Merck & Co. Inc, Rahway, NJ, USA

[2]Faculty of Pharmacy, Medical University Pleven

Background

HPV (human papillomavirus) infection is now recognized as the most prevalent sexually transmitted disease worldwide.[1,2]

Associated with HPV are the cancers of cervix (WHO ICD-10th revision code C53), vagina (ICD-10 C52), vulva (ICD-10 C51), penis (ICD-10 C60), anus (ICD-10 C21), oral cavity and lip (ICD-10 C00-C06), nasopharynx (ICD-10 C11), oropharynx (ICD-10 C09-C10), hypopharynx (ICD-10 C12-C13), and larynx (ICD-10 C32).[3]

HPV-related malignancies and illnesses impose a significant financial burden on health care systems worldwide.[4-8] Most expenditures are related to hospital care and are concentrated in the first year of care.[9,10] Data on economic burden of HPV-related cancers in Bulgaria are scarce. A previous retrospective study estimated that direct and indirect costs associated with cervical cancer in Bulgaria for a 3-year period amount up to €26M.[11] A model developed for 9 Central Eastern European countries estimates the number of HPV-related deaths and years of life lost in 2019 based on published HPV-attributable fractions. According to the results, the present value of the future lost productivity in Bulgaria due to cervical, anal, pharynx, and larynx cancers is €7M.[12]

Objectives

The study aims to identify direct health care costs of cervical, vaginal, vulvar, anal, penile, and H&N cancers in bulgaria and to calculate indirect costs and years of life lost associated with cervical, vaginal, vulvar, anal, penile and H&N cancers.

Methodology

- Prevalence-based cost of illness study
- Time horizon – 2018-2020
- Population – patients diagnosed with cervical, vaginal, vulvar, anal, penile. and H&N cancers (ICD-10 codes [C01], [C02], [C03], [C04], [C05], [C06], [C09], [C10], ([C11], [C12], [C13], [C21], [C32], [C51], [C52], [C53], [C60])
- Cost profile:
 - Direct costs (resource utilization for inpatient and outpatient care events related to treatment and follow-up)
 - Indirect costs due to productivity loss as percentage of GDP incurred to society due to cancer-specific premature mortality (human capital approach)
- Data sources:
 - National Health Insurance Fund (NHIF)
 - National Statistical Institute (NSI)
- Population

Table 1. Study sample

	2018	2019	2020
C01 Malignant neoplasm of base of tongue	40	37	33
C02 Malignant neoplasm of other and unspecified parts of tongue	128	113	114
C03 Malignant neoplasm of gum	34	31	39
C04 Malignant neoplasm of floor of mouth	79	77	83
C05 Malignant neoplasm of palate	31	34	26
C06 Malignant neoplasm of other and unspecified parts of mouth	27	27	31
C09 Malignant neoplasm of tonsils	92	108	123
C10 Malignant neoplasm of oropharynx	83	94	99
C11 Malignant neoplasm of nasopharynx	140	130	99
C12 Malignant neoplasm of pyriform sinus	0	3	0
C13 Malignant neoplasm of hypopharynx	94	110	110
C21 Malignant neoplasm of anus and anal canal	61	55	47
C32 Malignant neoplasm of larynx	312	288	255
C51 Malignant neoplasm of vulva	65	62	58
C52 Malignant neoplasm of vagina	29	27	22
C53 Malignant neoplasm of cervix uteri	916	904	937
C60 Malignant neoplasm of penis	27	22	24
Total	2,158	2,122	2,100

Source: NHIF

Results

Figure 1. Direct costs 2018-2020

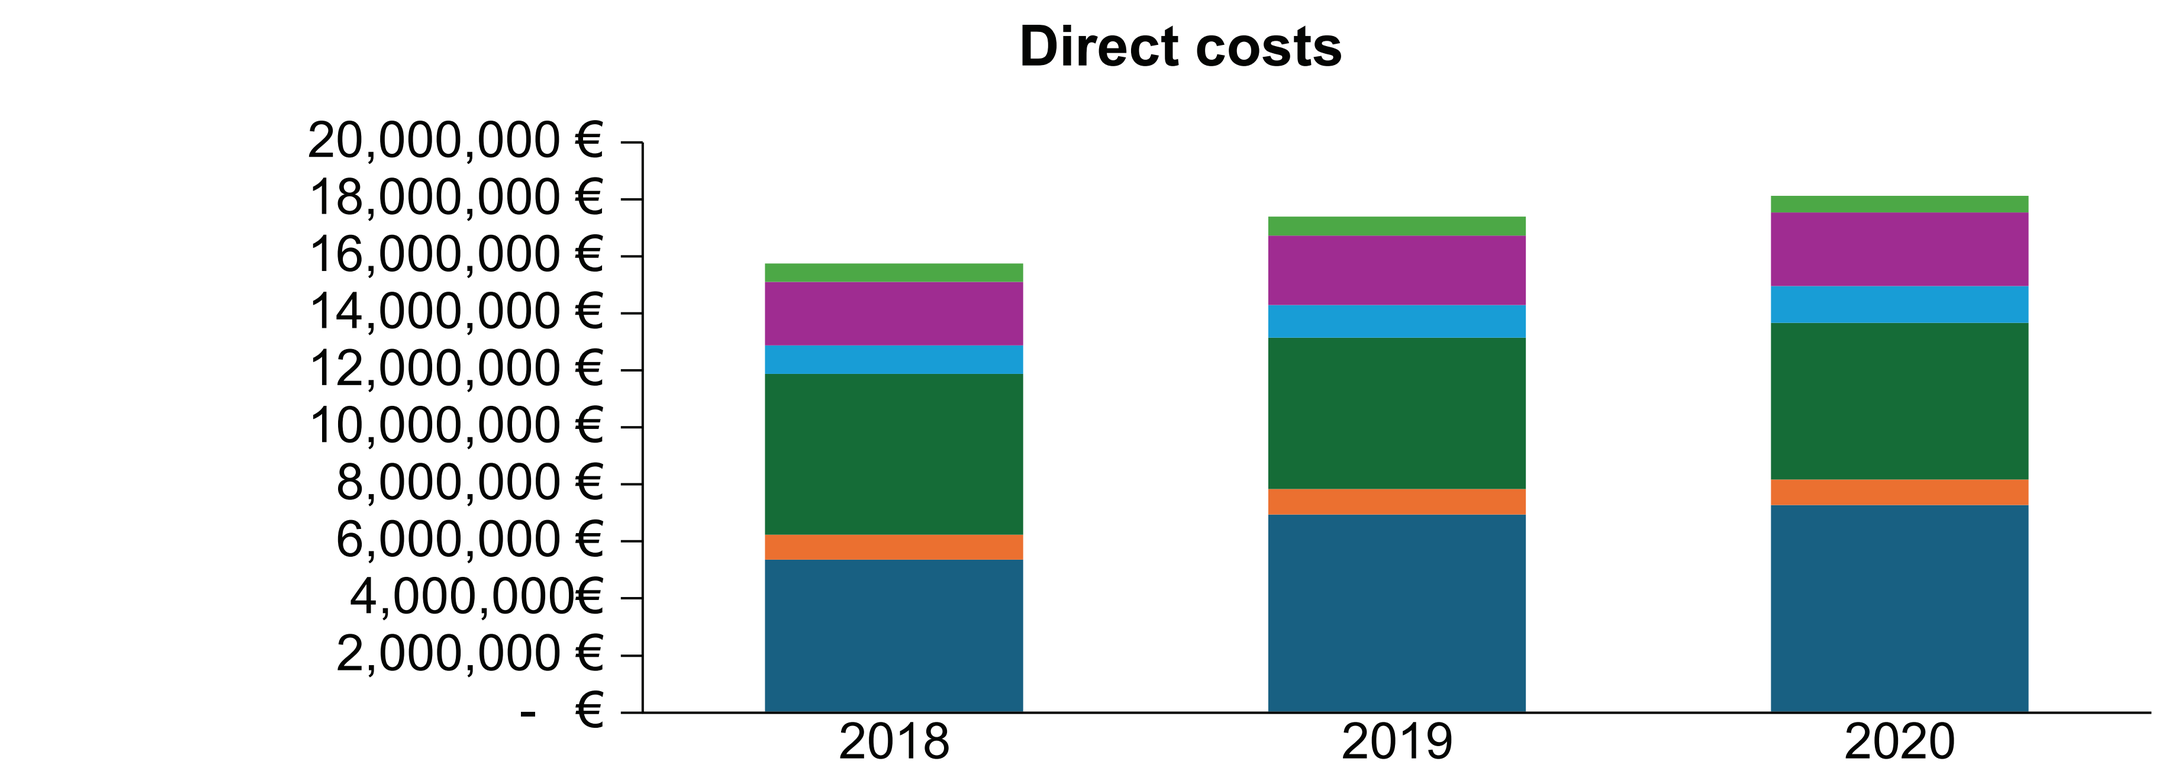
Direct costs
20,000,000 €
18,000,000 €
16,000,000 €
14,000,000 €
12,000,000 €
10,000,000 €
8,000,000 €
6,000,000 €
4,000,000€
2,000,000 €
- €
2018
2019
2020

