EE393

Economic Impact of COVID-19 Vaccination Programs on Healthcare Workers: A Comparative Model-Based Analysis in the US, UK, Japan, and the Netherlands

Orsolya Balogh,[1] Nicolas Van de Velde,[2] Yuriko Hagiwara,[3] Tjalke Westra,[4] Keya Joshi,[2] Cornelis Boersma,[5] Maarten Postma,[5] Ataru Igarashi,[6] Ekkehard Beck[2]*

[1]Moderna, Inc., London, UK; [2]Moderna, Inc., Boston, MA, USA; [3]Moderna, Inc., Tokyo, Japan; [4]Moderna, Inc., Amsterdam, Netherlands; [5]University of Groningen, Groningen, Netherlands; [6]University of Tokyo, Tokyo, Japan

*Presenting author.

BACKGROUND

- COVID-19 and seasonal influenza cause substantial public health and economic burden, particularly in healthcare, where absenteeism and reduced workforce productivity can severely disrupt essential services
- Vaccinating healthcare workers (HCWs) can minimize workforce disruption and ensure healthcare system resilience during respiratory disease outbreaks
- Vaccination coverage rates (VCRs) among HCWs remain suboptimal in many countries for both influenza and COVID-19
- COVID-19 disease burden is higher than influenza[1,2]; however, while HCW seasonal influenza vaccination is often mandated or recommended, similar policies for COVID-19 are under consideration by National Immunization Technical Advisory Groups and vary across countries
- Employer-led vaccination programs can effectively reduce absenteeism and minimize productivity loss by lowering the incidence of infections and long COVID; additionally, vaccinated healthcare workers are more likely to remain at work during periods of heightened winter demand, thereby supporting a resilient healthcare system

OBJECTIVE

- This study estimated the disease and economic impact of increasing the COVID-19 VCR among HCWs in the United States (US), United Kingdom (UK), Japan, and the Netherlands
 - Primary objective: impact on COVID-19 episodes, absenteeism, and productivity loss of increasing the VCR to 75% (the World Health Organization target VCR for influenza among HCWs[3]), versus the current VCR

METHODS

Model Overview

- An existing workplace vaccination model[4] was adapted to the HCW setting to assess COVID-19 HCW vaccination in the US, UK, Japan, and the Netherlands (**Figure 1**)

Figure 1. Workplace COVID-19 vaccination model structure

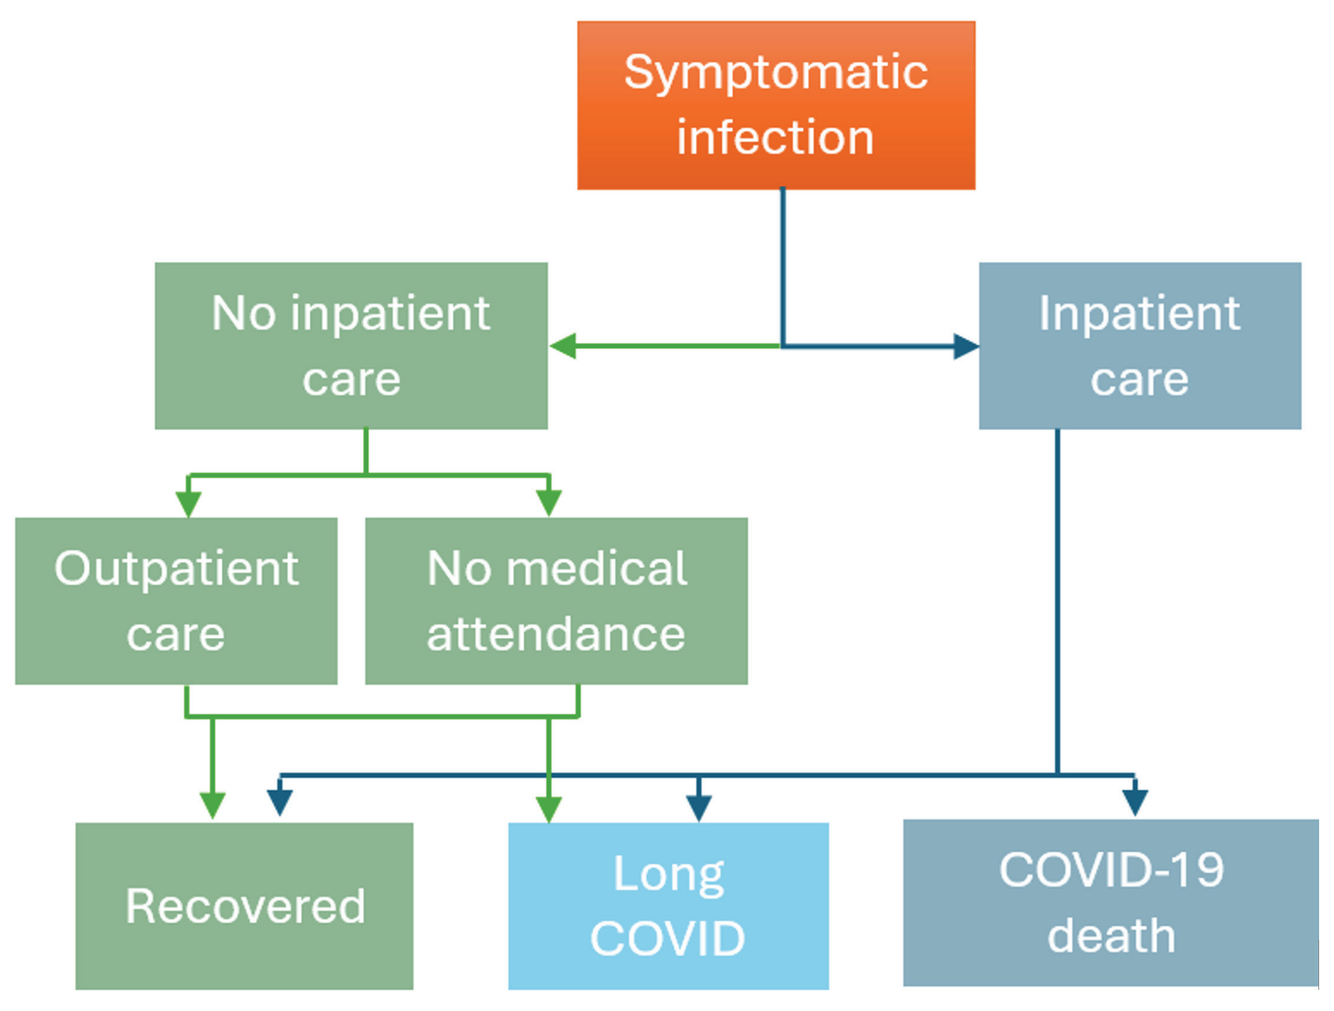
Symptomatic infection
No inpatient care
Inpatient care
Outpatient care
No medical attendance
Recovered
Long COVID
COVID-19 death

