Real-world data of severe viral lower respiratory tract disease: current landscape, challenges and future recommendations

Ekaterina Maslova[1]*, Monica Seif[2]*, Zohaib Akhter[2], Poppy Payne[2], Thomas Jennison[2], Susan Johnson[1], Neil Branscombe[1], Tim Holbrook[2]
[1]AstraZeneca, Cambridge UK, [2]Adelphi Real World, Bollington, UK
*Co-primary authors

PA 141474
Poster presented at ISPOR Europe;17th-20th November 2024; Barcelona, Spain

Why did we perform this research?

A 2019 burden of disease study reported 488.9 million lower respiratory tract disease (LRTD) cases globally, with a significant volume of 2.4 million deaths attributable to LRTD in adults [1]. The COVID-19 pandemic contributed further to the disease burden, especially among patients with severe viral LRTD.

Despite an immense global burden of LRTD, there is a scarcity of information available that outlines the capability of real-world data (RWD) sources to follow patients from symptom onset to hospitalisation and into recovery, while also capturing treatment pathways and respiratory events.

What was the objective of this research?

We conducted a comprehensive evaluation of the current global RWD landscape for severe viral LRTD in the acute respiratory care setting (specifically secondary care and post-discharge care), to identify suitable RWD sources for evidence generation activities, associated gaps and limitations.

How did we perform this research?

Proprietary and open data platforms explored using search term combinations, supplemented with in-depth interviews with data custodians

