Assessment of Health State Utilities Associated With Being a Caregiver for a Person With Alzheimer's Disease With Mild Cognitive Impairment or Dementia

PCR294

Scan the QR code or use this URL (https://lillyscience.lilly.com/congress/ispor-eu2024) for a list of all Lilly content presented at the congress. Other company and product names are trademarks of their respective owners.

Louis S. Matza[1], Timothy A. Howell[1], Mark Belger[2], Craig Ritchie[3,4], Philip R. Delio[5], Joseph A. Johnston[2], Antje Tockhorn-Heidenreich[2]

[1]Patient-Centered Research, Evidera, Wilmington, NC, US, [2]Eli Lilly and Company, Indianapolis, IN, US, [3]Scottish Brain Sciences, Edinburgh, UK, [4]Professor of Brain Health and Neurodegenerative Medicine, University of St Andrews, Scotland, UK, [5]Department of Neurology, Santa Barbara Cottage Hospital, Santa Barbara, CA, US.

Sponsored by Eli Lilly and Company

ISPOR EU 2024; Barcelona, Spain; November 17 – 20, 2024

OBJECTIVE

- The purpose of this study was to estimate health state utilities associated with being a caregiver for a person with mild cognitive impairment (MCI) or dementia associated with AD.

CONCLUSION

- Utilities followed expected patterns. Greater caregiving responsibility was associated with lower utilities. Mean utilities were lower for health states describing caring for a spouse/partner compared to health states describing caring for a parent with the same disease severity.
- Utilities for the US and UK samples were remarkably consistent, with no significant differences in utility between the two countries.
- This study adds to the existing literature on caregiver utilities. Methodological aspects of this study can inform future research on caregiver utility estimation.
- The utilities estimated in this study may be useful in models examining and comparing cost-effectiveness of treatments for MCI or dementia associated with AD.

STUDY DESIGN

- Six health state vignettes were drafted as described below in the Health State Development section (see example health state in **Figure 1**, and a complete list of health states in **Table 1**).
- These health states were valued in time trade-off (TTO) utility interviews conducted from October 2023 to February 2024 with samples of general population respondents in the UK and US.

Figure 1: Example of Health State- Health State D (Carer for a Spouse/Partner with Mild Cognitive Impairment)

	T
Your Spouse or Partner	• You live with your spouse. • It <u>**sometimes**</u> seems like something is not right with their **thinking and memory**.
Behavioral Impact	• You **sometimes** notice that the following things occur: ○ They forget and lose things more often than they used to. ○ They lose their train of thought. ○ They ask the same question repeatedly or engage in repetitive conversations.
Emotional Impact	• They are **sometimes frustrated** by their problems with memory and thinking. • Sometimes, they seem **anxious or depressed** about their thinking abilities. • Your spouse **does not have as much confidence** in their abilities as they used to.
Functional Impact	• Their ability to perform their **typical daily tasks is not impaired,** but subtle changes are noticeable to close family members. • They **show less interest** in activities they used to do. • They **avoid some activities**, such as driving in unfamiliar areas or at night. • They sometimes **get lost** in places they know well. • They sometimes **forget appointments** and social events. • They have **some difficulty organizing some tasks**, like **cooking a complete meal**. • They have made **some lifestyle adjustments to compensate** for the memory changes. For example, they write things down to ensure that they don't forget them.
Your Role	• You **do not have any daily caregiving responsibilitie**s. • You take on **more organizational responsibility** than you used to, like **planning things for both of you.**
Impact on You	• You sometimes try to **minimize your spouse's symptoms** in public by helping them remember things and answering questions for them. • You have to **remind them** of events or responsibilities more often than you used to. • You **watch their behavior** to try to better understand what the problem is and what the future might bring.
Emotional Impact	• You **sometimes** feel **irritated because of repetitive conversations** with your spouse. • You **worry about whether they can independently manage daily activities** like shopping. • You **worry about the future** of your spouse's condition and your relationship with them.
Healthcare	• Because of what you have noticed, you have started **searching on the internet for information** on memory disorders. • You have made **appointments with healthcare providers** to assess your spouse's condition.

