AGE-ADJUSTED YEARS OF POTENTIAL LIFE LOST (YPLL) RATES DUE TO OVARIAN CANCER IN CHILE DURING 2002-2019.

YPLL rates due to OC had overall declined. However, certain Chilean regions showed increasing rates in recent years.

EPH248

SCAN ME

Authors: dos Santos, FM[1], Caputo, M[1], Romero, JF[1]. Affiliations: [1]Medical Department, GSK, Santiago, Chile

Background & Aims

Since 2013, Ovarian Cancer (OC) was included in the Chilean Explicit Health Guarantees (GES) program, guaranteeing diagnosis and treatment (1). Advances in treatment and changes in clinical practice may impact on OC mortality. Therefore, the objective of this study was to evaluate the Years of Potential Life Lost (YPLL) due to OC in Chile considering 2002-2019 period.

Methods

Deaths: ICD-10 C56, C57.0, C48.1, C48.2 (2)
Population projection: National Statistics Institute (3).

Age-adjusted YPLL rates/100,000 habitants were estimated for Chile and its 7 most populous regions by using the direct method, applying age-specific rates in a population of interest to a standardized age distribution using the 2022 population as reference and considering 80-year life expectancy.

Postmenopausal women **(50-64)** and 3rd age **(65-79)** were analyzed.

The total period was evaluated and stratified into 2002-2013 (pre-GES) and 2014-2019 (post-GES) and assessed qualitative.

Results

Years of Potential Life Lost (considering 80 years as life expectancy)

50-64 years

The highest rate of AA-YPLL are in the post-menopause (50-64y) group due to the highest frequency of death.

Age group	Average AA-YPLL per 100.000 habs: Pre-GES: 2002-2013	Average AA-YPLL per 100.000 habs: Post-GES: 2014-2019	% of Change
50-64	44,60	43,22	-3%
65-79	19,08	19,16	0,4%

Postmenopausal and 3rd age AA-YPLL rates description in the 7 most populous Chilean regions. Results are shown as mean Pre-GES vs Post-GES (% of change)