	2018	2019	2020
Outpatient follow-up costs	651,585 €	667,072 €	574,350 €
Surgical treatment	2,224,294 €	2,443,040 €	2,598,510 €
Diagnostic and staging costs	994,580 €	1,138,606 €	1,271,281 €
Radiotherapy costs	5,639,289 €	5,312,491 €	5,509,364 €
Drug administration costs	873,757 €	896,586 €	897,903 €
Drug therapy costs	5,360,455 €	6,941,095 €	7,267,860 €

Figure 2. Distribution of drug acquisition costs by ICD-10 diagnosis (Pareto chart)

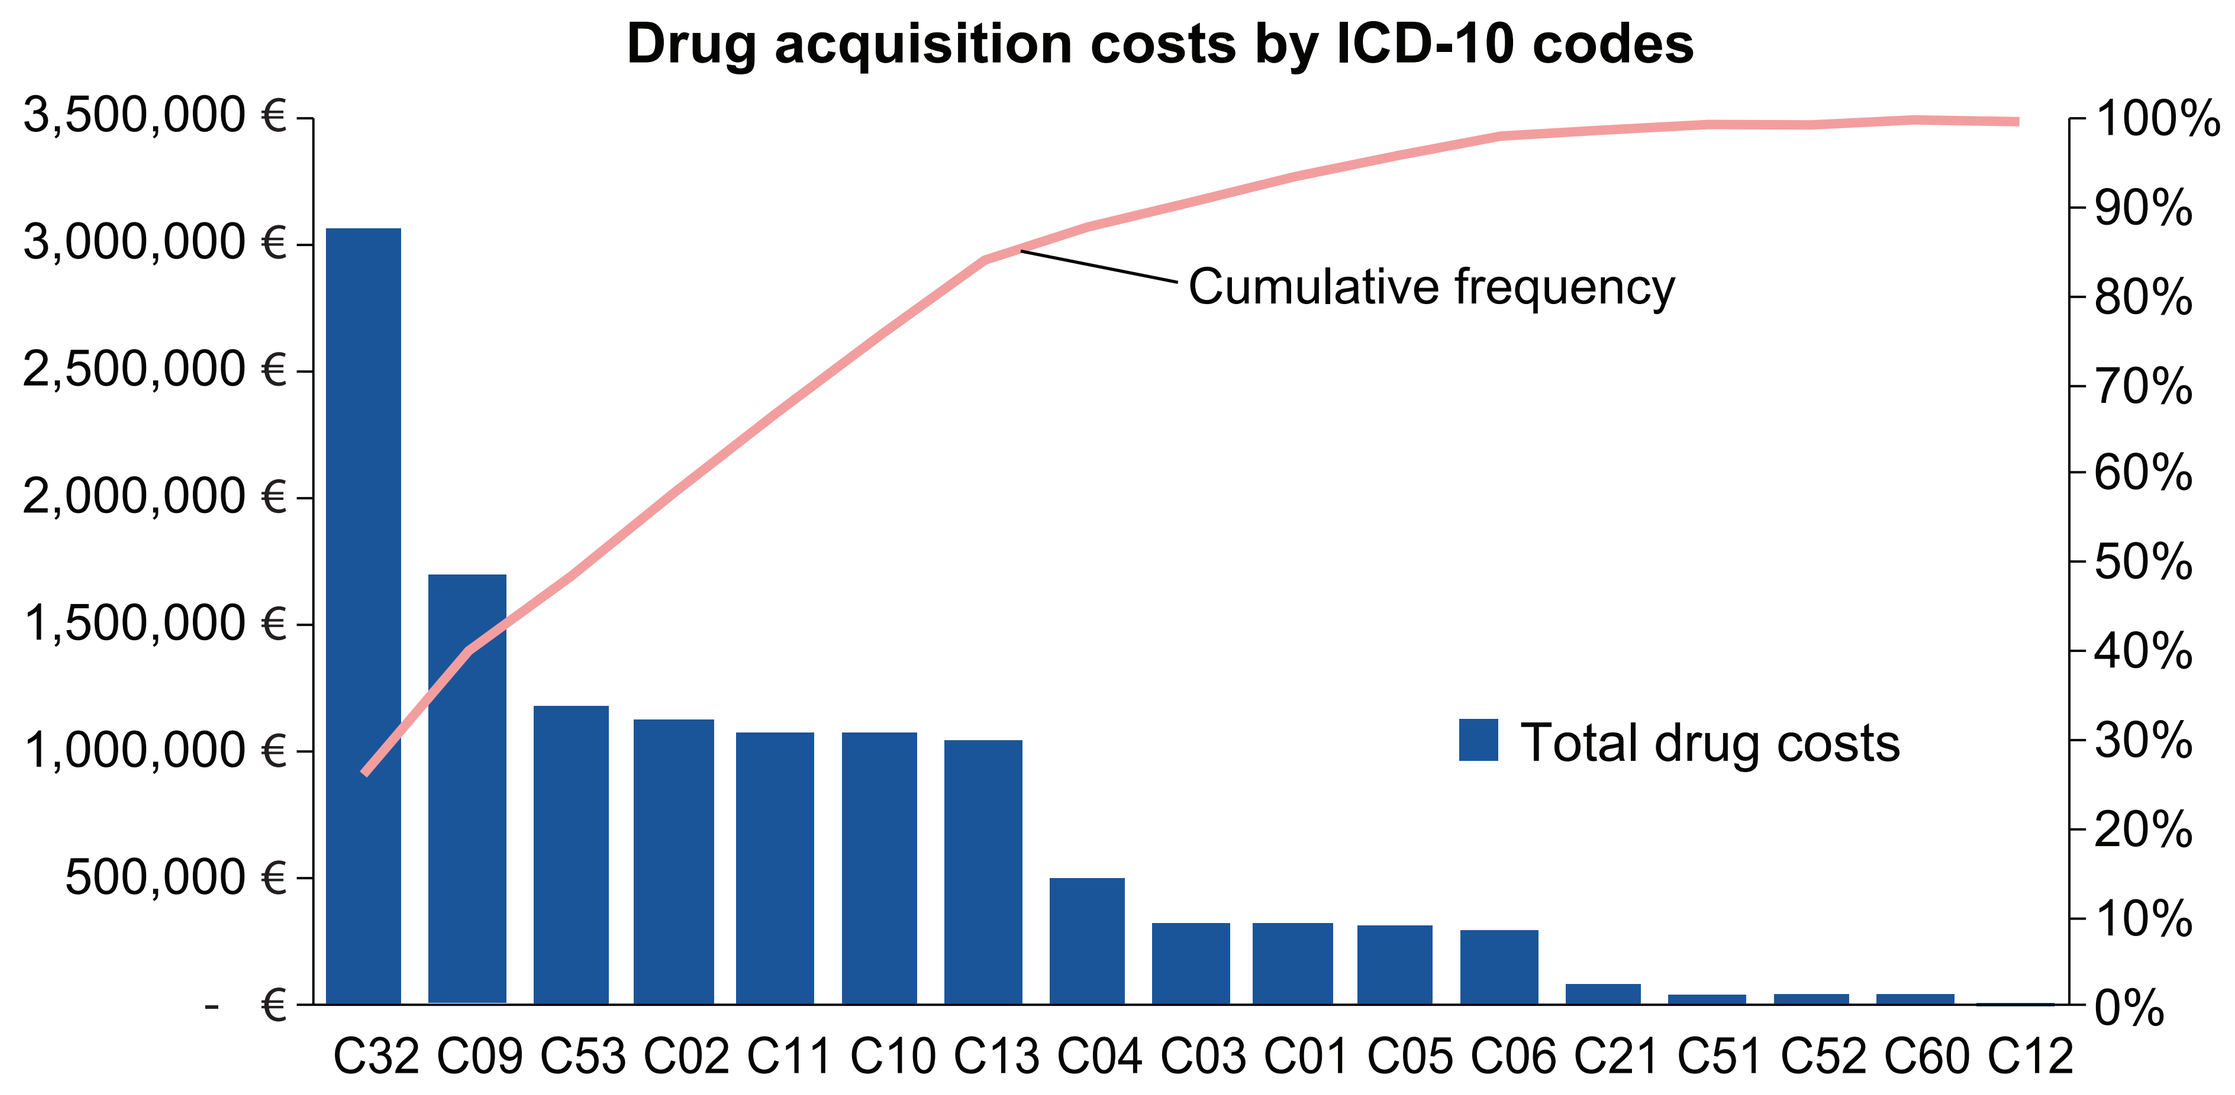
Drug acquisition costs by ICD-10 codes
3,500,000 €
3,000,000 €
2,500,000 €
2,000,000 €
1,500,000 €
1,000,000 €
500,000 €
- €
Cumulative frequency
Total drug costs
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
C32 C09 C53 C02 C11 C10 C13 C04 C03 C01 C05 C06 C21 C51 C52 C60 C12

