Introduction of a New Dengue Vaccine (TAK-003) in Brazil's Public Health System: Impact on Public Health and Cost-Effectiveness Across Different Age Groups

Mendes da Silva B[1], Shen J[2], Kharitonova E[3], Tytula A[4], Zawieja J[4], Towle P[2], Langer J[2], Hanley R[2]

[1]Takeda Farmacêutica Brasil, São Paulo, SP, Brazil, [2]Takeda Pharmaceuticals International AG, Zürich, ZH, Switzerland,
[3]External consultant hired by Putnam PHMR, Paris, France, [4]Putnam, Krakow, Poland

ISPOR

EPH68

Background

The incidence of dengue has grown substantially over the last century, and it is estimated that the observed growth will continue in the coming years, with over 1 billion new people at risk of dengue by 2080.[1,2] The increase in cases is driven by climate change, urbanization, and increased human migration.[3,4] Dengue fever represents a significant burden for the Brazilian population. Between January and May 2024, more than 4.5 million cases and 2300 deaths related to dengue had been reported, three and two times increase respectively, compared to the same period in 2023[5] (**Figure 1**). Given that there is no specific treatment for dengue and prevention programs based on vector control and individual care are insufficient on their own, there is a significant unmet public health need for a safe and effective vaccine, applicable regardless of the population's prior serological status, capable of substantially reducing the burden of this disease in the country and worldwide. TAK-003, a new dengue vaccine, has demonstrated efficacy in a robust clinical development program, and currently there is a need to understand its utility in a real-world public health setting. In the pivotal phase-3 trial (DEN-301), TAK-003 demonstrated efficacy against virologically confirmed dengue (VCD) and hospitalized cases up to 4.5 years.[6]

Figure 1. Dengue cases and deaths in the Brazilian population[5]

January to May, 2024

4.5 million cases ↑ 3x

2.3 thousand deaths ↑ 2x

Increase compared to the same period in 2023

Objectives

This study evaluates the public health impact and cost-effectiveness of vaccination strategies across different age groups, compared to no vaccination.

Methods

A **cost-effectiveness analysis** using a static model with a dynamic component was **performed** to assess the impact of vaccination with TAK-003 on epidemiologic, economic and quality of life outcomes in Brazil, capturing both direct and indirect protections[3,4]. The indirect protection was captured by the dynamic component of the model, which allowed varying the probability of infection at each model cycle based on the number of infections predicted at the previous cycle, unlike in a conventional static model where the probability of infection is constant over time. Model inputs were derived from Brazil's official databases and the model was fitted to the country's dengue epidemiology and an expansion factor of 1.8 was considered for both incidence and mortality to account for underreporting.[7] Vaccine efficacy was derived from DEN-301 study and extrapolated over 20 years to assess the impact over this period. Vaccination strategies were analyzed within each five-year age group across the age indication locally approved (**Figure 4**), assuming 90% coverage each year, which means that every year 90% of previously unvaccinated subjects would be vaccinated, and an illustrative cost of US$ 25 per dose. Benefits were measured by quality-adjusted life years (QALY). Both the payer's and society's perspectives were covered, in accordance with the World Health Organization (WHO) Guide to Standardizing Economic Evaluations of Immunization Programs[8]

Results

Model calibration was conducted by fitting the model to the average age-specific incidence based on data from 2014 to 2022, excluding the COVID pandemic years and Zika outbreaks that resulted in significant underreporting, and minimizing the difference between the predicted and observed incidence rates of symptomatic dengue in each specific age band, as per **Figure 2**. Furthermore, in the model the percentage of seropositivity at the age of 9 years was 19.5%, consistent with what is expected for this age group, taking into account the variations in disease incidence across the country (**Figure 3**).[9]

Figure 2. Model calibration according to the incidence curve: observed vs predicted values

