Real-World Evidence (RWE) Study on Burden, Healthcare Resource Utilization, & Cost of Illness Among Patients Diagnosed With Myopia in Germany: Retrospective, Longitudinal, Observational Cohort Claims Data Study

RWD177

Marc Pignot[1], Mohammed Dagher[1], Tarcyane Barata Garcia[2], Nils Kossack[2], Alexander K. Schuster[3], Kun Shi-van Wielink[4 #]

[1]ZEG – Berlin Center for Epidemiology and Health Research, Berlin, Germany

[2]WIG2 – Scientific Institute for Health Economics and Health System Research, Leipzig, Germany

[3]Department of Ophthalmology, University Medical Center Mainz, Geb. 101, Langenbeckstr. 1, 55131 Mainz, Germany

[4]Santen SA, Amsterdam, Netherlands

Presenting Author

BACKGROUND & INTRODUCTION

- Myopia (near-sightedness) is caused by eye elongation or increased refractive power, causing light to focus in front of the retina, resulting in blurred distance vision (Biswas et al., 2024). Defined by the International Myopia Institute as a spherical equivalent (SRE) ≤ -0.50 diopters (D); high myopia is ≤ -6.00 D (Flitcroft et al., 2019).
- Global myopia is increasing, with estimates suggesting half of the world population will be myopic by 2050, and 10% will have high myopia (Holden et al., 2016).
- Myopia can be corrected optically, but severe complications include myopic macular degeneration (MMD), retinal detachment (RD), cataracts, and open-angle glaucoma (OAG), potentially leading to irreversible vision loss (Haarman et al., 2020). MMD is the most significant complication, especially in high myopia. RD is the most sight-threatening among common peripheral retinal lesions (Haarman et al., 2020). Myopia is already the leading cause of irreversible visual impairment in the working population, with a major socio-economic impact (Naidoo et al., 2018).
- Limited real-world data on myopia in Germany hinders understanding of disease burden, healthcare resource utilization (HCRU), and costs. A retrospective real-world study is therefore designed to understand the patient characteristics, outcomes, health care resource utilization (HCRU) and cost.
- The WIG2 database is an anonymized healthcare claims database with longitudinal data from approx. 4.5 million Germans insured by one of various German statutory health insurer (SHIs).

METHOD

This retrospective observational study utilized data from the anonymized WIG2 benchmark database in Germany. Patients diagnosed with myopia by an ophthalmologist (ICD-10-GM H52.1) between 2018 and 2022 were categorized into mild/moderate and high myopia using a proxy algorithm based on prescription lenses. Age groups (in years) were classified as follows: preschool (0–5), primary school (6–9), secondary school (10–17), adults (18–59), elderly (60–75), and geriatric (76+). One-to-many propensity score matching was applied to ensure balanced comparisons between the mild/moderate and high myopia subgroups, accounting for non-ophthalmic healthcare resource utilization (HCRU), costs, and comorbidities. The matched samples were then analyzed to assess ophthalmic-related HCRU, costs, and the prevalence of ocular comorbidities in 2022.

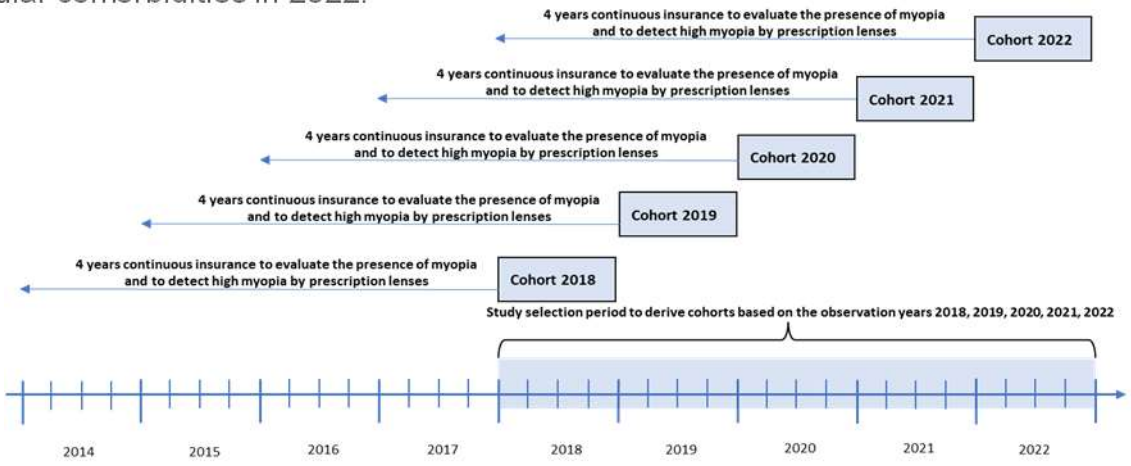
4 years continuous insurance to evaluate the presence of myopia and to detect high myopia by prescription lenses
Cohort 2022
4 years continuous insurance to evaluate the presence of myopia and to detect high myopia by prescription lenses
Cohort 2021
4 years continuous insurance to evaluate the presence of myopia and to detect high myopia by prescription lenses
Cohort 2020
4 years continuous insurance to evaluate the presence of myopia and to detect high myopia by prescription lenses
Cohort 2019
4 years continuous insurance to evaluate the presence of myopia and to detect high myopia by prescription lenses
Cohort 2018
Study selection period to derive cohorts based on the observation years 2018, 2019, 2020, 2021, 2022
2014
2015
2016
2017
2018
2019
2020
2021
2022

