Estimating the socioeconomic burden of paroxysmal nocturnal hemoglobinuria (PNH): Results from Germany and Japan.

SA60

Ioannis Athanasiou[1], Vassilis Fragoulakis[1], Michel Kroes[2], Takeo Dochi[2], Yoshinobu Kanda[3], Yasmin Taylor[4], Niall Hatchell[4]

[1] WifOR Institute, Athens, Greece [2] Novartis Pharma K.K., Tokyo, Japan [3] Department of Medicine, Jichi Medical University, Tochigi, Japan, [4] Adelphi Real World, Bollington, United Kingdom

Background

- Paroxysmal nocturnal hemoglobinuria (PNH) is a rare, acquired, potentially fatal hematopoietic stem cell disorder. PNH imposes a significant socioeconomic burden characterized by fatigue and hemolysis, and is associated with impact on quality of life and ability to work and perform activities of daily living [1].
- C5 inhibitors (C5i; i.e., eculizumab and ravulizumab) have been the standard of care for PNH for the past decade. However, while C5i provide patients with a normal life expectancy, studies conducted on European and Asian populations have shown that PNH symptomatology does not resolve even under the long-term use of these treatments [2,3]. Data on the socio-economic burden of PNH are scarce and will help to determine the true impact of the disease in cross-country analyses.

Objectives

The aim of this study was to estimate the health and monetized socioeconomic burden, in terms of lost productivity, of adult patients with PNH treated with C5i in Germany and Japan.

Methodology

- An open-cohort, dynamic, three-state transition model was developed in in Microsoft Excel 365™ spreadsheet using Visual Basic for Applications (VBA), in a lifetime horizon (Figure 1).
- The primary model outcomes which linked to productivity loss were a) number of red blood cell transfusions, b) percentage of patients with moderate-to-severe fatigue, c) hospital days and d) time spent for C5i IV infusion. The model cycle was one year, the initial age of patients was 18, and the lifetime horizon was 64 (for Germany) and 67 (for Japan), reflecting average life expectancy. The probability of death for PNH patients on C5i was assumed to be the same as for the general population.
- Productivity losses were defined as hours of paid and unpaid work lost due to the modeled outcomes, calculated based on productivity impairment rates derived from the literature. The Human Capital Approach was used to value total productivity losses [4], while the replacement cost approach was used to value unpaid labor losses [5].
- Average wages in the German and Japanese economies were used to monetize the socioeconomic burden. Clinical data on model outcomes and patient distribution on C5i treatments were collected from the "Adelphi PNH Disease Specific Programme (DSP) 2022 dataset" [6-9]. Disease epidemiology and patient demographics were based on the International PNH Registry, expert opinion, and published literature [10–13]. Employment rates, hours worked, time spent on unpaid work, wage information, time spent for C5i IV infusion, and mortality rates for the general population were obtained from the Statistisches Bundesamt in Germany, Eurostat, the Statistics Bureau of Japan, the Ministry of Health, Labor and Welfare (MHLW) in Japan, country-specific Time Use Surveys and published literature [14–18].
- Costs referred to the year 2022 and results were presented undiscounted in accordance with the related international literature [19,20].
- Scenario analysis, one-way sensitivity analysis at ±20% parameter variation from the mean and 1,000 bootstrapping iterations were also performed to assess the robustness of results and to produce 95% confidence intervals (95% CI).

