EE172

Estimating the Potential Lifetime Health and Economic Impact of V116, an Adult-Specific 21-Valent Pneumococcal Conjugate Vaccine, on Pneumococcal Diseases in Greece

Zinan Yi[1]; Ilias Gountas[2]; Eleana Tsoumani[2]; Georgios Trimis[2]; Kelly D. Johnson[1]; Kwame Owusu-Edusei[1]

[1]Merck & Co., Inc., Rahway, NJ, USA; [2]MSD Greece, Athens, Greece

Background

- *Streptococcus pneumoniae* is a Gram-positive bacterium that commonly colonizes the respiratory tract and can potentially cause pneumococcal disease (PD) in adults, with those considered immunocompromised/suppressed being at highest risk[1,2]
- PD, such as invasive pneumococcal disease (IPD) and non-bacteremic pneumococcal pneumonia (NBPP), is associated with high morbidity and mortality, which causes substantial health and economic burden on the healthcare system[1,2]
- Although available vaccines have largely reduced the burden of PD among adults, current data on PD show substantial residual burden attributable to serotypes they do not currently cover[1,2]
- V116 is a 21-valent pneumococcal conjugate vaccine (PCV) specifically designed for adults. It contains ***21*** serotypes: ***3, 6A, 7F, 8, 9N, 10A, 11A, 12F, 15A, 15C, 16F, 17F, 19A, 20A, 22F, 23A, 23B, 24F, 31, 33F,*** and ***35B***. ***Eight*** (***15A, 15C, 16F, 23A, 23B, 24F, 31, 35B***) of the 21 serotypes are unique and are not included in any currently licensed vaccines[3]
- According to published data in 2022, V116 serotypes account for a higher incidence of IPD vs PCV20, disease coverage for V116 and PCV20 are 75.9% and 55.4%, respectively, in individuals 65+ years in Greece, and the unique 8 serotypes were responsible for approximately 26.4%[2]

Objective

To quantify and compare the potential lifetime health and economic impact of vaccination with V116 vs PCV20 on IPD in adults aged 19 and older in Greece

Methods

- A multi-cohort Markov model (structure depicted in **Figure 1**) was built to estimate the lifetime (until death or turned 100 years old) IPD cases, NBPP inpatient (IP), and outpatient (OP) cases, IPD and NBPP IP related deaths, and the associated direct medical costs (in 2022 €) among current Greece adults aged 19-64 with risk conditions and 65+ (assuming 2022 population figures) of no vaccination or vaccinating with V116 or PCV20
- The study population was stratified into three mutually exclusive risk groups: low-risk (LR), at-risk (AR), and high-risk (HR) adults[4]
- The same serotype-specific vaccine effectiveness was assumed for the two vaccines. The waning of the effectiveness was assumed to be flat for the first 5 years and then linearly declines to zero in the following 10 years
- Vaccine coverage rates are the same for the two vaccines and for all risk groups – 40% for all age and risk groups[5]
- Discounting rate for costs is 3%. Other key model parameters are shown in **Table 1**
- Base case results were summarized for V116 and PCV20, as well as no vaccination
- One-way sensitivity analysis (OWSA) was conducted to assess the robustness of the results and to identify the most influential parameters on the total direct medical costs saved by V116 when compared to PCV20. Parameters varied in the OWSA included vaccine efficacy, direct medical treatment cost for IPD and NBPP by age and risk groups, IPD and NBPP IP fatality rates, and discounting rate for costs

Results

- In adults aged 19-64 with risk conditions, compared to no vaccination, V116 prevented 752 IPD cases and 82 IPD deaths – 45% more than the 520 IPD cases and 57 IPD deaths prevented by PCV20. The averted IPD cases by V116 saved €1.64 million in total medical costs – 45% higher than the €1.13 million costs averted by PCV20 (**Table 2**)
- In adults aged 65+, compared to no vaccination, V116 prevented 794 IPD cases and 124 IPD deaths – 45% more than the 549 IPD cases and 86 IPD deaths prevented by PCV20. The averted IPD cases by V116 saved €1.78 million in total medical costs – 45% higher than the €1.23 million costs averted by PCV20 (**Table 3**)
- Among the parameters included in the OWSA, the top sensitive parameters are NBPP treatment costs, efficacy, and discounting rate for costs (**Figure 2**)
- OWSA confirmed the robustness of the results. All scenarios showed a large lifetime direct medical cost saved by V116 when compared to PCV20 (**Figure 2**)