BACKGROUND

- Alzheimer's disease (AD) is the most common cause of dementia, accounting for 60% to 70% of cases.[1,2]
- Patients with AD dementia often require caregivers, a role which typically falls to a family member. Caregiving for people with AD dementia has been found to be associated with increased stress,[3,4] anxiety,[4-6] depression,[4-6] worsened mental functioning,[4] and lower quality of life.[7] However, the burden of caregiving may be reduced by interventions targeting psychiatric symptoms, behavioral disturbances, and quality of life in persons with AD dementia.[8]
- Although the burden of caregiving for a person with AD dementia has previously been quantified,[8-12] the effect caregiving has on an individual's utility has rarely been studied.[13,14]

Table 1: Health State Utilities

Health State Letter	Description
A	Caregiving for parent with MCI due to AD
B	Caregiving for parent with mild dementia due to AD
C	Caregiving for parent with moderate dementia due to AD
D	Caregiving for spouse/partner with MCI due to AD
E	Caregiving for spouse/partner with mild dementia due to AD
F	Caregiving for spouse/partner with moderate dementia due to AD

METHODS

Participants

- General population participants were required to meet the following criteria: (1) ≥18 years of age; (2) able to understand the assessment procedures; (3) able and willing to give informed consent; (4) able to complete the protocol requirements; and (5) a UK or US resident.

Health State Development

- Health state descriptions were developed based on literature review and multiple rounds of telephone interviews with five clinicians (3 neurologists and 2 psychiatrists in the UK and US with an average of 17.6 years of experience working clinically with MCI/AD demented patients).
- Health states were reviewed and refined in focus groups with members of Alzheimer Europe's European Working Group of People with Dementia and European Dementia Carers.
- Two of these focus groups were conducted, one with caregivers of parents with AD (n=4, all women, average age 59 years old) and another with caregivers of spouses/partners with AD (n=6, three women, average age 64 years old).

Pilot Study

- Prior to the main study, a pilot study was conducted to finalize the interview procedures and health state descriptions (N = 25; mean age = 41.2 years; 56% women).
- Participants completed health state valuation tasks and then provided feedback on health states and interview procedures. Almost all participants had no difficulty understanding the health states or the TTO task.
- Minor edits were made to the health states for clarity based on participant feedback.
- Because there is no consensus regarding methods for valuing caregiver health states, several TTO approaches were explored in the pilot study. Interviewers gathered feedback from participants about their understanding and comprehension of each approach, which was reviewed to inform the methods used in the main valuation study, including the upper anchor of the scale.

Procedures

- Participants first ranked the health states in order of preference. Then, they valued the health states in a TTO task. After completing the TTO portion of the interview, the participants also completed a demographic and clinical information form.
- After reviewing the performance of the various upper anchors attempted in the pilot study, the upper anchor of the utility scale was defined as an absence of emotional and logistical caregiver burden because the family member does not have MCI or dementia.

Results

Sample Description

- A total of 304 participants from four locations in the UK (Edinburgh [n=59], Leeds [n=72], London [n=116], and Bath [n=57]) and 202 participants from two locations in the US (Bethesda, MD, [n=108] and Chicago, IL [n=94]) completed the TTO utility elicitation.

Health State Rankings

- In the UK, health state A (parent with MCI) was ranked first by 65.8% of the sample. Health state D (spouse with MCI) was ranked highest by 31.3% of the sample. Health states C (parent with moderate AD) and F (spouse with moderate AD) were most commonly ranked lowest by 26.6% and 70.7% of the participants, respectively.
- In the US, health state A (parent with MCI) was ranked first by 55.0% of the sample. Health state D (spouse with MCI) was ranked highest by 38.6% of the sample. Health states C (parent with moderate AD) and F (spouse with moderate AD) were most commonly ranked lowest (35.6% and 59.9% of the sample, respectively).