- Inputs: country-specific population data, COVID-19 burden, healthcare resource use and probabilities from economic studies adapted to working age population (18-64 years) (**Table 1**)
 - Predicted probability of infection in unvaccinated population (2023-2024 season) from economic studies[5-7]
 - Observed COVID-19 VCR (2023-2024 season) was used
- Disease outcomes: symptomatic COVID-19 infections, COVID-19 outpatient visits, COVID-19 hospitalisations, COVID-19 deaths, long COVID (with only severe persistent cases assumed to affect work and result in absenteeism)
- Economic outcomes: absenteeism, productivity loss, and direct medical costs from employer and employee perspectives
- Vaccine effectiveness (VE) estimates: US nationwide retrospective real-world evidence study[8]
 - Baseline starting VE: 60.2% (hospitalisation), 33.1% (symptomatic infection)[8]
 - Monthly VE waning: 1.4% (hospitalisation), 4.8% (infection)[9]
- Scenario analyses: increased risk of a) infection and b) long COVID in HCWs; c) impact beyond wages of hospital staff absence on productivity loss for healthcare employers/systems
 - a) 2.5-fold increase in the risk of COVID-19 in HCWs versus the general population, based on UK data[10]
 - b) 2.6-fold increase in long COVID risk considering the increased risk of long COVID in patients with ≥3 COVID-19 infections[11]
 - c) wage multiplier of 1.4 for registered nurses in hospitals to account for productivity losses beyond wage[12]

Table 1. Key model inputs

	US	UK	Japan	Netherlands
Number of HCWs (N, millions)[13-16]	22.3	1.5	9.4	1.2
Infection rate, unvaccinated population (%)	20.7	37.5	27.9	12.9
COVID-19 VCR 2023/2024 (%)	17	30	32[a]	9
Severe long COVID with work time loss (%)	2.1	3.7	4.1	2.1
Productivity loss per day ($)[7,17-18]	262.9	171.5	123.4	210.2

HCW, healthcare worker; VCR, vaccine coverage rate.

[a]Based on projections and market research, we assumed 32% VCR for HCWs in Japan.

RESULTS

Disease Impact

- Increased vaccination coverage among HCWs is estimated to have substantial health benefits, preventing an additional 480,202 COVID-19 cases in the US; 59,257 in the UK; 267,157 in Japan; and 24,035 in the Netherlands compared to the current VCR (**Figure 2**)
 - This corresponds to approximately a 2.6-fold increase in the US, 1.5-fold in the UK, 1.4-fold in Japan, and 7.3-fold in the Netherlands

