The Payment for Performance Environment for Drugs and Medical Devices in Romania

HPR8

Radu CP[1], Dragoi L[2], Pana BC[3], Cheles C[4]

[1]Stryker Romania SRL, Bucharest, Romania, [2]Roche Romania SRL, Bucharest, Romania, [3] University of Medicine and Pharmacy Carol Davila Bucharest, Bucharest, Romania, [4]Gilead Sciences (GSR) S.R.L, Bucharest, Romania

INTRODUCTION

Within the National Health Strategy 2014 - 2020, Romania assumed some strategic directions to follow in healthcare, and one of the most important referred to "the development and implementation of an evidence-based drug policy to ensure equitable and sustainable population access to medication" [1].

OBJECTIVE

The objective of this research is to evaluate the readiness of the Romanian healthcare system to use the Payment for Performance (P4P) mechanisms in the reimbursement of drugs and medical devices.

METHOD

The authors performed an analysis of the Romanian legislation regarding the payment for performance for drugs and medical devices and an analysis of the existing local reimbursement models based on payment for performance, looking at public data sources.

RESULTS

- For drugs, there is one functional P4P mechanism in place, known as the "cost-volume-result" contract (CVRc).
- The CVRc represents a type of outcome-based payment agreement used with success for patients with Hepatitis C starting with 2015. By 2024 these CVRc expanded also to Hepatic Cirrhosis indication (table 1). Starting with 2021, the interferon-free non-respondents were also covered by one treatment and starting with 2022 the interferon-free pan-genotype by two treatments.
- From 2015 to 2023 over 50.000 patients received their therapies based on these CVRc and the success rate (cured patients) was around 96-98%[2, 3].
- CRVc used for the interferon-free treatment are mentioned in the national guidelines, launched in 2019, developed as part of the Ministry of Health and World Health Organization partnership to control the viral hepatitis in Romania ("The National Framework for control of viral hepatitis in Romania, 2019-2030")[3].
- There are other P4P initiatives like specific protocols with the Payer, risk-sharing agreements based on results like progression-free survival for oncology proposed by different stakeholders (Drugs Association, American Chamber of Commerce). Some of them are mentioned in the drug reimbursement methodology by the Innovation Fund (oncology) which will be developed in the future period.
- Regarding the medical devices, there is no functional mechanism in place. There are initiatives to promote the P4P proposed by different stakeholders (Medical Devices Association, American Chamber of Commerce etc.) to cover the reimbursement of innovative medical devices but are not yet implemented.

Year	Hepatitis C	Hepatic Cirrhosis
2015	2	0
2016	2	0
2017	3	3
2018	4	4
2019	4	4
2020	4	4
2021	4	4
2022	6	6
2023	4	4
2024	4	4

Table 1. The number of drugs with P4P mechanism by pathology in Romania, 2015-2024 (using Cost-Volume-Results contract) [4]

CONCLUSIONS

The P4P is just at the beginning in Romania. The experience gained in the last 10 years from the utilization of the "cost-volume-result" contracts for drugs provides the starting point for the future expansion of the P4P.

The main issues in expanding the P4P are related to the definition of the performance for different health interventions, the availability of easy-to-collect data and the administrative capacity to run such a payment mechanism.