Health State Utilities

- Mean (SD) utilities are presented in **Figure 2**. In both countries, all health state comparisons across severity levels (e.g., mild vs. moderate AD) were significant (p<0.0001). In the UK, utilities for all parent health states were significantly higher than corresponding spouse/partner health states (p<0.0001). In the US, utilities for parent mild and moderate AD dementia were significantly higher than corresponding spouse/partner health states (p<0.05), but there was no significant difference between parent and spouse MCI health states (p=0.08). There were no significant differences in utilities between samples in the UK and US.

Figure 2: Mean Health State Utilities (N=304 in UK, N=202 in US)

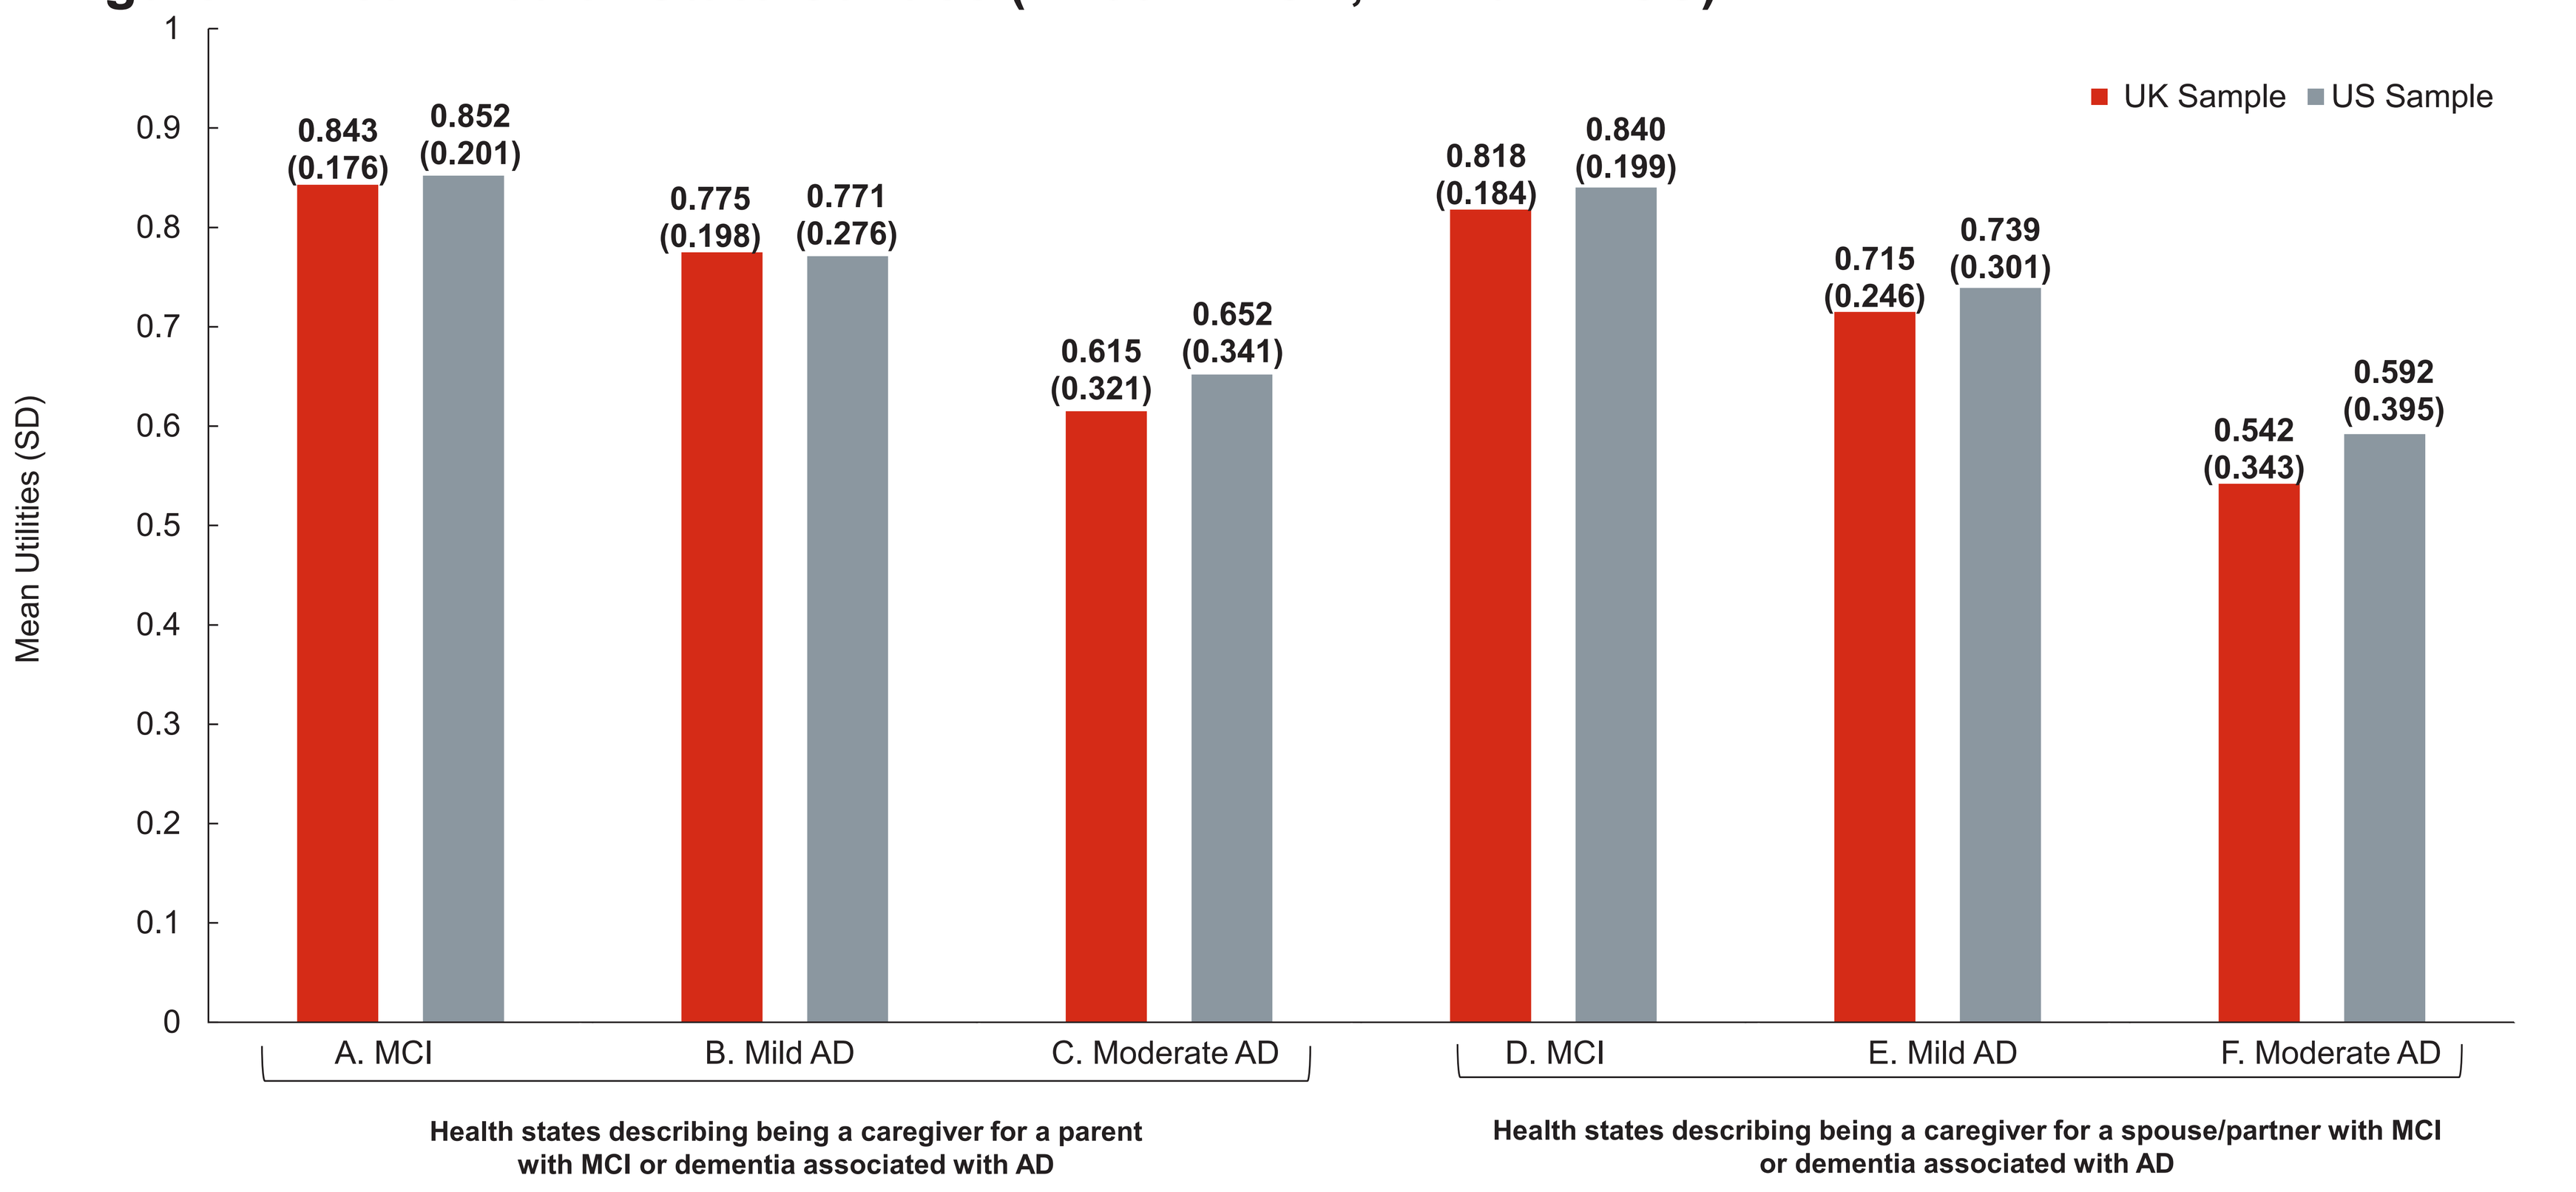

Mean Utilities (SD)
1
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0
UK Sample
US Sample
0.843 (0.176)
0.852 (0.201)
0.775 (0.198)
0.771 (0.276)
0.615 (0.321)
0.652 (0.341)
0.818 (0.184)
0.840 (0.199)
0.715 (0.246)
0.739 (0.301)
0.542 (0.343)
0.592 (0.395)
A. MCI
B. Mild AD
C. Moderate AD
D. MCI
E. Mild AD
F. Moderate AD
Health states describing being a caregiver for a parent with MCI or dementia associated with AD
Health states describing being a caregiver for a spouse/partner with MCI or dementia associated with AD

Abbreviations: AD = Alzheimer's dementia; MCI = mild cognitive impairment; SD = standard deviation; UK = United Kingdom; US = United States

