ADMINISTRATIVE FREQUENCY OF MIGRAINE AND CLINICAL CHARACTERISTICS IN A COLOMBIAN COHORT FROM 2018 TO 2022

EPH200

Rubio AC[*], Arciniegas J[*], Bolaños-López J[§], Gonzalez FJ, Mesa A[*], Bello C[§], Garcia M[*], Gomez D[§], Perez LE[§], Reyes Sanchez JM[*]

[*]Pfizer, Colombia. [§]Biociencias-Sura, Colombia

BACKGROUND

- Migraine is a disabling primary headache disorder[1] characterized by headaches and associated symptoms that can lead to considerable disruption of the professional and private lives of affected individuals. According to the GBD Study 2019, migraine alone was the second most common cause of disability and first among women under 50 years of age[2].
- Although migraine is a common disorder in urban communities in Latin America, the limited data on the prevalence of migraine in many countries are remarkable, possibly related to the lack of resources, underdiagnosis of migraine, and the limitations of study groups or epidemiologists with interest in this disease. Not all countries in Latin America have population-based epidemiological studies of migraine, and most of these studies were conducted many years ago, with few patients participating[3].
- Due to the limitations of population-based epidemiological studies of migraine and the emergence of novel medications, the development of research and analyses on local data on migraine are important and robust sources of information to support decision-making for health care stakeholders, researchers, and policymakers.

OBJECTIVE

- To estimate the frequency of migraine and measure its demographic and clinical characteristics in a Colombian cohort from electronic databases.

METHODS

- This non-interventional, retrospective, descriptive study was conducted in one Colombian Health Management Organization (HMO) from 2018 to 2022 with a 5-year follow-up period.
- Migraine patients were identified using the International Classification of Diseases, 10th version code G43, and the diagnosis was confirmed by a neurologist. The first recorded migraine diagnosis was defined as the index date.
- Medical records, claim databases and other electronic databases from the HMO were used to determine the clinical characteristics, treatments, and health care services.
- Administrative prevalence and incidence were estimated based on the number of cases and new cases each year from the con|sultation register in the database over the total number of insured patients in the HMO.

RESULTS

- A total of 89,227 patients were included in the study. Most of them were women (84.9%). The mean age of migraine diagnoses was 37.0 (Standard deviation 14.1). The most frequent cardiovascular disease in migraine patients was essential hypertension (21.3%). Essential hypertension was most frequent in men and occurred in 24.9% of patients with migraine. In people aged 55-74 years with migraine, essential hypertension was found in 61.8%, while in those aged ≥75 years, it was found in 88.3%. Ischemic heart disease and arrythmia were diagnosed in 4.2% and 4.1%, respectively, of the participants in the previous age group.
- Administrative prevalence of migraine started 1.7 (in 2018) and ended at 2.2 (in 2022) cases per 100 persons, showing an inverse U-shaped pattern, with the highest prevalence in 2020 (2.42, 95% Confidence interval (CI) 2.41 – 2.44 cases per 100 person).
- Administrative incidence presented a downward trend between 2019 and 2021 ranging between 0.9 and 0.032 cases per 100 patients, with a slight change in 2022, where it rose to 0.1.
- Incidence and prevalence were generally higher in the female and young adult age groups. Hypertension (21.3%), arrythmia (4.1%) and structural heart disease (3.4%) were the most common cardiovascular comorbidities, more frequent in male than female.

Figure 1. Administrative prevalence of migraine during the period of the study in Colombia discriminating by sex