Figure 1. Schematic diagram depicting the structure of the state-transition Markov model

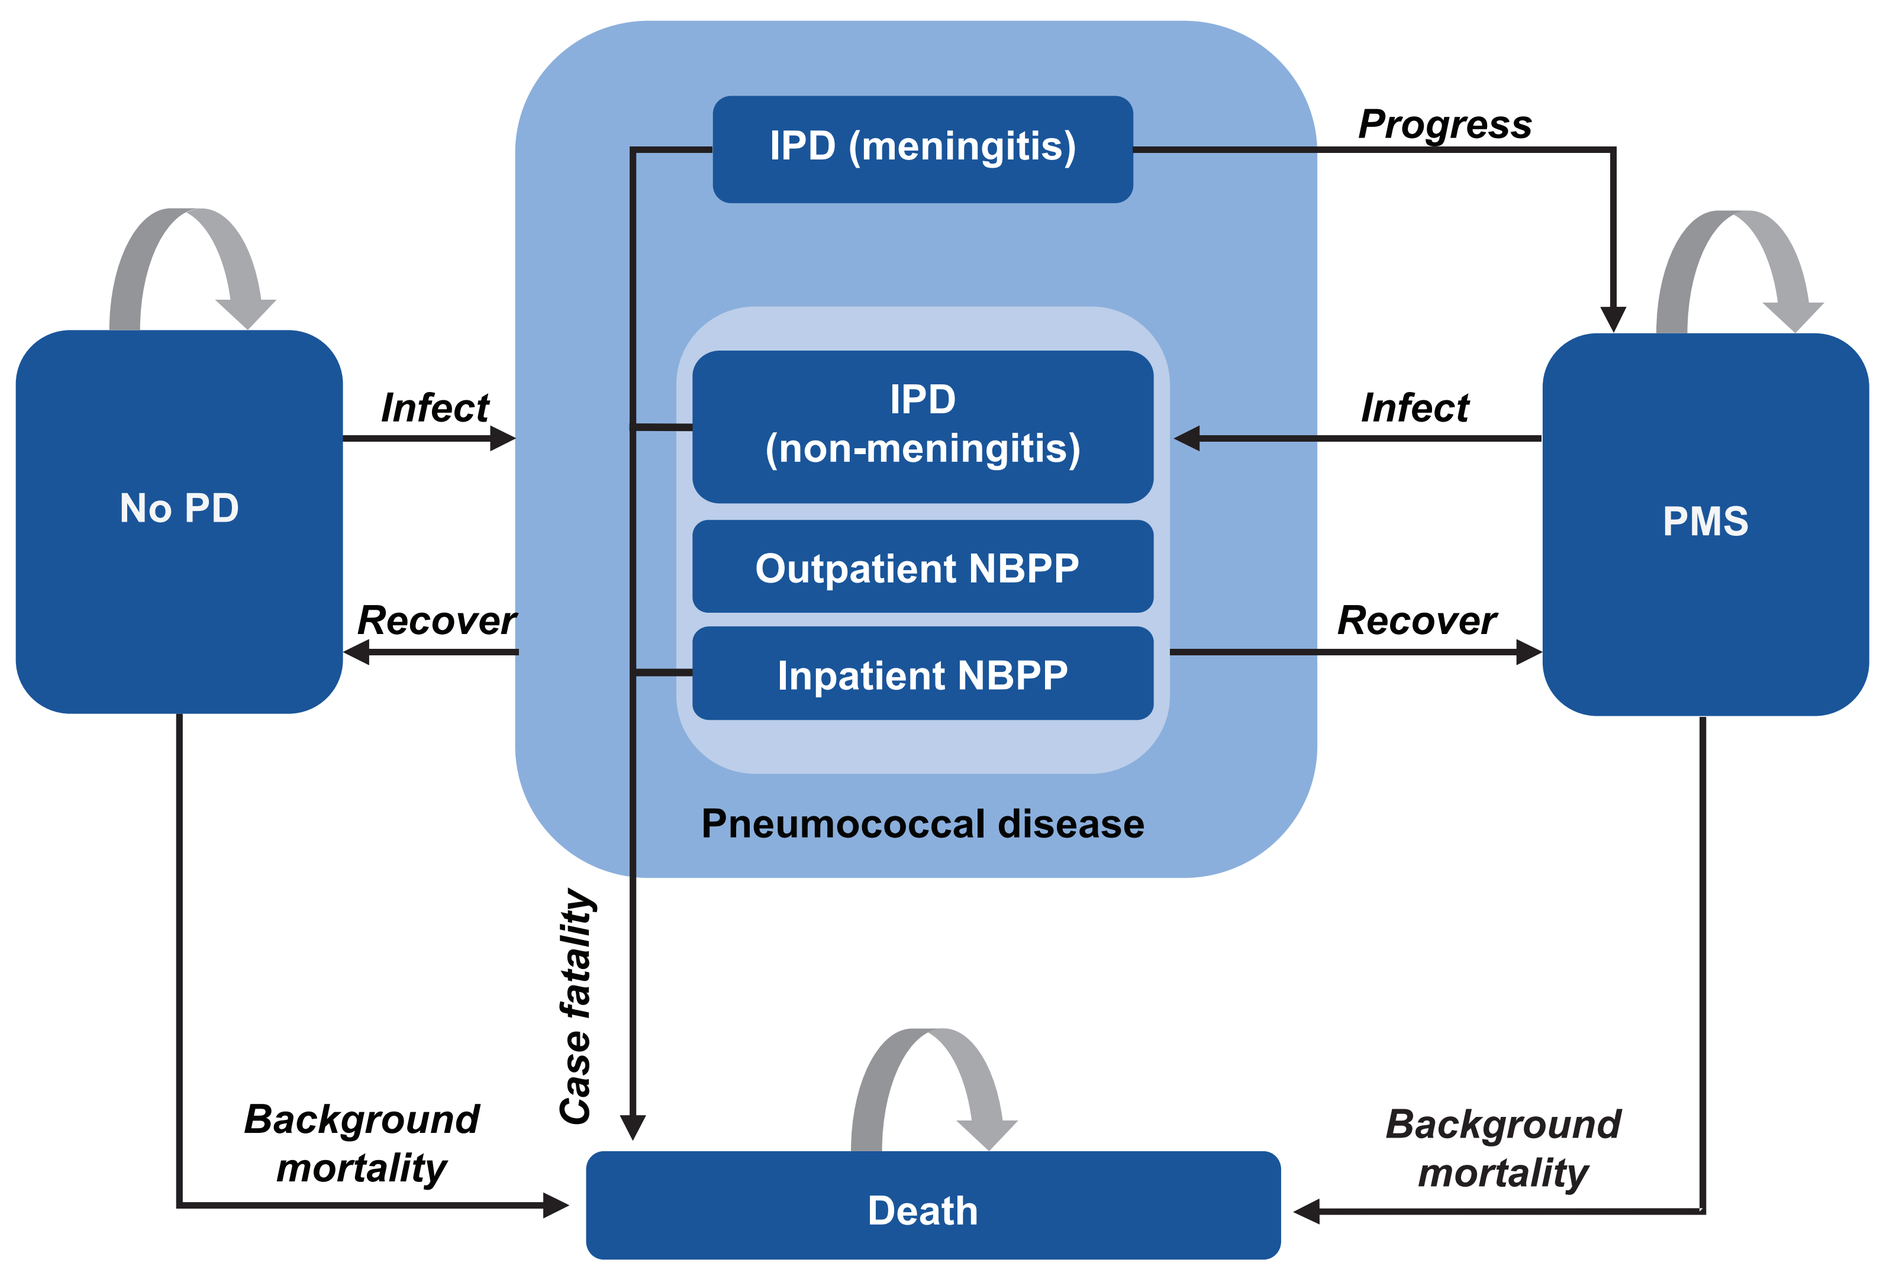
IPD (meningitis)
Progress
No PD
Infect
Recover
IPD
(non-meningitis)
Outpatient NBPP
Inpatient NBPP
Infect
Recover
PMS
Pneumococcal disease
Case fatality
Background
mortality
Death
Background
mortality

PD, pneumococcal disease; IPD, invasive pneumococcal disease; NBPP, non-bacteremic pneumococcal pneumonia; PMS, post meningitis sequelae.

