CO6

Projected Public Health Benefits of Increasing Vaccine Coverage for COVID-19 Among Adults ≥50 Years of Age in the United States

Keya Joshi, Ekkehard Beck, Nicolas Van de Velde, Darshan Mehta*

Moderna, Inc., Cambridge, MA, USA

*Presenting author

BACKGROUND

- COVID-19 continues to cause substantial morbidity and mortality, especially in older adults in the United States. Compared to influenza, the cumulative hospitalization rate for COVID-19 for the 2023-2024 season was 166 vs 97 per 100,000 and 814 vs 217 per 100,000 in adults 50-64 and 65+, respectively[1,2]
- Despite the elevated burden of COVID-19 disease in older adults, COVID-19 vaccination coverage rates (VCR) continue to decline over time. COVID-19 VCR for the 2023-2024 season for adults 50-64 and 65+ were only 23% and 35% respectively,[3] compared to 30.1% and 43.3% during the 2022-2023 season[4]
- Childhood VCR have benefitted from the introduction of combination vaccines.[5] Multiple influenza and COVID-19 adult combination vaccines are in development and could increase COVID-19 VCR among adults
- We aimed to assess the projected public health impact of increasing COVID-19 VCR in adults ≥50 years of age due to the introduction of an adult combination COVID-19 and influenza vaccine

OBJECTIVE

- To evaluate the projected public health impact of increasing COVID-19 VCR in adults ≥50 years of age in the United States, compared to observed coverage for the 2023-2024 season, over a 1-year time horizon from September 2024 to August 2025

METHODS

Study Design

- Incidence of SARS-CoV-2 infections (eg., asymptomatic and symptomatic) was estimated using a previously published Susceptible-Exposed-Infected-Recovered (SEIR) dynamic transmission model[6] (**Figure 1**). Due to changes in reporting and testing over time, incidence of symptomatic infection was further calibrated to align with observed COVID-19 hospitalization rates from the 2023-2024 season from COVID-NET[7]
- Outcomes associated with symptomatic SARS-CoV-2 infections were estimated using a previously published static decision-analytic model[8] (**Figure 2**)
- Baseline age-specific vaccination rates for adults ≥50 years of age for the 2023-2024 season were taken from the Centers for Disease Control and Prevention (CDC) COVID-19 Vaccination Dashboard (COVID VaxView)[3]
 - The additional public health benefits of increasing baseline VCR by an absolute 1, 5, and 10 percentage points, to match observed flu vaccination rates from the 2023-2024 season and to match the WHO-defined 70% healthy people target, compared to no vaccination, was assessed. **Table 1** provides baseline and increased coverage rates
- In the absence of 2024-2025 vaccine effectiveness (VE) for Spikevax we assumed that both the Spikevax Fall 2023 (XBB1.5) 2023-2024 season and Spikevax 2024-2025 season vaccines are similarly well matched against the circulating strains during each season. **Table 2** provides key model input parameters
 - Initial VE estimates informed by real-world evidence (RWE) analysis of Omicron XBB1.5 containing mRNA COVID-19 vaccination using Veradigm EHR and Komodo claims data from September – December 2023[9]
 - Monthly waning of vaccines assumed to be the same as original monovalent vaccines against BA.1 using a systematic literature review and meta regression[10]
- The static model predicted the occurrence of COVID-19–related disease outcomes including SARS-Cov-2–associated symptomatic infections, COVID-19–related hospitalizations, deaths, and cases of long COVID for the 2024-2025 season from September 2024 to August 2025
- Key epidemiology and disease burden input parameters were based on US data for the 2023-2024 season (**Table 2**). Disease dynamics for the 2024-2025 season were assumed to follow similar trends

Model Inputs

Table 1. COVID-19 Baseline Coverage and Increased Vaccine Coverage Scenarios

Age group	Baseline COVID-19 VCR[a]	+1% VCR	+5% VCR	+10% VCR	Flu VCR[b]	70% Healthy People target
50-64 years	24.96%	25.96%	29.96%	34.96%	49.10%	70.00%
65-74 years	39.96%	40.96%	44.96%	49.96%	67.60%	70.00%
75-84 years	41.60%	42.60%	46.60%	51.60%	73.70%	70.00%
85+ years	41.60%	42.60%	46.60%	51.60%	73.70%	70.00%

VCR, vaccine coverage rate.

[a]https://www.cdc.gov/vaccines/imz-managers/coverage/covidvaxview/interactive/adults.html
[b]https://www.cdc.gov/flu/fluvaxview/dashboard/vaccination-adult-coverage.html

Table 2. Key Input Parameters

Parameter	Value
Initial vaccine effectiveness	
Infection[a,9]	
50-64 years	35.3%
65+ years	38.7%
Hospitalization[9]	
50-64 years	61.1%
65+ years	60.5%
Monthly waning of vaccine effectiveness[10]	
Infection	4.75%
Hospitalization	1.37%
Hospitalization rate[9,11]	
50-64 years	1.90%
65-74 years	8.50%
75-84 years	15.75%
85+ years	23.50%
In-hospital mortality[12]	
50-64 years	3.37%
65-74 years	4.97%
75-84 years	5.27%
85+ years	5.27%

[a]Vaccine effectiveness against medically attended COVID-19 infections used as a proxy for COVID-19 infections.