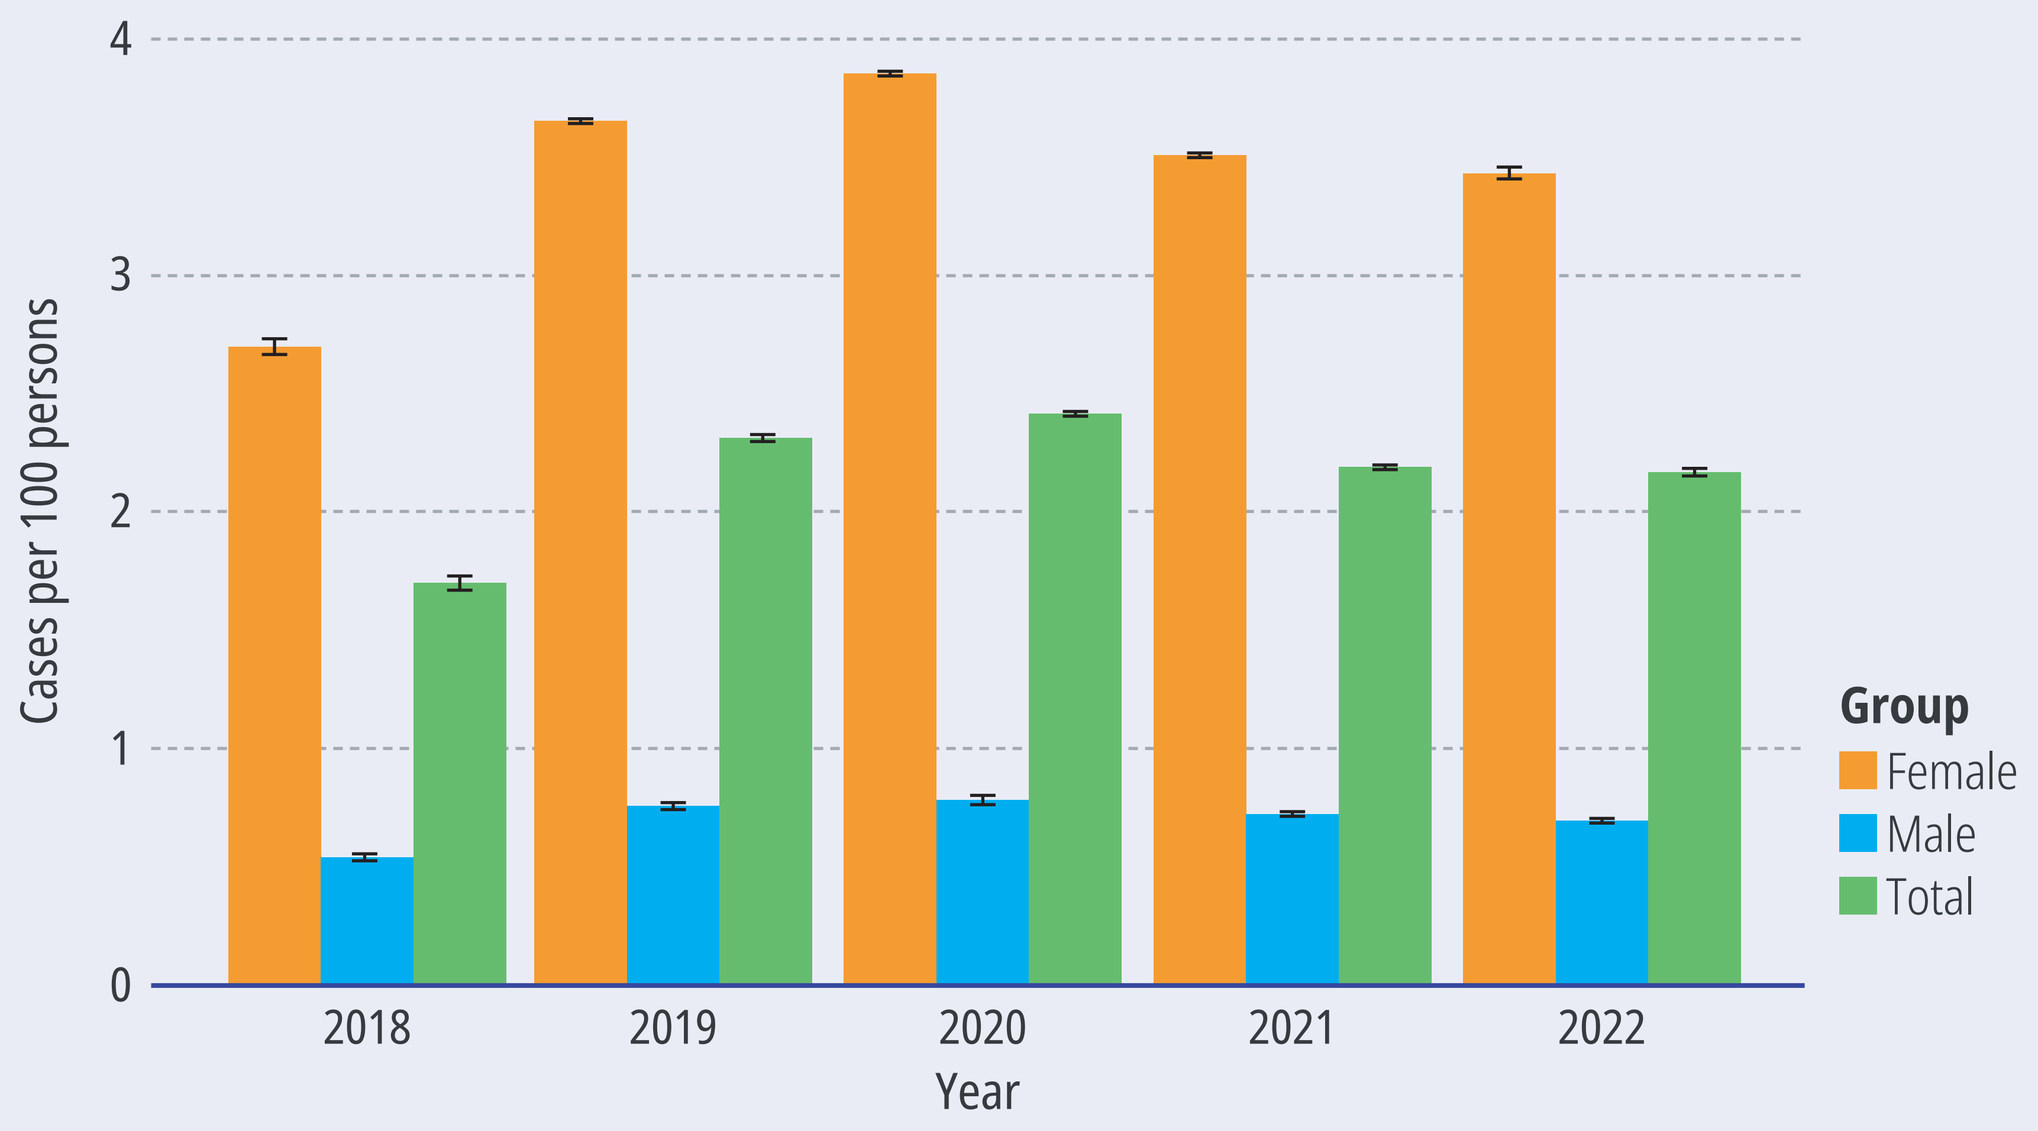
Cases per 100 persons
0
1
2
3
4
2018
2019
2020
2021
2022
Year
Group
Female
Male
Total