Table 1. Key model parameters[4]

	Low risk	At risk	High risk
Risk group split			
19-49	68.80%	20.60%	10.60%
50-64	47.20%	33.90%	18.90%
65-74	29.40%	43.10%	27.50%
75	16.10%	55.00%	28.90%
IPD incidence (per 100,000 person-years)			
19-49	2.5	8.4	24.0
50-64	6.2	21.0	60.0
65-74	8.6	24.0	37.7
75-84	10.2	28.5	44.7
85-100	14.8	41.5	65.2
NBPP IP incidence (per 100,000 person-years)			
19-49	69	249	467
50-64	199	720	1,349
65–74	471	1,424	1,910
75–84	722	2,183	2,928
85+	1,192	3,601	4,830
NBPP OP incidence (per 100,000 person-years)			
19-49	187	676	1,266
50-64	236	851	1,594
65–74	523	1,579	2,144
75–84	805	2,434	3,303
85+	1,121	3,338	4,598
Case fatality rate	**IPD**	**NBPP IP**	
19-49	0.06836	0.0085	
50-64	0.11717	0.0234	
65-74	0.13118	0.0476	
75-84	0.13517	0.0832	
85-100	0.20995	0.0912	
IPD direct medical cost			
19-64	€2,316	€2,460	€2,820
65+	€2,316	€2,604	€2,892
NBPP IP direct medical cost			
19-64	€229	€645	€2,122
65+	€547	€1,048	€2,338
NBPP OP direct medical cost			
19-64	€14	€12	€2,122
65+	€13	€9	€2,338

Low risk, healthy; at risk, chronic conditions; high risk, immunocompromised, cochlear implant, and CSF leakage. NBPP, non-bacteremic pneumococcal pneumonia; IP, inpatient; IPD, invasive pneumococcal disease; OP, outpatient.

Table 2. Estimated lifetime clinical and economic outcomes when vaccinating with V116 and PCV20 for ages 19-64 with risk conditions

	V116	PCV20	No vaccine	Cases/costs averted, compared to “no vaccine”		Additional reduction by V116, compared to PCV20
				by V116	by PCV20	
Clinical outcomes (undiscounted)						
IPD cases	25,910	26,142	26,663	752	520	232 (44.5%)
NBPP-IP cases	1,296,580	1,300,973	1,310,926	14,346	9,953	4,392 (44.1%)
NBPP-OP cases	1,503,061	1,509,334	1,523,549	20,488	14,215	6,273 (44.1%)
IPD deaths	3,427	3,452	3,509	82	57	25 (44.5%)
NBPP IP deaths	79,624	79,750	80,033	409	284	125 (44.1%)
Economic outcomes (discounted; in million €)						
Direct cost - IPD	37	37	38	2	1	1 (44.5%)
Direct cost - NBPP IP	918	922	931	13	9	4 (44.3%)
Direct cost - NBPP OP	757	760	767	10	7	3 (44.3%)

IP, inpatient; IPD, invasive pneumococcal disease; NBPP, non-bacteremic pneumococcal pneumonia; OP, outpatient; PCV20, 20-valent pneumococcal conjugate vaccine; PMS, post meningitis sequelae; V116, an investigational 21-valent pneumococcal conjugate vaccine.

