COST-EFFECTIVENESS ANALYSIS OF CORRECT VS IMPROPER USE OF RIVASTIGMINE TRANSDERMAL PATCH IN PATIENTS WITH ALZHEIMER'S DISEASE IN COLOMBIA.

EE116

Romero M[1] , Cuellar K[1] , Acero G[1] , Díaz-Cely A[1] , Cuadros R[2], Patiño A[3].

1. **ProyectaMe**. Colombia. 2. **Asociación Colombiana de Gerontología y Geriatría.** 3. **Knight Therapeutics**. Colombia.

INTRODUCTION

Alzheimer´s disease (AD) **is becoming one of the leading causes of death in the world**, due to world population ageing and increase in incidence and prevalence. It is expected that annual **cost of (AD) reach US $2.8 trillion in 2030**. In Colombia Alzheimer´s disease is the **7th cause of mortality** for all the population groups, and the **4th cause** of disability adjusted life years (DALYs) in elderly population (>70 years old). **Rivastigmine transdermal patch is used to treat mild to severe AD. Proper administration of this patch is crucial for archiving desired therapeutics effects and minimizing adverse events**. Improper use, including erroneous application of the patch and incorrect titration, can lead to substantial impact on therapeutic outcomes and increased healthcare costs.(1,2)

OBJECTIVE

Evaluate the cost associated with the improper use (erroneous application and incorrect titration) of the rivastigmine patch in patients with AD.

