RWD122

Understanding the burden of sleep disturbances and vasomotor symptoms on work productivity and healthcare resource utilization among women experiencing menopause in the EU: a National Health and Wellness study

Nils Schoof[1], Joehl Nguyen[2], Kushal Modi[3], Sheila Drakeley[3], Lin Yang[3], Victoria Banks[4*], Kelly Genga[5], Kristina Rosa Bolling[2], Carsten Möller[1], Andromachi Giannopoulou[6], Carina Dinkel-Keuthage[1]

1. Bayer, Berlin, Germany; 2. Bayer, Whippany, NJ, USA; 3. Oracle Life Sciences, Austin, TX, USA; 4. Bayer, Reading, UK; 5. Bayer, Sao Paulo, Brazil; Bayer, Basel, Switzerland *At the time of the study.

INTRODUCTION

Sleep disturbances (SDB) and vasomotor symptoms (VMS), also referred to as hot flashes or night sweats (hot flashes during night), are common in menopausal women and pose significant economic and resource challenges. These issues can lead to substantial declines in workplace productivity and higher healthcare expenses. Additionally, SDB and VMS can negatively impact a woman's mood and overall well-being. **This study assessed the impact of sleep disturbances and vasomotor symptoms on work productivity and activity impairment (WPAI) and healthcare resource use (HCRU) in postmenopausal women with VMS compared to those without VMS, in the EU and UK.**

METHODS

Data were analyzed from postmenopausal women aged 40–65 who participated in the internet-based EU (including France, Germany, Italy, and Spain) and UK National Health & Wellness Survey in either 2017 or 2020 (which were the most recent years with data on sleep-related patient-reported outcomes). Women were classified as postmenopausal if their menstrual bleeding had stopped for more than 12 months. They were then categorized based on the presence or absence of VMS, defined as reporting hot flashes or night sweats in the past year, and subsequently assessed for sleep disturbances. Those with SDB were identified as having self-reported sleep issues in the last year or experiencing symptoms at least weekly, such as trouble falling asleep, night-time awakenings, and poor sleep quality, which may be associated with menopause. WPAI scores were evaluated only for employed respondents, focusing on absenteeism, presenteeism, and overall work productivity; activity impairment scores included all respondents. **WPAI scores are shown in Figure 2 as percentages, with higher scores indicating greater impairment. Healthcare resource use (HCRU) outcomes comprised self-reported healthcare provider visits, emergency room visits, and hospitalizations in the past six months.** Multivariable analyses examined the relationship between VMS, sleep disturbances, and WPAI and HCRU outcomes.

