The Burden-of-Illness of Duchenne Muscular Dystrophy in Japan: A Socioeconomic Survey

EE472

Shimizu-Motohashi Y[1], Merla V[2], Nagano M[3], Shima D[3], **Posner N[2]**, Evans J[4]

[1]National Center of Neurology and Psychiatry, Kodaira, Tokyo, Japan [2]Pfizer Inc., New York, NY, USA [3]Pfizer Japan Inc.,Tokyo, Japan [4]HCD Economics, Knutsford, UK

Presented at ISPOR EU 2024• November 17-20 • In-person

Objectives

1) Estimate the average per-patient cost, including direct medical, direct non-medical, and indirect cost, of Duchenne muscular dystrophy (DMD) from a societal perspective in Japan,
2) Quantify the health-related quality of life (HRQoL) impact of DMD in Japan on patients and caregivers.

Conclusions

In Japan, as patient disease severity increased, total societal cost increased, driven by direct non-medical cost.

While trends in HRQoL scores suggested substantial humanistic impact as DMD progressed, further work is required to identify key drivers of these trends, with larger sample sizes.

Background

- Duchenne muscular dystrophy (DMD) is a severe degenerative neuromuscular condition, which causes a variety of problems such as muscular weakness, respiratory failure, and loss of ambulation
- Median patient life expectancy for DMD is estimated at 22 years[1]
- In Japan, the prevalence of DMD is 1.9 to 3.4 per 100,000 individuals and it is estimated that there are 3,000 to 4,000 patients with DMD[2]
- DMD is associated with significant socioeconomic burden due to the intensity of care required and multi-organ involvement[3]
- There is a lack of real-world data on the cost burden of DMD placed on healthcare systems, patients, and their caregivers, particularly in Japan

Materials and Methods

- Physicians completed an electronic case report form (eCRF) using patients' medical records. Information collected included demographics, clinical, and healthcare resource utilization (HRU)
- All data were collected with respect to the past 12 months
- For physicians to be eligible they needed a location of practice in Japan, a primary specialty in neurology or other DMD specialities, qualified in their medical specialty for at least three years, and personally responsible for the management of paediatric and/or adult patients with DMD
- Patient eligibility criteria included: Male, diagnosed with DMD (via muscle biopsy or genetic test), and diagnosed at least 12 months before date of consultation (index date)
- Patients and their caregivers were recruited separately via the Japanese Muscular Dystrophy Association to complete Patient Public Involvement and Engagement forms (PPIE)
- For patients with cognitive delay or below 18 years, caregivers were asked to complete the study on the patient's behalf and provide informed consent
- The PPIE collected data including quality of life and direct non-medical and indirect costs, related to patients (PPIE – P) and their caregivers (PPIE – C), over the last 12 months
- Patient reported outcomes were included in the PPIE which collected data on HRQoL (EQ5D-5L, DMDQoL)

Analysis

- Country-specific unit costs were multiplied by HRU to estimate direct medical cost from the eCRF
- All EQ5D versions utility scores were calculated using Japanese value sets. DMDQoL utility scores were calculated using the only value set available (UK)
- Mean societal cost and HRQoL were calculated per patient and stratified using the Project HERCULES natural history model[4], as displayed in **Figure 1**

