THE HIDDEN BURDEN OF MIGRAINE: ANALYSIS USING A FISCAL FRAMEWORK IN COLOMBIA

EE249

Rui Martins[a,b], Zrinka Orlovic[a,c], Ana Teresa Paquete[a], Juan Sebastian Estrada[d], Juliana Villarreal Ramírez[d], Juan Manuel Reyes-Sánchez[d], Jair Arciniegas[d], Natalia Castaño[d]

[a]Global Market Access Solutions, St-Prex, Switzerland; [b]University Medical Center Groningen, University of Groningen, Groningen, The Netherlands; [c]Faculty of Economics and Business – Zagreb, University of Zagreb; [d]Pfizer Colombia

INTRODUCTION

- Migraine is a highly prevalent and incapacitating neurological disorder often underdiagnosed and undertreated. Consequently, despite leading to considerable disability and lost productivity, migraine is often associated with relatively low healthcare costs[1].
- Focusing on direct healthcare costs alone undermines the true economic burden of migraine[2].
- This study adapted a published modelling framework previously used to inform health policy in Sweden[3] and the UK[4].

OBJECTIVE

- To assess the economic burden of migraine from the perspective of the Colombian government.

METHODS

- The model simulated the annual fiscal consequences of migraine in the entire Colombian population. It captured tax revenue and transfer payments (disability benefits and public healthcare expenditure) – Figure 1.
- This was achieved by applying migraine prevalence[5] to age and sex-specific demographic distributions[6]. Inputs informing labor market participation[7], absenteeism losses[8], and healthcare cost[9,10] were extracted from a recent published Colombian study and official databases. All costs were expressed in 2022 Colombian pesos (COP).
- The impact of migraine on employment status and absenteeism was sourced from international peer-reviewed publications (proxy) given that Colombian-specific data were unavailable. Relative risks were extracted for employment[11,12] and disability[13]. Annual workdays missed were used to estimate absenteeism[14].
- The fiscal burden associated to the entire cohort with prevalent migraine was reported as annual incremental fiscal consequences to the Colombian government.