RESULTS

A total of 537,506 myopia patients were identified in the WIG2 dataset from 2018 to 2022. Of these, 524,756 patients met the inclusion criteria for the low-to-moderate myopia cohort, while 25,465 patients were included in the high myopia cohort.

The overall prevalence of myopia (across all severity groups) in the WIG2-DB and SHI populations from 2018 to 2022 is reported, stratified by the entire population, as well as by sex. Myopia prevalence was consistently higher in females compared to males across all years. Over the study period, there was a slight decline in overall myopia prevalence for both sexes. In the WIG2-DB population, myopia prevalence decreased from 13.52% in 2018 to 13.24% in 2022, while in the SHI population, it declined from 13.75% to 13.28% over the same period. A similar trend was observed in the sex-stratified data, with females in the SHI population showing a decrease in prevalence from 16.55% to 16.06%, and males decreasing from 10.73% to 10.3%. (Figure 2)

The overall myopia incidence from 2018 to 2022, measured across different age groups in both the WIG2-DB and SHI populations, along with their respective 95% confidence intervals (CIs), showed an age-related increase. Myopia incidence peaked in secondary school students and adults. In 2022, the incidence among secondary school students was approximately 2.31% (95% CI: [2.25-2.38] DB, [2.24-2.38] SHI). Preschool children consistently had the lowest incidence rates, starting at 0.20% in 2018 (95% CI: [0.18-0.23] DB, [0.19-0.23] SHI) and rising to 0.43% in 2022 (95% CI: [0.39-0.47] DB, [0.40-0.47] SHI). The data indicates an overall upward trend in myopia incidence over the years, with marked differences among age groups. Figure 3.

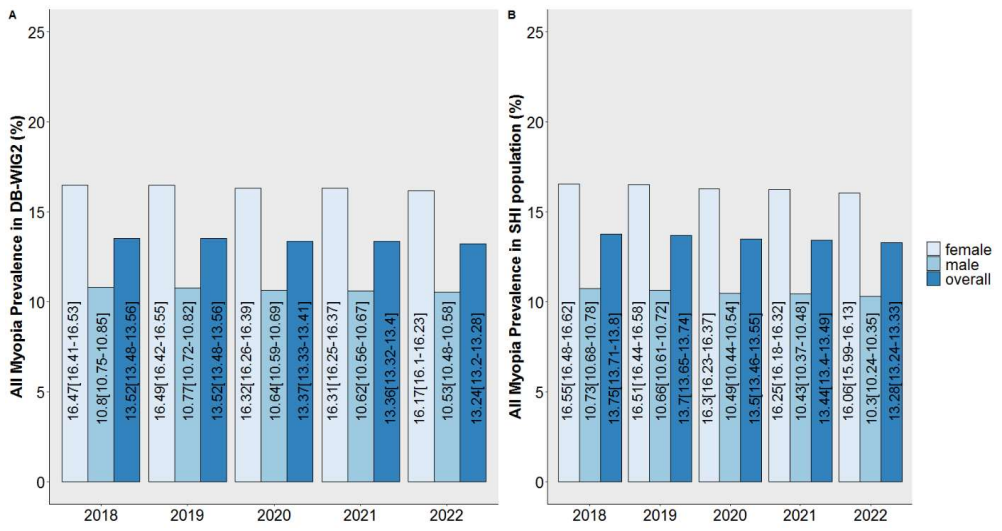
A
All Myopia Prevalence in DB-WIG2 (%)
16.47[16.41-16.53]
10.8[10.75-10.85]
13.52[13.48-13.56]
16.49[16.42-16.55]
10.77[10.72-10.82]
13.52[13.48-13.56]
16.32[16.26-16.39]
10.64[10.59-10.69]
13.37[13.33-13.41]
16.31[16.25-16.37]
10.62[10.56-10.67]
13.36[13.32-13.4]
16.17[16.1-16.23]
10.53[10.48-10.58]
13.24[13.2-13.28]
2018
2019
2020
2021
2022
B
All Myopia Prevalence in SHI population (%)
16.55[16.48-16.62]
10.73[10.68-10.78]
13.75[13.71-13.8]
16.51[16.44-16.58]
10.66[10.61-10.72]
13.7[13.65-13.74]
16.3[16.23-16.37]
10.49[10.44-10.54]
13.5[13.46-13.55]
16.25[16.18-16.32]
10.43[10.37-10.48]
13.44[13.4-13.49]
16.06[15.99-16.13]
10.3[10.24-10.35]
13.28[13.24-13.33]
2018
2019
2020
2021
2022
female
male
overall

