RWD157

A real-world study of Multiple Myeloma Patients With at least 3 Prior Treatment Lines and Triple Class Exposed Disease in Finland: Outcomes and Need for Healthcare Resource Use

Mervi Putkonen[1], Akseli Kivioja[2], Johanna Vikkula[3], Marika Waltari[2], Riikka Mattila[3], Katja Närhi[2], Jonna Eeva[4], Anu Partanen[5],
[1]Turku University Hospital, Department of Hematology, Turku, Finland, [2]Johnson & Johnson, Espoo, Finland, [3]Medaffcon Oy, Espoo, Finland,
[4]Tampere University Hospital, Department of Hematology, Tampere, Finland, [5]Kuopio University Hospital, Department of Medicine, Kuopio, Finland

Background

Multiple myeloma (MM) is an aggressive hematologic malignancy characterized by malignant plasma cell proliferation in the bone marrow. Patients who have received three or more prior lines of therapy and are exposed to proteasome inhibitors (PIs), immunomodulatory drugs (IMIDs), and anti-CD38 antibodies (triple class exposed, [TCE]) face a particular high unmet medical need as they have limited treatment options and clinical outcomes are typically poor. This study aims to understand current clinical practice in Finland for TCE-MM patients, by evaluating patient characteristics, treatment patters and outcomes as well as healthcare resource use (HCRU). This information is crucial for optimizing MM care and informing healthcare policy and resource allocation.

Results:

Patient demographics and treatment history:

- 1733 incident patients with MM and receiving pharmacological MM treatment (Table 1)
- 51 patients had received 3 prior treatment lines, were TCE and started following treatment (Table 1)
- Median age of 67 years (IQR 60-73) at TCE-MM
- 38 patients (75%) had 3-4 prior treatment lines
- No standard treatment can be defined for this group. 31 different treatments were used, with PCd (16%) and Kd (10%) being most common (Table 2)

Treatment outcomes:

- Median OS was 10.3 months (95% CI 7.4-17.9; Fig 1)
- Median TTNT for patients with TCE-MM was 4.4 months (95% CI 3.3-8.6; Fig 2)

Healthcare resource use (HCRU):

- The number of outpatient specialized care contacts per month was 6 (95% CI 4-7) for patients with TCE-MM and 3 (95% CI 3-3) for all patients with MM (Fig 3)
- The number of inpatient days per month was 2 (95% CI 1-3) for patients with TCE-MM and 1 (95% CI 1-1) for all patients with MM (Fig 3)
- The number of contacts at primary care for patients with TCE-MM was 2 (95% CI 1-4) and 4 (95% CI 4-5) for all patients with MM, respectively (Fig 3)
- The corresponding costs per patient month (PPM) were **4 002€** (95% CI 3 012-5 379) for patients with TCE-MM and **2 139€** (95% CI 2 054-2 223) for all patients with MM.

Table 1. Characteristics of multiple myeloma (MM) patients at two key points: at the time of their initial MM diagnosis and when their disease has progressed to become TCE-MM.

Variable		MM	TCE-MM	Missing*
n		1733	51	
Age at index, years, median [IQR]		71 [63, 78]	67 [60, 73]	0 %
Female sex, N (%)		834 (48)	21 (41)	0 %
MM type, N (%)	IgG	799 (51)	29 (57)	0 %
	Non-IgG/ unknown	934 (49)	22 (43)	
ECOG, N (%)	0	159 (22)	9 (32)	45 %
	1	266 (36)	8 (29)	
	2+	307 (42)	9 (39)	
Cytogenetic risk, N (%)	Standard	760 (77)	32 (71)	12 %
	High	224 (23)	13 (29)	
ISS, N (%)	I	224 (17)	13 (28)	8 %
	II	602 (44)	19 (40)	
	III	530 (39)	15 (32)	

**calculated for TCE-MM cohort*

Table 2. Treatment characteristics of TCE-MM-patients

Variable		TCE-MM
Number of prior treatment lines, median [IQR]		3 [3, 5]
Exposure status, N (%)	triple exposed	51 (100)
	quad exposed	38 (75)
	penta exposed	14 (28)
Prior SCT, N (%)		38 (75)
Time from MM diagnosis, years, N (%)	<6	44 (86)
	≥6	7 (14)
Time from MM diagnosis, years, median [IQR]		3 [2, 5]
Length of follow-up, months, median [IQR]		8 [3, 20]

Conclusions

- Based on the amount of index treatments, standard of care for TCE-MM cannot be defined.
- **Poor Prognosis:** Despite advancements in MM treatment, TCE-MM patients have a median OS of only 10.3 months and a median TTNT of 4.4 months.
- **High Burden of Care**: TCE-MM patients exhibit higher number of outpatient specialized care contacts and inpatient days compared to the general MM patient population.
- **Increased Costs:** The costs per patient month for TCE-MM patients are nearly double those for general MM patients, reflecting the intensive care and management required for this heavily pretreated group.
- The results in this study are in line with those in the LocoMMotion study (Mateos et al. Leukemia, 2022. https://doi.org/10.1038/s41375-022-01531-2)
- These findings underscore the vulnerable position of patients with TCE disease, and the need for more effective treatment strategies for this patient population.

Methods

- This retrospective study included MM and TCE-MM patients diagnosed in Finland (2013 – 2022).
 - Patients were identified and EHR data collected from four hospital district data lakes (Helsinki and Uusimaa, Southwest Finland, Pirkanmaa and Northern Savo).
 - Data on primary care visits, and reimbursed drug purchases were collected from the National institute of health and welfare, and Social insurance institution, respectively.
- TCE-MM cohort was formed of MM patients with ≥3 prior treatment lines and exposure to PI, IMID, and anti-CD38 antibody. Two index dates were defined for the cohorts: initial MM diagnosis and start of a new treatment line after progressing to TCE. Patients were followed from index until death or Dec 31, 2022 (end of study, EOS).
- Overall survival (OS) was analyzed as time from index until death (event), or EOS (censoring). Time to next treatment (TTNT) was analyzed as time from index until start of a new treatment line (event), death (competing risk), or EOS (censoring). All-cause HCRU was analyzed per patient month (PPM; total HCRU divided by the length of follow-up) with costs being estimated using unit costs. Treatment lines were defined according to Rajkumar SV, et al. (Blood. 2015 Aug 13;126(7):921-2).