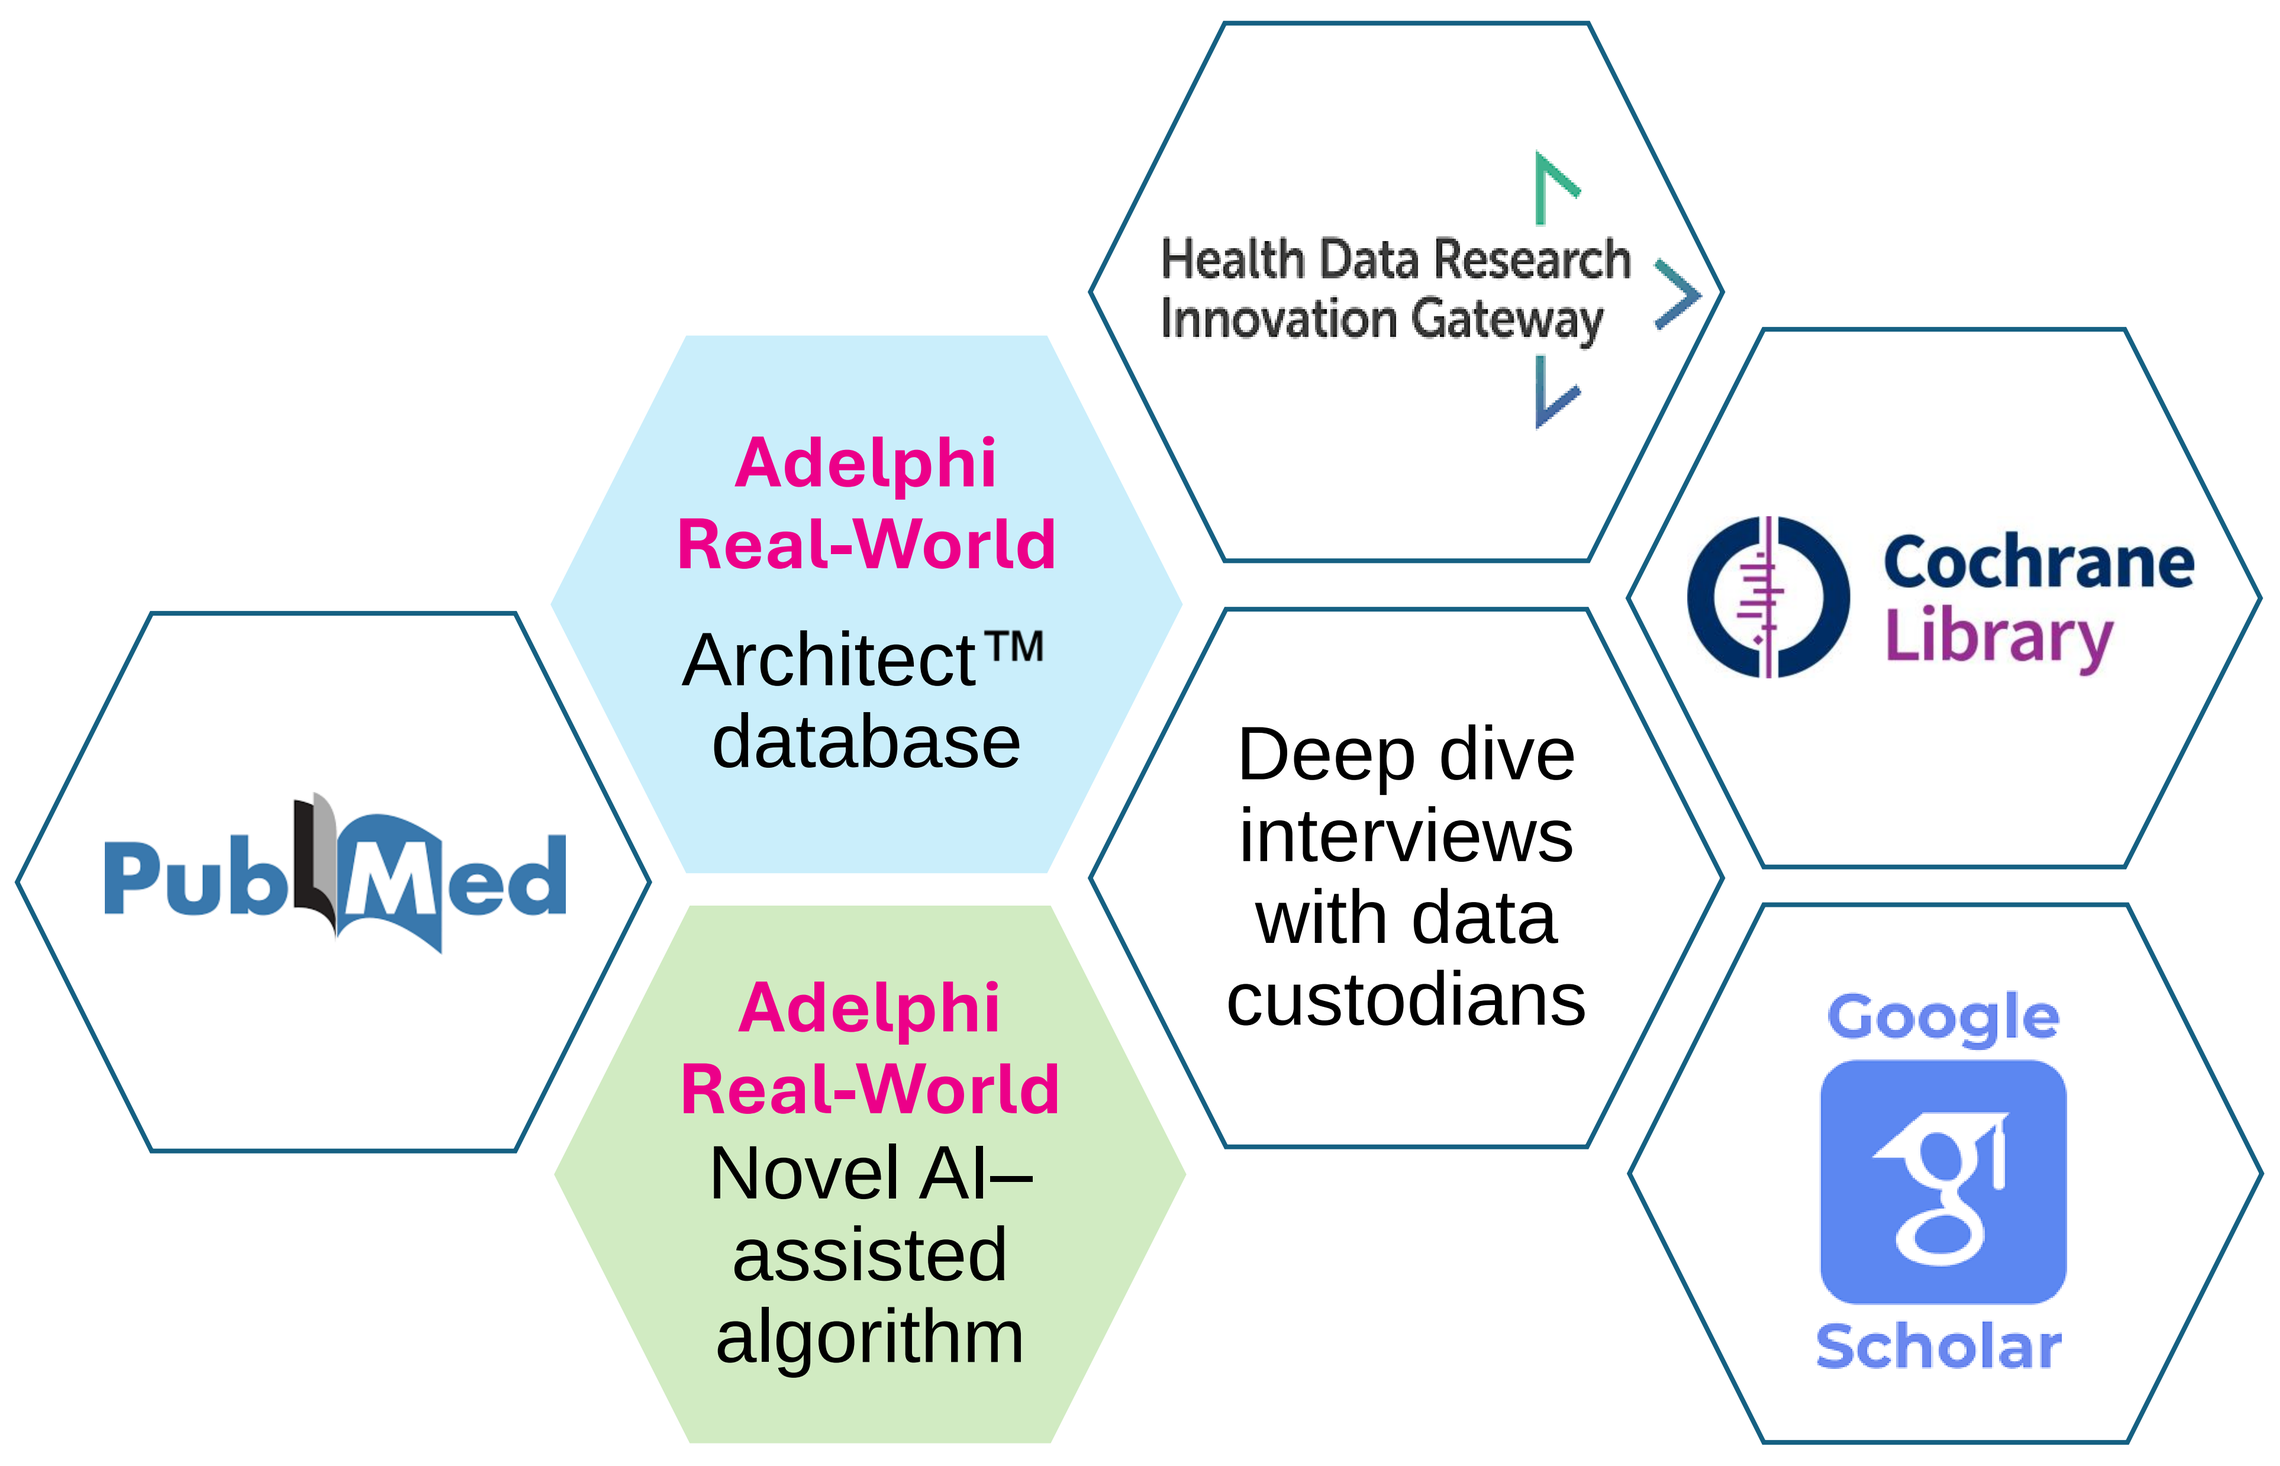
Health Data Research Innovation Gateway
Adelphi Real-World
Architect™ database
Cochrane Library
PubMed
Deep dive interviews with data custodians
Adelphi Real-World
Novel AI–assisted algorithm
Google Scholar