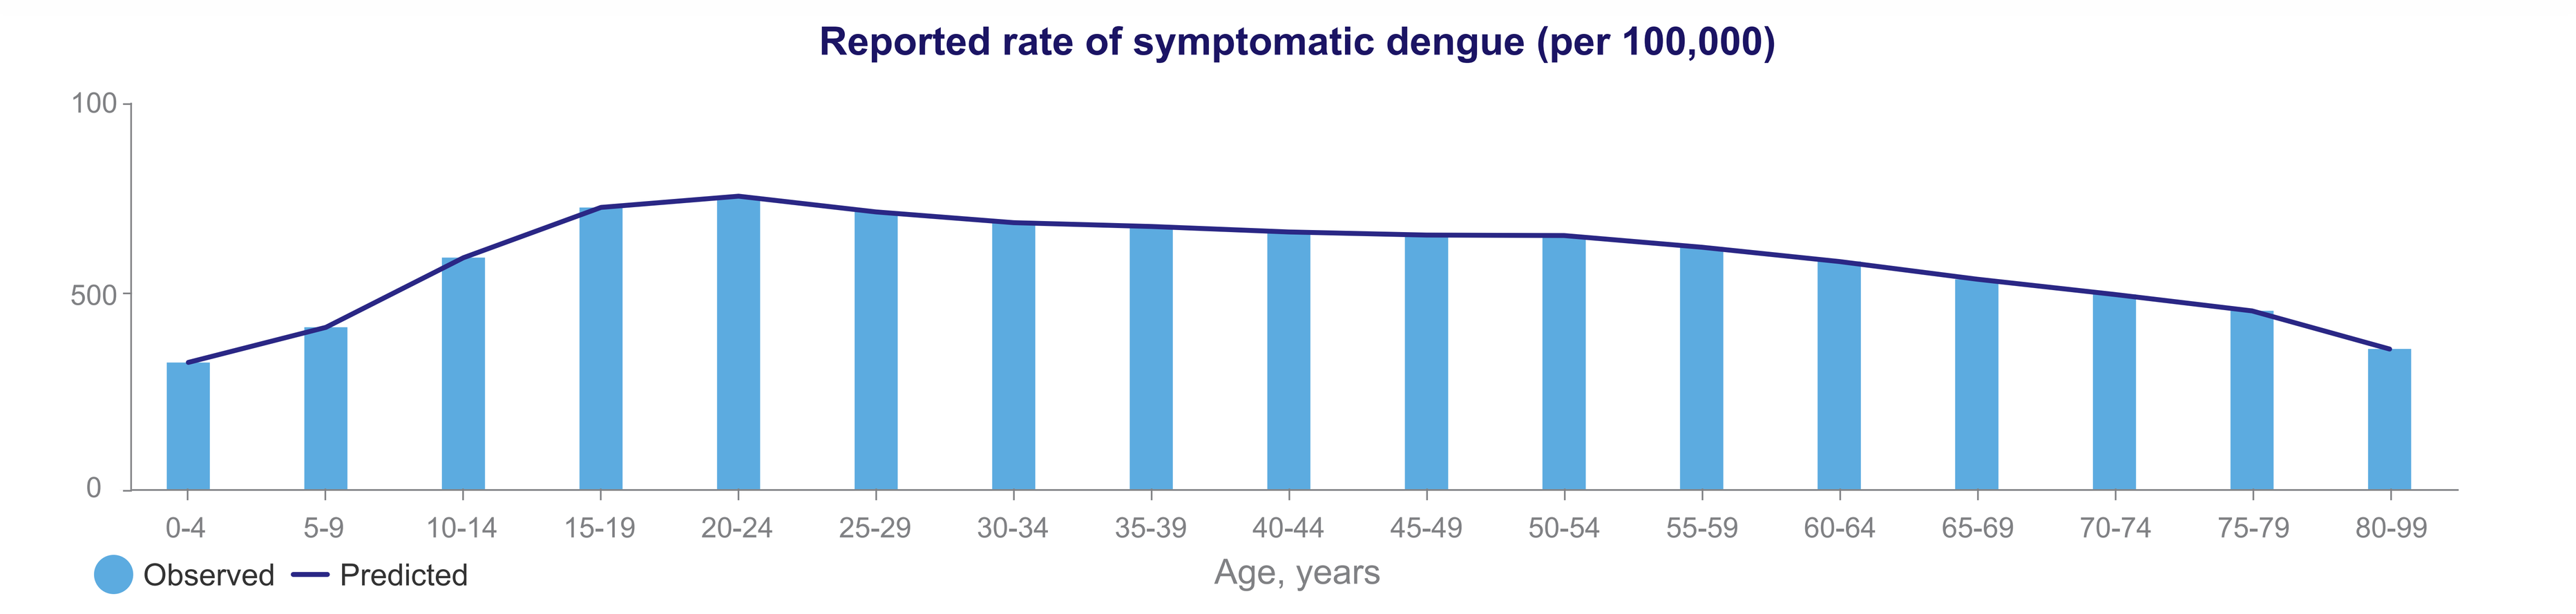
Reported rate of symptomatic dengue (per 100,000)
100
500
0
0-4
5-9
10-14
15-19
20-24
25-29
30-34
35-39
40-44
45-49
50-54
55-59
60-64
65-69
70-74
75-79
80-99
Observed
Predicted
Age, years