Figure 1. Project HERCULES natural history model

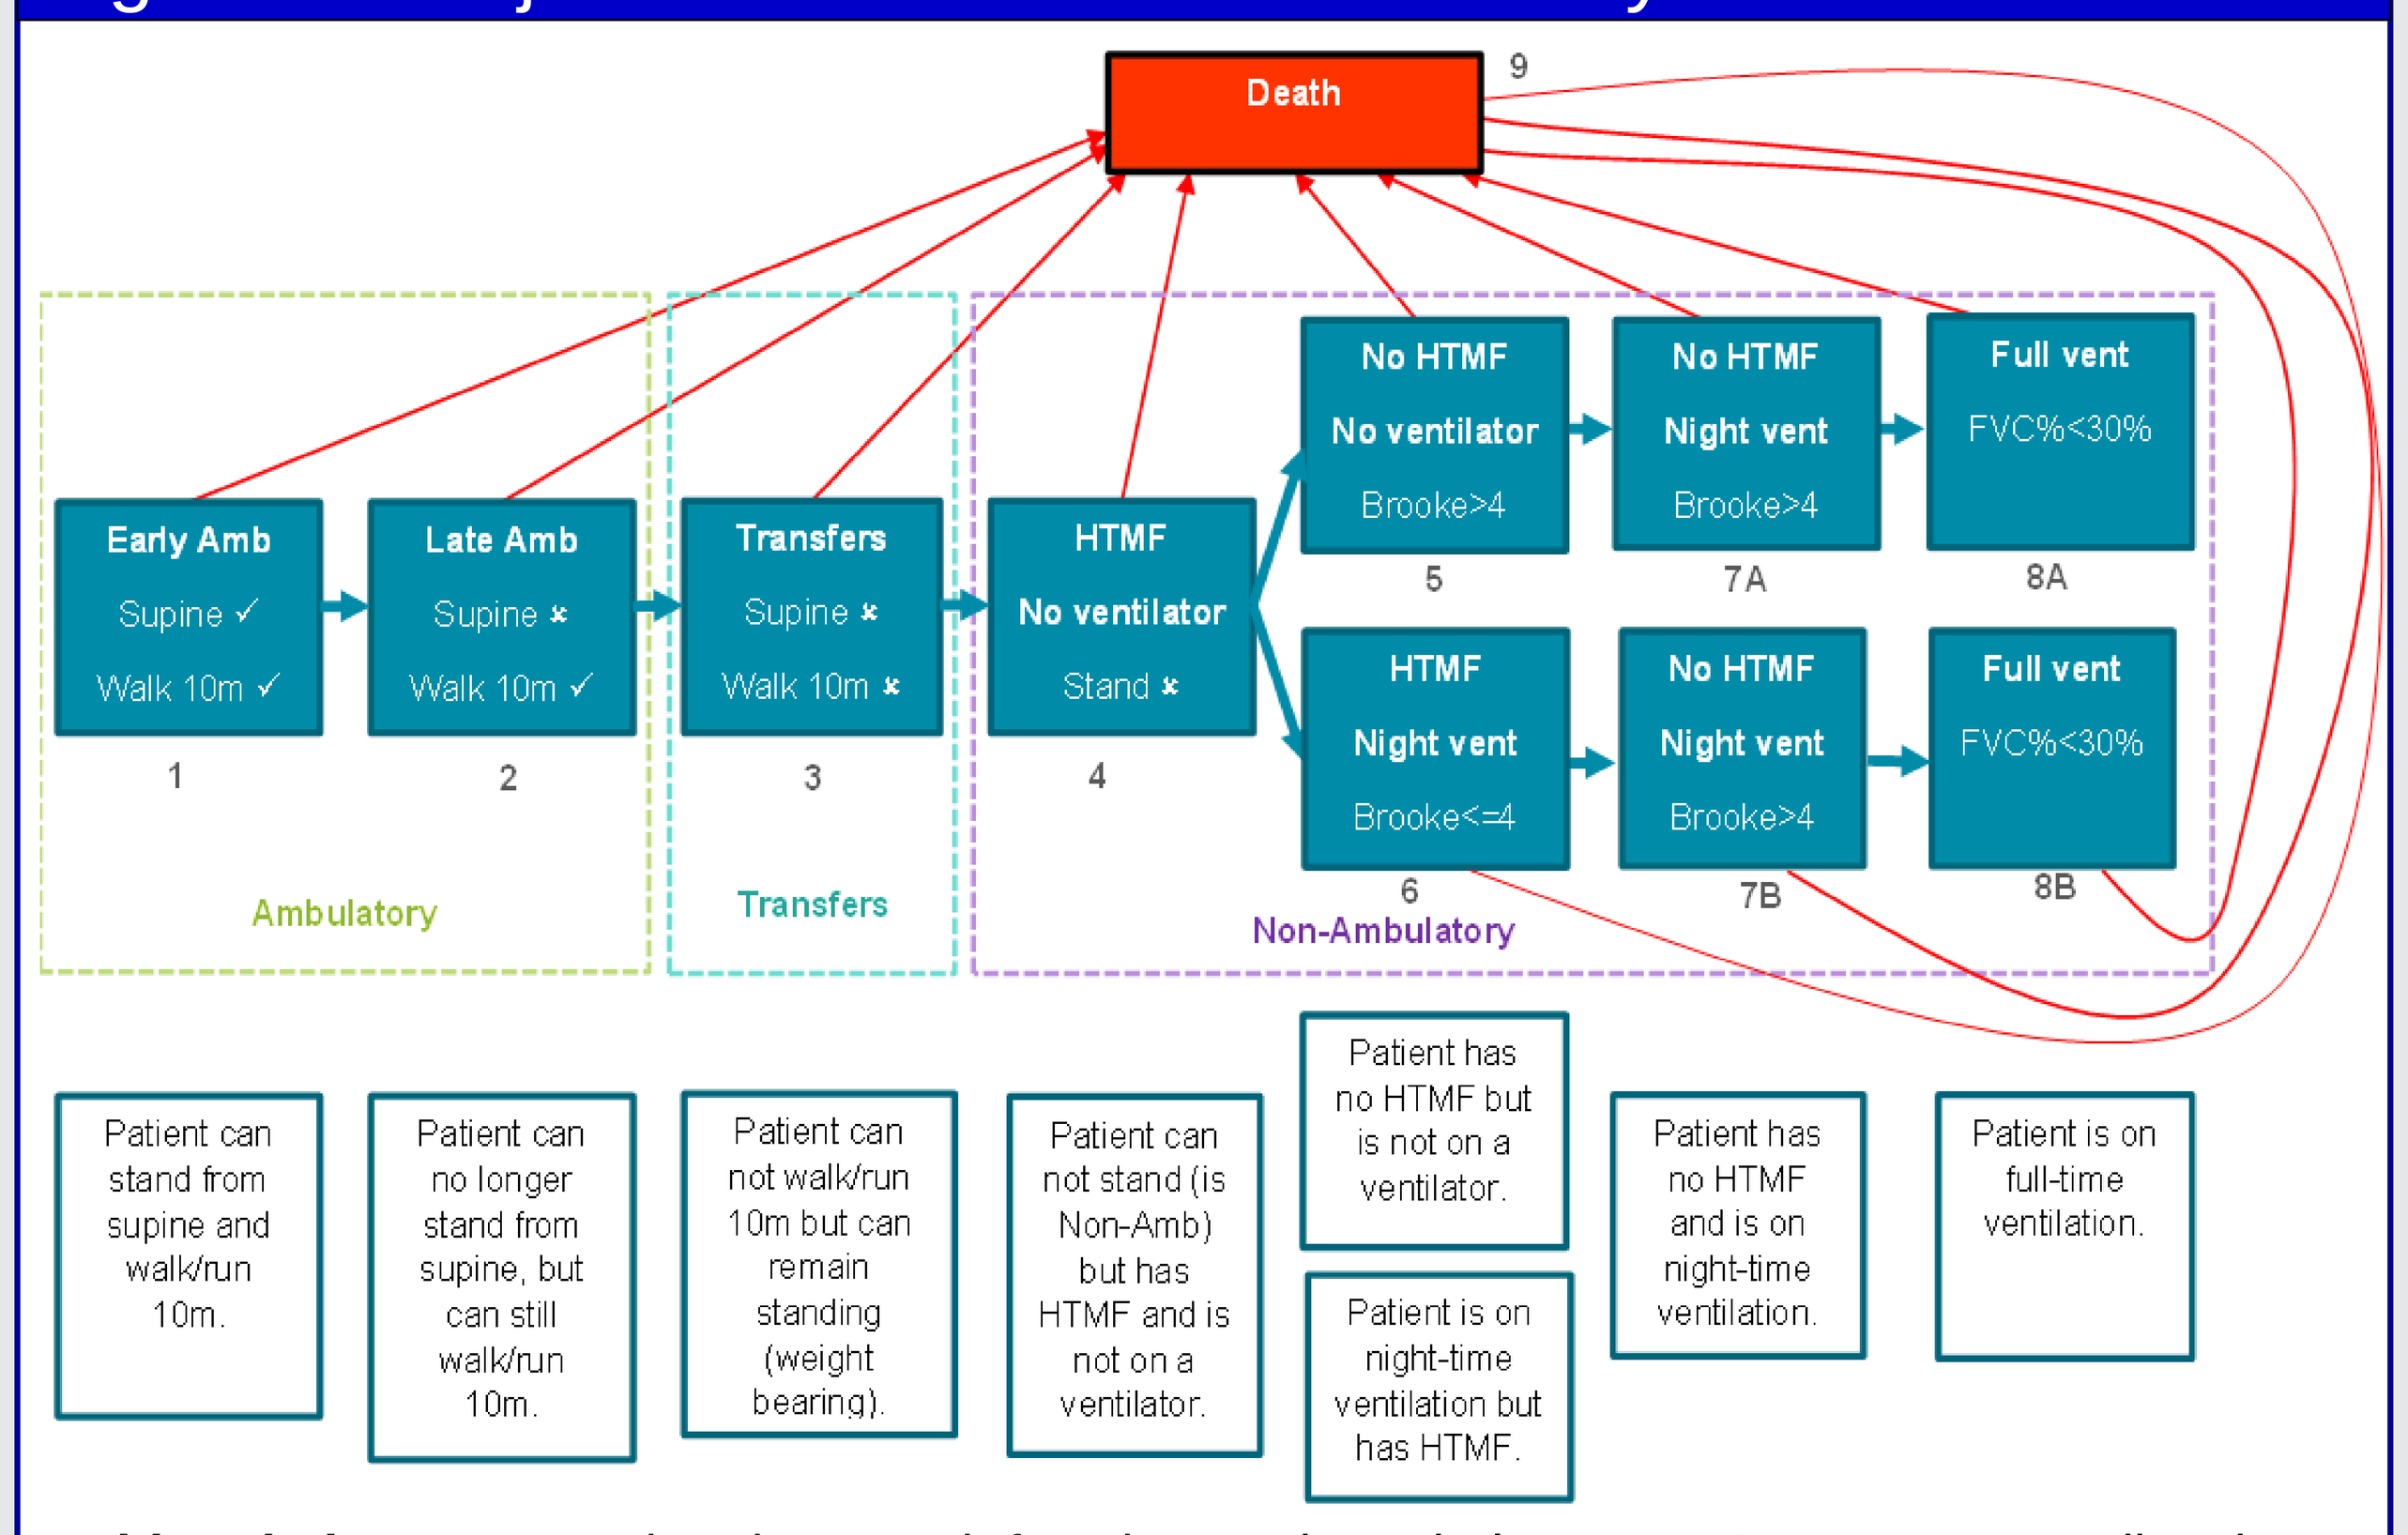
Death
9
No HTMF
No ventilator
Brooke>4
5
No HTMF
Night vent
Brooke>4
7A
Full vent
FVC%<30%
8A
Early Amb
Supine ✓
Walk 10m ✓
1
Late Amb
Supine ✗
Walk 10m ✓
2
Transfers
Supine ✗
Walk 10m ✗
3
HTMF
No ventilator
Stand ✗
4
HTMF
Night vent
Brooke<=4
6
No HTMF
Night vent
Brooke>4
7B
Full vent
FVC%<30%
8B
Ambulatory
Transfers
Non-Ambulatory
Patient can stand from supine and walk/run 10m.
Patient can no longer stand from supine, but can still walk/run 10m.
Patient can not walk/run 10m but can remain standing (weight bearing).
Patient can not stand (is Non-Amb) but has HTMF and is not on a ventilator.
Patient has no HTMF but is not on a ventilator.
Patient is on night-time ventilation but has HTMF.
Patient has no HTMF and is on night-time ventilation.
Patient is on full-time ventilation.

