Cost-Minimization and Budget Impact Analysis of Abiraterone (Tarosyn® and Avitosyn®) as Second Line Treatment for Metastatic Prostate Cancer Resistant to Castration in Mexico

Sánchez y Rodríguez, Tanya Pamela[1], Domínguez-Cruces, Carlos[1], Oliver-Mercado, Ingrid[1], Monteon Batalla, Isabel[1], Paladio-Hernández, José Ángel[2]
[1]Synthon, Tlaquepaque, Jalisco, México, [2]PalaGod Health Supply, Guadalajara, Jalisco, México

EE476

INTRODUCTION

Evidence indicates that treatment with abiraterone, cabazitaxel, or enzalutamide in patients with metastatic castration-resistant prostate cancer (mCRPC), demonstrated an increase in survival by approximately 3-5 months and recommends the use of all three drugs equally.

The objective of this analysis was to estimate the potential reduction in costs and the identification of the net budget impact of the adoption of new Abiraterone (Tarosyn® and Avitosyn®) as 2nd line treatment of mCRPC at the Mexican Social Security Institute (IMSS) in Mexico.

METHODS

A Cost Minimization Analysis (CMA) was conducted along with a 5-year Budget Impact Analysis (BIA).

A 5-year Budget Impact Analysis (BIA) model was developed to estimate the incremental BI for 100 patients in treatment for 2nd line mCRPC at the IMSS with the adoption of new Abiraterone (Tarosyn® and Avitosyn®), compared to abiraterone, cabazitaxel, or enzalutamide.

Illustration 1. Cost-minimization and Budget Impact Model.

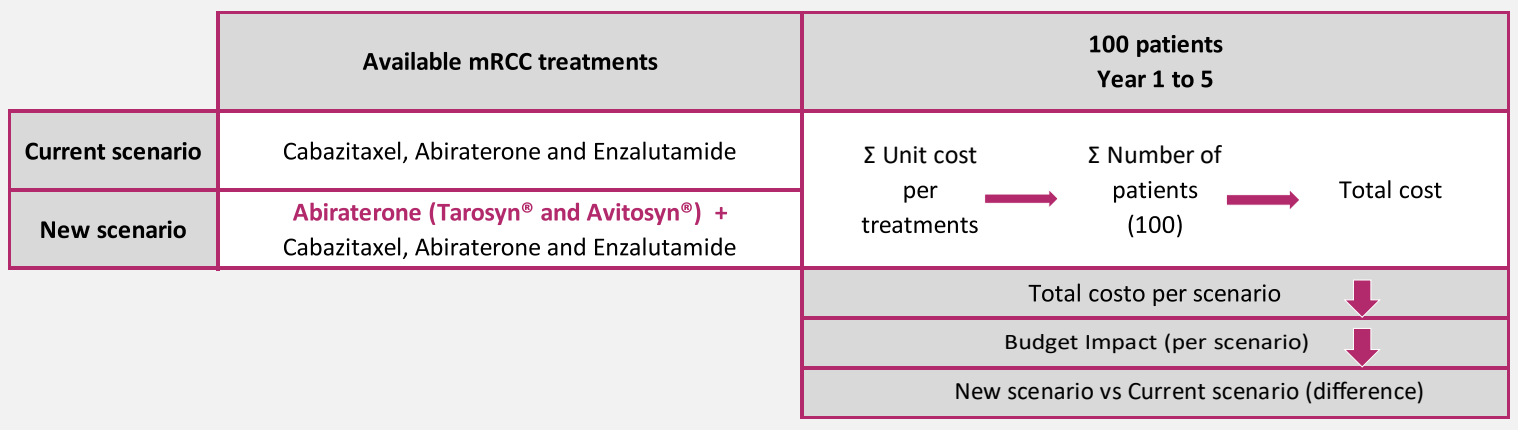
Available mRCC treatments
100 patients
Year 1 to 5
Current scenario
Cabazitaxel, Abiraterone and Enzalutamide
New scenario
Abiraterone (Tarosyn® and Avitosyn®) +
Cabazitaxel, Abiraterone and Enzalutamide
Σ Unit cost per treatments
Σ Number of patients (100)
Total cost
Total costo per scenario
Budget Impact (per scenario)
New scenario vs Current scenario (difference)

BIA assumptions include that new Abiraterone (Tarosyn® and Avitosyn®) would gradually gain market share until it reaches 100% of new or switching patients in the fifth year.

Illustration 2. Available mCRPC treatments, cost differences.