METHODS

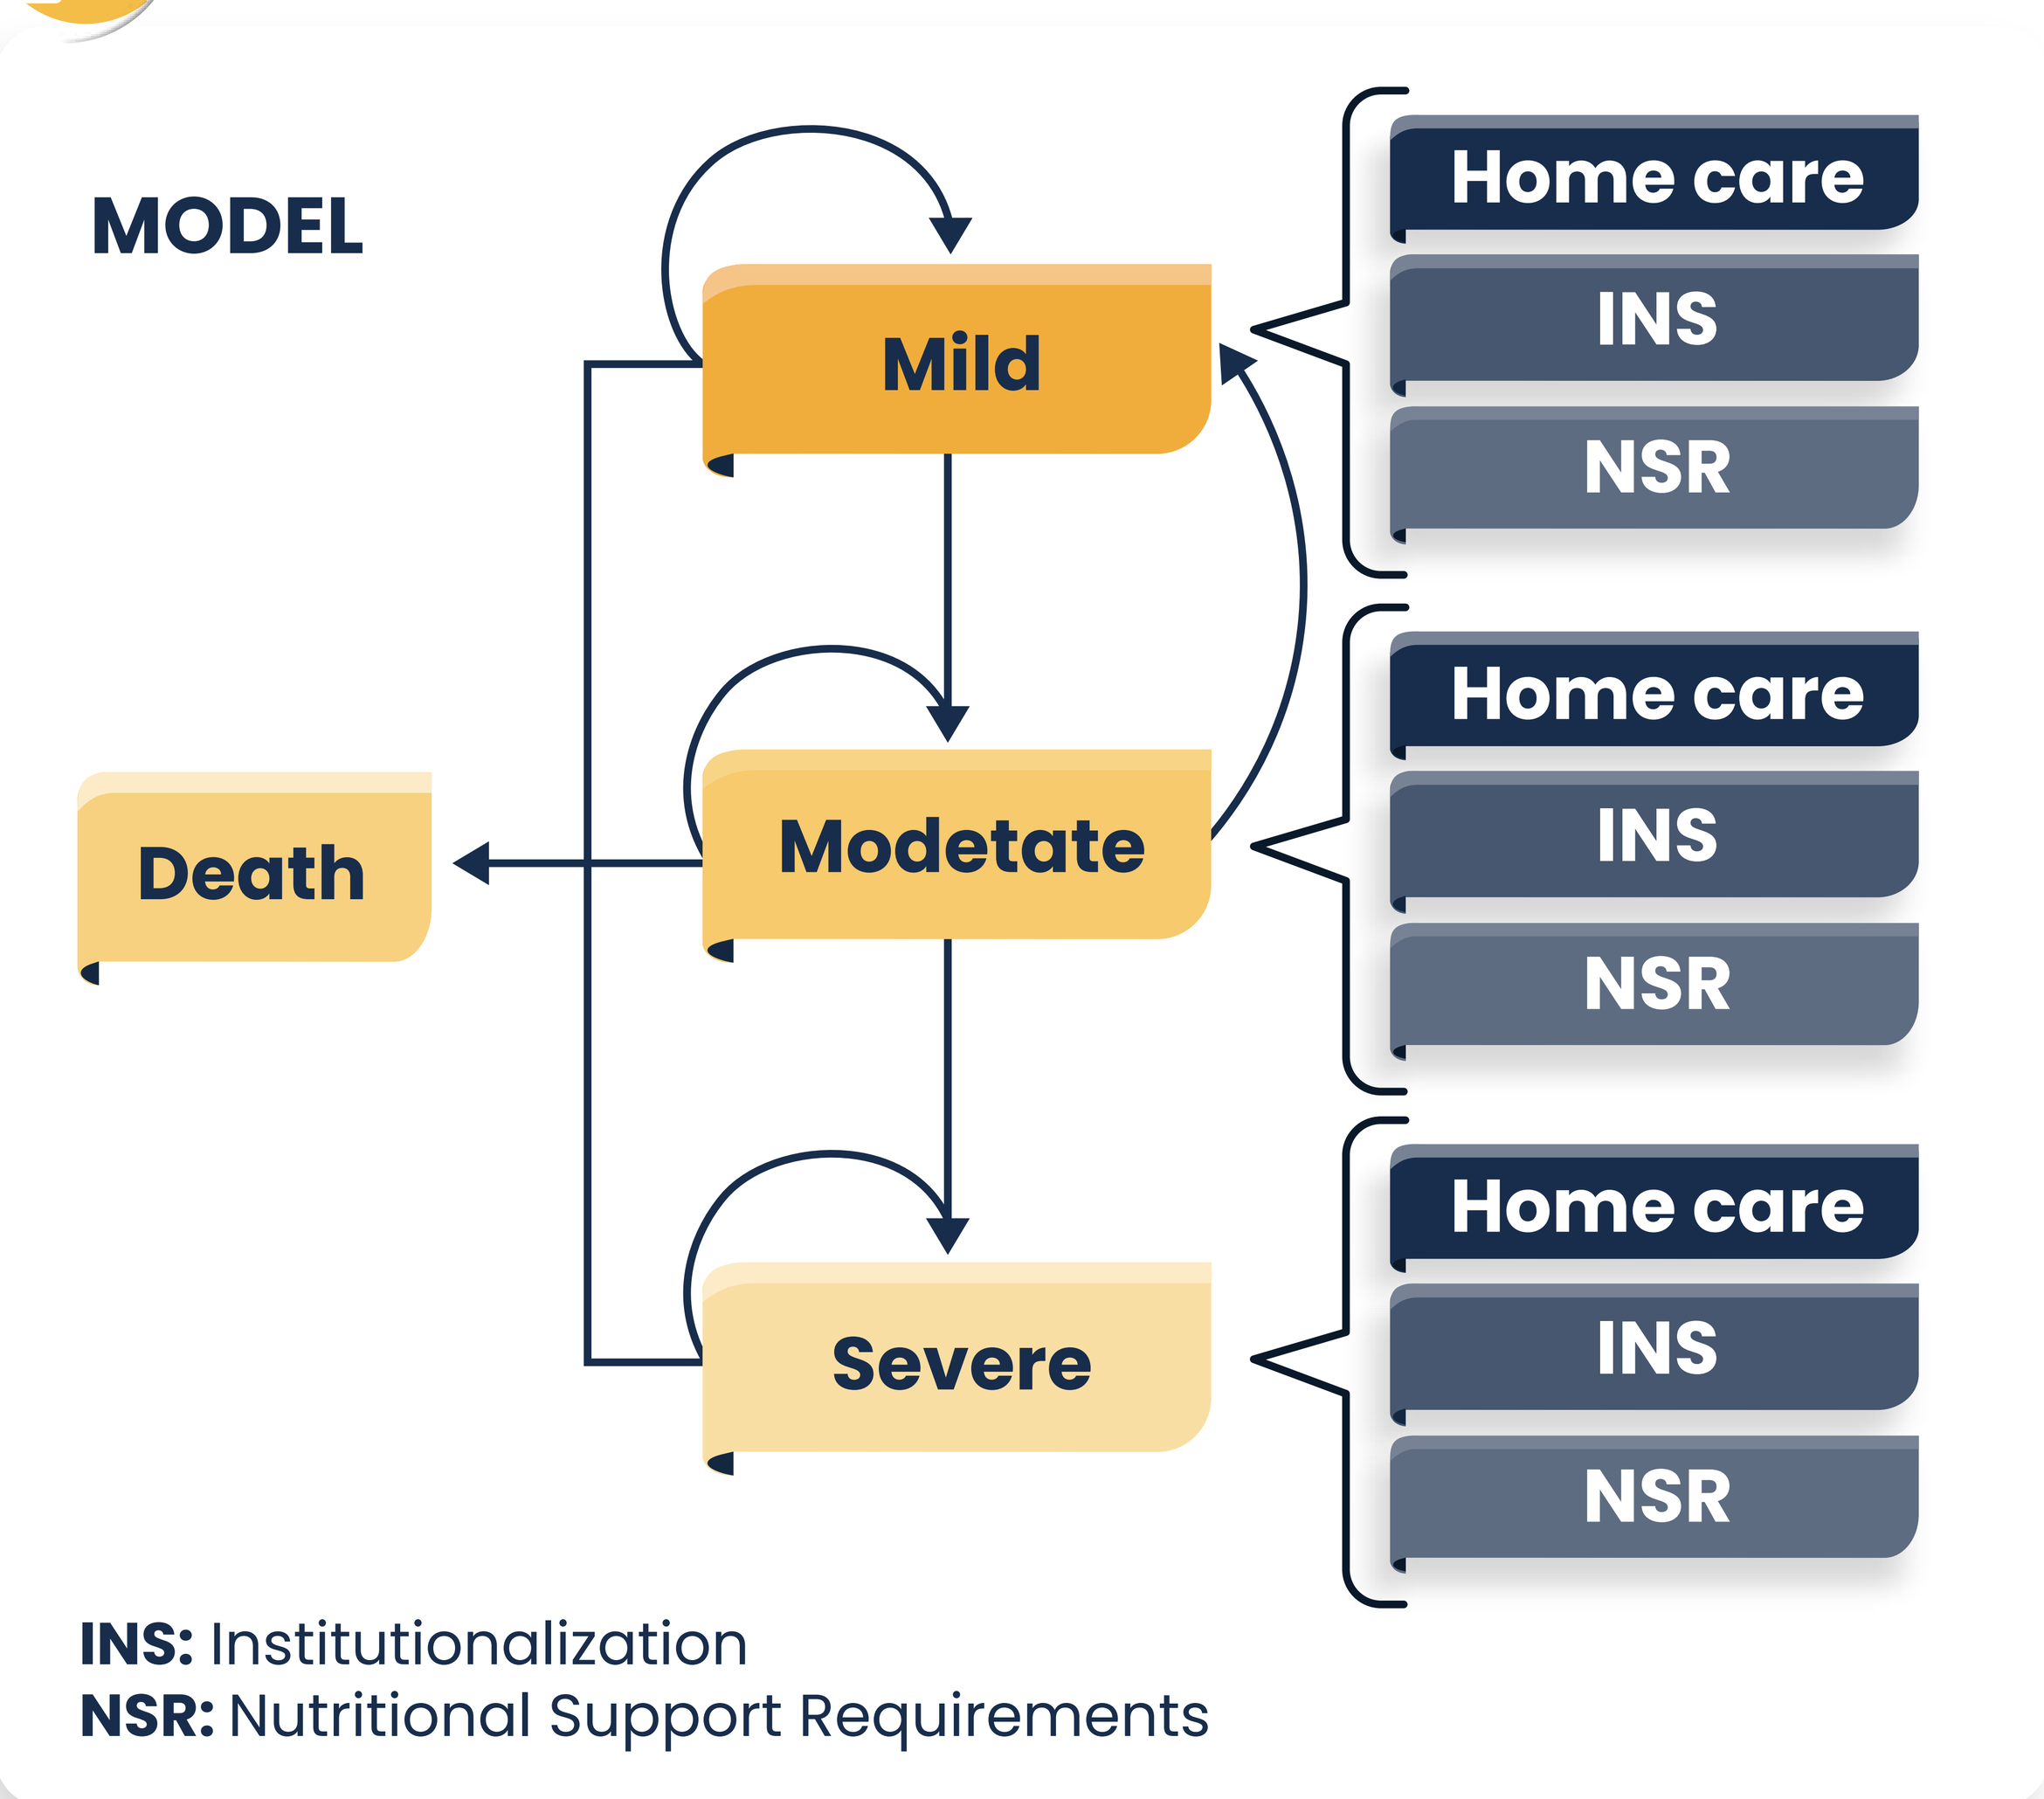
MODEL
Mild
Modetate
Severe
Death
Home care
INS
NSR
Home care
INS
NSR
Home care
INS
NSR
INS: Institutionalization
NSR: Nutritional Support Requirements