Source: NHIF

Years of life lost

Figure 3. Years of life lost by ICD-10 codes (Pareto chart)

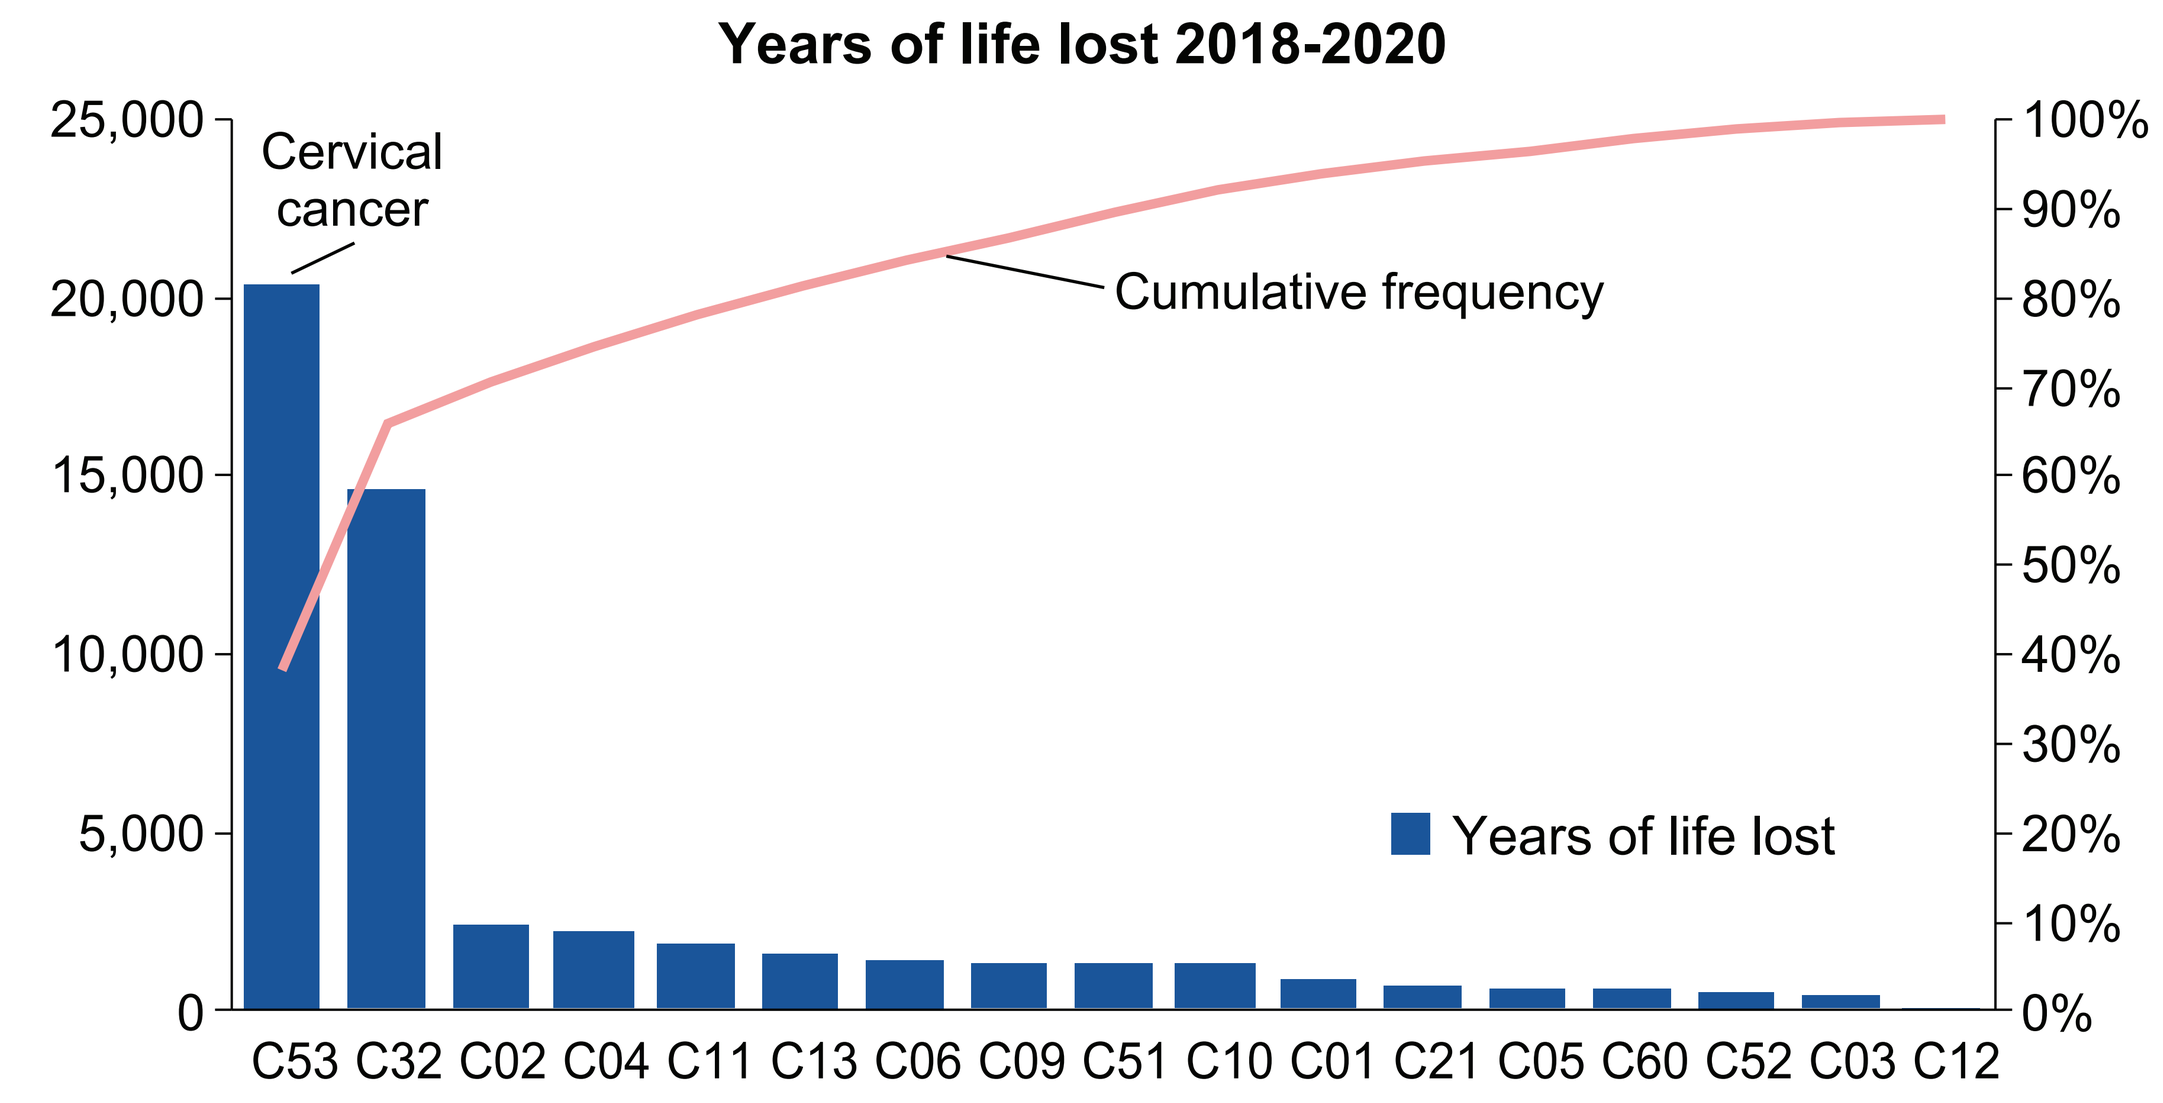
Years of life lost 2018-2020
25,000
20,000
15,000
10,000
5,000
0
Cervical cancer
Cumulative frequency
Years of life lost
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
C53 C32 C02 C04 C11 C13 C06 C09 C51 C10 C01 C21 C05 C60 C52 C03 C12