Figure 2. Administrative incidence of migraine during the period of the study in Colombia discriminating by sex

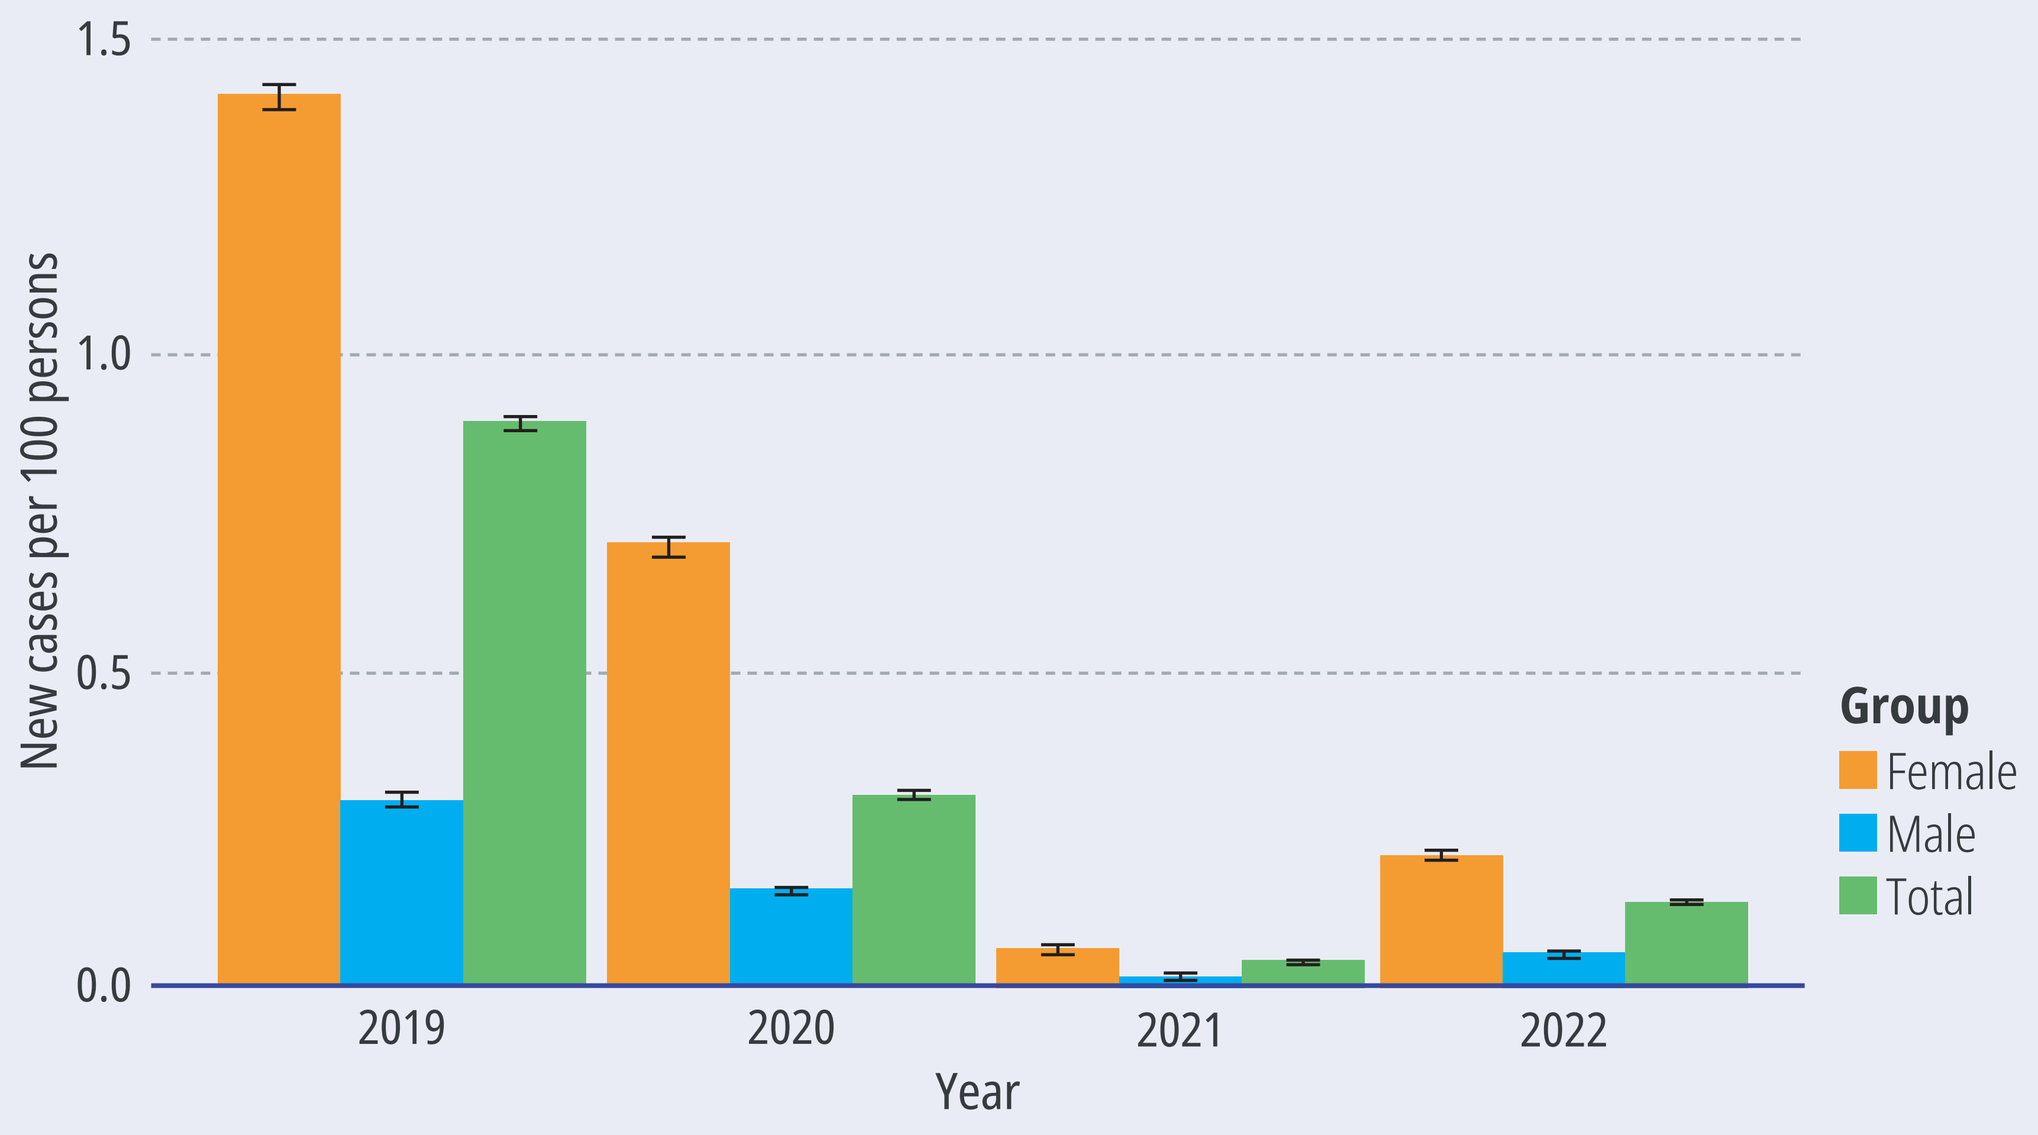
New cases per 100 persons
0.0
0.5
1.0
1.5
2019
2020
2021
2022
Year
Group
Female
Male
Total