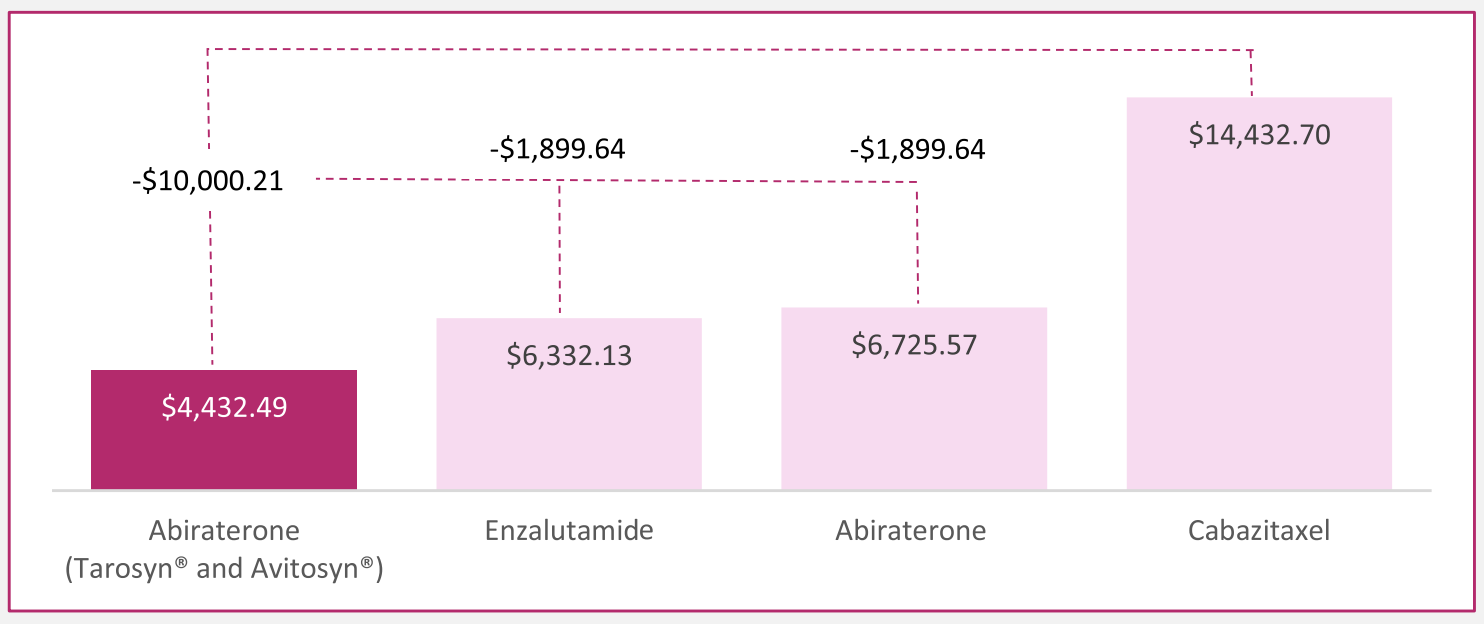
-$10,000.21
-$1,899.64
-$1,899.64
$14,432.70
$4,432.49
$6,332.13
$6,725.57
Abiraterone
(Tarosyn® and Avitosyn®)
Enzalutamide
Abiraterone
Cabazitaxel

All costs are in 2024 USD.

Table 1. Patient distribution per year (current and new scenario).

Current scenario	Market share year 1	Market share year 2	Market share year 3	Market share year 4	Market share year 5
Cabazitaxel	5%	7%	9%	10%	15%
Abiraterone	45%	40%	40%	40%	40%
Enzalutamide	50%	53%	51%	50%	45%
Total	100%	100%	100%	100%	100%
New scenario	**Market share year 1**	**Market share year 2**	**Market share year 3**	**Market share year 4**	**Market share year 5**
Cabazitaxel	5%	5%	5%	0%	0%
Abiraterone	15%	10%	5%	5%	0%
Enzalutamide	20%	15%	10%	5%	0%
Abiraterone (Tarosyn® and Avitosyn®)	60%	70%	80%	90%	100%
Total	100%	100%	100%	100%	100%

RESULTS

In the CMA, the calculated costs per year treatment per patient were $4,954.33 for new Abiraterone (Tarosyn® and Avitosyn®), basal abiraterone $11,644.13, cabazitaxel $9,252.93 and enzalutamide $12,719.45. Savings associated to new Abiraterone (Tarosyn® and Avitosyn®) are in average $6,251.17 per patient.

