EE23

Healthcare resource utilisation (HCRU) and medical costs among patients with endometrial cancer in a real-world setting in Finland (the FIRE study)

Digital poster

SCAN ME

Barbara Mascialino[1], Juhani Aakko[2], Kai Kysenius[2], Samuli Tuominen[2], Juhana Idänpään-Heikkilä[3], Sari Käkelä[3], Saara Tikka[3], Dirk Schneider[4], Sakari Hietanen[5], Heini Lassus[6], Annika Auranen[7]

[1]GSK, Verona, Italy; [2]Medaffcon Oy, Espoo, Finland; [3]GSK, Helsinki, Finland; [4]GSK, Baar Onyx, Switzerland; [5]Turku University Hospital, Turku, Finland; [6]Helsinki University Hospital, Helsinki, Finland; [7]Tays Cancer Centre, Tampere University Hospital and Tampere University, Tampere, Finland

Background

- EC is the fourth most common cancer in women in developed countries. Every year, close to 100,000 new cases are registered across the European Union and the United Kingdom.[1]
- In Finland, approximately 900 cases of EC are diagnosed annually, primarily in women >65 years of age.[2]
- EC can be classified as dMMR or MMRp, based on absence or presence of proteins that have a crucial role in the DNA mismatch repair process.[3]
 - ESMO recommends the MMR IHC panel as the first method for MMR testing.[4]
 - In Finland, IHC-based MMR testing for patients with EC has been recommended by FINGOG since 2018.[5]
- There is currently a lack of real-world data on the characteristics, treatment patterns and clinical outcomes for patients with EC with known MMR status in Finland.
 - Obtaining real-world data is important to understand patient unmet needs, inform clinical trial design and guide the assessment of novel therapeutic strategies within this treatment landscape.

Aims

- The retrospective FIRE study used healthcare data to assess the real-world clinical characteristics, treatment patterns, outcomes and HCRU for patients with EC in Finland.
- Here, we focus primarily on HCRU and associated medical costs for patients with advanced or recurrent EC.

Methods

- FIRE is a multicentre, retrospective, non-interventional study of patients with EC based on electronic medical records from Helsinki, Turku and Tampere University Hospitals.
- Here, we report findings for patients diagnosed with incident recurrent or advanced EC receiving 1L platinum- and taxane-based treatments in the listed hospitals (LOT1-PT cohort).
- The study observation period started from availability of medication data (1 January 2010 for Turku and 1 January 2014 for Helsinki and Tampere) until the end of the study (30 June 2022) or death.
- Patient demographics, clinical characteristics, treatment patterns, HCRU data (outpatient visits, radiotherapy visits, inpatient admissions) and cost per patient are reported; cost calculations were based on the cost per day of inpatient episodes and cost per visit of outpatient visits.
- Data are presented for the overall LOT1-PT cohort and stratified by MMR status, where available.