Figure 1. Susceptible-Exposed-Infectious-Recovered (SEIR) Dynamic Transmission Model Structure

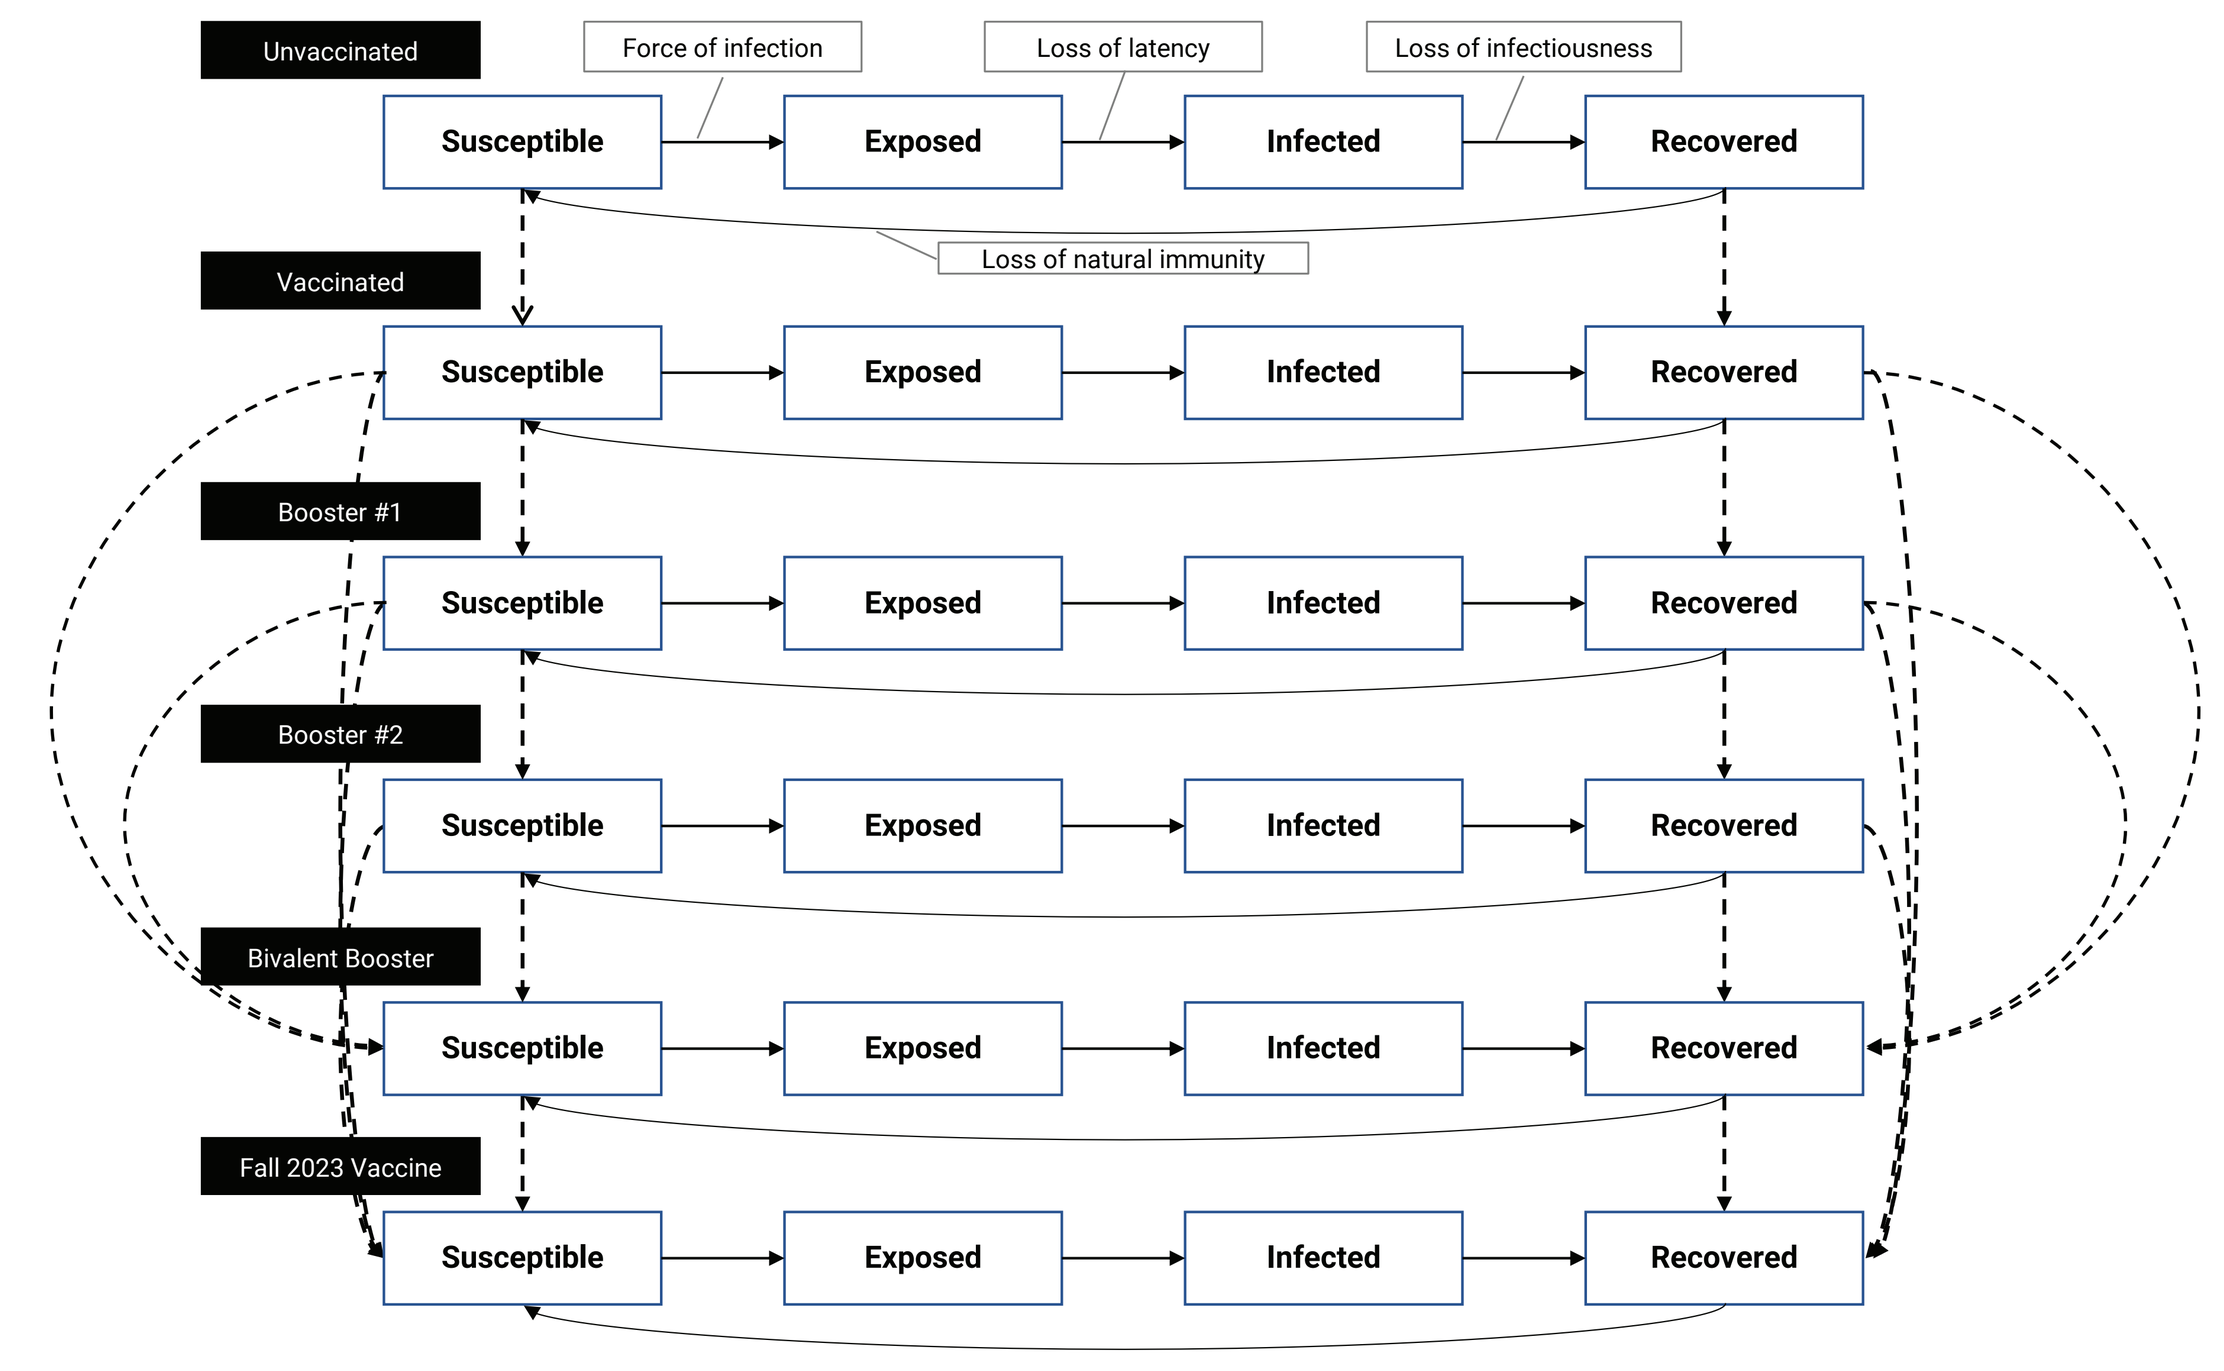

Unvaccinated
Force of infection
Loss of latency
Loss of infectiousness
Loss of natural immunity
Vaccinated
Booster #1
Booster #2
Bivalent Booster
Fall 2023 Vaccine
Susceptible
Exposed
Infected
Recovered

Figure 2. (A) Static Model Structure and (B) Infection Consequence Decision Tree Structure

A)