Outcomes measures of interest for LRTD RWD sources

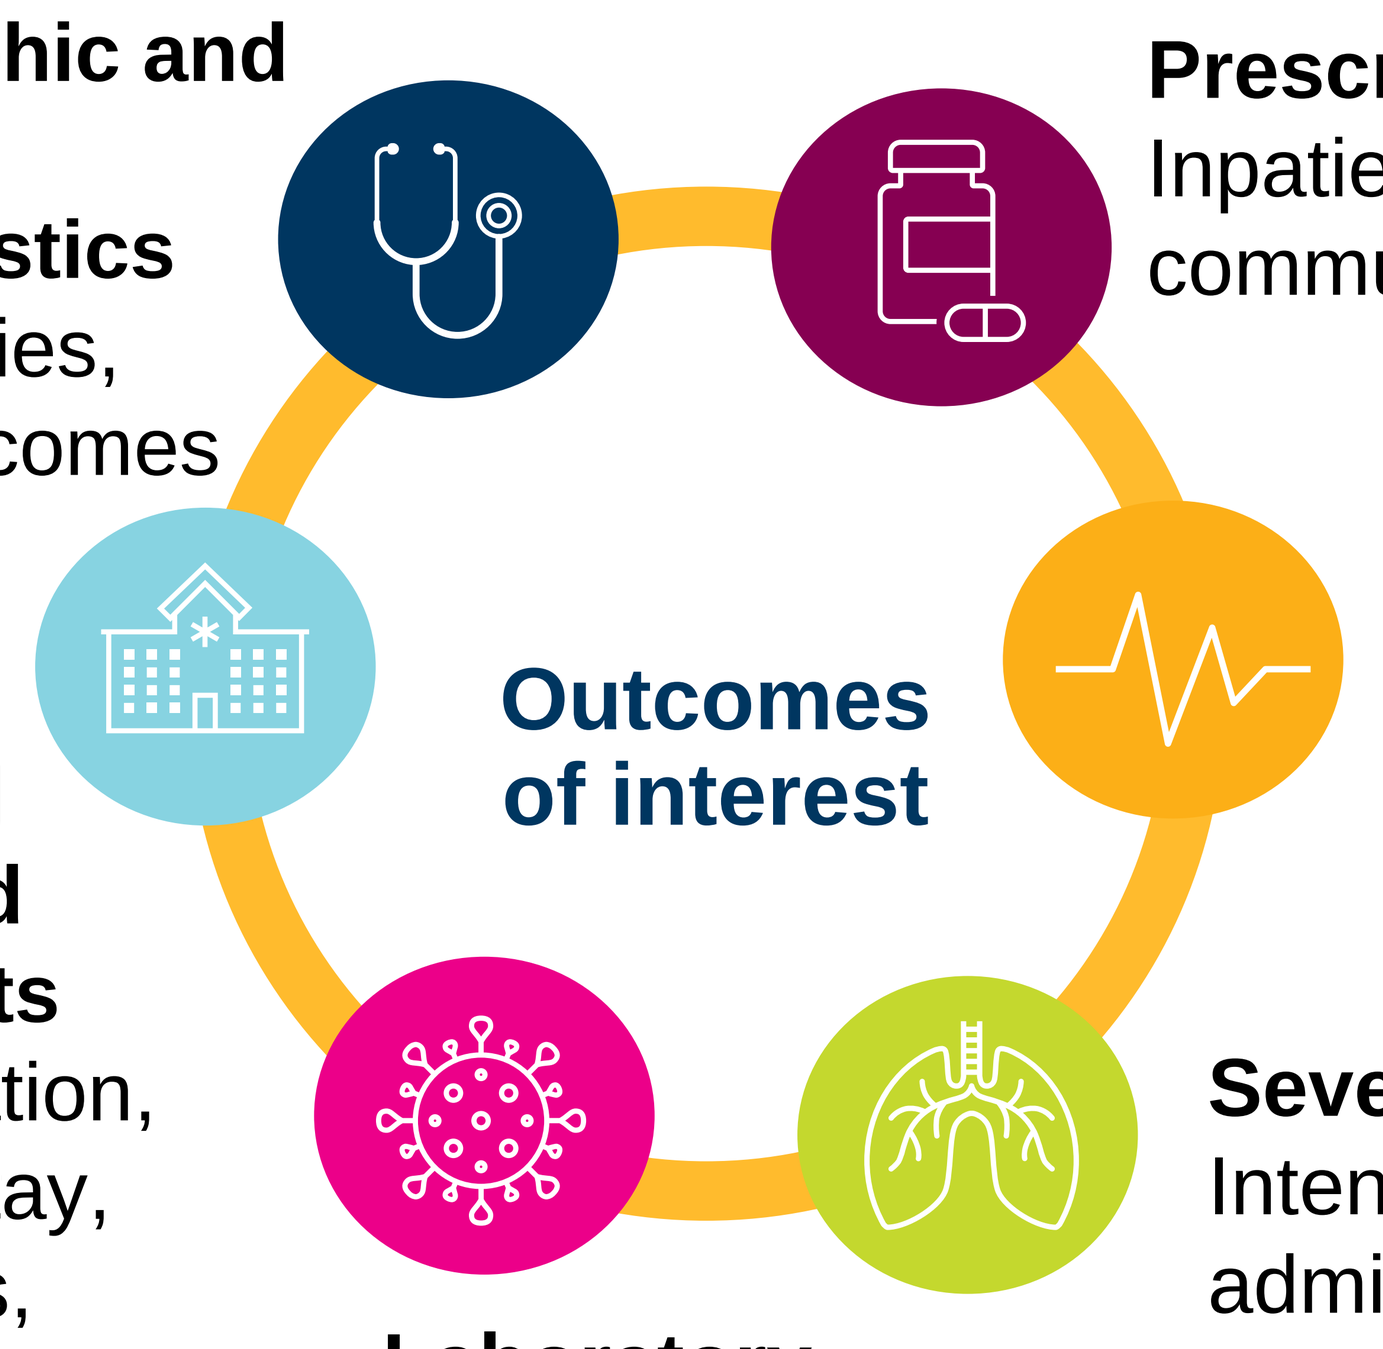
Outcomes of interest

Demographic and clinical characteristics
Comorbidities, clinical outcomes

Prescriptions
Inpatient, outpatient and community prescriptions

Mortality/survival
All-cause or disease-specific

HCRU and associated direct costs
Hospitalisation, length of stay, procedures, prescriptions

Laboratory testing
Viral aetiology

Severity measures
Intensive care unit admissions, oxygen support, disease-specific severity scales, hospital severity indicators

Three-tiered evaluation of data sources to short-list

1= Definitive i.e., Source of value to extract on being a **high-quality** source
2= Valuable source but includes caveats such as:
- Several other higher quality sources in the same country
- Meets one of the flexible* exclusion criteria
- May be a source with future potential
- Insufficient available information to classify as high-value.