RESULTS

Figure 1. Prevalence of VMS and sleep disturbances

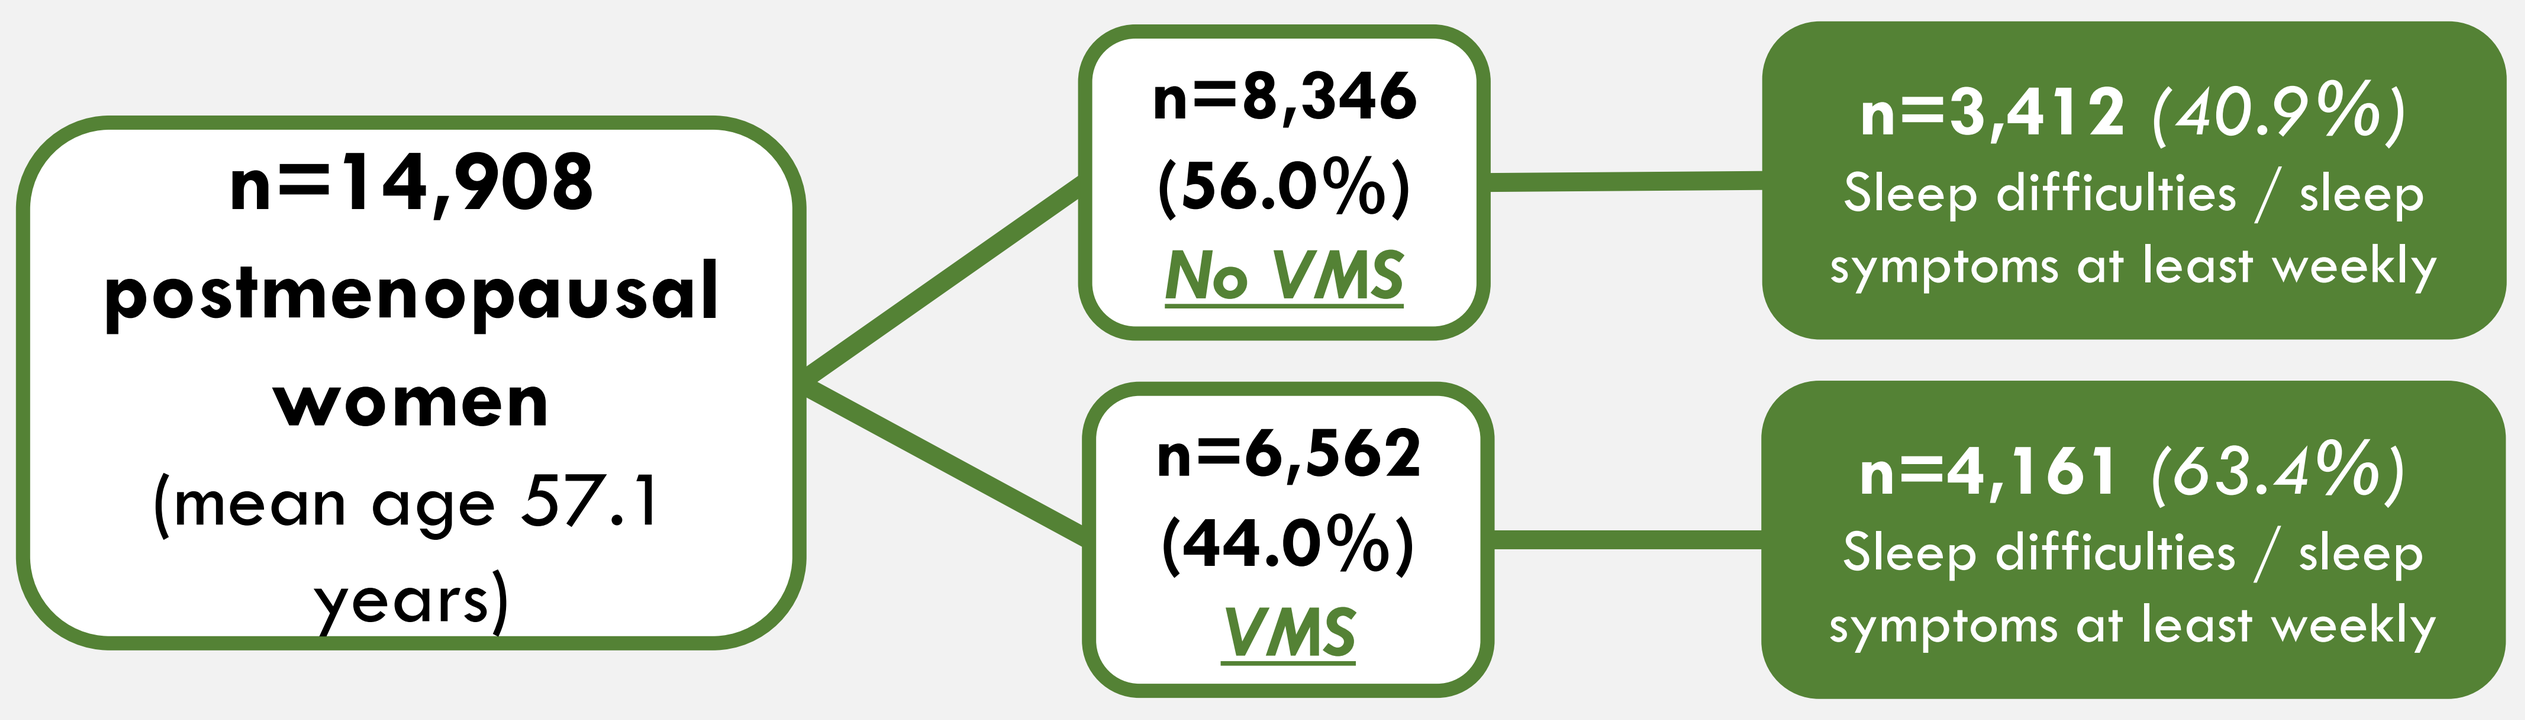
n=14,908
postmenopausal
women
(mean age 57.1
years)
n=8,346
(56.0%)
No VMS
n=6,562
(44.0%)
VMS
n=3,412 (40.9%)
Sleep difficulties / sleep
symptoms at least weekly
n=4,161 (63.4%)
Sleep difficulties / sleep
symptoms at least weekly