Figure 1. The three-state transition model

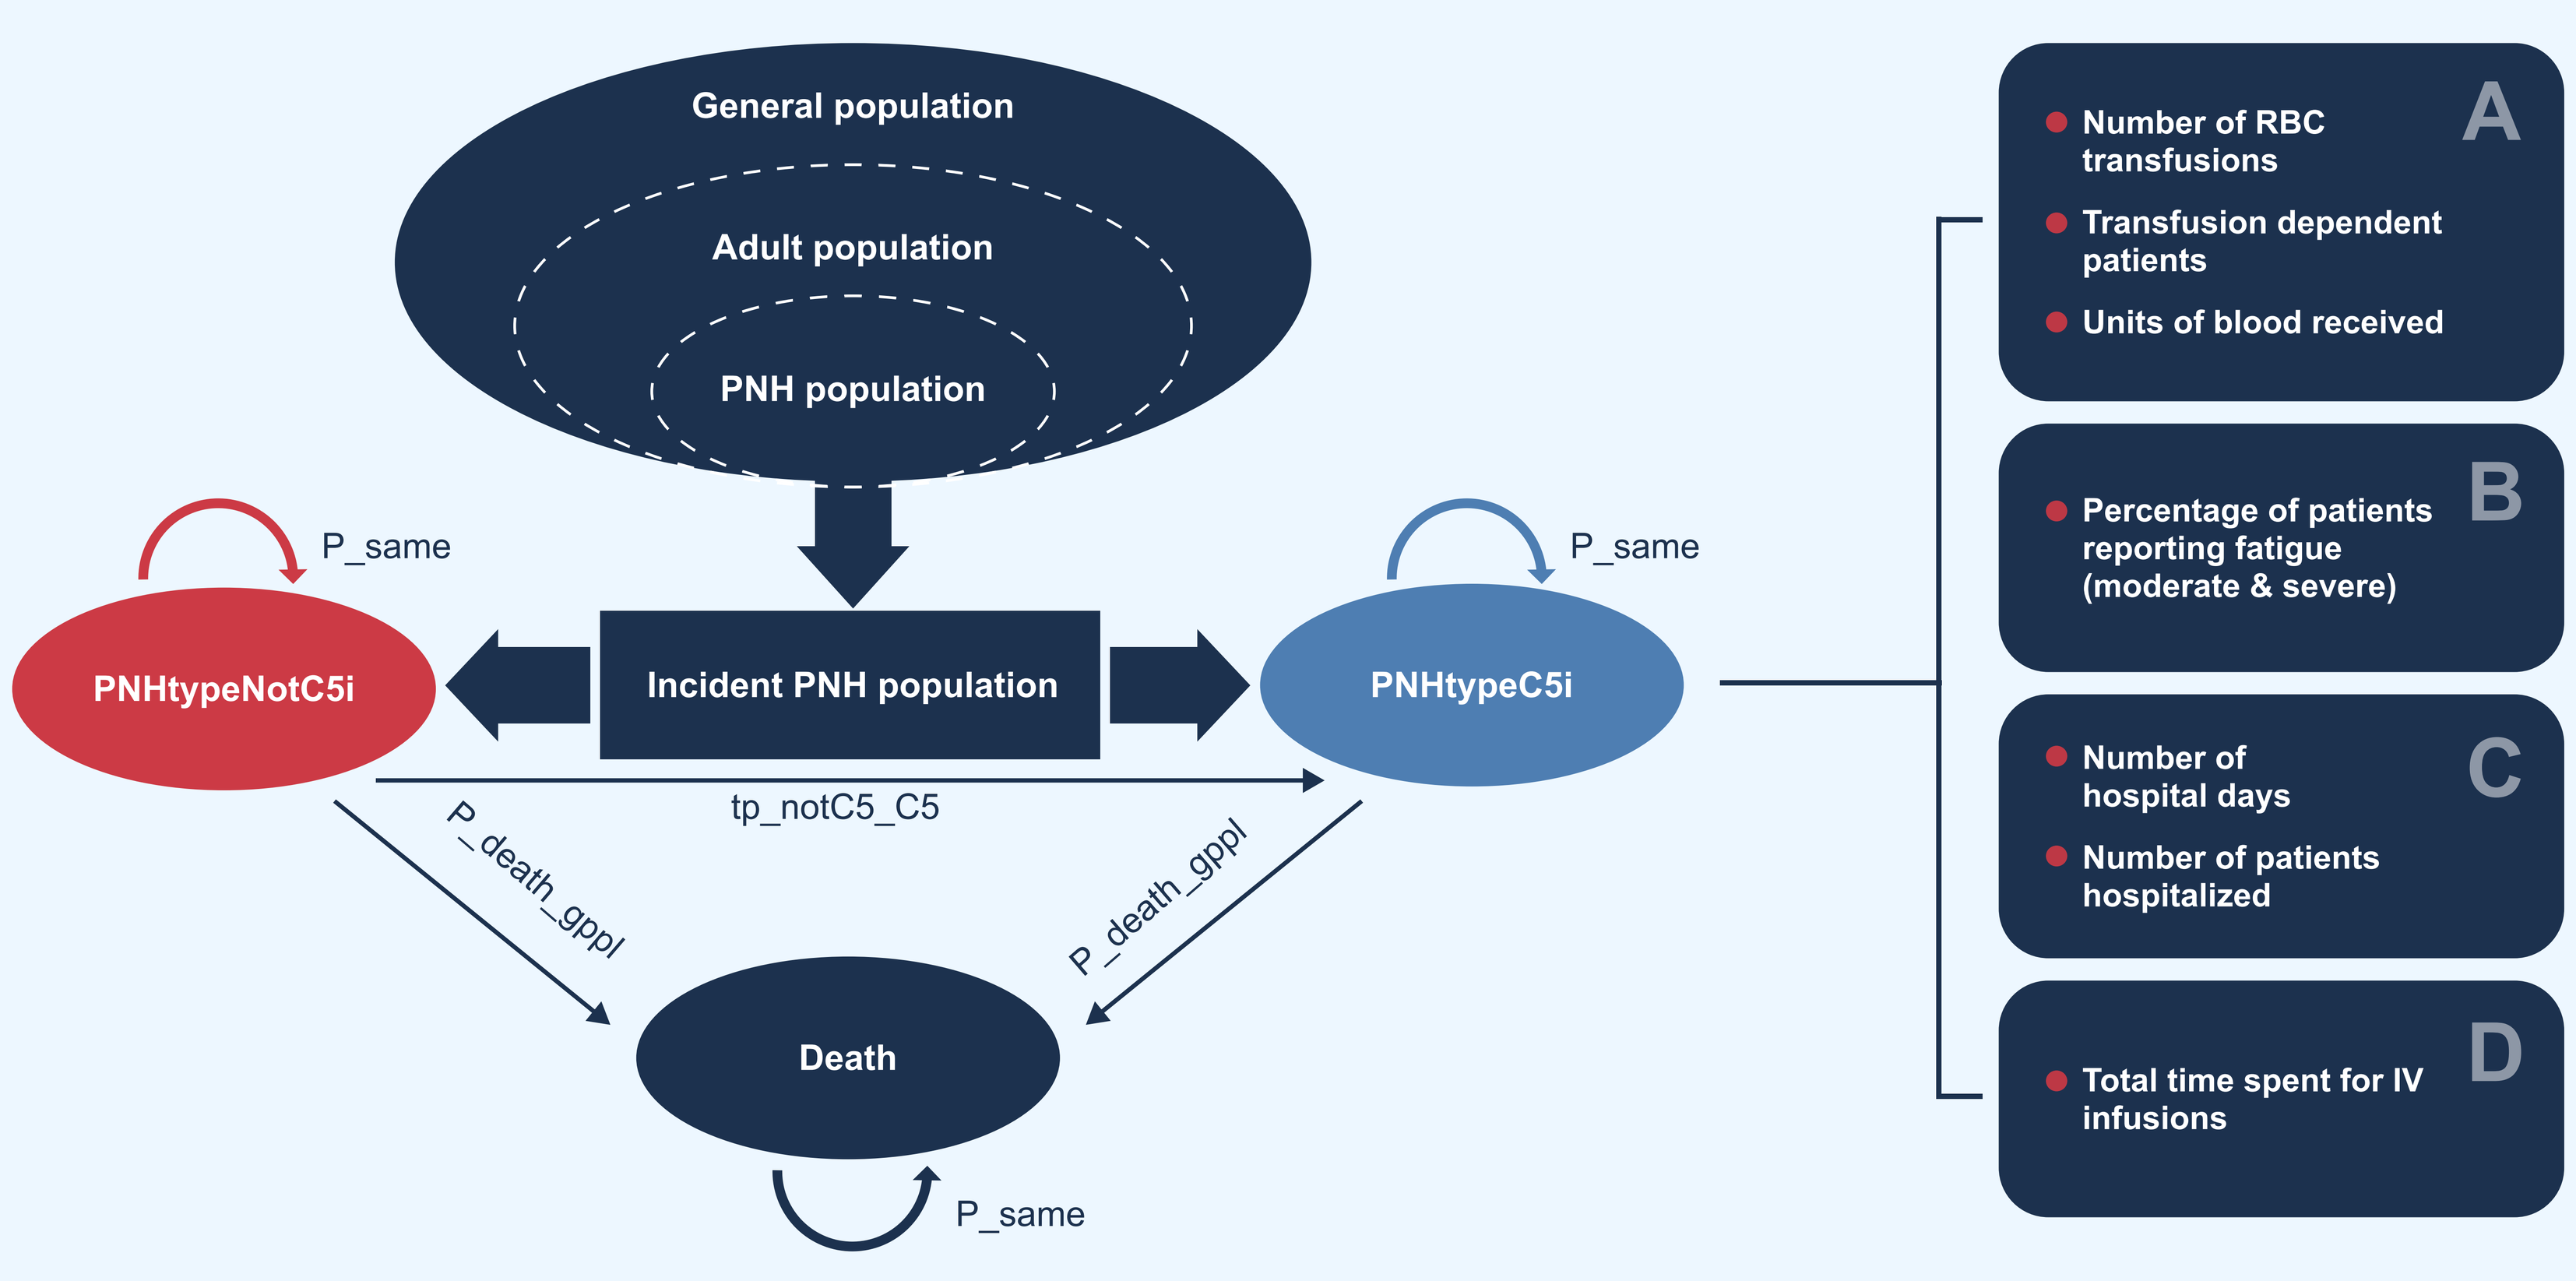
General population
Adult population
PNH population
P_same
PNHtypeNotC5i
Incident PNH population
PNHtypeC5i
P_same
tp_notC5_C5
P_death_gppl
P_death_gppl
Death
P_same
A
Number of RBC transfusions
Transfusion dependent patients
Units of blood received
B
Percentage of patients reporting fatigue (moderate & severe)
C
Number of hospital days
Number of patients hospitalized
D
Total time spent for IV infusions