Map of Finland

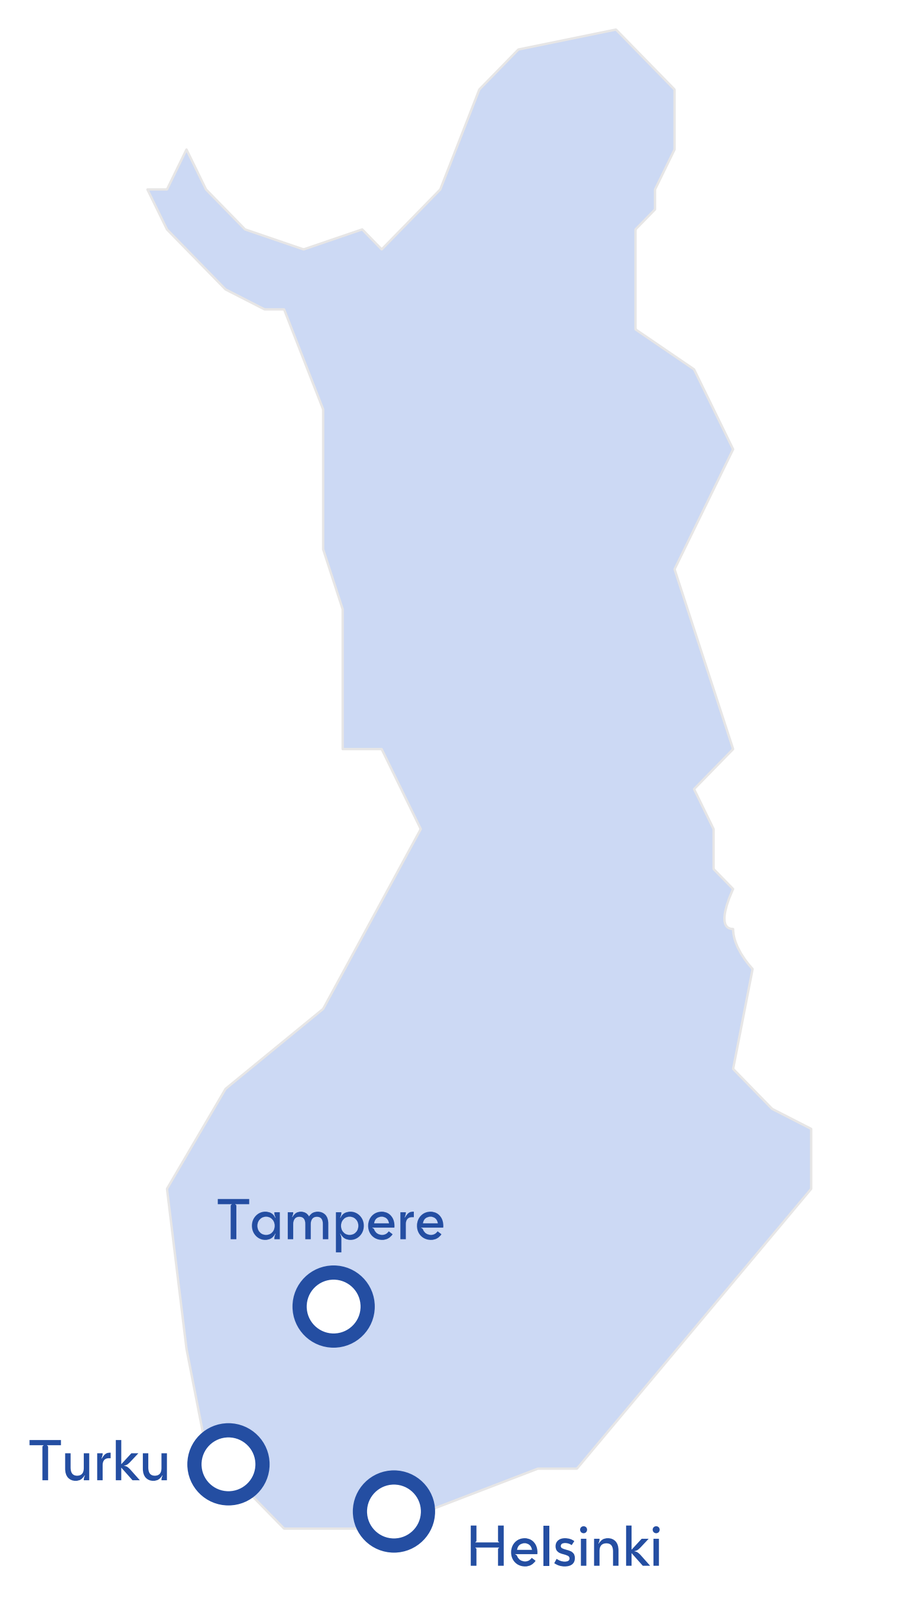
Tampere
Turku
Helsinki

Results

Baseline characteristics

- The FIRE LOT1-PT cohort included 266 patients with advanced or recurrent EC.
- Patient demographic and clinical characteristics are detailed in the **Table**.
- In the overall LOT1-PT population:
 - The mean (SD) age at diagnosis was 67 (9) years.
 - At study index date, 80.1% of patients had Stage III EC and 19.9% had Stage IV EC.
 - 86.1% of patients underwent surgery for EC (hysterectomy).
- In total, 62 patients (23.3%) in the overall LOT1-PT cohort had data on MMR status:
 - Among patients with available data on MMR status, 19 (30.6%) patients had dMMR tumours and 43 (69.4%) patients had MMRp tumours.
 - The observation that >30% of patients had dMMR tumours was likely a result of MMR status not being available for 204 (76.7%) patients.
 - The mean (SD) age at diagnosis was 64 (10) years for the dMMR group and 67 (10) years for the MMRp group.
 - Sixteen (84.2%) and 41 (95.3%) patients with dMMR and MMRp tumours underwent hysterectomy, respectively.

Table: Baseline characteristics for the LOT1-PT cohort and by MMR status

n (%)†	LOT1-PT (N=266)	LOT1-PT dMMR (n=19)	LOT1-PT MMRp (n=43)
Age at diagnosis (years), mean (SD)	67 (9)	64 (10)	67 (10)
Follow-up length (years), mean (SD)	4 (2.6)	3 (2.9)	2 (1.7)
Surgery for EC (hysterectomy)	229 (86.1)	16 (84.2)	41 (95.3)
Prior radiotherapy	9 (3.4)	0 (0.0)	<5 (<11.6)
Stage at index			
III	213 (80.1)	>12 (>63.2)	32 (74.4)
IV	53 (19.9)	<5 (<26.3)	11 (25.6)
MMR status			
MMRp	43 (69.4)‡	0 (0.0)	43 (100.0)
dMMR	19 (30.6)‡	19 (100.0)	0 (0.0)
Missing	204 (76.7)	0 (0.0)	0 (0.0)
Comorbidities			
Diabetes	21 (8.4)	<5 (<27.8)	<5 (<11.6)
Diabetes without complications	14 (5.6)	<5 (<27.8)	<5 (<11.6)

†Data presented as n (%) unless otherwise specified; ‡Percentage based on 62 patients with available data on MMR status.

Treatment patterns

- The most common treatments received in the 1L, 2L, 3L and 4L settings are shown in **Figure 1**.
 - The most common 2L treatment regimens were platinum + taxane chemotherapy (8.6% of patients in the LOT1-PT cohort), 'other' regimens such as gemcitabine and bevacizumab (8.3% of patients), and doxorubicin monotherapy (6.4% of patients).

HCRU

- During the first treatment year, there was an average (95% CI) of:
 - 24.5 (22.8–26.2) EC-related days with outpatient contact per patient year (dMMR: 28.4 [21.6–35.6]; MMRp: 29.1 [24.2–33.8]; **Figure 2**).
 - 11.9 (10.4–13.4) EC-related radiotherapy contacts per patient year (dMMR: 12.8 [6.4–20.2]; MMRp: 10.2 [6.5–14.3]; **Figure 3**).
 - 3.3 (2.5–4.1) inpatient days per patient year (dMMR: 5.5 [2.4–9.8]; MMRp: 2.8 [1.3–4.7]; **Figure 4**).
 - Inpatient utilisation was driven by patients with dMMR tumours in Years 1 and 2.
 - Of note, data on inpatient days during Years 4 and 5 reflect patients whose MMR status was unknown.