Figure 3. Seroprevalence by age predicted by the model

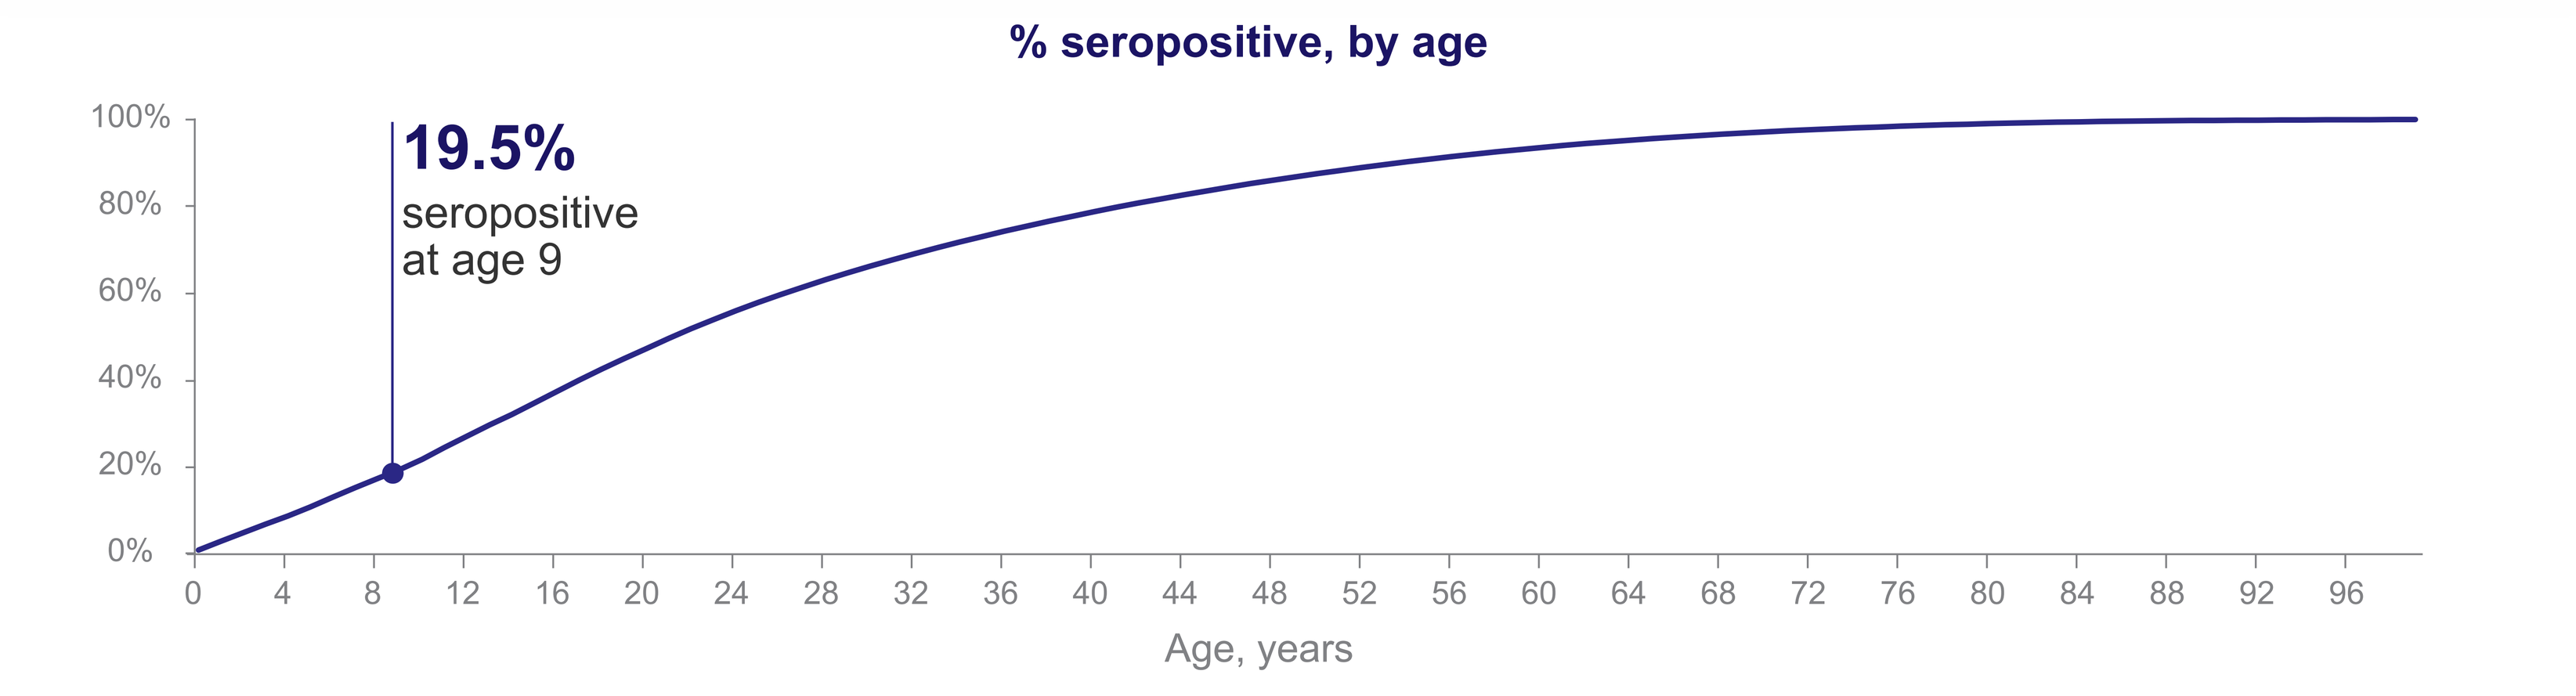
% seropositive, by age
100%
80%
60%
40%
20%
0%
19.5%
seropositive
at age 9
0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76 80 84 88 92 96
Age, years