3 = Exclude i.e., relatively poor-quality source

**unless they are the only best source available for a country*

What did we find?

- Overall, 458 viral LRTD RWD sources were identified following triage.
- Among these, 75 data sources were evaluated for final in-depth exploration.
- Of those 75, the majority data sources were disease agnostic (n=66; 88.0%)
- Electronic health records [EHR] were found as the most common data type (n=21; 28.0%; **Figure 1).**
- Most represented regions/ countries were Europe (n=27; 36.0%), and the United States [US] (n=16; 21.3%; **Figure 2** and **3**).
- Outcome availability between data sources varied, with clinical outcomes (n=69; 92.0%) and mortality (n=61; 81.3%) being the most common while patient reported outcome measures [PROMs] (n=13; 17.3%) and non-clinical specialist care (n=29; 38.6%) being the least.
- Access to data remains a limitation within the conduct of real-world research, with 7 data sources (11.0%) reported as "unclear or restricted access" to the data.

Figure 1. Final distribution of valuable LRTD RWD sources (n=75) by type of data source

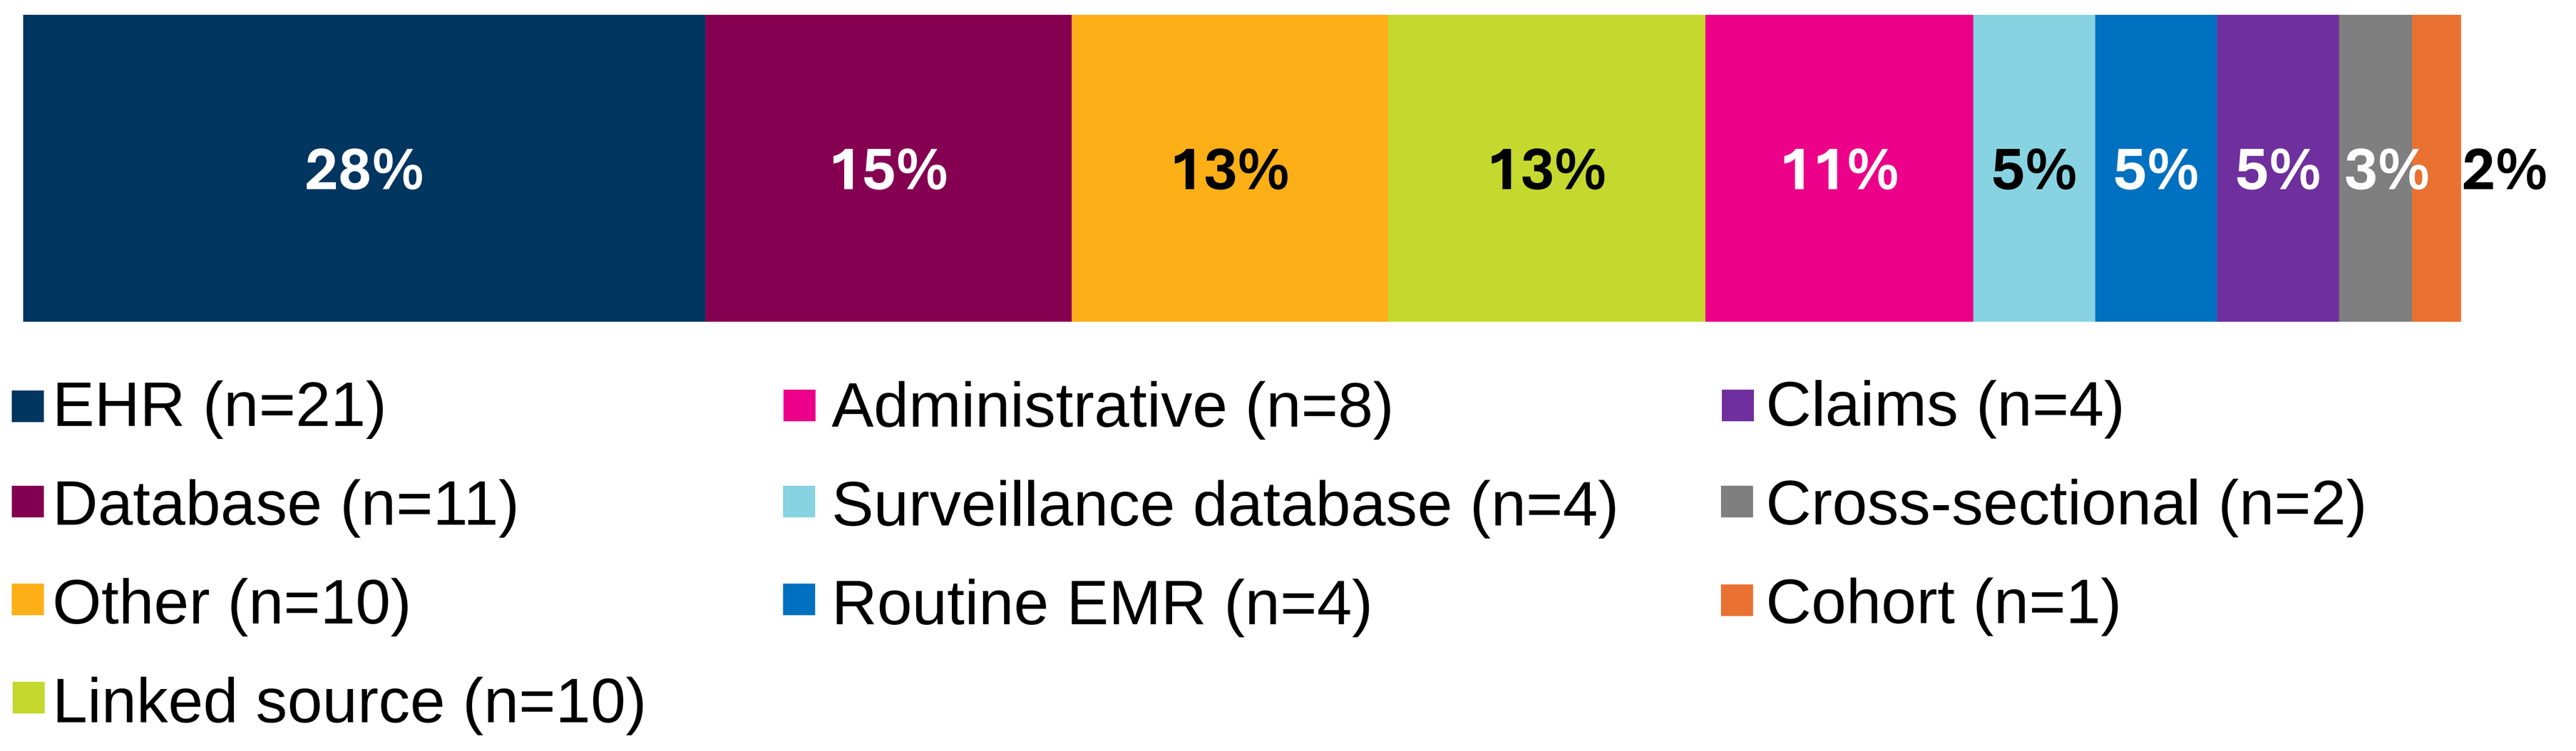
28%
15%
13%
13%
11%
5%
5%
5%
3%
2%
EHR (n=21)
Database (n=11)
Other (n=10)
Linked source (n=10)
Administrative (n=8)
Surveillance database (n=4)
Routine EMR (n=4)
Claims (n=4)
Cross-sectional (n=2)
Cohort (n=1)