Figure 4. Total years of life lost

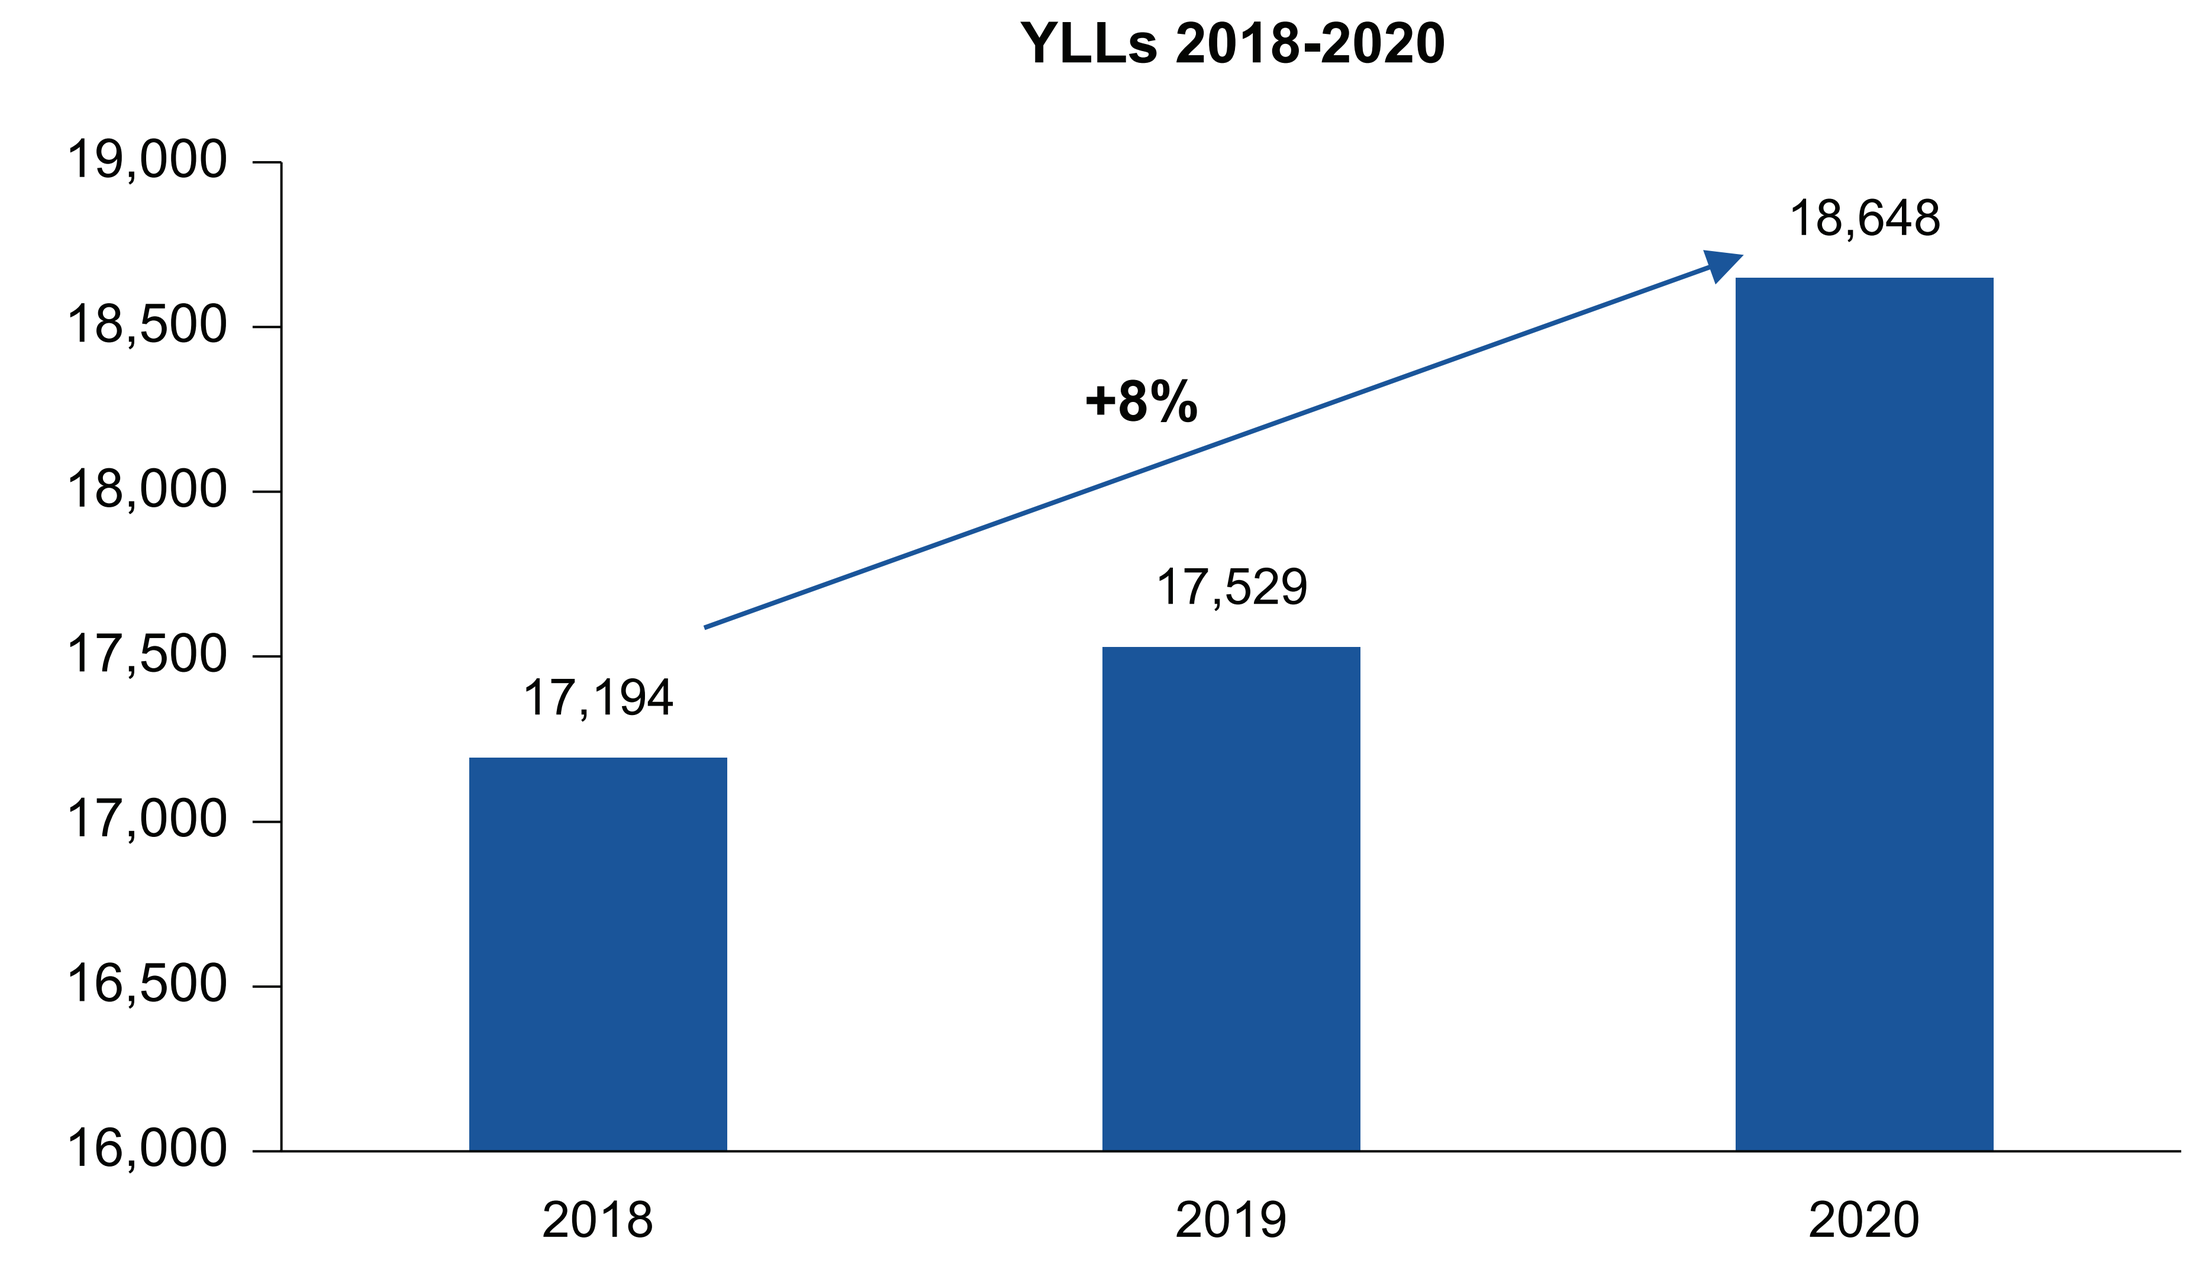
YLLs 2018-2020
19,000
18,500
18,000
17,500
17,000
16,500
16,000
17,194
17,529
18,648
+8%
2018
2019
2020

Source: NSI

Figure 5. Years of working life lost by ICD-10 codes (Pareto chart)