Modeling results revealed that implementing vaccination strategies with TAK-003 holds the potential to decrease by up to 26% and 29% symptomatic cases[10] and hospitalizations[11,12] respectively, compared to no vaccination, depending on the age strategy assessed, **even when vaccinating narrow five-year groups** (**Figure 4**).

Figure 4. Proportion of symptomatic and hospitalized cases avoided vs no vaccination per routine 5 years age-group simulated over a 20 year time horizon

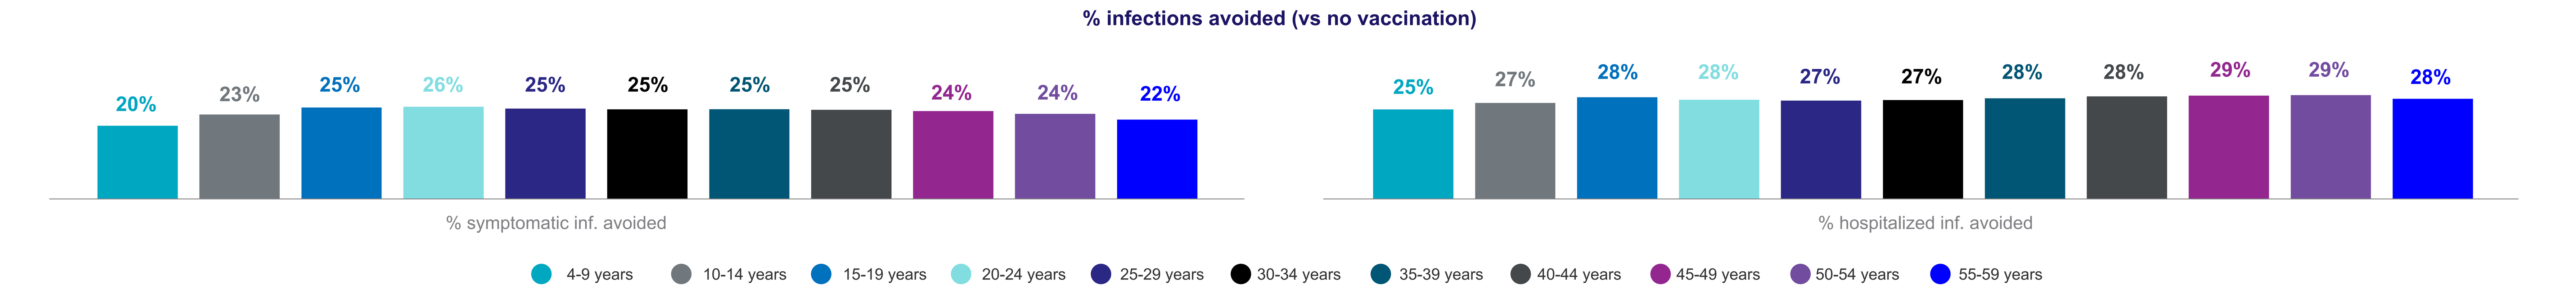
% infections avoided (vs no vaccination)
20% 23% 25% 26% 25% 25% 25% 25% 24% 24% 22%
% symptomatic inf. avoided
25% 27% 28% 28% 27% 27% 28% 28% 29% 29% 28%
% hospitalized inf. avoided
4-9 years
10-14 years
15-19 years
20-24 years
25-29 years
30-34 years
35-39 years
40-44 years
45-49 years
50-54 years
55-59 years