Abbreviations: HTMF: hand-to-mouth function; Amb: ambulatory; FVC%: percent predicted forced vital capacity

Results

Sample

- The DMD burden-of-illness (BOI) Japan sample consisted of 63 eCRFs with 38 PPIEs
- **Table 1** displays sample size and key demographics of the DMD BOI Japan sample

Table 1. Study sample size and key demographics

Characteristics	eCRF	PPIE
Number of complete forms	63	38
Mean (SD) age	15.9 (11.6)	17.5 (15.6)
Mean (SD) age at diagnosis	4.3 (3.4)	N/A*
Mean (SD) BMI	17.5 (3.0)	N/A*

*age of diagnosis and height and weight were not captured in the PPIE
Abbreviations: eCRF: electronic case report form; n: number; BMI: body mass index; SD: standard deviation; PPIE: public involvement and engagement form

- Both eCRF and PPIE samples were stratified by the natural history of disease model (**Figure 2**)

Figure 2. Sample distribution by disease stage

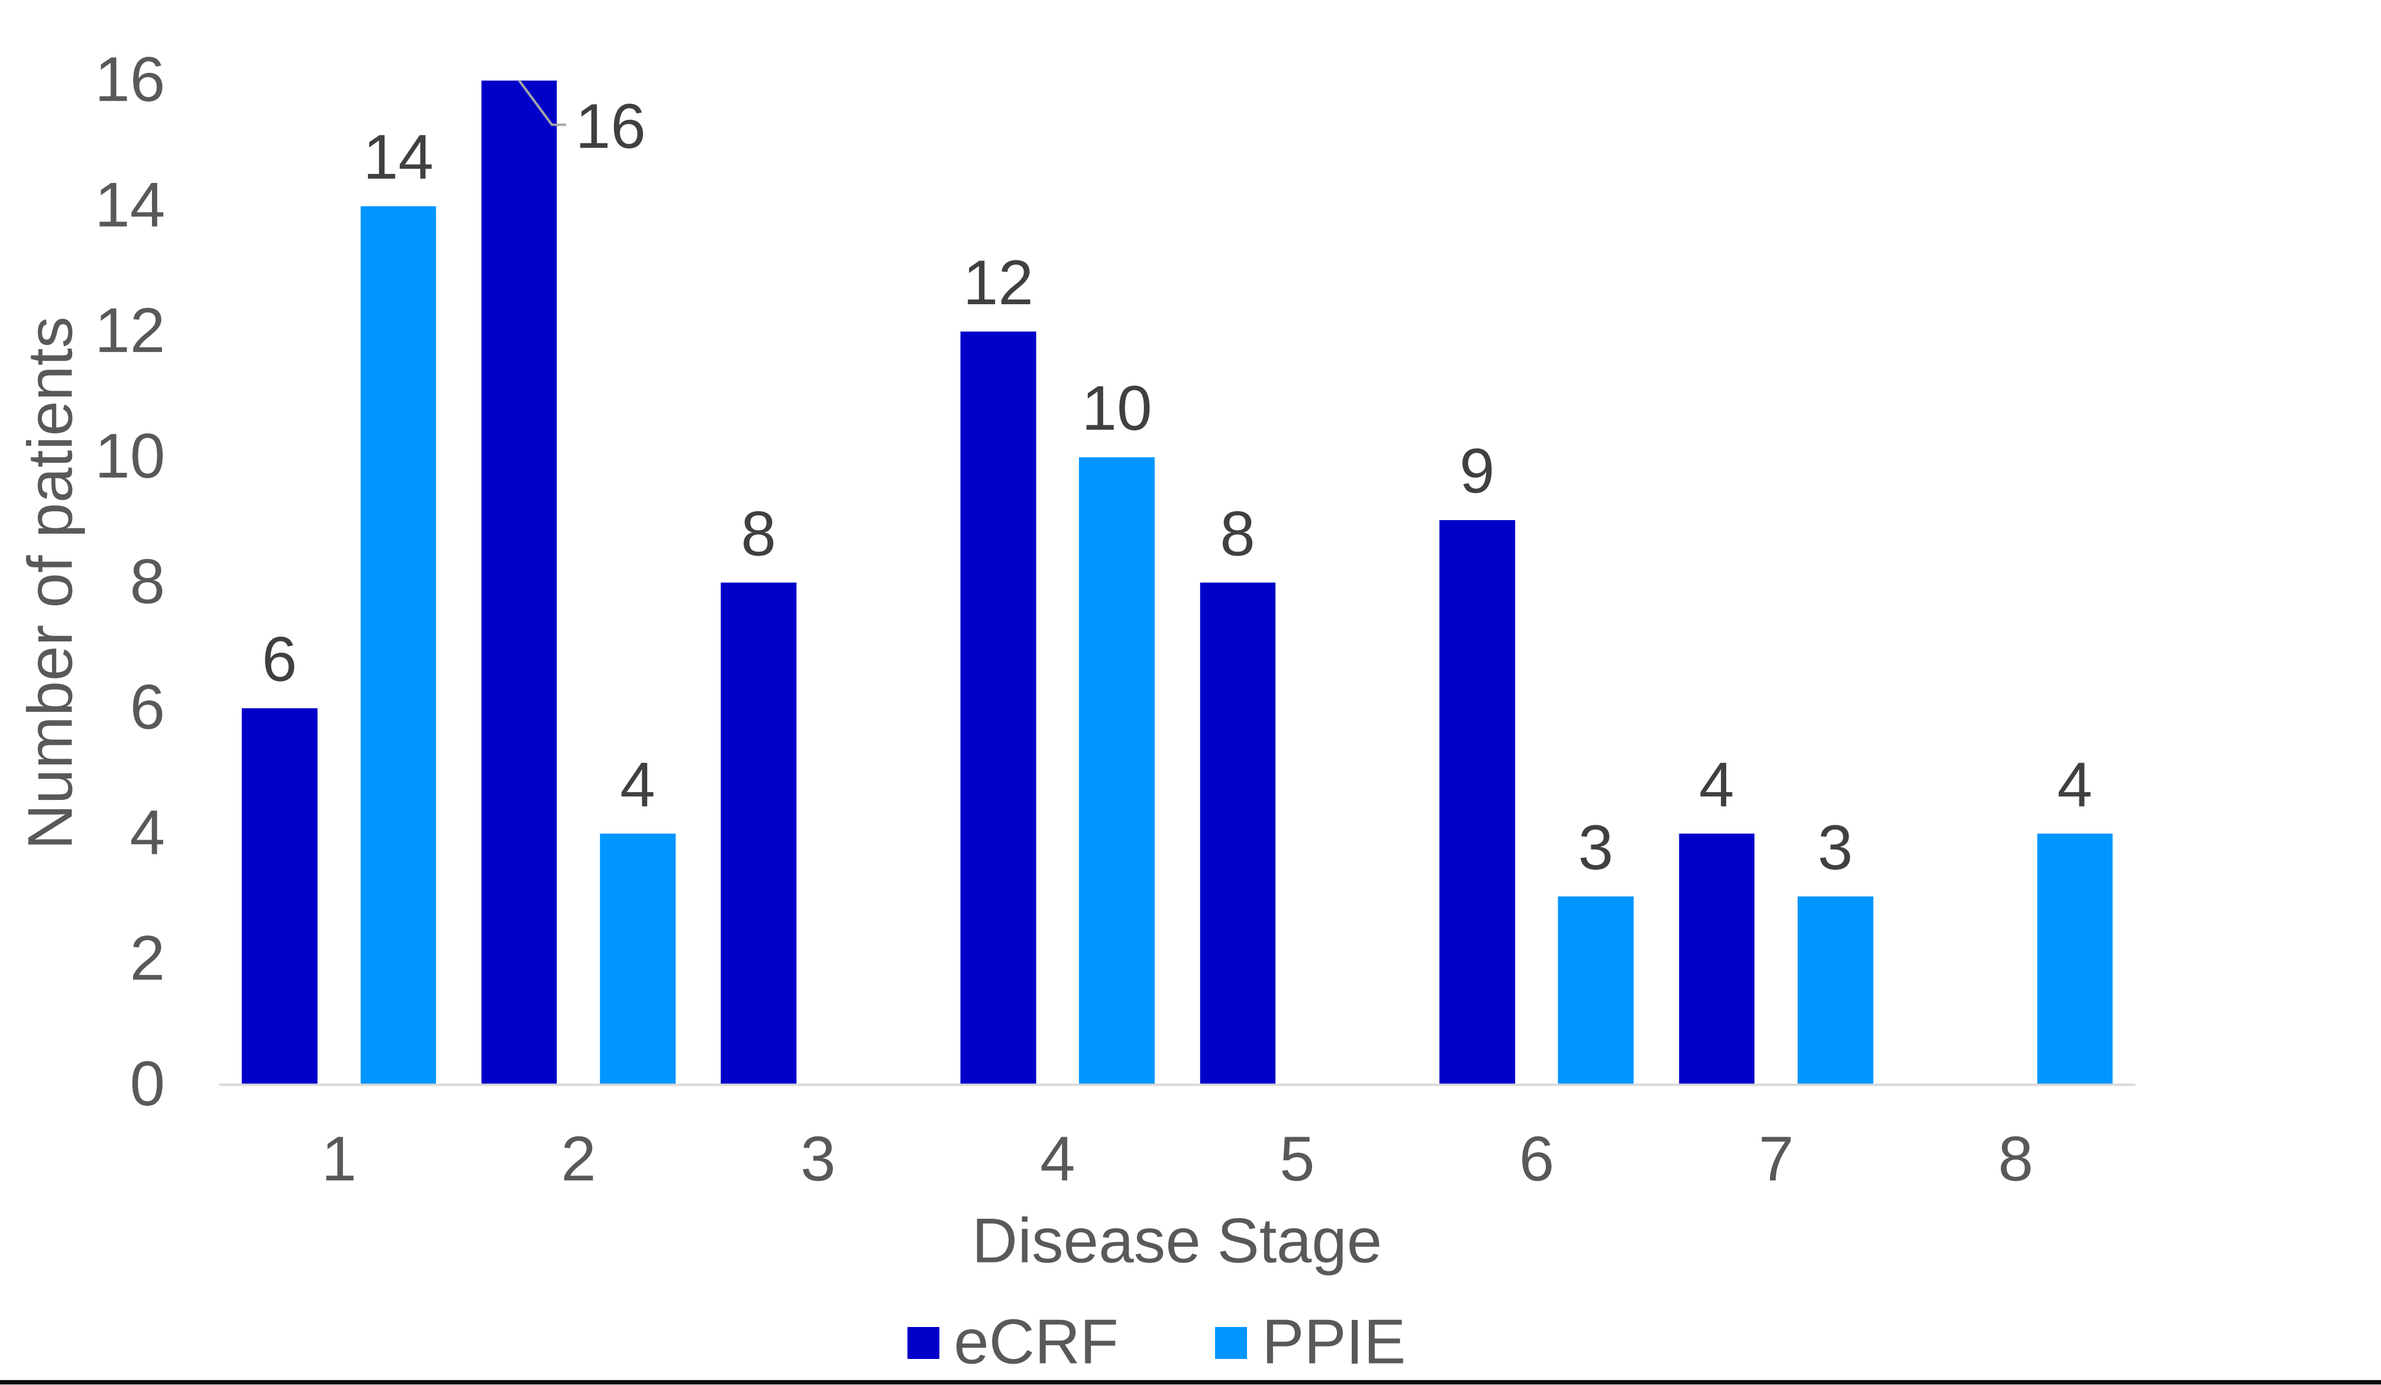
Number of patients
16
14
12
10
8
6
4
2
0
6
14
16
4
8
12
10
8
9
3
4
3
4
1
2
3
4
5
6
7
8
Disease Stage
eCRF
PPIE