__PNHtypeNotC5i:__ Patients having a PNH type which does not require systematic treatment with C5 inhibitors; __PNHtypeC5i:__ Patients having a PNH type which require systematic treatment with C5 inhibitors; __tp_notC5_C5:__ Transition probability of patients who do not require treatment to those who require treatment with C5 inhibitors; __P_death_gppl:__ General population probability of death

Results

- In 2023, there were an estimated 825 and 1,040 patients diagnosed with PNH who received C5i in Germany and Japan, respectively. According to the International PNH Registry, the mean age of patients in each country was 45.1 years (standard deviation: 18.1), and women were slightly more likely to have PNH than men (53% vs. 47%).
- The total monetized socioeconomic burden of PNH patients on C5i over the lifetime horizon was estimated at €813,9m (95% CI: €750-€930m.) and ¥242.6b (95%CI: ¥201-¥280b.) for Germany and Japan, respectively.
- The average per patient-year monetized burden was €3,890 (95% CI: €3,430-€4,365) and ¥210,178 (95% CI: ¥182,959-¥238,251) for Germany and Japan, respectively. Table 1 shows the health and socioeconomic burden for the whole population and per patient, per year.
- "Percentage of patients with moderate-to-severe fatigue" and "time spent for C5i IV infusion" were the major drivers of socioeconomic burden, accounting for 52.7% (19.6%) and 37.6% (56.5%) of the total monetized burden in Germany (Japan). Unpaid work losses represented 49.8% (42.6%) of the total productivity costs in Germany (Japan) (Figure 2, Figure 3).
- One-way sensitivity analysis indicated that fatigue and time spent for C5i IV infusion were the main factors influencing the annual monetized burden, in both countries (Figure 4, Figure 5).
- The probabilistic analysis results varied from -20.1% (-18.7%) to 9.8% (9.9%) as compared with the base case analysis in Germany (Japan).

Table 1. Burden of PNH patients on C5i in Germany and Japan, per year

	Estimated outcome	Total population, per year: Germany Mean (95% CI)	Total population, per year: Japan Mean (95% CI)	Per patient, per year: Germany Mean (95% CI)	Per patient, per year: Japan Mean (95% CI)
Health Burden	Number of RBC transfusions	113 (91-132)	285 (236-348)	1.48 (1.45-1.57)	1.22 (1.19-1.39)
	Number of RBC units transfused	199 (162-236)	756 (628-897)	2.61 (2.32-2.72)	3.16 (2.88-3.42)
	Hospitalizations	9 (8-10)	191 (162-224)	0.055 (0.054-0.063)	0.23 (0.22-0.26)
	Hospitalization days	54 (36-74)	1,438 (1,018-1,905)	6.05 (4.15-8.05)	7.52 (5.51-9.44)
	Patients reporting moderate-to- severe fatigue	57 (46-67)	58 (47-71)	0.35 (0.30-0.40)	0.07 (0.06-0.08)
	Time spent for C5i IV infusion (hours)	12,421 (9,732-15,143)	65,057 (51,506-78,233)	77 (64-93)	78 (63-92)
Socioeconomic Burden	Total paid work time lost (working months-Total population, working days-Per patient)	124 (98-152)	406 (321-508)	25.94 (23.88-28.87)	26.51 (24.31-28.98)
	Total unpaid work time lost (Working month- Total population, days-Per patient)	57 (47-67)	135 (113-157)	9.30 (8.76-10.42)	10.43 (9.58-11.80)
Monetized socioeconomic Burden	Total monetized socioeconomic burden	€644,792 (€531,538-€750,333)	¥182,440,534 (151,088,567-211,713,003)	€3,890 (€3,430-€4,365)	¥210,178 (¥182,959-¥238,251)