The modeling results show that TAK-003 significantly reduces the costs associated with dengue management (**Figure 5**). This is important because the direct and indirect costs associated with dengue create significant financial strain, making the reduction of its economic burden a top priority for the WHO.[13]

Figure 5. Predicted cost savings among the simulated 5 years age-group vaccination scenarios over 20 year time horizon (versus no vaccination)

Medical costs
Savings up to
US$ 429,904,696
(20-24 yo scenario)

Non-medical costs
Savings up to
US$ 102,923,203
(20-24 yo scenario)

Productivity lost cost
Savings up to
US$ 1,717,907,116
(15-19 yo scenario)

School absenteeism cost
Savings up to
US$ 123,783,401
(10-14 yo scenario)

TAK-003 vaccination was dominant in most scenarios from a societal perspective, with up to **353,748 QALYs** gained and savings of up to US$ 260 million (**Figure 6**). From a payer's perspective, TAK-003 was cost-effective across all evaluated scenarios, considering the **US$ 8,264/QALY** threshold set by the Brazilian Health Technology Assessment (HTA) agency (**Figure 7**)[14].

Figure 6. Cost-effectiveness plane (societal perspective)

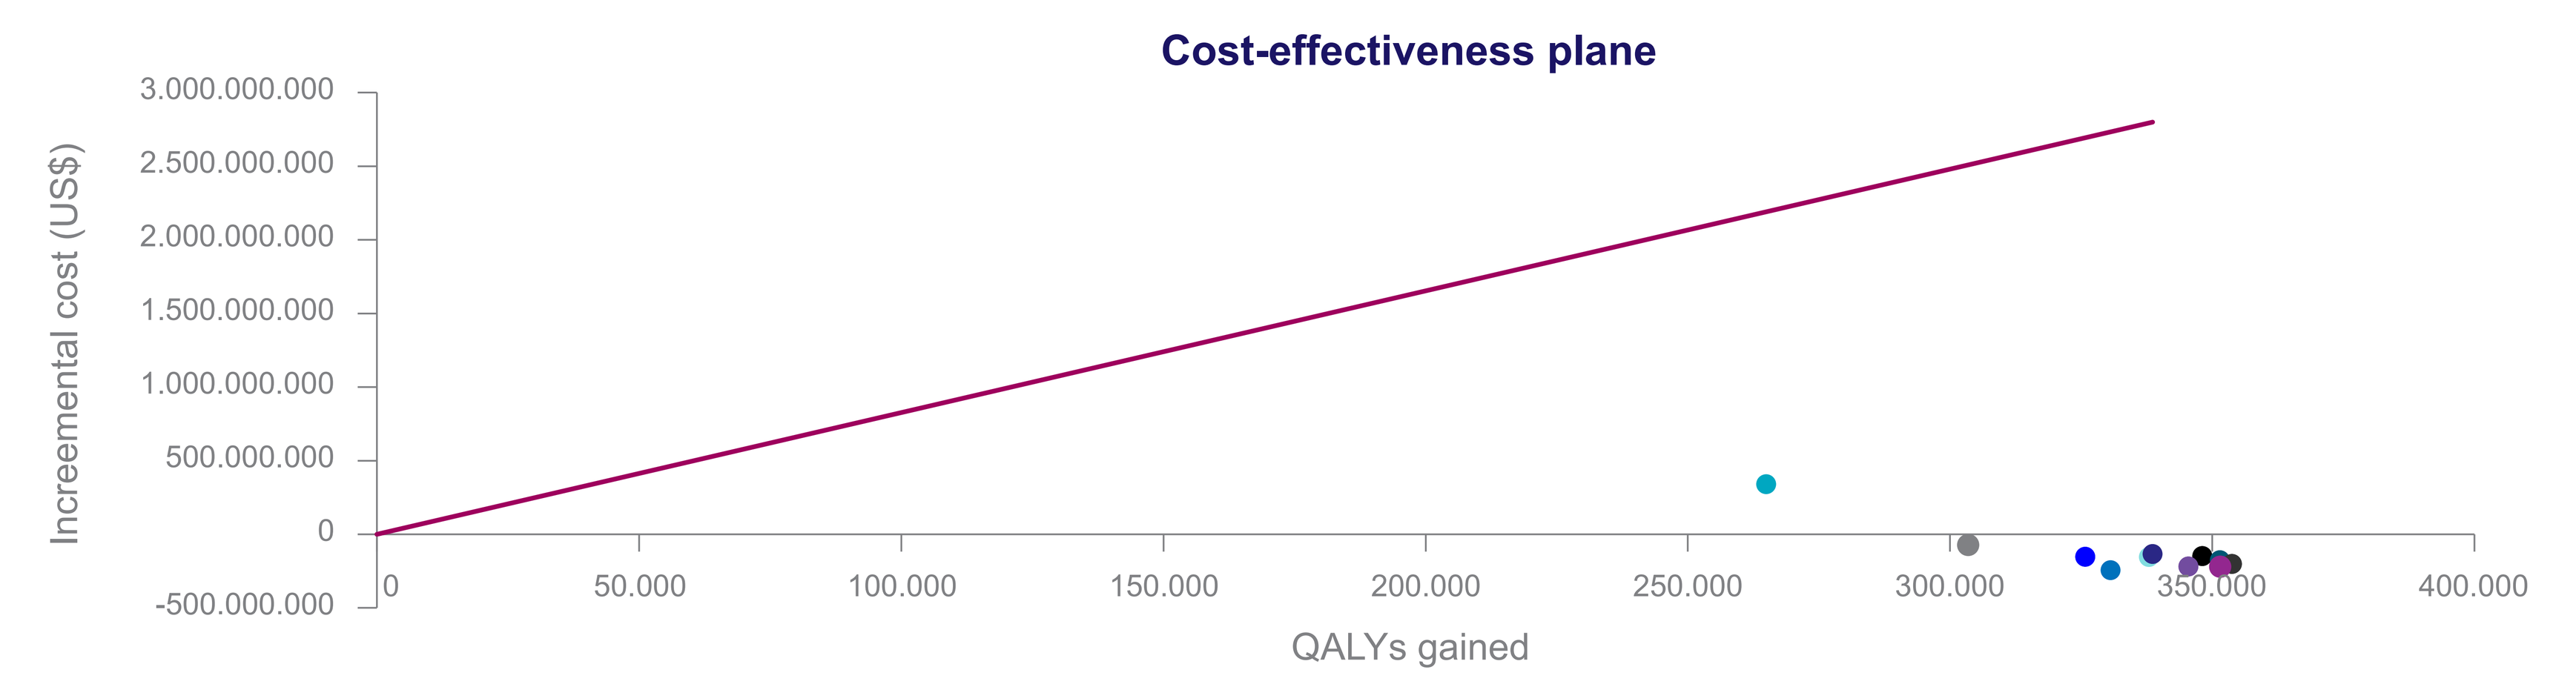
Cost-effectiveness plane
Increemental cost (US$)
3.000.000.000
2.500.000.000
2.000.000.000
1.500.000.000
1.000.000.000
500.000.000
0
-500.000.000
0 50.000 100.000 150.000 200.000 250.000 300.000 350.000 400.000
QALYs gained