Figure 2. COVID-19 episodes averted with target vs current VCR

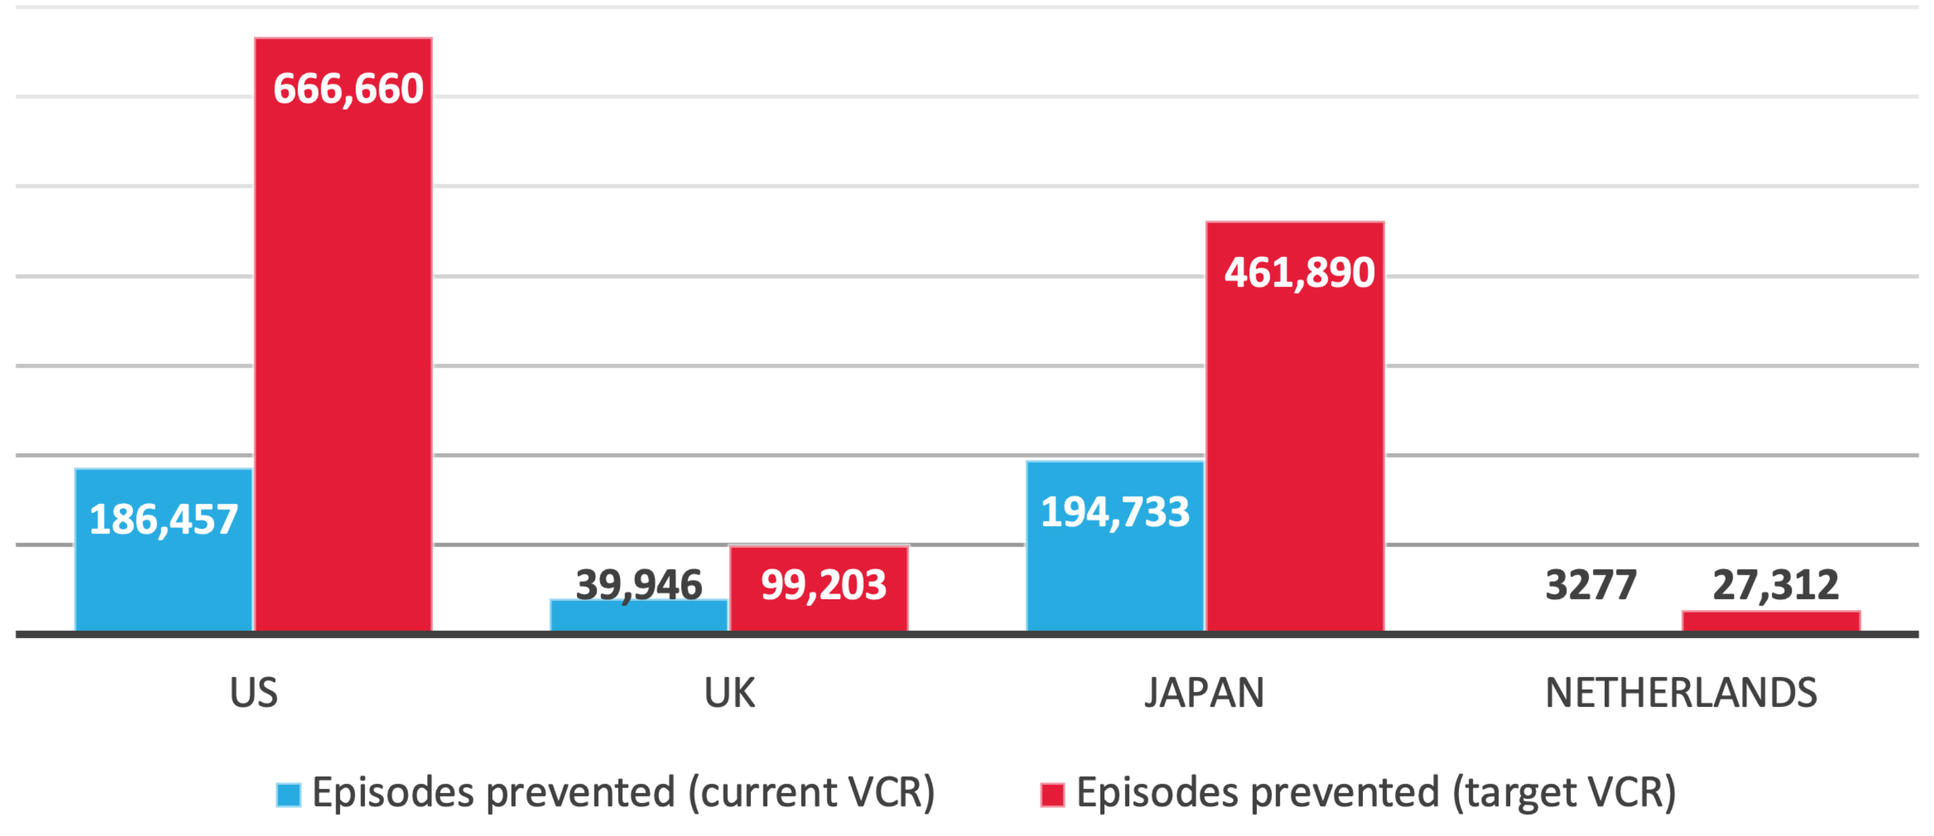
666,660
186,457
39,946
99,203
194,733
461,890
3277
27,312
US
UK
JAPAN
NETHERLANDS
Episodes prevented (current VCR)
Episodes prevented (target VCR)

