Cost-effectiveness of mRNA COVID-19 vaccination (mRNA-1273) in Preventing COVID-19 Infections, Hospitalizations, and Deaths Among Older Adults in Italy: A Static Health Economic Model

EE573

F. RUMI[1], M. BASILE[1], K. JOSHI[2], E. BECK[2], S. SCHOLZ[2], E. DI BRINO[1] and A. PONZIANELLI[3]
1 Altems Advisory, spin-off of the Università Cattolica del Sacro Cuore, Rome, Italy; 2 Moderna, Inc., Cambridge, MA, USA; 3 Moderna Italy, Rome, Italy.

BACKGROUND

- The COVID-19 pandemic has had a profound economic impact on Italy, severely straining both the healthcare system and the broader economy. In the early stages, healthcare costs soared due to the high demand for intensive and prolonged hospitalizations, compounded by productivity losses from illness and quarantine measures.
- The introduction of vaccination alleviated some of these costs by reducing the severity of cases and decreasing hospitalization rates. However, managing COVID-19 and addressing its long-term effects continue to present a significant financial burden. In the post-pandemic phase, cost-effectiveness analysis plays a pivotal role in healthcare decision-making, aiming to optimize resource allocation while maximizing health outcomes at reduced costs.
- This is especially relevant as the disease burden remains substantial in 2023/2024, compounded by low vaccine uptake. Real-world evidence also suggests that among mRNA-vaccines, mRNA-1273 may be more effective in protecting vulnerable populations including older adults, underscoring its value in terms of cost-effectiveness.

OBJECTIVE

- This study evaluates the cost-effectiveness of an mRNA-1273 vaccination campaign for the Fall 2023/2024 season, comparing it to both no vaccination and a campaign using the mRNA BNT162b2 vaccine. The aim is to provide valuable insights into the economic and health benefits of these vaccination strategies, supporting informed public health decision-making. Specifically, the study examines:
- The effectiveness of mRNA-1273 in reducing infections, hospitalizations, long COVID cases, and deaths among Italian adults aged ≥60, compared to no vaccination.
- A comparison of the cost-effectiveness and public health impact between Moderna's mRNA-1273 vaccine and Pfizer-BioNTech's BNT162b2 vaccine.

METHODS

- A decision-analytic model assessed COVID-19 outcomes in Italians aged 60+ by comparing vaccination (mRNA-1273 and BNT162b2) with non-vaccination. The model, using a static decision tree, evaluated infections, hospitalizations, deaths, long COVID cases, and QALY losses over October 2023–September 2024.
- mRNA-1273, specifically targeting Omicron XBB.1.5, showed higher effectiveness in preventing hospitalizations and infections than BNT162b2, especially in adults aged 65+ (Kavikondala, 2024).
- According to Kopel et al. (2024), mRNA-1273 showed an absolute VE of 60.5% (95% CI: 53.3%-66.6%) against hospitalizations and 38.7% (95% CI: 35.4%-41.9%) against any medically attended COVID-19.
- Additionally, a relative risk (RR) analysis for mRNA1273 vs Pfizer-BioNTech by Kavikondala (2024) estimated an RR of 0.74 (95% CI: 0.62-0.88) for infection and 0.69 (95% CI: 0.53-0.89) for hospitalization among adults aged 65+. Considering RRs of Kavikondala and VE of Kopel et al for mRNA-1273, VE for BNT126b2 was estimated at 42.75% against hospitalizations and 17.16% for any medically attended COVID-19.
- mRNA-1273 Total Cost: €74.15, including acquisition and administration, with no wastage due to its pre-filled syringe format.
- BNT162b2 Total Cost: €75.98 assumed, including acquisition, administration, and an assumed 16.67% wastage rate, reflecting the challenges of multi-dose vials, as supported by literature on MDV wastage (Pessoa-Gonçalves et al., 2023; Shinozuka et al., 2021; Lazarus et al., 2022).
- Cost Assessment: COVID-19 treatment, management of long COVID and vaccine adverse effects.
- Utility Inputs: International studies provided QALY losses from COVID-19-related morbidity, post-infection effects, and vaccine side effects, ensuring quality of life impacts are accurately represented.

RESULTS

mRNA-1273 vs no vaccination

- The mRNA-1273 vaccine effectively reduced symptomatic COVID-19 cases, hospitalizations, deaths, and long COVID cases. Although vaccination costs were higher than no vaccination, the health benefits and reduced complication expenses justified the investment.
- The Incremental Cost-Effectiveness Ratio (ICER) was €18,689 per QALY, below Italy's WTP threshold of €35,000 per QALY, indicating high cost-effectiveness **(Figure 1)**.
- Even with a dose price of €107.00, the ICER stayed below the threshold (€34,966 per QALY), confirming cost-effectiveness.
- Vaccination notably reduced total QALYs lost, reinforcing its public health value.
- Sensitivity analysis highlighted hospitalization rates as the most uncertain parameter, but no ICER exceeded the WTP threshold **(Figure 2)**. Probabilistic sensitivity analysis showed most simulations were cost-effective, with incremental costs of €100-400 million and incremental QALYs of 5,000,000-25,000,000.
- The CEAC indicated around 98% of simulations were cost-effective at the €35,000 threshold, highlighting the vaccine's economic value in Italy's COVID-19 management **(Figure 3)**.

mRNA-1273 vs BNT162b2

- The mRNA-1273 vaccine outperformed BNT162b2 in reducing symptomatic COVID-19 infections, hospitalizations, and deaths. mRNA-1273 was associated with lower healthcare costs and fewer total QALYs lost compared to BNT162b2.
- To match mRNA-1273's clinical prevention outcomes, BNT162b2 would require approximately 15% higher vaccination coverage.
- mRNA-1273 is estimated to prevent an additional 44,020 infections, 7,628 hospitalizations, and 1,054 deaths over BNT162b2.
- This enhanced effectiveness translates to a value-based price premium of €45.99 for mRNA-1273 at a €35,000 per QALY willingness-to-pay threshold.