Table 3. Estimated lifetime clinical and economic outcomes when vaccinating with V116 and PCV20 for ages 65+

	V116	PCV20	No vaccine	Cases/costs averted, compared to “no vaccine”		Additional reduction by V116, compared to PCV20
				by V116	by PCV20	
Clinical outcomes (undiscounted)						
IPD cases	8,519	8,764	9,313	794	549	245 (44.6%)
NBPP IP cases	644,732	655,046	678,219	33,487	23,173	10,314 (44.6%)
NBPP OP cases	664,080	674,961	699,407	35,326	24,446	10,881 (44.6%)
IPD deaths	1,433	1,471	1,557	124	86	38 (44.6%)
NBPP IP deaths	53,524	54,335	56,157	2,632	1,822	811 (44.6%)
Economic outcomes (discounted; in million €)						
Direct cost - IPD	17	18	19	2	1	1 (44.6%)
Direct cost - NBPP IP	740	751	775	34	24	10 (44.4%)
Direct cost – NBPP OP	490	494	503	13	9	4 (44.6%)

IP, inpatient; IPD, invasive pneumococcal disease; NBPP, non-bacteremic pneumococcal pneumonia; OP, outpatient; PCV20, 20-valent pneumococcal conjugate vaccine; V116, an investigational 21-valent pneumococcal conjugate vaccine.

Figure 2. Sensitivity analysis–estimated lifetime direct medical cost in ages 19-64 with risk conditions and 65+ saved by V116, compared with PCV20 (2022. €; discounted)