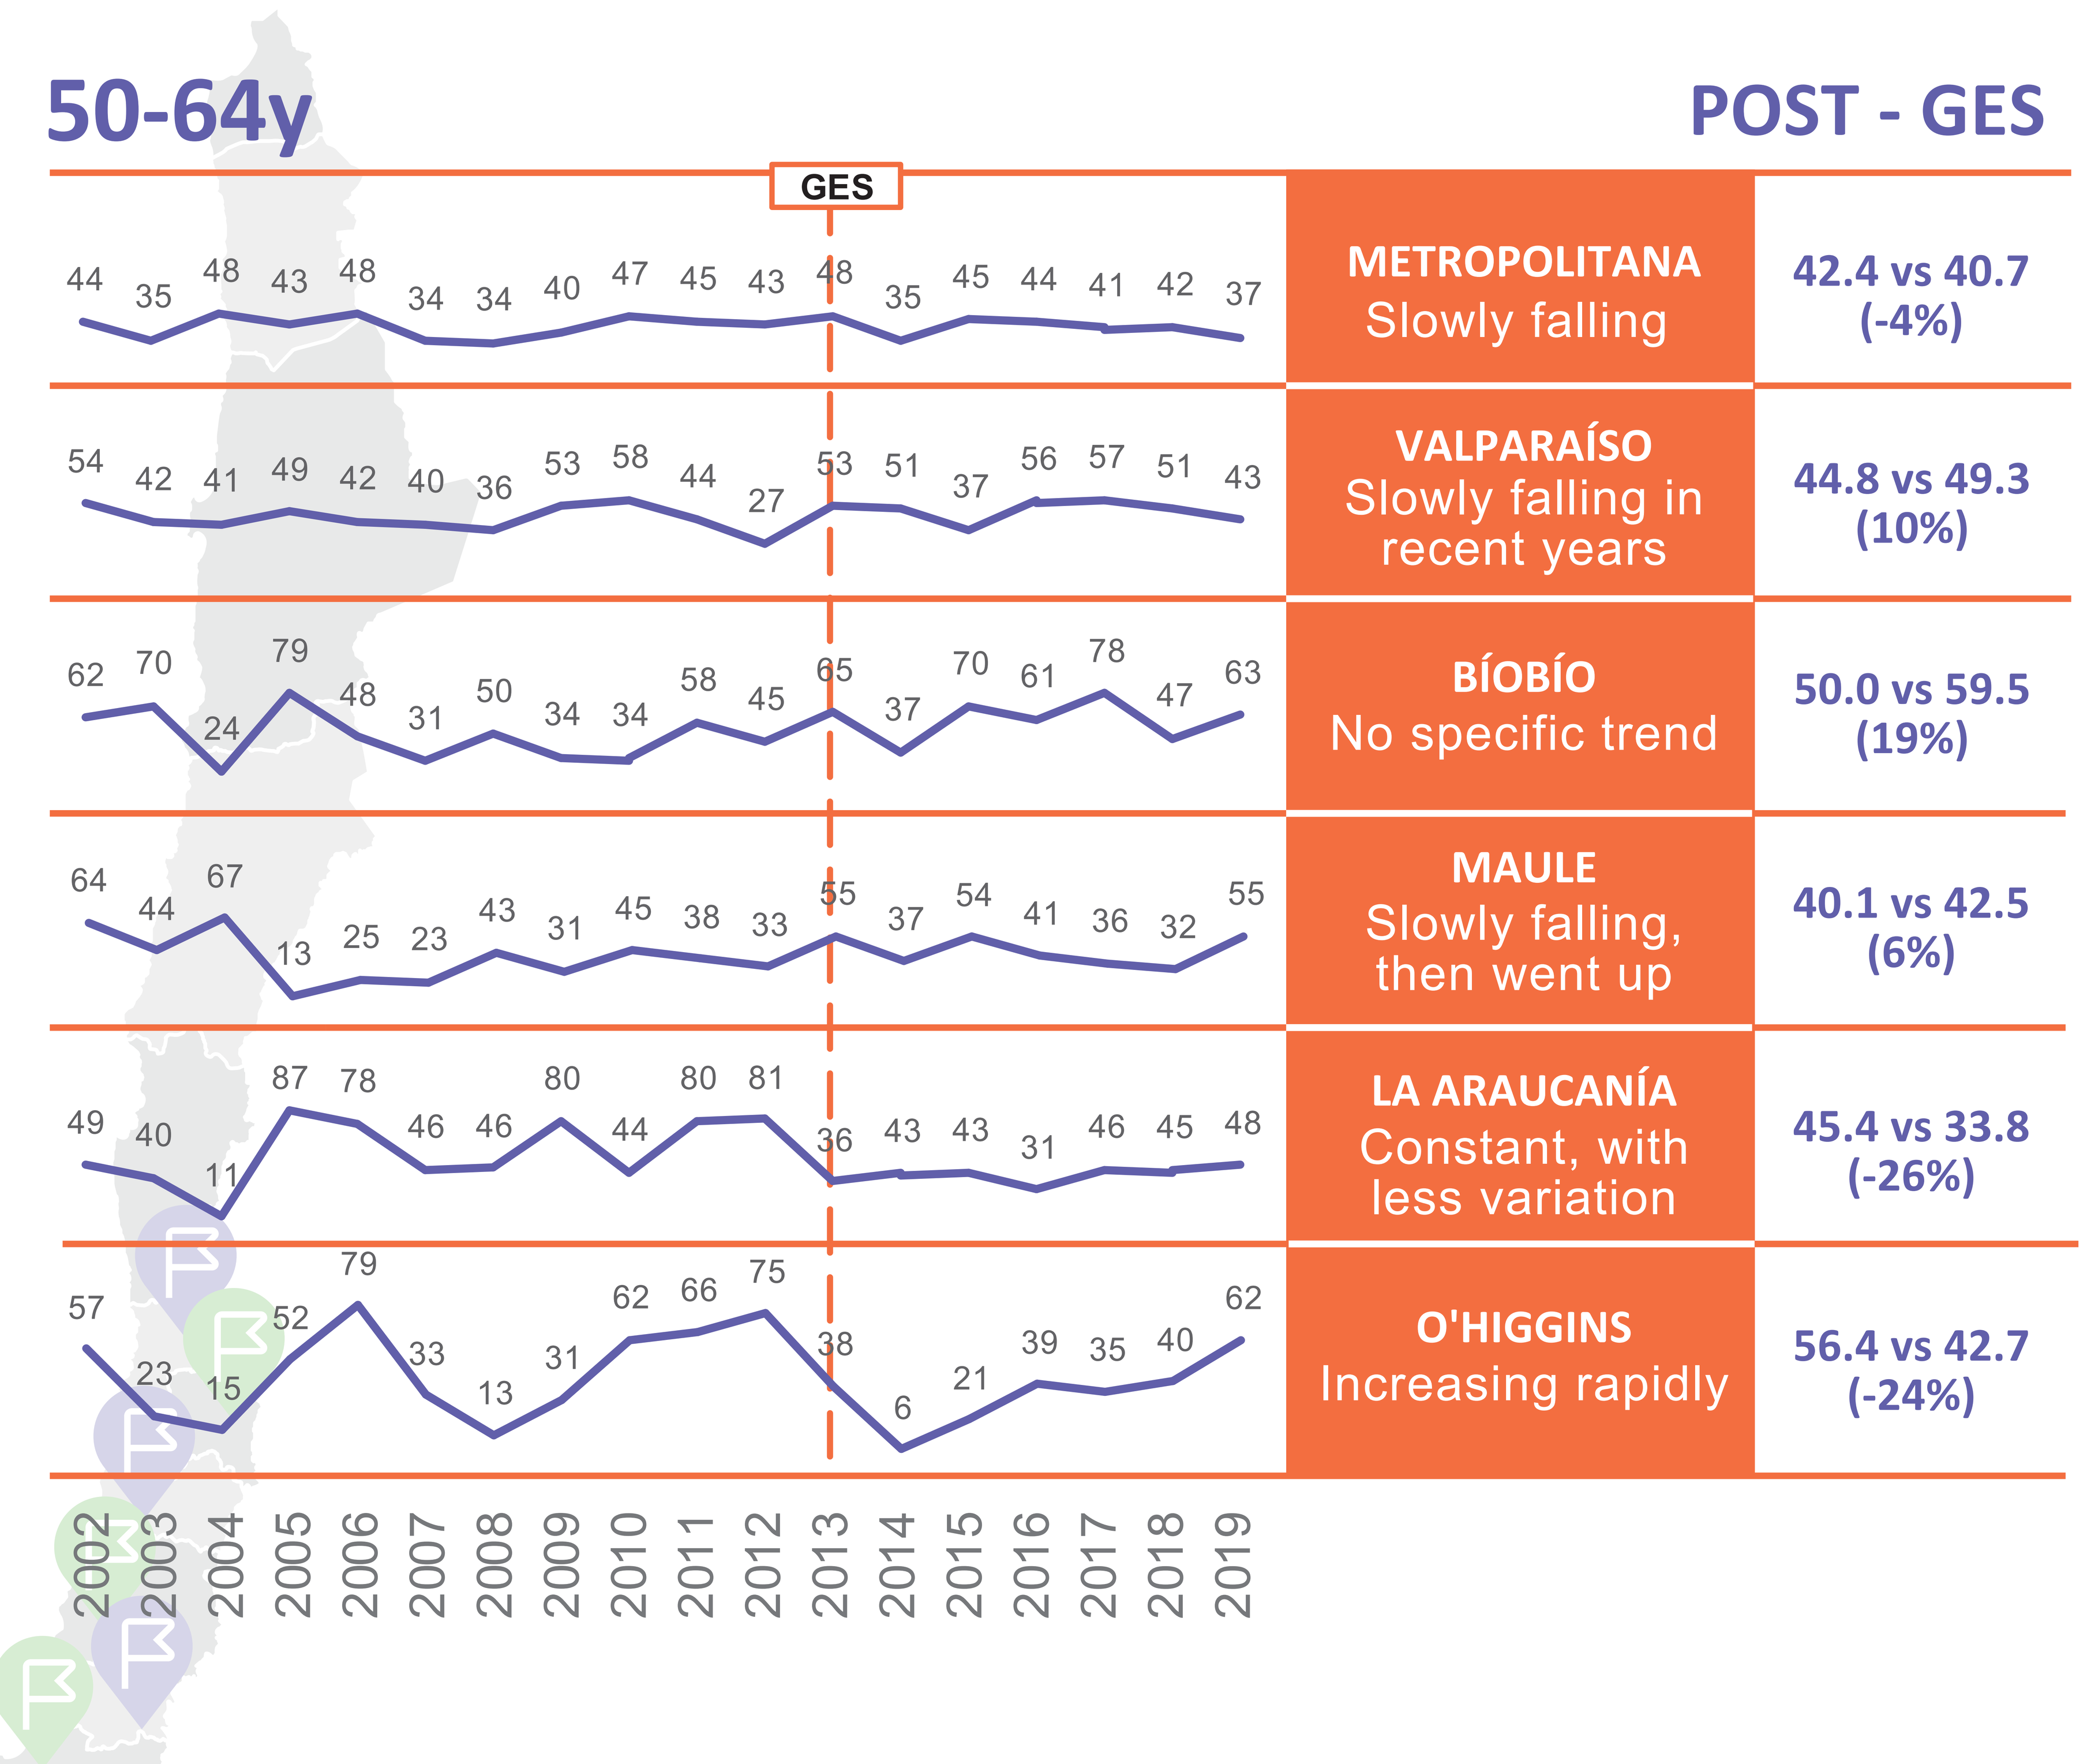
50-64y
POST - GES
GES
44 35 48 43 48 34 34 40 47 45 43 48 35 45 44 41 42 37
METROPOLITANA
Slowly falling
42.4 vs 40.7 (-4%)
54 42 41 49 42 40 36 53 58 44 27 53 51 37 56 57 51 43
VALPARAÍSO
Slowly falling in recent years
44.8 vs 49.3 (10%)
62 70 24 79 48 31 50 34 34 58 45 65 37 70 61 78 47 63
BÍOBÍO
No specific trend
50.0 vs 59.5 (19%)
64 44 67 13 25 23 43 31 45 38 33 55 37 54 41 36 32 55
MAULE
Slowly falling, then went up
40.1 vs 42.5 (6%)
49 40 11 87 78 46 46 80 44 80 81 36 43 43 31 46 45 48
LA ARAUCANÍA
Constant, with less variation
45.4 vs 33.8 (-26%)
57 23 15 52 79 33 13 31 62 66 75 38 6 21 39 35 40 62
O'HIGGINS
Increasing rapidly
56.4 vs 42.7 (-24%)
2002 2003 2004 2005 2006 2007 2008 2009 2010 2011 2012 2013 2014 2015 2016 2017 2018 2019