Figure 2. Final distribution of valuable LRTD RWD sources (n=75) by country

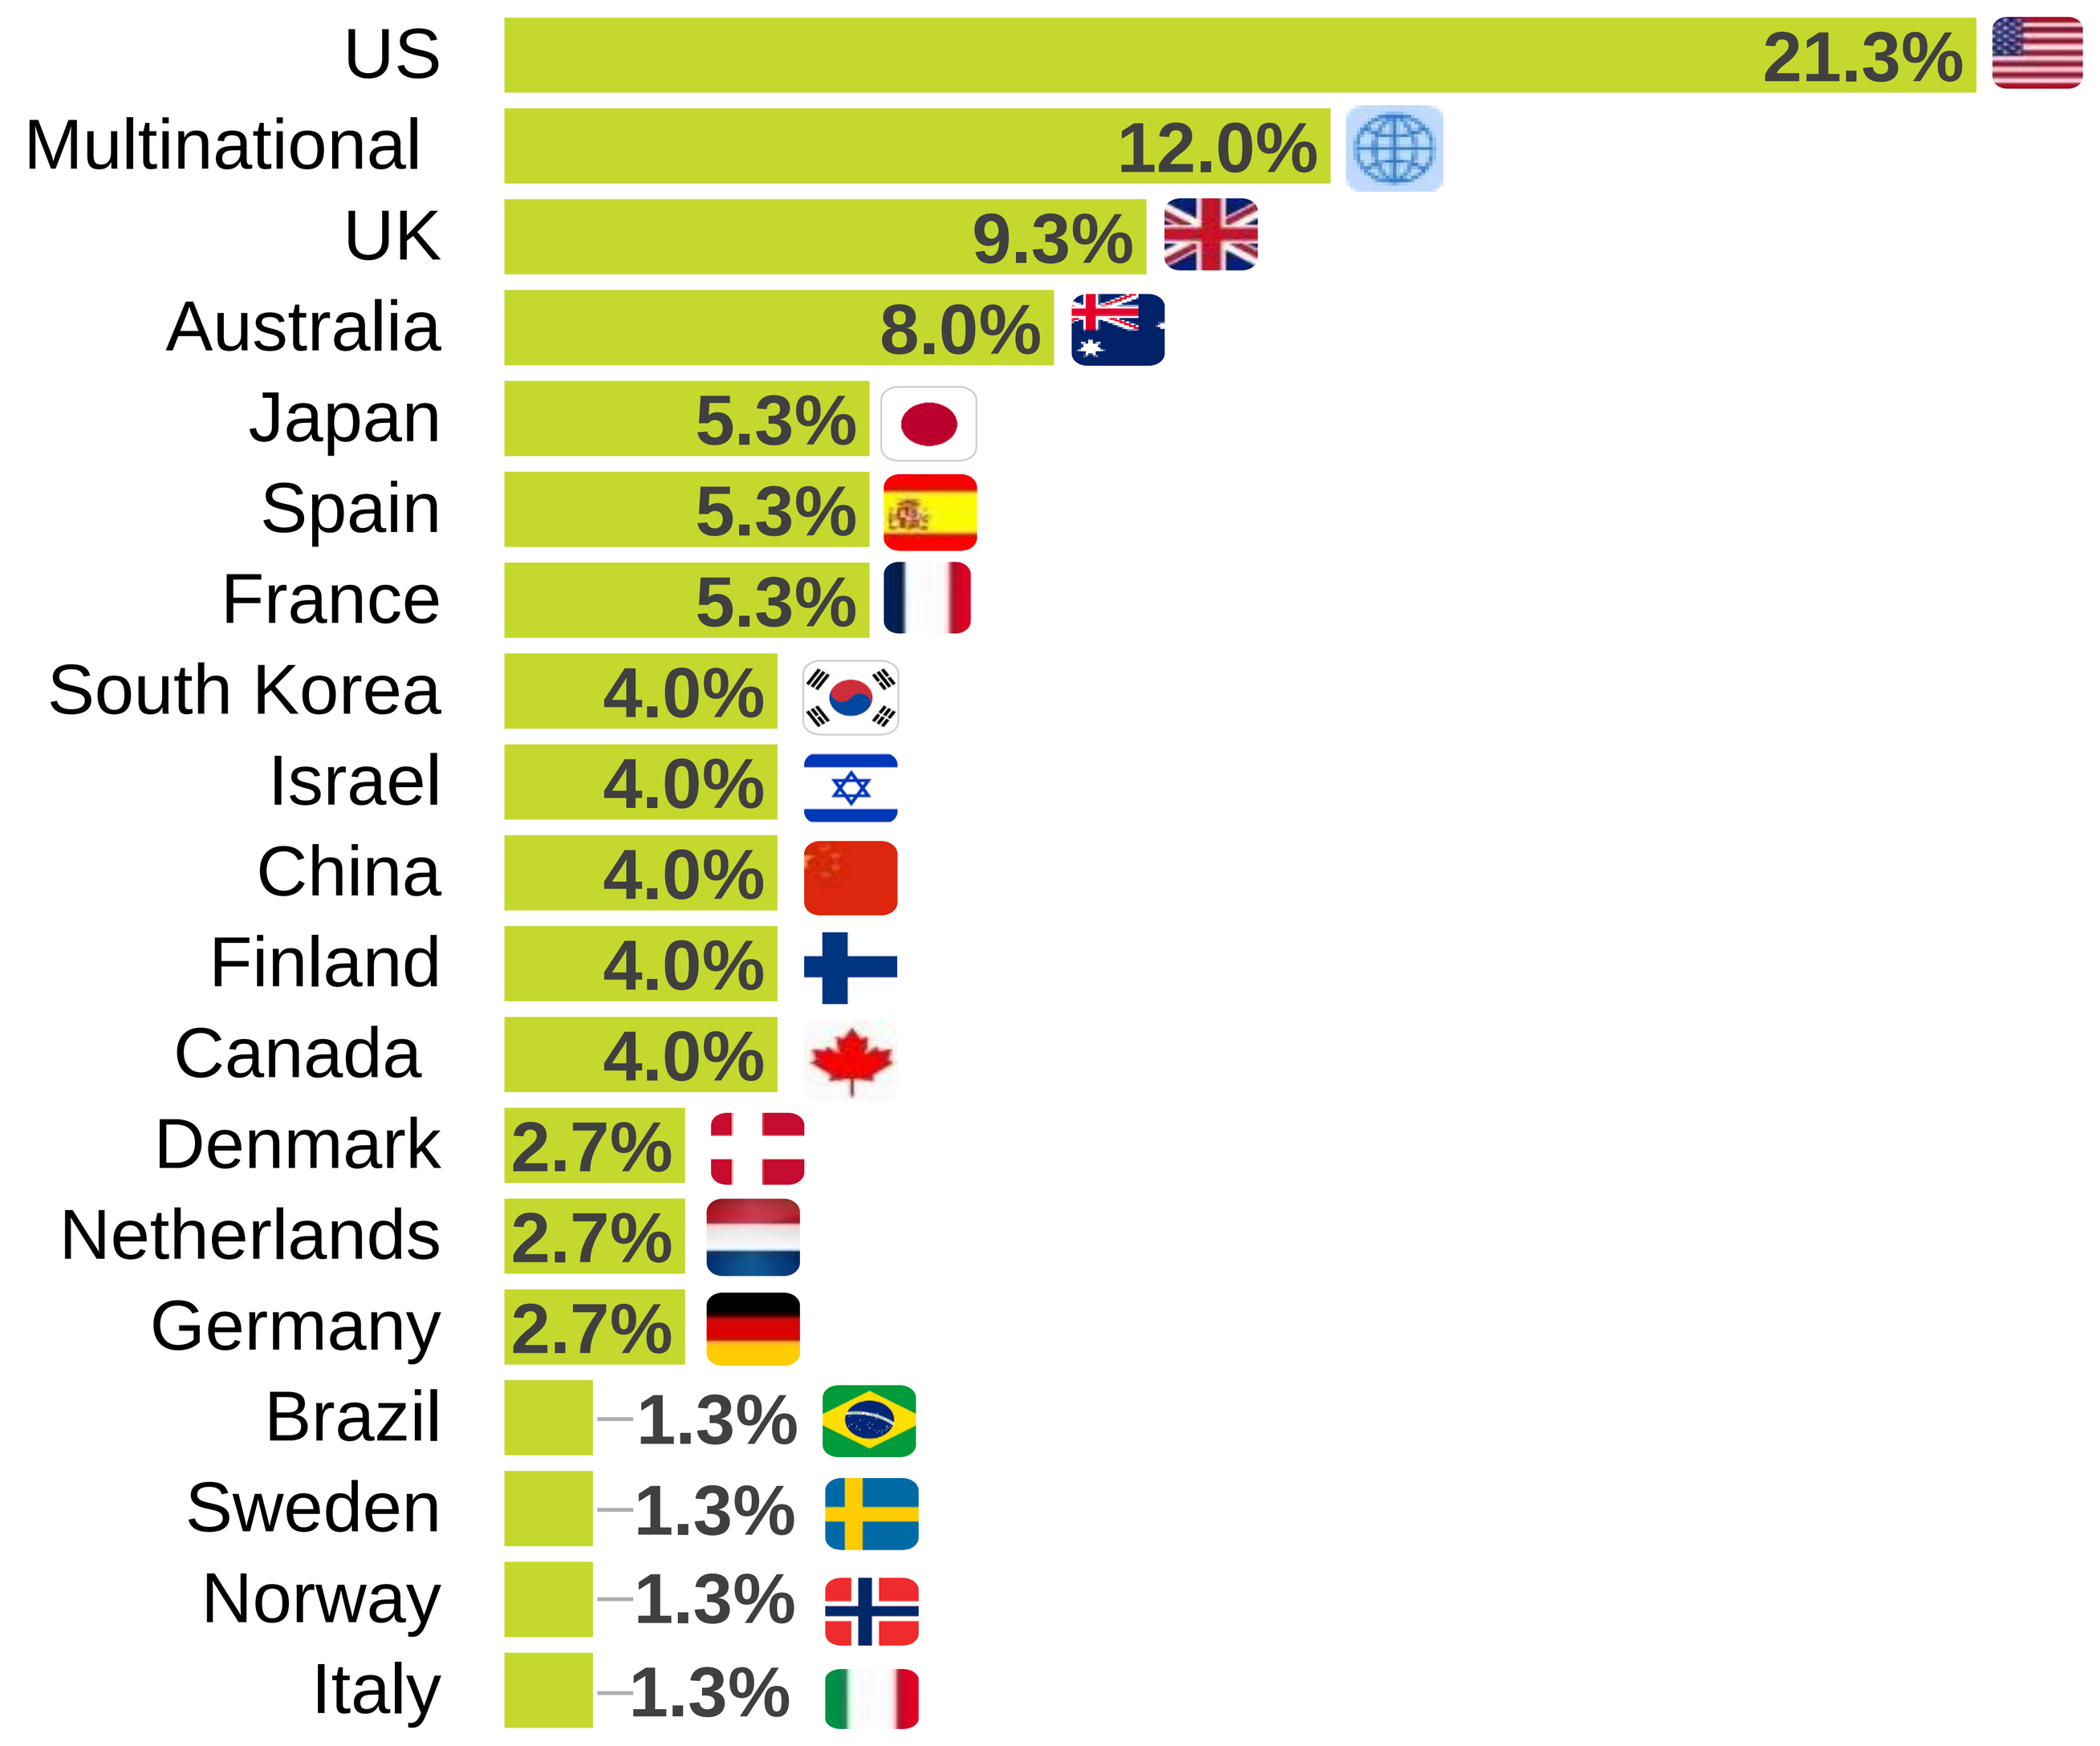
US 21.3%
Multinational 12.0%
UK 9.3%
Australia 8.0%
Japan 5.3%
Spain 5.3%
France 5.3%
South Korea 4.0%
Israel 4.0%
China 4.0%
Finland 4.0%
Canada 4.0%
Denmark 2.7%
Netherlands 2.7%
Germany 2.7%
Brazil 1.3%
Sweden 1.3%
Norway 1.3%
Italy 1.3%