Table 2: Demographic Characteristics

Characteristics	UK Sample (N=304)	US Sample (N=202)
Age (years), (mean, SD)	46.7 (15.7)	46.4 (16.2)
Gender (n, %)		
Male	139 (45.7%)	100 (49.5%)
Female	164 (53.9%)	96 (47.5%)
Nonbinary	1 (0.3%)	6 (3.0%)
Ethnic Background (n, %)		
Hispanic or Latino		19 (9.4%)
Not Hispanic or Latino		182 (90.1%)
Missing		1 (0.5%)
Racial Background (n, %)		
Asian/Asian British/<u>Asian American</u>	24 (7.9%)	<u>27 (13.4%)</u>
Black/African/Caribbean/Black British/<u>African American</u>	8 (2.6%)	<u>33 (16.3%)</u>
Mixed/Multiple ethnic groups	12 (3.9%)	<u>7 (3.5%)</u>
White	249 (81.9%)	<u>123 (60.9%)</u>
Other	11 (3.6%)	<u>12 (5.9%)</u>
Marital Status (n, %)		
Single	101 (33.2%)	83 (41.1%)
Married	97 (31.9%)	66 (32.7%)
Other	106 (34.9%)	53 (26.2%)
Employment Status (n, %)		
Full-time work	116 (38.2%)	91 (45.0%)
Part-time work	76 (25.0%)	44 (21.8%)
Other	112 (36.8%)	67 (33.2%)
Education Level (n, %)		
No university degree	93 (30.6%)	59 (29.2%)
University degree	211 (69.4%)	143 (70.8%)
Participant had a family member with MCI diagnosis	53 (17.4%)	49 (24.3%)
Participant had a family member with AD diagnosis	75 (24.7%)	54 (26.7%)
Participant had experience as a caregiver for a person with dementia due to MCI or AD	60 (19.7%)	55 (27.2%)

Abbreviations: UK = United Kingdom; US = United States

Limitations

- Utilities are derived from general population preferences for hypothetical health state vignettes, rather than the experiences of actual caregivers. Comparability between these values and those derived from caregivers is unknown.
- Assessment of caregiver utilities is still an emerging field with no consensus on the preferred methodology for utility elicitation. Further research is required to provide more confidence in these methods.

References

1. Gauthier S, Webster C, et al. *World Alzheimer Report 2022.* https://www.alzint.org/u/World-Alzheimer-Report-2022.pdf
2. Tahami Monfared AA, Byrnes MJ, et al. *Neurol Ther*. 2022;11(2):525-551. doi:10.1007/s40120-022-00335-x
3. Anand KS, Dhikav V, et al. *Ann Indian Acad Neurol*. 2016;19(1):58-62. doi:10.4103/0972-2327.167695
4. Correa MS, Vedovelli K, et al. *Neuroscience*. 2015;286:371-382. doi:10.1016/j.neuroscience.2014.11.052
5. Laks J, Goren A, et al. *Int J Geriatr Psychiatry*. 2016;31(2):176-185. doi:10.1002/gps.4309
6. Sallim AB, Sayampanathan AA, et al. *J Am Med Dir Assoc*. 2015;16(12):1034-1041. doi:10.1016/j.jamda.2015.09.007
7. Garzón-Maldonado FJ, et al. *Neurologia (Engl Ed)*. 2017;32(8):508-515. doi:10.1016/j.nrleng.2016.02.011
8. Mohamed S, Rosenheck R, et al. *Am J Geriatr Psychiatry*. 2010;18(10):917-927. doi:10.1097/JGP.0b013e3181d5745d
9. Allegri RF, Sarasola D, et al. *Neuropsychiatr Dis Treat*. 2006;2(1):105-110.
10. Folquitto JC, Marques RCG, et al. *Dement Neuropsychol*. 2013;7(3):258-262. doi:10.1590/S1980-57642013DN70300005
11. Pena-Longobardo LM, Oliva-Moreno J. *J Alzheimers Dis*. 2015;43(4):1293-1302. doi:10.3233/JAD-141374
12. Shin H, Youn J, et al. *J Geriatr Psychiatry Neurol*. 2012;25(4):222-226. doi:10.1177/0891988712464819
13. Belger M, Dell'Agnello G, et al. PCR24. *Value Health*. 2022;25(7)doi:10.1016/j.jval.2022.04.1371
14. Belger M, Dell'Agnello G, et al. PCR59. *Value Health*. 2022;25(7):S551-S552. doi:10.1016/j.jval.2022.04.1404

Acknowledgments: Editorial support was provided by Amara Tiebout and graphic design support was provided by Richard Leason.

Disclosures: Funding for this research was provided by Eli Lilly & Co.

Copyright ©2024 Eli Lilly and Company. All rights reserved.

Untitled-6 1

11/11/2024 11:24:37 AM