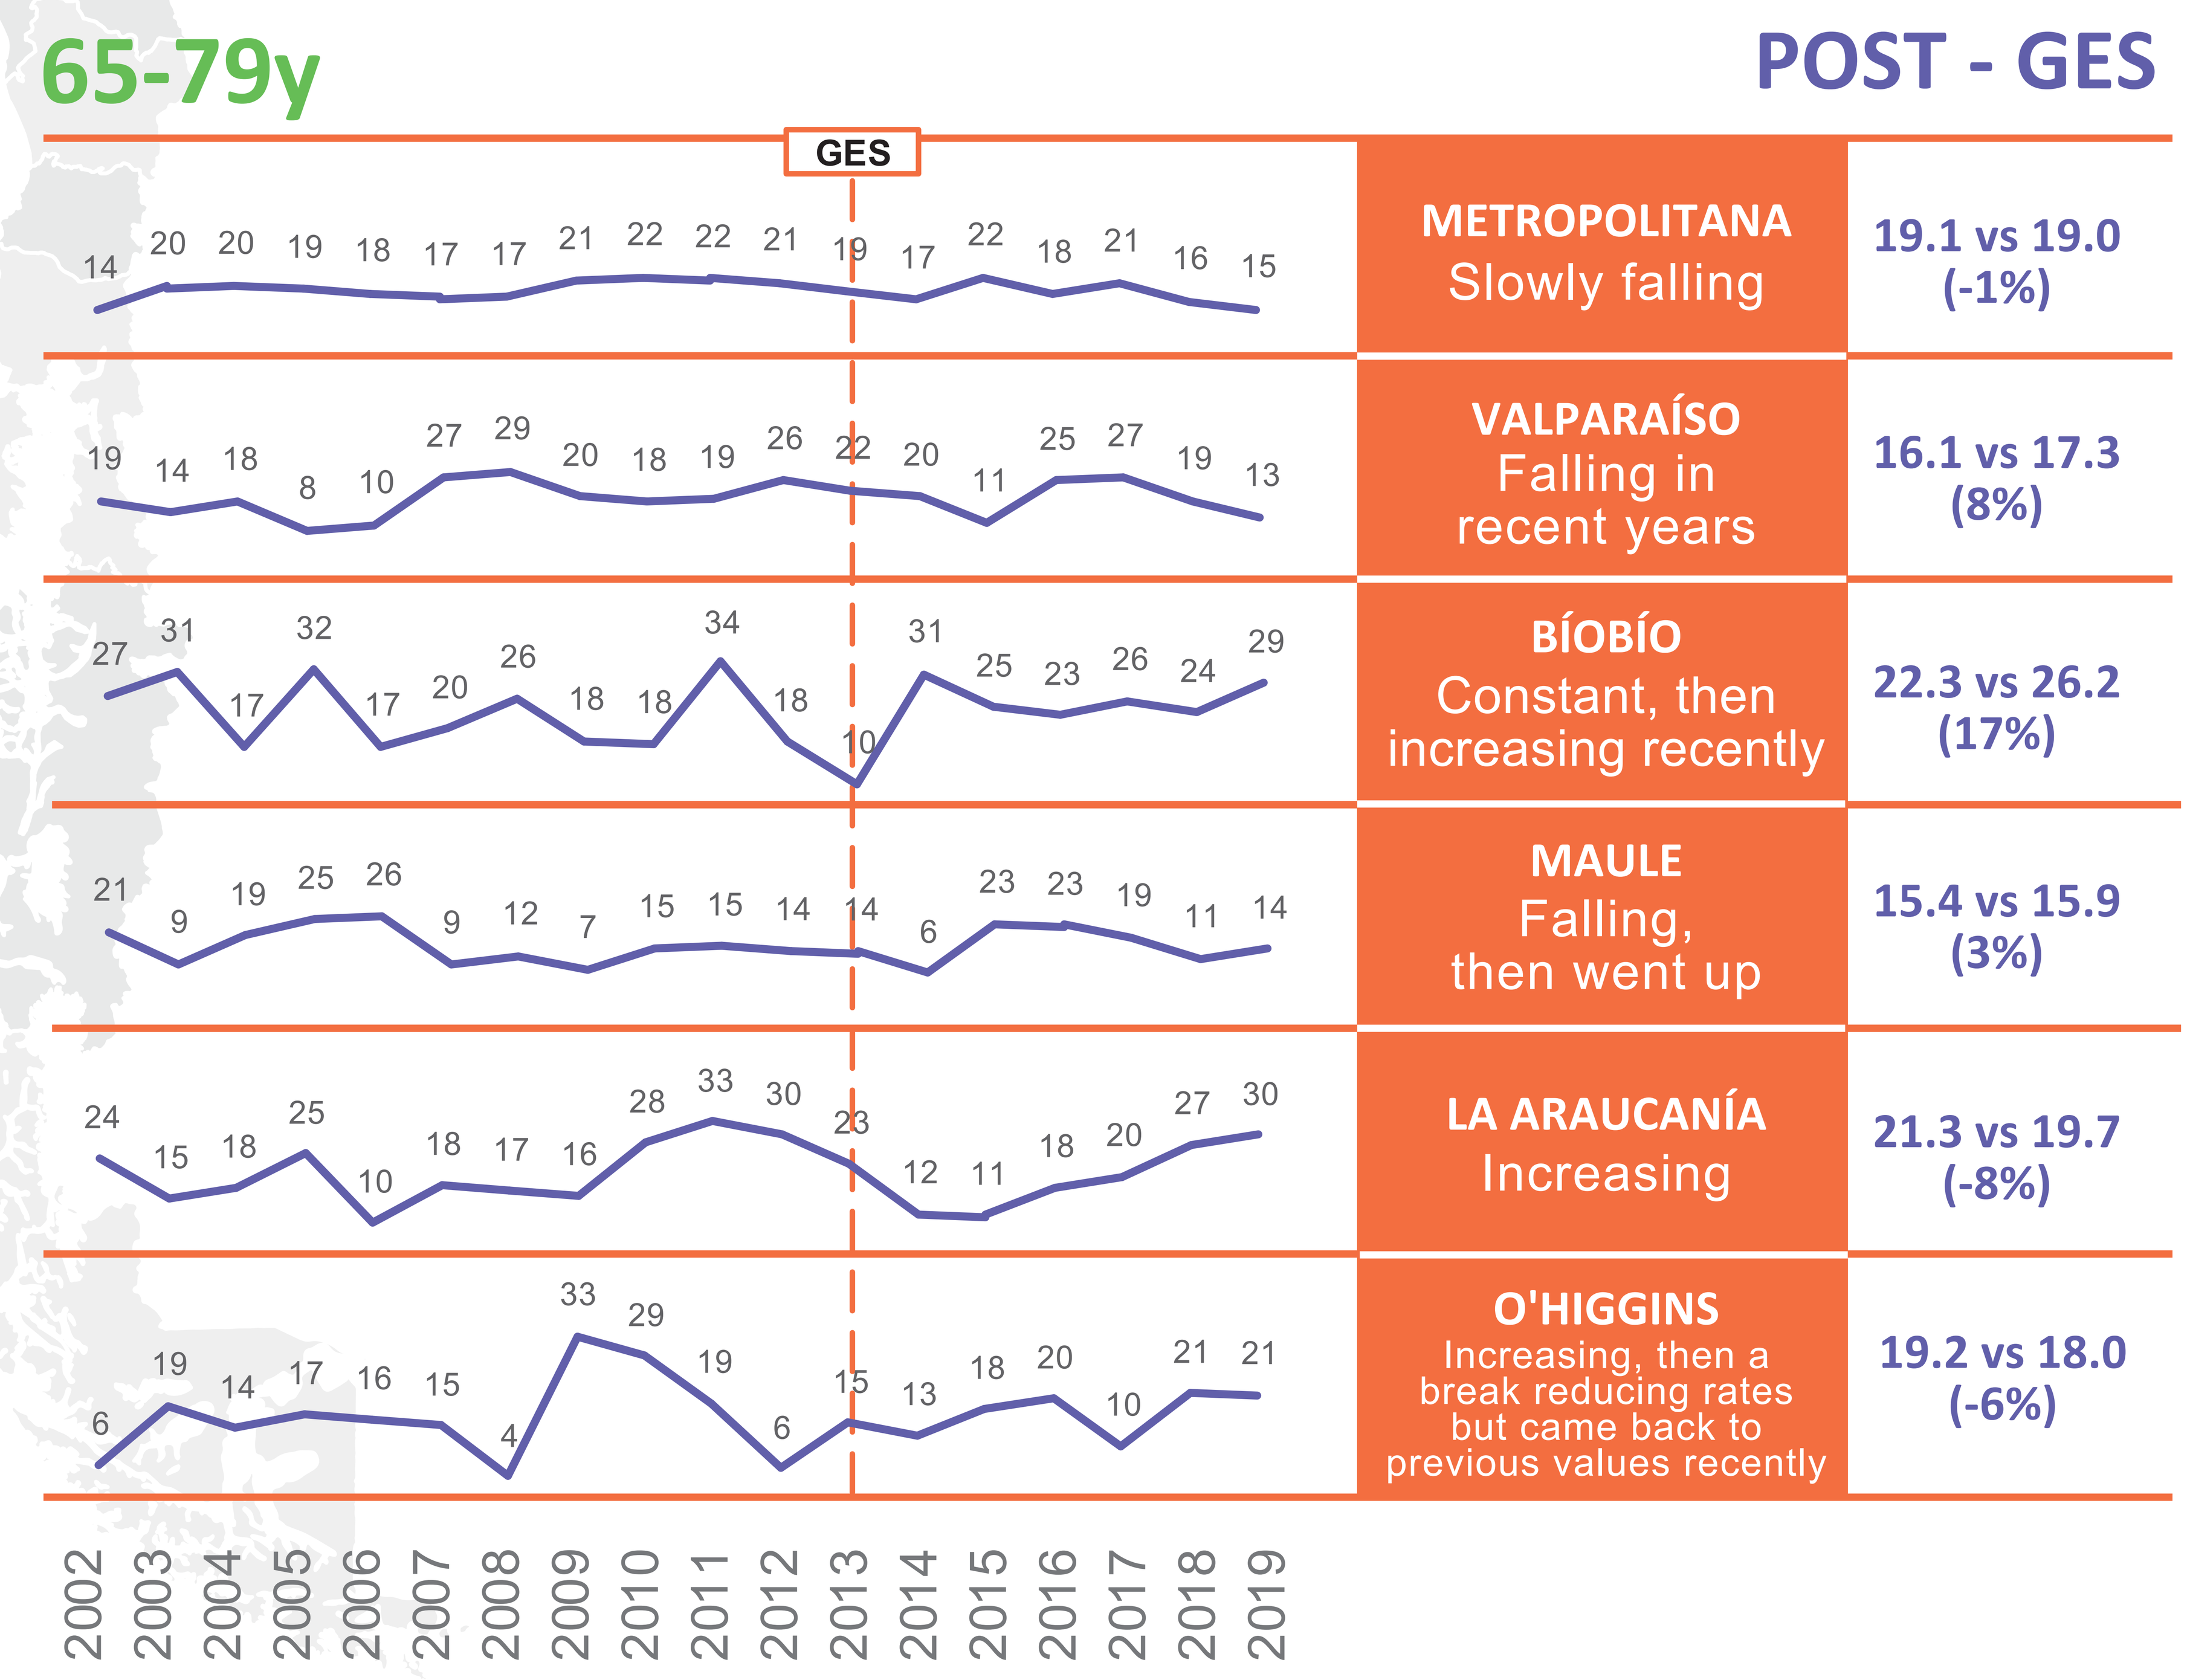
65-79y
POST - GES
GES
14 20 20 19 18 17 17 21 22 22 21 19 17 22 18 21 16 15
METROPOLITANA
Slowly falling
19.1 vs 19.0 (-1%)
19 14 18 8 10 27 29 20 18 19 26 22 20 11 25 27 19 13
VALPARAÍSO
Falling in recent years
16.1 vs 17.3 (8%)
27 31 17 32 17 20 26 18 18 34 18 10 31 25 23 26 24 29
BÍOBÍO
Constant, then increasing recently
22.3 vs 26.2 (17%)
21 9 19 25 26 9 12 7 15 15 14 14 6 23 23 19 11 14
MAULE
Falling, then went up
15.4 vs 15.9 (3%)
24 15 18 25 10 18 17 16 28 33 30 23 12 11 18 20 27 30
LA ARAUCANÍA
Increasing
21.3 vs 19.7 (-8%)
6 19 14 17 16 15 4 33 29 19 6 15 13 18 20 10 21 21
O'HIGGINS
Increasing, then a break reducing rates but came back to previous values recently
19.2 vs 18.0 (-6%)
2002 2003 2004 2005 2006 2007 2008 2009 2010 2011 2012 2013 2014 2015 2016 2017 2018 2019