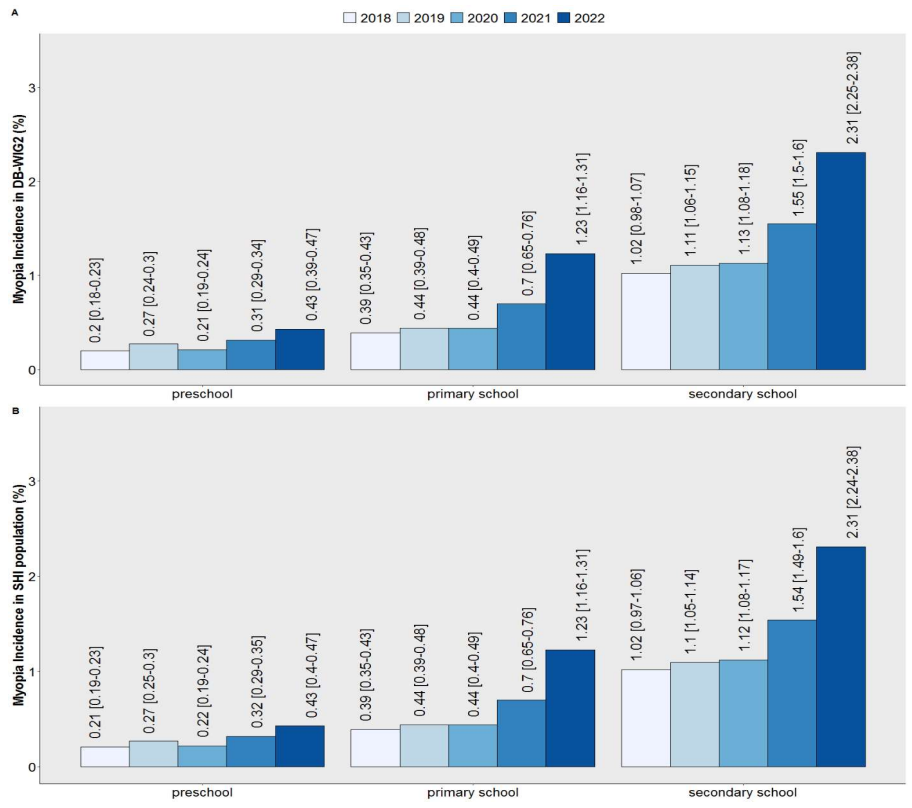
2018 2019 2020 2021 2022
A
Myopia Incidence in DB-WIG2 (%)
0.2 [0.18-0.23]
0.27 [0.24-0.3]
0.21 [0.19-0.24]
0.31 [0.29-0.34]
0.43 [0.39-0.47]
0.39 [0.35-0.43]
0.44 [0.39-0.48]
0.44 [0.4-0.49]
0.7 [0.65-0.76]
1.23 [1.16-1.31]
1.02 [0.98-1.07]
1.11 [1.06-1.15]
1.13 [1.08-1.18]
1.55 [1.5-1.6]
2.31 [2.25-2.38]
preschool
primary school
secondary school
B
Myopia Incidence in SHI population (%)
0.21 [0.19-0.23]
0.27 [0.25-0.3]
0.22 [0.19-0.24]
0.32 [0.29-0.35]
0.43 [0.4-0.47]
0.39 [0.35-0.43]
0.44 [0.39-0.48]
0.44 [0.4-0.49]
0.7 [0.65-0.76]
1.23 [1.16-1.31]
1.02 [0.97-1.06]
1.1 [1.05-1.14]
1.12 [1.08-1.17]
1.54 [1.49-1.6]
2.31 [2.24-2.38]
preschool
primary school
secondary school

