Public health impact of a simplified booster vaccine schedule against tick-borne encephalitis (TBE) in Austria – A modeling analysis

EPH83

Holly Yu, MSPH[1], Maryna Ivets, PhD[2], Jennifer Branner, MSc[2], Harish Madhava, MD[3], Andreas Pilz, PhD[4], Frederick J. Angulo, DVM, PhD[1], Jennifer C. Moïsi, PhD[5], Malina Müller, PhD[2]

[1] Pfizer, Inc., Collegeville, PA, USA [2] WifOR Institute, Darmstadt, Germany [3] Pfizer, Inc., Walton Oaks, UK [4] Pfizer, Inc., Vienna, Austria [5] Pfizer, Inc., Paris, France

Background

- Tick-borne encephalitis (TBE) is a human viral infectious disease that can affect the central nervous system (CNS), causing long-term neurological symptoms and even death.[1] The TBE incidence in humans has risen significantly over the past decades, and endemic regions have expanded.[1]
- Since there is no specific antiviral treatment for TBE, vaccination is essential to effectively reduce TBE morbidity and mortality.[2,3] Despite this, TBE vaccination uptake varies considerably across many countries (especially endemic), and compliance with the recommended booster vaccination schedule remains suboptimal, even in Austria where the completion rate of primary immunization is high.[4]
- FSME-Immun vaccine (TicoVac®) is currently licensed and recommended against TBE in Austria in a three-dose primary series with an initial booster dose after three years for all ages, and subsequent booster doses every three years for those ≥60 and every five years for those <60 years of age (Figure 1).
- Implementation of this booster vaccination schedule is not simple and therefore, a simplification is expected to improve TBE vaccine uptake and completion. Additionally, by reducing the number of vaccine doses a person receives over their lifetime, there would be fewer associated adverse events.

Objective

- This study investigates a potential change in the vaccination schedule for FSME-Immun and its impact on public health.
- Given the high effectiveness of FSME-Immun, we developed a model to assess the public health impact of a simplified schedule in Austria. In the proposed simplified schedule, after completion of the primary series and first booster dose, subsequent FSME-Immun boosters would be indicated every 10 years in all age groups (Figure 1).