Figure 3. Final distribution of valuable LRTD RWD sources (n=75) by region

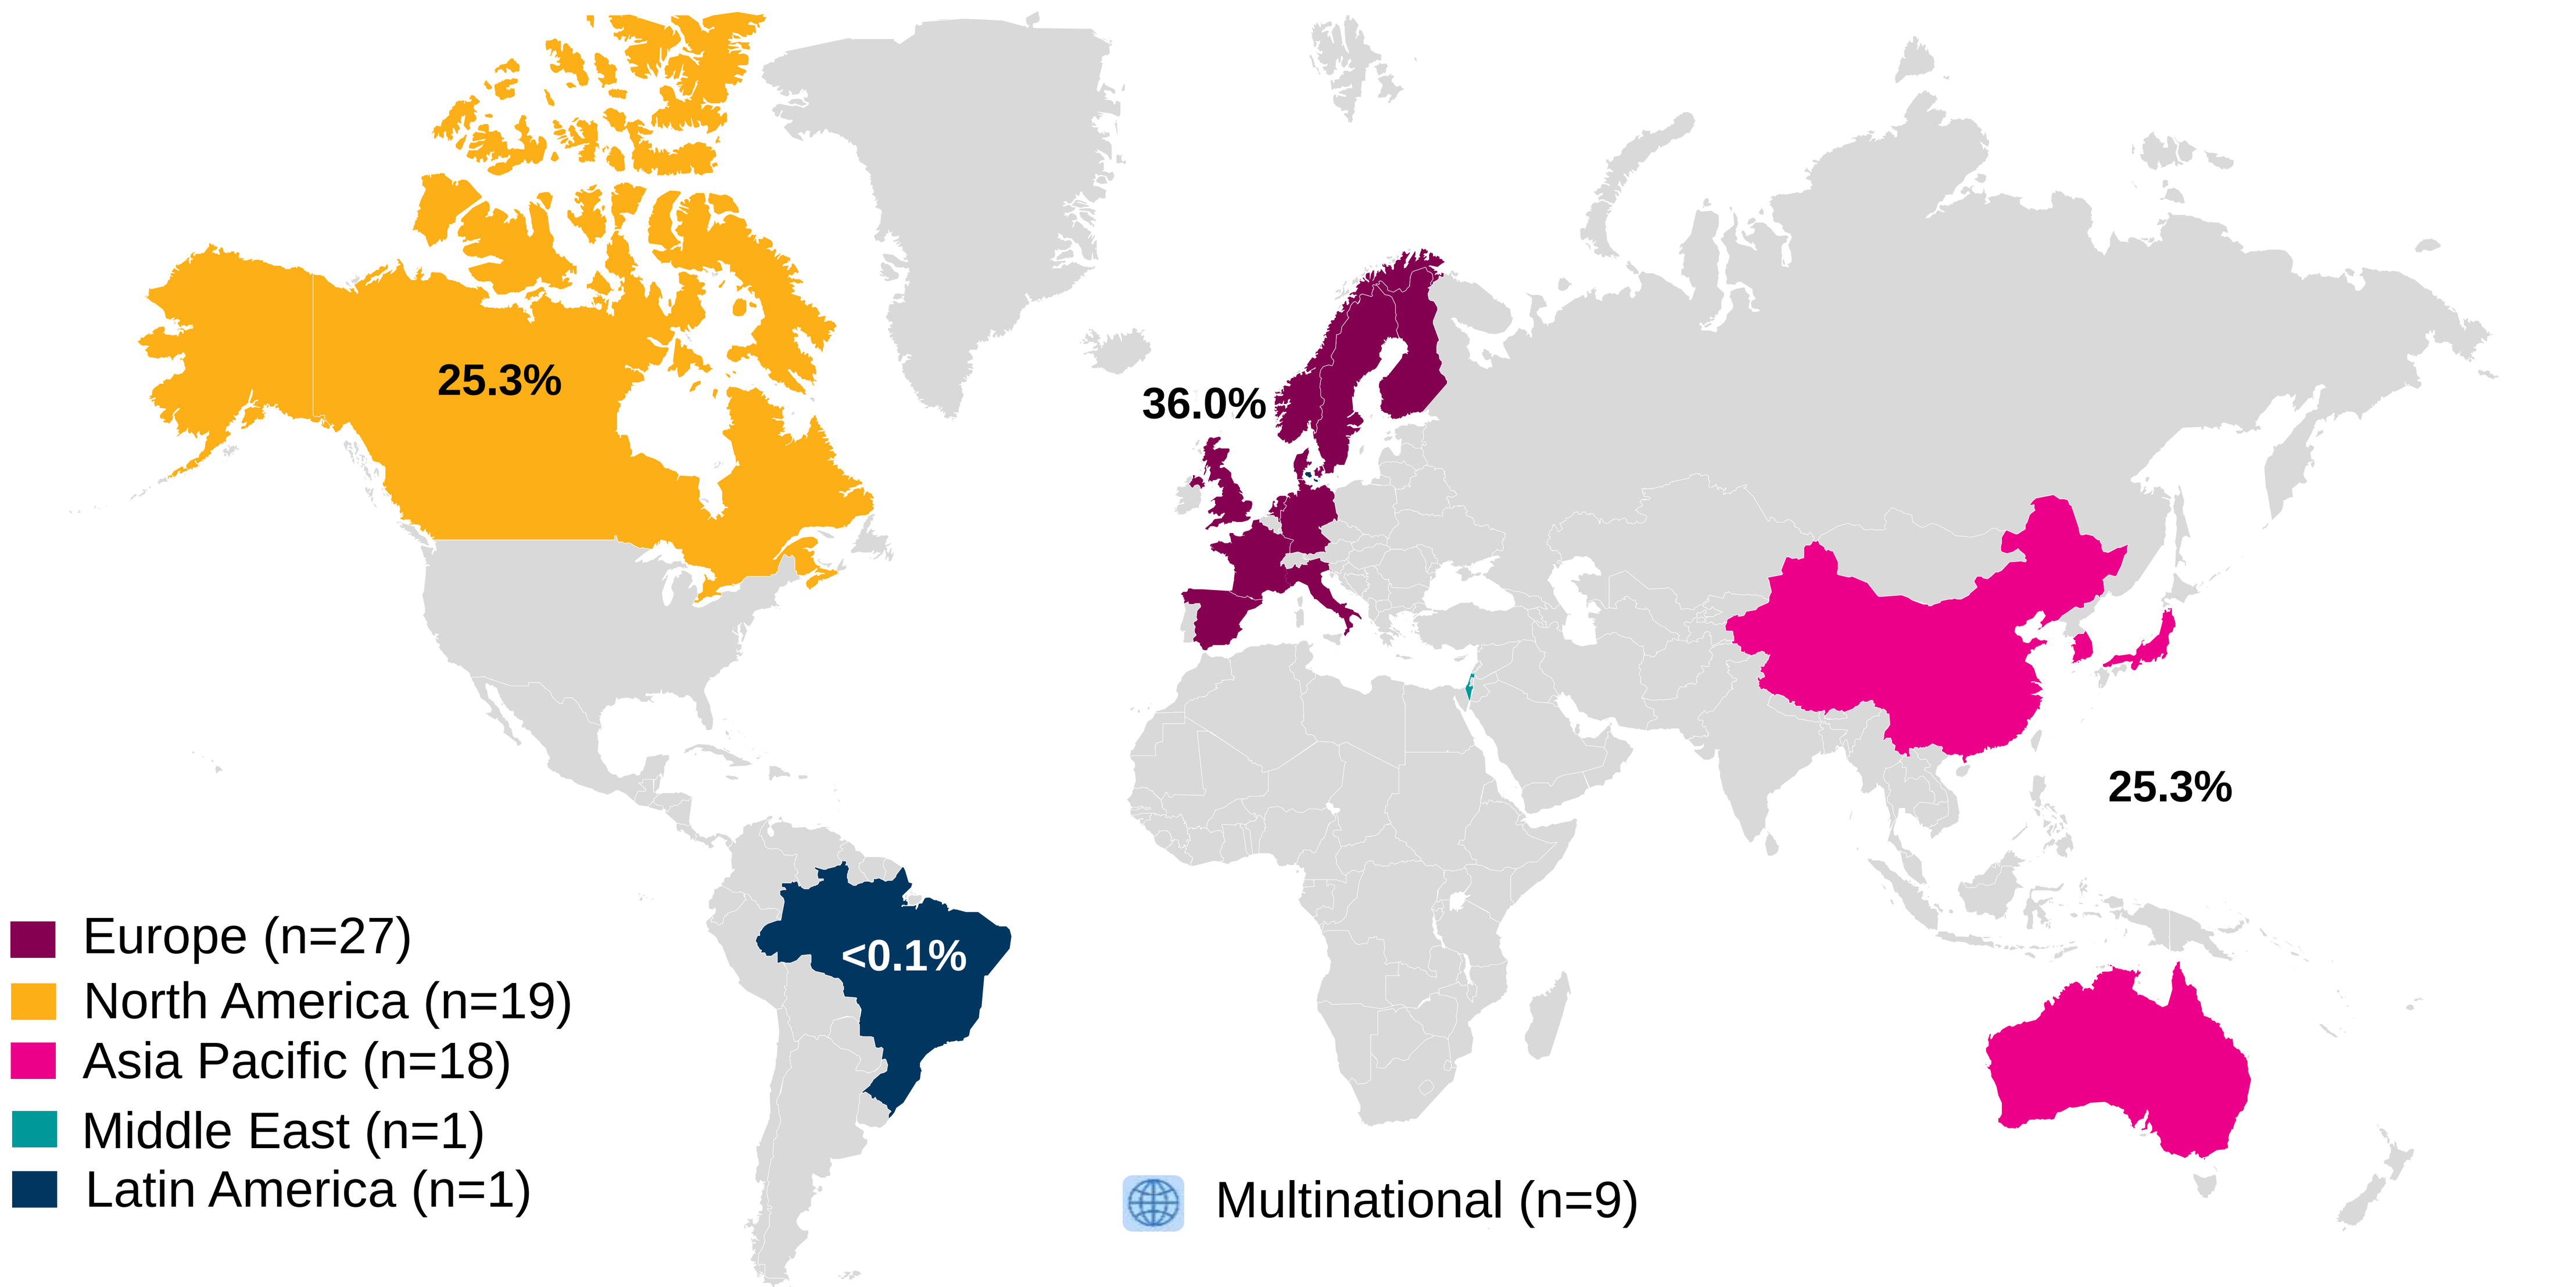
25.3%
36.0%
25.3%
<0.1%
Europe (n=27)
North America (n=19)
Asia Pacific (n=18)
Middle East (n=1)
Latin America (n=1)
Multinational (n=9)