Figure 1. Probabilistic Sensitivity Analysis (PSA)*

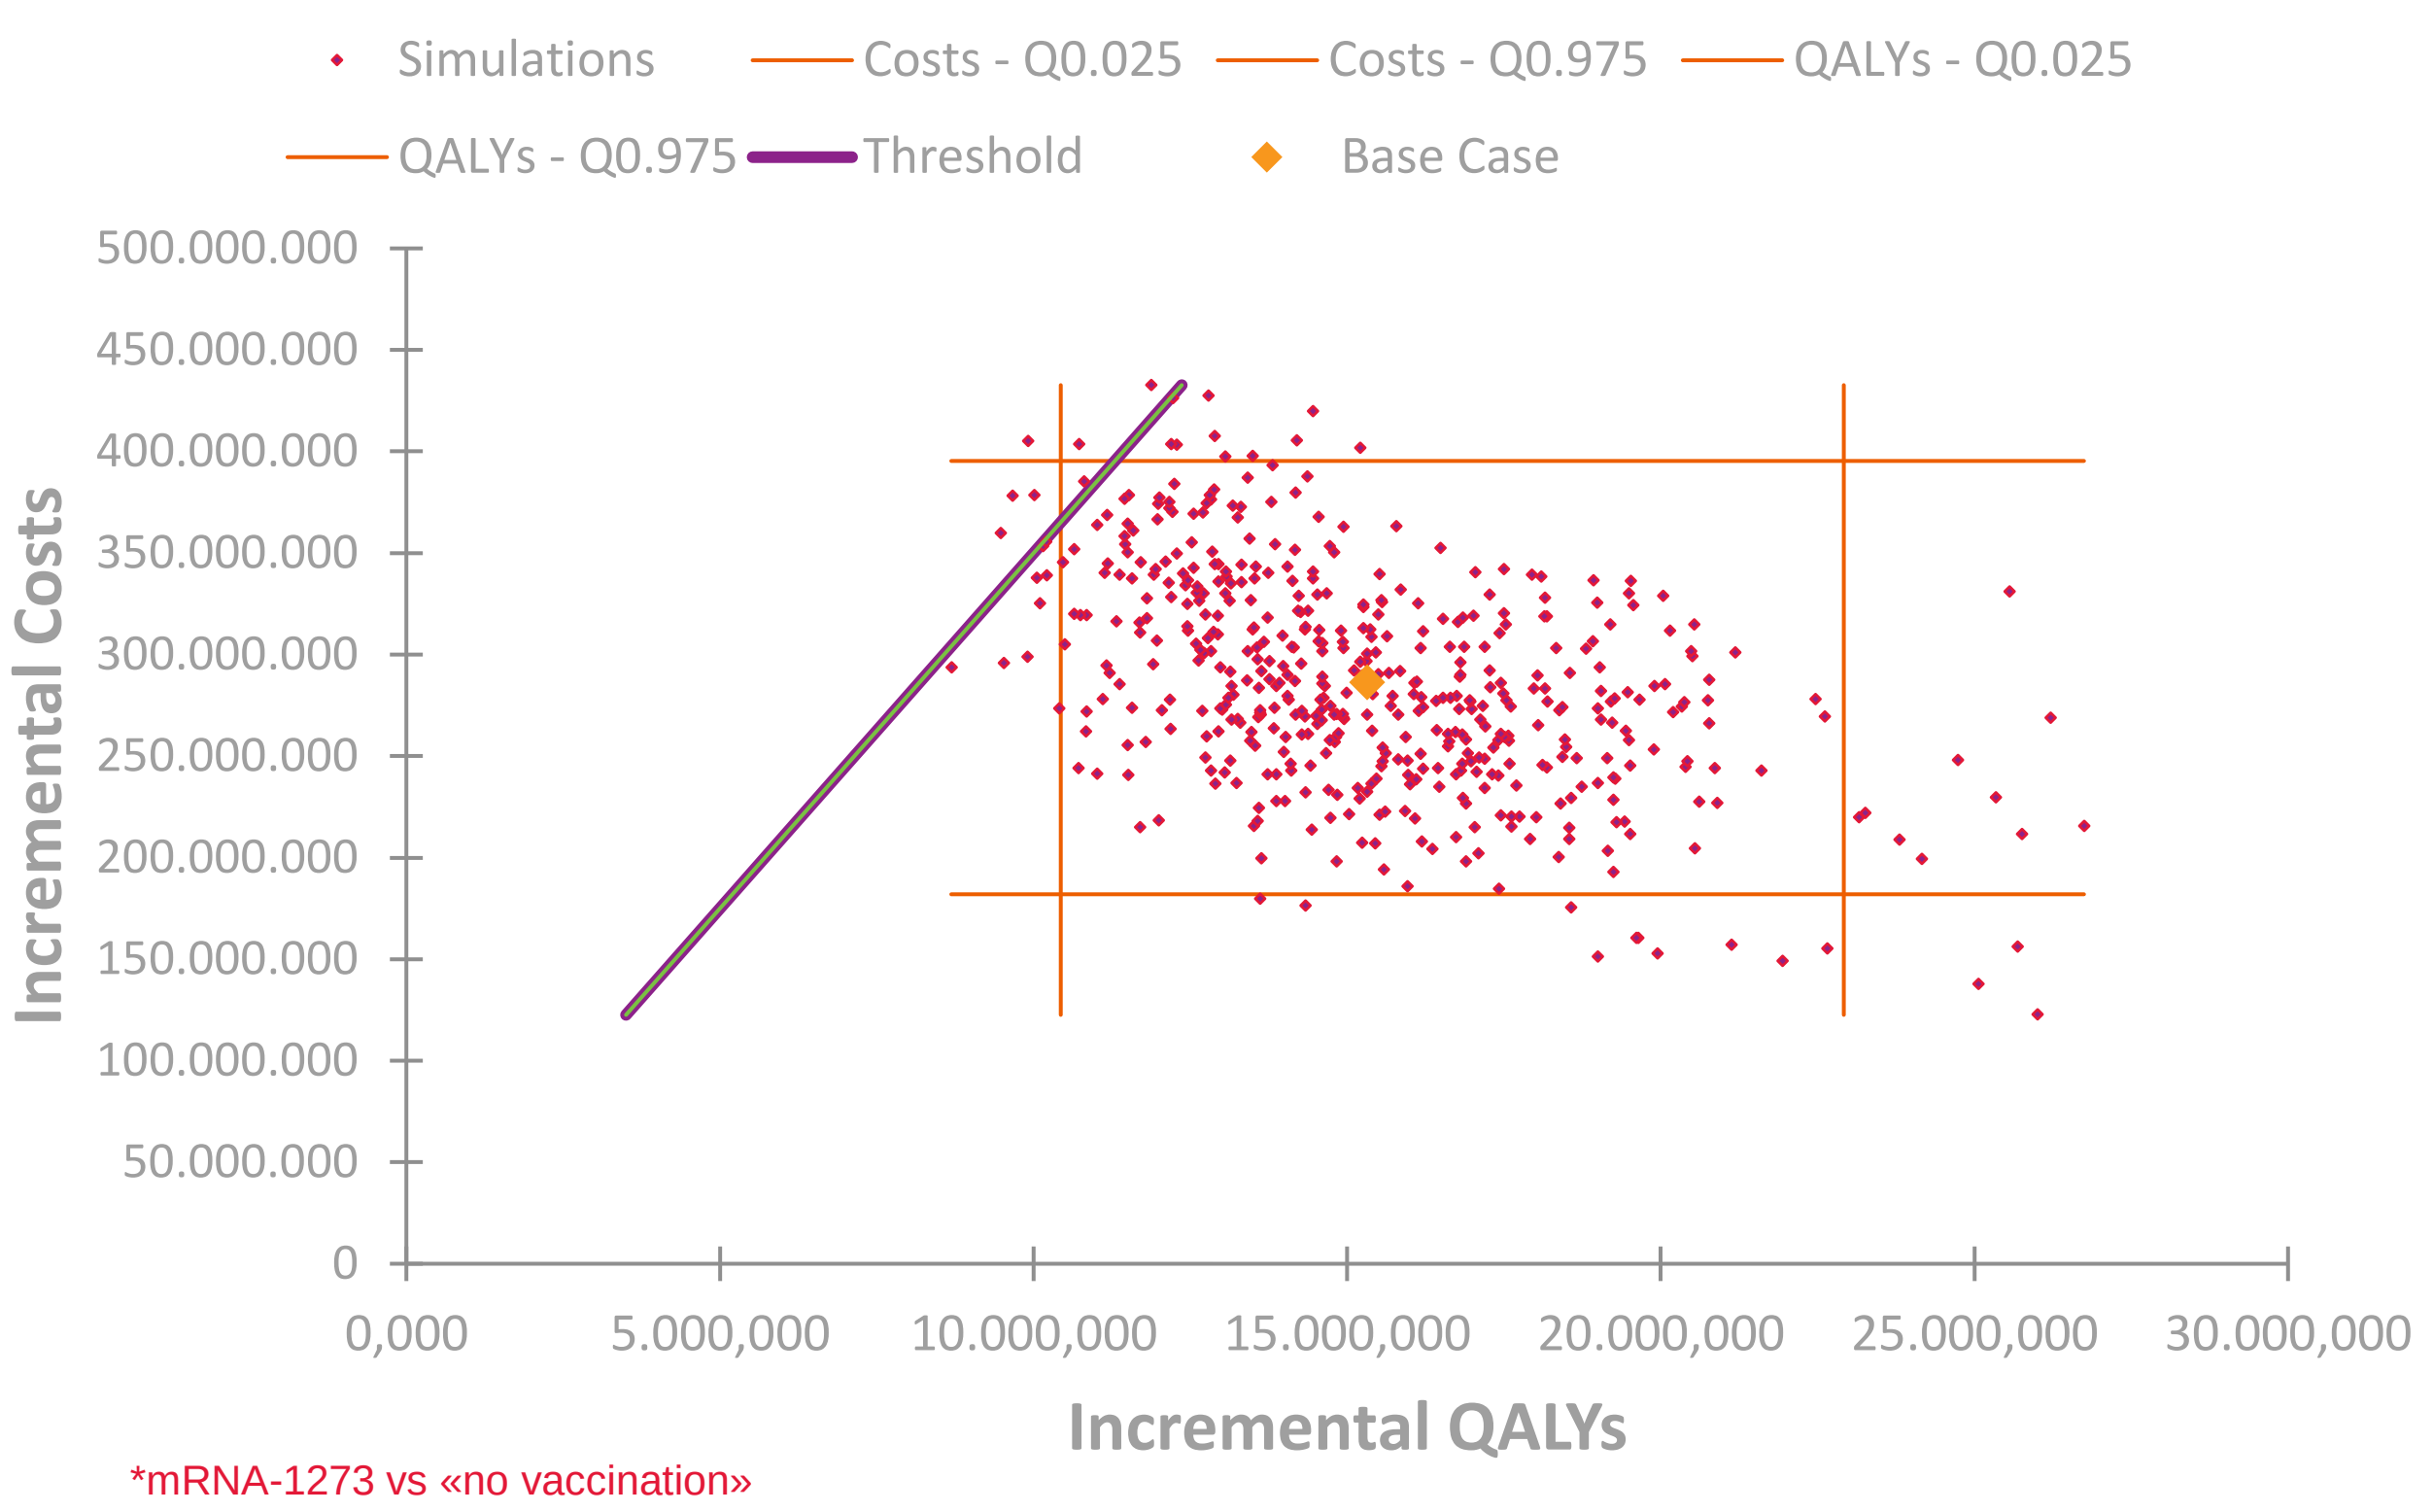
Simulations
Costs - Q0.025
Costs - Q0.975
QALYs - Q0.025
QALYs - Q0.975
Threshold
Base Case
Incremental Costs
500.000.000
450.000.000
400.000.000
350.000.000
300.000.000
250.000.000
200.000.000
150.000.000
100.000.000
50.000.000
0
0,000
5.000,000
10.000,000
15.000,000
20.000,000
25.000,000
30.000,000
Incremental QALYs