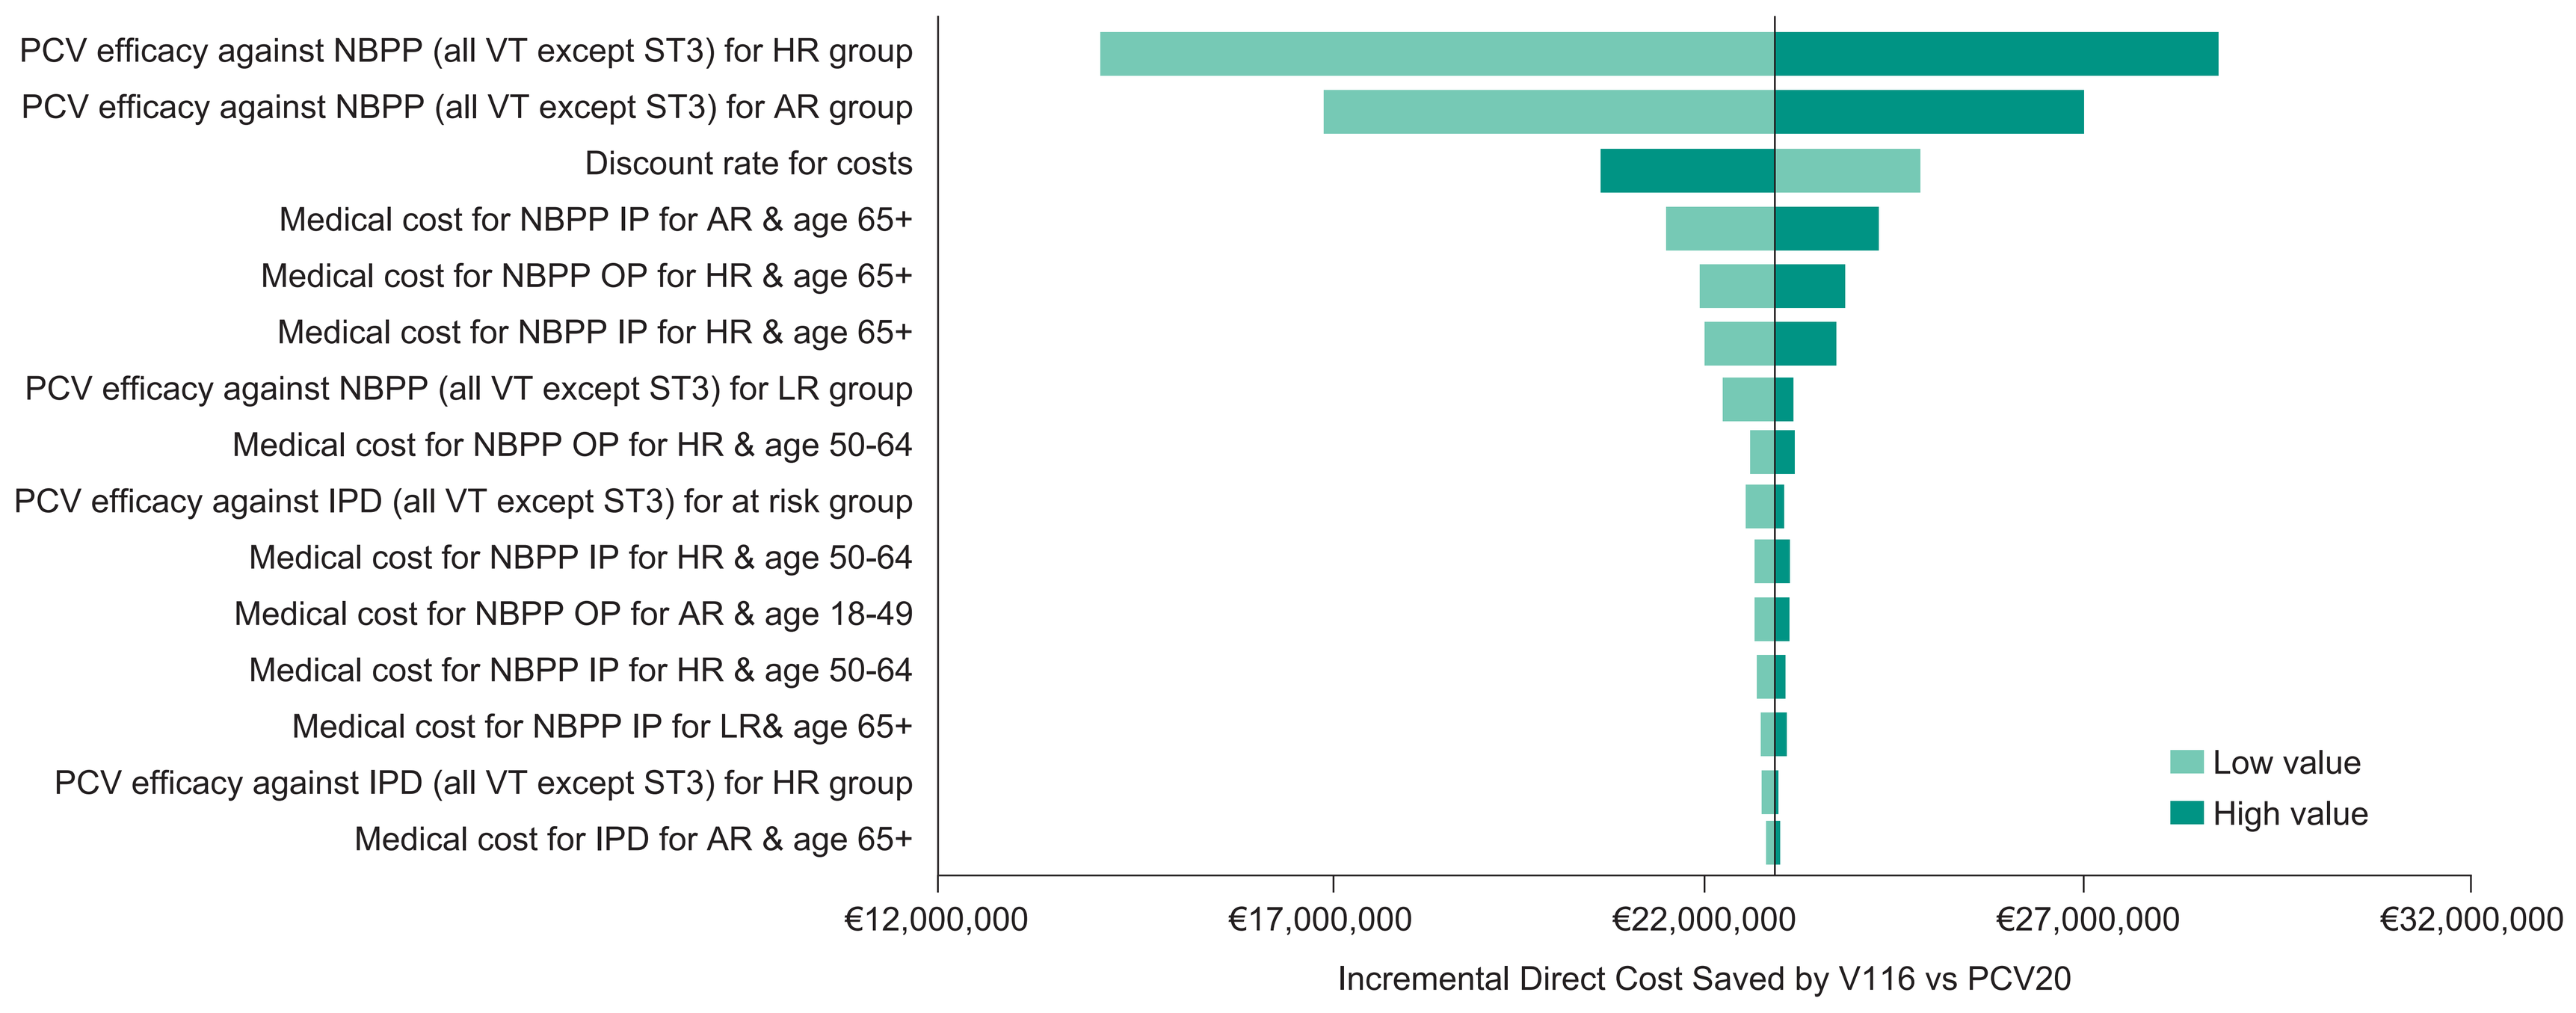
PCV efficacy against NBPP (all VT except ST3) for HR group
PCV efficacy against NBPP (all VT except ST3) for AR group
Discount rate for costs
Medical cost for NBPP IP for AR & age 65+
Medical cost for NBPP OP for HR & age 65+
Medical cost for NBPP IP for HR & age 65+
PCV efficacy against NBPP (all VT except ST3) for LR group
Medical cost for NBPP OP for HR & age 50-64
PCV efficacy against IPD (all VT except ST3) for at risk group
Medical cost for NBPP IP for HR & age 50-64
Medical cost for NBPP OP for AR & age 18-49
Medical cost for NBPP IP for HR & age 50-64
Medical cost for NBPP IP for LR& age 65+
PCV efficacy against IPD (all VT except ST3) for HR group
Medical cost for IPD for AR & age 65+
Low value
High value
€12,000,000
€17,000,000
€22,000,000
€27,000,000
€32,000,000
Incremental Direct Cost Saved by V116 vs PCV20