Figure 2. Paid and unpaid work losses as % of total monetized burden in Germany

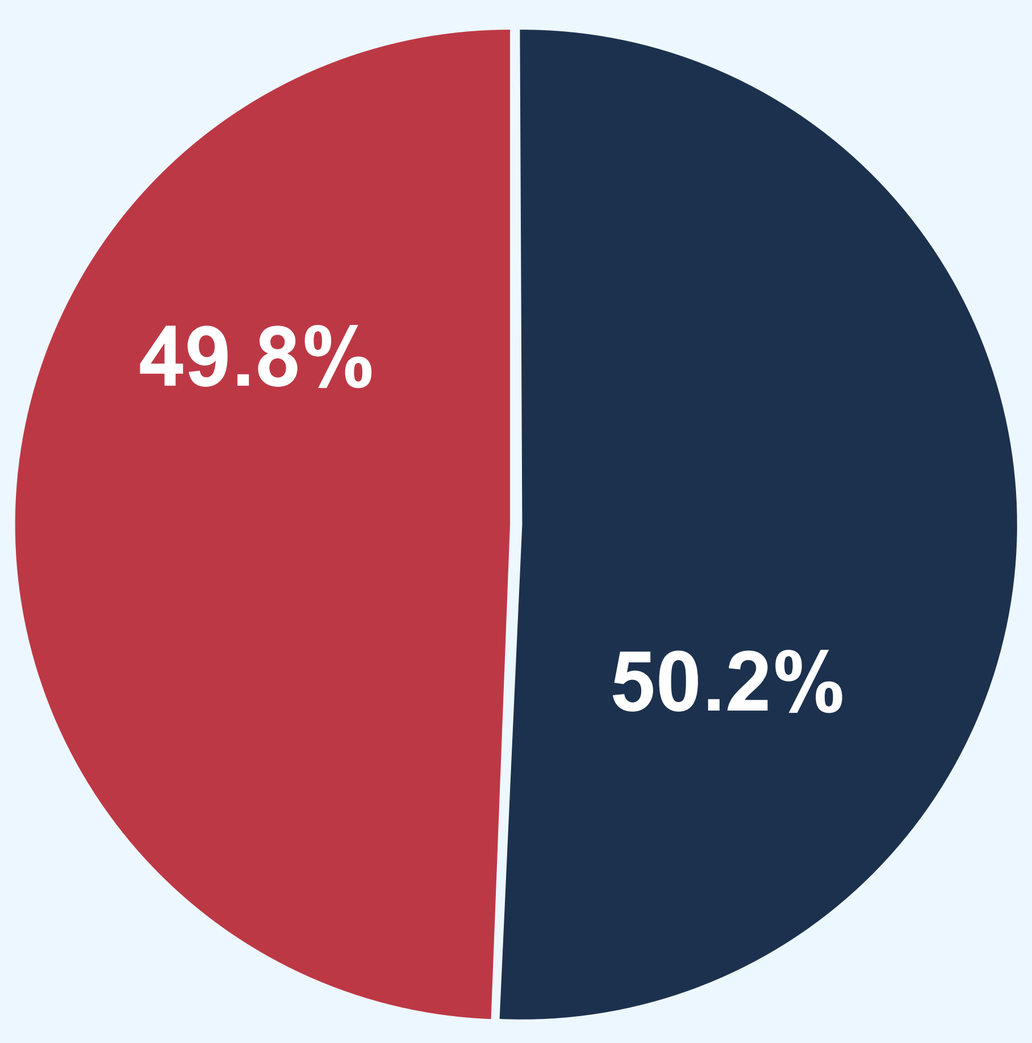
49.8%
50.2%

Figure 3. Paid and unpaid work losses as % of total monetized burden in Japan

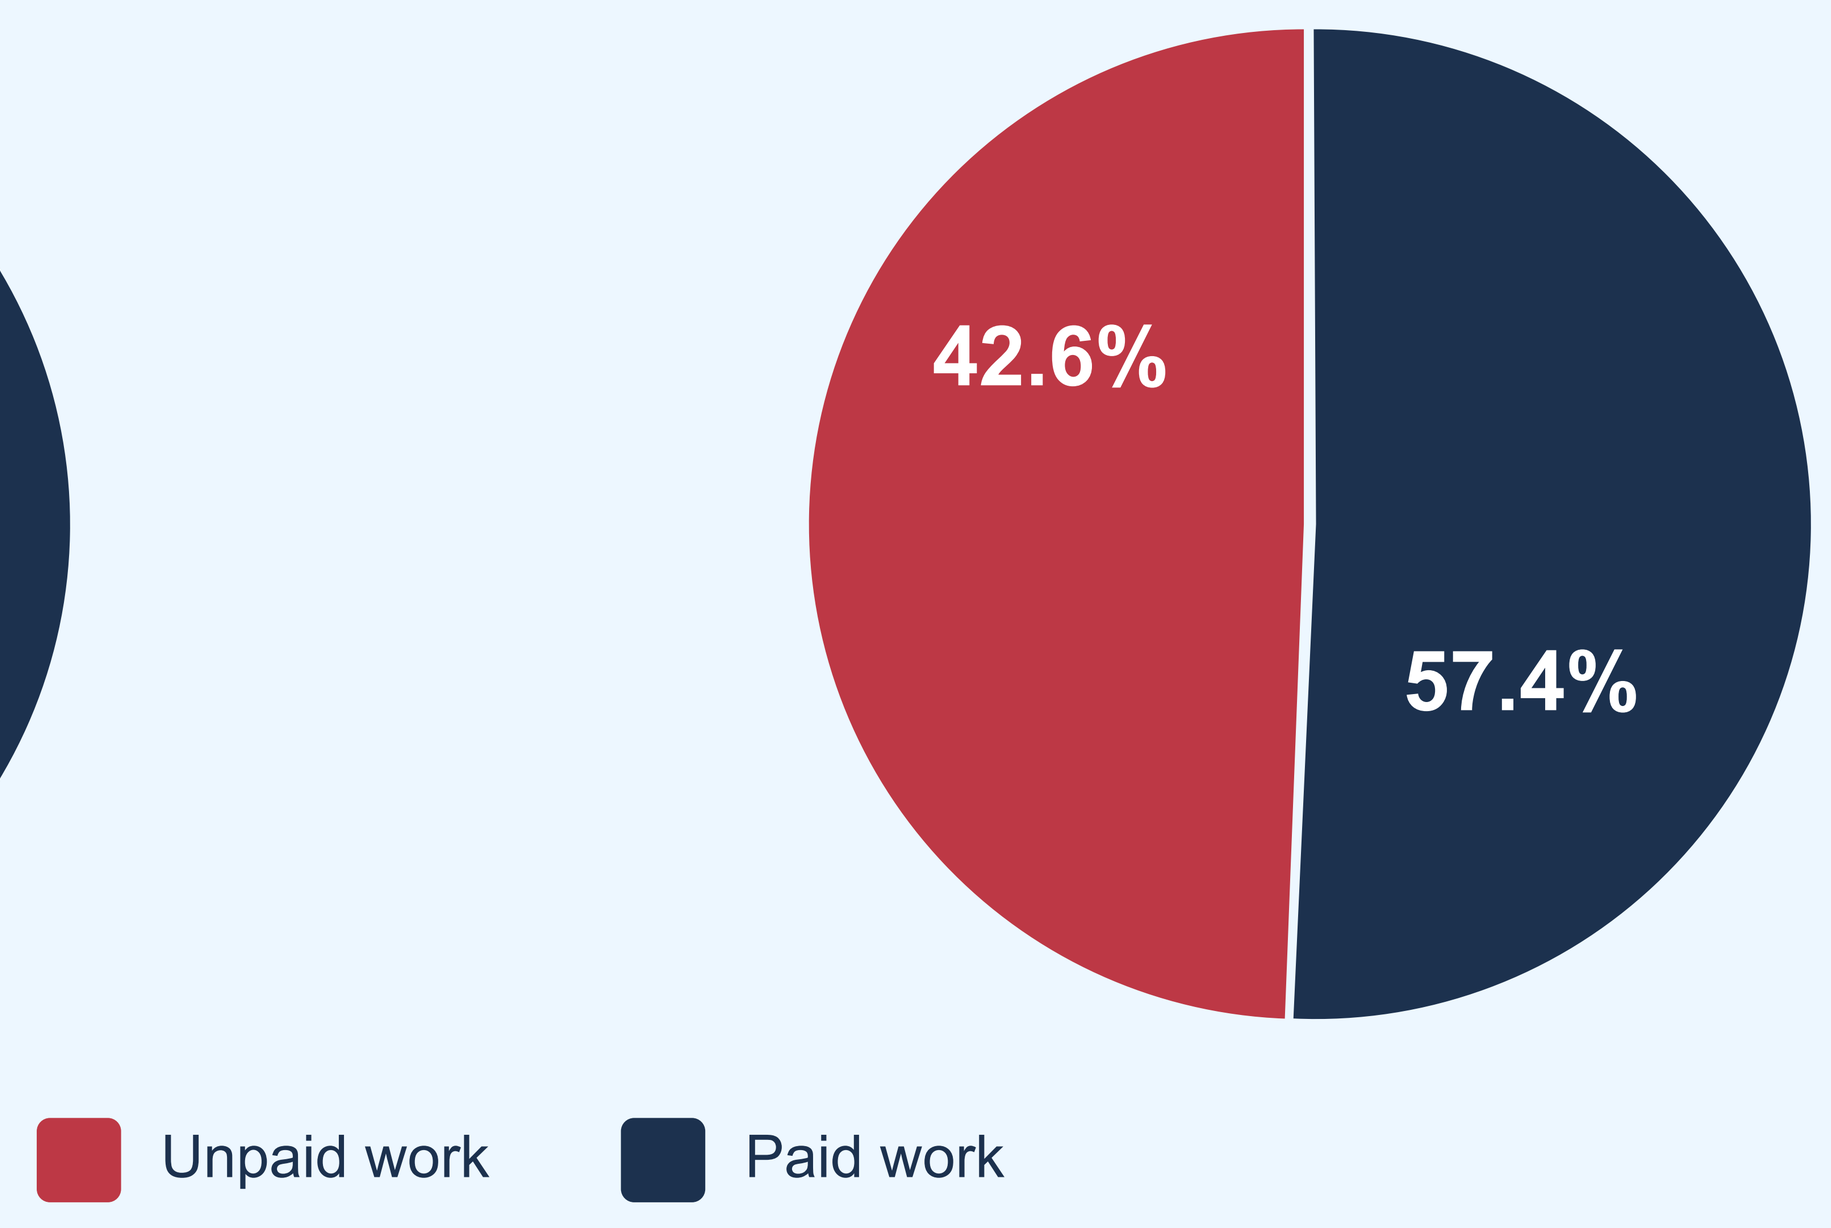