*mRNA-1273 vs «no vaccination»

Figure 2. Deterministic Sensitivity Analysis (DSA)*

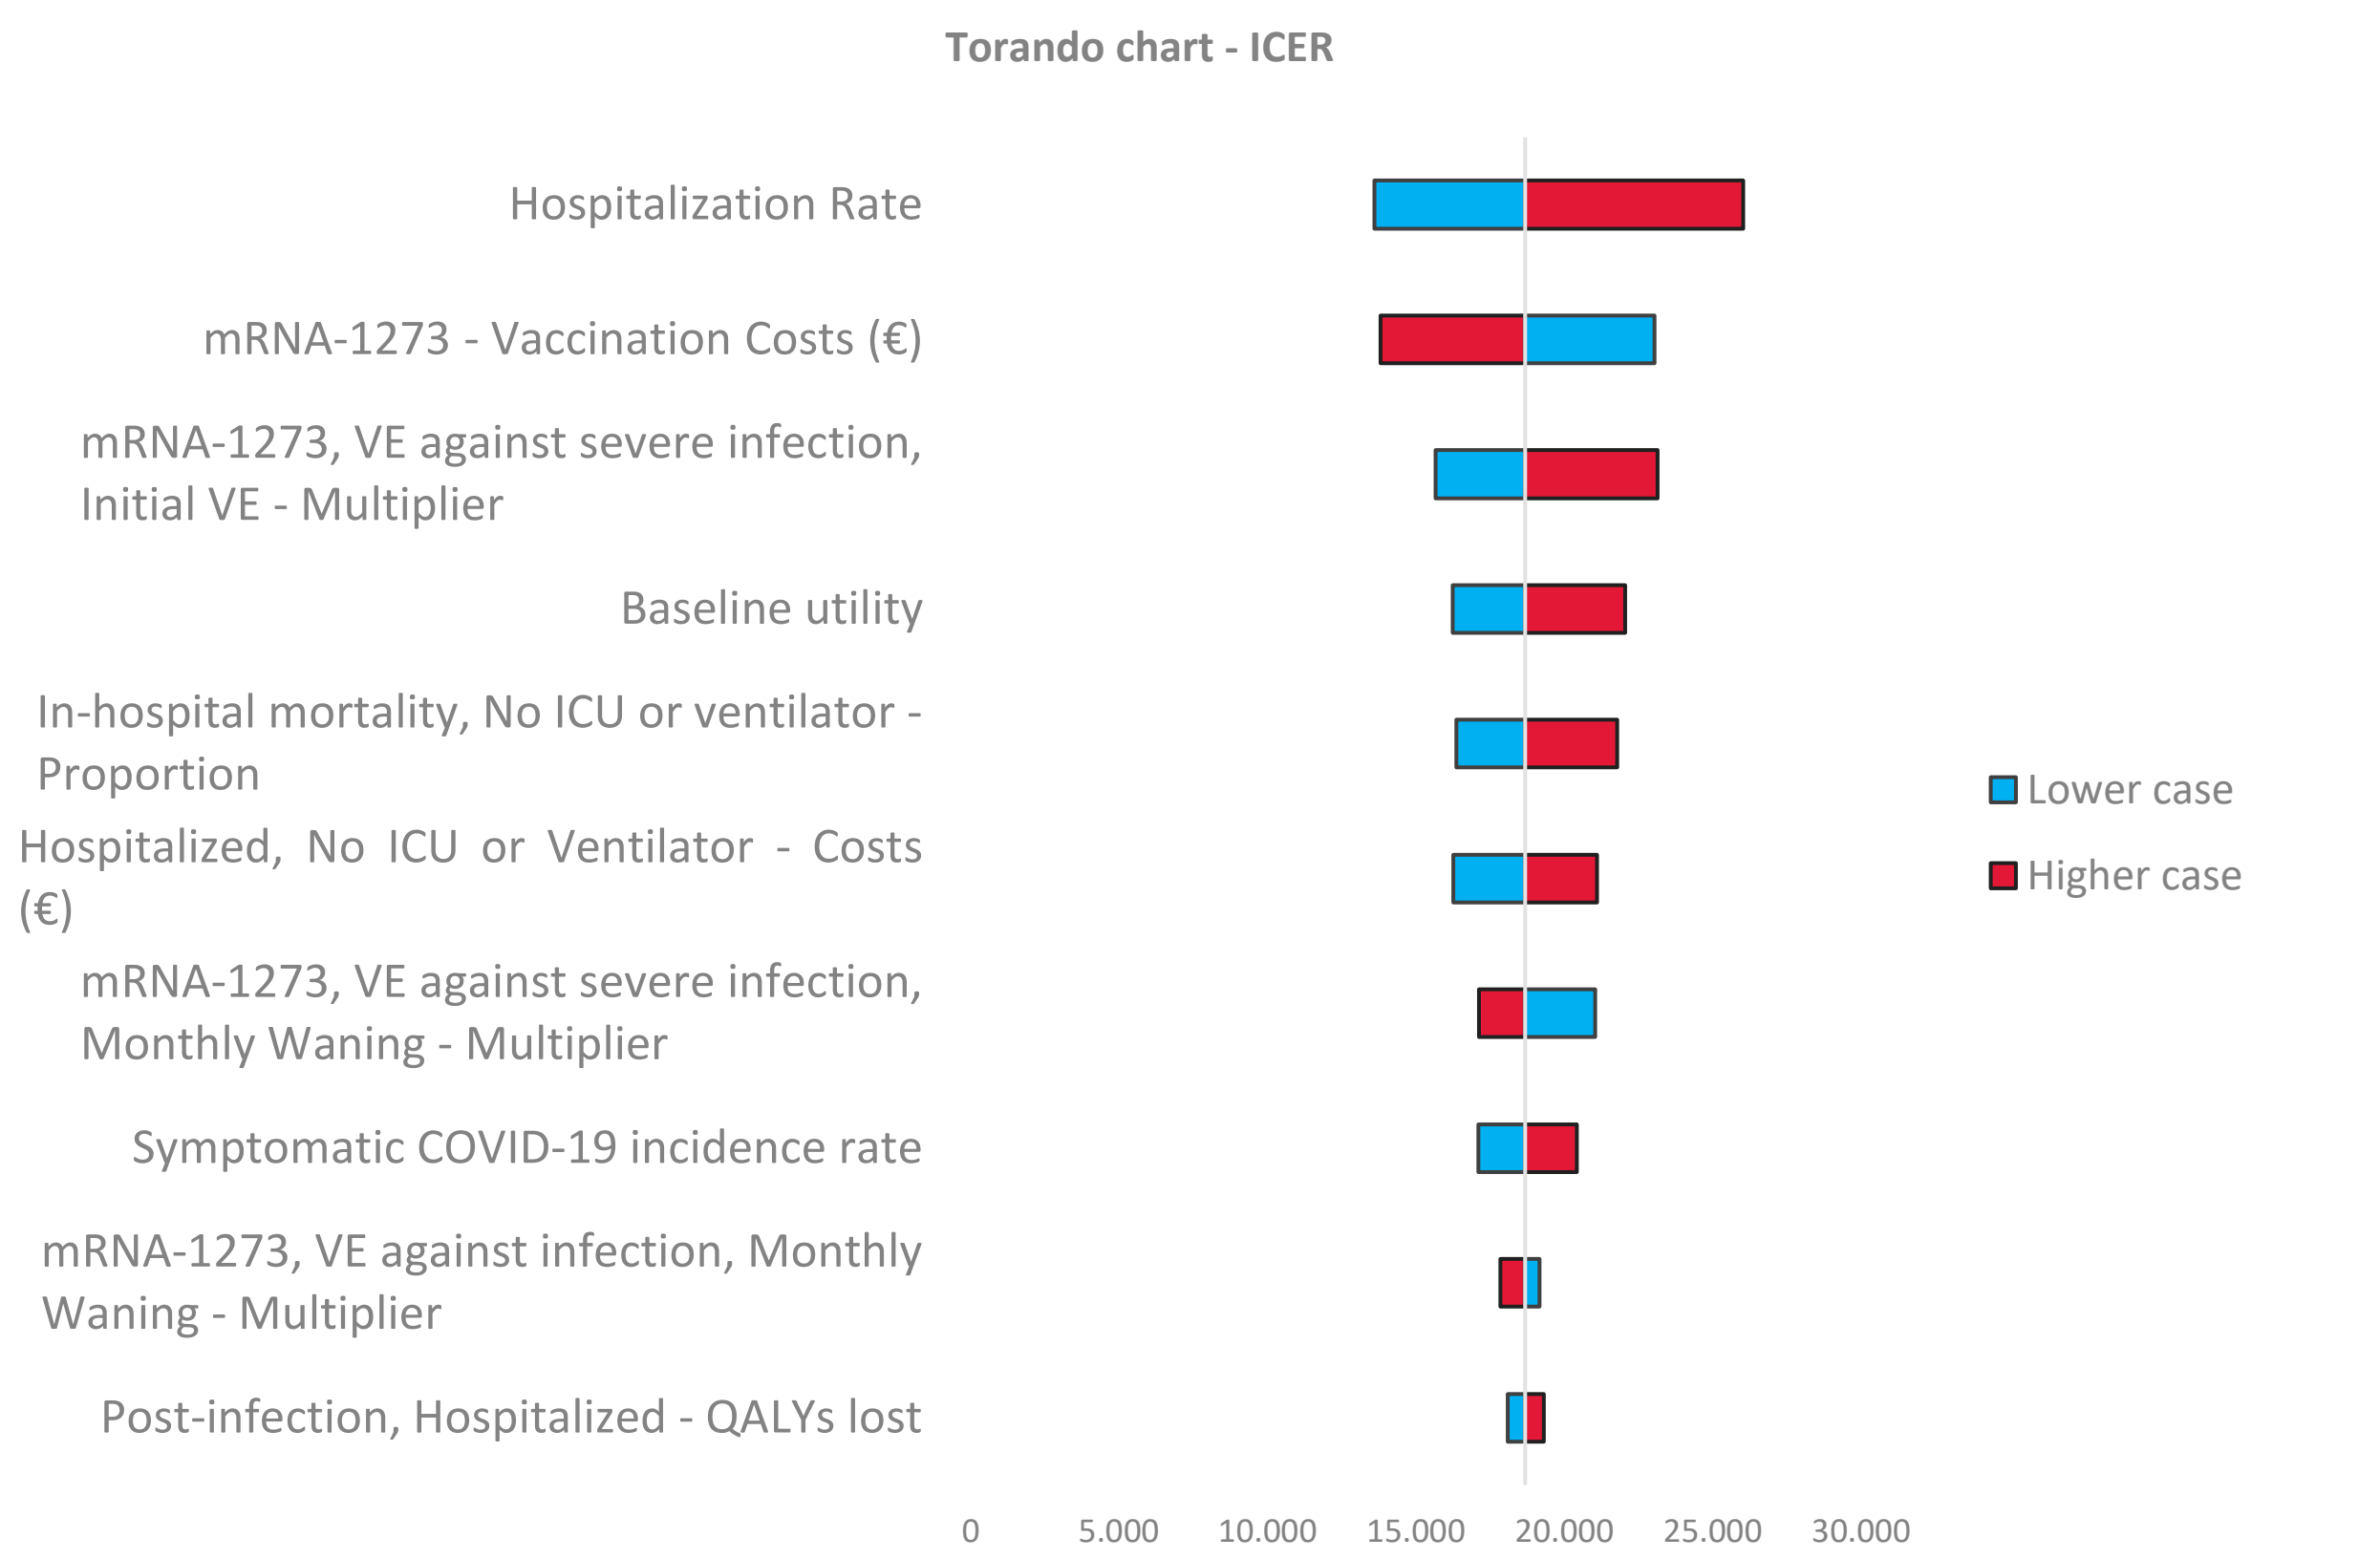
Torando chart - ICER
Hospitalization Rate
mRNA-1273 - Vaccination Costs (€)
mRNA-1273, VE against severe infection, Initial VE - Multiplier
Baseline utility
In-hospital mortality, No ICU or ventilator - Proportion
Hospitalized, No ICU or Ventilator - Costs (€)
mRNA-1273, VE against severe infection, Monthly Waning - Multiplier
Symptomatic COVID-19 incidence rate
mRNA-1273, VE against infection, Monthly Waning - Multiplier
Post-infection, Hospitalized - QALYs lost
0
5.000
10.000
15.000
20.000
25.000
30.000
Lower case
Higher case