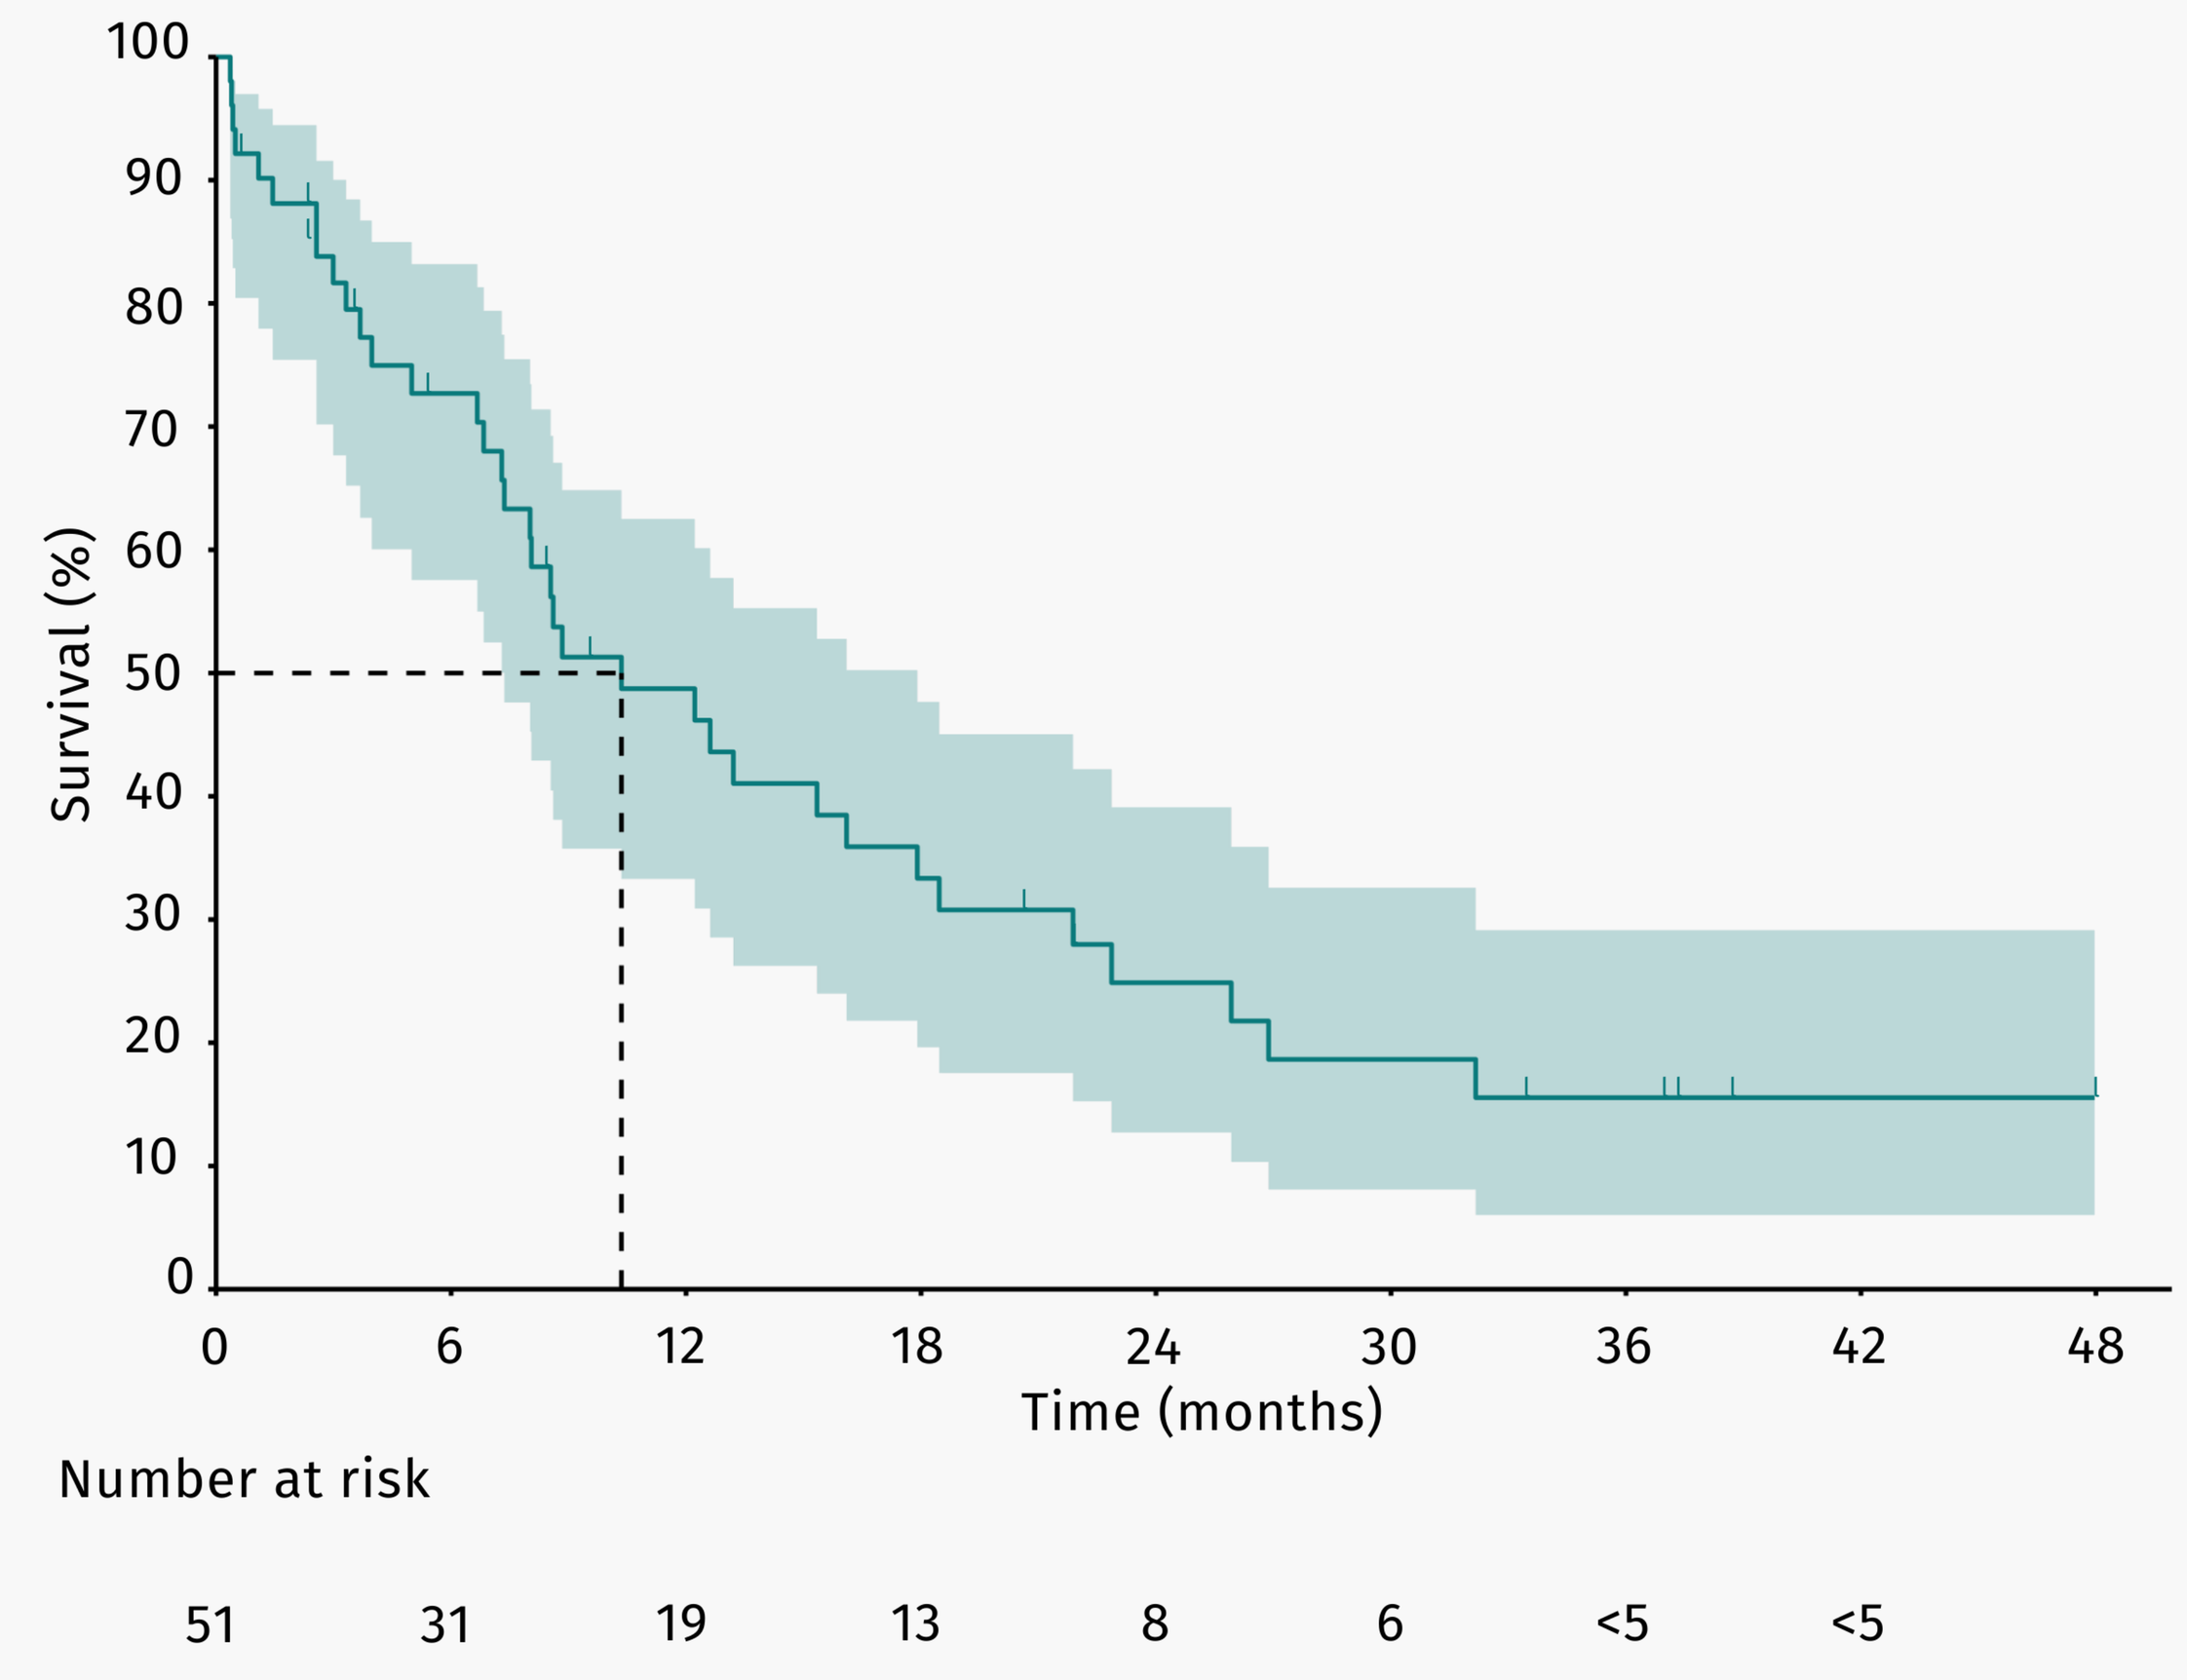
Survival (%)
100
90
80
70
60
50
40
30
20
10
0
0
6
12
18
24
30
36
42
48
Time (months)
Number at risk
51
31
19
13
8
6
<5
<5