42.6%
57.4%
Unpaid work
Paid work

Figure 4. One-way Sensitivity Analysis per patient, per year, Germany

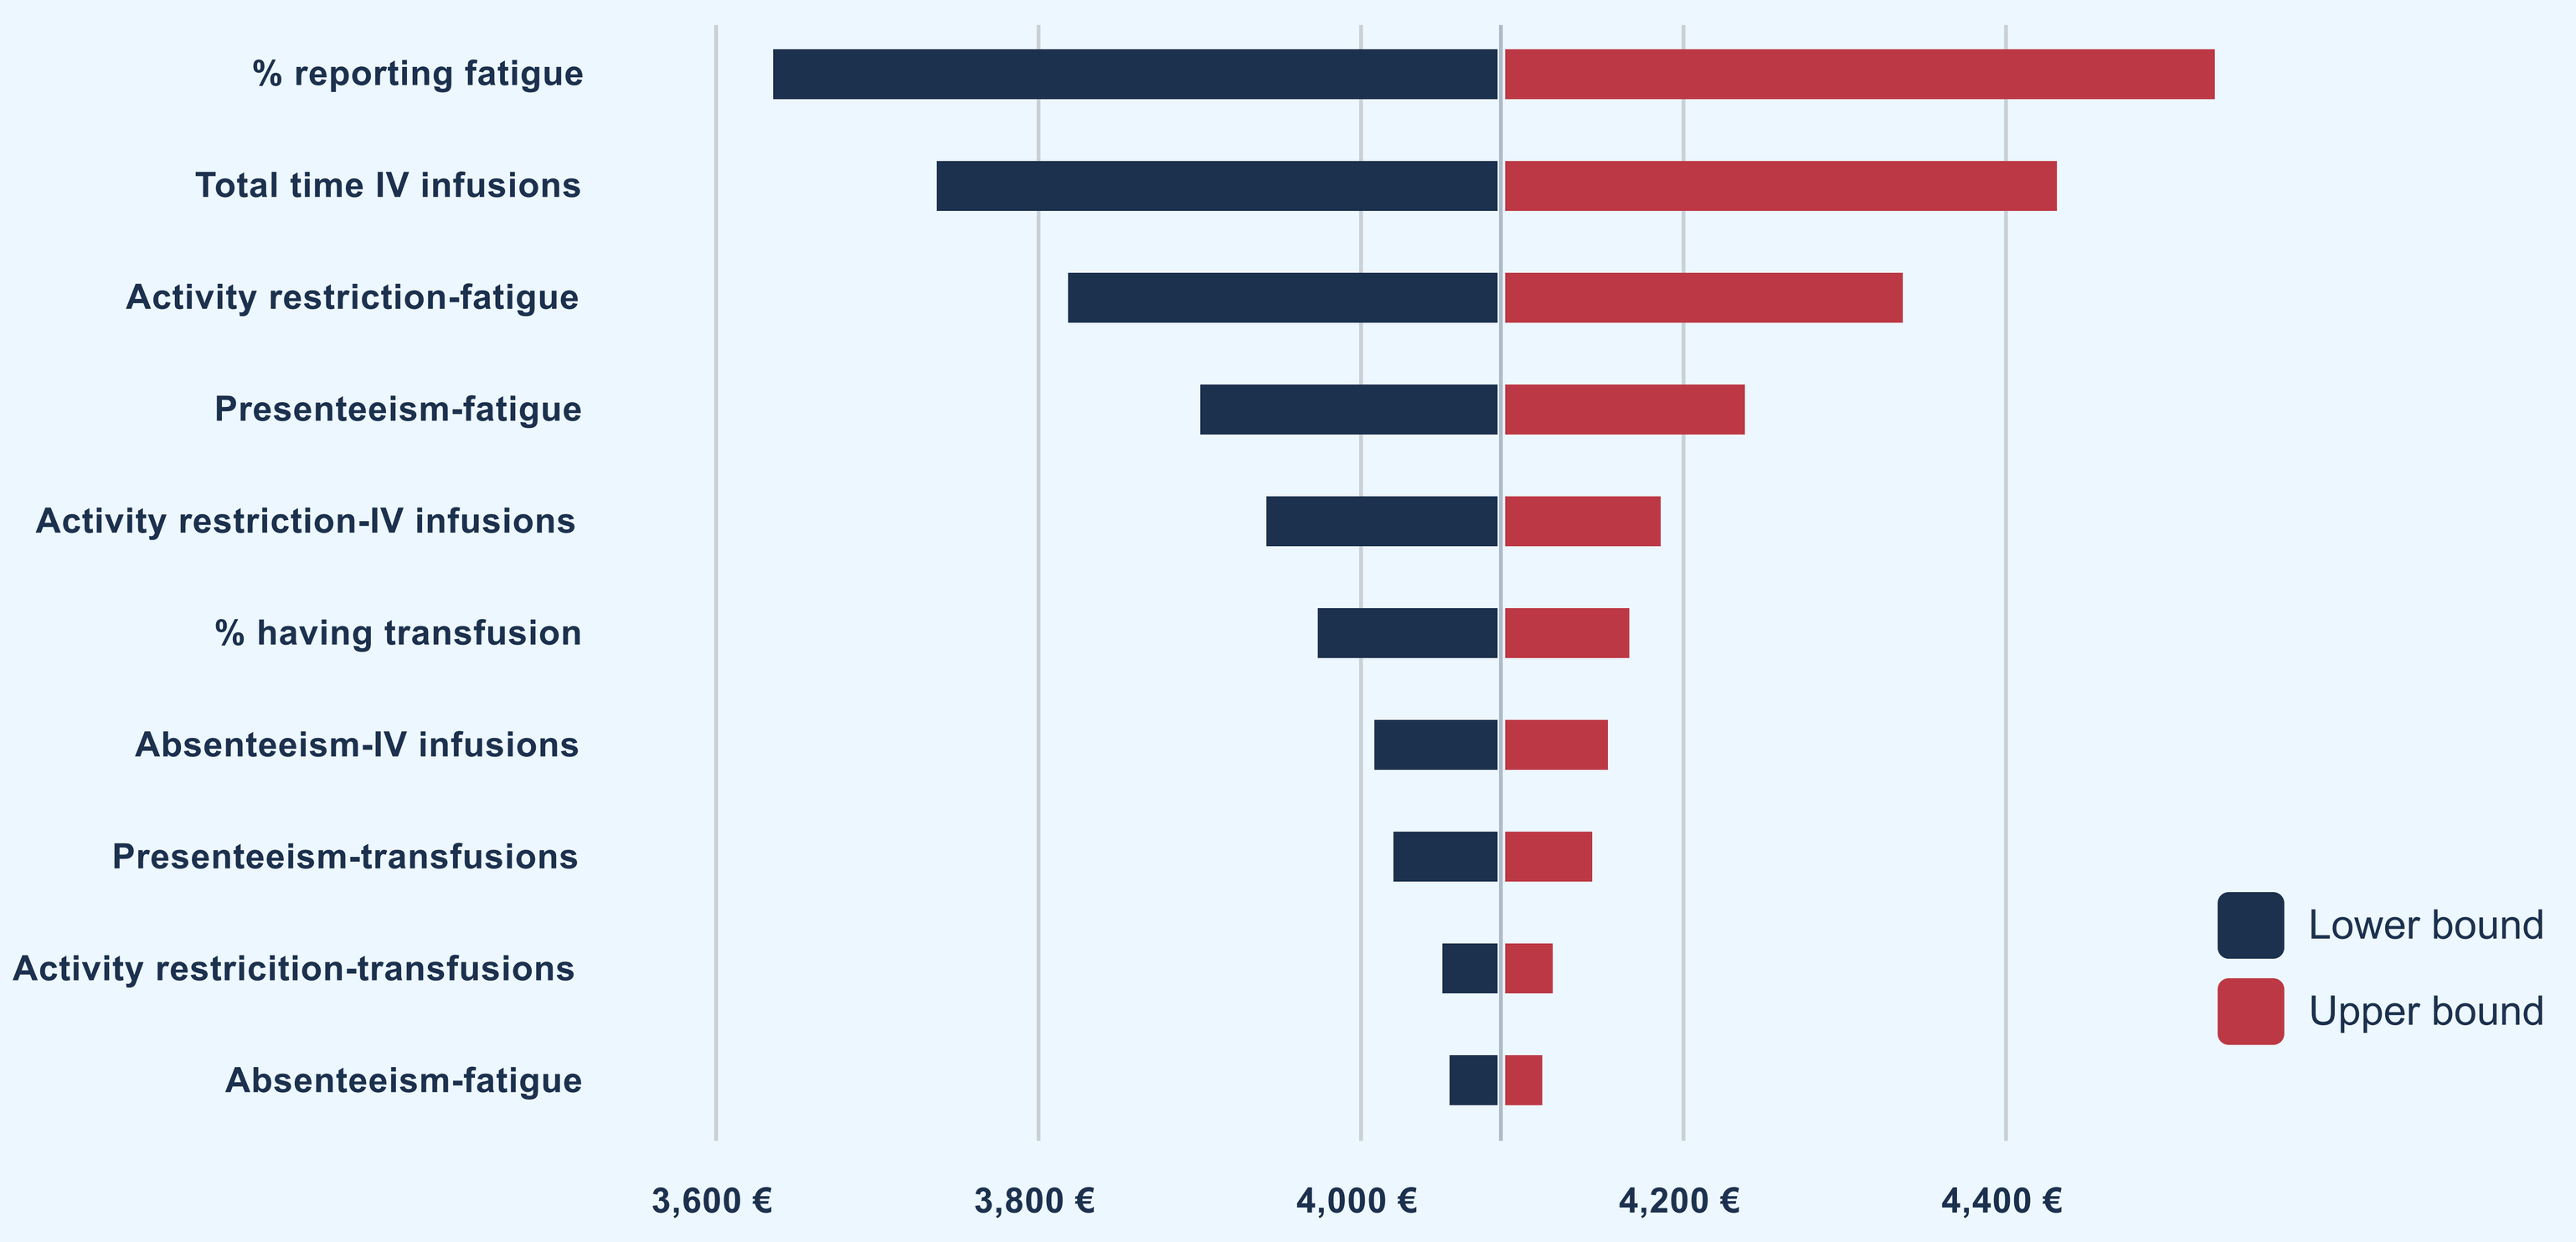
% reporting fatigue
Total time IV infusions
Activity restriction-fatigue
Presenteeism-fatigue
Activity restriction-IV infusions
% having transfusion
Absenteeism-IV infusions
Presenteeism-transfusions
Activity restricition-transfusions
Absenteeism-fatigue
3,600 €
3,800 €
4,000 €
4,200 €
4,400 €
Lower bound
Upper bound