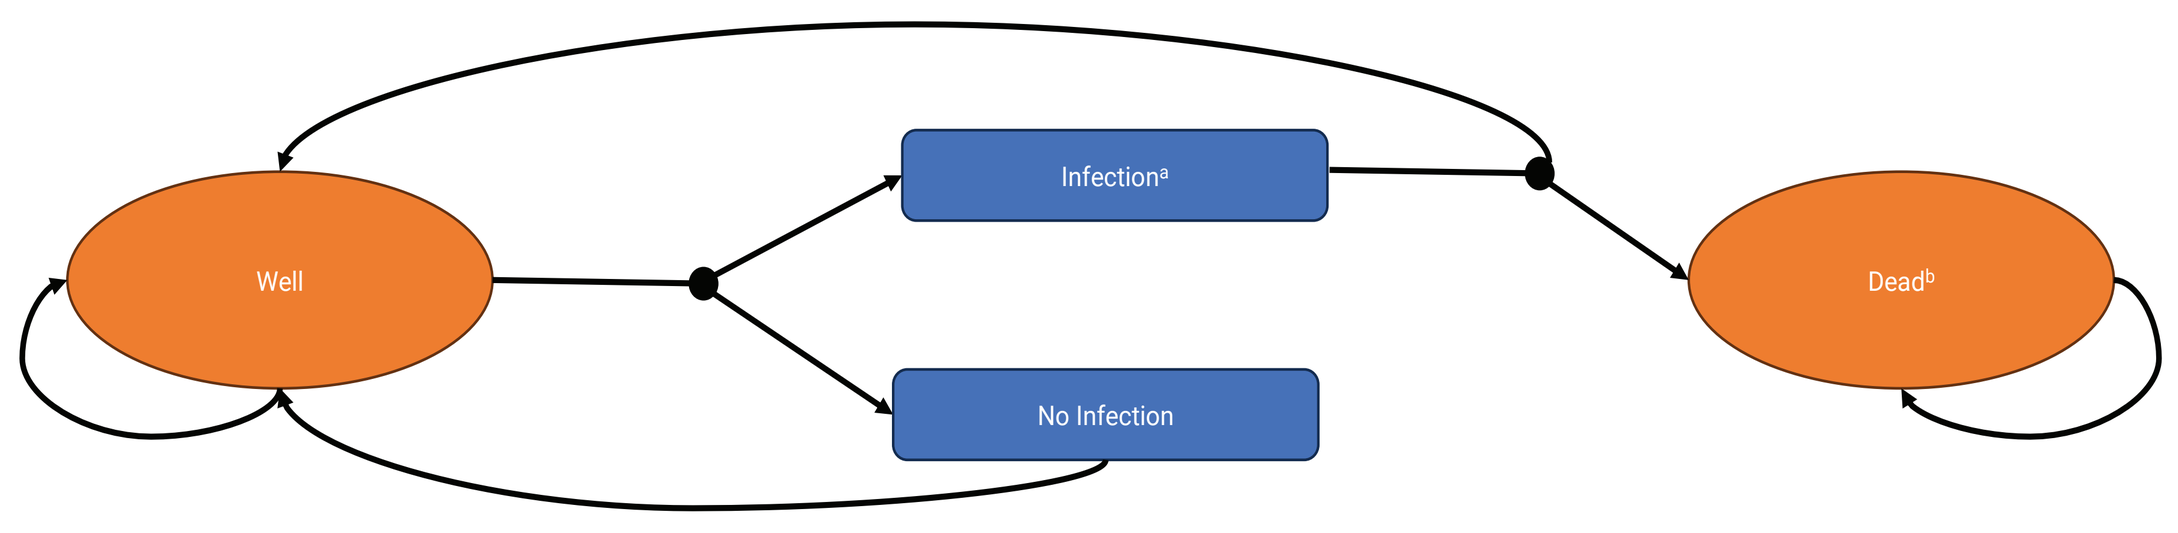

Well
Infection[a]
No Infection
Dead[b]

B)