- Across the 5-year study period, the mean follow-up period was 3.7 years (SD: 1.2), and within this timeframe, most patients were first seen by a general practitioner (82.6%, 73,657/89,227), and only 8.9% of patients (7,901/89,227) were first seen by a neurologist (first visit). Approximately 5.7% (5,053/89,227) of patients visited an emergency specialist. During the follow-up visits, for patients who continued to go to visits until the last observed visit, patients seen by a neurologist represented a greater share (11%, 458/4,274), and visits to emergency specialists decreased to 3.6% (155/4,274) (**Figure 3**)

Figure 3. Utilization of the health professional group in the initial diagnosis and follow-up. A) all migraine population; B) percentage of specialists according to health care.

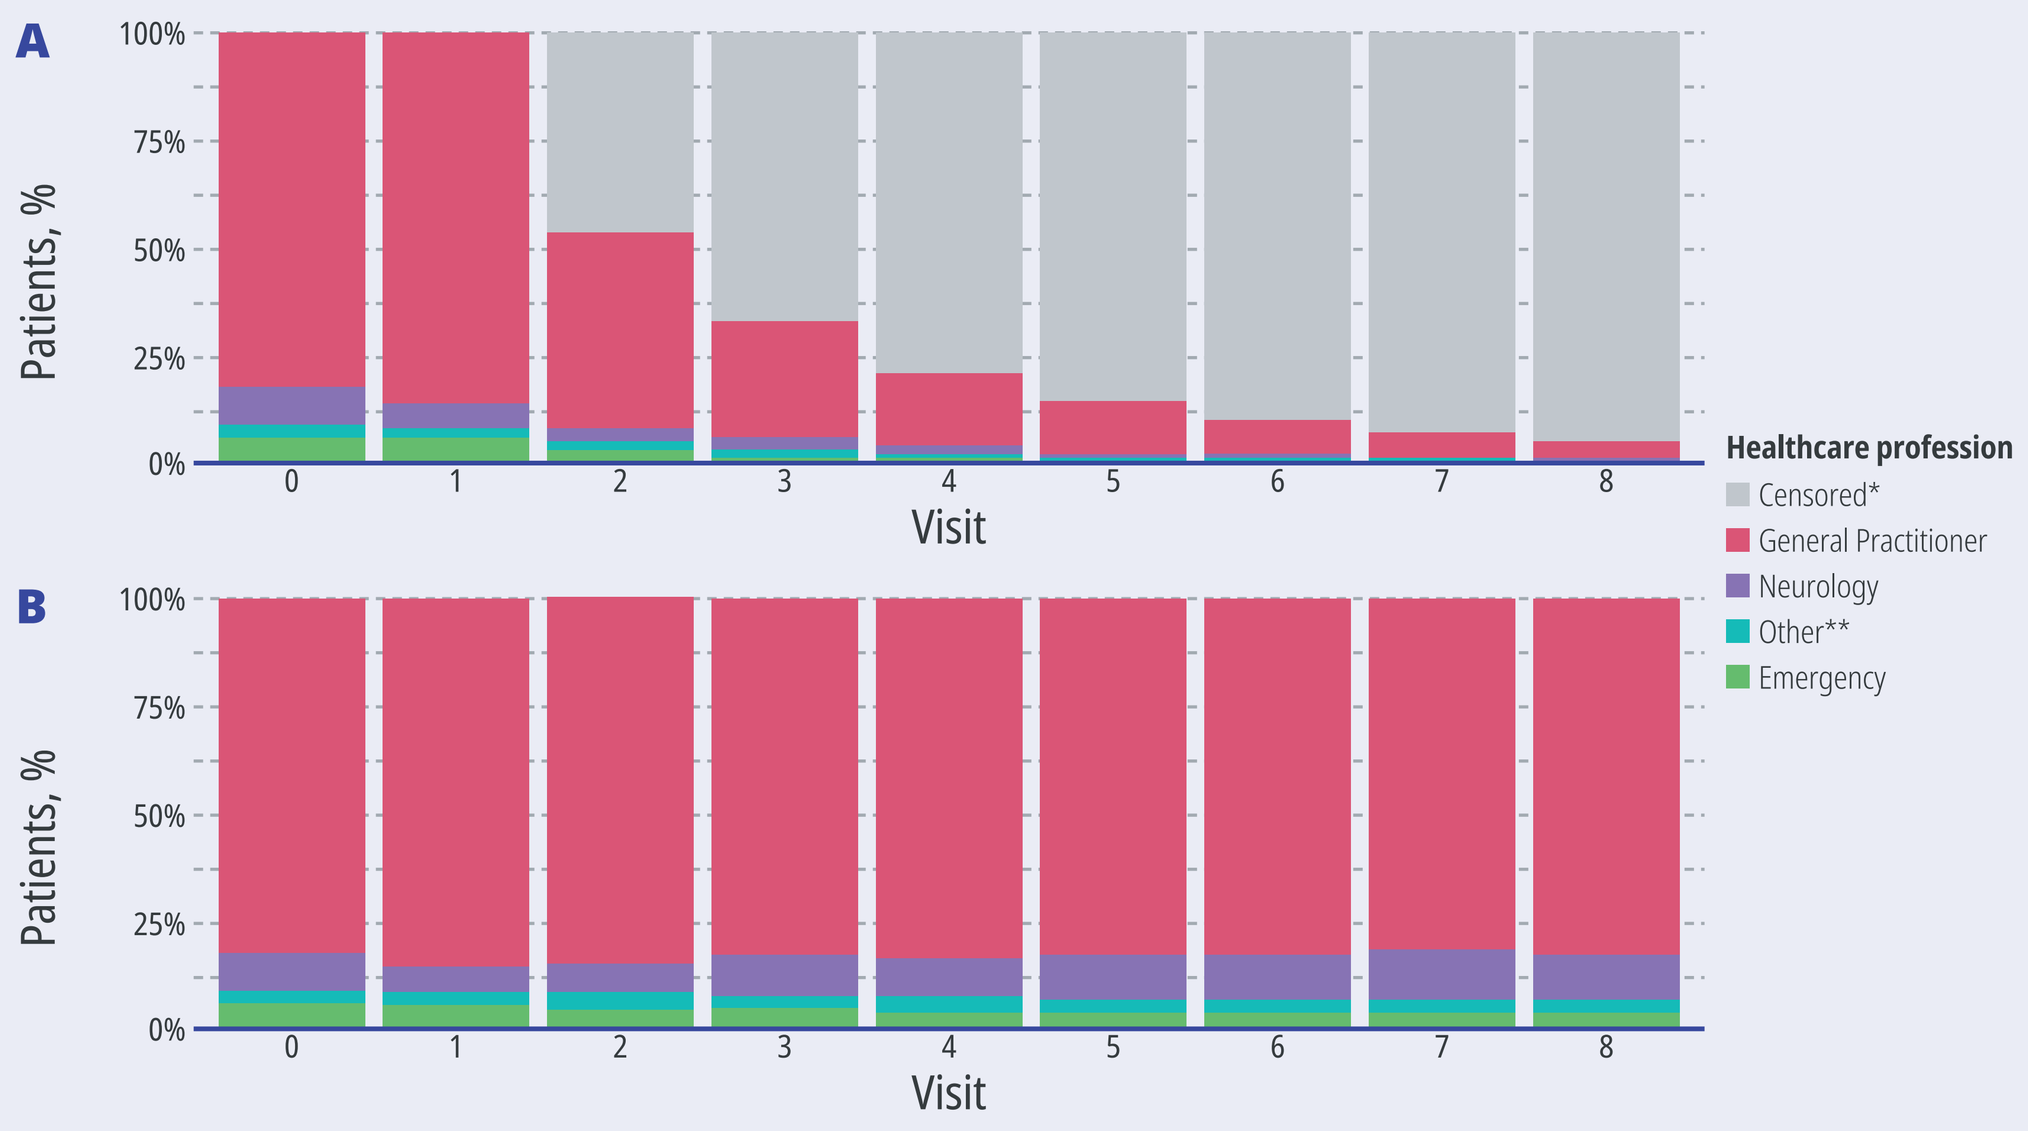
A
B
Patients, %
0%
25%
50%
75%
100%
0
1
2
3
4
5
6
7
8
Visit
Healthcare profession
Censored*
General Practitioner
Neurology
Other**
Emergency