Economic Impact

- Increased COVID-19 vaccination coverage among HCWs is estimated to provide substantial cost savings from reduced COVID-19 absenteeism (**Figure 3**) and improved workforce productivity in HCWs (**Figure 4**)

Figure 3. Absent days averted by cause per 1000 HCWs (target vs current VCR)

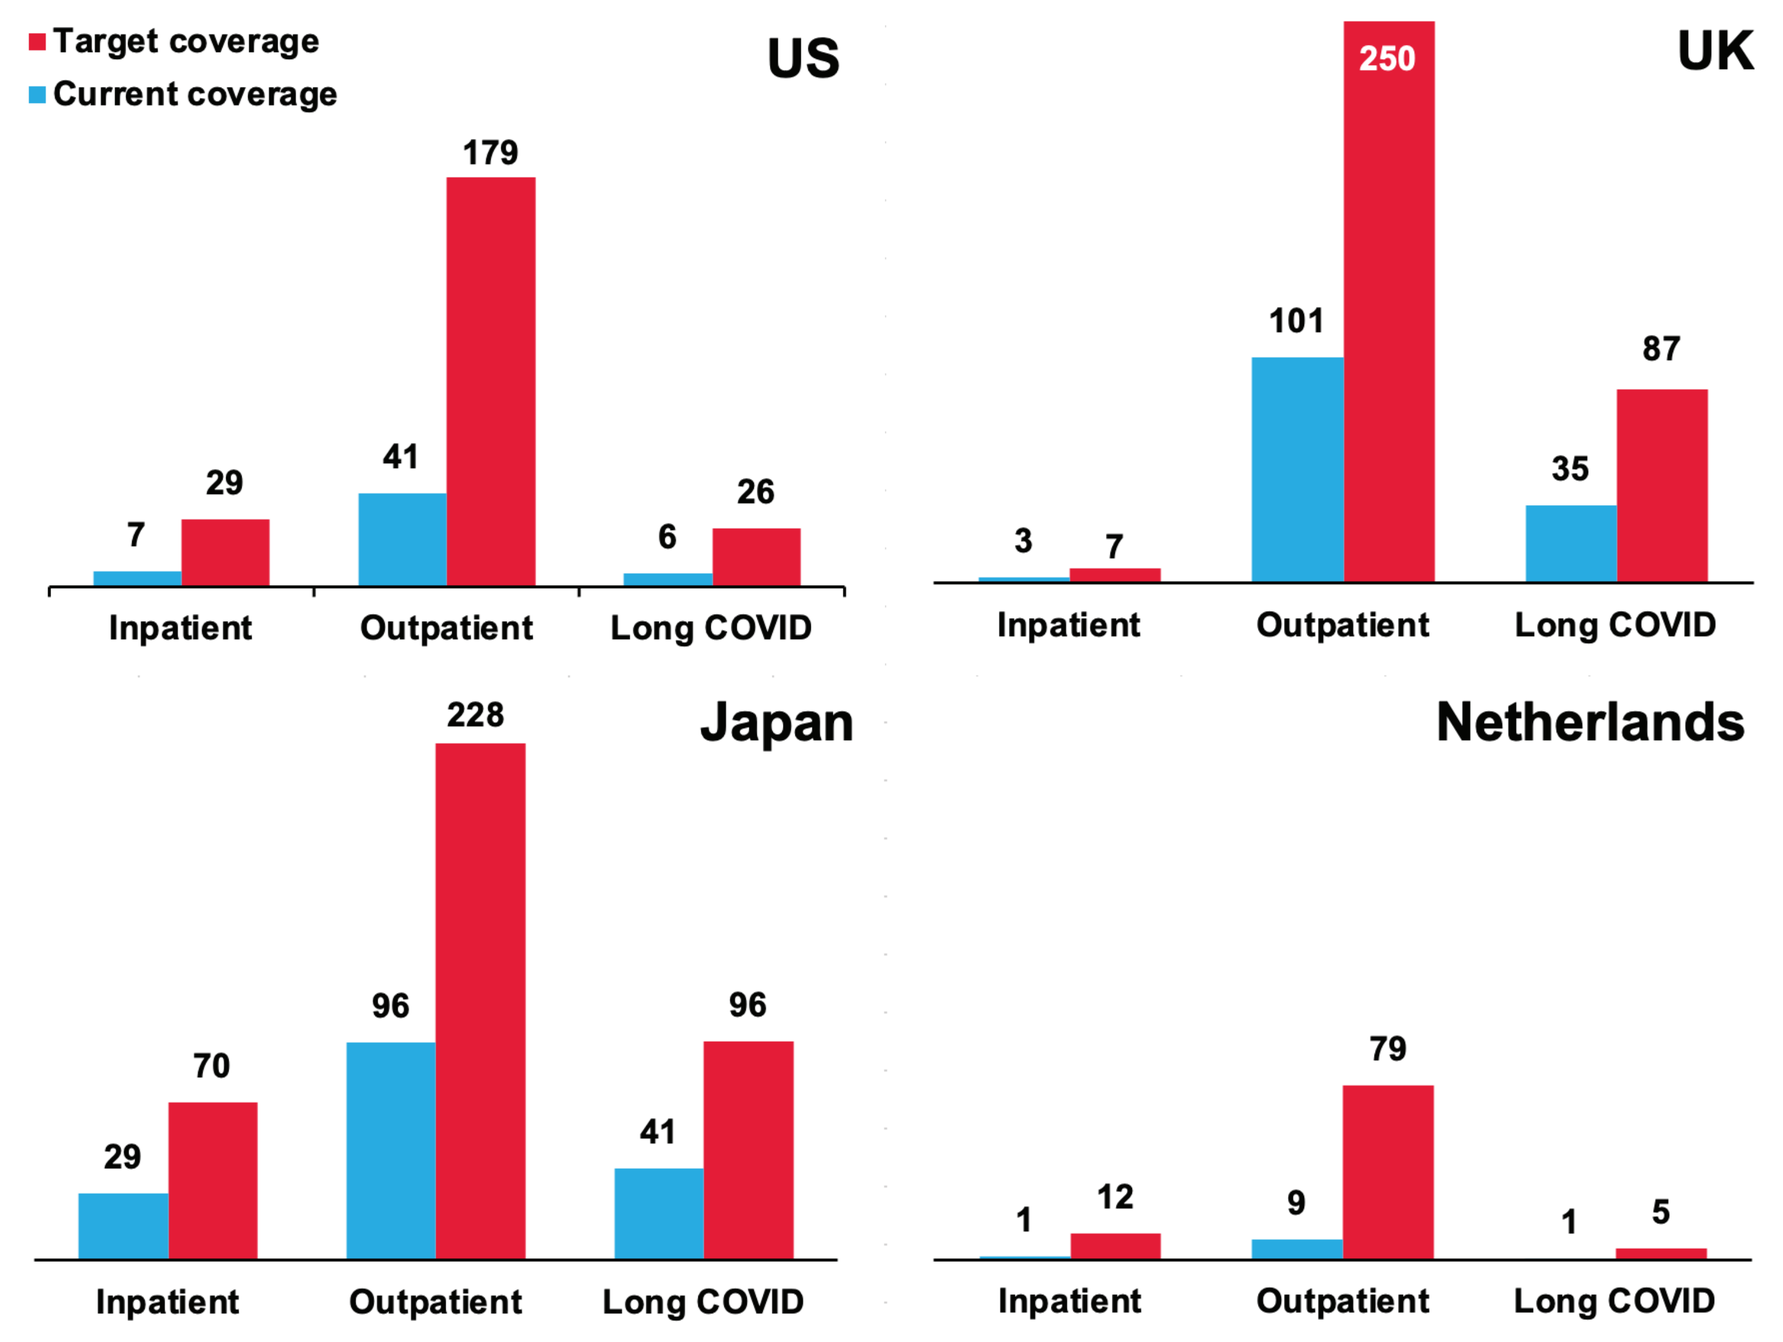
Target coverage
Current coverage
US
7
29
41
179
6
26
Inpatient
Outpatient
Long COVID
UK
3
7
101
250
35
87
Inpatient
Outpatient
Long COVID
Japan
29
70
96
228
41
96
Inpatient
Outpatient
Long COVID
Netherlands
1
12
9
79
1
5
Inpatient
Outpatient
Long COVID