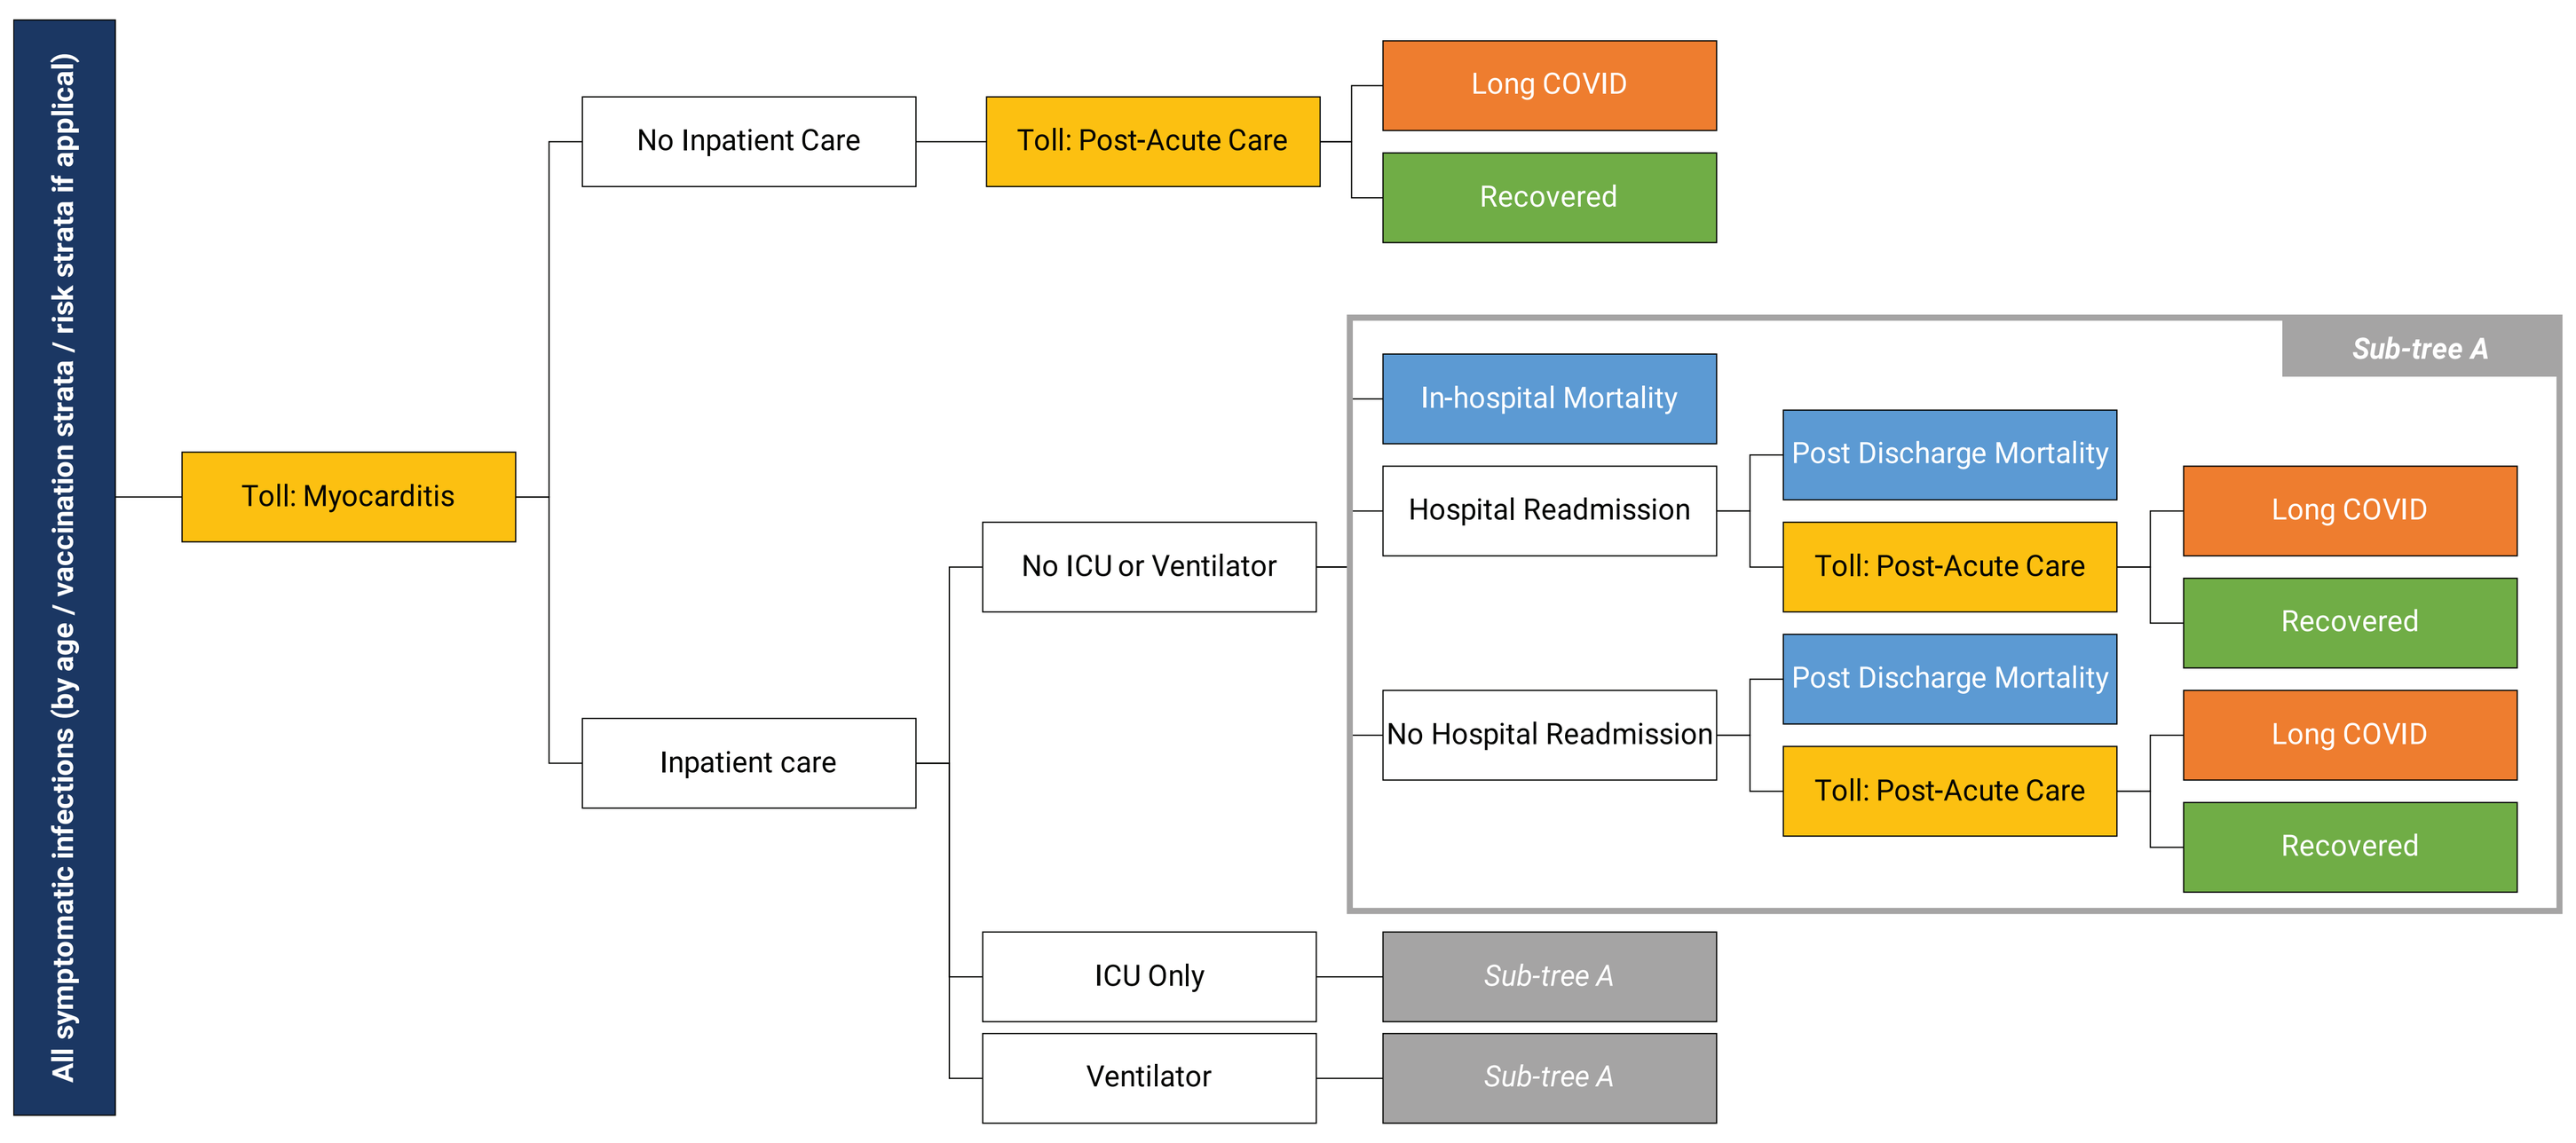

All symptomatic infections (by age / vaccination strata / risk strata if applical)
Toll: Myocarditis
No Inpatient Care
Toll: Post-Acute Care
Long COVID
Recovered
Inpatient care
No ICU or Ventilator
ICU Only
Ventilator
Sub-tree A
In-hospital Mortality
Hospital Readmission
No Hospital Readmission
Post Discharge Mortality
Toll: Post-Acute Care
Long COVID
Recovered

[a]See infection consequence decision tree structure (**Figure 2B**).
[b]Death due to COVID-19.