Baseline Characteristics		SDB and VMS		No SDB, but with VMS		SDB, but no VMS		No SDB or VMS	
Age, Mean, SD		55.8 (5.3)		55.9 (5.3)		58.2 (5.9)		57.9 (6.0)	
Employed (yes), n, %		2212	53.2%	1434	59.7%	1615	47.3%	2619	53.1%
Region, n, %	**UK**	1246	29.9%	478	19.9%	917	26.9%	959	19.4%
	Spain	365	8.8%	342	14.2%	271	7.9%	534	10.8%
	Italy	480	11.5%	496	20.7%	279	8.2%	680	13.8%
	Germany	1111	26.7%	589	24.5%	811	23.8%	1276	25.9%
	France	959	23.0%	496	20.7%	1134	33.2%	1485	30.1%
Insurance (yes) n, %		3812	91.6%	2191	91.3%	3134	91.9%	4581	92.8%
CCI, Mean, SD		.54	.99	.32	.77	.48	.93	.26	.66

Among 14,908 postmenopausal women (mean age: 57.1, SD: 5.8), 44.0% self-reported VMS and 50.8% self-reported SDB; 27.9% experienced both VMS and SDB. 52.9% were employed.

In Figure 2, employed women experiencing both VMS and SDB showed the highest absenteeism and presenteeism compared to those experiencing neither (Estimated Marginal Means (EMM): 6.6% vs. 4.9%, p=0.026, 17.4% vs. 12.9%, p<0.001, respectively). Similarly, work productivity (among employed women) and activity impairment (among all women) was highest among the VMS with SDB group compared to those experiencing neither (EMM: 18.8% vs.14.4%, p<0.001, 28.1% vs. 20.9%, p<0.001, respectively).

Figure 3 depicts the odds ratios (OR) for HCRU use of the cohorts compared to the reference group of No VMS or SDB. The odds of an office-based HCP visits was highest among women experiencing sleep-related symptoms, with or without VMS (OR 1.62 and 1.67, p<0.001) compared to those experiencing neither.

Figure 2. Marginal mean estimates for WPAI scales: Absenteeism, Presenteeism, Work Productivity, and Activity Impairment
Adjusted for confounder variables

