A comparison of generalized risk adjusted cost-effectiveness analysis (GRACE) vs. traditional cost-effective analysis (CEA) estimates for the treatment of early-onset metachromatic leukodystrophy (MLD)

EE102

Ortega L[1], Incerti D[1], Bean K[2], Fields[2], Pang F[2], Lakdawalla D[3]

[1] EntityRisk Inc, Princeton, NJ, US; [2] Orchard Therapeutics, London, UK; [3] University of Southern California Schaeffer Center for Health Policy and Economics, CA, US

BACKGROUND & OBJECTIVES

- MLD is an ultra-rare and fatal inherited neurodegenerative disease, caused by a deficiency of the arylsulfatase A (ARSA) enzyme resulting in a build-up of toxic sulfatides. This leads to rapid motor and cognitive decline and premature death, particularly in early-onset MLD (late infantile (LI) and early juvenile (EJ)). The four forms of MLD are based on the age of symptom onset: late infantile (LI ≤ 30 months); early juvenile (EJ >30 months to <7 years); late juvenile (LJ ≥7 to 16 years), and finally adult onset (≥ 17 years).[1]
- Atidarsagene autotemcel (arsa-cel; OTL-200), an *ex vivo* autologous hematopoietic stem cell (HSC) gene therapy, is an effective disease modifying therapy that enables children with early-onset MLD to retain motor and cognitive function.[2]
- Traditional cost utility analyses (CUA) have shown substantial incremental QALY gains for arsa-cel vs. best supportive care;[3,4] however, the utility function underpinning traditional CUA does not incorporate risk preferences and uncertainty in outcomes. For example, a very sick patient may seek quality of life gains more desperately than more years of life in a bad state—but traditional CUA ignores this linkage.
- GRACE generalizes traditional CEA by accommodating any empirically valid utility function over health-related quality-of-life (HRQoL).

The aim of this study was to compare cost-effectiveness estimates from traditional CEA to those produced by GRACE for arsa-cel vs. best supportive care (BSC) for the treatment of early-onset MLD.

METHODS

- Traditional CEA and GRACE models were estimated from a US perspective for a combined MLD cohort encompassing pre-symptomatic late infantile (PS-LI: 51%) and early juvenile patients (PS-EJ: 23%), along with early symptomatic early juvenile patients (ES-EJ: 26%) based on the distribution of patients in the arsa-cel clinical trial programme.
- Non-negative HRQoL values initially proposed by the Institute of Clinical and Economic Review (ICER) during its appraisal of arsa-cel were used in the analysis rather than the utility values reported in the literature,[6] as the parametrization of the utility functions in the GRACE model have yet to be validated for health states worse than death.
- Estimates reported in Mulligan et al. (2024)[5] were used to parameterize expo-power and constant relative risk aversion (CRRA) GRACE utility functions over health.
- Figure 1 illustrates how the GRACE parameterized utility function compares to traditional CEA, where marginal improvements in utility values between 0.235 and 0.78 are valued more highly and therefore carry a greater weight than marginal gains in health-related quality of life (HRQoL) for relatively healthy treatment profiles (utility values >0.78) or for patients in very poor health states (utility values <0.235).
- This is in contrast to traditional CEA, which values marginal gains in HRQoL equally irrespective of the baseline health state, such that an improvement in utility value from 0.8 to 0.95 is considered equal to an improvement from 0.55 to 0.7. However, patients value HRQoL gains differently based on disease severity, and GRACE methodology is able to account for this.

Figure 1: Comparison of the parametrized expo-power utility function in the GRACE model to the utility function applied in traditional CEA