AR, at risk; HR, high risk; IP, inpatient; IPD, invasive pneumococcal disease; LR, low risk; NBPP, non-bacteremic pneumococcal pneumonia; PCV, pneumococcal conjugate vaccine; OP, outpatient; ST3, serotype 3; VT, vaccine type.

Conclusions

- In both age groups, compared to PCV20, V116 led to a greater reduction of both the health and economic burden associated with IPD and NBPP in Greece
- The addition of V116 to the national vaccination recommendations has the potential to substantially reduce the health and economic burden associated with PD among adults in Greece, compared to PCV20

Corresponding author

Zinan Yi; email: zinan.yi@merck.com

Disclosures

All authors are employees of Merck Sharp & Dohme LLC or MSD Greece subsidiaries of Merck & Co., Inc., Rahway, NJ, USA and may hold stock or stock options in Merck & Co., Inc., Rahway, NJ, USA.

V116 was developed by Merck & Co., Inc., Rahway, NJ, USA.

Presented at ISPOR Europe; Barcelona, Spain; 17-20 November 2024.

References

1. Maraki S, et al. *Infect Chemother.* 2018;50(4):328-339.
2. Xirogianni A, et al. *Vaccine.* 2022;40(34):5079-5087.
3. Stoecker C. Economic assessment of PCV15 & PCV20. Presented at ACIP; Atlanta, GA, USA; June 25, 2021.
4. Gourzoulidis G, et al. *Front Public Health.* 2023;11:1229524.
5. Tountas et al. *Panhellenic Health Survey*, Hellas Health VIII. https://www.nextdeal.gr/sites/default/files/sitefiles_2021-12/hellas_health_viii_synenteyxi_typoy.pdf

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the Congress or the author of this poster.

https://bit.ly/3UxodzS

Copyright © 2024 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.