Figure 1: Treatment patterns in the LOT1-PT cohort

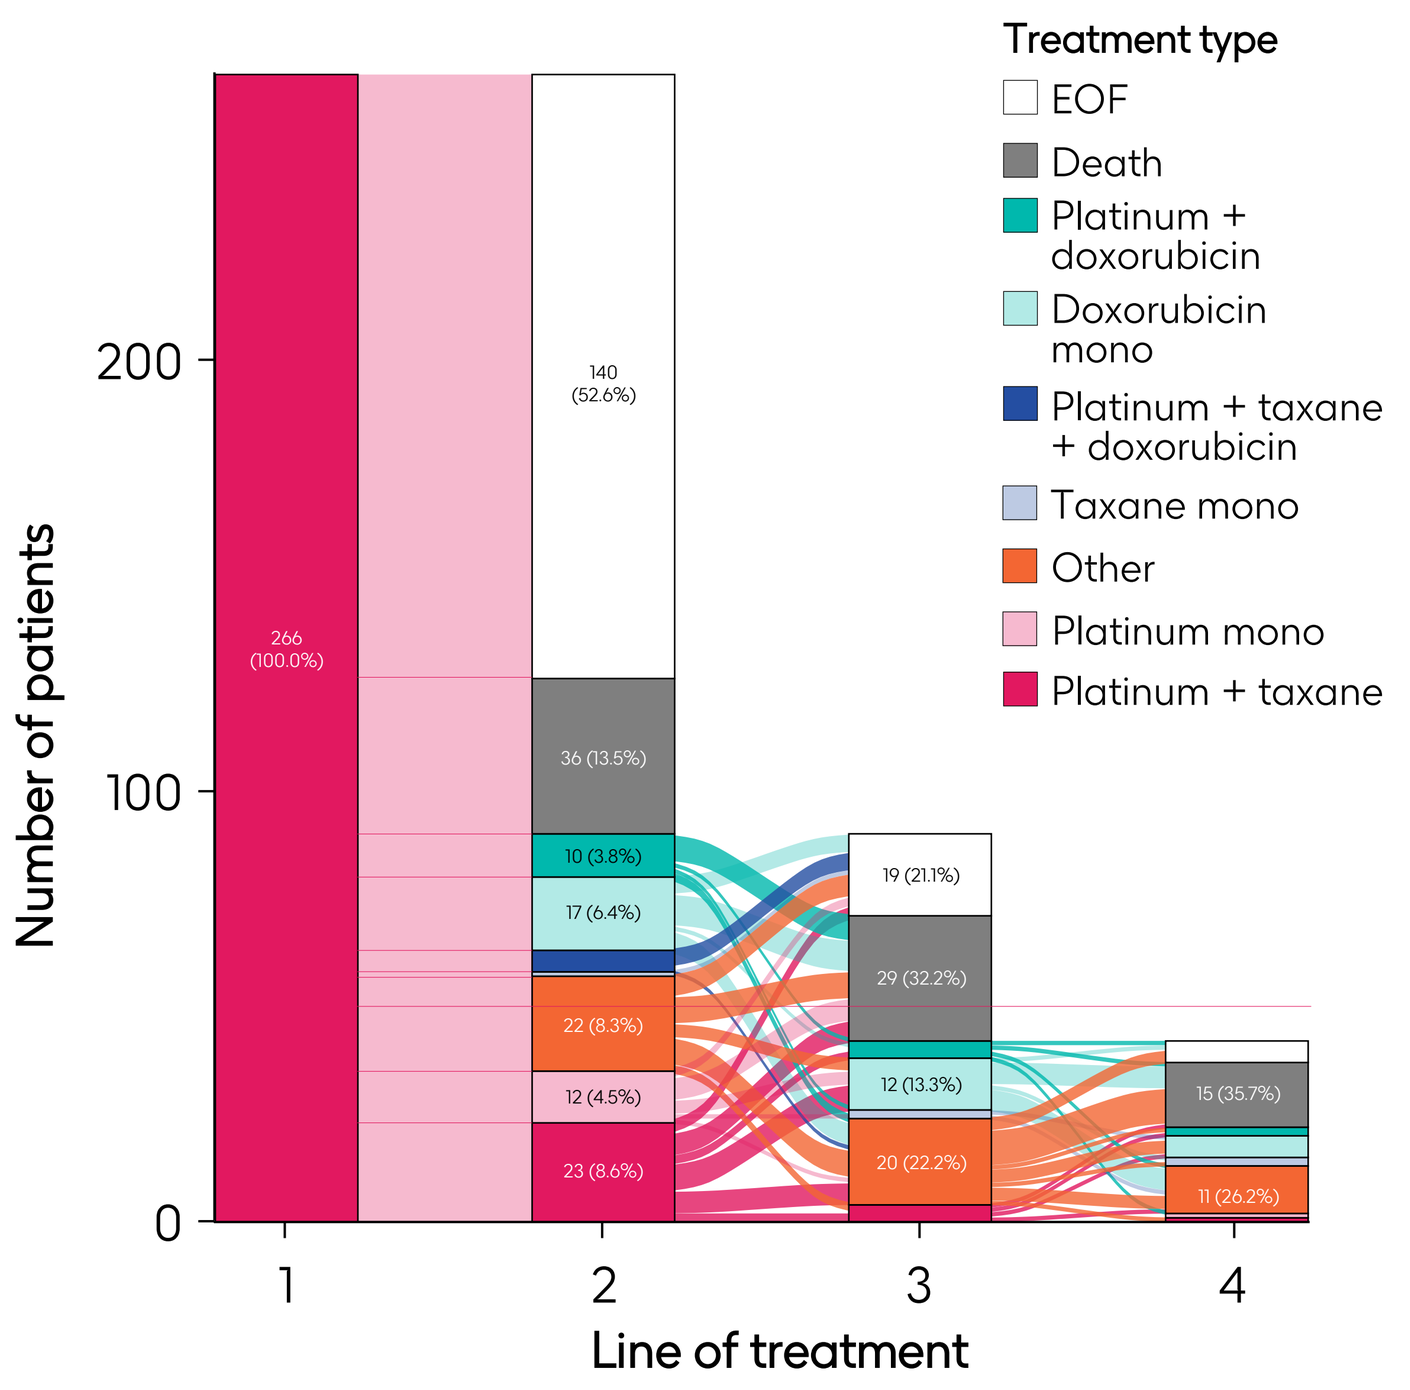
Treatment type
EOF
Death
Platinum + doxorubicin
Doxorubicin mono
Platinum + taxane + doxorubicin
Taxane mono
Other
Platinum mono
Platinum + taxane
Number of patients
Line of treatment

Figure 2: Outpatient contact days per year in the LOT1-PT cohort and by MMR status

