EE196

Has the introduction of PARPis changed healthcare resource utilisation (HCRU) and medical costs among patients with high-grade serous ovarian cancer (HGSOC) in the real-life setting in Finland?

Barbara Mascialino[1], Mari Lahelma[2], Karita Salo[2], Matti Rajala[2], Eija Heikkilä[2], Fredrik Herse[2], Juhana Idänpään-Heikkilä[3], Sari Käkelä[3], Saara Tikka[3], Dirk Schneider[4], Sakari Hietanen[5], Mikko Loukovaara[6], Annika Auranen[7]

[1]GSK, Verona, Italy; [2]Nordic Healthcare Group, Helsinki, Finland; [3]GSK, Espoo, Finland; [4]GSK, Baar Onyx, Switzerland; [5]Turku University Hospital, Turku, Finland; [6]Helsinki University Hospital, Helsinki, Finland; [7]Tays Cancer Centre, Tampere University Hospital and Tampere University, Tampere, Finland

Digital poster

SCAN ME

Background

- OC is the eighth most common cancer in women globally and, in Finland, approximately 457 new cases of OC and 300 deaths from OC were reported in 2022.[1,2]
- Standard of care treatment of OC has shifted from chemotherapy and bevacizumab to PARPis and combination strategies.[3]
- Real-world outcomes data are important to understand the clinical relevance of new treatment approaches.
- OCRWE-Finland was a retrospective study that assessed the real-world burden of disease, treatment patterns, outcomes and HCRU for patients with OC in Finland.
 - Data from OCRWE-Finland using medical records from 2014–2019, during a time when bevacizumab was the only available maintenance treatment for OC, showed that mean HCRU cost per patient during the first year after diagnosis was highest for patients with Stage III–IV HGSOC with visible residual disease (€23,700).[4]

Aims

- The aim of this analysis was to provide updated data from OCRWE-Finland, over a more recent time period (2019–2023), to analyse the impact of PARPis on HCRU and associated medical costs for patients with HGSOC living in Finland.

Methods

- This multicentre, retrospective, non-interventional study collected medical records from Helsinki, Turku and Tampere University Hospitals.
- Patients with newly diagnosed HGSOC who received treatment at these hospitals during 2019–2023 were included, covering around 50% of patients with HGSOC in Finland.
- Patient demographics and characteristics, treatment patterns, HCRU outcomes (outpatient visits, emergency department visits, inpatient admissions) and costs were collected and analysed.

Results

- This analysis included 738 patients with HGSOC diagnosed between 2019 and 2023.
- Patient demographic and clinical characteristics are detailed in **Table 1**.
 - Mean (SD) age was 70 (10) years.
 - 72.3% of patients had Stage III or IV disease at diagnosis.
 - *BRCA* mutation testing rate was low; 49.2% of patients had unknown *BRCA* status, 43.1% of patients were *BRCA*wt and 7.7% were *BRCA*m.

Table 1: Baseline demographics and clinical characteristics

	Patients (N=738)
Demographic characteristics	
Mean age at first diagnosis (SD), years	70.3 (10.2)
Age at diagnosis, n (%)	
<40 years	5 (0.7)
40–59 years	104 (14.1)
60–79 years	503 (68.2)
≥80 years	126 (17.1)
BMI, n (%)	
Underweight: <18.5 kg/m²	12 (1.6)
Normal weight: 18.5–24.9 kg/m²	152 (20.6)
Overweight: 25–29.9 kg/m²	106 (14.4)
Obese: >30.0 kg/m²	69 (9.3)
Unknown	399 (54.1)
Year of initial OC diagnosis, n (%)	
2019	173 (23.4)
2020	129 (17.5)
2021	139 (18.8)
2022	147 (19.9)
2023	150 (20.3)
Geographic region, n (%)	
Helsinki	389 (52.7)
Tampere	180 (24.4)
Turku	169 (22.9)
Clinical characteristics	
FIGO stage at initial diagnosis, n (%)	
I	38 (5.1)
II	31 (4.2)
III	258 (35.0)
IV	275 (37.3)
Unknown	136 (18.4)
BRCA mutation status, n (%)	
*BRCA*m	57 (7.7)
*BRCA*wt	318 (43.1)
Unknown	363 (49.2)
Residual tumour, n (%)	
No visible residual disease	215 (29.1)
Visible residual disease	153 (20.7)
Unknown	93 (12.6)
Patients without surgery	277 (37.5)
HRD status, n (%)	
Positive	68 (9.2)
Negative	105 (14.2)
Unknown	565 (76.6)

Treatment patterns

- Surgery plus adjuvant chemotherapy (42.1%) was the most common treatment used in 1L (**Figure 1**).
- In the 1L maintenance setting, patients received either bevacizumab (41.1%), PARPi (3.6%), bevacizumab plus PARPi (8.5%) or active surveillance (46.7%) (**Figure 2**).
- Treatment in 2L and 3L comprised platinum-based chemotherapy or other chemotherapy, although a large proportion of surviving patients received no 2L or 3L treatment (50.1% and 71.7% of patients, respectively; **Figure 1**).