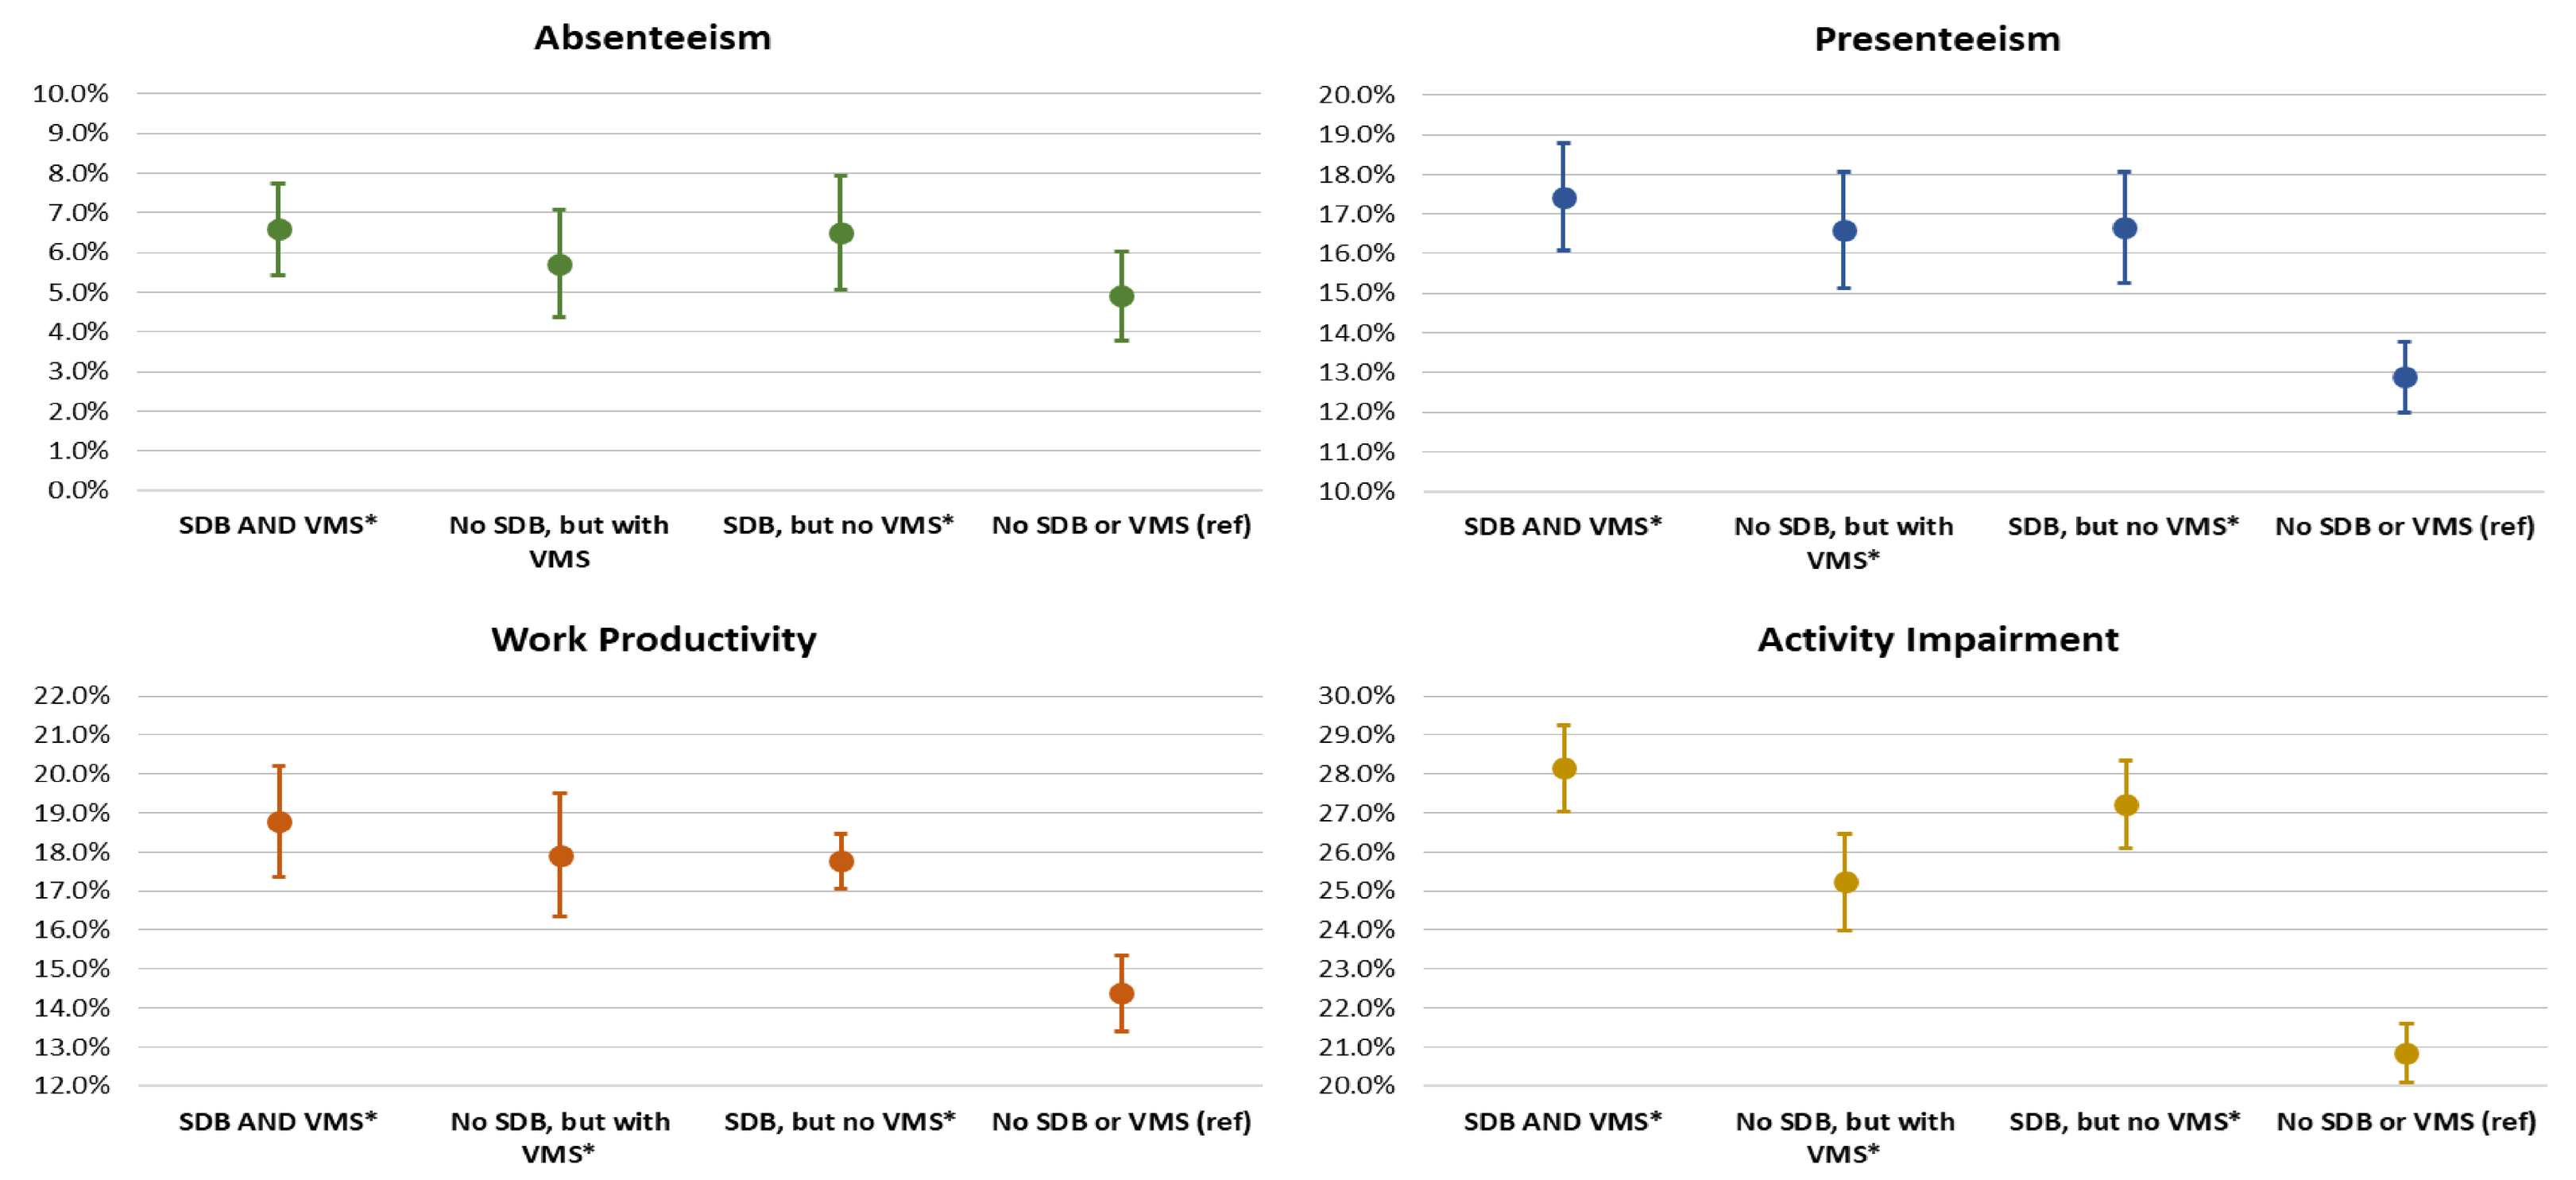
Absenteeism
Presenteeism
Work Productivity
Activity Impairment
SDB AND VMS*
No SDB, but with VMS
No SDB, but with VMS*
SDB, but no VMS*
No SDB or VMS (ref)