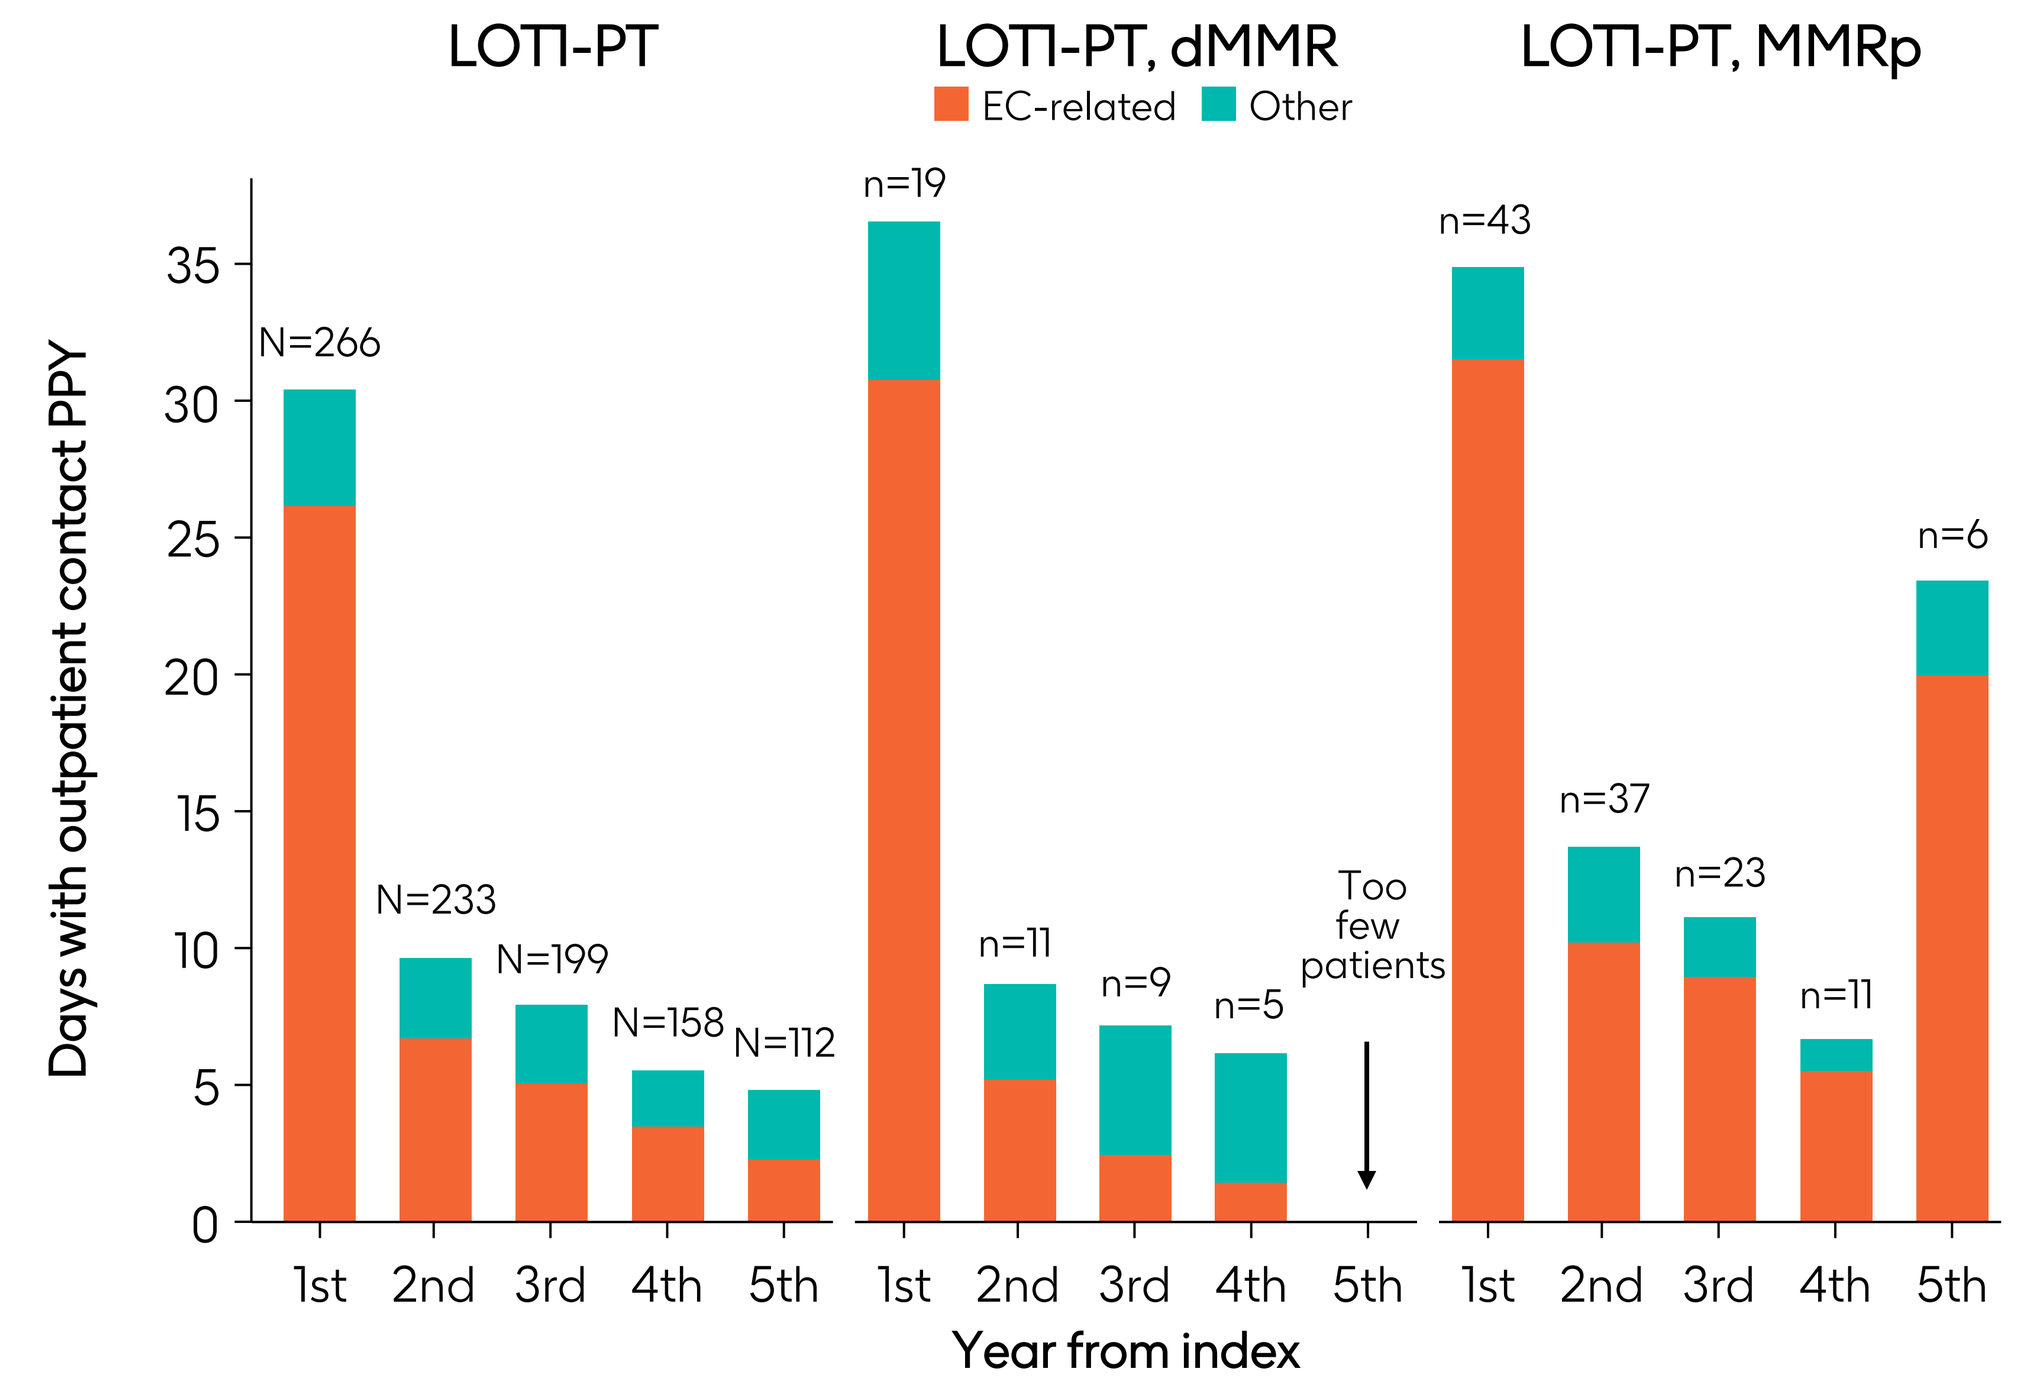
LOT1-PT
LOT1-PT, dMMR
LOT1-PT, MMRp
EC-related
Other
Days with outpatient contact PPY
Too few patients
Year from index