Figure 1: Treatment by line of therapy

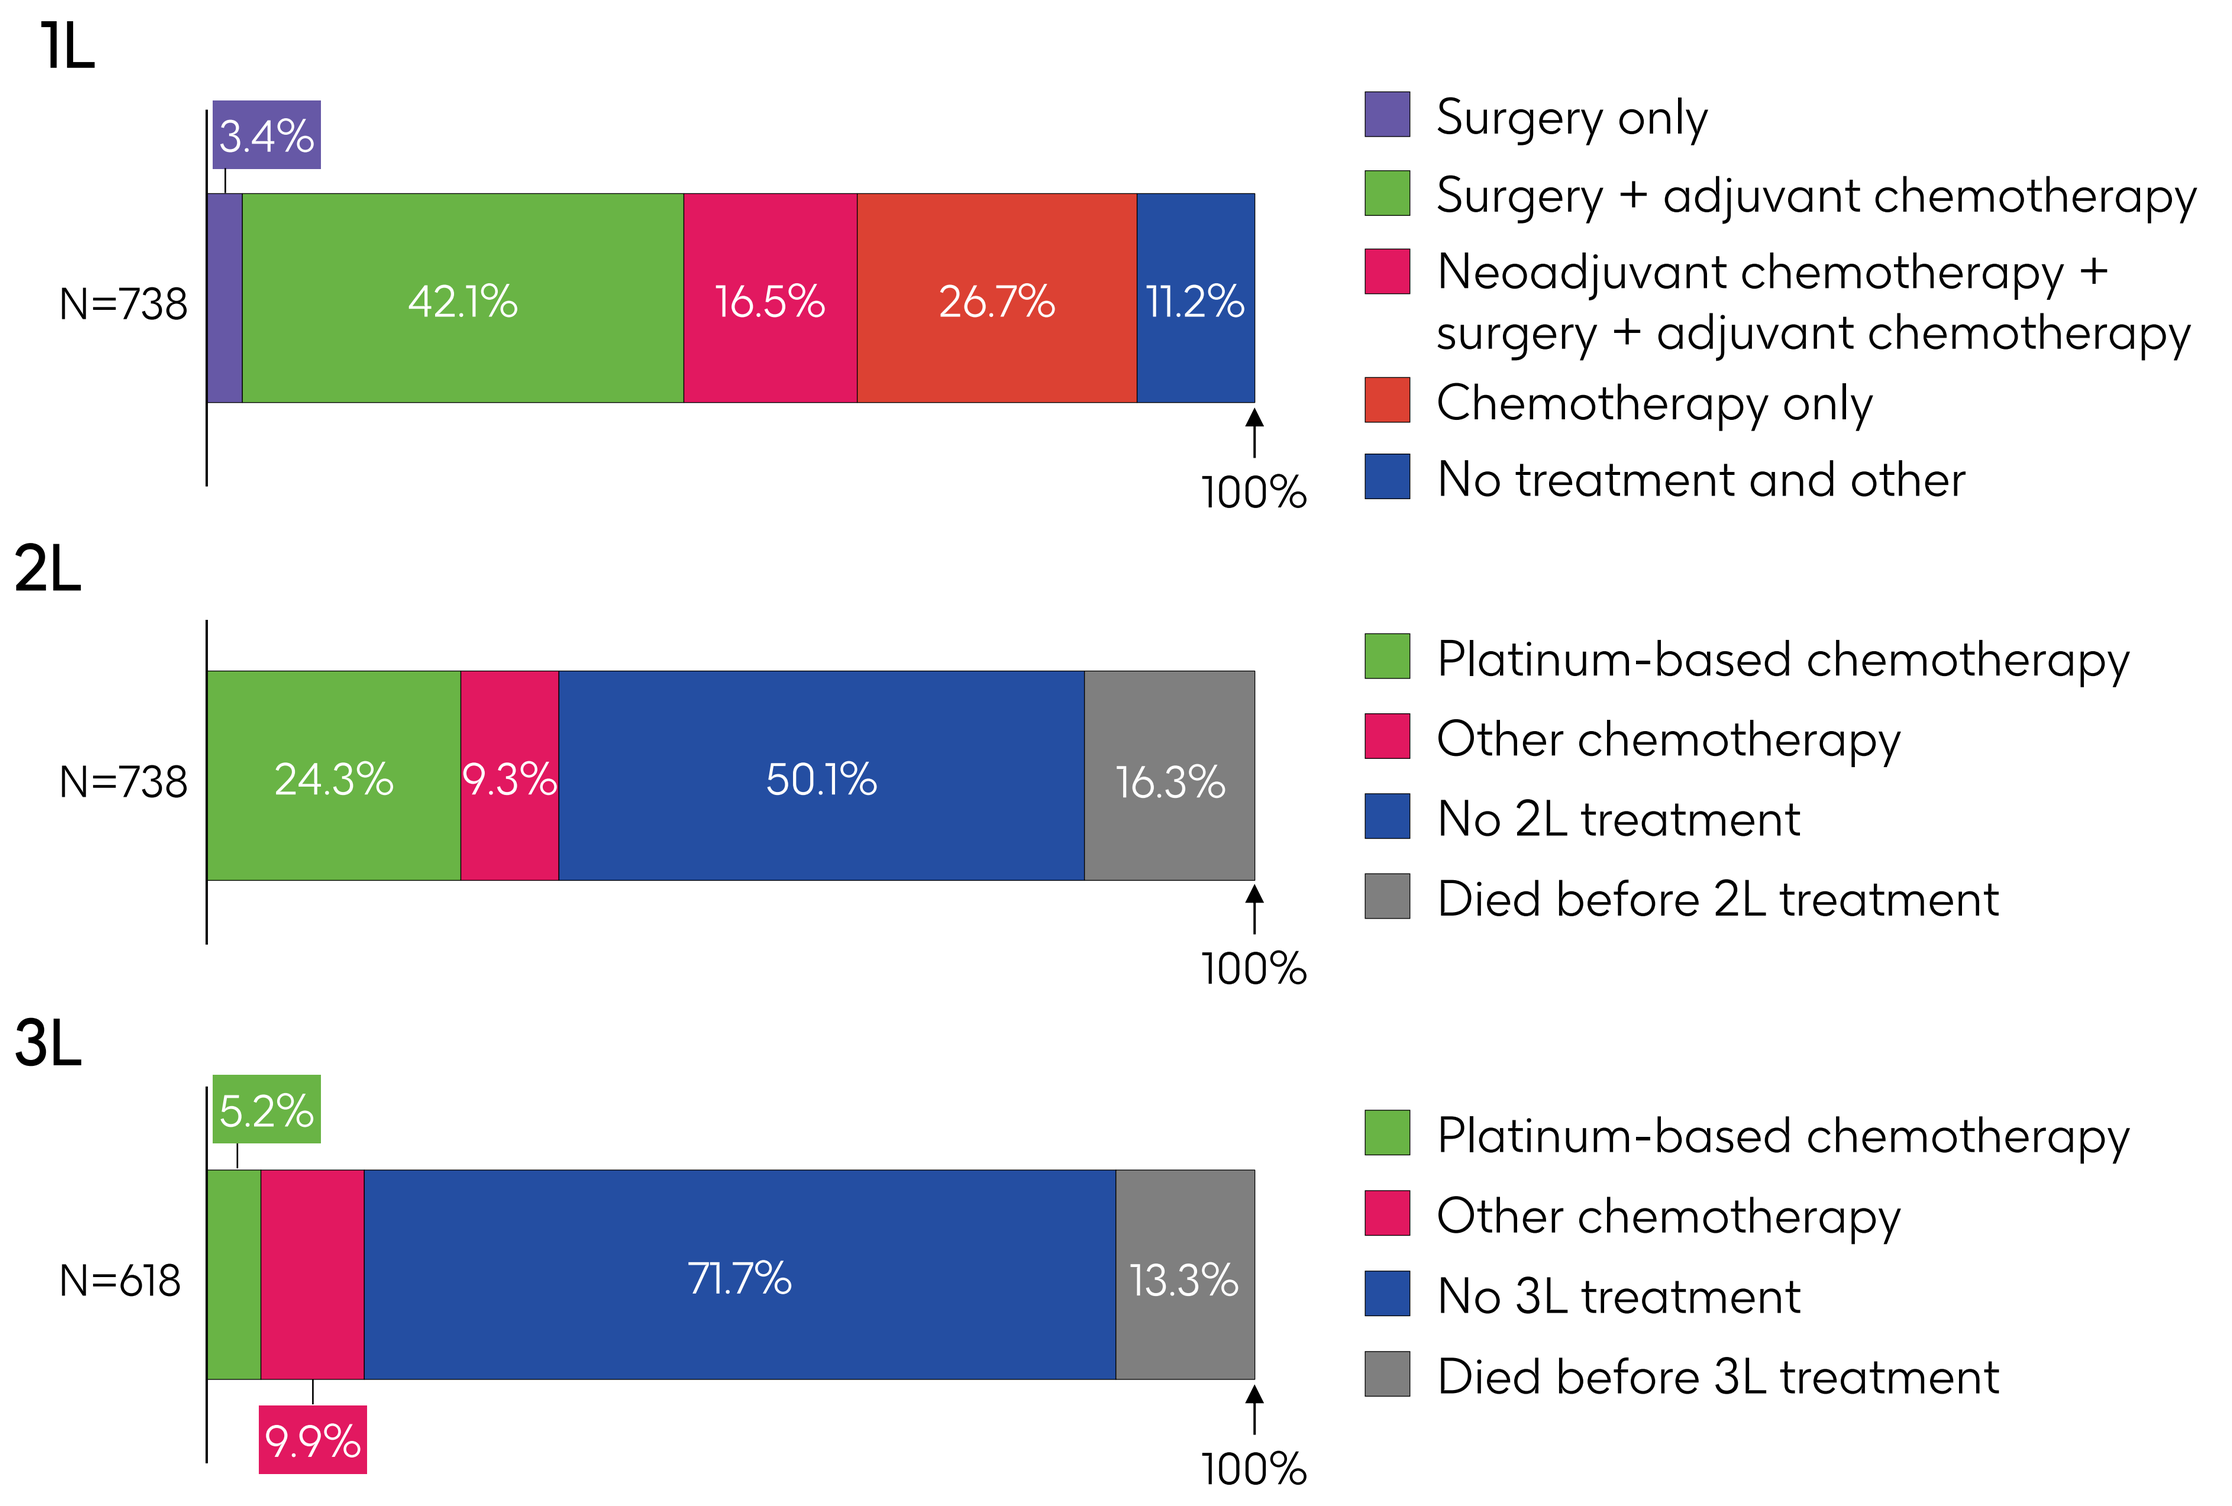
1L
N=738
3.4%
42.1%
16.5%
26.7%
11.2%
100%
Surgery only
Surgery + adjuvant chemotherapy
Neoadjuvant chemotherapy + surgery + adjuvant chemotherapy
Chemotherapy only
No treatment and other
2L
N=738
24.3%
9.3%
50.1%
16.3%
100%
Platinum-based chemotherapy
Other chemotherapy
No 2L treatment
Died before 2L treatment
3L
N=618
5.2%
9.9%
71.7%
13.3%
100%
Platinum-based chemotherapy
Other chemotherapy
No 3L treatment
Died before 3L treatment