*** Indicates p < 0.05 when compared to No SDB or VMS (reference group)**

Figure 3. Odds Ratio of a HCP visit, ER visit, or hospitalization in the past 6 months compared to those with neither SDB or VMS
Adjusted for confounder variables

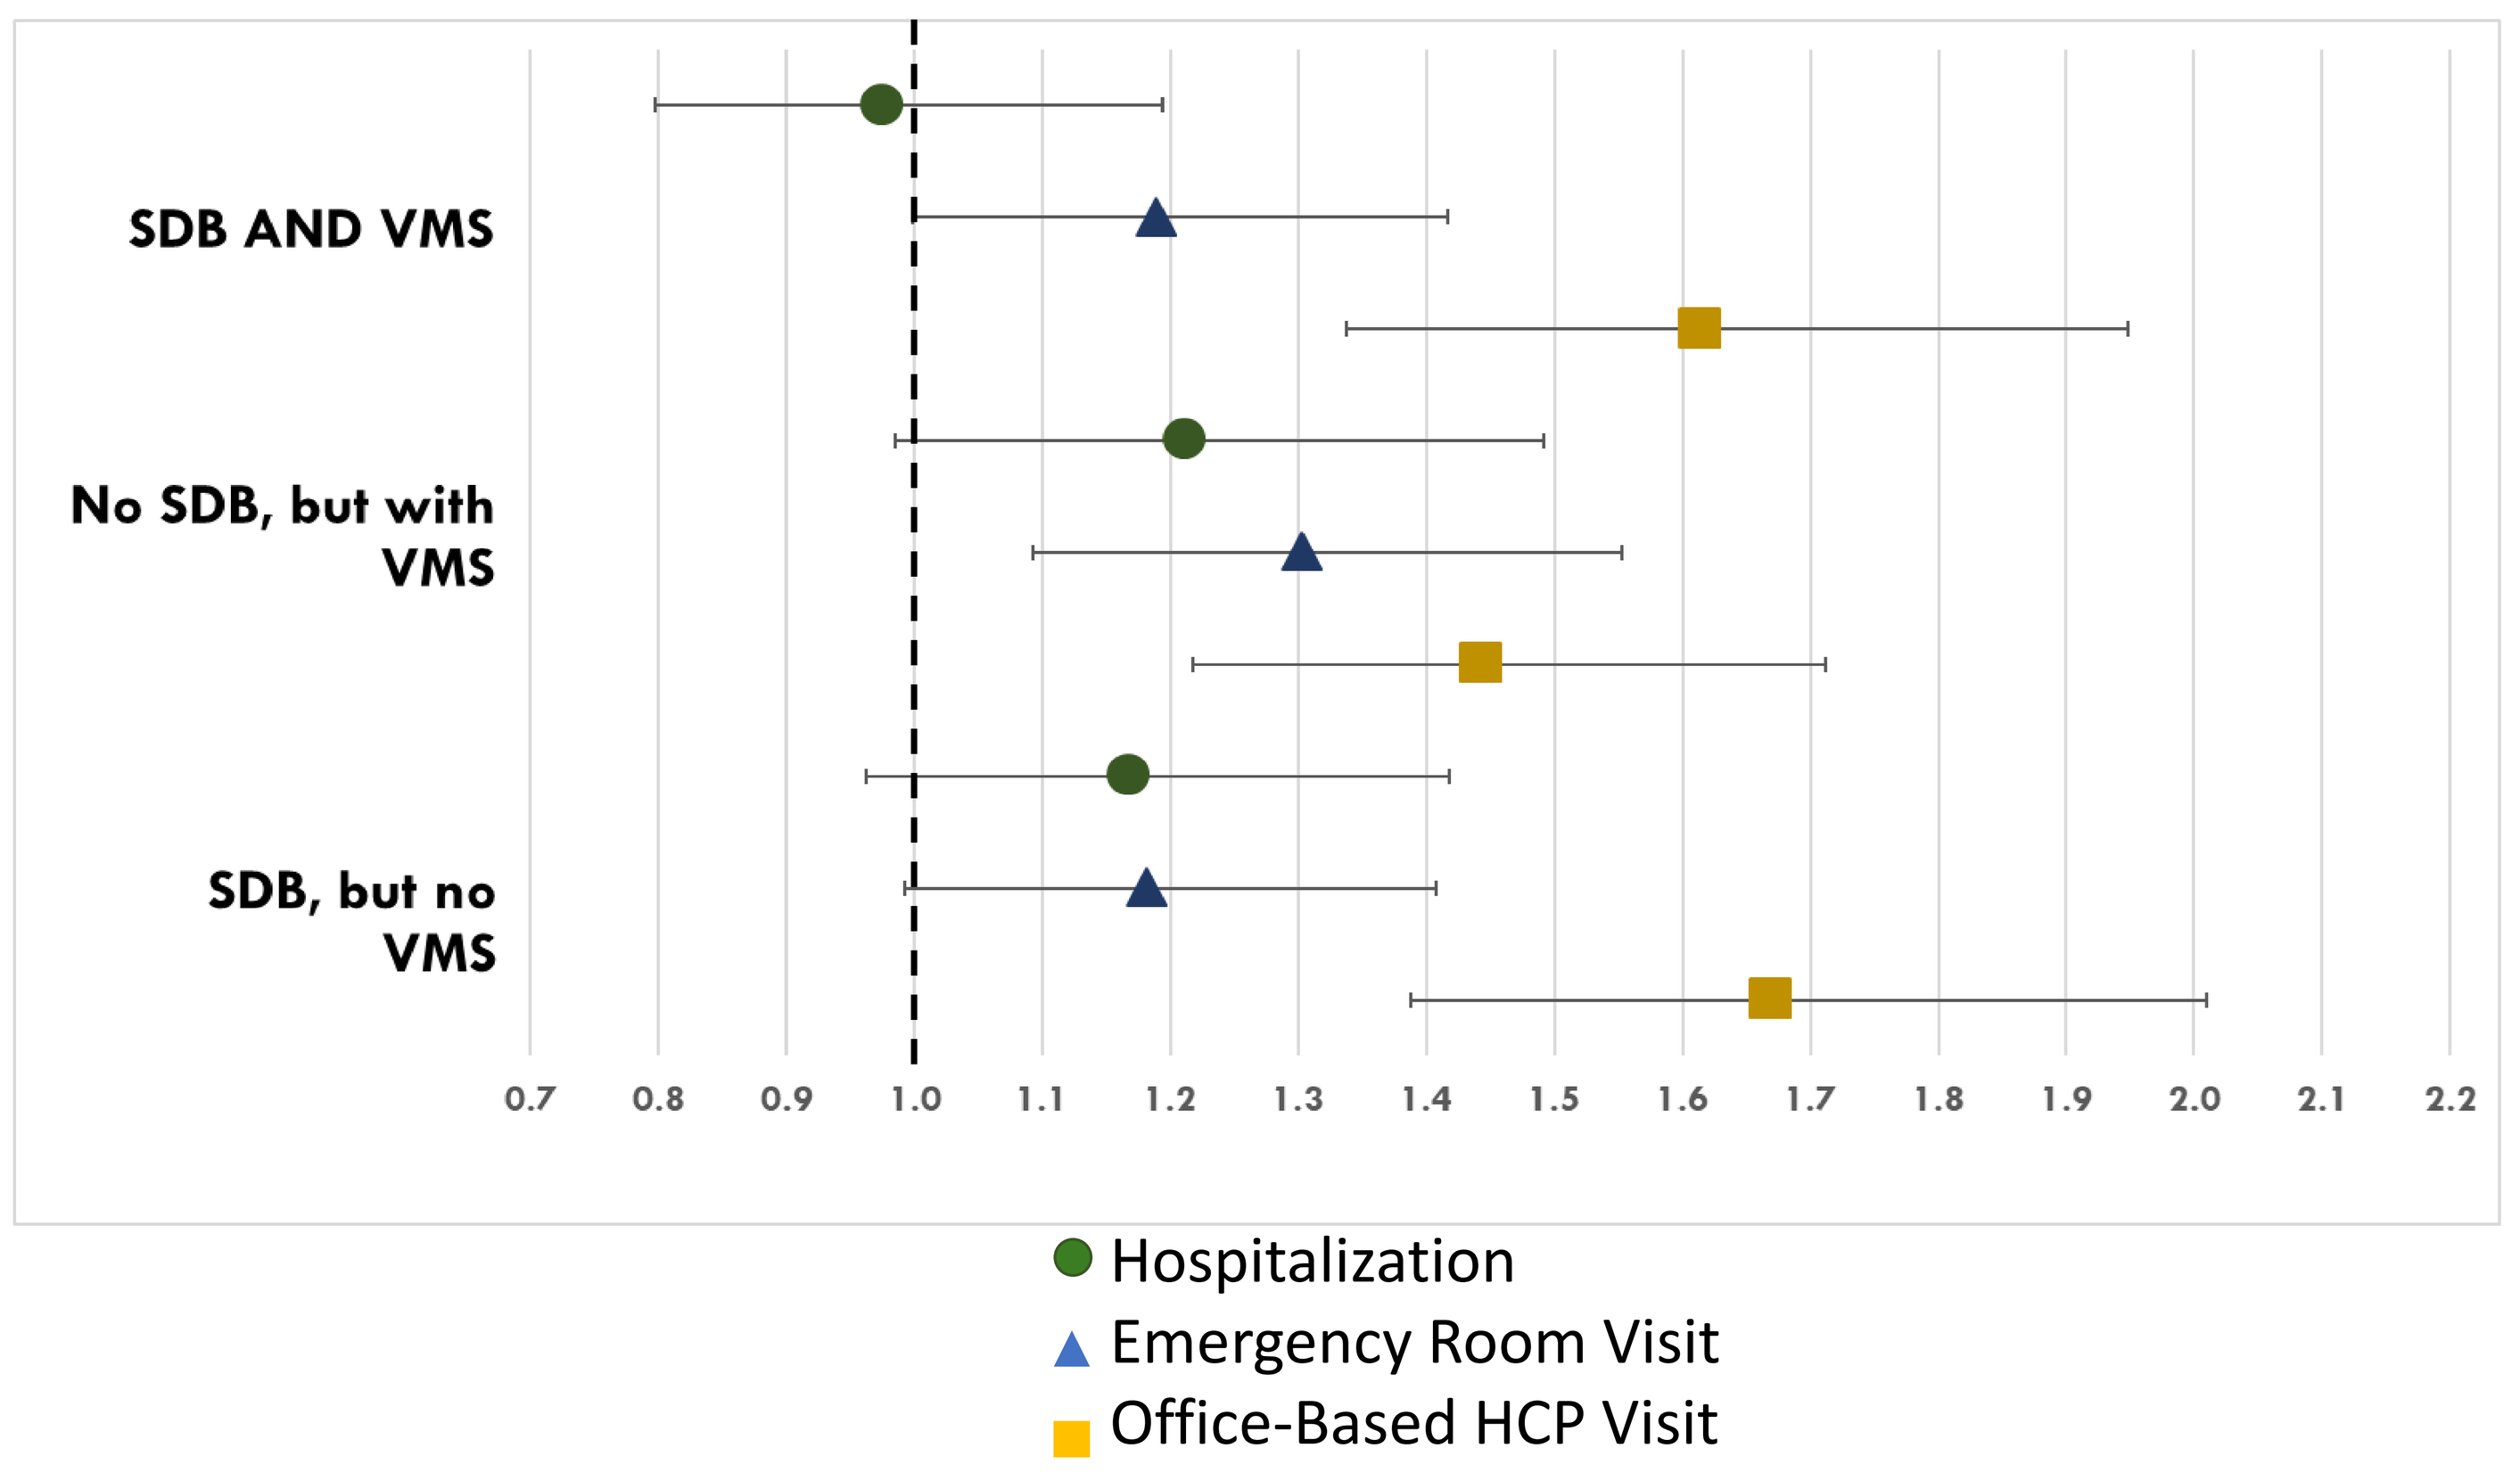
SDB AND VMS
No SDB, but with VMS
SDB, but no VMS
0.7 0.8 0.9 1.0 1.1 1.2 1.3 1.4 1.5 1.6 1.7 1.8 1.9 2.0 2.1 2.2
Hospitalization
Emergency Room Visit
Office-Based HCP Visit