Figure 3: Radiotherapy contacts per year in the LOT1-PT cohort

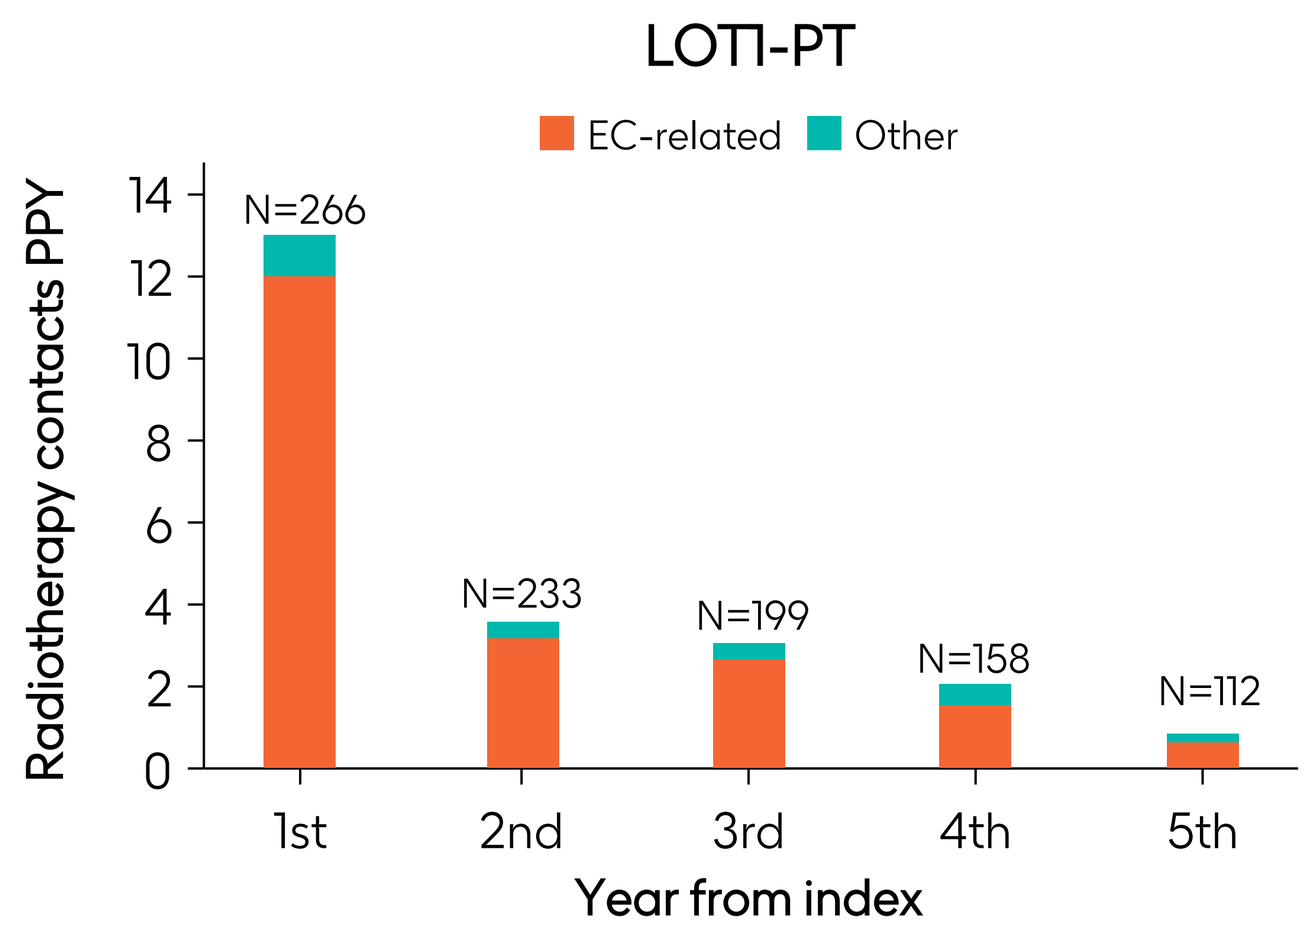
LOT1-PT
EC-related
Other
Radiotherapy contacts PPY
Year from index