RESULTS

- The model predicted that, when the COVID-19 VCR was increased by 5% and 10% per year, there would be an additional 85,635 to 171,270 SARS-CoV-2 symptomatic infections averted; 13,562 to 27,123 of COVID-19–related hospitalizations averted; 1,494 to 2,987 of COVID-19–related deaths averted; and 24,664 to 49,328 long COVID-19 cases averted (**Figure 3**)
- When the COVID-19 VCR matched the observed influenza VCR, the model predicted there would be an additional 451,483 SARS-CoV-2 infections averted; 78,625 COVID-19–related hospitalizations averted; 8687 COVID-19–related deaths averted; and 127,824 long COVID cases averted (**Figure 3**)

Figure 3. Incremental Number of SARS-CoV-2 Symptomatic Infections, Long COVID Cases, COVID-19–Related Hospitalizations and Deaths Averted With Varying Levels of COVID-19 VCR Increase

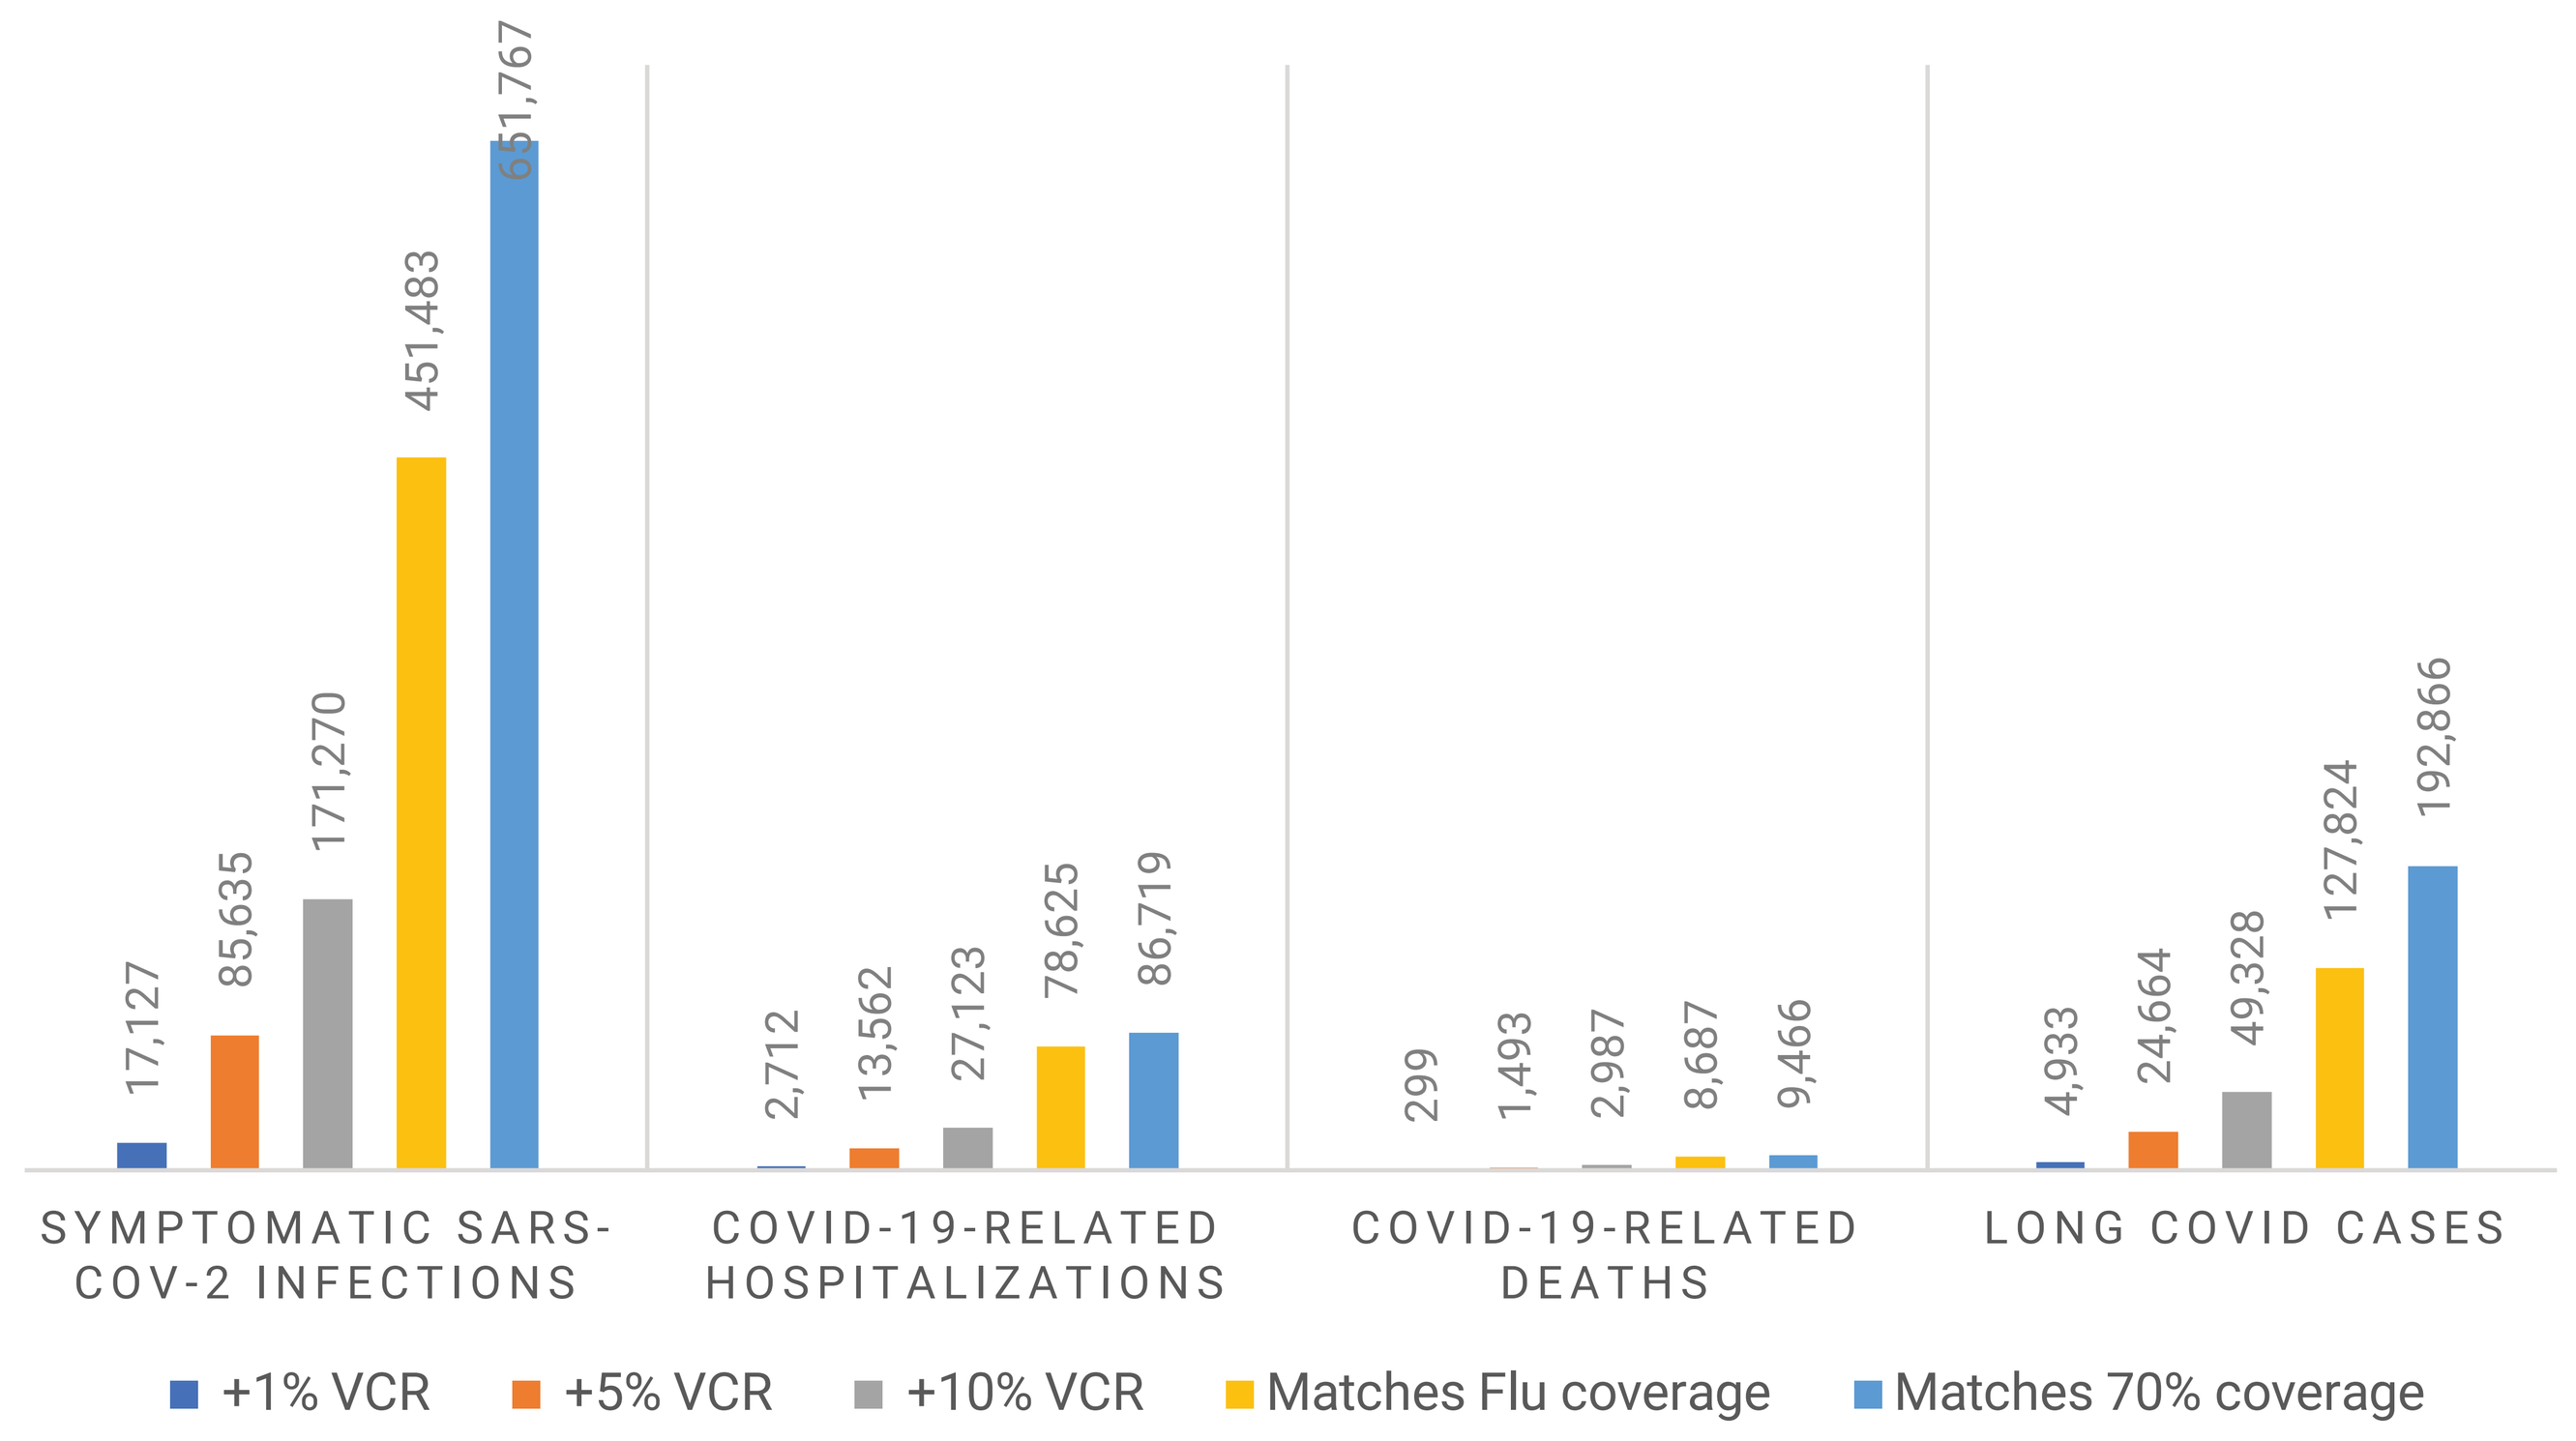

17,127
85,635
171,270
451,483
651,767
2,712
13,562
27,123
78,625
86,719
299
1,493
2,987
8,687
9,466
4,933
24,664
49,328
127,824
192,866
SYMPTOMATIC SARS-COV-2 INFECTIONS
COVID-19-RELATED HOSPITALIZATIONS
COVID-19-RELATED DEATHS
LONG COVID CASES
+1% VCR
+5% VCR
+10% VCR
Matches Flu coverage
Matches 70% coverage

VCR, vaccine coverage rate.

CONCLUSIONS

- Results from this modelling analysis show increasing COVID-19 VCR in adults ≥50 years of age in the United States could lead to substantial reductions in COVID-19–related disease burden
- Findings may be limited due to the uncertainty associated with 2023-2024 COVID-19 epidemiology, vaccination uptake, and VE of mRNA-1273
- Full impact of increasing COVID-19 VCR might be underestimated, with the magnitude increasing at higher VCR, as results are based on a static model and do not consider population level benefits due to indirect protection of vaccination