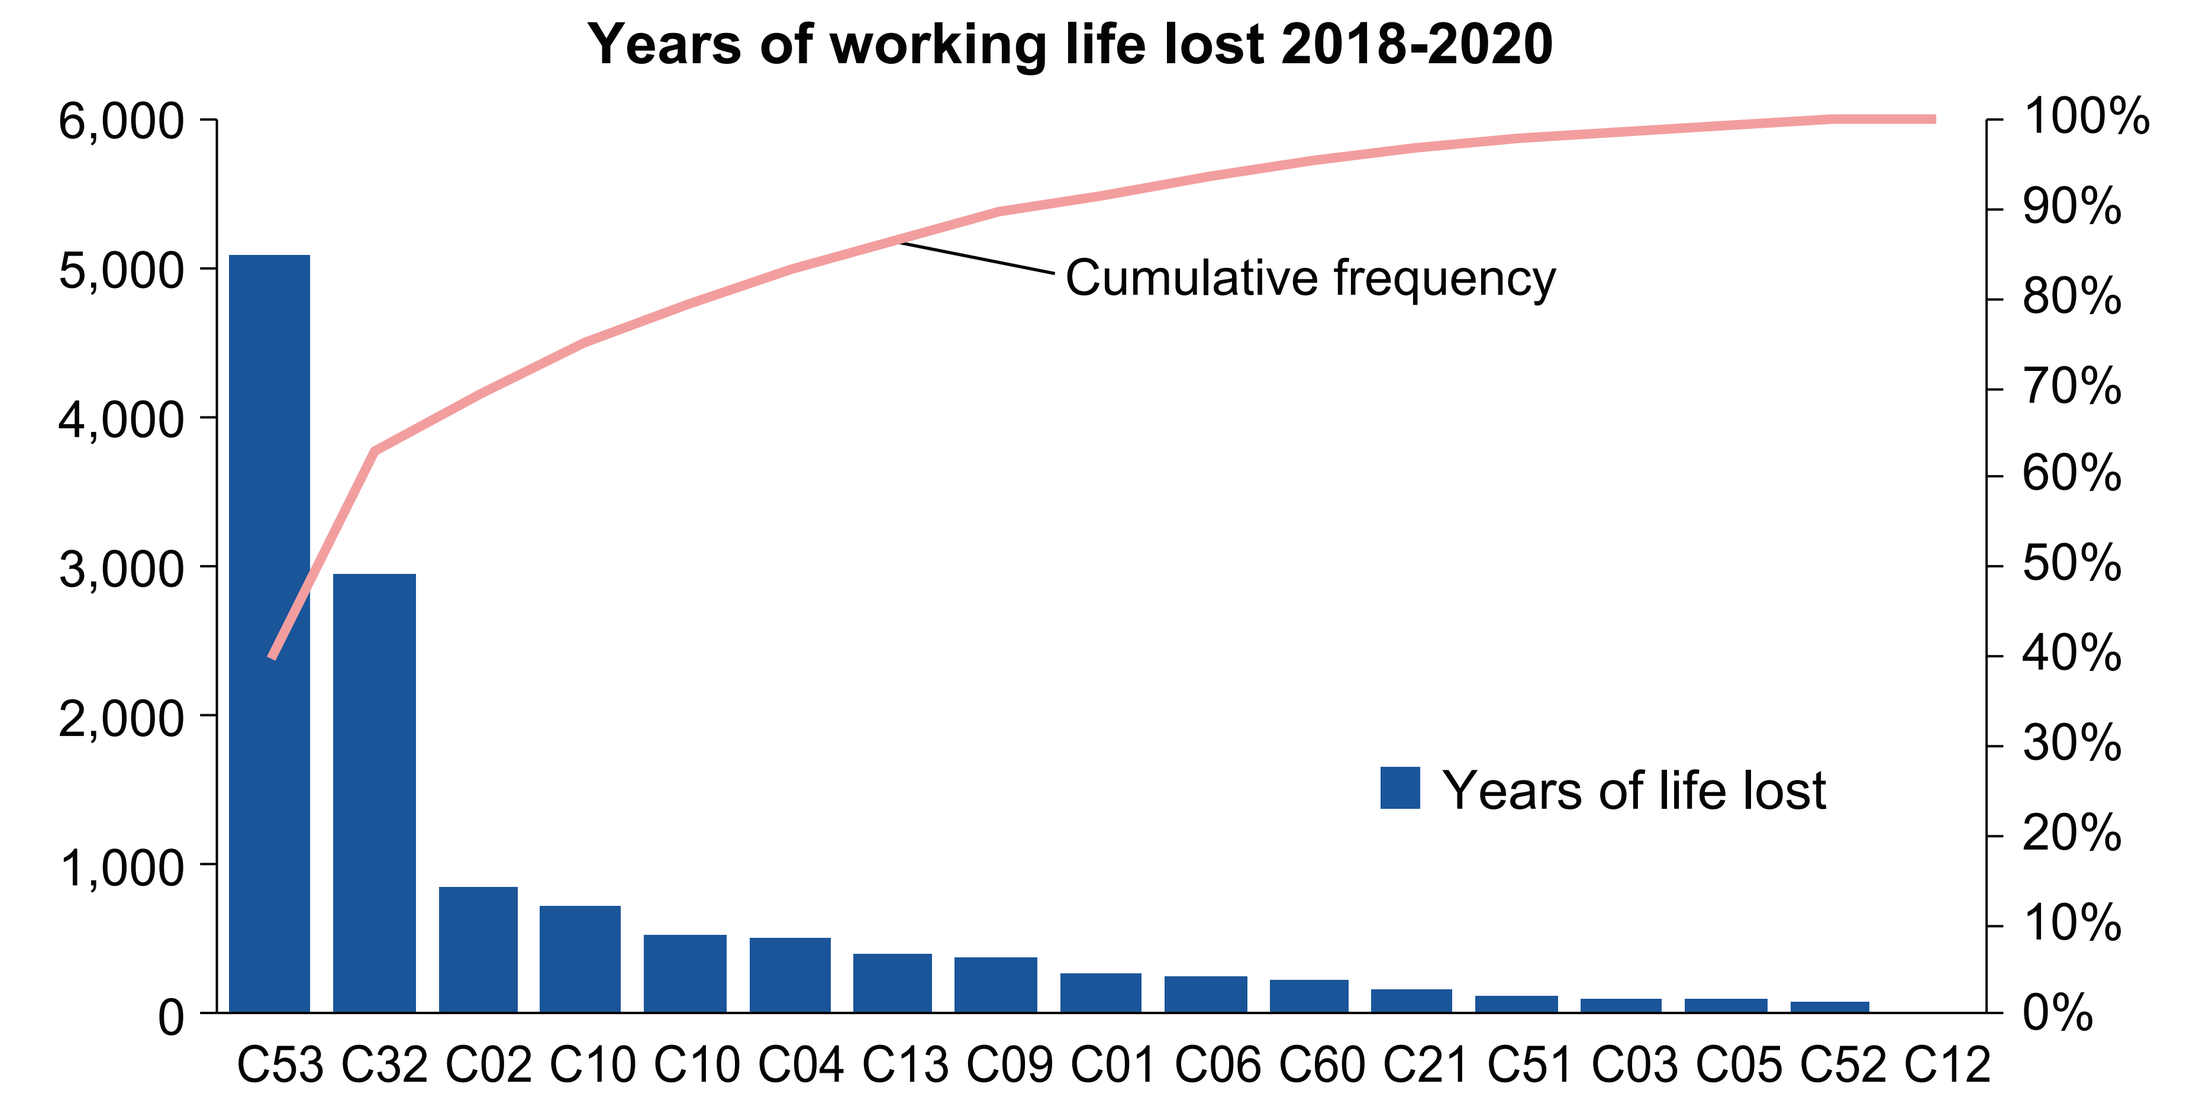
Years of working life lost 2018-2020
6,000
5,000
4,000
3,000
2,000
1,000
0
Cumulative frequency
Years of life lost
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
C53 C32 C02 C10 C10 C04 C13 C09 C01 C06 C60 C21 C51 C03 C05 C52 C12