Figure 1. *Overall survival of patients with TCE-MM. Shaded areas represent 95% CI.*

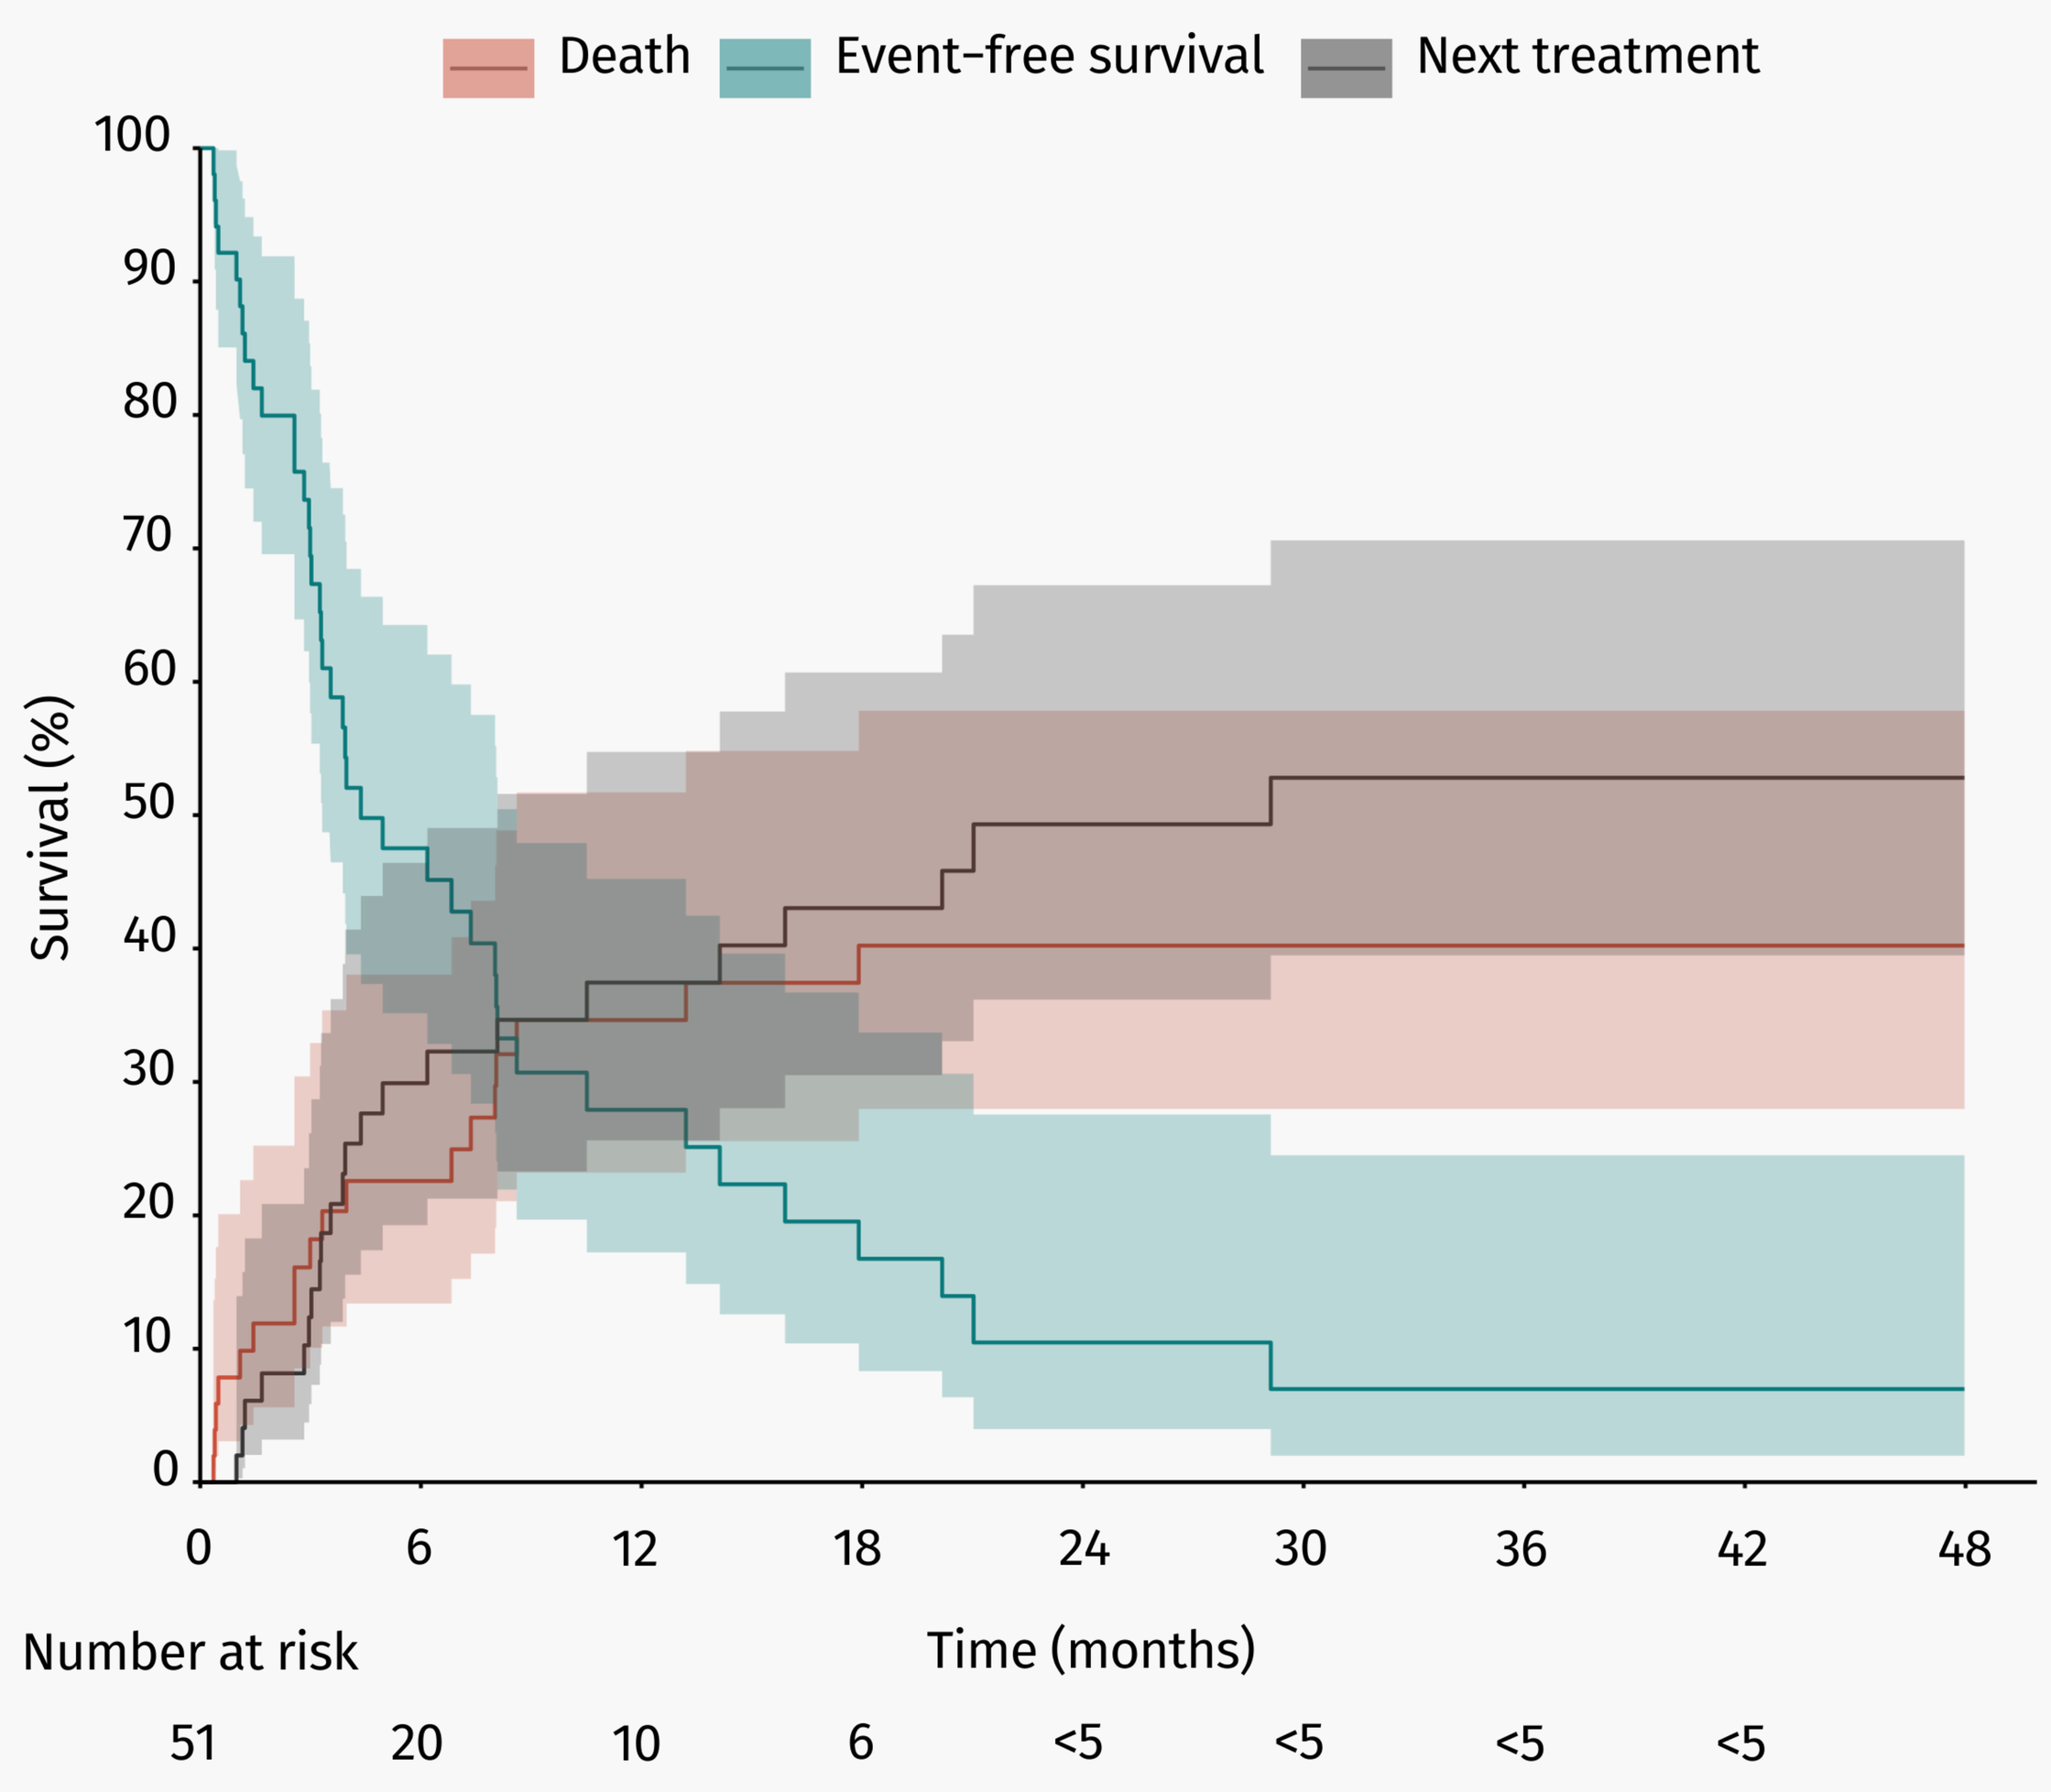
Death
Event-free survival
Next treatment
Survival (%)
100
90
80
70
60
50
40
30
20
10
0
0
6
12
18
24
30
36
42
48
Number at risk
Time (months)
51
20
10
6
<5
<5
<5
<5