Figure 4: Inpatient days per year in the LOT1-PT cohort and by MMR status

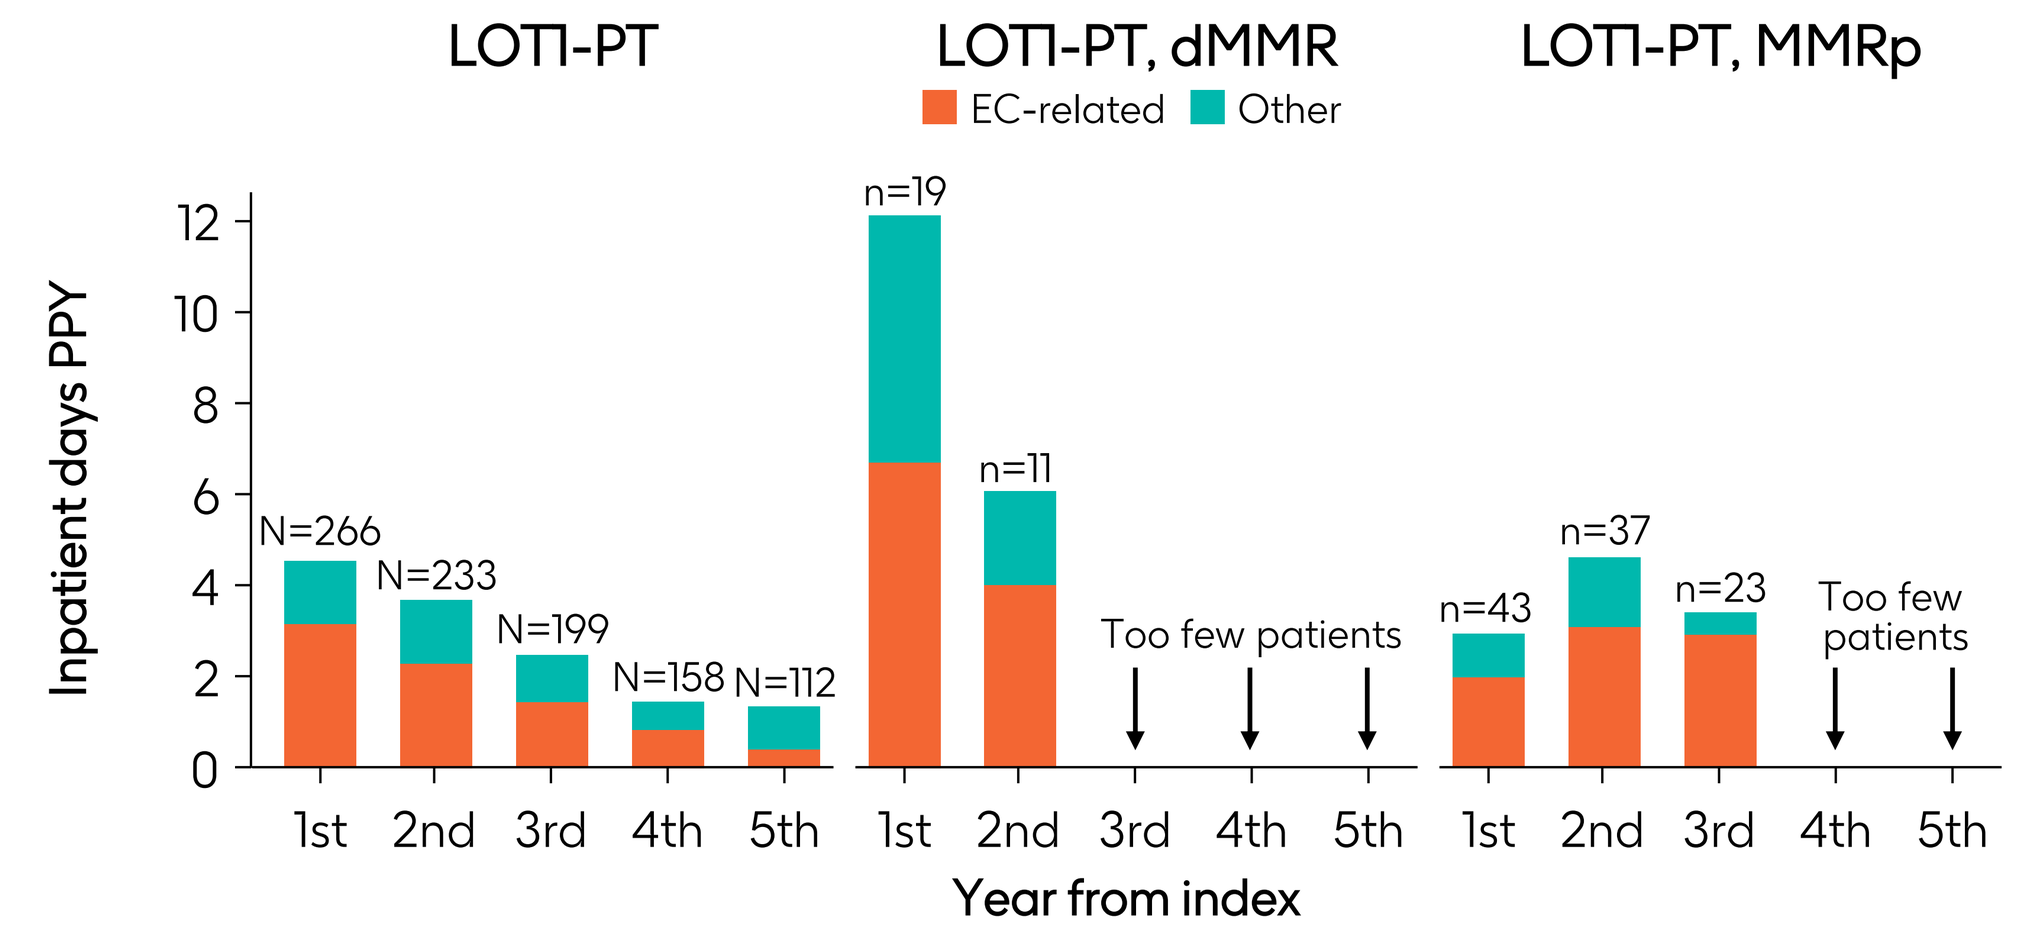
LOT1-PT
LOT1-PT, dMMR
LOT1-PT, MMRp
EC-related
Other
Inpatient days PPY
Too few patients
Year from index

Cost

- Average (95% CI) total EC-related cost of all treatment resources was €5973 (€5478–6532) per patient year (dMMR: €8069 [€5642–12,802]; MMRp: €10,383 [€8466–12,676]; **Figure 5**).
 - Average total EC-related costs per patient year over Years 1–5 are shown in **Figure 6**.
- In the first year of follow-up, the average cost per patient year was €10,783 (€10,000–11,590; dMMR: €12,513 [€9460–15,706]; MMRp: €13,165 [€10,781–15,609]) for outpatient contacts, €4466 (€3856–5087; dMMR: €4940 [€2286–8017]; MMRp: €3930 [€2421–5587]) for radiotherapy contacts and €843 (€664–1038; dMMR: €1133 [€532–1895]; MMRp: €552 [€299–858]) for hospitalisations.
- Average costs per patient year decreased in the second year to €2666 (€2153–3219; dMMR: €2073 [€778–3927]; MMRp: €3530 [€2076–5324]) for outpatient contacts, €1043 (€829–1285; dMMR: €1414 [€476–2896]; MMRp: €1429 [€542–2534]) for radiotherapy contacts and €437 (€278–627; dMMR: €1552 [€131–3397]; MMRp: €555 [€214–974]) for hospitalisations.