Figure 5. One-way Sensitivity Analysis per patient, per year, Japan

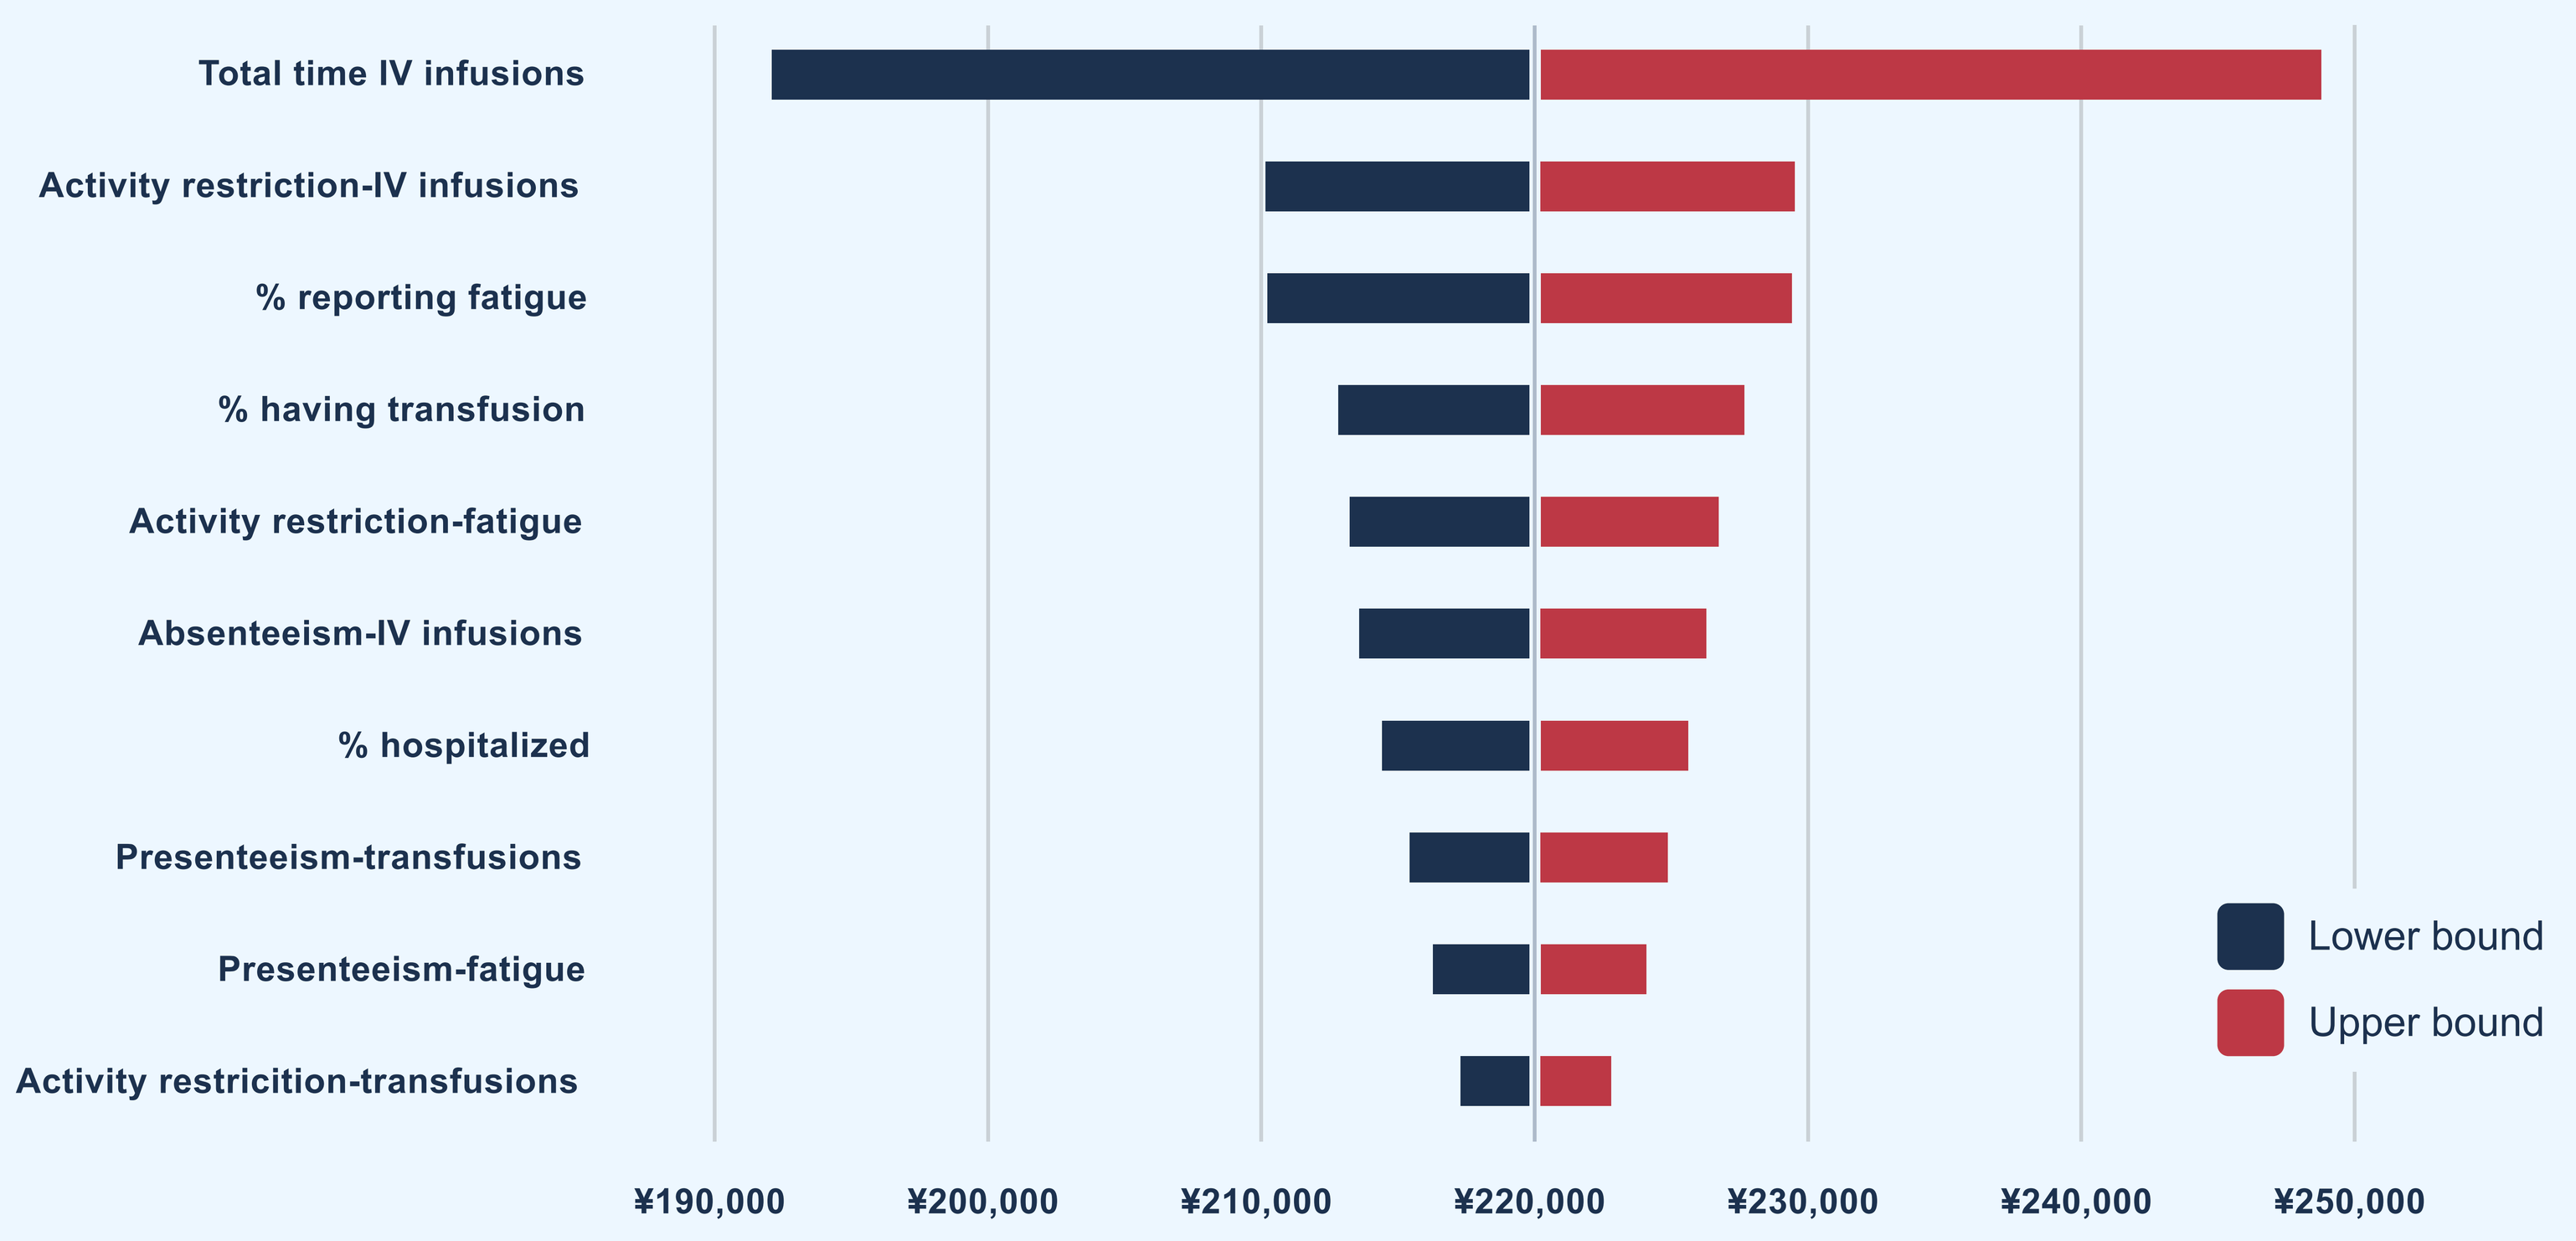
Total time IV infusions
Activity restriction-IV infusions
% reporting fatigue
% having transfusion
Activity restriction-fatigue
Absenteeism-IV infusions
% hospitalized
Presenteeism-transfusions
Presenteeism-fatigue
Activity restricition-transfusions
¥190,000
¥200,000
¥210,000
¥220,000
¥230,000
¥240,000
¥250,000
Lower bound
Upper bound