- The most common 2L maintenance treatments were bevacizumab alone (11.4% of patients) or PARPi alone (10.9% of patients; **Figure 2**).
- The most common 3L maintenance regimen was bevacizumab plus PARPi (10.3% of patients; **Figure 2**)

Figure 2: Maintenance treatment by line of therapy

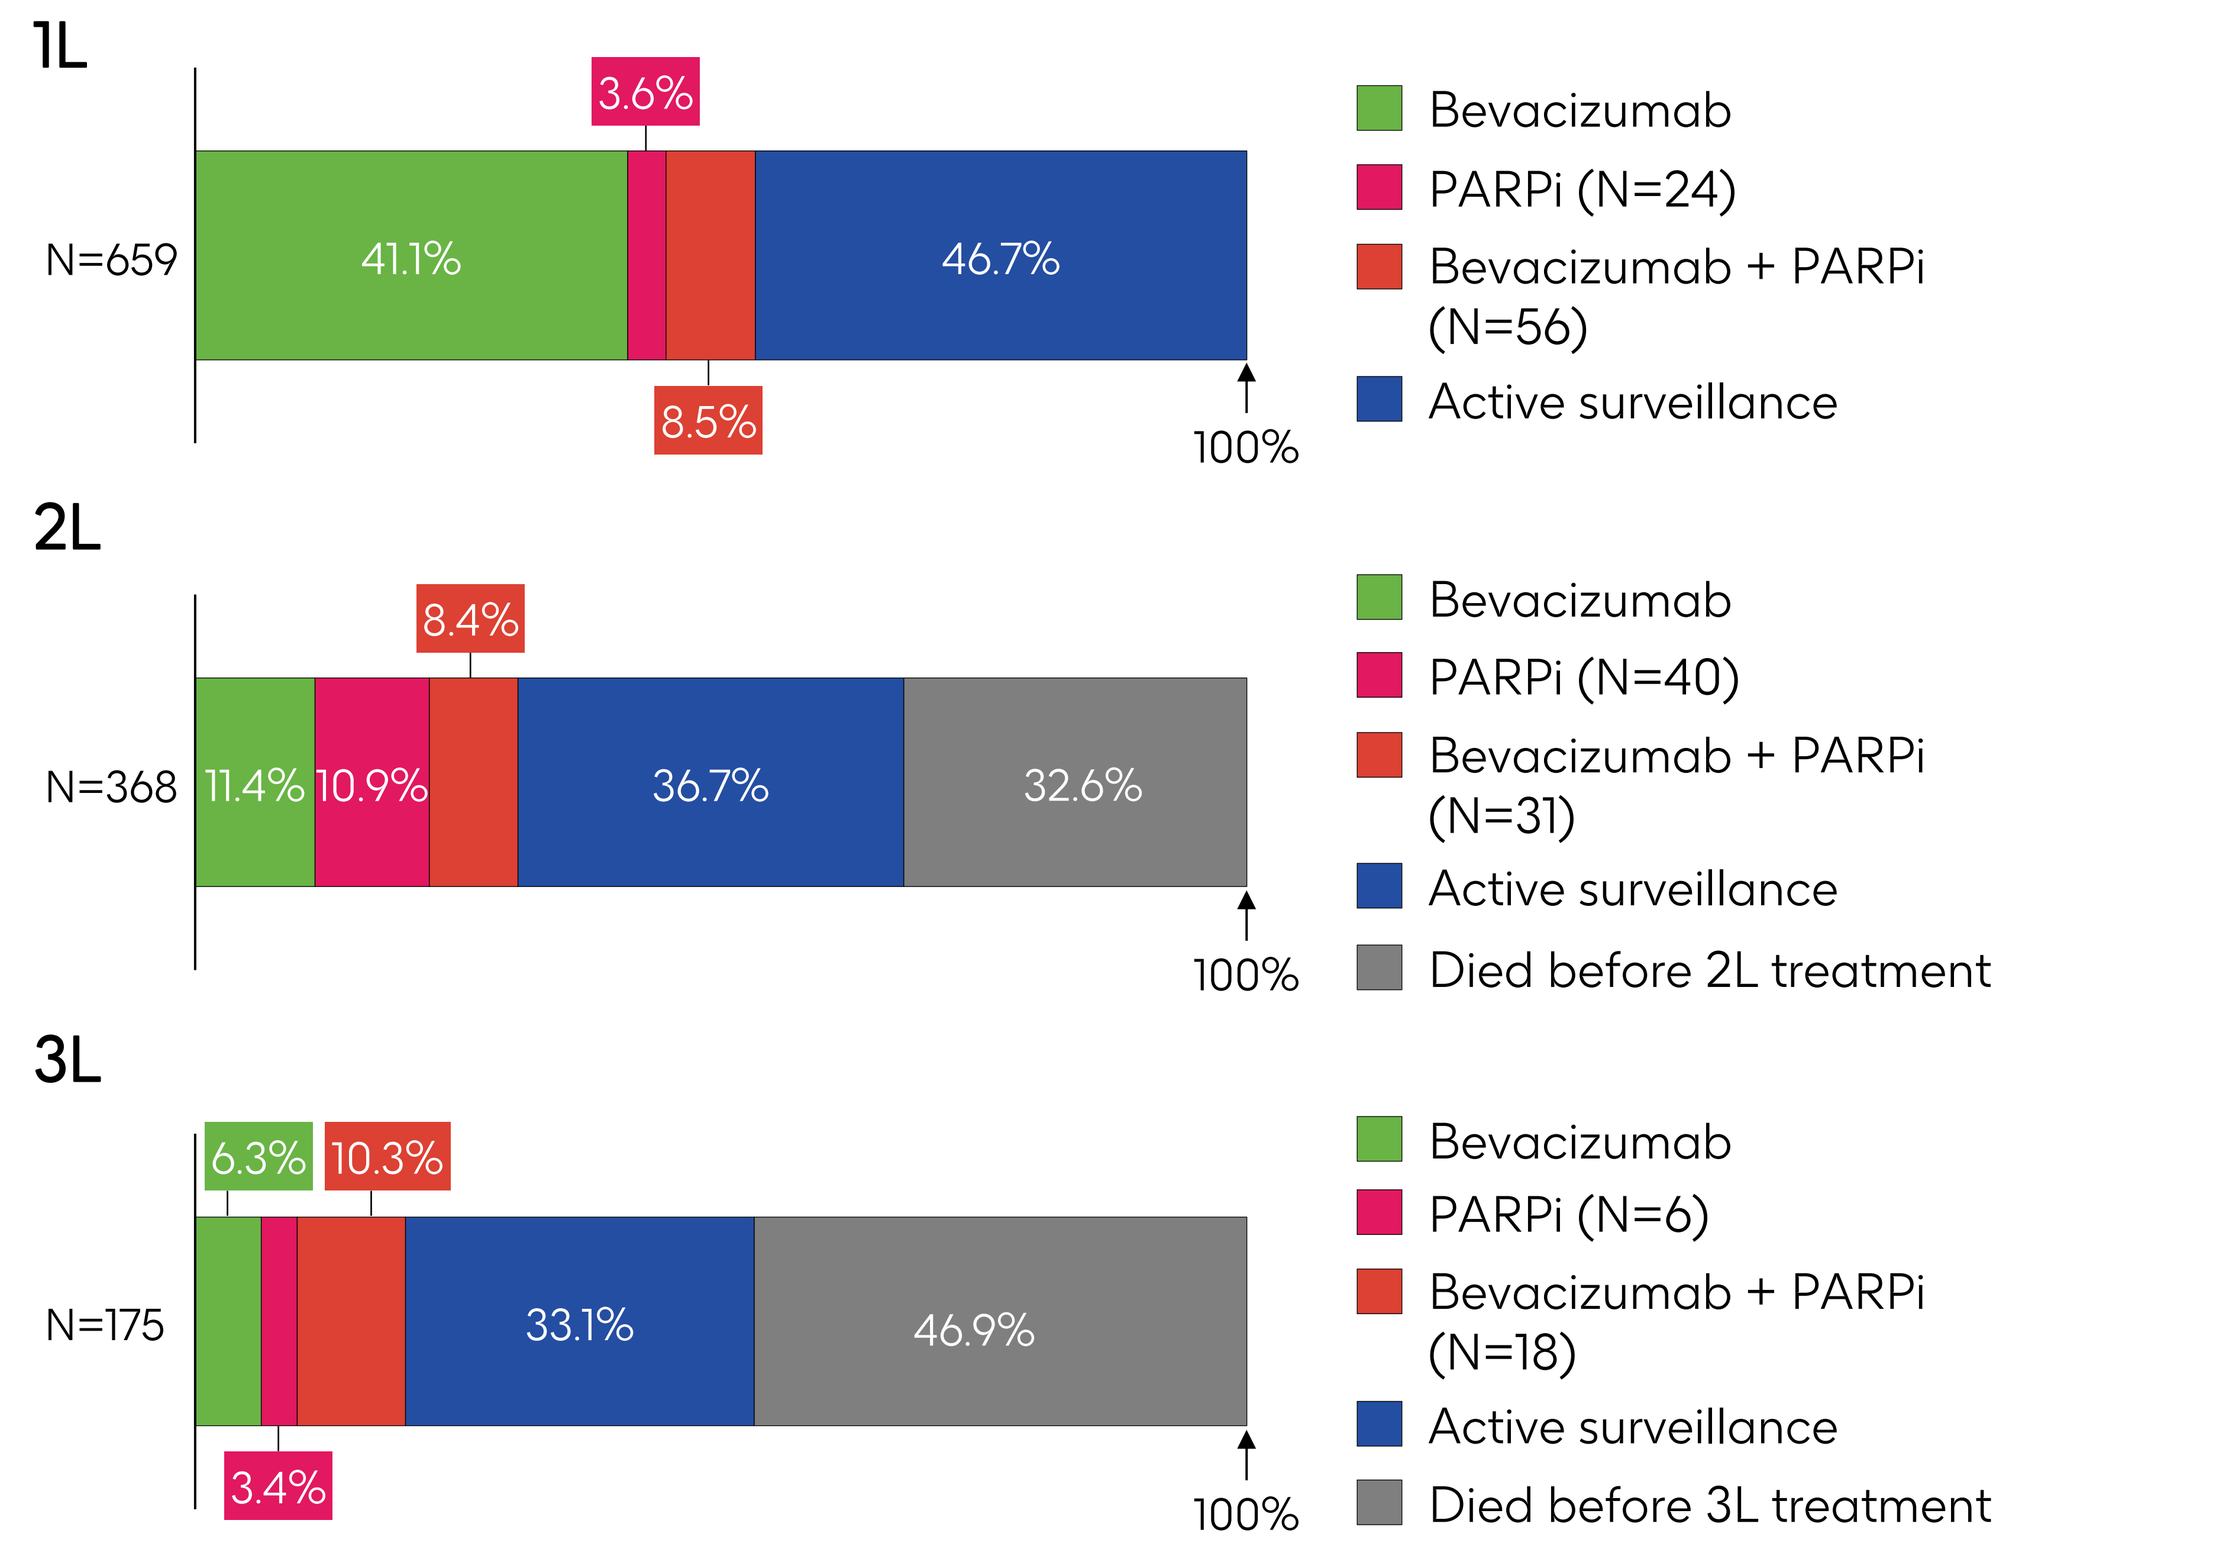
1L
N=659
3.6%
41.1%
46.7%
8.5%
100%
Bevacizumab
PARPi (N=24)
Bevacizumab + PARPi (N=56)
Active surveillance
2L
N=368
8.4%
11.4%
10.9%
36.7%
32.6%
100%
Bevacizumab
PARPi (N=40)
Bevacizumab + PARPi (N=31)
Active surveillance
Died before 2L treatment
3L
N=175
6.3%
10.3%
33.1%
46.9%
3.4%
100%
Bevacizumab
PARPi (N=6)
Bevacizumab + PARPi (N=18)
Active surveillance
Died before 3L treatment

HCRU

- HCRU was highest during the first year following diagnosis (**Figure 3A–C**).
- During the first year after HGSOC diagnosis, among patients receiving maintenance therapy:
 - Patients receiving PARPi monotherapy had the lowest average number of outpatient visits per patient (10.8), while the rate of outpatient visits was highest in patients receiving bevacizumab monotherapy (17.0) (**Figure 3A**).
 - Average rate of emergency department visits per patient during the first treatment year was also lower in patients receiving PARPi monotherapy (0.5) than in patients receiving bevacizumab monotherapy (0.9) (**Figure 3B**).
 - Patients had around one inpatient admission, regardless of the maintenance therapy regimen received (**Figure 3C**).