Figure 7. Cost-effectiveness plane (payer perspective)

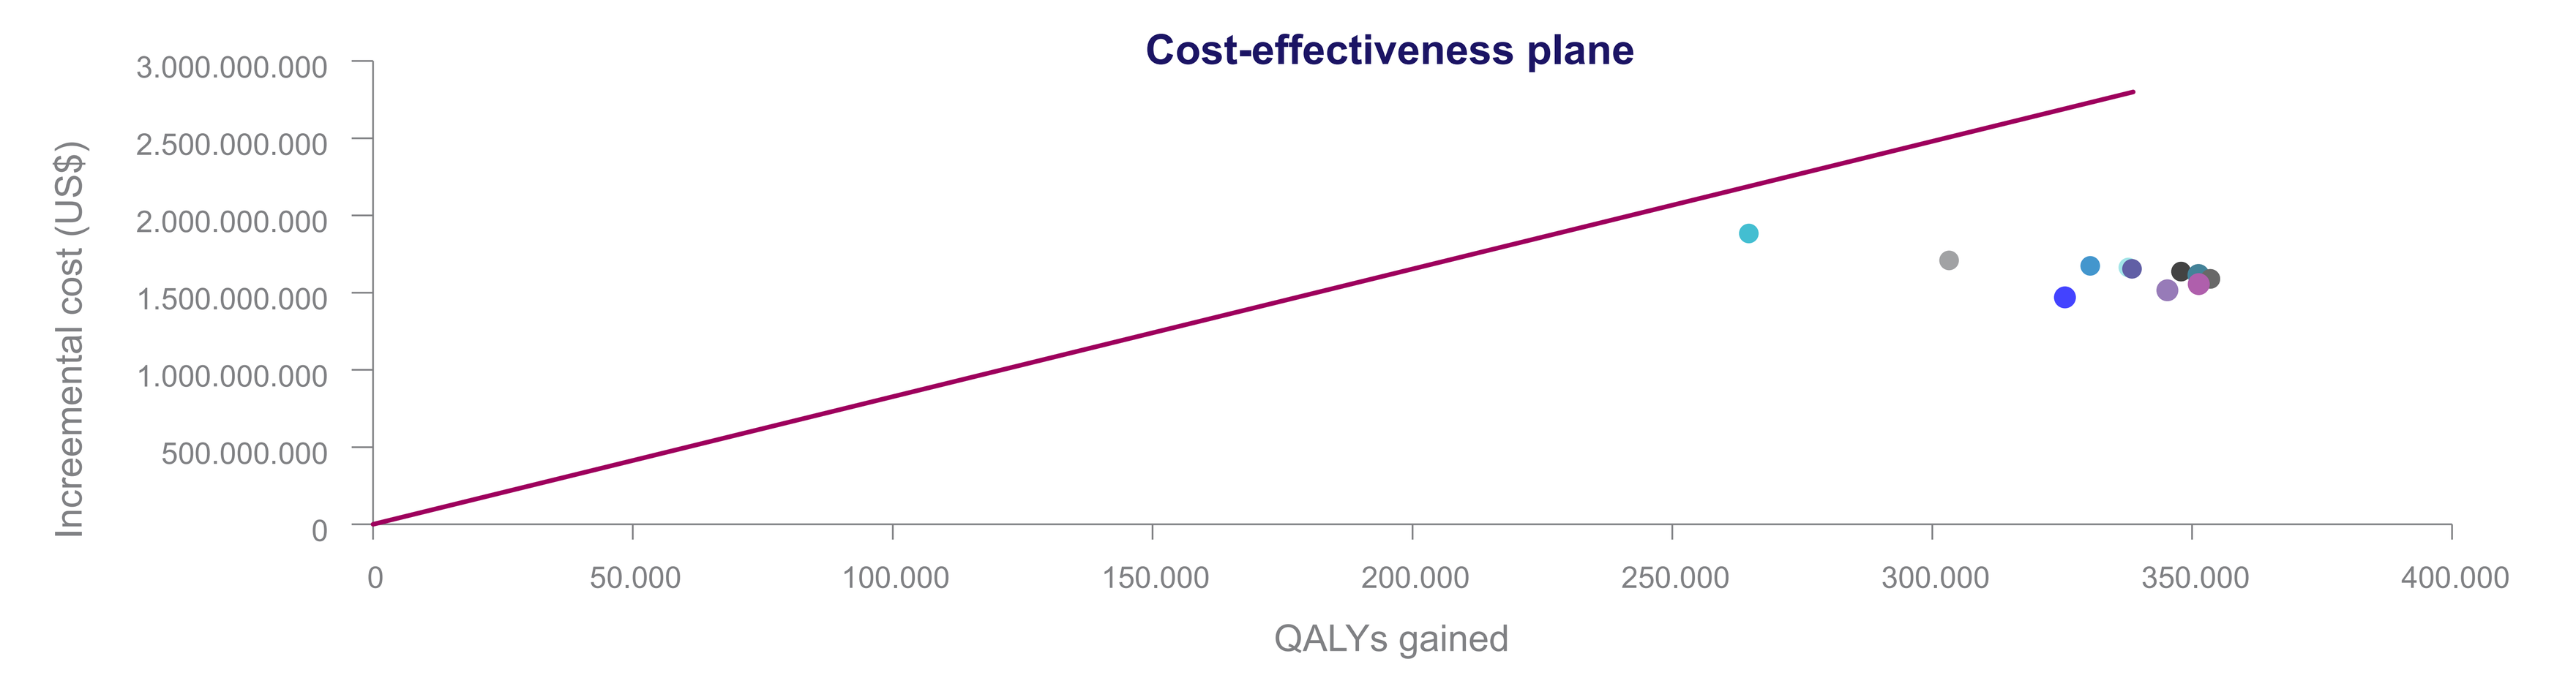
Cost-effectiveness plane
Increemental cost (US$)
3.000.000.000
2.500.000.000
2.000.000.000
1.500.000.000
1.000.000.000
500.000.000
0
0 50.000 100.000 150.000 200.000 250.000 300.000 350.000 400.000
QALYs gained

4-9 years · 10-14 years · 15-19 years · 20-24 years · 25-29 years · 30-34 years · 35-39 years · 40-44 years · 45-49 years · 50-54 years · 55-59 years · Willingness to pay

The budgetary impact of its inclusion in the national immunization schedule is largely offset when considering the broader social perspective, including the macroeconomic cost context, and has even greater potential considering that the benefits of a vaccination program grow exponentially in the long term.