CONCLUSIONS

The prevalence of sleep conditions, including menopause-related sleep disturbances, was high overall, regardless of the presence or absence of VMS. Sleep disturbances were a large driver in WPAI and HCRU burden among women experiencing menopause, independently of VMS.

Among postmenopausal women, **presenteeism, absenteeism and activity impairment were highest among postmenopausal women with sleep disturbances than those without.**

Independent of the presence of VMS, **the odds of an office-based HCP visit was highest among postmenopausal women experiencing sleep-related symptoms** compared to those experiencing neither.

Addressing sleep disturbances in menopausal women is vital **for reducing economic strain and resource burden while promoting overall health and well-being.**

DISCLOSURES

This study was funded by Bayer AG, Berlin, Germany. NS, JN, KG, KRB, CM, AG, and CDK are employees of Bayer. VB and LH are former employees of Bayer. KM, LY, and SD are employees of Oracle Life Sciences.

REFERENCES

1. Moorhead, S., et al. (2021). The Impact of Menopausal Symptoms on Work Productivity and Healthcare Utilization. Menopause, 28(6), 731-739; **2.** Freeman, E. W., et al. (2017). Vasomotor Symptoms and Sleep Disturbances: The Interplay and Impact on Quality of Life. Journal of Women's Health, 26(7), 749-757;

ACKNOWLEDGEMENTS

The authors would like to acknowledge Oracle Life Sciences (Austin, Texas, USA) for their assistance in processing and analyzing the data. Medical writing support was provided by Oracle Life Sciences, funded by Bayer.