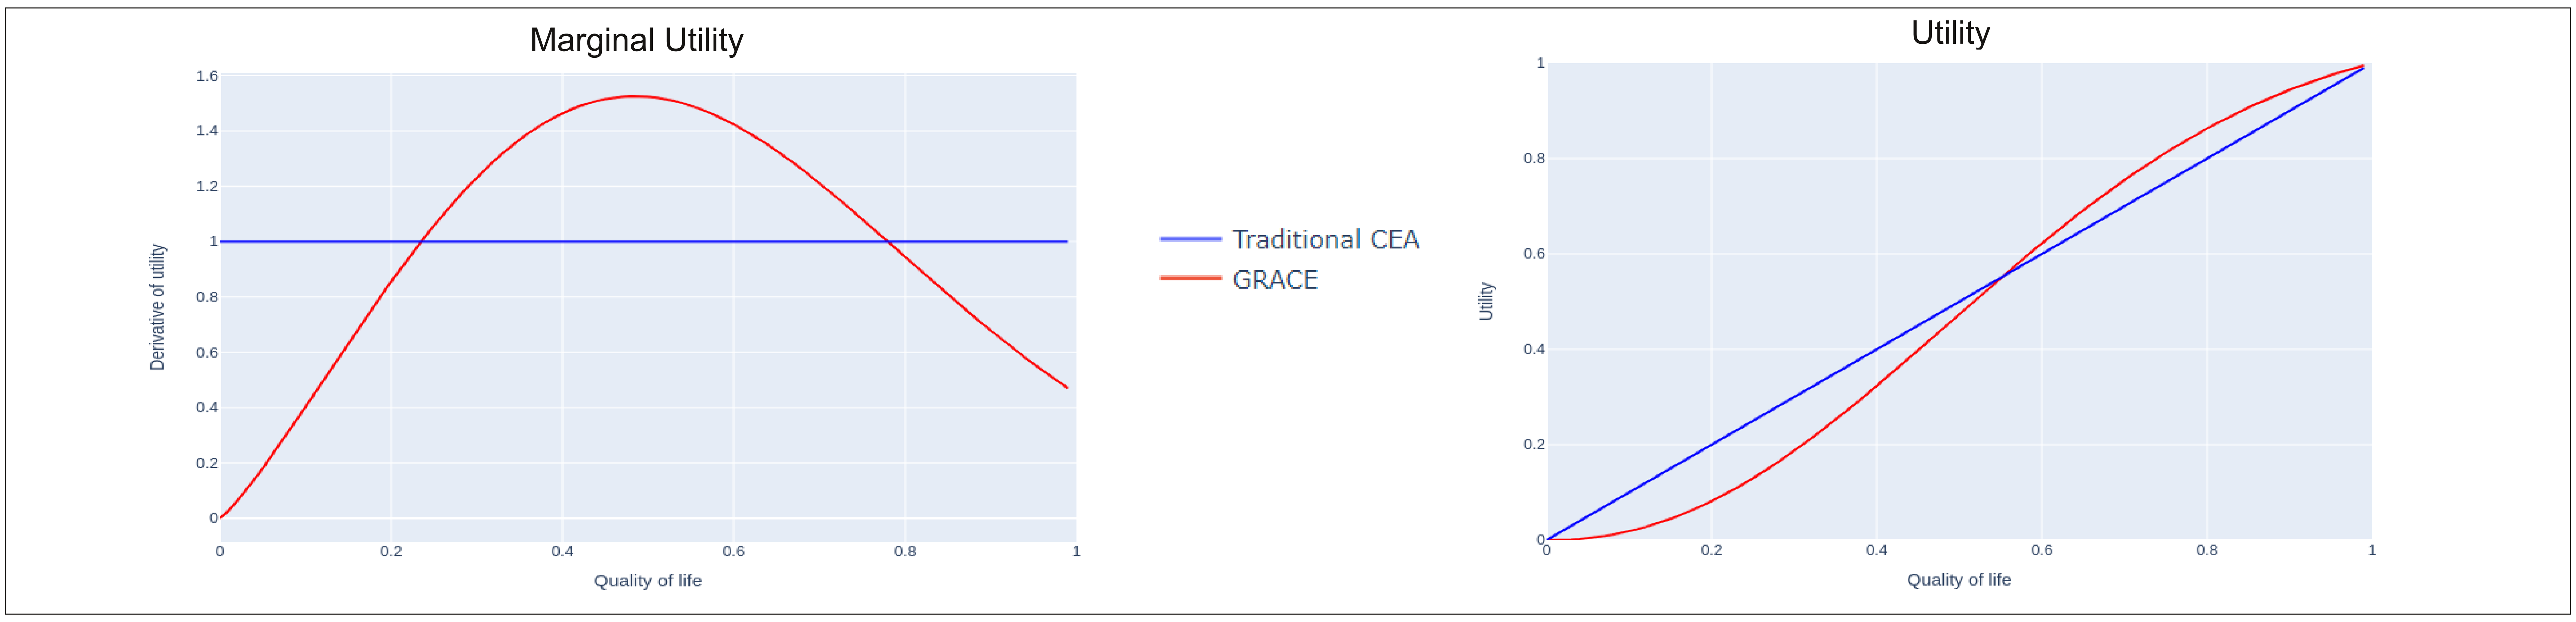
Marginal Utility
Derivative of utility
Quality of life
Traditional CEA
GRACE
Utility
Utility
Quality of life

- Traditional CEA is measured from the perspective of a consumer with "perfect" health in the baseline period. However, in reality individuals do not always have perfect health. By incorporating a disability adjustment allowing individuals to have less than perfect health at baseline, the marginal utility of consumption is lowered, which in turn makes gains in health more valuable. Consequently, analyses adjusted for the effect of pre-existing disability on willingness to pay for health were also performed.