A cost-effectiveness analysis was developed using a Markov model that included health states based on **severity** (mild, moderate, severe) and considered transitions between these states, mortality, **institutionalization** (INS) which is the remission of a patient to a specialized center for dementia care, and nutritional support requirements (NSR). **Four scenarios were evaluated:** 1) no treatment, 2) improper use due to incorrect placement of patch, 3) improper use due to don't apply titration, using 9mg patch (4.5 mg/day) during the whole-time horizon, and 4) proper titration until reach a 27mg patch (13.3 mg/day). **Transition probabilities were obtained from clinical trials and published literature.**(3-10)

COSTS

COSTS INCLUDED IN THE MODEL ARE:

- Institutionalization (INS)
- Rivastigmine patch use
- Laboratory tests and use of other drugs (Memantine)
- Therapy — Therapy: Pshysical, occupational, respiratory
- Medical Visit — Geriatric, Psychiatric, Nurologist, Psychologist, Nutritionist, GP.
- Nutritional suppor requirements (NSR)

Frequencies were determined by expert consensus and costs were calculated based on Sismed 2024 Q1 for Rivastigmine and Memantine and market prices for INS, NSR, medical visits, laboratory tests. A USD – COP exchange rate of $4.095 was used.

EFFECTIVENESS

Effectiveness was calculated based on the Mini-Mental State Exam (MMSE) and translated into Life Years (LY) and Quality Adjusted Life Years (QALY) based on literature. Differential in effectiveness associated to the improper use of the patch (erroneous application and incorrect titration) was obtained from literature(7,8).

SENSITIVITY ANALYSIS

Sensitivity analyses were performed to assess the robustness of the model across 5 and 10 year time horizon.

RESULTS

Over a 15-year time horizon, institutionalization (INS) and Nutritional support requirements (NSR) together represent at least half of total cost (between 50% and 85%), in contrast Rivastigmine comprising less than 40% of total cost (between 29% and 38%). Comparing alternatives, the proper use of the patch (27mg) is the most cost-saving intervention with the lowest total cost among all treatment options. This cost-reduction is driven, mainly by decreases in INS costs (from $12,116 to $11,333) and NSR costs (from $7,102 to $4,732) when compared to no treatment, erroneous patch application (INS: $15,545; NSR: $5,841) and incorrect titration (INS: $15,726; NSR: $5,883) (Figure 1).