Figure 3: HCRU outcomes†

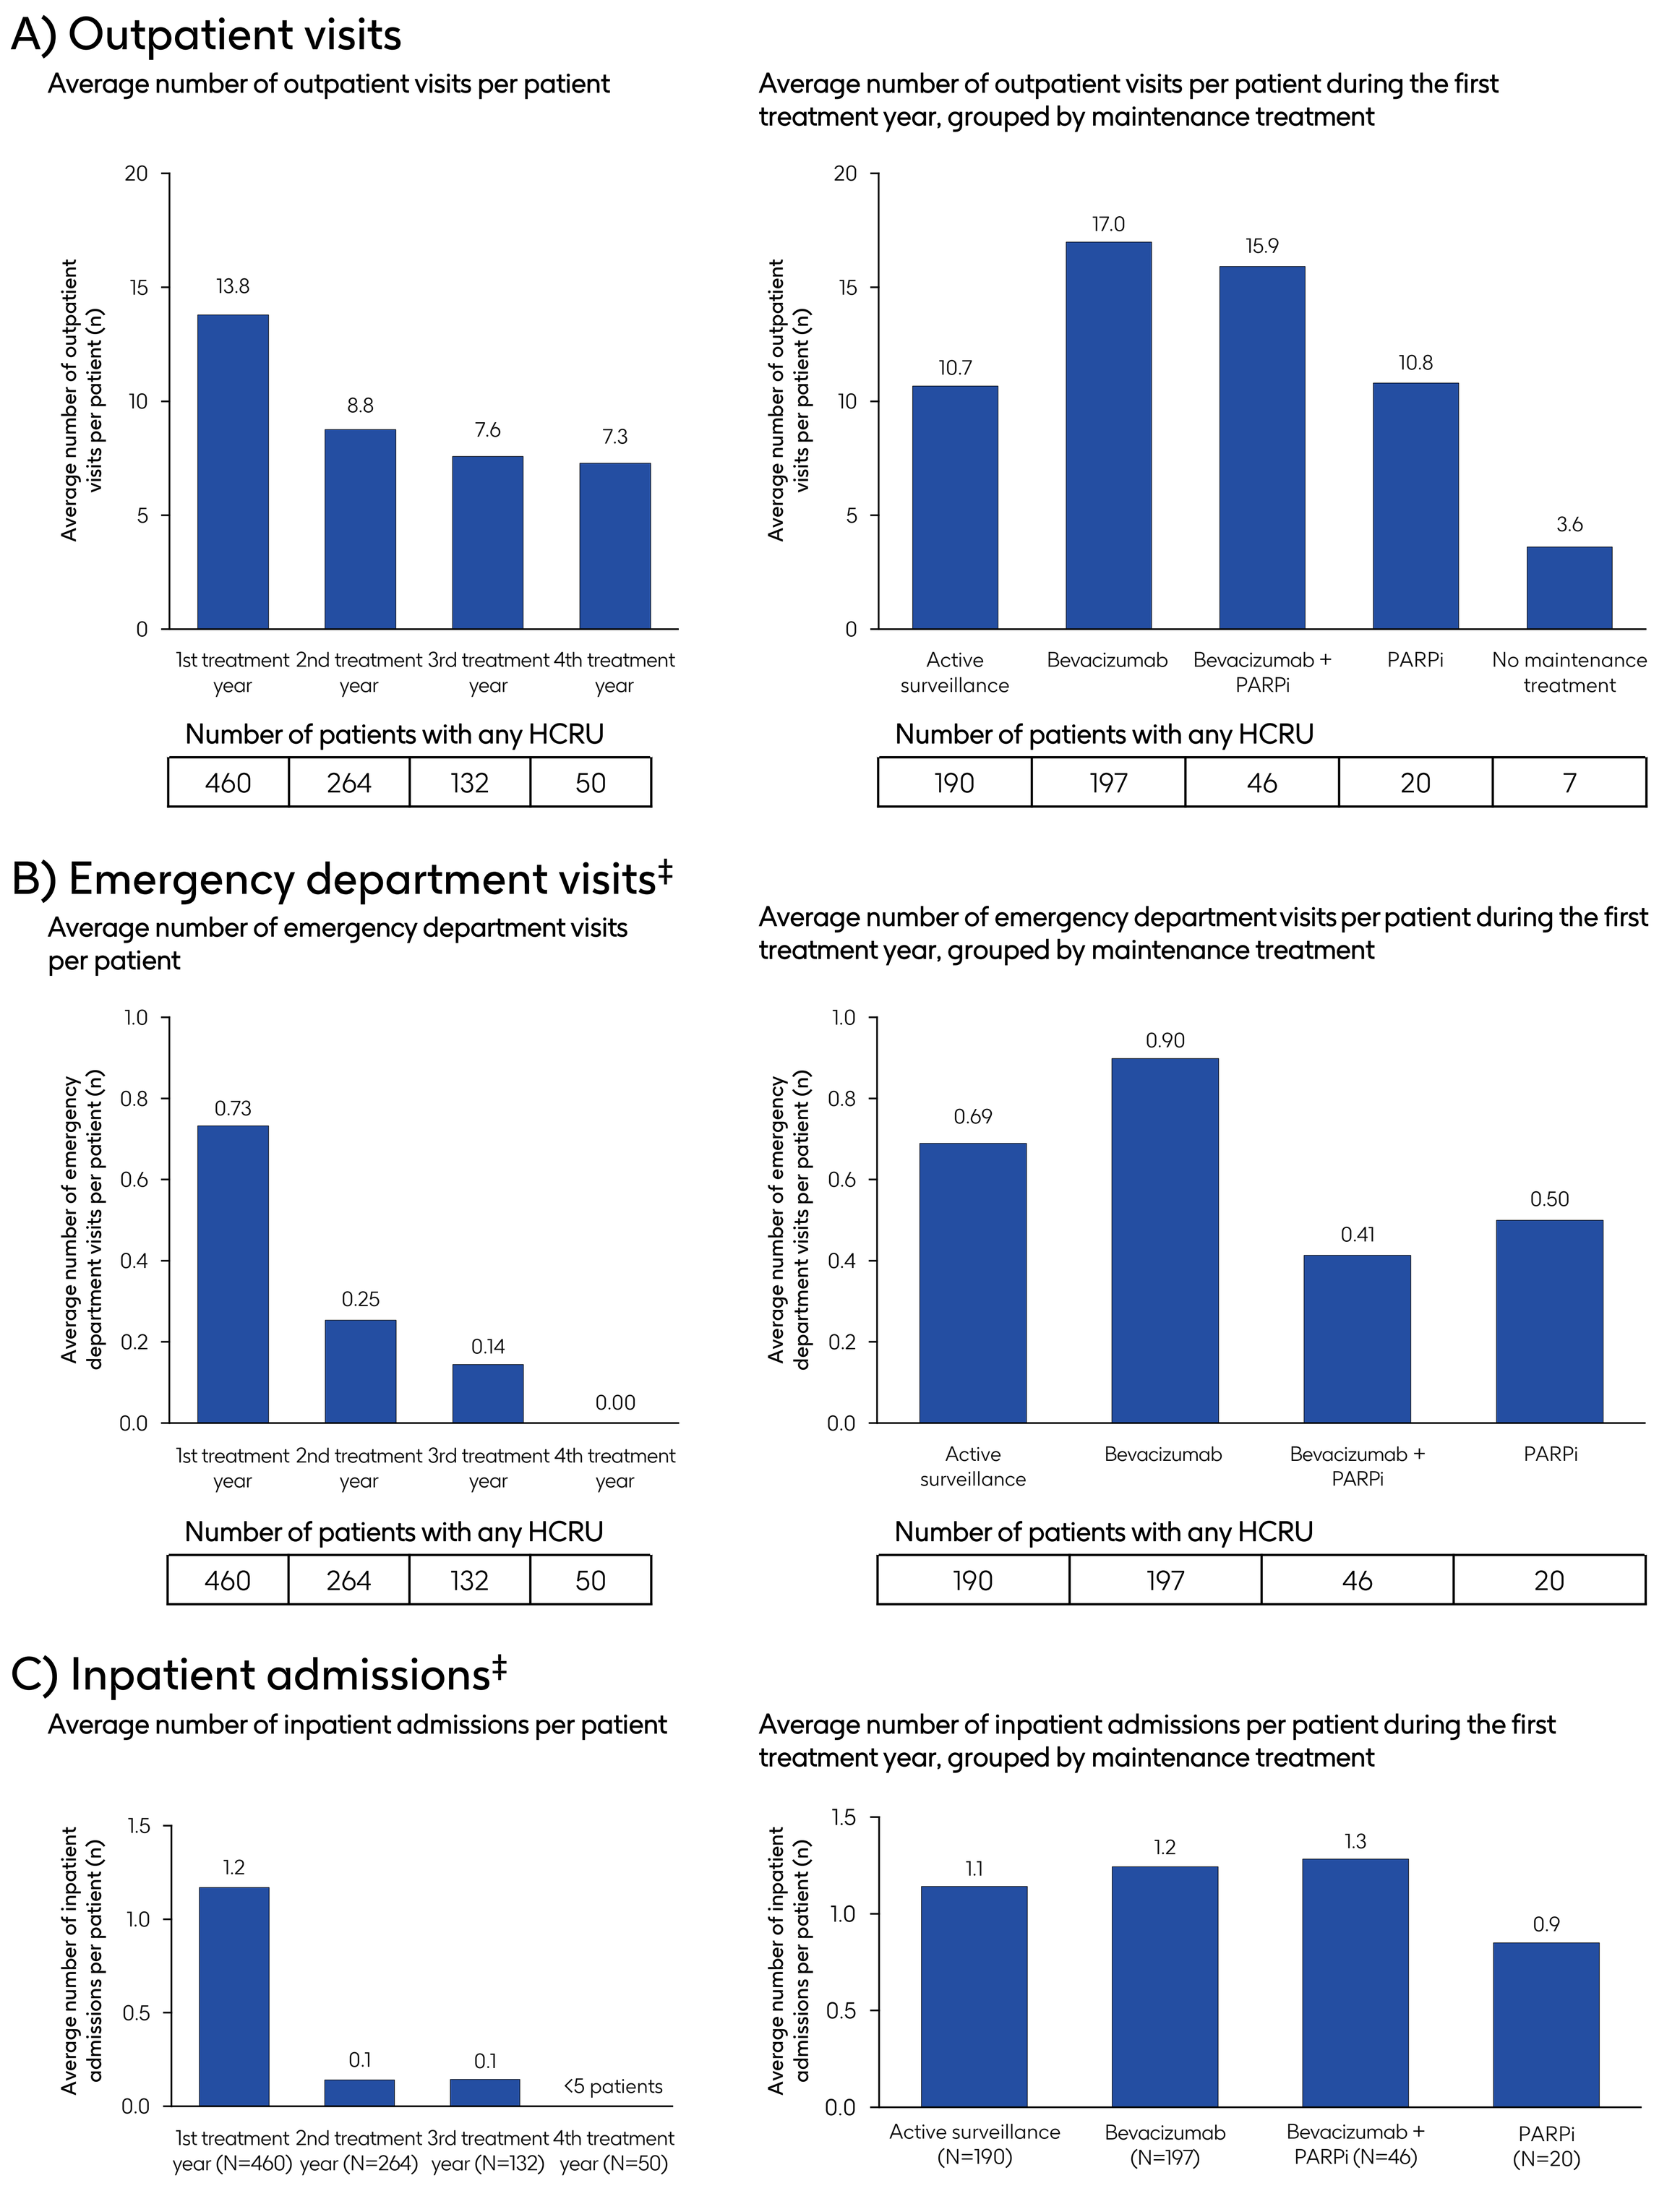
A) Outpatient visits
Average number of outpatient visits per patient
Average number of outpatient visits per patient (n)
13.8
8.8
7.6
7.3
1st treatment year
2nd treatment year
3rd treatment year
4th treatment year
Number of patients with any HCRU
460
264
132
50
Average number of outpatient visits per patient during the first treatment year, grouped by maintenance treatment
10.7
17.0
15.9
10.8
3.6
Active surveillance
Bevacizumab
Bevacizumab + PARPi
PARPi
No maintenance treatment
Number of patients with any HCRU
190
197
46
20
7
B) Emergency department visits‡
Average number of emergency department visits per patient
Average number of emergency department visits per patient (n)
0.73
0.25
0.14
0.00
1st treatment year
2nd treatment year
3rd treatment year
4th treatment year
Number of patients with any HCRU
460
264
132
50
Average number of emergency department visits per patient during the first treatment year, grouped by maintenance treatment
0.69
0.90
0.41
0.50
Active surveillance
Bevacizumab
Bevacizumab + PARPi
PARPi
Number of patients with any HCRU
190
197
46
20
C) Inpatient admissions‡
Average number of inpatient admissions per patient
Average number of inpatient admissions per patient (n)
1.2
0.1
0.1
<5 patients
1st treatment year (N=460)
2nd treatment year (N=264)
3rd treatment year (N=132)
4th treatment year (N=50)
Average number of inpatient admissions per patient during the first treatment year, grouped by maintenance treatment
1.1
1.2
1.3
0.9
Active surveillance (N=190)
Bevacizumab (N=197)
Bevacizumab + PARPi (N=46)
PARPi (N=20)

†HCRU was calculated only for patients who had follow-up for the whole treatment year; ‡Only emergency department visits/inpatient admissions that occurred during the active treatment line plus 30 days are included.