Conclusions

Mean AA-YPLL rates post-GES had an overall decrease. However, in certain regions rising rates were observed in recent years. These results must be further studied to understand the causes and design appropriate public health actions.

References:
1. Minister of Health. GES 71. Cáncer de ovario epitelial. [cited 2024 Sep 30]. Available at: https://auge.minsal.cl/problemasdesalud/index/71.
2. Minister of Health. Department of Health Statistics and Information. [cited 2024 Sep 30]. Available at: https://deis.minsal.cl/.
3. National Istitute of Statistics. Population Projections. [cited 2024 Sep 30]. Available at: https://www.ine.gob.cl/estadisticas/sociales/demografia-y-vitales/proyecciones-de-poblacion

Acknowledgements:
We would like to acknowledge Elif Coskuncay from GSK, London, UK for her inputs in the poster, Igloolab for their support on poster design and OPEN Health Communications for the coordination of the review & approval, which was funded by GSK, in accordance with Good Publication Practice (GPP) guidelines (www.ismpp.org/gpp-2022).

Disclosures:
This study was funded by GSK (study ID: 222625). Felipe Moraes and Milva Caputo are employed by GSK and do not hold financial equities in GSK. Jose Romero is an employee of GSK and holds financial equities in GSK.

ISPOR Europe | 17-20 Nov 2024 | Barcelona, Spain

Presenting author: Felipe Moraes dos Santos, felipe.s.moraes@gsk.com

GSK