Figure 2. *Competing risk model for TTNT from TCE-MM. Teal: TTNT (event free survival; patients who have not started next treatment line and are alive), Black: next treatment started, Red: death before next treatment.*

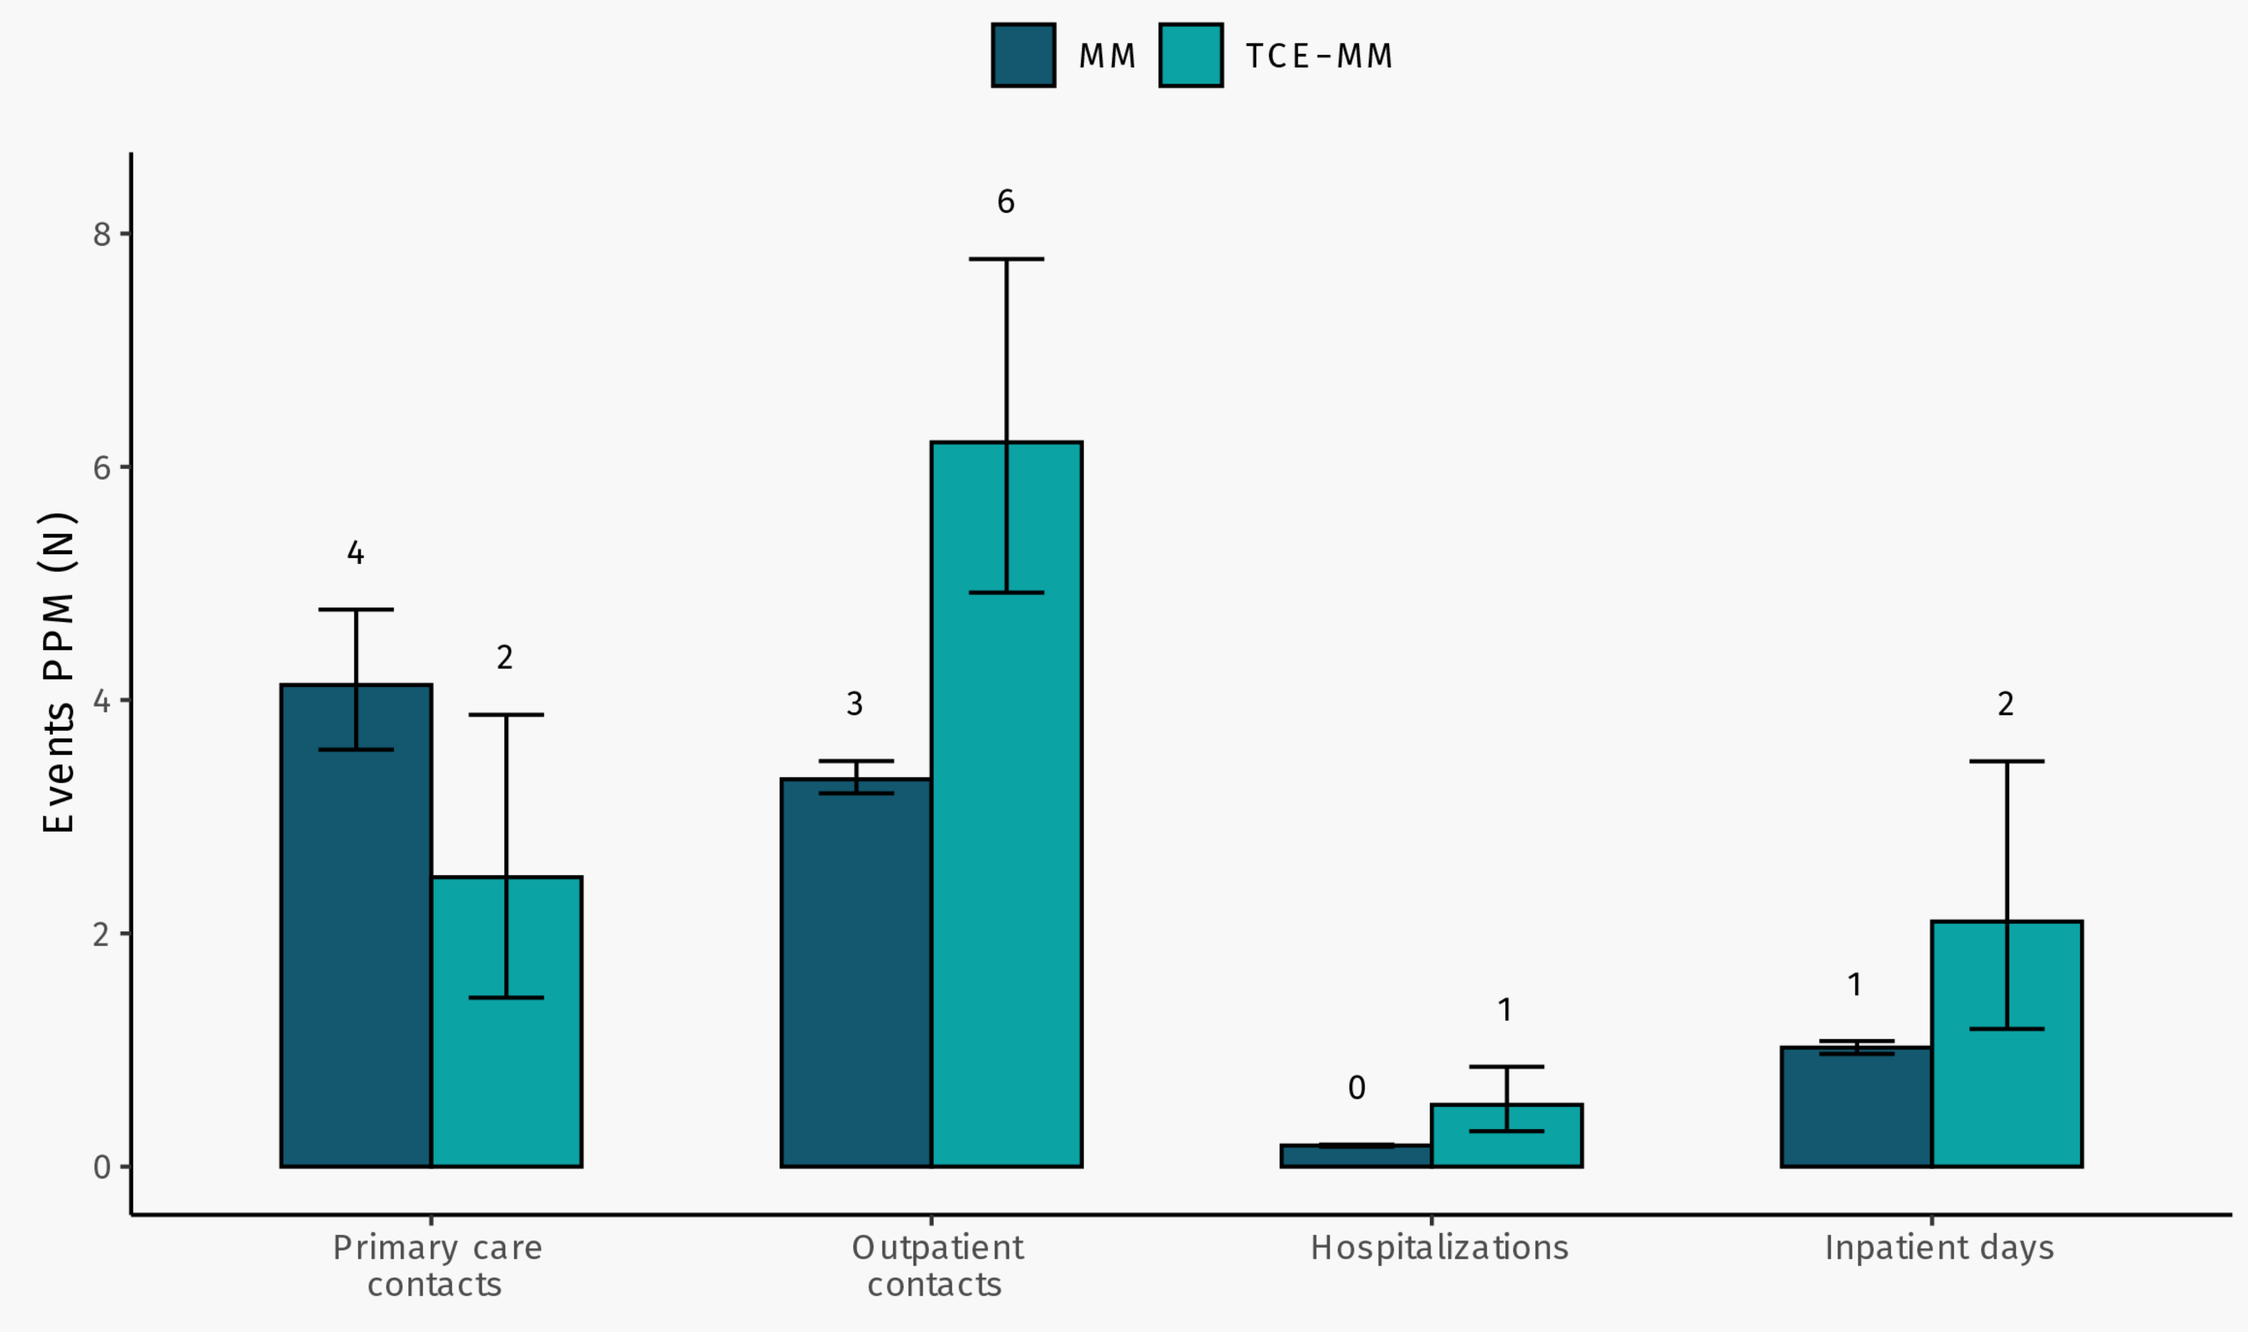
MM
TCE-MM
Events PPM (N)
8
6
4
2
0
4
2
3
6
0
1
1
2
Primary care contacts
Outpatient contacts
Hospitalizations
Inpatient days