Cost

- The average cost per patient during the first year after diagnosis was approximately €12,300 for patients receiving PARPi monotherapy, €22,900 for bevacizumab plus PARPi and €25,500 for bevacizumab monotherapy (**Figure 4**).

Figure 4: Cost outcomes†,‡

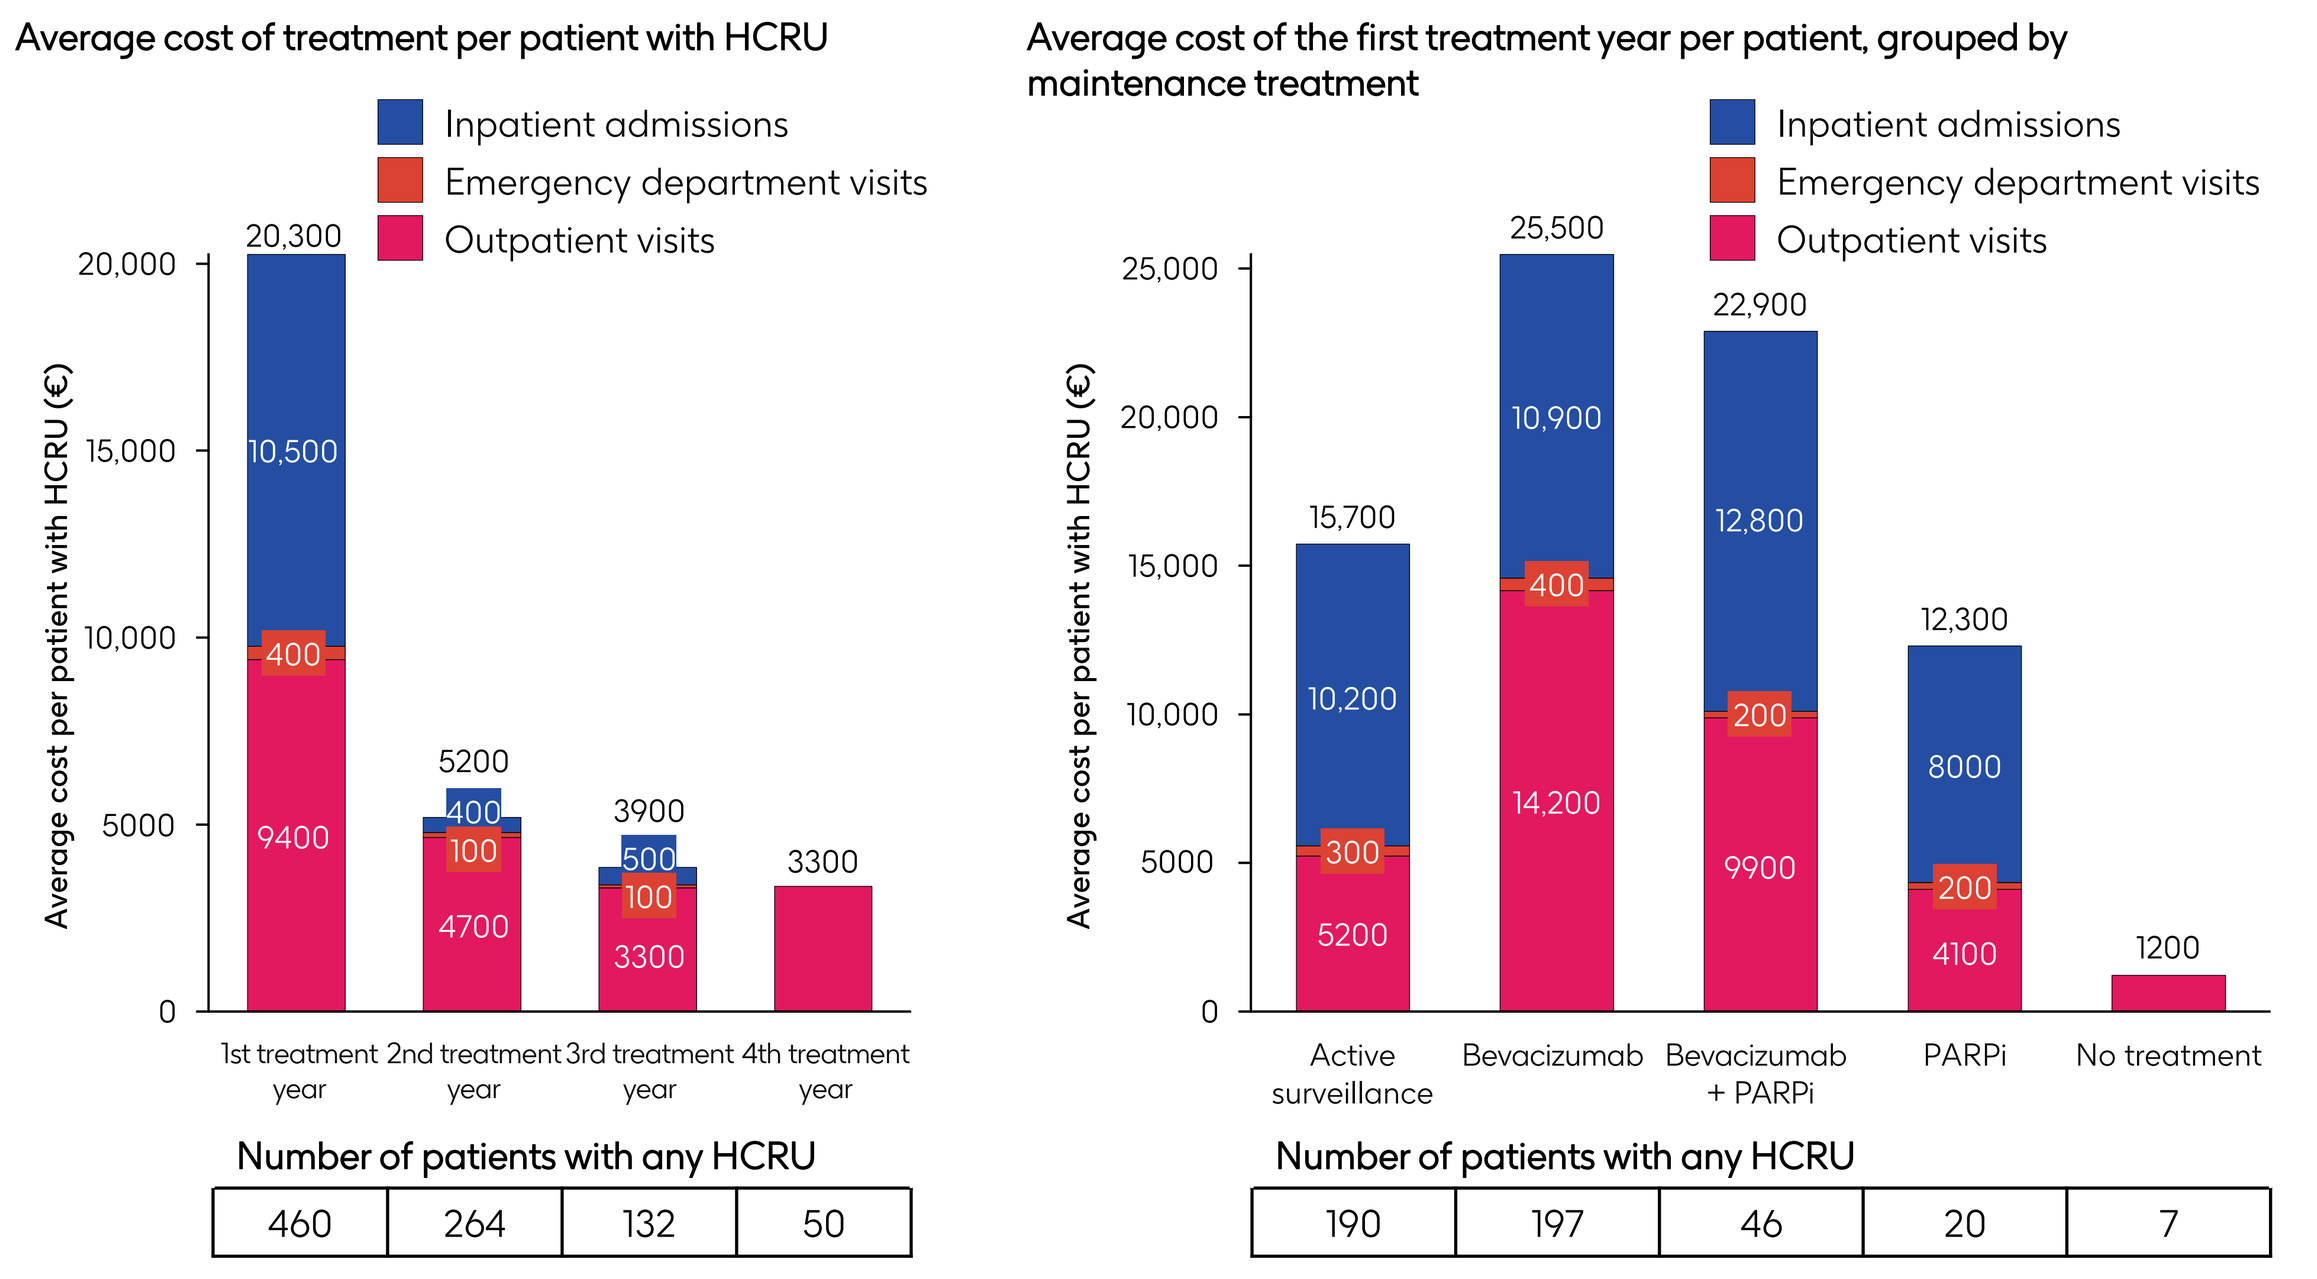
Average cost of treatment per patient with HCRU
Inpatient admissions
Emergency department visits
Outpatient visits
Average cost per patient with HCRU (€)
20,300
10,500
400
9400
5200
400
100
4700
3900
500
100
3300
3300
1st treatment year
2nd treatment year
3rd treatment year
4th treatment year
Number of patients with any HCRU
460
264
132
50
Average cost of the first treatment year per patient, grouped by maintenance treatment
15,700
10,200
300
5200
25,500
10,900
400
14,200
22,900
12,800
200
9900
12,300
8000
200
4100
1200
Active surveillance
Bevacizumab
Bevacizumab + PARPi
PARPi
No treatment
Number of patients with any HCRU
190
197
46
20
7