In 2022, the mean annual healthcare resource utilization (HCRU) and the proportion of individuals with high myopia-associated visual disorders were higher in people with high myopia compared to those with mild/moderate myopia across all age groups. Individuals with high myopia had a greater average number of ophthalmic visits, with geriatric patients averaging 2.93 visits per year, compared to 2.18 visits for those with mild/moderate myopia. Elderly and geriatric patients with high myopia also required significantly more ophthalmic hospital days (0.16/year and 0.06/year, respectively) and hospitalizations (0.05/year and 0.02/year) than those with mild/moderate myopia (0.05/year and 0.02/year, and 0.02/year and 0.01/year, respectively).High myopia was associated with a greater prevalence of visual disorders, such as myopic macular degeneration, especially among elderly (0.16%) and geriatric (0.12%) patients. Additionally, individuals with high myopia incurred higher ophthalmic expenses across all age groups, with the geriatric cohort experiencing significantly elevated costs across all categories. Figure 4.

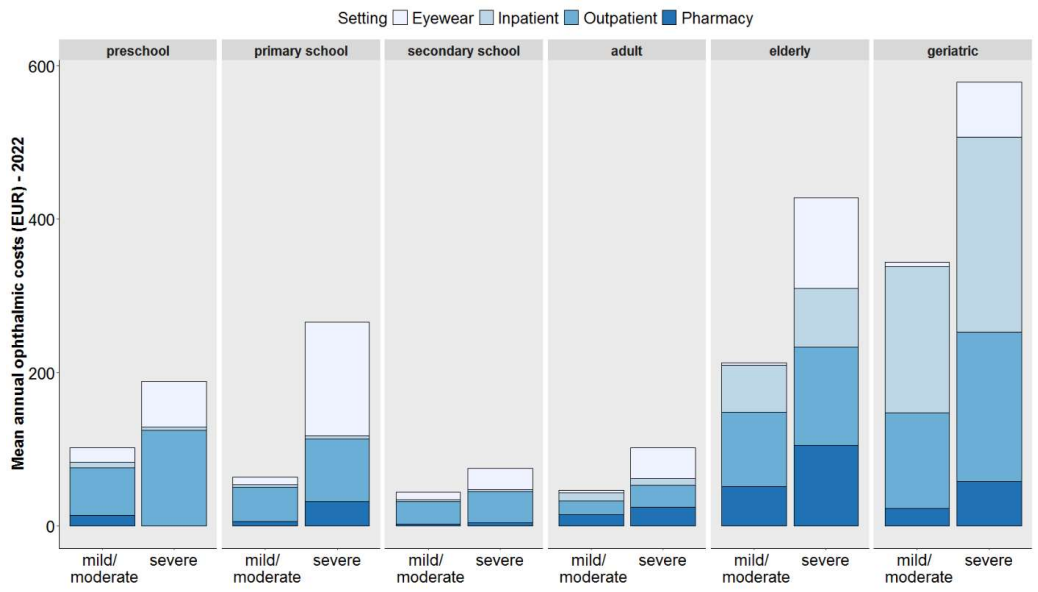
Setting Eyewear Inpatient Outpatient Pharmacy
preschool
primary school
secondary school
adult
elderly
geriatric
Mean annual ophthalmic costs (EUR) - 2022
600
400
200
0
mild/ moderate
severe

CONCLUSIONS

Myopia prevalence was highest among secondary school students, though it decreased over time.

Myopia incidence followed an age-related pattern, with secondary school students showing the highest rates and preschool children the lowest. Despite a decline, secondary school students maintained the highest prevalence of high myopia.

Individuals with high myopia had more ophthalmic visits across all age groups.

Elderly and geriatric patients with high myopia required more hospital days and hospitalizations than those with mild/moderate myopia.

High myopia was linked to a greater prevalence of visual disorders and incurred higher ophthalmic expenses across all age groups, especially in the geriatric cohort.

The higher costs for outpatient care, inpatient care, pharmacy, and eyewear, highlighting the need for better healthcare strategies.

Several limitations still need to be explored.

ACKNOWLEDGMENT

- This study was supported by Santen GmbH.
- The WIG2 Institute, which owns the rights to the research database, has conducted the data analysis contracted by ZEG Berlin.

Wissenschaftliches Institut für Gesundheitsökonomie und Gesundheitssystemforschung

ISPOR Europe 2024 #ISPOREurope