- Adult combination vaccines against influenza and COVID-19 could help increase COVID-19 vaccination rates and reduce burden on the healthcare system, especially during winter surge settings

References

1. Centers for Disease Control and Prevention (CDC). COVID-NET Laboratory-Confirmed COVID-19 Hospitalizations. https://covid.cdc.gov/covid-data-tracker/#covidnet-hospitalization-network. Accessed October 21, 2024.
2. Centers for Disease Control and Prevention (CDC). FluView Interactive: Laboratory-Confirmed Influenza Hospitalizations. https://gis.cdc.gov/GRASP/Fluview/FluHospRates.html. Accessed October 21, 2024.
3. Centers for Disease Control and Prevention (CDC). COVIDVaxView: Adult Vaccination Coverage and Intent. https://www.cdc.gov/covidvaxview/weekly-dashboard/adult-vaccination-coverage.html. Accessed October 21, 2024.
4. Lu P, Zhou T, Santibanez TA, et al. COVID-19 bivalent booster vaccination coverage and intent to receive booster vaccination among adolescents and adults — United States, November–December 2022. *MMWR Morb Mortal Wkly Rep*. 2023;72:190–198. DOI: http://dx.doi.org/10.15585/mmwr.mm7207a5
5. Kurosky SK, Davis KL, Krishnarajah G. Effect of combination vaccines on completion and compliance of childhood vaccinations in the United States, *Hum Vaccin Immunother*. 2017; 13(11): 2494-2502, doi:10.1080/21645515.2017.1362515.
6. Kohli MA, Maschio M, Joshi K, et al. The potential clinical impact and cost-effectiveness of the updated COVID-19 mRNA fall 2023 vaccines in the United States. *J Med Econ*. 2023;26(1):1532-1545. doi:10.1080/13696998.2023.2281083.
7. Centers for Disease Control and Prevention. COVID-NET Laboratory-Confirmed COVID-19 Hospitalizations. https://covid.cdc.gov/covid-data-tracker/#covidnet-hospitalization-network. Accessed May 30, 2024.
