PCR82

Care pathways of surgical patients with Renal Cell Carcinoma (RCC) in Greek Public Hospitals

Topic: Patient-Centered Research, SDC: Oncology, STA: Surgery

Kaitelidou D[1], Mitropoulos D[2], Kydonaki K[2], Spanomichou D[2], Katsikatsos P2, Petsioti S[2], Siskou O[3], Galanis P[4], Peleka P[4], Charalambous G[5], Mangioris G[4], Angelopoulou DM[1], Karaiskou A[1], KolliopoulouG[1], Kostaras D6, Karokis A[6], Yfantopoulos N[6], Gountas I[6], Lazouras K[6], Emmanouil G[6], Paloukis K[6], **Konstantakopoulou O[1]**

[1]Center for Health Services Management and Evaluation, Nursing Department, National and Kapodistrian University of Athens, Athens, Greece, [2]Laiko" University hospital of Athens, Athens, Greece, [3]University of Piraeus, Piraeus, Greece, [4]National and Kapodistrian University of Athens, Athens, Greece, [5]Hippocratio Hospital of Athens, ATHENS, Greece, [6]MSD Greece, Athens, Greece,

Introduction

- Healthcare process mapping is a new and important form of clinical audit that examines how hospitals manage the patients' journey, identify problems and suggest improvements. Understanding how the patient moves within the healthcare system is a particularly important factor in identifying gaps and barriers that hinder patients' access to healthcare services.[1]
- Mapping the patients' journey constitutes one of the most effective ways to determine patients' navigation problems in health services, including gaps and barriers that hinder their prompt access to care.[2]

Objectives

- The aim of this study was to map Renal Cell Carcinoma (RCC) surgical patients' journey in the Greek health system, to explore key stakeholders' perceptions and identify best practices to streamline the patient journey process.
- Here, we present study outcomes based on observations of the RCC patients' journey within a Greek public hospital's surgical unit and on health professionals' perspective.

Methods

- An observational study design based on the Lean Thinking principles and a qualitative design with semi-structured interviews were used.
- Study population consisted of 24 patients and 8 healthcare staff members from the public hospital's surgical unit.
- Data was collected in 2023 in collaboration with hospital key stakeholders.

Results

- In total, 24 RCC surgical patients were included in the study. The mean age of the study patients was 68.3 years (SD=9.9), 54.2% were men and 95.8% had Greek nationality. Almost 4 out of 10 patients (37.5%) resided in a different district from this cancer center, and 83.3% had public health insurance while 8.3% had no insurance. Almost 9 out of 10 patients (87.5%) suffered from some other chronic disease and 25% had to stop (either permanently – 8.3% - or temporarily – 16.7%) working because of the disease.
- Regarding the pre-hospital journey, 25% of patients reported delays of over 3 months in contacting a PCP/GP after noticing that something was wrong. Additionally, 16.7% experienced long delays in getting diagnostic tests and referrals to specialists. System delays in starting treatment affected 25% of patients due to unavailability of specific surgeons, operating rooms, and ICU beds.

- The steps of the procedure for the admission of the patient to the hospital, to get operated on as planned, are shown in **Figure 1**.
- For the in-hospital journey, based on the three time-estimates, i.e., the Optimistic Time (O) (the minimum time required to complete an activity under the best possible conditions), the Most Likely Time (M) (the most realistic estimate of the time required based on normal conditions and past experience), and the Pessimistic Time (P) (the maximum time required under the worst possible conditions), the Expected Time (ET) admission procedure time was calculated, which was 78.2 minutes in total. Longer waiting times were observed until the medical history recording and completing their admission papers (up to 39 minutes) (**Table 1**).

- Health care professionals identified the main challenges as:
 - a lack of nursing, medical, and administrative staff, and
 - limited availability of operating rooms for RCC surgical patients.