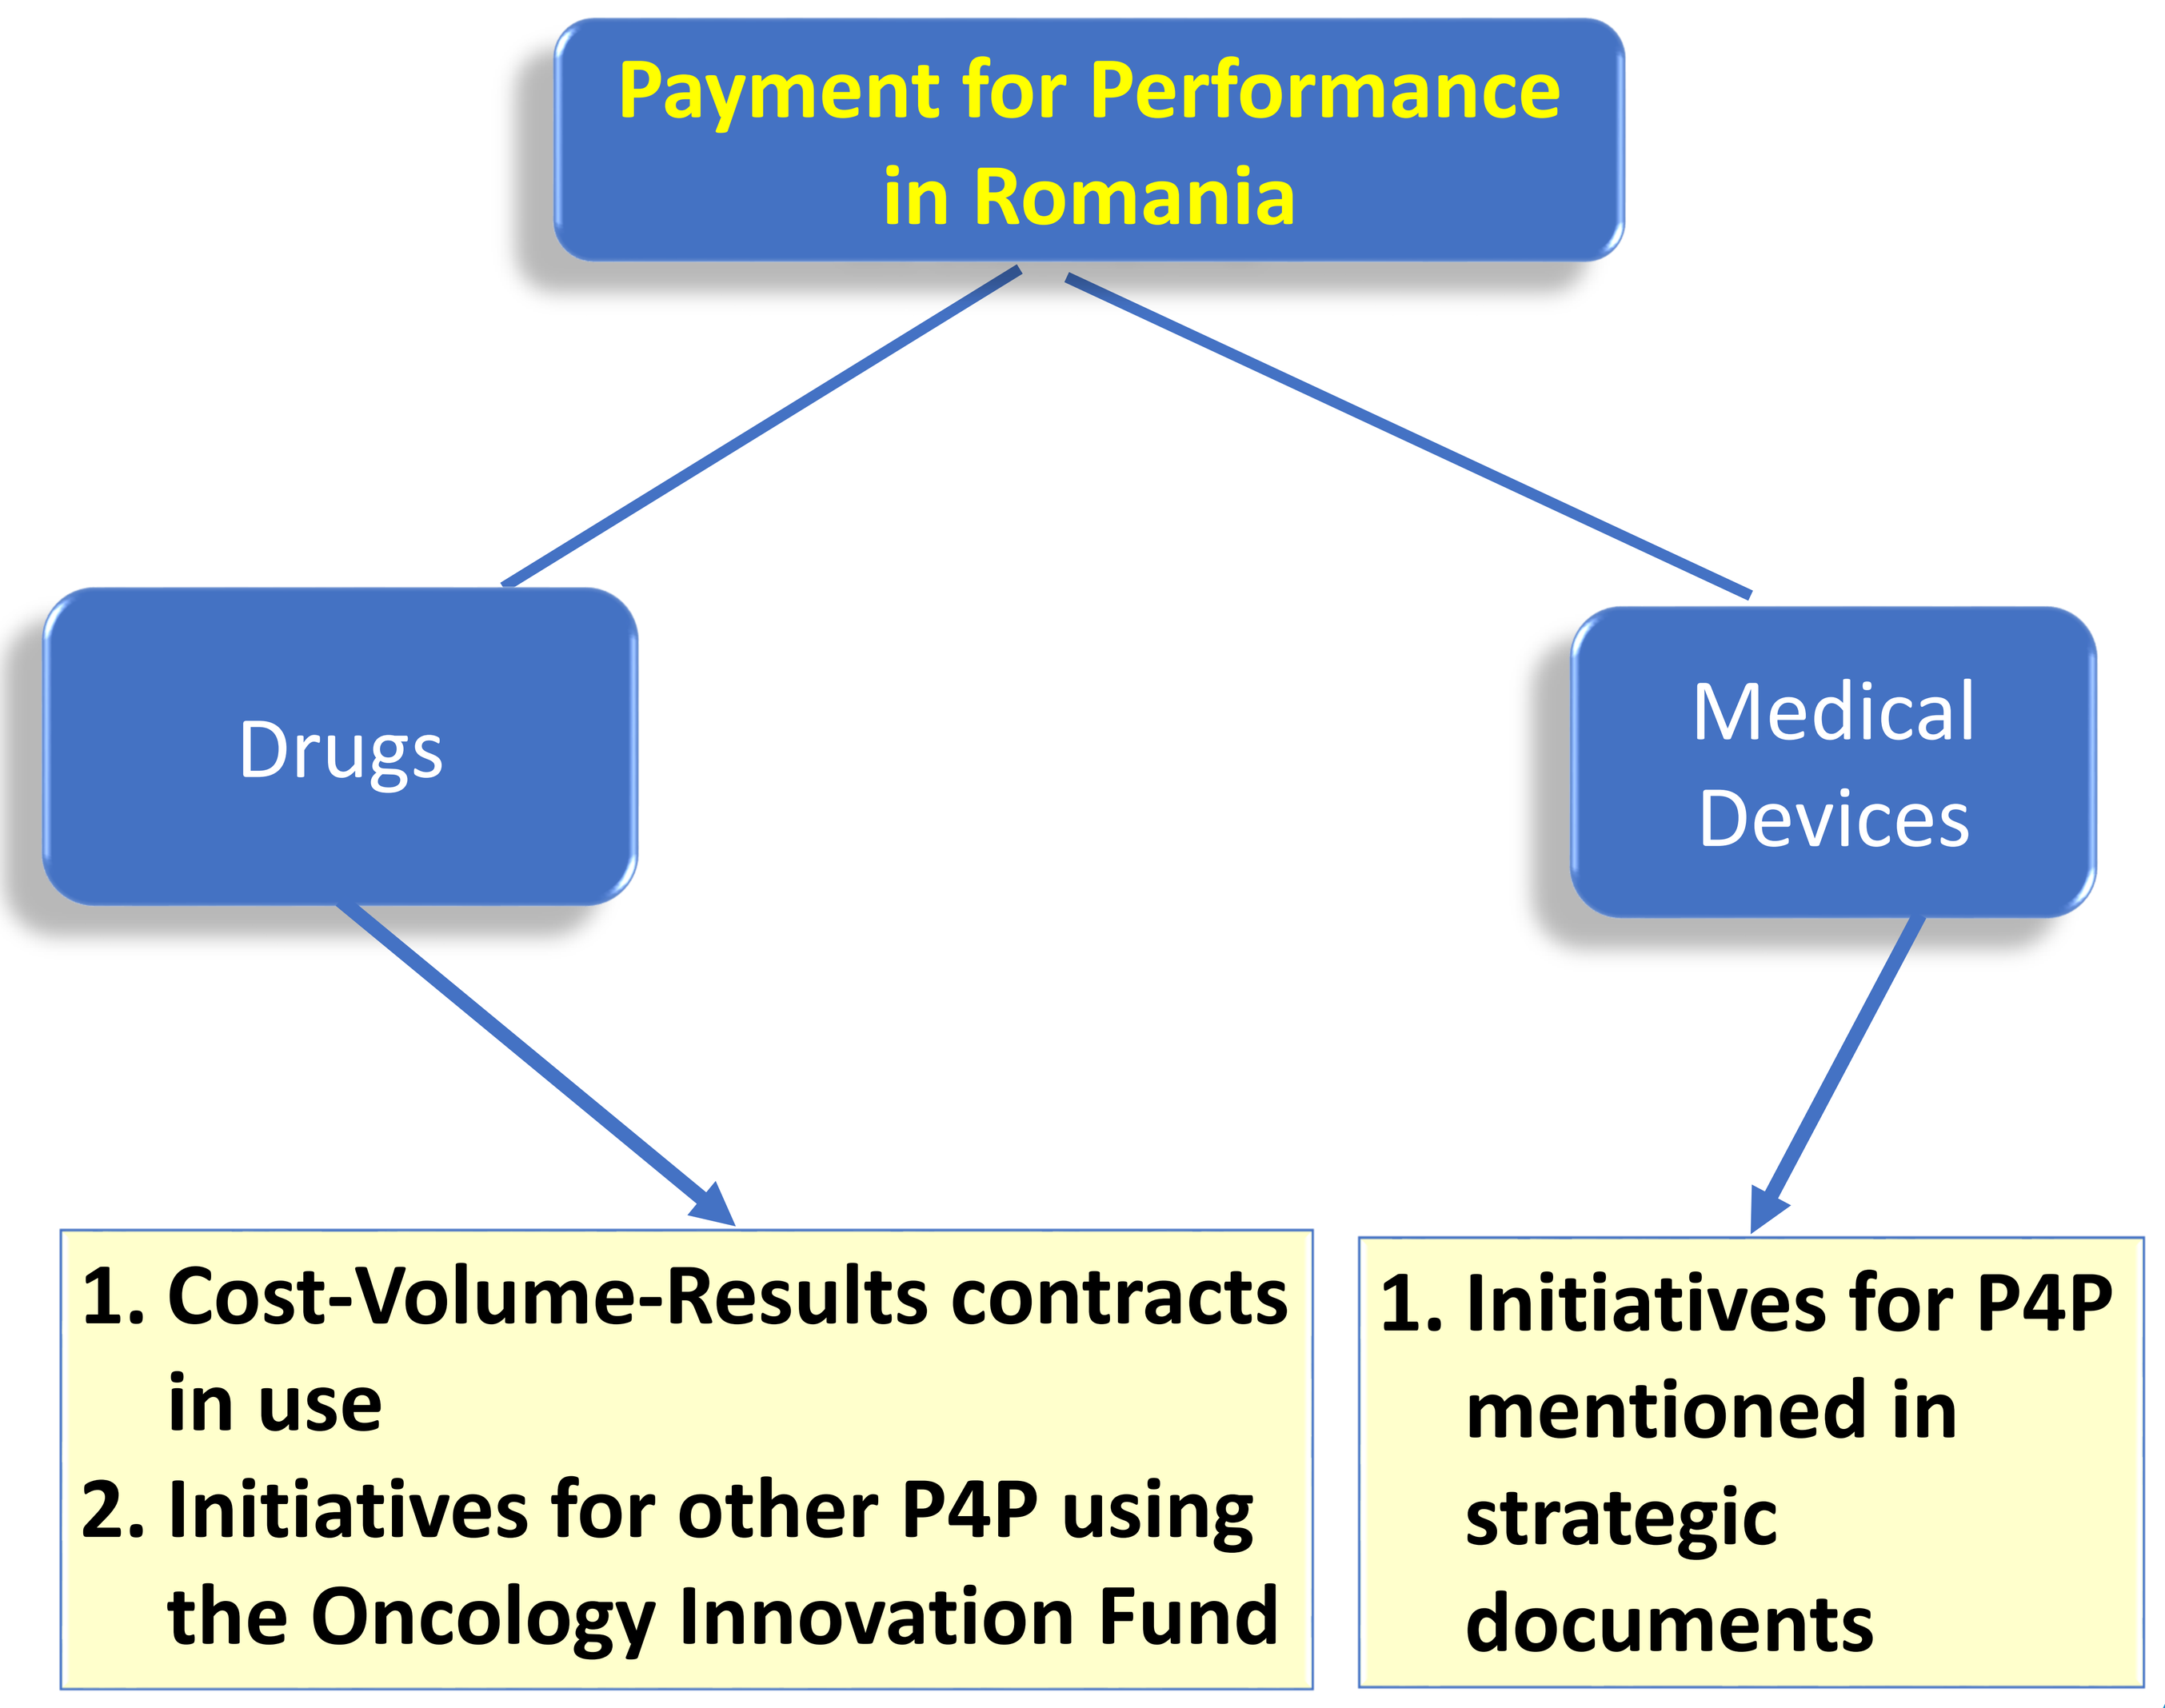
Payment for Performance in Romania
Drugs
Medical Devices
1. Cost-Volume-Results contracts in use
2. Initiatives for other P4P using the Oncology Innovation Fund
1. Initiatives for P4P mentioned in strategic documents

REFERENCES

[1] *https://farmaciajournal.com/issue-articles/the-outcomes-of-managed-entry-agreements-in-romania-from-2015-to-2022/*

[2] *https://hotnews.ro/romnia-are-printre-cele-mai-multe-cazuri-de-hepatita-din-europa-n-timp-ce-statele-unite-vor-sa-elimine-hepatita-c-pna-n-2030-39417*

[3] *Ordinul MS nr. 588/2019 pentru aprobarea Planului-cadru naţional privind controlul hepatitelor virale în România pentru perioada 2019-2030, published in Romanian Official Gazette no. 332/02.05.2019*

[4] *Government Decision no. 720/2008 regarding the Reimbursement List of drugs covered by the social health insurance system in Romania*

CONTACT INFO

ciprianpaul.radu@stryker.com

ISPOR Europe 2024 Conference, November 17-20, Barcelona, Spain

ISPOR2024 HPR-8 Health Policy & Regulatory Ciprian Paul Radu

ISPOR Europe 2024 17 - 20 November | Barcelona, Spain

PosterSessionOnline.com SPREADING KNOWLEDGE