How might this impact the current LRTD data landscape?

- **Of the countries in scope, several including Italy and China were found to have limited robust RWD sources to recommend, specifically for viral LRTD evidence needs.**
- **Despite identifying numerous sources globally, limited access and specialised datasets limit wider research opportunities which are crucial for surveillance and pandemic preparedness. Newer initiatives encourage open access via national electronic health systems and common data models, conditional on ensuring patient protection and confidentiality, as well as increased inclusion of PROMs to elicit patient outcomes.**

Abbreviations

AI: Artificial intelligence; EHR: electronic health records; EMR: electronic medical record; EU5: France, Germany, Italy, Spain, United Kingdom; ICU: intensive care unit; LoS: length of stay; LRTD: Lower respiratory tract disease; RWD: real-world data; PROs: patient reported outcomes; UK: United Kingdom; US: United States

Reference: [1] Safiri S et al. Global burden of lower respiratory infections during the last three decades. Front Public Health. 2023 Jan 9;10:1028525.

Acknowledgments

Editorial support (in the form of writing assistance, including preparation of the draft poster under the direction and guidance of the authors, collating and incorporating authors' comments for each draft, assembling tables and figures, grammatical editing, and referencing) was provided by Adelphi Real World. The project was funded by AstraZeneca.

Disclosures

This study was funded by AstraZeneca. The authors declare the following real or perceived conflicts of interest in relation to this presentation: EM, NB and SJ are employees of AstraZeneca and/or hold financial equities in AstraZeneca. TH, MS, ZA, TJ and PP are employees of Adelphi Real World. Adelphi Real World is a business that provides consulting and other research services to pharmaceutical, device, government, and non-government organizations, which received funding from AstraZeneca to conduct the project.

Confidential - Not for Public Consumption or Distribution