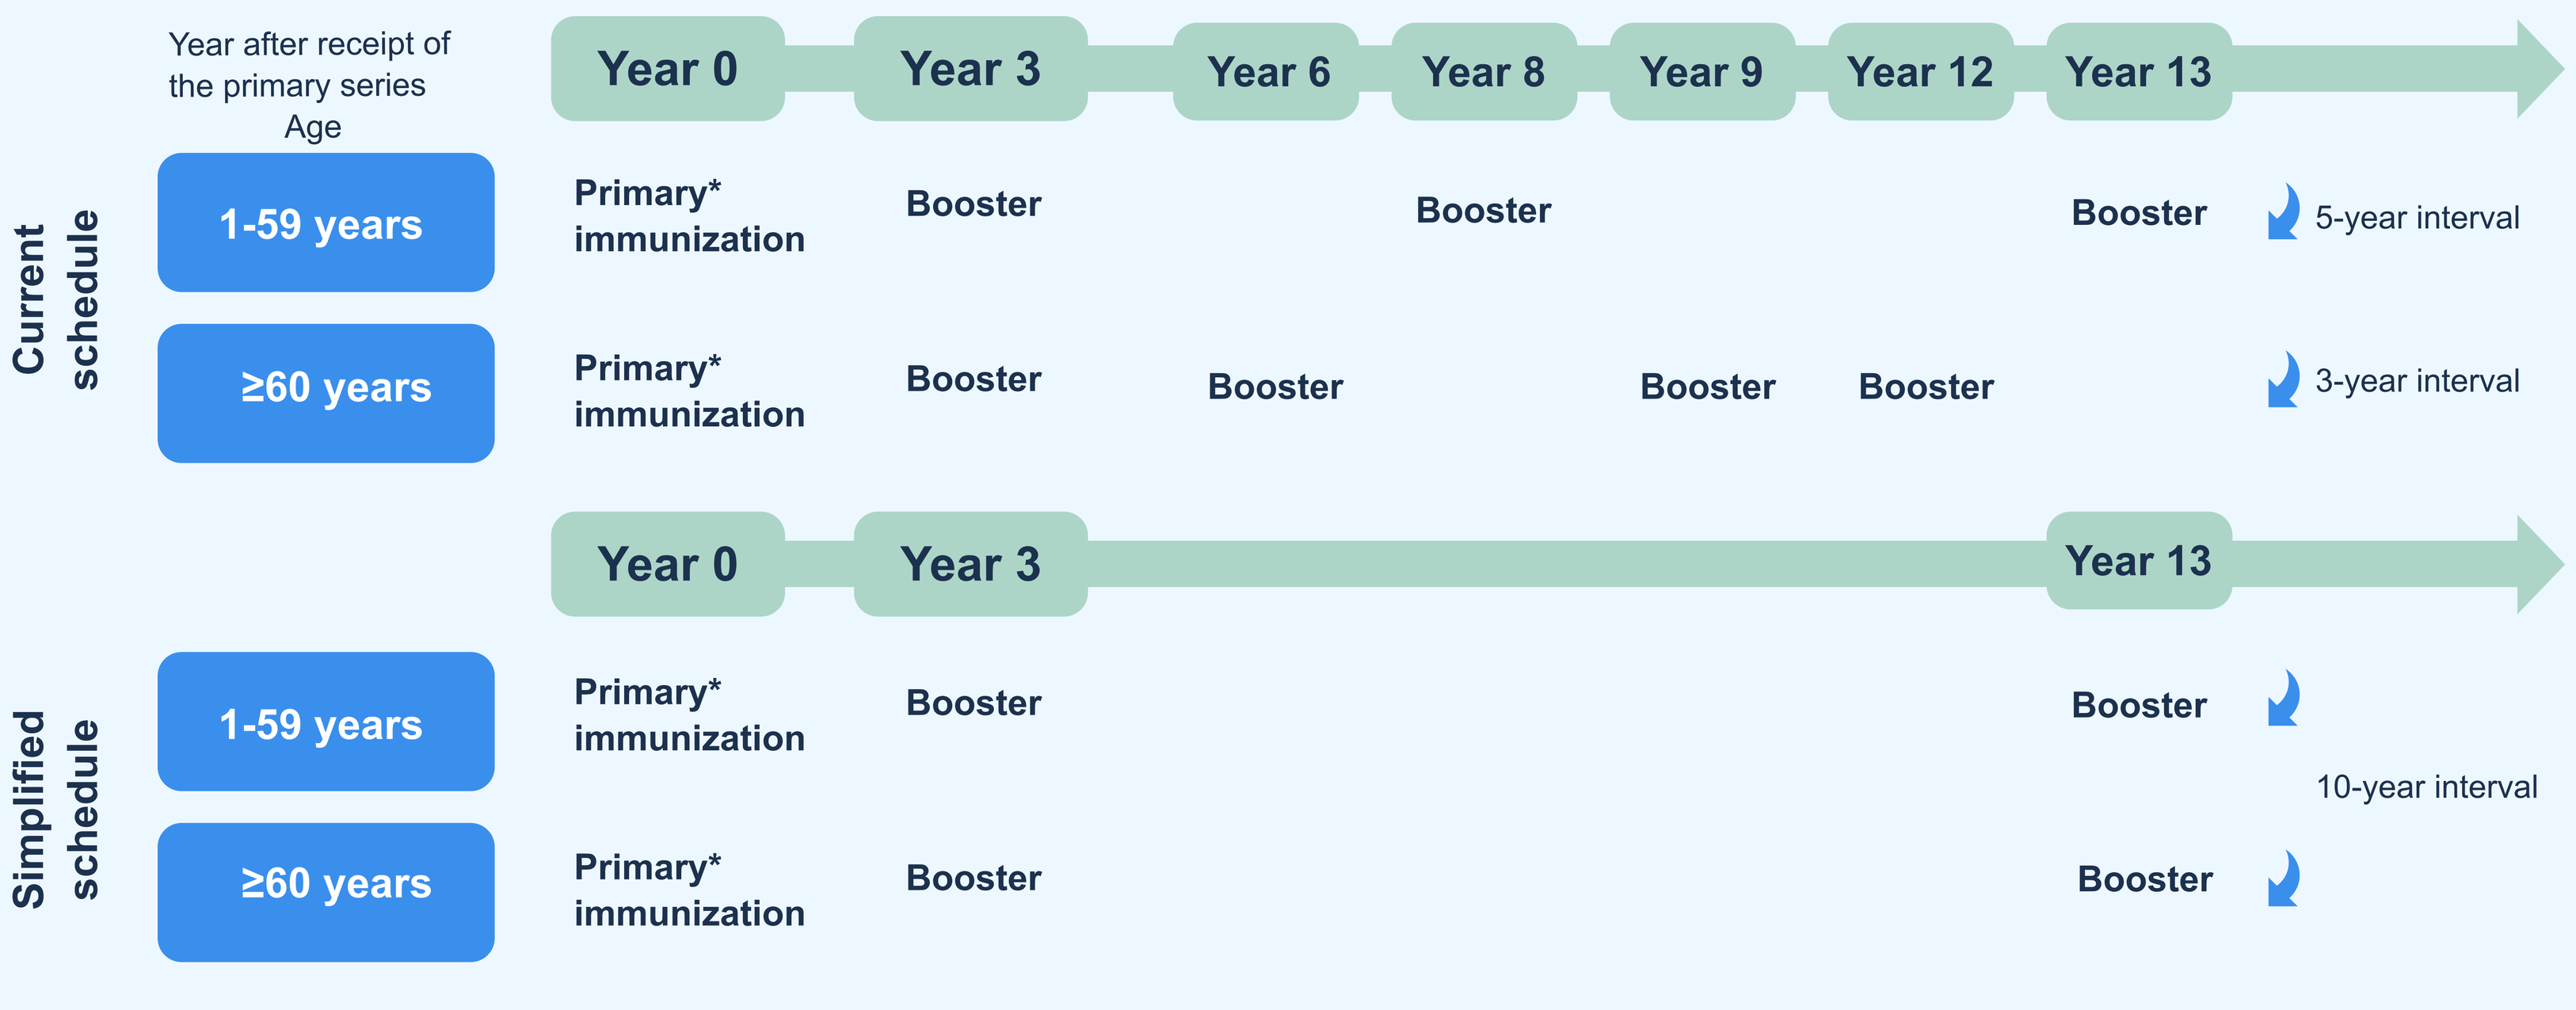

Year after receipt of the primary series
Age
Year 0
Year 3
Year 6
Year 8
Year 9
Year 12
Year 13
Current schedule
1-59 years
Primary* immunization
Booster
Booster
Booster
5-year interval
≥60 years
Primary* immunization
Booster
Booster
Booster
Booster
3-year interval
Year 0
Year 3
Year 13
Simplified schedule
1-59 years
Primary* immunization
Booster
Booster
10-year interval
≥60 years
Primary* immunization
Booster
Booster

Figure 1: Current recommendation and simplified vaccination schedule
**Primary immunization refers to three vaccinations according to the conventional schedule.*

Methods

- We adapted a previously developed closed-cohort Markov model[5] to Austria to track age cohorts over their lifetimes from 1-85 years, simulating the incremental public health impact due to the change in vaccination schedule (Figure 2).
- We compared the current schedule and the simplified schedule against no vaccination. Vaccine effectiveness (VE) data and transition probabilities were derived from the literature; age-specific risk and infection rates were incorporated assuming the TBEV European (TBEV-EU) subtype as the primary infection agent (Table 1).
- In sensitivity analyses, we assumed higher VE and that the reduced frequency of subsequent boosters in the simplified schedule would result in increased vaccine uptake and completion as compared to the current schedule. Outcomes were the number of vaccine doses administered, adverse events (AEs), TBE cases, and associated deaths.