*mRNA-1273 vs «no vaccination»

Figure 3. Cost-effectiveness acceptability curve (CEAC)*

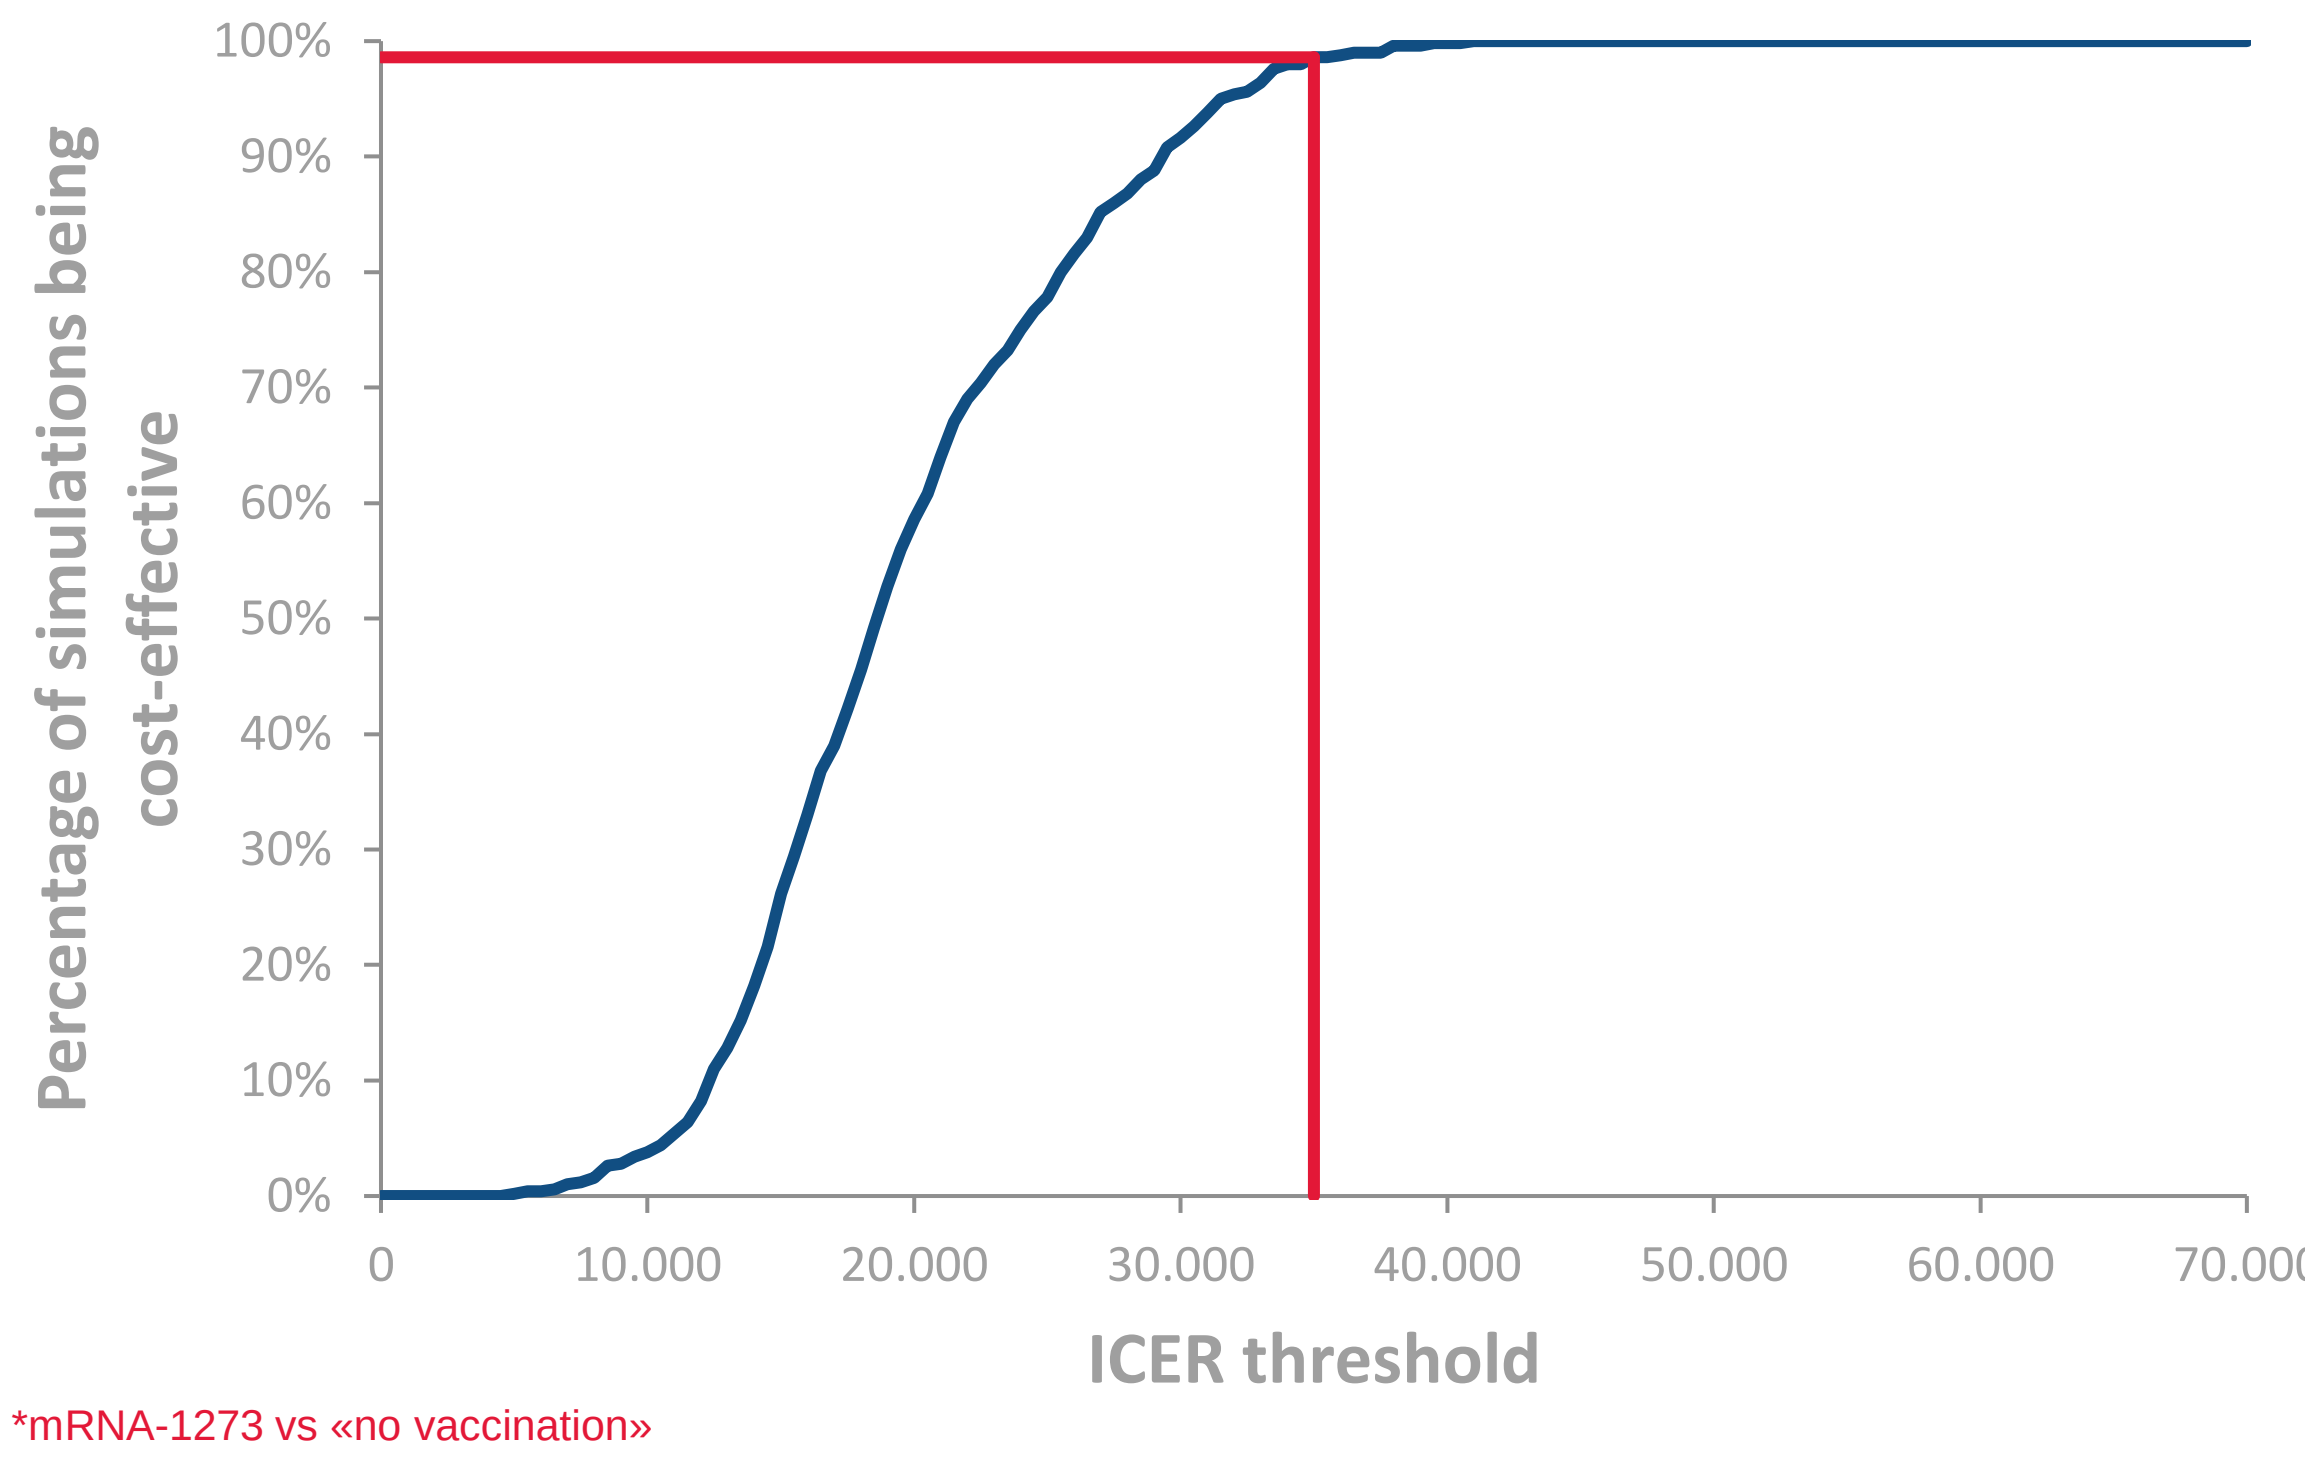
Percentage of simulations being cost-effective
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
0
10.000
20.000
30.000
40.000
50.000
60.000
70.000
ICER threshold