* Censored (patients with no new healt care during subsequent follow-up)
** Other: Intenist, family physician occupational physician, anesthetists, Gynecologists, Nephrologists, Ophthalmologists, Psychiatrist/psychoterapist, General surgery, Endocrinologist, Neurosurgery, Bioenergetics, Neurosurgery, dermatology, physiatry, gastroenterology, and allergology.

CONCLUSION

- The administrative prevalence and incidence of migraine were lower compared to other studies, indicating that many migraine patients are untreated by the HMO. The prevalence demonstrated an inverse U-shaped-pattern over the study's follow-up period.
- The participation of neurologists in the management of migraine patients was very low, and increasing the involvement of neurologists in migraine management will improve the effectiveness of therapy, considering the conditions and characteristics of patients with migraine.

REFERENCES

1. (IHS) HCCotIHS. Headache Classification Committee of the International Headache Society (IHS) The International Classification of Headache Disorders, 3rd edition. Cephalalgia. 2018;38(1):1-211.
2. Steiner TJ, Stovner LJ, Jensen R, Uluduz D, Katsarava Z. Migraine remains second among the world's causes of disability, and first among young women: findings from GBD2019. J Headache Pain. 2020;21(137).
3. Navarro-Pérez MP, Marín-Gracia M, Bellosta-Diago E, Santos-Lasaosa S. [Epidemiology of migraine in Spain and Latin America]. Rev Neurol. 2020;71(3):10-118.

Financial statement: This study was funded by Pfizer. Poster presented at ISPOR Europe 2024, Barcelona, Spain, 17-20 November 2024