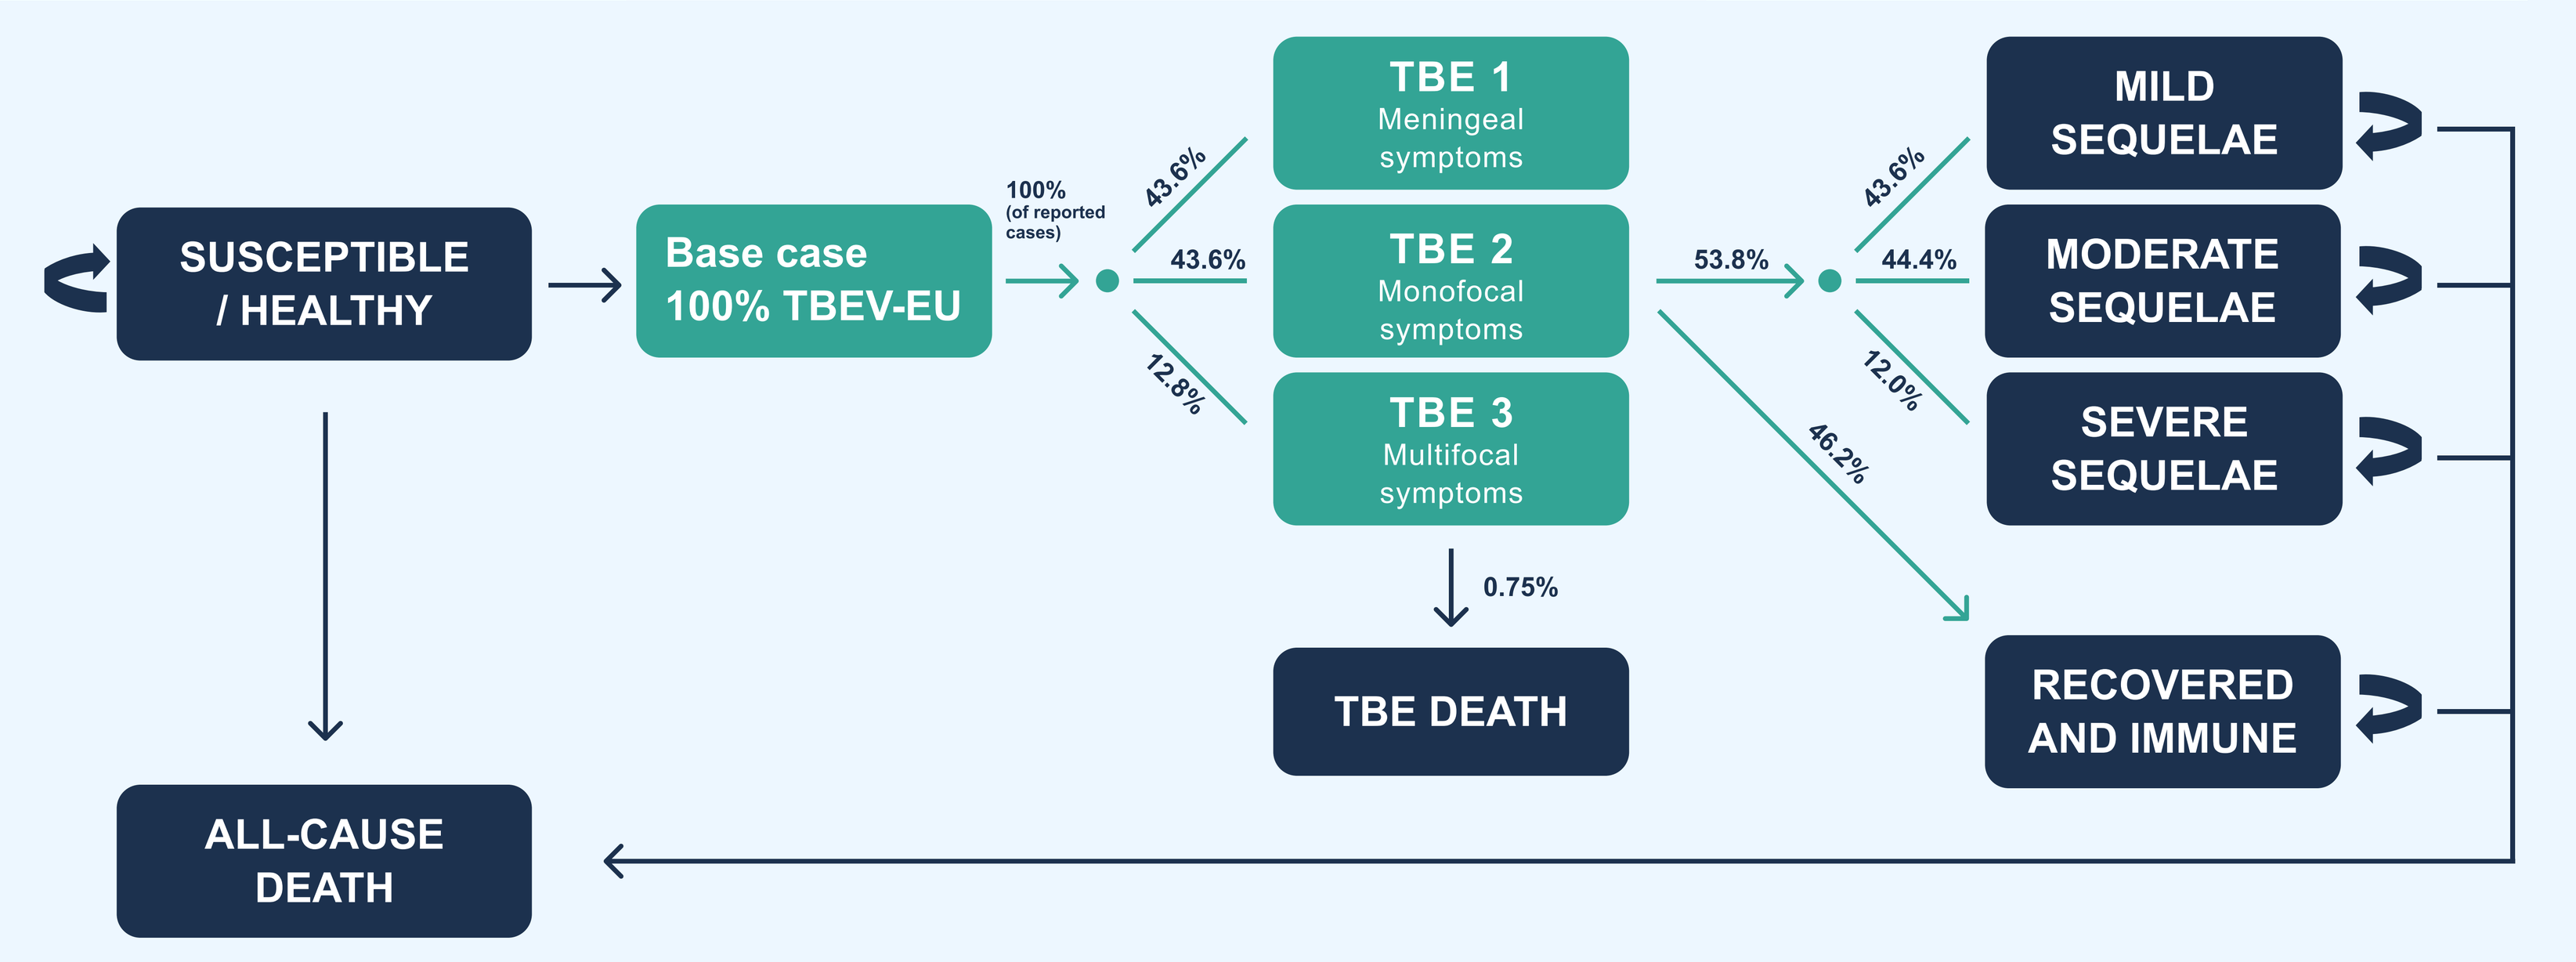

Pre-infection (susceptible)
TBE manifestation (1 cycle)
Sequelae or recovered (permanent state)
SUSCEPTIBLE / HEALTHY
Base case 100% TBEV-EU
100% (of reported cases)
43.6%
43.6%
12.8%
TBE 1 Meningeal symptoms
TBE 2 Monofocal symptoms
TBE 3 Multifocal symptoms
0.75%
TBE DEATH
53.8%
43.6%
44.4%
12.0%
46.2%
MILD SEQUELAE
MODERATE SEQUELAE
SEVERE SEQUELAE
RECOVERED AND IMMUNE
ALL-CAUSE DEATH

Figure 2: Model structure

Input parameter	Current vaccination schedule	Simplified vaccination schedule
Completion of the three-dose primary vaccination series[4]	60%	
Completion of the first booster after primary series, and subsequent boosters[4]	85%	90% Based on assumption of behavioral change
Booster vaccination interval	3 years (age ≥60) 5 years (age <60)	10 years (all ages)
Vaccine effectiveness (VE)[6]	Consistent vaccine effectiveness of 96.6% following the initial primary immunization series in year 0. This effectiveness remains constant at 96.6% in case of compliance. Thus, no waning is assumed.	This effectiveness remains constant at 96.6% until the receipt of the first booster. Afterwards and for upcoming boosters, there is a gradual decline in effectiveness after 3 years (i.e., in year 6, 16, 26, etc.) at a rate of 1.1% annually, reaching 88.6% just before the receipt of the subsequent booster, scheduled every 10 years (i.e., in year 13, 23, etc.).
Yearly waning rate if noncompliant[6]	1.1%	
Age-specific risk of TBEV infection[7,8]	*Age group / TBEV infection risk*: 1 to 6: 0.0000131; 7 to 14: 0.0000205; 15 to 20: 0.0000113; 21 to 30: 0.0000151; 31 to 40: 0.0000113; 41 to 50: 0.0000144; 51 to 60: 0.0000186; 61 to 70: 0.0000492; 71 to 80: 0.0000219; 81 to 85: 0.0000222	$\frac{\text{TBE cases}_{\text{age group}}}{\text{Population}_{\text{age group}}}$
Risk of adverse events after vaccination (frequency)[9]	Very common (≥1/10) Common (≥1/100 to <1/10) Uncommon (≥1/1,000 to <1/100) Rare (≥1/10,000 to <1/1,000)	