- Increasing the COVID-19 VCR in HCWs produced potential savings of $1 billion (US), $47 million (UK), $280 million (Japan), and $21 million (the Netherlands) in direct medical and indirect (lost productivity) costs

Figure 4. Lost productivity costs averted ($) in the base case and scenario analyses (target vs current VCR): Employer and employee perspectives

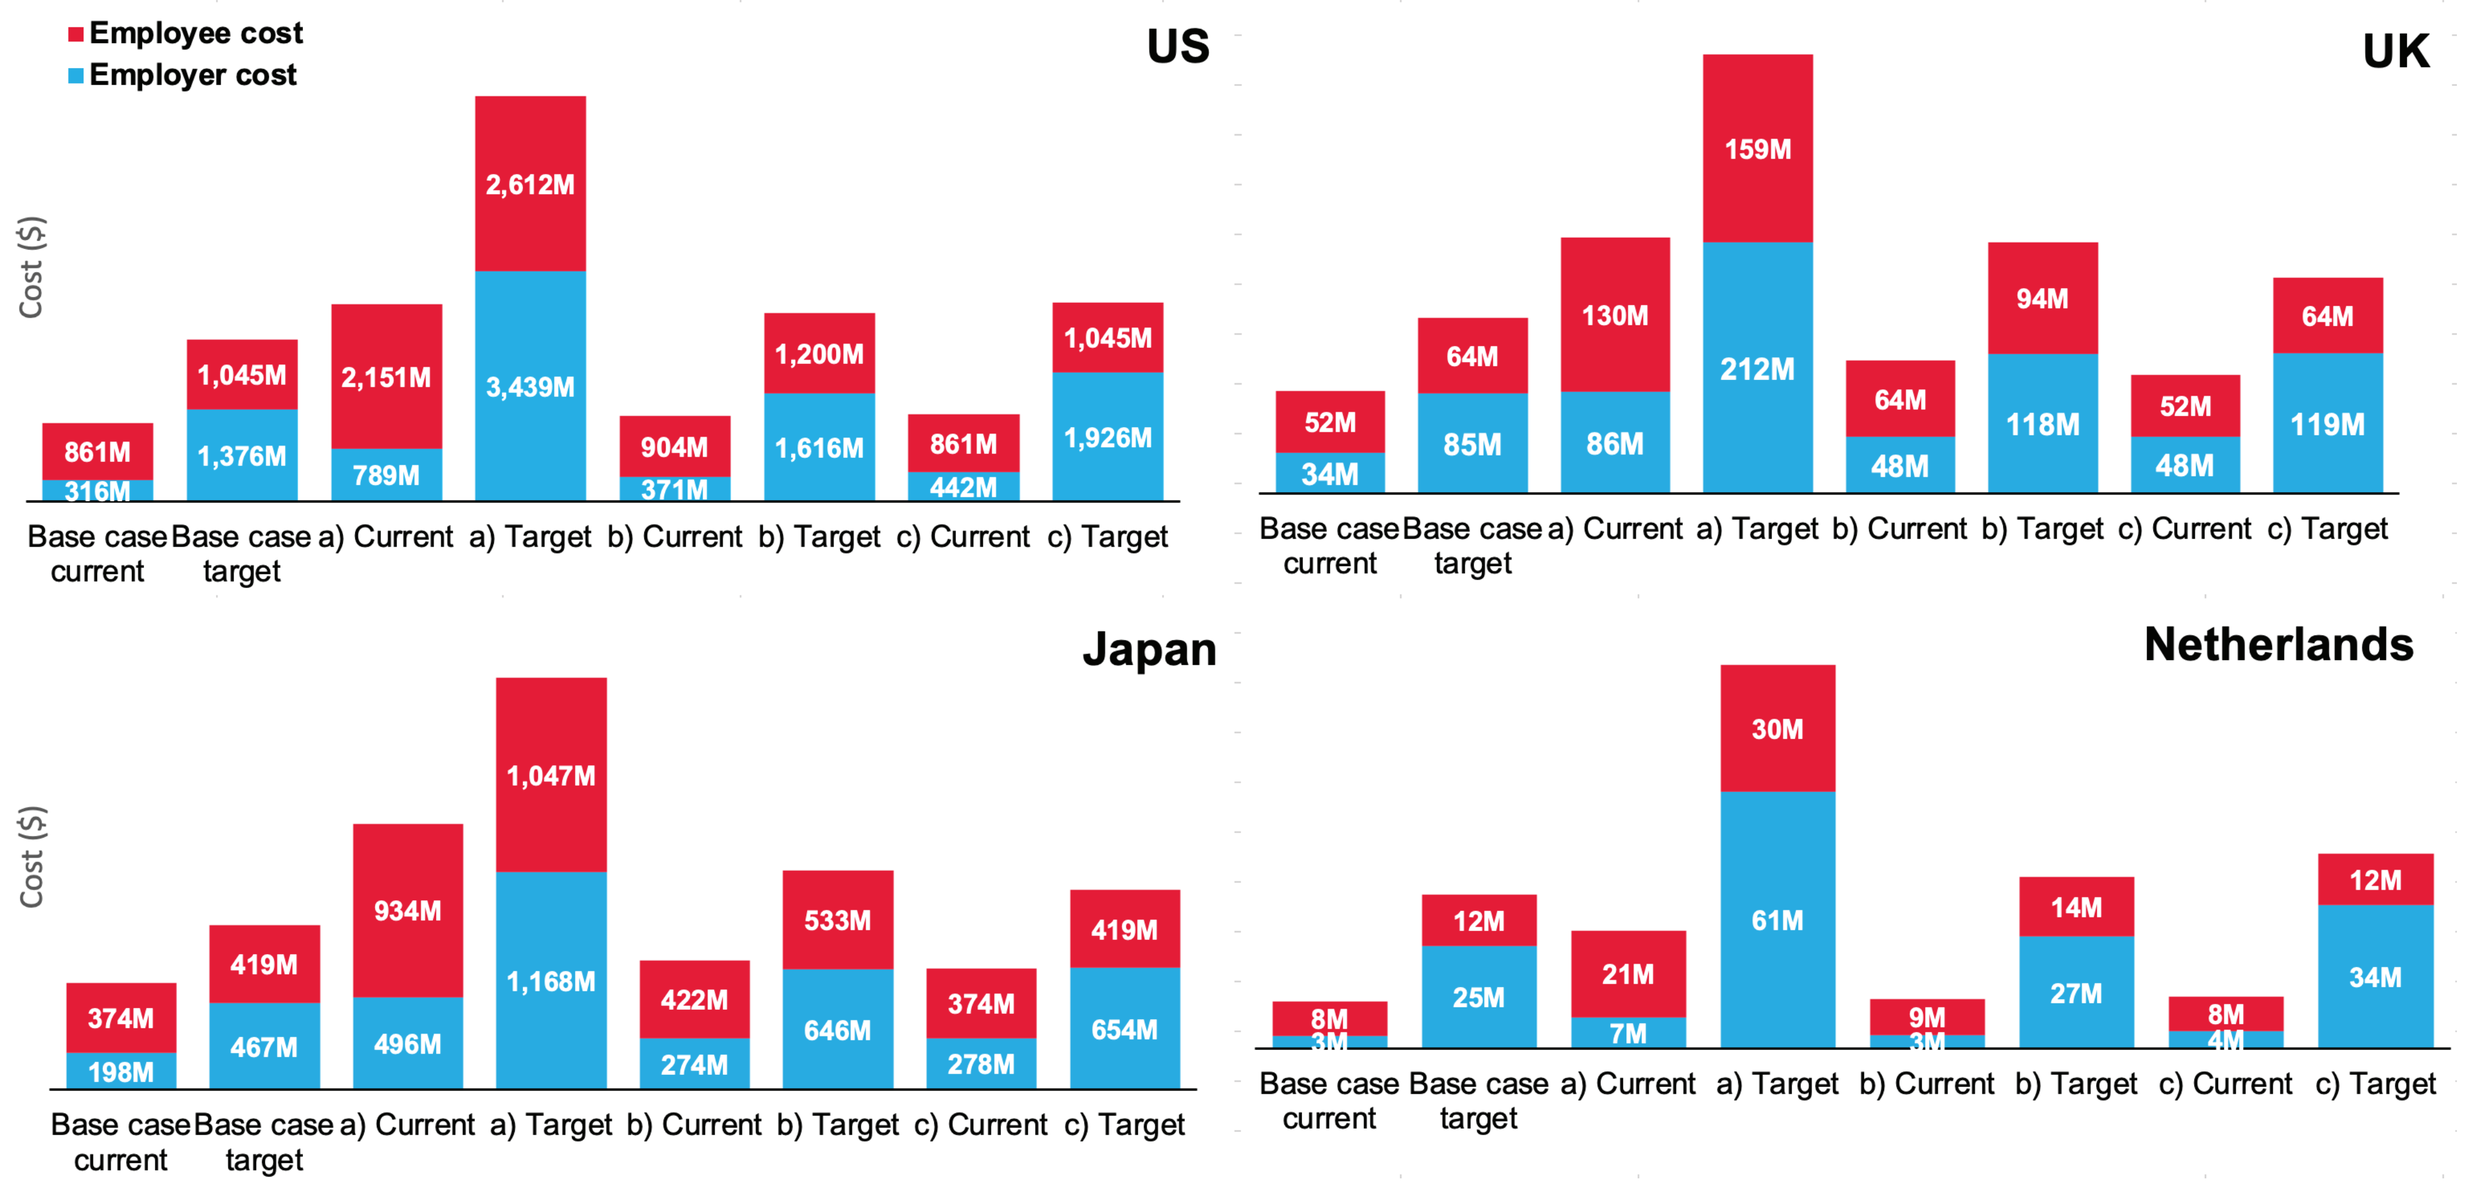
Employee cost
Employer cost
US
Cost ($)
861M
316M
1,045M
1,376M
2,151M
789M
2,612M
3,439M
904M
371M
1,200M
1,616M
861M
442M
1,045M
1,926M
Base case current
Base case target
a) Current
a) Target
b) Current
b) Target
c) Current
c) Target
UK
52M
34M
64M
85M
130M
86M
159M
212M
64M
48M
94M
118M
52M
48M
64M
119M
Japan
374M
198M
419M
467M
934M
496M
1,047M
1,168M
422M
274M
533M
646M
374M
278M
419M
654M
Netherlands
8M
3M
12M
25M
21M
7M
30M
61M
9M
3M
14M
27M
8M
4M
12M
34M