Source: NSI

Figure 6. Total years of working life lost

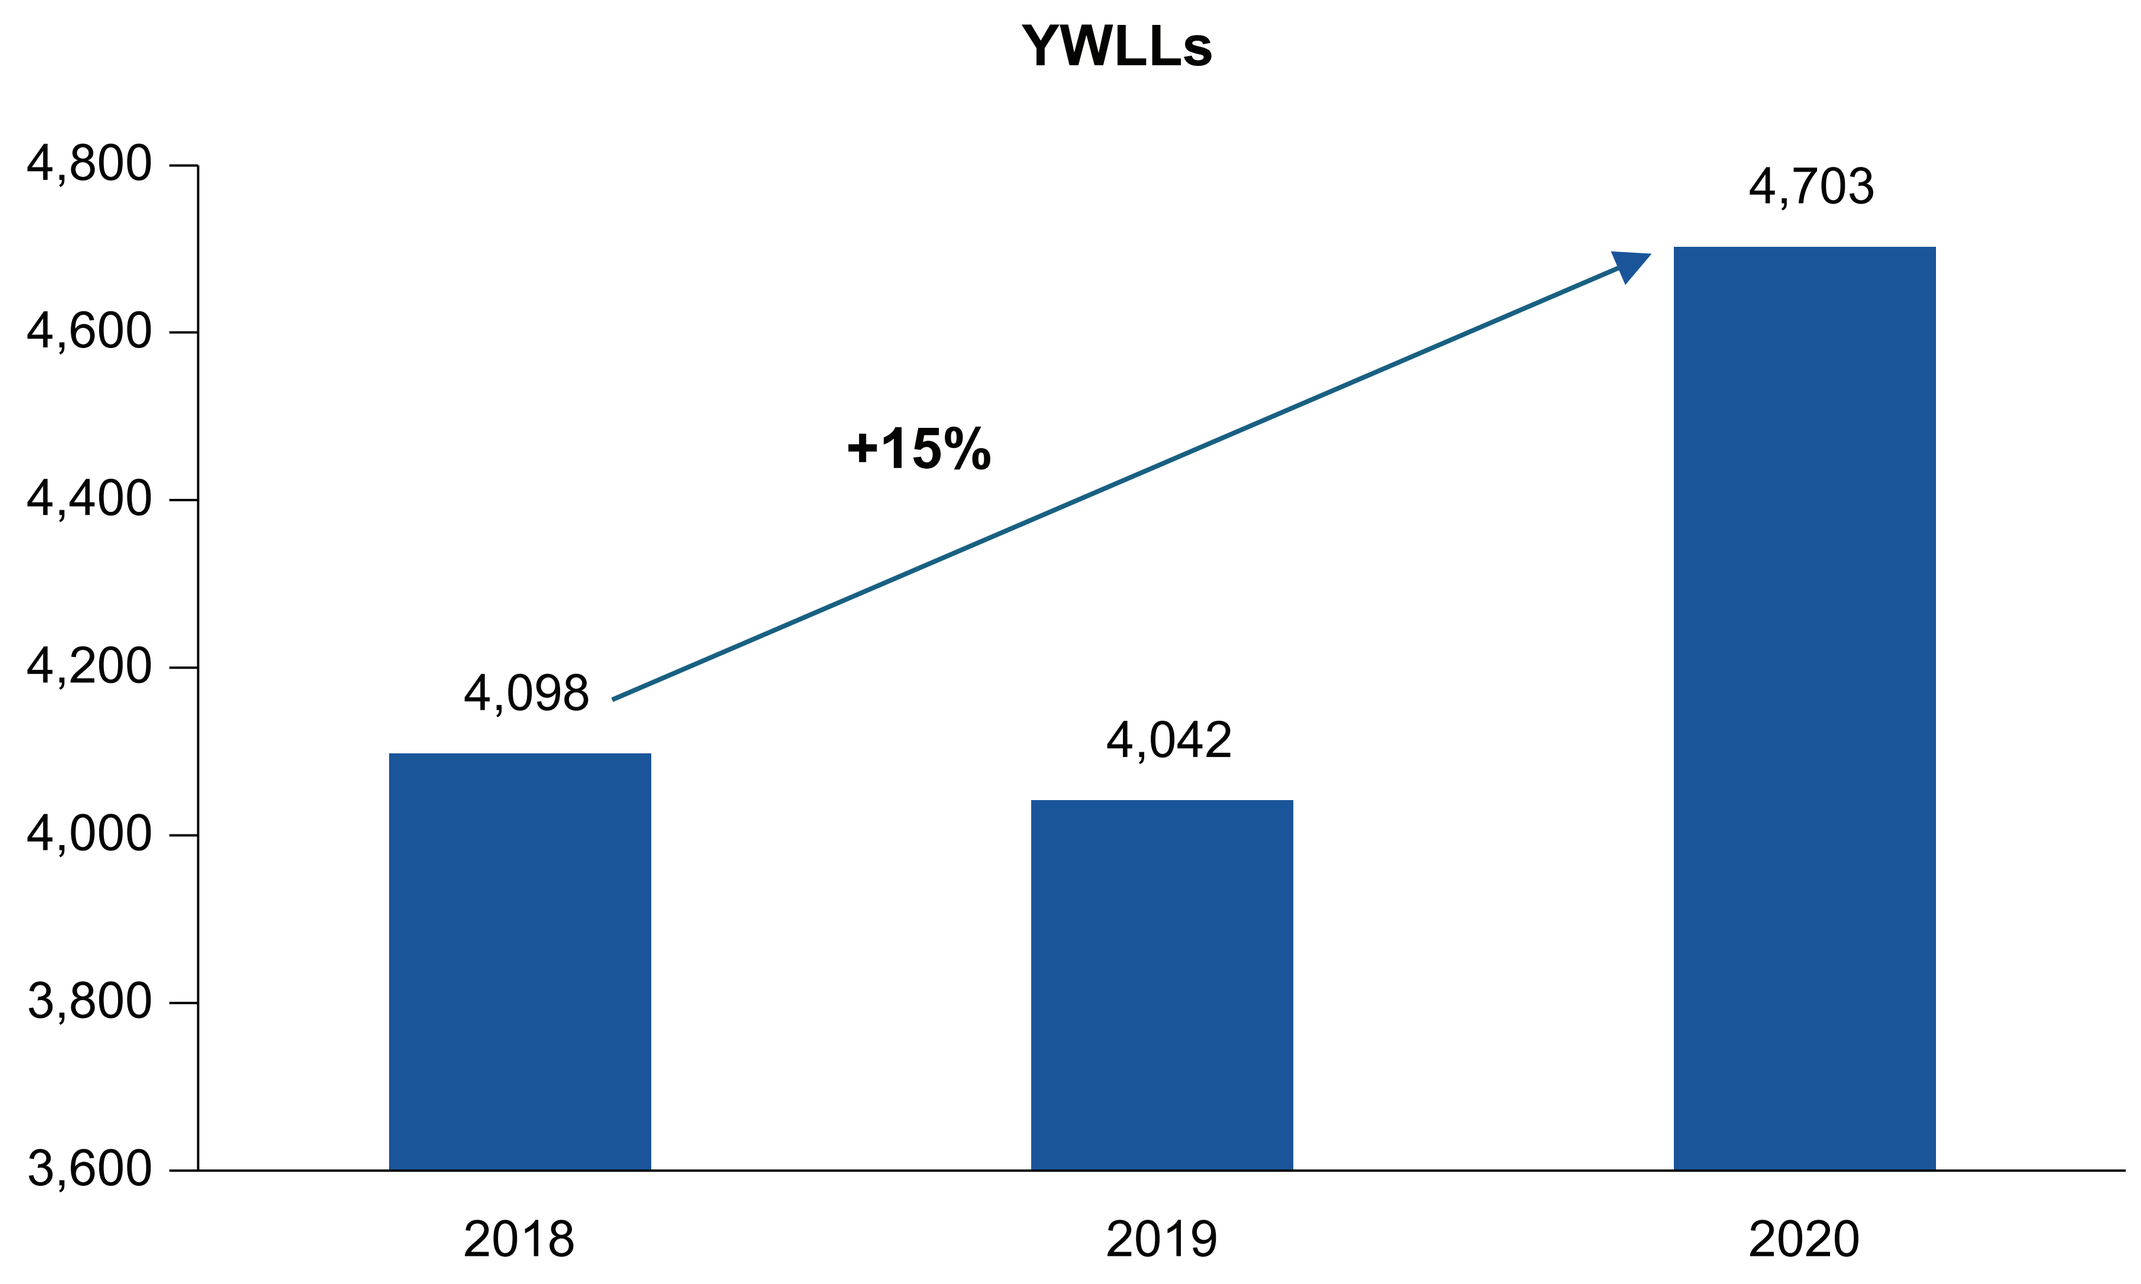
YWLLs
4,800
4,600
4,400
4,200
4,000
3,800
3,600
4,098
4,042
4,703
+15%
2018
2019
2020