Table 1: Main input parameters in the base case model

Results

Over a period of 85 years, the simplified schedule compared to the current schedule (Table 2) would result in

- 13.98 million fewer vaccine doses received,
- a total of 2.17 million fewer "very common", "common", "uncommon", and "rare" AEs,
- 55 additional TBE cases (0.64 per year), and
- 0.21 additional TBE-related deaths (0.002 per year).

Outcomes	No vaccination scenario	Current schedule	Simplified schedule
Vaccine doses administered (overall)	0	49,545,387	35,563,418
Adverse events midpoint (min, max)			
Very common (≥1/10)	0	≥4,954,539	≥3,556,342
Common (≥1/100 to <1/10)	0	2,477,269 (495,454; 4,954,539)	1,778,171 (355,634; 3,556,342)
Uncommon (≥1/1,000 to <1/100)	0	247,727 (49,545; 495,454)	177,817 (35,563; 355,634)
Rare (≥1/10,000 to <1/1,000)	0	24,773 (4,955; 49,545)	17,782 (3,556; 35,563)
Number of TBE cases	8,476	3,790	3,845
Number of TBE-related deaths	31.79	14.21	14.42

Table 2: Base case results over 85 years

In sensitivity analyses, the simplified schedule prevented more TBE cases and deaths than the current schedule if an increase in uptake and completion for the primary series and /or booster vaccination takes place (Table 3).

Outcomes	Current schedule: If higher vaccine effectiveness of 99.1%[10] vs. 96.6%	Simplified schedule: If higher vaccine effectiveness of 99.1%[10] vs. 96.6%	Simplified schedule: If higher primary completion 65% vs. 60%	Simplified schedule: If higher booster completion 95% vs. 85%	Simplified schedule: If higher primary 65% + booster completion 95%
Vaccine doses administered (overall)	49,545,412	35,563,441	38,527,059	36,666,680	39,722,261
Adverse events midpoint (min, max)					
Very common (≥1/10)	≥4,954,541	≥3,556,344	≥3,852,706	≥3,666,668	≥3,972,226
Common (≥1/100 to <1/10)	2,477,271 (495,454; 4,954,541)	1,778,172 (355,634; 3,556,344)	1,926,353 (385,271; 3,852,706)	1,833,334 (366,667; 3,666,668)	1,986,113 (397,223; 3,972,226)
Uncommon (≥1/1,000 to <1/100)	247,727 (49,545; 495,454)	177,817 (35,563; 355,634)	192,635 (38,527; 385,271)	183,333 (36,667; 366,667)	198,611 (39,722; 397,223)
Rare (≥1/10,000 to <1/1,000)	24,773 (4,955; 49,545)	17,782 (3,556; 35,563)	19,264 (3,853; 38,527)	18,333 (3,667; 36,667)	19,861 (3,972; 39,722)
Number of TBE cases	3,510	3,528	3,459	3,776	3,384
Number of TBE-related deaths	13.16	13.23	12.97	14.16	12.69