Conclusions

TAK-003 represents a major advancement in dengue prevention and offers a sustainable long-term solution to address the growing dengue burden in the Brazilian population. The modelling shows that TAK-003 vaccination program lead to a significant reduction in disease burden and costs, regardless of the age group evaluated, and serve as an additional effective dengue control measure for the study population. Through this analysis, which considered the vaccination of narrow groups composed of five individual ages, it is plausible to expect that vaccinating larger groups has the potential to prevent even more infections and generate greater savings, both directly and indirectly, related to disease management.

HTA: Health Technology Assessment; **CP:** cross-protection; **DALY:** disability-adjusted life-year; **DEN:** Dengue; **DSA:** deterministic sensitivity analysis; **PSA:** probabilistic sensitivity analysis; **QALY:** quality-adjusted life-year; **TAK:** Takeda; **VCD:** virologically confirmed dengue, **WHO:** World Health Organization.

References: **1.** Messina JP, Brady OJ, Scott TW, Zou C, Pigott DM, Duda KA, et al. Global spread of dengue virus types: mapping the 70 year history. Trends Microbiol. março de 2014;22(3):138–46; **2.** Huy R, Buchy P, Conan A, Ngan C, Ong S, Ali R, et al. National dengue surveillance in Cambodia 1980-2008: epidemiological and virological trends and the impact of vector control. Bull World Health Organ. setembro de 2010;88(9):650–7; **3.** Ryan SJ, Carlson CJ, Mordecai EA, Johnson LR. Global expansion and redistribution of Aedes-borne virus transmission risk with climate change. PLoS Negl Trop Dis. março de 2019;13(3):e0007213–e0007213; **4.** Messina JP, Brady OJ, Golding N, Kraemer MUG, Wint GRW, Ray SE, et al. The current and future global distribution and population at risk of dengue. Nat Microbiol. setembro de 2019;4(9):1508–15; **5.** Ministério da Saúde – Brasil. 26 estados brasileiros registram queda ou estabilidade na incidência de dengue, Available at: https://www.gov.br/saude/pt-br/assuntos/noticias/2024/maio/26-estados-brasileiros-registram-queda-ou-estabilidade-na-incidencia-de-dengue. Acessed in September 2024; **6.** Tricou V, Yu D, Reynales H, et al. Long-term efficacy and safety of a tetravalent dengue vaccine (TAK-003): 4·5-year results from a phase 3, randomised, double-blind, placebo-controlled trial. Lancet Glob Health. 2024;12(2):e257-e270; **7.** Coudeville, Laurent et al. Potential impact of dengue vaccination: Insights from two large-scale phase III trials with a tetravalent dengue vaccine. Vaccine 34 50 (2016): 6426-6435; **8.** Walker DG, Hutubessy R, Beutels P. WHO Guide for standardisation of economic evaluations of immunization programmes. Vaccine. março de 2010;28(11):2356–9; **9.** Junior JBS, Massad E, Lobao-Neto A, et al. Epidemiology and costs of dengue in Brazil: a systematic literature review. Int J Infect Dis IJID Off Publ Int Soc Infect Dis. 2022;122:521–8; **10.** Flasche S, Jit M, Rodríguez-Barraquer I, Coudeville L, Recker M, Koelle K, et al. The LongTerm Safety, Public Health Impact, and Cost-Effectiveness of Routine Vaccination with a Recombinant, Live-Attenuated Dengue Vaccine (Dengvaxia): A Model Comparison Study. PLoS Med. 2016;13(11):e1002181–e100218; **11.** Notificação S de I de A de. Portal SINAN 2023. Available at: https://portalsinan.saude.gov.br/. Acessed in September 2024; **12.** Coelho GE, Leal PL, Cerroni M de P, Simplicio ACR, Siqueira JBJ. Sensitivity of the Dengue Surveillance System in Brazil for Detecting Hospitalized Cases. PLoS Negl Trop Dis. 2016;10(5):e0004705–e0004705; **13.** (PAHO) PAHO. Health in the Americas 2023. Available at: https://hia.paho.org/en. Acessed in September 2024.**14.** Ministério da Saúde. O uso de liminares de custo-efetividade nas decisões em saúde: Recomendações da comissão nacional de incorporação de tecnologias no SUS. Available at: https://www.gov.br/conitec/pt-br/midias/consultas/relatorios/2022/20220620_relatorio_oficina_limiares_2022.pdf. Acessed in November 2024.

Aknowledgments and Disclosures: BS, JS, PT, JL and RH are Takeda employees. AT and JZ are employees of Putnam. EK is an external consultant hired by Putnam.
Funding statement: This study was funded by Takeda Pharmaceuticals International AG and Takeda Farmacêutica Brasil. The model was developed in partnership with Putnam and funded by Takeda Pharmaceuticals International AG.