Figure 3. *Healthcare resource use of patients with MM (teal) and TCE-MM (turquoise). HCRU is presented as per patient month (PPM). Whiskers represent 95% CI.*

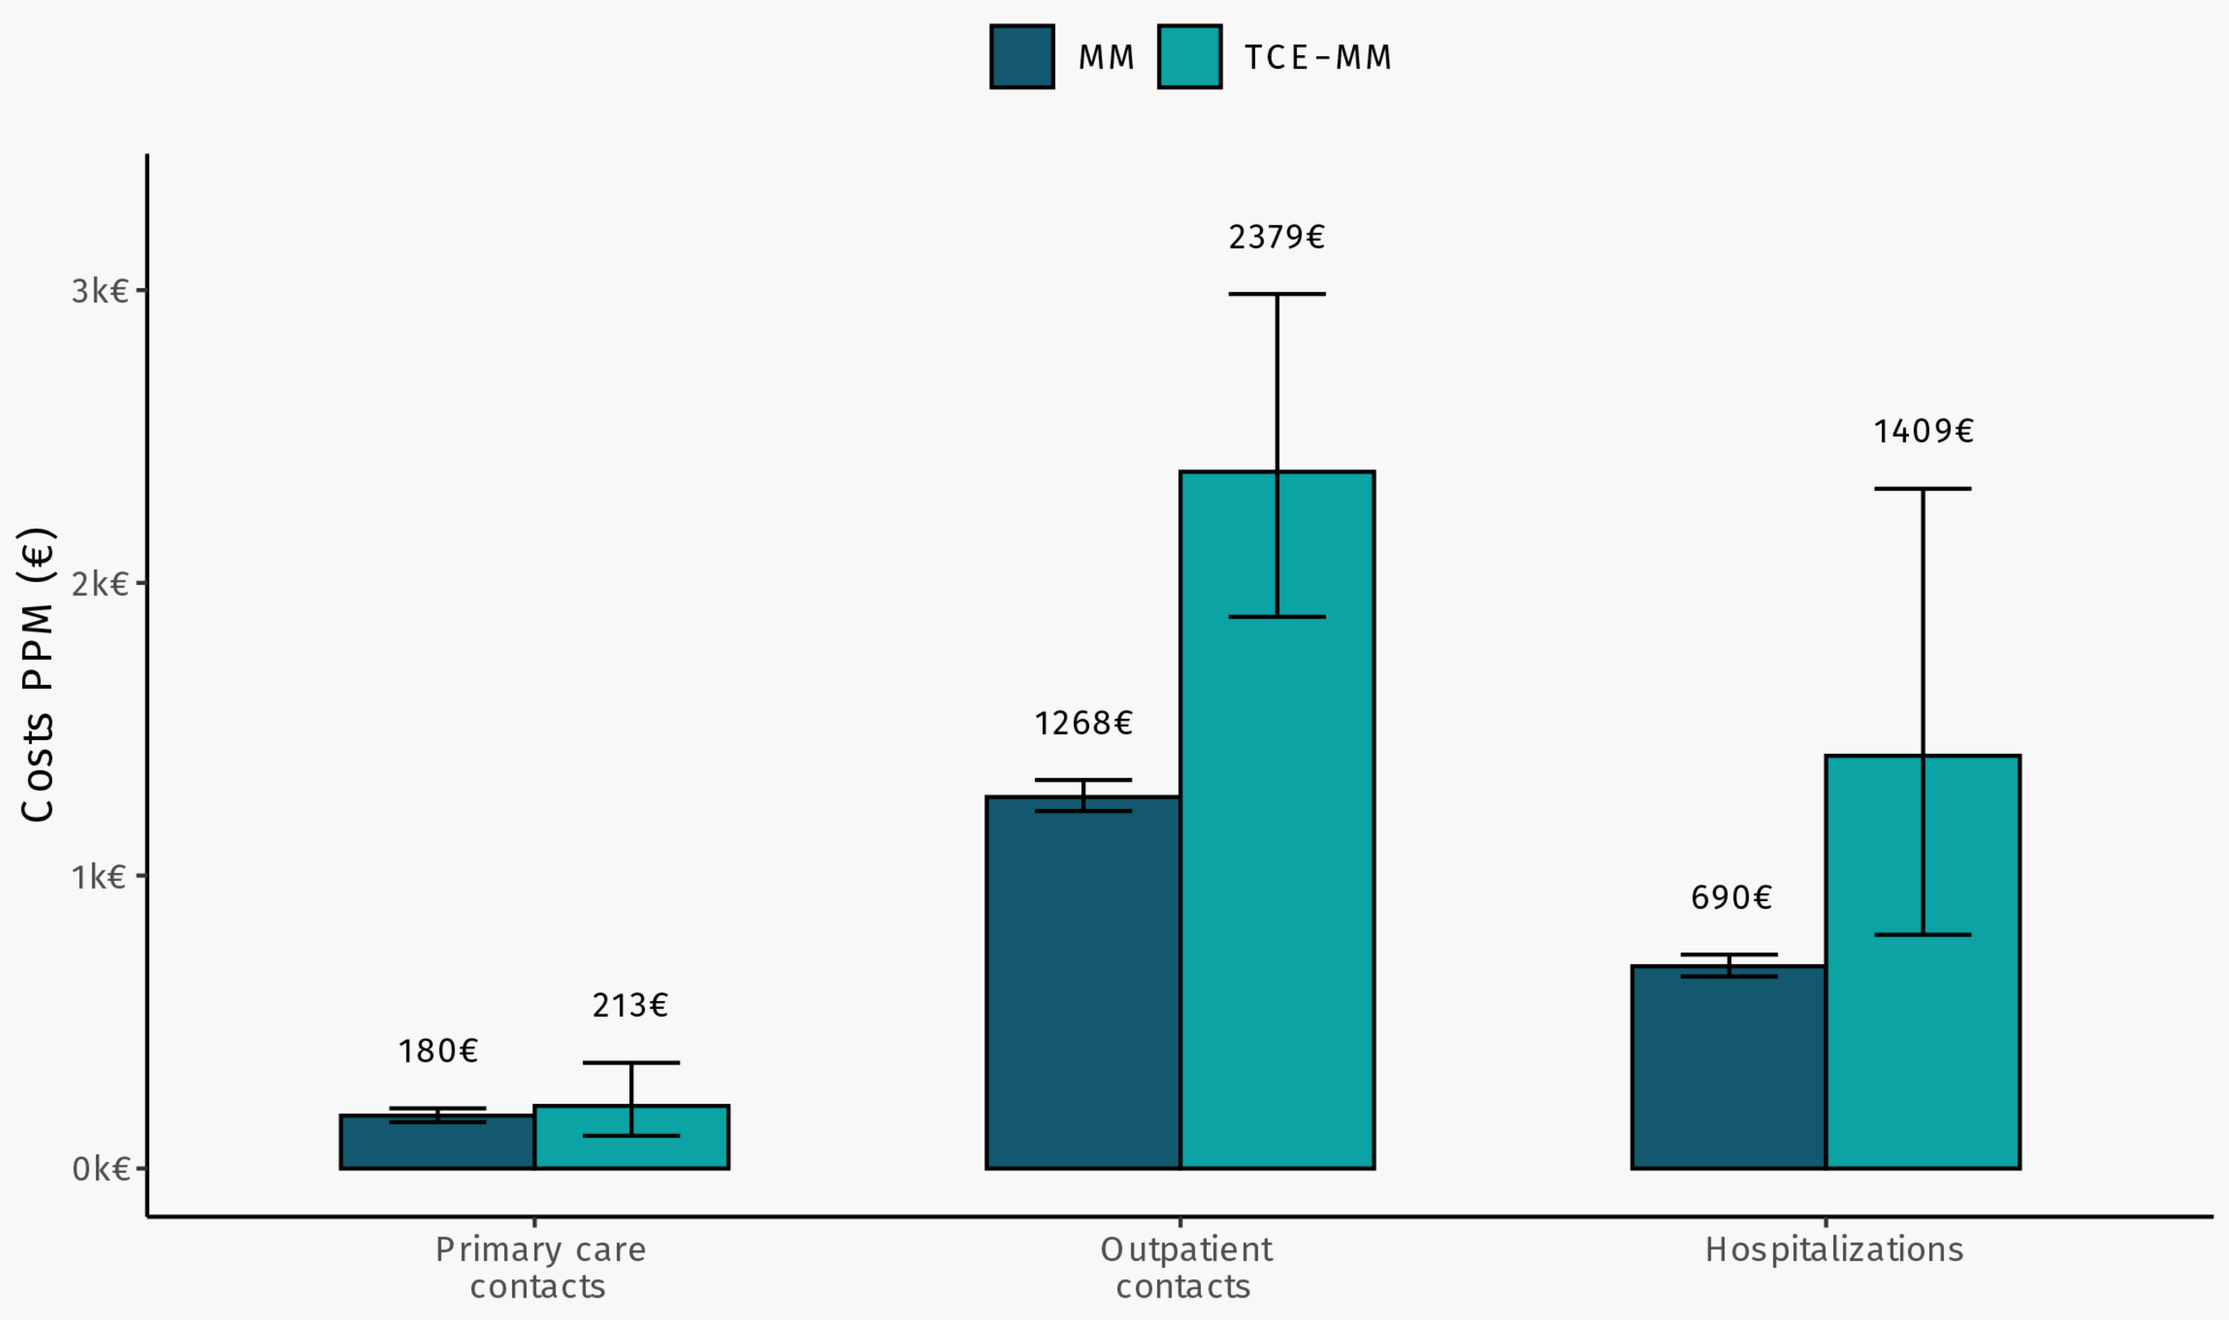
MM
TCE-MM
Costs PPM (€)
3k€
2k€
1k€
0k€
180€
213€
1268€
2379€
690€
1409€
Primary care contacts
Outpatient contacts
Hospitalizations

Figure 4. *Healthcare resource use as costs for patients with MM (teal) and TCE-MM (turquoise). Costs are presented in € per patient month (PPM). Whiskers represent 95% CI.*

Funding: This work was supported by J&J innovative medicine EMEA and Finland as a company sponsored study. The sponsor and its employee had no access to the data at any time or any role in the data management and data analysis of the study.