Table 3: Sensitivity analysis results over 85 years

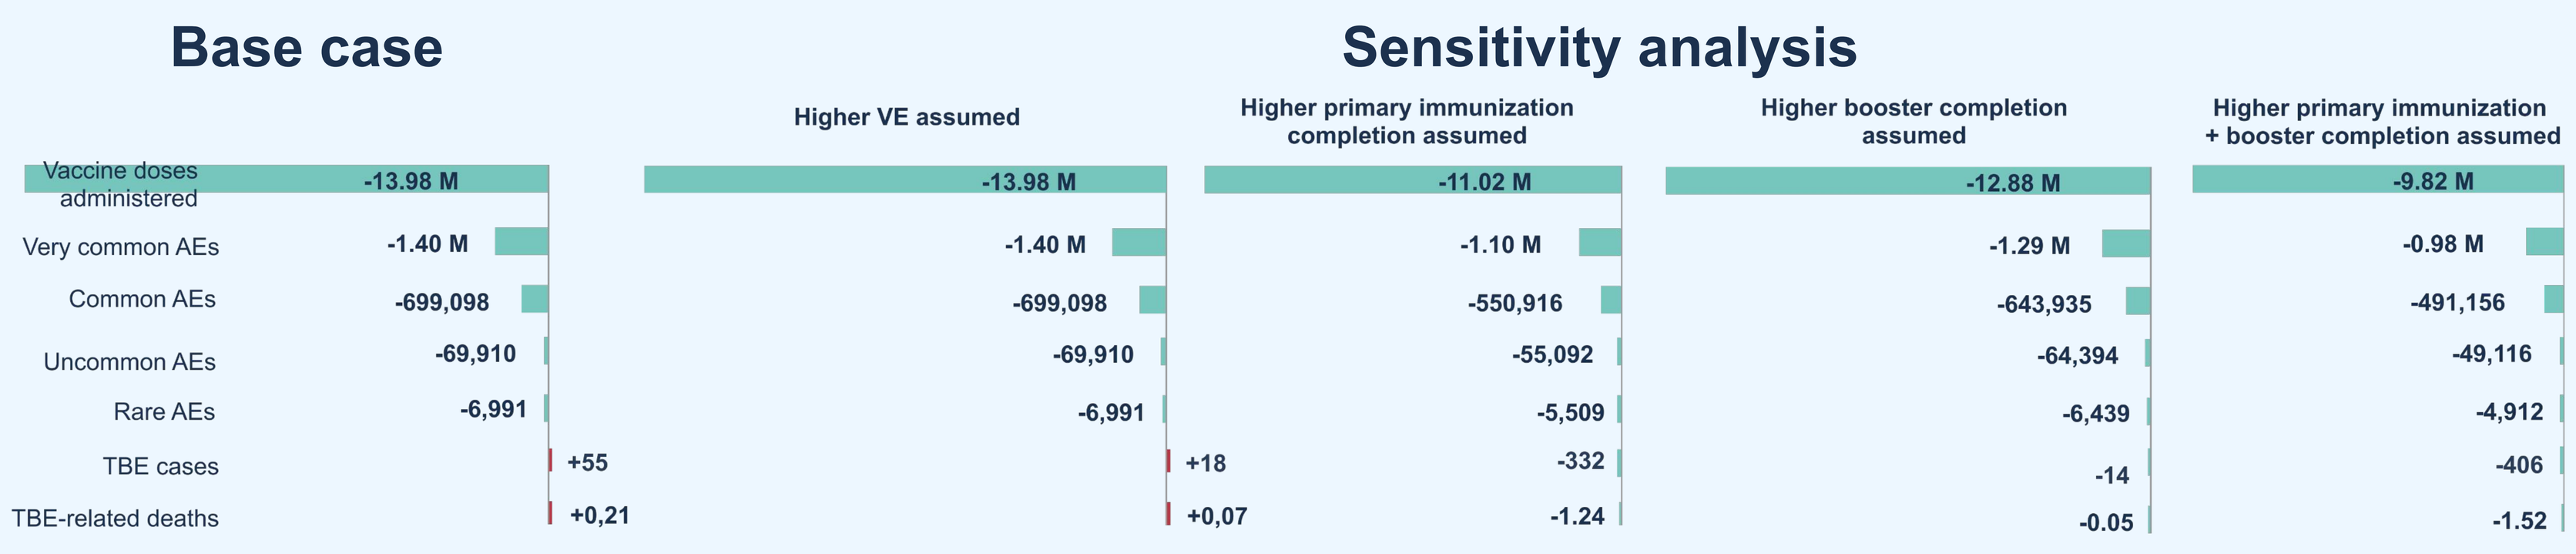

Base case
Sensitivity analysis
Higher VE assumed
Higher primary immunization completion assumed
Higher booster completion assumed
Higher primary immunization + booster completion assumed
Vaccine doses administered
Very common AEs
Common AEs
Uncommon AEs
Rare AEs
TBE cases
TBE-related deaths
-13.98 M
-1.40 M
-699,098
-69,910
-6,991
+55
+0,21
-13.98 M
-1.40 M
-699,098
-69,910
-6,991
+18
+0,07
-11.02 M
-1.10 M
-550,916
-55,092
-5,509
-332
-1.24
-12.88 M
-1.29 M
-643,935
-64,394
-6,439
-14
-0.05
-9.82 M
-0.98 M
-491,156
-49,116
-4,912
-406
-1.52