Abbreviations: eCRF: electronic case report form; PPIE: public involvement and engagement form

- All disease stages (DS) had at least one form completed for them. Only disease stages 3, 5 and 8 didn't have both eCRF and PPIE data available.
- DS 2 and 1 were the most prevalent for eCRFs and PPIEs respectively, with 16 eCRFs in DS 2 and 14 PPIEs in DS 1
- In the eCRF sample, 35% of patients were ambulatory (DS 1-2) in comparison with the PPIE sample where 47% of patients were ambulatory

Societal Cost

- Mean direct medical, direct non-medical, and indirect, and societal cost per patient over the last 12 months is presented and stratified by DS in **Table 2**
- Only medical device use cost was available for direct medical cost in DS 8. No direct non-medical or indirect cost data were available for DS 3 and 5
- Across the sample, direct non-medical cost contributed the highest proportion of the total cost (75%), followed by indirect cost (15%) and direct medical cost (10%)
- Mean direct non-medical and indirect costs were highest in DS 8, while direct medical cost was highest in DS 6
- Overall, the total societal cost of DMD increased as severity increased through the DS, peaking at ¥19,184,381 in DS 8

Health-related quality of life

- Of the 38 patients who completed a PPIE form, 13 (34%) completed the EQ5D-5L
- Mean EQ5D-5L utility scores per patient are presented in **Figure 3**, stratified by disease stage
- As DMD severity progressed, mean EQ5D-5L utility score decreased
- Mean DMDQoL utility scores were available for 28 patients (74%) and showed large decreases in HRQoL in the later disease stages
- DS 7 had a mean DMDQoL utility score of 0.31 and DS 8 reported the lowest DMDQoL utility score of 0.03, in comparison to the sample average of 0.5