Figure 1. Base case 15-year time horizon, cost by category (US $)

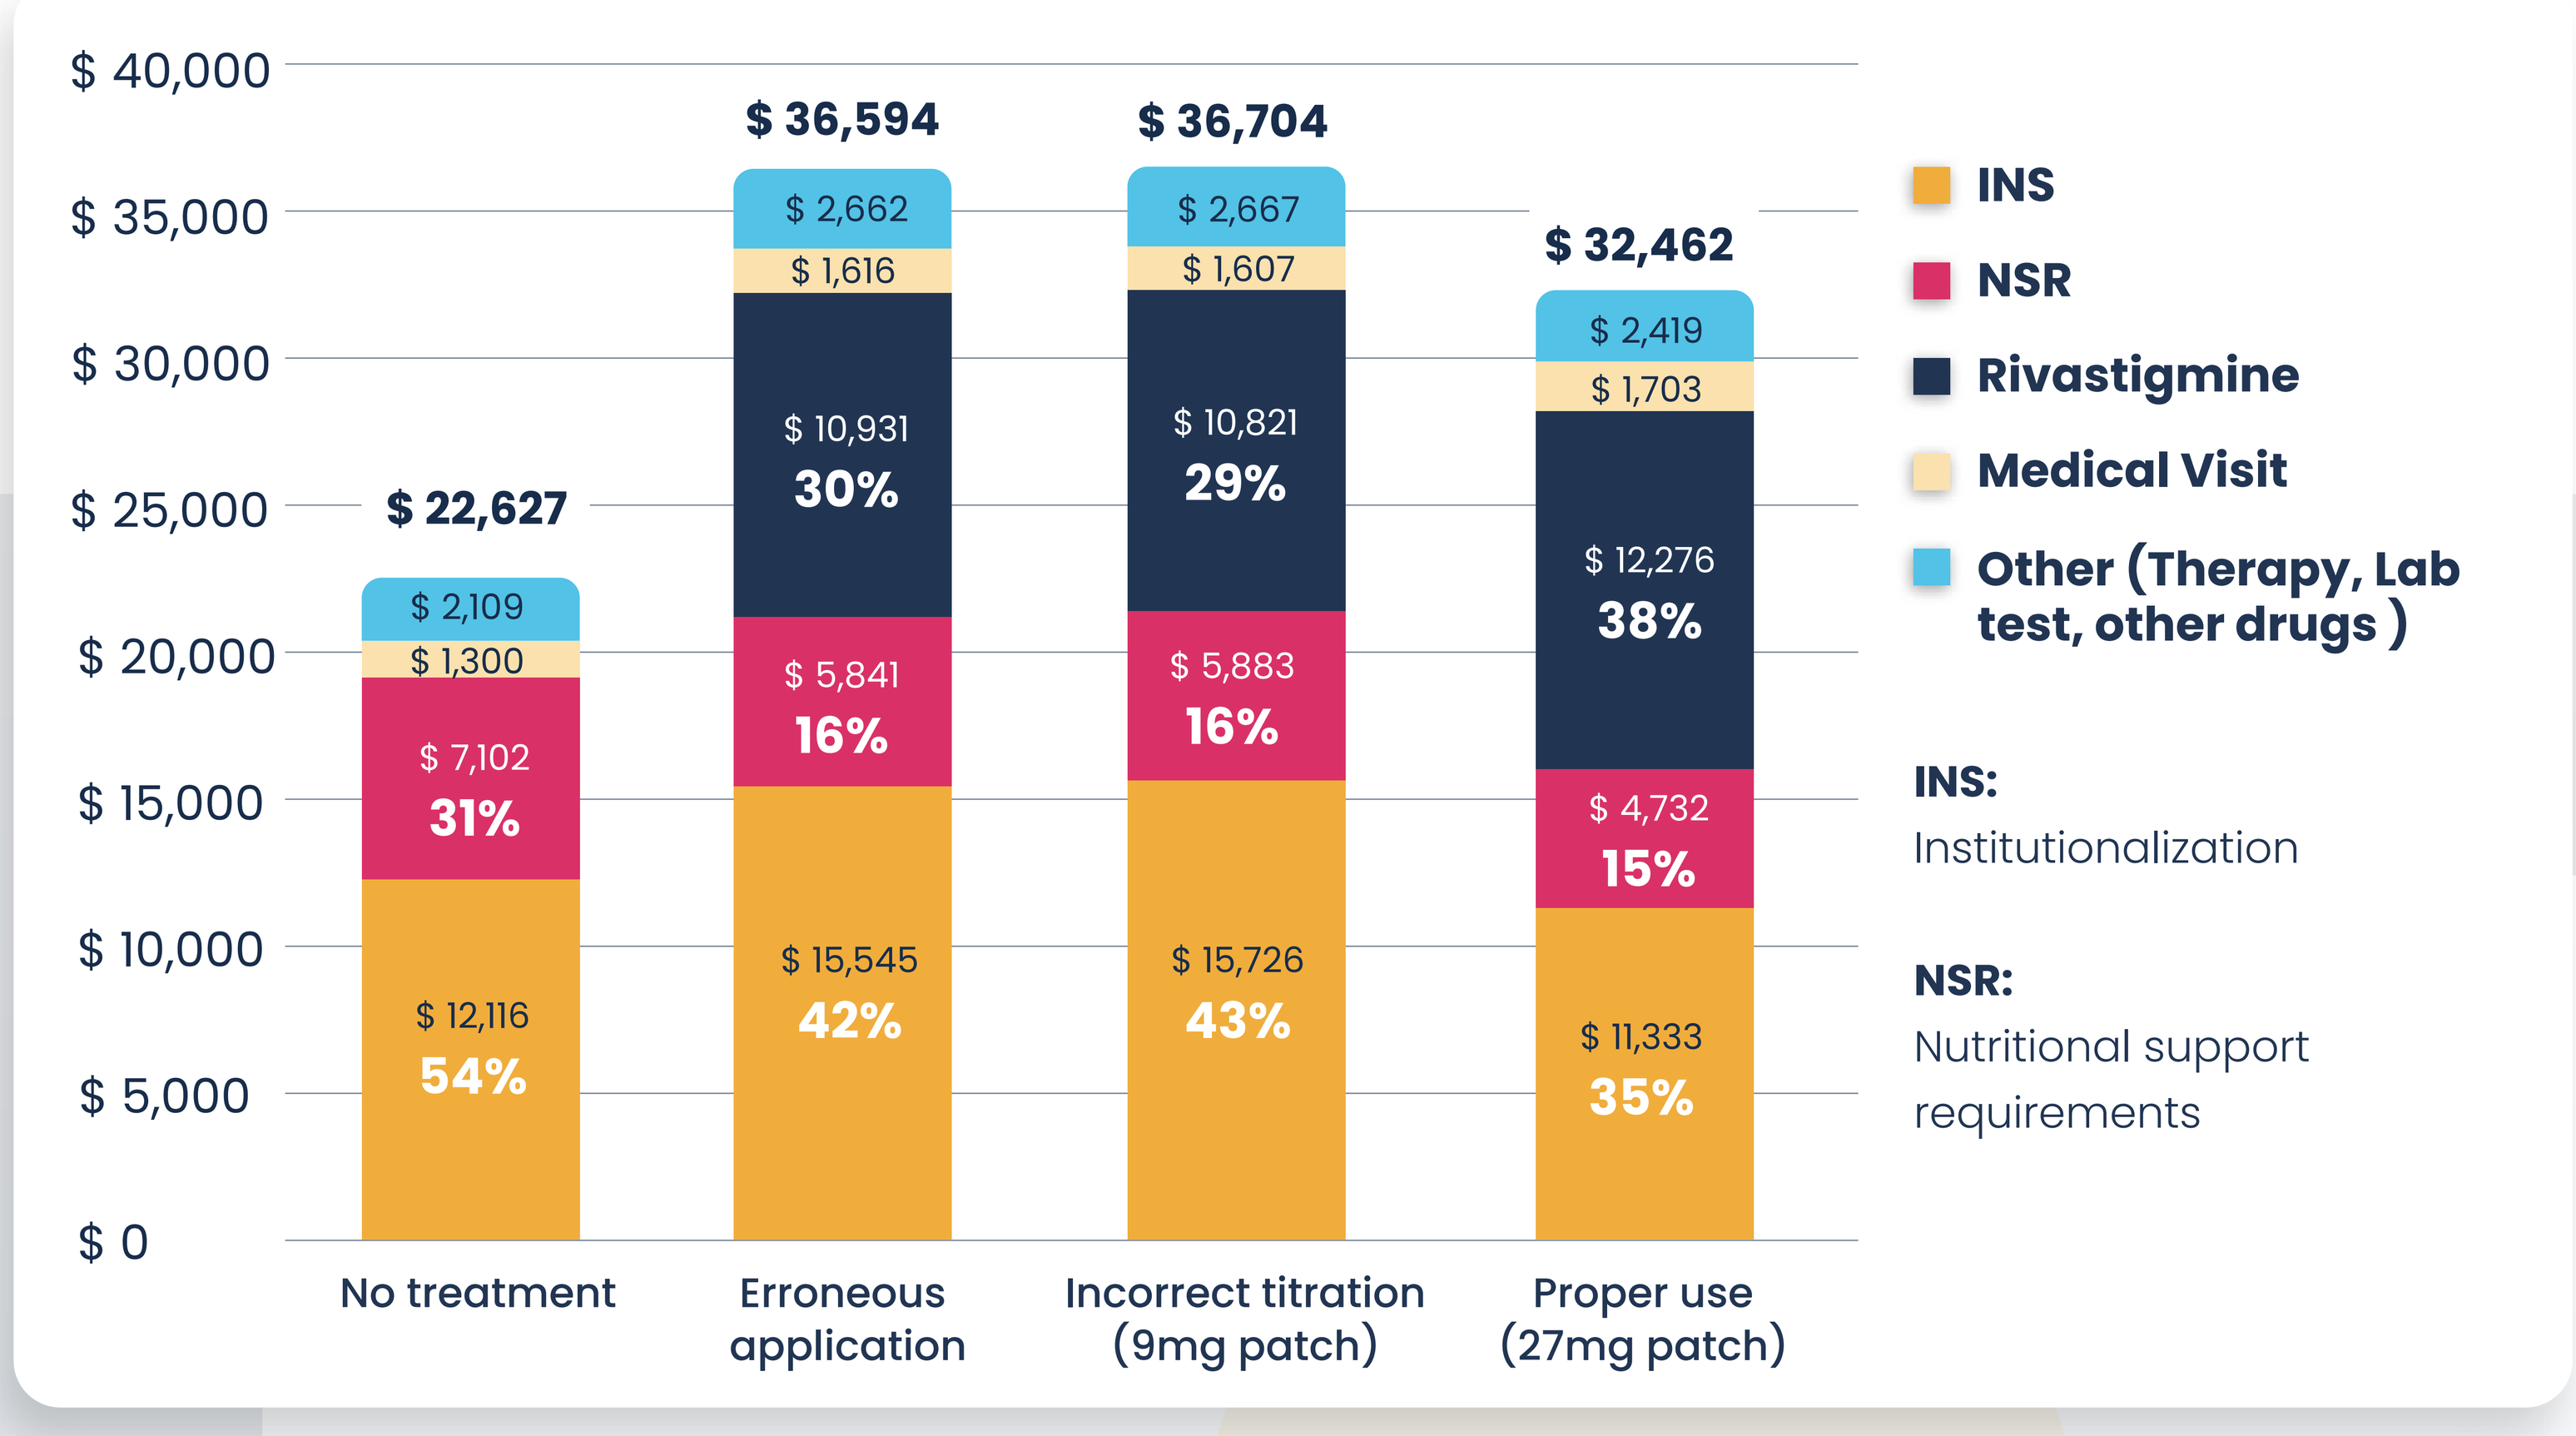
$ 40,000
$ 35,000
$ 30,000
$ 25,000
$ 20,000
$ 15,000
$ 10,000
$ 5,000
$ 0
$ 22,627
$ 2,109
$ 1,300
$ 7,102
31%
$ 12,116
54%
$ 36,594
$ 2,662
$ 1,616
$ 10,931
30%
$ 5,841
16%
$ 15,545
42%
$ 36,704
$ 2,667
$ 1,607
$ 10,821
29%
$ 5,883
16%
$ 15,726
43%
$ 32,462
$ 2,419
$ 1,703
$ 12,276
38%
$ 4,732
15%
$ 11,333
35%
No treatment
Erroneous application
Incorrect titration (9mg patch)
Proper use (27mg patch)
INS
NSR
Rivastigmine
Medical Visit
Other (Therapy, Lab test, other drugs)
INS:
Institutionalization
NSR:
Nutritional support requirements