Figure 3: Incremental results over a time horizon of 85 years for the base case and variations of the sensitivity analysis

Discussion

- This is the first study to evaluate how a simplification in FSME-Immun subsequent booster schedule would impact public health in Austria – a TBE-endemic country.
- The base case model provides conservative estimates, largely driven by our assumptions on vaccine effectiveness and of only a 5 percentage points increase for the booster completion rates in the simplified booster schedule.
- The most robust Austrian-specific data were used in this model, such as TBE incidence rates and TBE vaccine completion rates. While vaccine effectiveness data are available for some European countries,[6,11] they are lacking for Austria.

Conclusions

- The simplified vaccination schedule would significantly reduce the number of vaccine doses administered and associated AEs without a meaningful increase in TBE cases or deaths over a lifetime.
- In our sensitivity analyses, we evaluated the impact of various assumptions on the outcomes, such as higher vaccine effectiveness, an increased completion of the primary series, first booster and subsequent boosters for the simplified schedule (Table 3). All of these resulted in a positive impact compared to the current schedule on the number of vaccine doses and associated AEs. The analyses showed a small decrease in the number of TBE cases and deaths if higher completion was assumed (Figure 3).
- Changing FSME-Immun subsequent booster schedule to every 10 years for all ages could simplify vaccine implementation and improve uptake and compliance while achieving better public health outcomes.

1. Riccardi N, Antonello RM, Luzzati R, Zajkowska J, Di Bella S, Giacobbe DR. Tick-borne encephalitis in Europe: a brief update on epidemiology, diagnosis, prevention, and treatment. Eur J Intern Med. 2019;62:1-6. doi:10.1016/j.ejim.2019.01.004
2. Beauté J, Spiteri G, Warns-Petit E, Zeller H. Tick-borne encephalitis in Europe, 2012 to 2016. Eurosurveillance. 2018;23(45). doi:10.2807/1560-7917.ES.2018.23.45.1800201
3. Angulo FJ, Zhang P, Halsby K, et al. A systematic literature review of the effectiveness of tick-borne encephalitis vaccines in Europe. Vaccine. 2023;41(47):6914-6921. doi:10.1016/j.vaccine.2023.10.014
4. Pilz A, Erber W, Schmitt HJ. Vaccine uptake in 20 countries in Europe 2020: Focus on tick-borne encephalitis (TBE). Ticks Tick-Borne Dis. 2023;14(1):102059. doi:10.1016/j.ttbdis.2022.102059
5. Müller M, Lintener H, Henkel V, et al. Does the Vaccination against Tick-Borne Encephalitis Offer Good Value for Money for Incidence Rates below the WHO Threshold for Endemicity? A Case Study for Germany. Vaccines. 2024;12(10):1165. doi:10.3390/vaccines12101165
6. Nygren TM, Pilic A, Böhmer MM, et al. Tick-borne encephalitis vaccine effectiveness and barriers to vaccination in Germany. Sci Rep. 2022;12(1):11706. doi:10.1038/s41598-022-15447-5
7. Eurostat. Bevölkerung Am 1. Januar Nach Alter Und Geschlecht. European Commission; 2022. Accessed February 3, 2023. https://ec.europa.eu/eurostat/databrowser/view/DEMO_PJAN/default/table?lang=de&category=demo.demo_pop
8. Aberle J, Kundi M. TBE in Austria: Extended Assessment of Clinical Course Including Sequelae and Vaccination Status of Virologically Verified TBE Cases Based on Active Surveillance. Medizinische Universität Wien; 2023.
9. Summary of Product Characteristics (SPC) FSME-IMMUN 0.5 Ml Suspension for Injection in a Pre-Filled Syringe, Tick-Borne Encephalitis Vaccine (Whole Virus, Inactivated).
10. Zavadska D, Freimane Z, Karelis G, et al. Effectiveness of tick-borne encephalitis vaccination in Latvia, 2018–2020: an observational study. Clin Microbiol Infect. Published online July 2023:S1198743X23003099. doi:10.1016/j.cmi.2023.06.028
11. Erber W, Khan F, Zavadska D, et al. Effectiveness of TBE vaccination in southern Germany and Latvia. Vaccine. 2022;40(5):819-825. doi:10.1016/j.vaccine.2021.12.028

This analysis was funded by Pfizer.