Discussion and conclusion

- The analysis focused on the productivity losses associated with PNH from the patient's perspective. It didn't take into account disease management costs, such as routine doctor visits and drug costs, or the burden on the healthcare system. The results should only be considered in the context of German and Japanese socio-economic factors and disease management guidelines.
- Our study underlines that even with the systematic use of C5i, there is still a significant burden of disease associated with PNH, highlighting the need for improved PNH therapies. Furthermore, the results emphasize the relevance of including unpaid work in the evaluation of burden of disease studies to help obtain a broader picture of the assessed impact. Alternative treatment options may be beneficial to enhance PNH patient outcomes and reduce humanistic and economic burden in Germany and Japan.

1. Bektas, M., Copley-Merriman, C., Khan, S., Sarda, S. P. & Shammo, J. M. Paroxysmal nocturnal hemoglobinuria: patient journey and burden of disease. J Manag Care Spec Pharm 26, S8–S14 (2020).
2. Panse, J. et al. The burden of illness of patients with paroxysmal nocturnal haemoglobinuria receiving C5 inhibitors in France, Germany and the United Kingdom: Patient-reported insights on symptoms and quality of life. European J of Haematology 109, 351–363 (2022).
3. Obara, N., Usuki, K., Hayashi, T., Fujii, M. & Ikezoe, T. Burden of illness in Japanese patients with paroxysmal nocturnal hemoglobinuria receiving C5 inhibitors. Int J Hematol 119, 255–264 (2024).
4. Human capital approach. in Encyclopedia of Public Health (ed. Kirch, W.) 697–698 (Springer Netherlands, Dordrecht, 2008). doi:10.1007/978-1-4020-5614-7_1583.
5. van den Berg, B. et al. Economic valuation of informal care: Lessons from the application of the opportunity costs and proxy good methods. Social Science & Medicine 62, 835–845 (2006).
6. Anderson P, Higgins V, Courcy J, Doslikova K, Davis VA, Karavali M, et al. Real-world evidence generation from patients, their caregivers and physicians supporting clinical, regulatory and guideline decisions: an update on Disease Specific Programmes. Curr Med Res Opin. 2023;39(12):1707-15.
7. Babineaux SM, Curtis B, Holbrook T, Milligan G, Piercy J. Evidence for validity of a national physician and patient-reported, cross-sectional survey in China and UK: the Disease Specific Programme. BMJ Open. 2016;6(8):e010352.
8. Anderson P, Benford M, Harris N, Karavali M, Piercy J. Real-world physician and patient behaviour across countries: Disease-Specific Programmes - a means to understand. Curr Med Res Opin. 2008;24(11):3063-72.
9. AHiggins V, Piercy J, Roughley A, Milligan G, Leith A, Siddall J, et al. Trends in medication use in patients with type 2 diabetes mellitus: a long-term view of real-world treatment between 2000 and 2015. Diabetes, Metabolic Syndrome and Obesity: Targets and Therapy. 2016;Volume 9:371-80.
10. Schrezenmeier, H. et al. Baseline clinical characteristics and disease burden in patients with paroxysmal nocturnal hemoglobinuria (PNH): updated analysis from the International PNH Registry. Ann Hematol 99, 1505–1514 (2020).
11. Dingli, D. et al. Relationship of paroxysmal nocturnal hemoglobinuria (PNH) granulocyte clone size to disease burden and risk of major vascular events in untreated patients: results from the International PNH Registry. Ann Hematol 102, 1637–1644 (2023).
12. Hill, A., DeZern, A. E., Kinoshita, T. & Brodsky, R. A. Paroxysmal nocturnal haemoglobinuria. Nat Rev Dis Primers3, 17028 (2017).
13. Farooq, Q., Saleem, M. W., Khan, Z. U. & Hadi, N. Paroxysmal Nocturnal Hemoglobinuria: A Diagnostic "Zero-Sum-Game". Cureus (2020) doi:10.7759/cureus.11956.
14. Eurostat. Time spent, participation time and participation rate in the main activity by sex and age group 2010 - tus_00age. (2022).
15. Eurostat. Average number of usual weekly hours of work in main job, by sex, age, professional status, full-time/part-time and occupation - 2021 – LFSQ_EWHUIS. (2022).
16. Ministry of Health, Labour and Welfare (MHLW). Basic Survey on Wage Structure. (2021).
17. Statistics Bureau of Japan. Annual Report on the Labour Force Survey 2020. https://www.stat.go.jp/english/data/roudou/report/2020/index.html (2020).
18. Levy, A. R. et al. Comparison of Lost Productivity Due to Eculizumab and Ravulizumab Treatments for Paroxysmal Nocturnal Hemoglobinuria in France, Germany, Italy, Russia, Spain, the United Kingdom, and the United States. Blood 134, 4803–4803 (2019).
19. Arrow, K. J. et al. Should Governments Use a Declining Discount Rate in Project Analysis? Review of Environmental Economics and Policy 8, 145–163 (2014).
20. Gollier, C. Discounting and Growth. American Economic Review 104, 534–537 (2014).

Acknowledgements: This study was commissioned and sponsored by Novartis.