Figure 1. The steps of the procedure for the admission of the patient to the hospital, to get operated on as planned

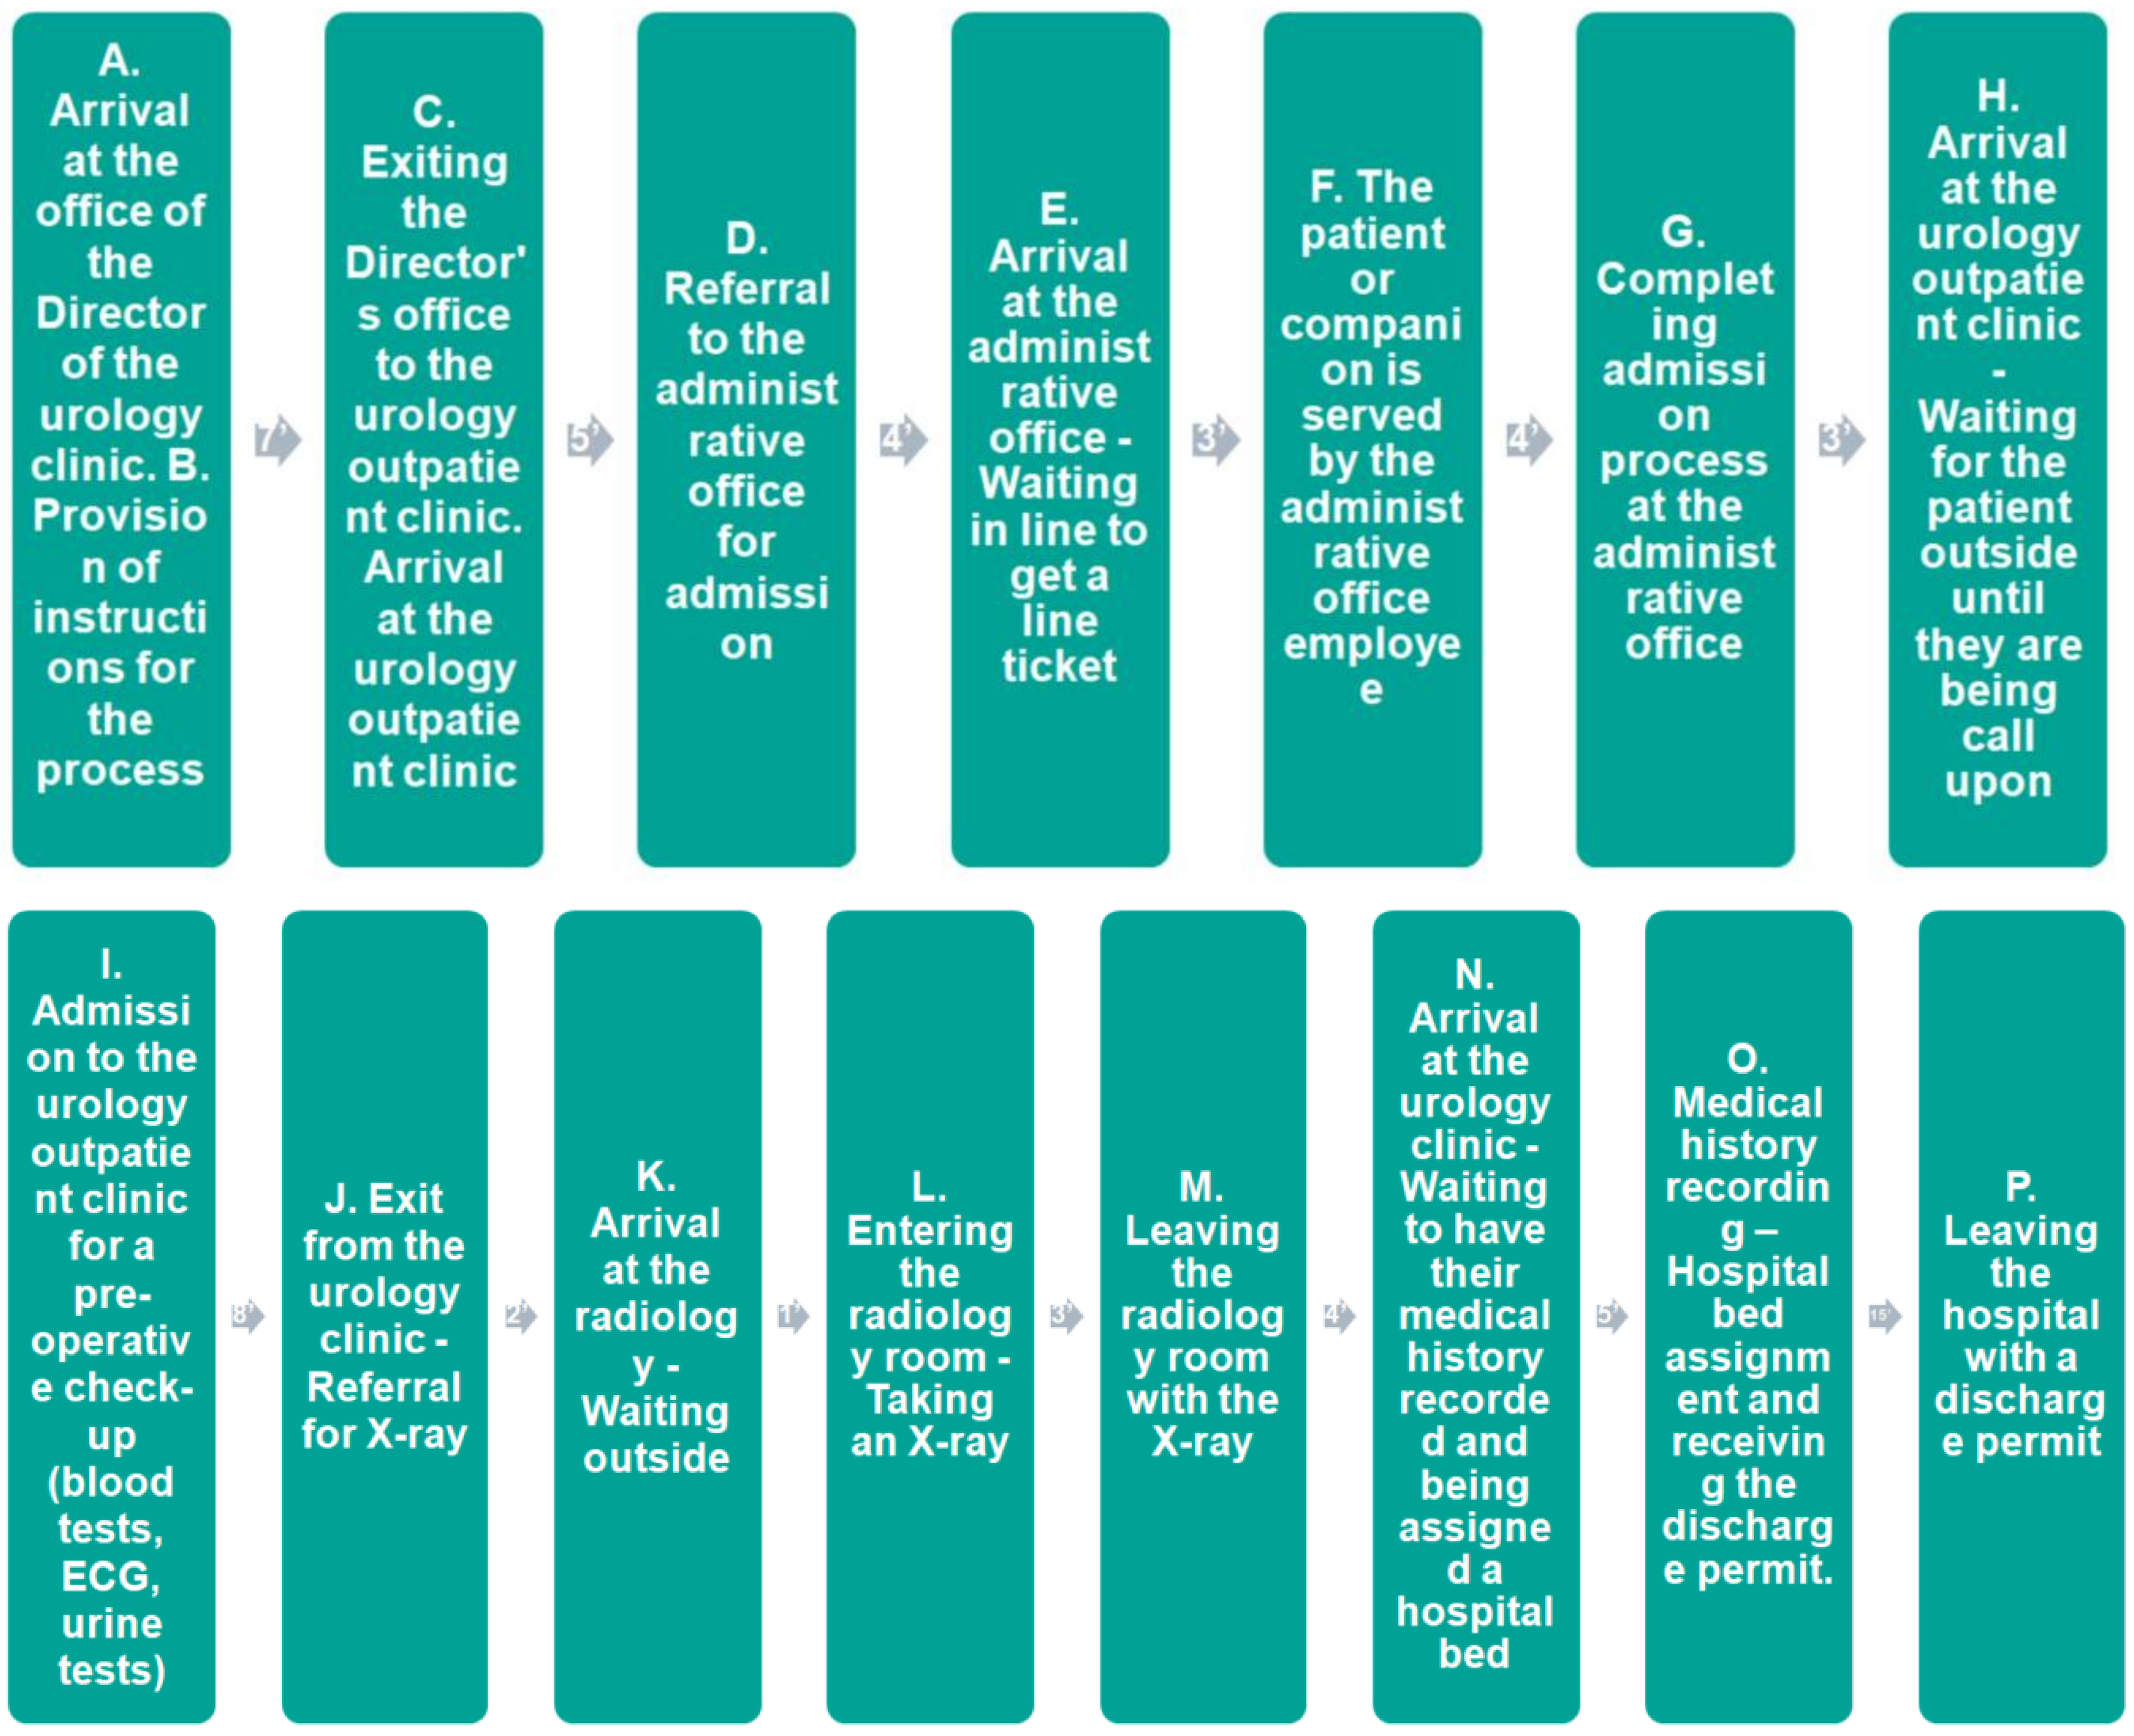
A. Arrival at the office of the Director of the urology clinic. B. Provision of instructions for the process
C. Exiting the Director's office to the urology outpatient clinic. Arrival at the urology outpatient clinic
D. Referral to the administrative office for admission
E. Arrival at the administrative office - Waiting in line to get a line ticket
F. The patient or companion is served by the administrative office employee
G. Completing admission process at the administrative office
H. Arrival at the urology outpatient clinic - Waiting for the patient outside until they are being call upon
I. Admission to the urology outpatient clinic for a pre-operative check-up (blood tests, ECG, urine tests)
J. Exit from the urology clinic - Referral for X-ray
K. Arrival at the radiology - Waiting outside
L. Entering the radiology room - Taking an X-ray
M. Leaving the radiology room with the X-ray
N. Arrival at the urology clinic - Waiting to have their medical history recorded and being assigned a hospital bed
O. Medical history recording – Hospital bed assignment and receiving the discharge permit.
P. Leaving the hospital with a discharge permit