Figure 5: Average total cost of treatment in the LOT1-PT cohort and by MMR status

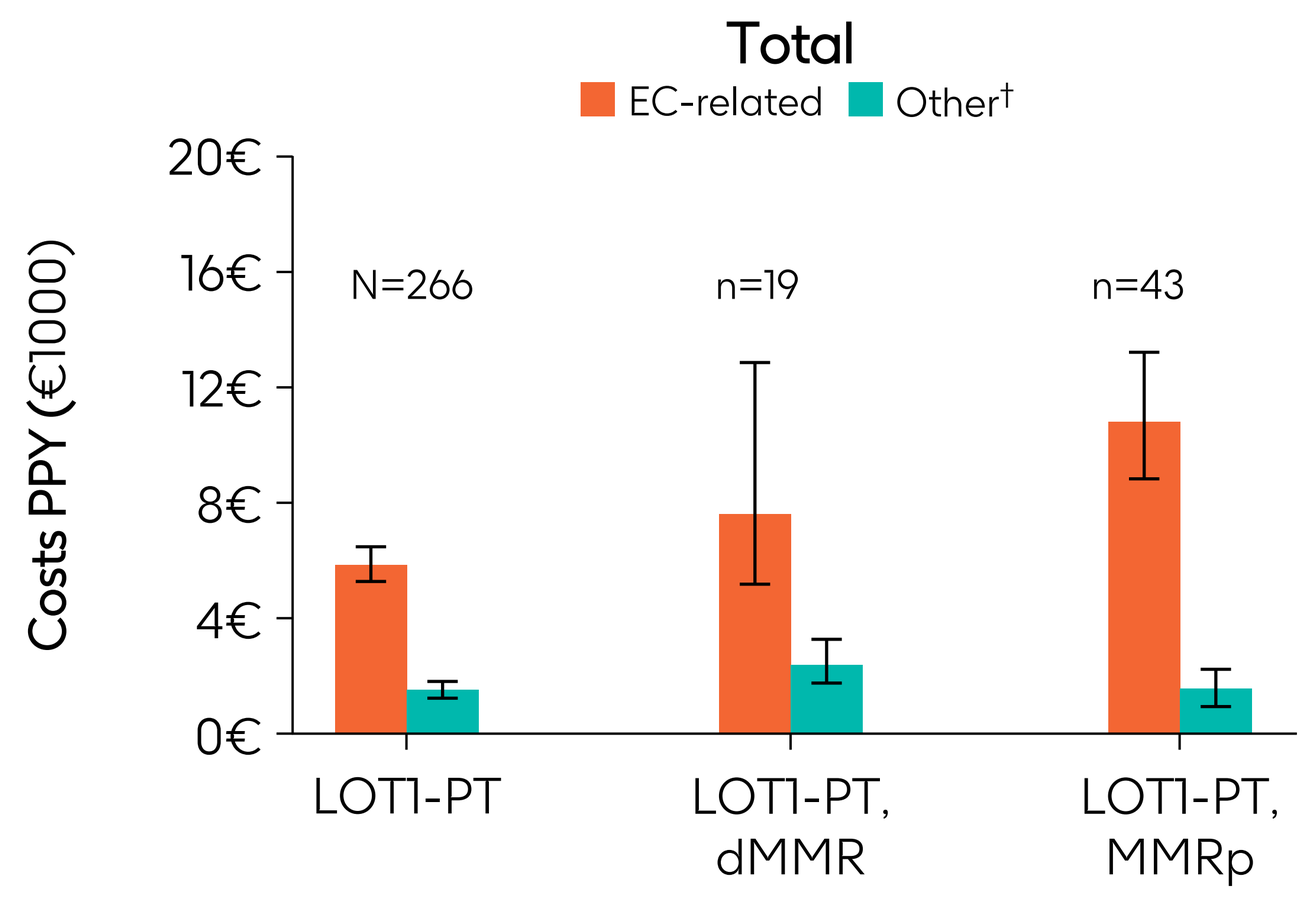
Total
EC-related
Other†
Costs PPY (€1000)
LOT1-PT
LOT1-PT, dMMR
LOT1-PT, MMRp

†Includes treatment costs not specifically related to EC.

Figure 6: Average total costs per year in the LOT1-PT cohort and by MMR status

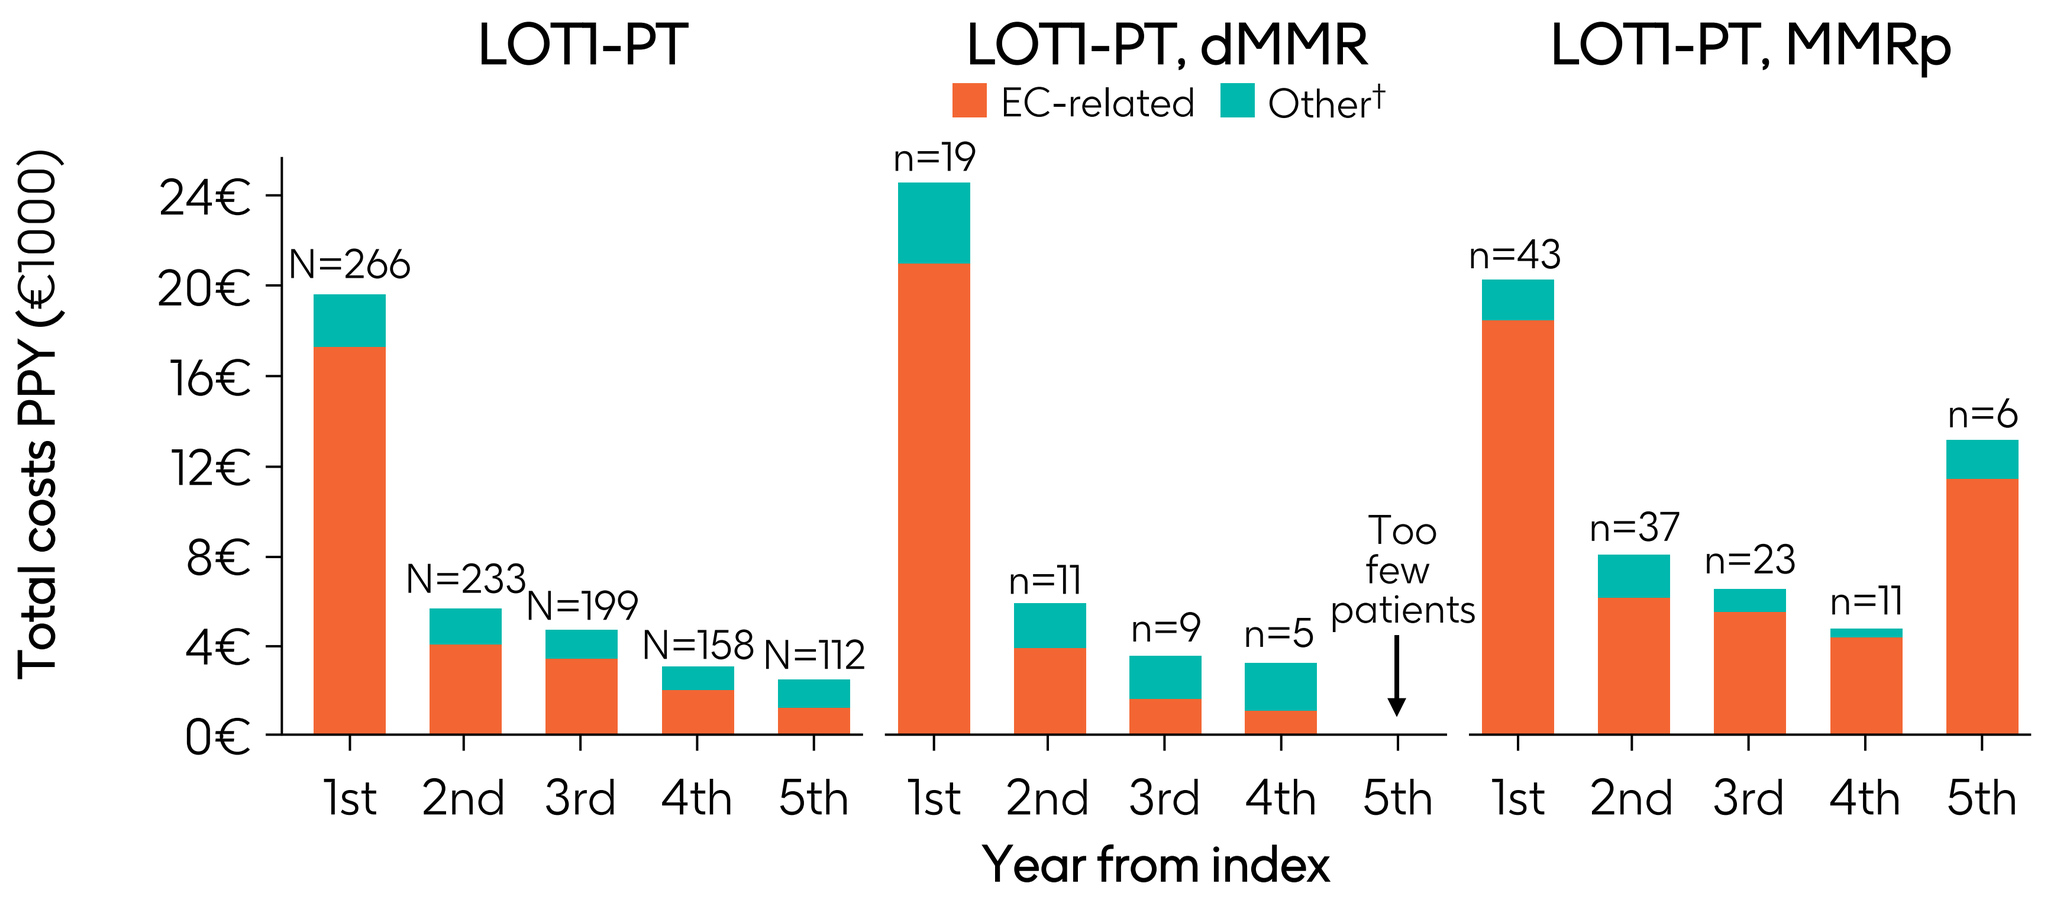
LOT1-PT
LOT1-PT, dMMR
LOT1-PT, MMRp
EC-related
Other†
Total costs PPY (€1000)
Too few patients
Year from index