*mRNA-1273 vs «no vaccination»

CONCLUSIONS

- The study evaluates the cost-effectiveness of a potential Fall 2023/2024 vaccination campaign using mRNA-1273 compared to no vaccination and the BNT162b2 vaccine in the Italian population aged 60 and older. The findings indicate that mRNA-1273 significantly reduces symptomatic infections, hospitalizations, and deaths.
- The analysis showed that mRNA-1273 resulted in less cost and significantly more prevented COVID-19 cases, hospitalisations and deaths than BNT162b2. In conclusion, adopting mRNA-1273 strengthens Italy's public health response, offering higher clinical protection and sustainable costs.
- This highlights the importance of effective vaccination strategies to protect the elderly and optimize healthcare resources. Despite the analysis done for the 2023/2024 season, these findings may be helpful to inform implementation of 2024-2025 and future COVID-19 vaccination campaigns in Italy as based on these findings mRNA-1273 could substantially enhance COVID-19 prevention in adults aged 60 and older. A campaign using mRNA-1273 would not only be highly cost-effective but also provide better protection for older adults compared to BNT162b2, ensuring more sustainable healthcare policies and improved outcomes. However, limitations exist.
- The analysis relies on real-world vaccine effectiveness (VE) estimates, which may not fully capture variant-specific differences or the evolving pandemic. Additionally, healthcare costs and QALY estimates are uncertain, especially for long COVID. Sensitivity analyses confirm the robustness of findings but highlight the need for ongoing research to refine cost models and address these uncertainties.

References

1. Kavikondala S, Comparative Effectiveness of mRNA-1273 and BNT162b2 COVID-19 Vaccines Among Older Adults: Systematic Literature Review and Meta-Analysis Using the GRADE Framework. Infect Dis Ther. 2024 Apr;13(4):779-811.
2. Joshi, K, Clinical impact and cost-effectiveness of the updated COVID-19 mRNA Autumn 2023 vaccines in Germany. Journal of Medical Economics, 27(1), 39–50.
3. Kopel, Effectiveness of the 2023-2024 Omicron XBB.1.5-containing mRNA COVID-19 vaccine (mRNA-1273.815) in preventing COVID-19-related hospitalizations and medical encounters among adults in the United States: An interim analysis. medRxiv

Acknowledgments

We extend our gratitude to the individuals and institutions who contributed to this study. We are grateful to Moderna Europe and Italian teams, for their technical and analytical support throughout this research. This study was funded by Moderna Inc., whose support included assistance in study design and data analysis.

Disclosures

This study was funded by Moderna Inc. The funder had a role in the study design, analysis, interpretation of data, writing of the report, and the decision to submit the article for publication. KJ, EB, SS, and AP are employees of Moderna, Inc. and may hold stock/stock options in the company.

ABSTRACT PLAIN LANGUAGE SUMMARY

Please scan the QR code for an infographic plain language summary of the submitted abstract. Copies of this summary obtained through the QR code are for personal use only and may not be reproduced without written permission of the authors. For additional information, please contact Anna Ponzianelli (Anna.Ponzianelli@modernatx.com)

International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe; November 17-20, 2024; Barcelona, Spain