Figure 3. Mean EQ5D-5L utility score by disease stage

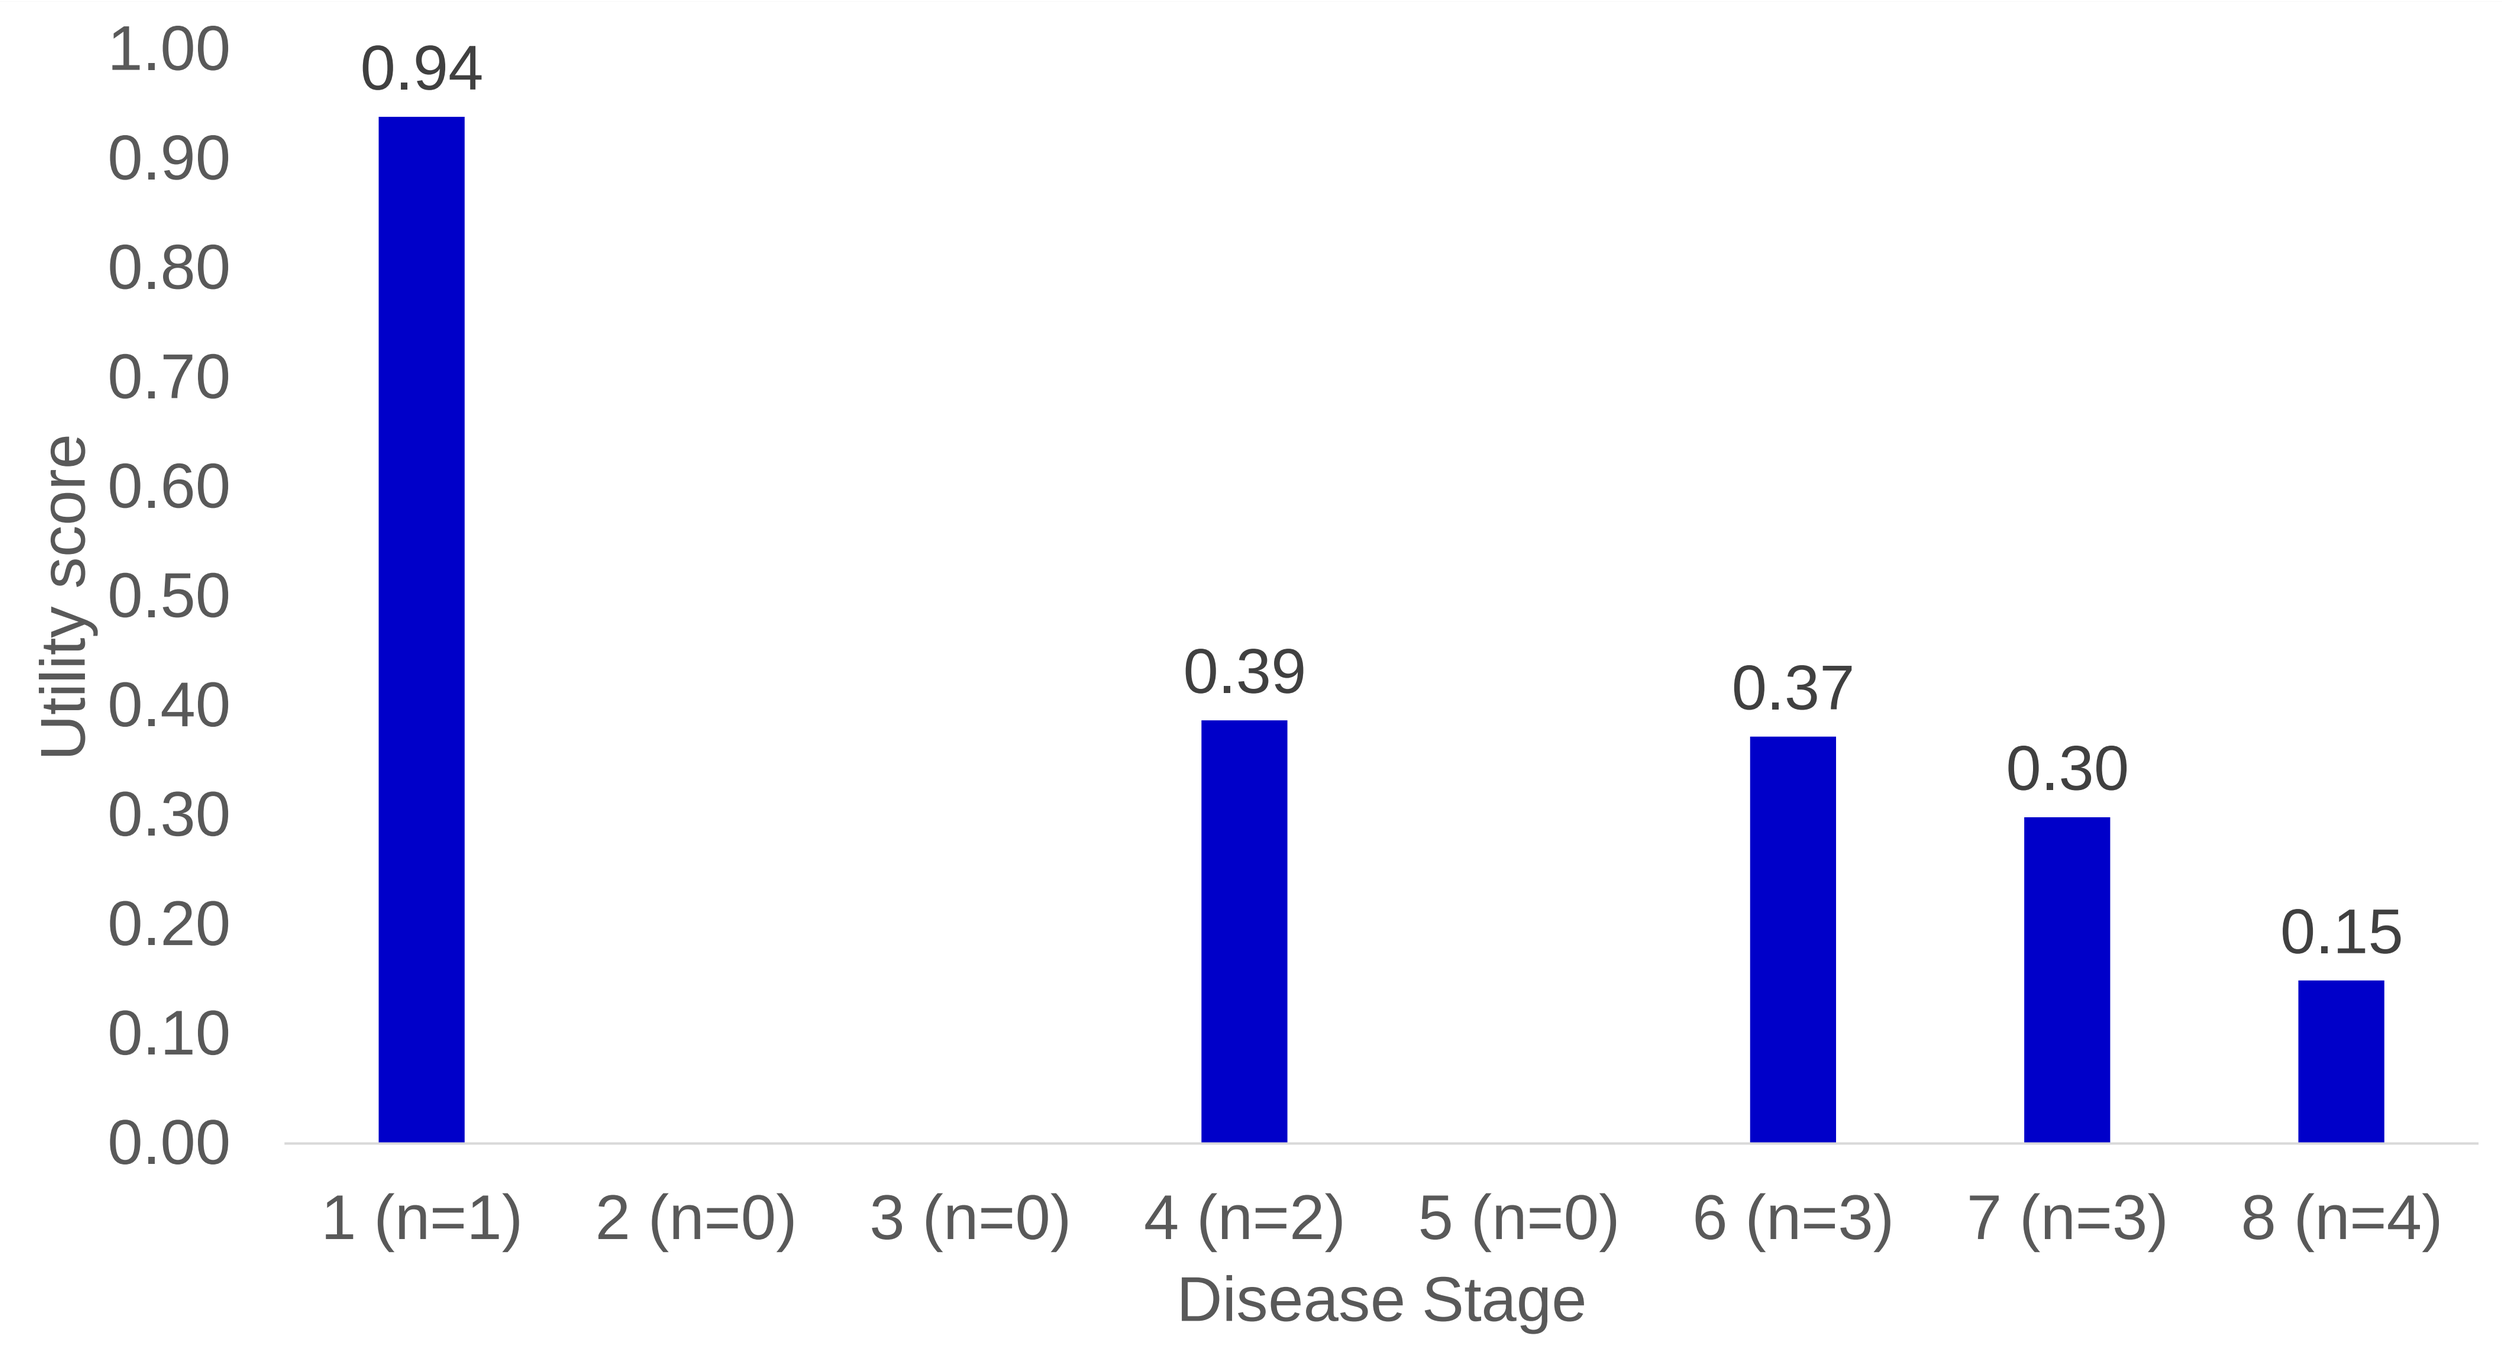
Utility score
1.00
0.90
0.80
0.70
0.60
0.50
0.40
0.30
0.20
0.10
0.00
0.94
0.39
0.37
0.30
0.15
1 (n=1)
2 (n=0)
3 (n=0)
4 (n=2)
5 (n=0)
6 (n=3)
7 (n=3)
8 (n=4)
Disease Stage