Table 2. CMA results

mCRP treatment	Total cost (per year)	Incremental cost	Times vs new treatment	Patients treated with new treatment
Abiraterone (Tarosyn® and Avitosyn®)	**$4,954.33**	**Reference**		
Cabazitaxel	$9,252.93	-$4,298.60	1.87	-0.87
Abiraterone	$11,644.13	-$6,689.80	2.35	-1.35
Enzalutamide	$12,719.45	-$7,765.12	2.57	-1.57

Probabilistic Sensitivity Analysis (SA) was conducted with 1,000 random simulations, with new Abiraterone (Tarosyn® and Avitosyn®) being less costly than its comparators in all simulations.

Table 3. Patient distribution and cost 1-5 years (current *vs* new scenario).

Current scenario	Year 1	Year 2	Year 3	Year 4	Year 5
Cabazitaxel	$72,163.50	$101,028.90	$129,894.30	$144,327.00	$216,490.50
Abiraterone	$302,650.65	$269,022.80	$269,022.80	$269,022.80	$269,022.80
Enzalutamide	$316,606.50	$335,602.89	$322,938.63	$316,606.50	$284,945.85
Total	$691,421	$705,655	$721,856	$729,956	$770,459
New scenario	**Year 1**	**Year 2**	**Year 3**	**Year 4**	**Year 5**
Cabazitaxel	$72,163.50	$72,163.50	$72,163.50	$0.00	$0.00
Abiraterone	$101,864.70	$67,909.80	$33,954.90	$33,954.90	$0.00
Enzalutamide	$131,565.57	$98,674.18	$65,782.78	$32,891.39	$0.00
Abiraterone (Tarosyn® and Avitosyn®)	$276,288	$322,336	$368,384	$414,432	$460,479
Total	$581,881	$561,083	$540,285	$481,278	$460,479

Additionally, the BIA result indicates that new Abiraterone (Tarosyn® and Avitosyn®) is a cost-saving intervention, with an average predicted five-year saving-impact of -$984,970 for every 100-patient cohort.

Table 4. BI difference 1-5 years (current *vs* new scenario).

Difference between scenarios	Year 1	Year 2	Year 3	Year 4	Year 5
Total cost					
Current scenario	$691,420.65	$705,654.59	$721,855.73	$729,956.30	$770,459.15
New scenario	$581,881.46	$561,083.12	$540,284.78	$481,277.83	$460,479.49
Difference (total cost)	**-$109,539.19**	**-$144,571.47**	**-$181,570.95**	**-$248,678.47**	**-$309,979.66**
Budget impact					
Current scenario	0.00101%	0.00103%	0.00105%	0.00106%	0.00112%
New scenario	0.00085%	0.00082%	0.00079%	0.00070%	0.00067%
(Difference BI)	**-0.00016%**	**-0.00021%**	**-0.00026%**	**-0.00036%**	**-0.00045%**

The difference between scenarios indicated average savings generated by new Abiraterone (Tarosyn® and Avitosyn®) for -$286,221. The BI for IMSS is -0.00020% in average from year 1-5.

CONCLUSION

Results obtained by both CMA and BIA, concluded that the adoption of new Abiraterone (Tarosyn® and Avitosyn®) by the IMSS in the Mexican public healthcare system has a positive impact on its budget, considering the low cost when compared to other metastatic castration-resistant prostate cancer drugs without any prejudice regarding efficacy and safety parameters.

BIA result also indicates that Abiraterone (Tarosyn® and Avitosyn®) is a cost-saving intervention.

Results increasing 10% unit price of Abiraterone (Tarosyn® and Avitosyn®) were robust in the SA.

REFERENCES

1. *Fizazi K, Scher HI, Molina A, Logothetis CJ, Chi KN, Jones RJ, Staffurth JN, North S, Vogelzang NJ, Saad F, et al. Abiraterone acetate for treatment of metastatic castration-resistant prostate cancer: final overall survival analysis of the COU-AA-301 randomised, double-blind, placebo-controlled phase 3 study. Lancet Oncol. 2012 Oct;13(10):983-92.*

Synthon
Contact
Tanya Pamela Sánchez y Rodríguez
Pamela.sanchez@synthon.com

Acknowledgment
palagod Health Supply

Presented at ISPOR Europe 2024 Barcelona, España (17-20 November 2024). This analysis was sponsored by Synthon México. The prices of Abiraterone (Tarosyn® and Avitosyn®) used in this Budget Impact Analysis are not binding.