The model showed that the 27mg rivastigmine patch with proper titration resulted in better outcomes in terms of Years of Life Saved (YL) 12.42, and Quality-Adjusted Life Years (QALY) 1.58. Demonstrating that it is a cost-effective intervention compared to no treatment and was dominant (lower total costs and higher effectiveness) over the related options of erroneus application and incorrect titration (Table 1). The sensitivity analysis showed that the results are stable across different time horizons, 5 and 10 years.

Table 1. Cost-effectiveness results at 15 years

	LY	QALY	Costs	Incremental effectiveness	Incremental Cost	ICUR
No treatment	8.81	0.86	$22,627	0.72	9,263.93	13,666
Erroneous application	11.02	1.04	$36,594	0.54	- 4,133	Dominant
Incorrect titration (9mg patch)	10.90	1.00	$36,704	0.58	- 4,243	Dominant
Proper use (27mg patch)	12.42	1.58	$32,462			

LY: Life of Years Saved **| QALY:** Qality Adjusted life Years **| ICUR:** Incremental Cost-utility ratio

CONCLUSIONS AND DISCUSSION

- Alzheimer´s Disease (AD) has a significant clinical and economic burden in Colombia, and it is expected to increase in the following years.
- Literature reports that severe stages are more expensive, and the main driver of this cost increase is the caregiver cost, which in our model is addressed by the institutionalization of the patient; Prada et al. 2014 reports that progress from mild to severe stage increase 15 times total costs of AD, in our model this is close to 5,4 times.
- Providing patients with Alzheimer's disease who are candidates for treatment with rivastigmine within an optimal management scheme for administration and titration is a cost-effective strategy. Inadequate use reduces the patch's effectiveness, incurs substantial additional costs to the healthcare system, and results in a deterioration of patients' quality of life.(11)
- Most of patients with AD suffer from 1 or more comorbidities and AD deterioration may have a larger impact on their health outcomes and costs, Prada et al. 2017 highlighted an increase between 1.79 and 4.47 times in total cost for severe AD patients depending on the number of comorbidities. Comorbidities are not included into the model and addressing this will improve results precision.(12)
- Initiatives that improve comprehensive care for AD patients and allows early diagnosis and correct treatment will impact not only health outcomes but also quality of life and costs.

REFERENCES:

References: 1. Alzheimer's Disease International. 2024. World Alzheimer Report 2024. - 2. Global Burden of Disease 2021. IHME 2024. - 3. Neumann PJ, et al. Neurology. 2001. 25;57(6):957-64. - 4. Albanese E, et al. Alzheimers Dement. 2013. Nov;9(6):649-56. - 5. Fuh JL, et al. Int J Geriatr Psychiatry. 2004. 19(3):266-70. - 6. St John, P. D., et al. Age and ageing. 2002. 31(5), 373-378. - 7. Helmer C, et al. Dement Geriatr Cogn Disord. 2007. 23(3):168-74 - 8. Furiya, Y., et al. Dem. & Ger Cogn Dis Extra. 2018. 8(1), 77-84. - 9. Lefevre, G. et al. 2007. The Journal of Clinical Pharmacology, 47(4), 471-478. - 10. Grossberg, G. T. et al. 2011. International journal of clinical practice, 65(4), 465-471. - 11. Prada SI, et al. 2017.1835–40. - 12. Prada, S. I., et al. 2014. 30(4), 247-255.

- This material is for exclusive use by the prescribing medical body.
- Additional information available with medical information area, e-mail: infomed.colombia@knighttx.com
- ©Biotoscana Farma S.A. All rights reserved.
- Total or partial reproduction is prohibited without authorization of the owner.