Scenarios:

a) 2.5-fold increase in the risk of COVID-19 in HCWs versus the general population

b) 2.6-fold increase in long COVID risk considering the increased risk of long COVID in patients with ≥3 COVID-19 infections

c) Wage multiplier of 1.4 for registered nurses in hospitals to account for productivity losses beyond wage

VCR, vaccine coverage rate.

CONCLUSIONS

- Substantial health and economic benefits were predicted with increased COVID-19 vaccination coverage due to reductions in absenteeism and productivity losses among HCWs[19]
- High vaccination coverage among HCWs is crucial to ensure the continued operation of healthcare systems during periods of increased respiratory disease activity
- This study aims to provide evidence to inform formal evaluations in countries where COVID-19 vaccination for HCWs is either routinely recommended or not yet adopted as standard practice, supporting data-driven policy decisions regarding vaccination programs for HCWs
- During high absenteeism periods, such as COVID-19 peaks, the burden on hospital bed availability increases significantly. Unscheduled absences due to illness in HCWs are strongly correlated with a reduction in hospital staffing capacity, impacting the ability to manage patient loads effectively and secure bed availability. Thus, HCW vaccination programs play a critical role in reducing absenteeism and in maintaining hospital operations during peak seasons
- Limitations: the model used assumptions on epidemiology, vaccine effectiveness for HCWs, and exposure risk, which may vary across populations and healthcare settings. In the scenario analysis, different multipliers may apply to various types of HCWs, but these distinctions were not specifically addressed. The model did not account for permanent workforce exits due to long COVID, potentially underestimating long-term effects on the workforce. Additionally, the model did not quantify transmission within hospital settings. The analysis considered the net benefit of reducing disease burden but did not include direct costs of vaccination, which may influence the overall estimates
- Targeted vaccination strategies in HCWs remain a key component to maintain the functionality and resilience of healthcare systems, allowing them to operate effectively
- This study can also support recommendations to extend vaccination efforts to other high-risk groups, helping to enhance system-wide resilience and mitigate the broader economic impacts of illness

References

1. Xie et al. (2024) DOI: 10.1016/S1473-3099(23)00684-9
2. Kopel et al. (2024) DOI: 10.3390/diseases12010016
3. World Health Organization. (https://www.who.int/europe/activities/managing-seasonal-vaccination-policies-and-coverage-in-the-european-region) Accessed 28-10-2024
4. Beck et al. (2024) DOI: 10.20944/preprints202407.1919.v1
5. Kohli et al. (2024) DOI: 10.1101/2024.08.19.24312243
6. Fust et al. (2024) DOI: 10.3390/vaccines12040434
7. Zeevat et al. (2024) DOI: 10.1101/2024.09.26.24314420
8. Kopel et al. (2024) DOI: 10.1101/2024.04.10.24305549
9. Higdon et al. (2022) DOI: 10.1016/S1473-3099(22)00409-1
10. Strain et al. (2021) DOI: 10.1136/bmjopen-2020-042225
11. Mansi et al. (2024) DOI: 10.3390/vaccines12091072
12. Nicholson et al. (2006) DOI: 10.1002/hec.1052
13. Bell et al. (2023) DOI: 10.15585/mmwr.mm7245a5
14. NHS England. (https://digital.nhs.uk/data-and-information/publications/statistical/nhs-workforce-statistics/january-2024) Accessed 28-10-2024
15. Ministry of Health Labor and Welfare Japan. (https://www.mhlw.go.jp/wp/hakusyo/kousei/21/dl/1-01.pdf) Accessed 28-10-2024
16. Organisation for Economic Co-operation and Development. (https://www.oecd.org/netherlands/health-at-a-glance-Netherlands-EN.pdf) Accessed 28-10-2024
17. Working days US. (https://www.workingdays.us/workingdays_holidays_2023.htm) Accessed 28-10-2024
18. ONS Statistics 2023. (https://www.workingdays.us/workingdays_holidays_2023.htm) Accessed 28-10-2024
19. Maltezou et al. (2023) DOI: 10.3390/healthcare11222950

Acknowledgments

Medical writing and editorial assistance for this poster were provided by Anja Varjacic, PhD, of MEDiSTRAVA in accordance with Good Publication Practice (GPP 2022) guidelines, funded by Moderna, Inc., and under the direction of the authors. This study was supported by Moderna, Inc.

Disclosures

OB, NVV, YH, TW, KJ, and EB are employees of Moderna and hold stocks/stock options in the company.

PLAIN LANGUAGE SUMMARY

This study examines how increasing COVID-19 vaccination rates for healthcare workers can improve health outcomes and provide economic benefits in four countries: the US, UK, Japan, and the Netherlands.

By raising vaccination rates to 75% (compared to currently observed rates), fewer healthcare workers are likely to become infected with COVID-19 or develop severe long-term effects, such as long COVID. This helps to reduce the number of sick days, keeping more staff available to deliver patient care.

The study found that in the US, increasing vaccination rates could prevent more than 4 million sick days, resulting in potential savings of $1 billion due to fewer sick days and lower healthcare costs. Similar savings were observed in other countries, with significant reductions in infections, hospitalisations, and long COVID.

Ensuring that healthcare workers are protected enables hospitals to maintain operations more effectively, especially during peak periods like winter, when healthcare demand is highest. Overall, increasing vaccination rates leads to healthier staff and lowers costs for both employers and healthcare systems.

Presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe; 17-20 November 2024; Barcelona, Spain