8. Lee A, Jayasundara K, Kohli MA et al. Clinical and economic impact of updated Fall 2023 COVID-19 vaccines in the immunocompromised population in Canada. *medRxiv*. 2023.11.10.23298369; doi: https://doi.org/10.1101/2023.11.10.23298369
9. Kopel H, Araujo AB, Bogdanov A, Zeng N, Winer I, Winer-Jones J, et al. Effectiveness of the 2023-2024 Omicron XBB.1.5-containing mRNA COVID-19 vaccine (mRNA-1273.815) in preventing COVID-19-related hospitalizations and medical encounters among adults in the United States: an interim analysis. *medRxiv*. 2024:2024.04.10.24305549
10. Higdon MM, Baidya A, Walter KK, Patel MK, Issa H, Espié E, et al. Duration of effectiveness of vaccination against COVID-19 caused by the omicron variant. *Lancet Infect Dis*. 2022;22(8):1114-6.
11. Moderna Data on File (DOF). Analysis of the Optum CMD data with patients enrolled from Sep 12, 2023 to Feb 29, 2024 and who have not received any COVID-19 XBB1.5 vaccination.
12. Centers for Disease Control and Prevention (CDC). COVID Data Tracker. https://covid.cdc.gov/covid-data-tracker/#datatracker-home. Accessed October 21, 2024.

Acknowledgments

Editorial assistance was provided by Clare Miller, PhD, of MEDiSTRAVA and was funded Moderna, Inc. This study was funded by Moderna, Inc.

Disclosures

KJ, EB, NVV, and DM are employees of Moderna, Inc., and hold stock/stock options in the company.

ADDITIONAL INFORMATION

Copies of this presentation obtained through the QR code are for personal use only and may not be reproduced without permission of the authors.

For additional information, please contact Darshan.Mehta (Darshan.Mehta@modernatx.com).

Presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe; 17-20 November 2024; Barcelona, Spain