RESULTS

- The incremental cost-effectiveness ratios (ICERs) and incremental QALYs for arsa-cel vs. best supportive care for the individual MLD phenotypes and the combined cohort from both the GRACE models and traditional CEA are presented In Table 1 below. As incremental costs between the two types of model are the same, these have not been included. Table 2 presents the results including the disability adjustment.

Table 1: Incremental QALYs and ICERS for arsa-cel vs BSC from GRACE and traditional CEA

Population	Traditional CEA	GRACE	Difference QALYs
Utility function: Expo-power			
PS-LI	20.01	20.34	0.33
PS-EJ	26.91	26.87	-0.04
ES-EJ	10.53	10.51	-0.02
Combined	19.13	19.29	0.16
Utility function: CRRA			
PS-LI	20.01	21.46	1.45
PS-EJ	26.91	26.68	-0.23
ES-EJ	10.53	11.36	0.83
Combined	19.13	20.04	0.91

Population	Traditional CEA	GRACE	Difference ICER
Utility function: Expo-power			
PS-LI	$114,502	$112,684	-$1,818
PS-EJ	$72,460	$72,551	$91
ES-EJ	$270,622	$271,071	$449
Combined	$123,247	$122,273	-$974
Utility function: CRRA			
PS-LI	$114,502	$106,784	-$7,718
PS-EJ	$72,460	$73,071	$611
ES-EJ	$270,622	$250,792	-$19,830
Combined	$123,247	$117,695	-$5,552

Table 2: Incremental QALYs and ICERS for arsa-cel vs. BSC including the disability adjustment

Population	Traditional CEA	GRACE	Difference QALYs
Utility function: Expo-power			
PS-LI	23.41	22.32	-1.09
PS-EJ	31.47	29.5	-1.97
ES-EJ	12.32	11.54	-0.78
Combined	22.38	21.17	-1.21
Utility function: CRRA			
PS-LI	23.41	24.01	0.6
PS-EJ	31.47	29.86	-1.61
ES-EJ	12.32	12.72	0.4
Combined	22.38	22.42	0.04

Population	Traditional CEA	GRACE	Difference ICER
Utility function: Expo-power			
PS-LI	$97,900	$102,647	$4,747
PS-EJ	$61,953	$66,089	$4,136
ES-EJ	$231,381	$246,927	$15,546
Combined	$105,376	$111,382	$6,006
Utility function: CRRA			
PS-LI	$97,900	$95,427	-$2,473
PS-EJ	$61,953	$65,299	$3,346
ES-EJ	$231,381	$224,119	-$7,262
Combined	$105,376	$105,177	-$199

DISCUSSION & CONCLUSION

- **This is one of the first applications of GRACE to an ultra-rare disease. In this instance, incremental QALYs and the cost-effectiveness estimates from the GRACE analysis were similar to those generated from traditional CEA after exploring several scenarios.**
- **The results suggest that economic valuations of arsa-cel are robust to generalizations of utility over quality of life beyond what is permitted by traditional CEA.**
- **The rough equality between GRACE and traditional CEA occurs because the GRACE utility function in Mulligan *et al.* (2024) implies higher values of health improvement for quality of life levels between 0.235 and 0.78, but lower values for quality of life below 0.235. Treatment with arsa-cel results in quality of life improvements that cover both these intervals, resulting in competing effects of GRACE on the value of arsa-cel. These effects roughly offset each other here.**
- **A future scenario will investigate the impact of negative HRQoL values in the GRACE framework.**

References

1. Fumagalli F *et al.* 2021. *J Inherit Metab Dis* 44(5):1151-1164
4. NICE HST18. March 2022. https://www.nice.org.uk/guidance/hst18

Libmeldy (atidarsagene autotemcel, OTL-200) received approval from the European Commission on 17 December 2020, in the UK on 1