†Includes treatment costs not specifically related to EC.

Conclusions

EC treatment patterns in Finland were generally aligned with ESMO treatment guidelines at the time of the study.

EC-related HCRU and healthcare costs were highest in the first year after diagnosis in Finland and decreased in subsequent years.

- Increased HCRU and costs in the first year of treatment have also been noted with other gynaecological cancers in Finland.[6]

We observed differences in HCRU and costs between patients with dMMR and MMRp tumours, suggesting higher total costs in patients with MMRp tumours.

- However, MMR testing was not recommended in Finland during the earlier years of the study, so MMR status was only available for a minority of patients; therefore, it is not possible to draw conclusions based on these findings.
- When MMR status was reported in electronic medical records, retrieval of these data was difficult, due to inconsistencies in the format of reporting across hospitals.

Since this study was conducted, the treatment landscape for EC has evolved towards targeted therapy with PD-(L)1 inhibitors.

- Our findings highlight the importance of consistent and standardised reporting of biomarker data (especially MMR status) within datasets, as modern targeted oncology treatments rely on precision medicine and molecular profiling.

Acknowledgements

Writing and editorial support, under the direction of the authors, were provided by Martina Kusi-Mensah, PharmD, Francesca Murphy, BA, and Ian Norton, PhD, all of Scion (a division of Prime, London, UK). Writing and editorial support provided by Prime was funded by GSK. This study was sponsored by GSK (study 216979).

Conflicts of interest/disclosures

J.A., K.K. and S.T. are employed by Medaffcon Oy (Medaffcon OY received payment for study planning and execution from GSK). B.M., J.I., S.T. and D.S. are employed by GSK and hold financial equities in GSK. S.K. was employed by GSK at the time of the study. S.H. and H.L declare consulting fees or advisory board participation from GSK, MSD, Eisai and AstraZeneca. A.A. declares advisory board participation for GSK, MSD and AstraZeneca.

Abbreviations

1L, first line; 2L, second line; 3L, third line; 4L, fourth line; CI, confidence interval; dMMR, deficient mismatch repair; DNA, deoxyribonucleic acid; EC, endometrial cancer; EOF, end of follow-up; ESMO, European Society for Medical Oncology; FINGOG, Finnish Gynaecological Oncology Specialists; HCRU, healthcare resource utilisation; IHC, immunohistochemistry; LOT, line of treatment; MMR, mismatch repair; MMRp, mismatch repair proficient; mono, monotherapy; PD-(L)1, programmed cell death protein (ligand)-1; PPY, per patient year; PT, platinum + taxane; SD, standard deviation.

References

1. Colombo N, et al. *Ann Oncol.* 2013;24 Suppl 6:VI33–VI38; 2. Kohdunrungon syöpä. In: Kaikki syövästä. https://www.kaikkisyovasta.fi/tietoasyovasta/ syopataudit/kohdunrungon-syopa/. Accessed November 2024; 3. Lee V, et al. *Oncologist.* 2016;21:1200–1211; 4. Luchini C, et al. *Ann Oncol.* 2019;30:1232–1243; 5. FINGOG työryhmä. 2022. Gynekologisen syövän KELPO HOITO. https://gynekologiyhdistys.fi/fingog-kelpo-hoito-suositus-2024-on-julkaistu/. Accessed November 2024; 6. Mascialino B, et al. Poster EE196 presented at ISPOR Europe, Barcelona, Spain, 17–20 November 2024.

Presented at the Professional Society for Health Economics and Outcomes Research (ISPOR) Europe Meeting; 17–20 November 2024; Barcelona, Spain.

Presenting author:
Barbara Mascialino, barbara.x.mascialino@gsk.com