Indirect costs due to productivity loss

Table 2. Indirect costs due to productivity loss

	2018	2019	2020
N of all deaths due to the investigated malignancies (all ages)	1,053	1,069	1,115
N of deaths male	620	606	601
N of deaths female	433	463	514
N of deaths up to 64 years of age (males)	217	273	272
N of deaths up to 61 years of age (females)	170	173	222
Years of life lost (male)	9,317	8,760	8,164
Years of life lost (female)	8,252	8,769	10,484
Mean YLL per male	15.03	14.46	13.58
Mean YLL per female	19.06	18.94	20.40
Years of working life lost (male)	2,179	2,136	2,311
Years of working life lost (female)	1,919	1,906	2,392
Mean years of working life lost per male	10.04	7.82	8.50
Mean years of working life lost per female	11.29	11.02	10.77
GDP per employed– current prices (EUR)*	15,965 €	17,421 €	17,768 €
Lost GDP (not produced GDP), EUR per year	6,178,545 €	7,769,594 €	8,777,309 €

Limitations

- Data on costs are provided by the Bulgarian National Health Insurance Fund which provides data from the public sector only. The results do not include out-of-pocket patients' costs for outpatient visits, diagnostic tests, and follow-ups which are suspected to be considerable. This makes our estimates conservative as the true economic burden of cervical, vulvar, vaginal, anal, penile, and H&N cancers in Bulgaria is expected to be even higher
- It is impossible to measure the effect of COVID-19 on cervical, vaginal, vulvar, penile, anal, and H&N cancer–related deaths in 2020, so it may be suspected that the years of life and productivity lost in 2020 are overestimated
- The above analyses were performed considering the fact that it is not possible to estimate with high certainty the HPV-attributable proportion of each of the health outcomes we assessed in the current study

Conclusions

- The calculated economic burden of HPV-related malignancies in Bulgaria from 2018 to 2020 amounts to 51.3 million euros
- The economic burden of vaginal, vulvar, penile, anal, and H&N cancers in Bulgaria is considerable and primarily driven by medicines and radiotherapy costs
- For the study period, a total 53,371 years of life were lost (17,194 in 2018, 17,529 in 2019, and 18,648 in 2020)
- These findings indicate that there is a need for nationwide actions to reduce the ongoing consequences of cancer, including neoplasms related to HPV. At least, additional resources should be allocated to areas where an approved vaccine approach is accessible, as it provides protection against HPV-related diseases like cervical, vaginal, vulvar, and anal malignancies

Funding

This study was funded by MSD Bulgaria.

Ethical approval

The current study has obtained approval from the Ethics Committee for Clinical Trials (Ministry of Health, Republic of Bulgaria) as a noninterventional cost of illness study (notification letter # ECCT-5569/date: 13.12.2023).

Contact information

Assoc. Prof. Hristina Lebanova, PhD

Medical University – Pleven, Bulgaria

References

1. Winer RL, et al. In Cervical Cancer: From Etiology to Prevention. 2004: 143-187.
2. Liu YQ, et al. *Int J Clin Exp Med.* 2015;8:16007-16014.
3. Pešut E, et al. *Viruses.* 2021;13(11):2234. doi:10.3390/v13112234
4. Préaud E, et al. *J Med Econ.* 2013;16:763-776. doi:10.3111/13696998.2013.79.3691
5. Chesson HW, et al. *Vaccine.* 2012;30:6016-6019. doi:10.1016/j.vaccine.2012.07.056
6. Borget I, et al. *Vaccine.* 2011;29:5245-5249. doi:10.1016/j.vaccine.2011.05.018
7. Ki M, et al. *Vaccine.* 2018;36:4633-4640. doi:10.1016/j.vaccine.2018.06.046
8. Baio G, et al. *PLoS One.* 2012;7:e49699. doi:10.1371/journal.pone.0049699
9. Abramowitz L, et al. *PLoS One.* 2018;13:1-19. doi:10.1371/journal.pone.0202564
10. Saxena K, et al. *J Med Econ.* 2022;25:299-308. doi:10.1080/13696998.2022.2041855
11. Lebanova H, et al. *Int J Environ Res Public Health.* 2023;20(3):2746. doi:10.3390/ijerph20032746
12. Sabale U, et al. *J Med Econ.* 2024;27:1-8. doi:10.1080/13696998.2024.2341572

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the Congress or the author of this poster.

https://bit.ly/4dr8Uzl

Presented at ISPOR Europe; Barcelona, Spain; 17-20 November 2024.

Copyright © 2024 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.