†HCRU was calculated only for patients who had follow-up for the whole treatment year; ‡Only emergency department visits/inpatient admissions that occurred during the active treatment line plus 30 days are included. All costs are in 2023 Euros.

Conclusions

Similar to analyses of OCRWE-Finland conducted from 2014–2019,[4] HCRU and costs per patient remained high from 2019–2023 for patients with HGSOC, especially in the first treatment year.

In the first year following diagnosis:
- Patients treated with PARPi monotherapy had fewer outpatient visits compared with those who received bevacizumab monotherapy or bevacizumab plus PARPi, although this is likely due to the different routes of administration for these regimens.
- Emergency department visits were also less frequent with PARPi monotherapy than bevacizumab monotherapy.

Patients receiving PARPi monotherapy had lower medical costs per patient than other maintenance treatment approaches in the first year following diagnosis.

Presented at the Professional Society for Health Economics and Outcomes Research (ISPOR) Europe Meeting; 17–20 November 2024; Barcelona, Spain.

Presenting author:
Barbara Mascialino, barbara.x.mascialino@gsk.com

Acknowledgements

Writing and editorial support, under the direction of the authors, were provided by Lyndsey Wood, BSc, Francesca Murphy, BA, and Ian Norton, PhD, all of Scion (a division of Prime, London, UK). Writing and editorial support provided by Prime was funded by GSK. This study was sponsored by GSK (study 218281).

Conflicts of interest/disclosures

Barbara Mascialino is an employee of GSK and holds financial equities in GSK. **Mari Lahelma** discloses study sponsorship from GSK. **Karita Salo** was employed by NHG Finland at the time of the study. **Matti Rajala** discloses consulting fees from GSK paid to their employer, NHG Finland Oy. **Eija Heikkilä** discloses consulting fees from GSK paid to their employer, NHG Finland Oy. **Fredrik Herse** reports consulting fees from GSK paid to their employer, NHG Finland Oy. **Juhana Idänpään-Heikkilä** is an employee of GSK and holds financial equities in GSK. **Sari Käkelä** was employed by GSK at the time of the study. **Saara Tikka** is employed by GSK and holds financial equities in GSK. **Dirk Schneider** is employed by GSK and holds financial equities in GSK. **Sakari Hietanen** reports personal fees from GSK; consulting fees from AstraZeneca, GSK, and Orion Pharma; honoraria from AstraZeneca; and payment for expert testimony from AstraZeneca and GSK. **Mikko Loukovaara** reports consulting fees from AstraZeneca and GSK; meeting support from AstraZeneca; advisory board participation from AstraZeneca and GSK; and is an unpaid member of the Finnish Gynecologic Oncology Group. **Annika Auranen** reports advisory board participation for GSK, MSD and AstraZeneca.

Abbreviations

1L, first line; 2L, second line; 3L, third line; BMI, body mass index; *BRCA*, breast cancer gene; *BRCA*m, *BRCA* mutation; *BRCA*wt, *BRCA* wild type; FIGO, International Federation of Gynecology and Obstetrics; HCRU, healthcare resource utilisation; HGSOC, high-grade serous ovarian cancer; HRD, homologous recombination deficiency; OC, ovarian cancer; PARPi, poly (ADP-ribose) polymerase inhibitor; SD, standard deviation.

References

1. World Cancer Research Fund International. Ovarian cancer statistics; 2022. https://www.wcrf.org/cancer-trends/ovarian-cancer-statistics. Accessed November 2024; 2. Global Cancer Observatory. Finland 2022. https://gco.iarc.who.int/media/globocan/factsheets/populations/246-finland-fact-sheet.pdf. Accessed November 2024; 3. O'Malley DM, et al. *Target Oncol.* 2023;18:471–503; 4. Rauhamaa H, et al. Poster EE64 presented at ISPOR Europe 2022, Vienna, Austria.