Table 1. Duration of the procedure for the admission of the patient to the hospital, to get operated on as planned (minimum, maximum, median, and expected time per step of the procedure)

	Optimistic Time (O) Minimum time (mins)	Pessimistic Time (P) Maximum time (mins)	Most Likely Time (M) Median time (mins)	Expected Time (ET) ET = (O + 4M + P) / 6
A. Arrival at the office of the Director of the urology clinic				
B. Provision of instructions for the process	7	10	7	7.5
C. Exiting the Director's office to the urology outpatient clinic. Arrival at the urology outpatient clinic	3	7	5	5.0
D. Referral to the administrative office for admission.	2	6	4	4.0
E. Arrival at the administrative office - Waiting in line to get a line ticket	0	10	3	3.7
F. The patient or companion is served by the administrative office employee.	3	4	4	3.8
G. Completing admission process at the administrative office.	1	3	3	2.7
H. Arrival at the urology outpatient clinic - Waiting for the patient outside until they are being call upon.	1	23	2	5.3
I. Admission to the urology outpatient clinic for a pre-operative check-up (blood tests, ECG, urine tests).	7	10	8	8.2
J. Exit from the urology clinic - Referral for X-ray.	1	11	2	3.3
K. Arrival at the radiology - Waiting outside.	1	8	1	2.2
L. Entering the radiology room - Taking an X-ray.	2	7	3	3.5
M. Leaving the radiology room with the X-ray.	3	7	4	4.3
N. Arrival at the urology clinic - Waiting to have their medical history recorded and being assigned a hospital bed.	4	39	5	10.5
O. Medical history recording – Hospital bed assignment and receiving the discharge permit.	10	15	15	14.2
P. Leaving the hospital with a discharge permit.				
TOTAL	**45.0**	**160.0**	**66.0**	**78.2**

Conclusions

- Addressing the challenges in the provision of care to RCC surgical patients may require multidisciplinary efforts, including investments in healthcare infrastructure, staffing, and administrative processes.
- The benefits refer to reducing delays and enhancing patient outcomes and quality of healthcare for RCC patients.

Disclosures

This study was funded by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA

References

1. Simonse LWL, Albayrak A, Starre S. *Patient journey method for integrated service design* 2019, Design for Health; **3**(1).
2. Trebble TM, Hansi N, Hydes T, Smith MA, Baker M. *Process mapping the patient journey: an introduction* 2010; BMJ **341**.

Copyright © 2024 Merck & Co., Inc., Rahway, USA and its affiliates. All rights reserved