Table 2. Mean cost in the last 12 months by disease stage of DMD

Cost (¥) in the last 12 months	Disease Stage 1	2	3	4	5	6	7	8	Total
Mean direct medical	¥42,059	¥514,648	¥489,467	¥1,112,729	¥532,248	¥1,661,008	¥1,388,418	¥840,847	¥814,492
Mean direct non-medical	¥2,293,064	¥5,199,981	-	¥6,002,276	-	¥2,917,648	¥13,200,000	¥15,200,000	¥5,849,454
Mean indirect	¥330,315	¥1,278,513	-	¥964,627	-	¥2,720,240	¥2,076,450	¥3,143,534	¥1,136,346
Mean societal*	**¥2,665,438**	**¥6,993,142**	**-**	**¥8,079,632**	**-**	**¥7,298,896**	**¥16,664,868**	**¥19,184,381**	**¥7,800,292**

*estimated as the sum of the averages of each cost type
Abbreviations: DMD: Duchenne Muscular Dystrophy; SD: standard deviation

References: 1. Broomfield J, et al. Neurology. 2021 Dec 7; 97(23) **2.** TAIHO PHARMA press release Jan 5 **3.** Landfeldt E, et al. Neurology 2014 83, 529–536 . **4.** Broomfield J, et al.. Brain Behav. 2023 13(12): e3331

Disclosures: YMS has been a Sub-investigator in clinical trials for Pfizer, Sarepta Therapeutics, Nippon Shinyaku, Taiho Pharma, and PTC therapeutics; has been a principle investigator for Kaneka; and has received consultancy fees from Pfizer. VM, MN, and DP are employees of Pfizer. NP is an employee of Pfizer and owns stock in the company. JE is an employee of Prime HCD.