Figure 1. Model Structure

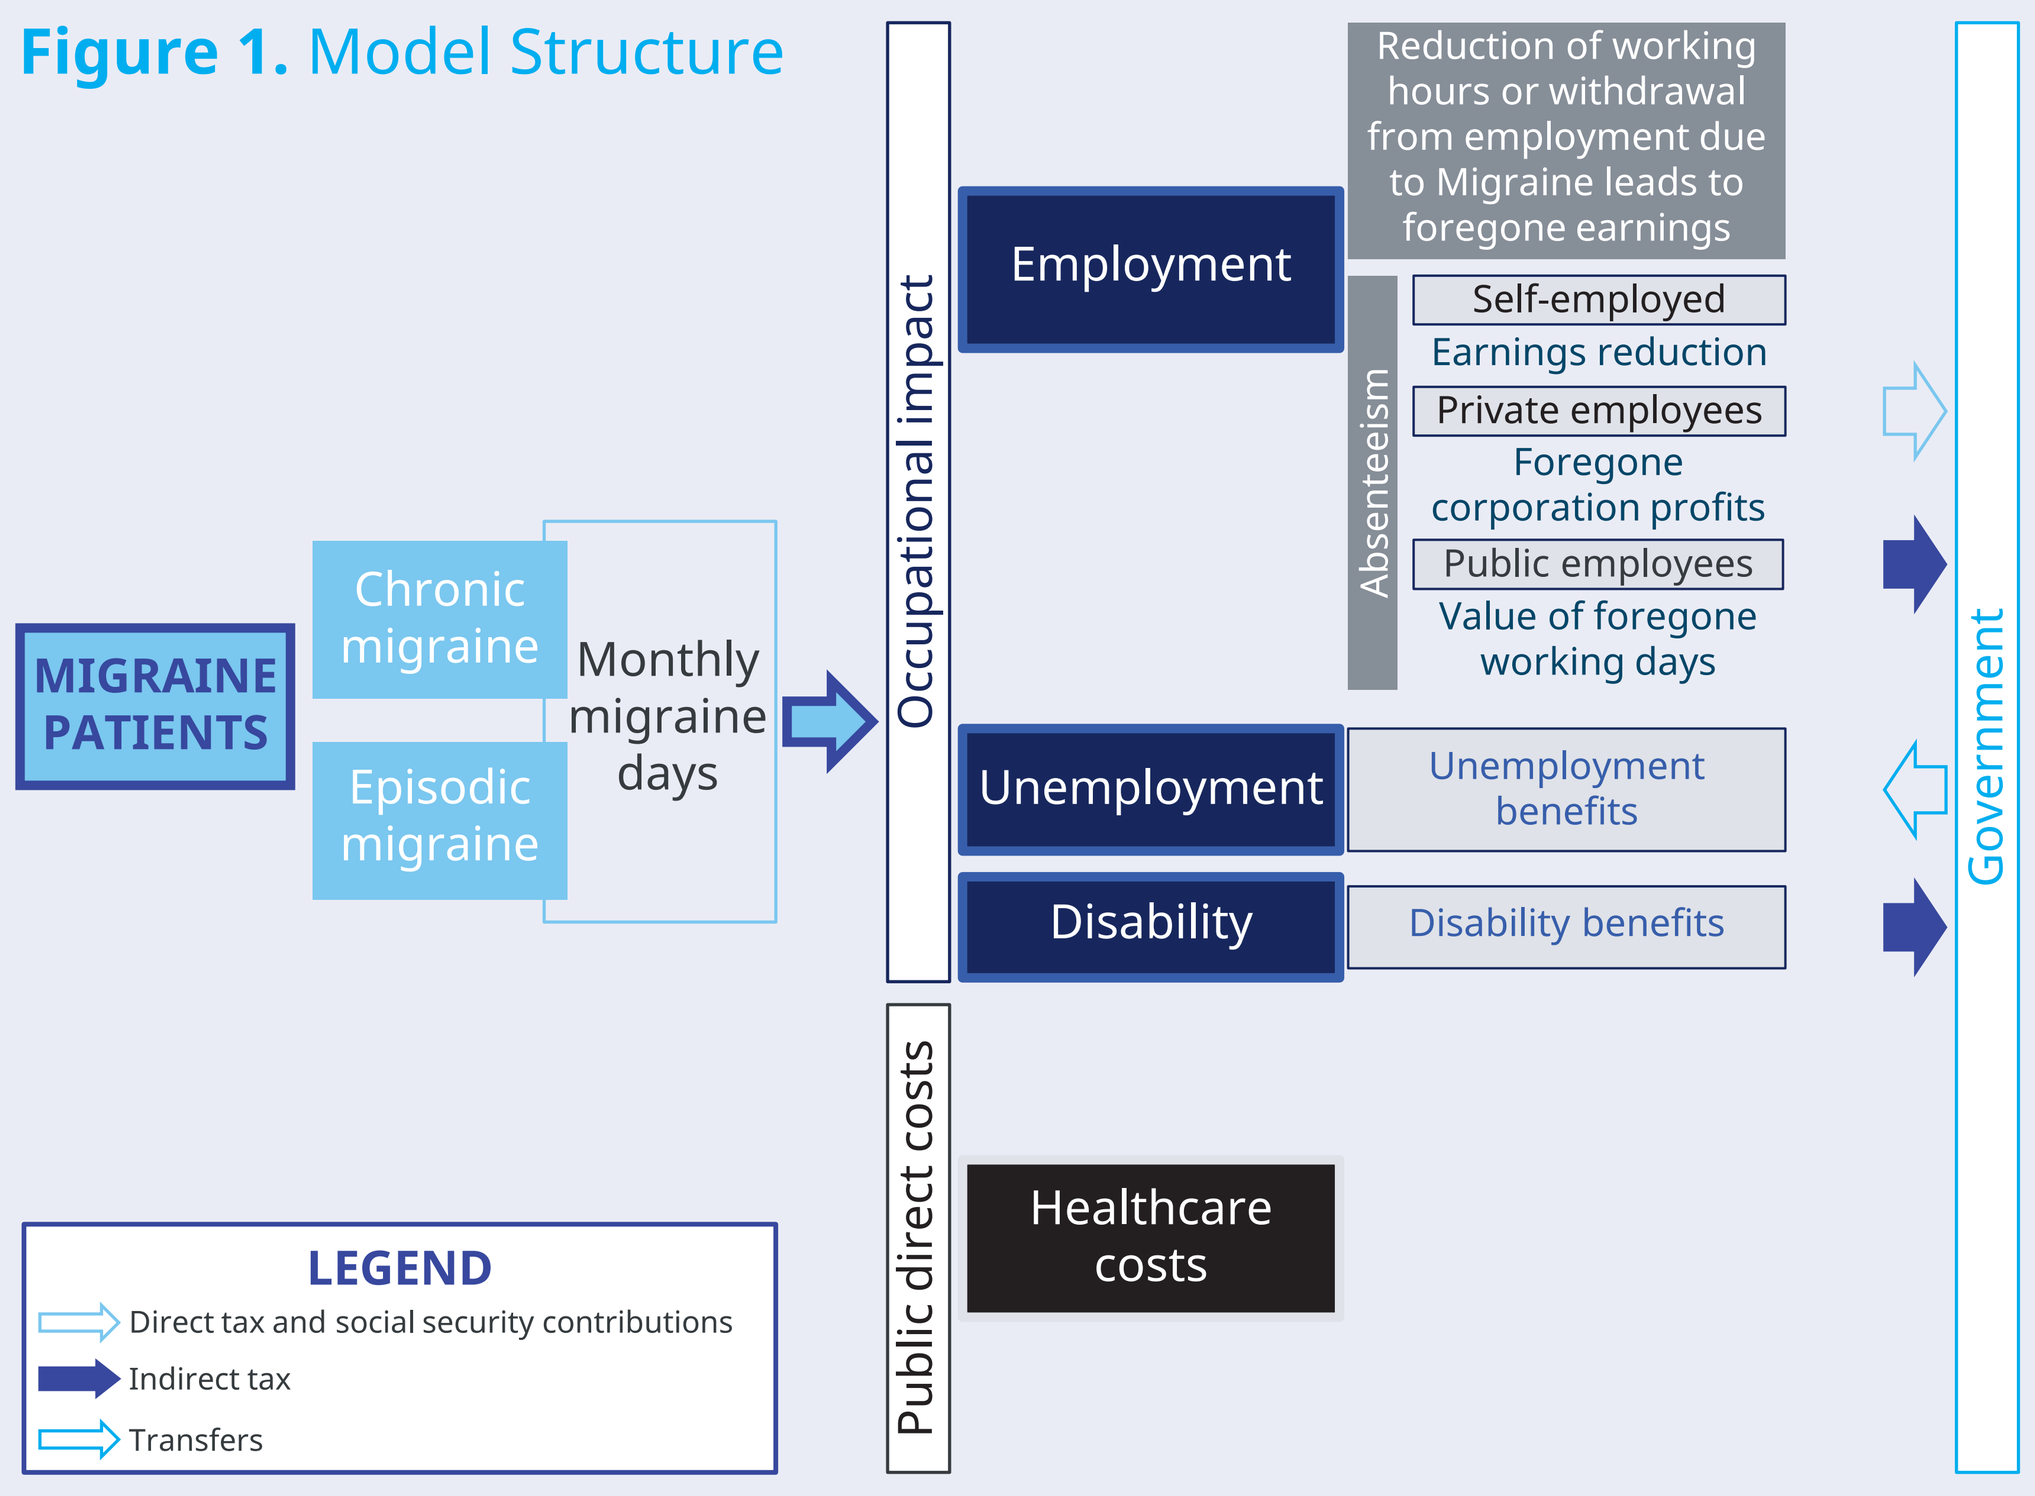
MIGRAINE PATIENTS
Chronic migraine
Episodic migraine
Monthly migraine days
Occupational impact
Public direct costs
Employment
Unemployment
Disability
Healthcare costs
Reduction of working hours or withdrawal from employment due to Migraine leads to foregone earnings
Absenteeism
Self-employed
Earnings reduction
Private employees
Foregone corporation profits
Public employees
Value of foregone working days
Unemployment benefits
Disability benefits
Government
LEGEND
Direct tax and social security contributions
Indirect tax
Transfers

RESULTS

- A total of 813,208 people aged 18 years and older were estimated to be affected by migraine annually, 84.9% of all migraine patients are females. The model predicted that these people would experience 7,426,448 migraine days, including both episodic and chronic migraine (Figure 2).

Figure 2. Estimated prevalence of migraine in Colombia

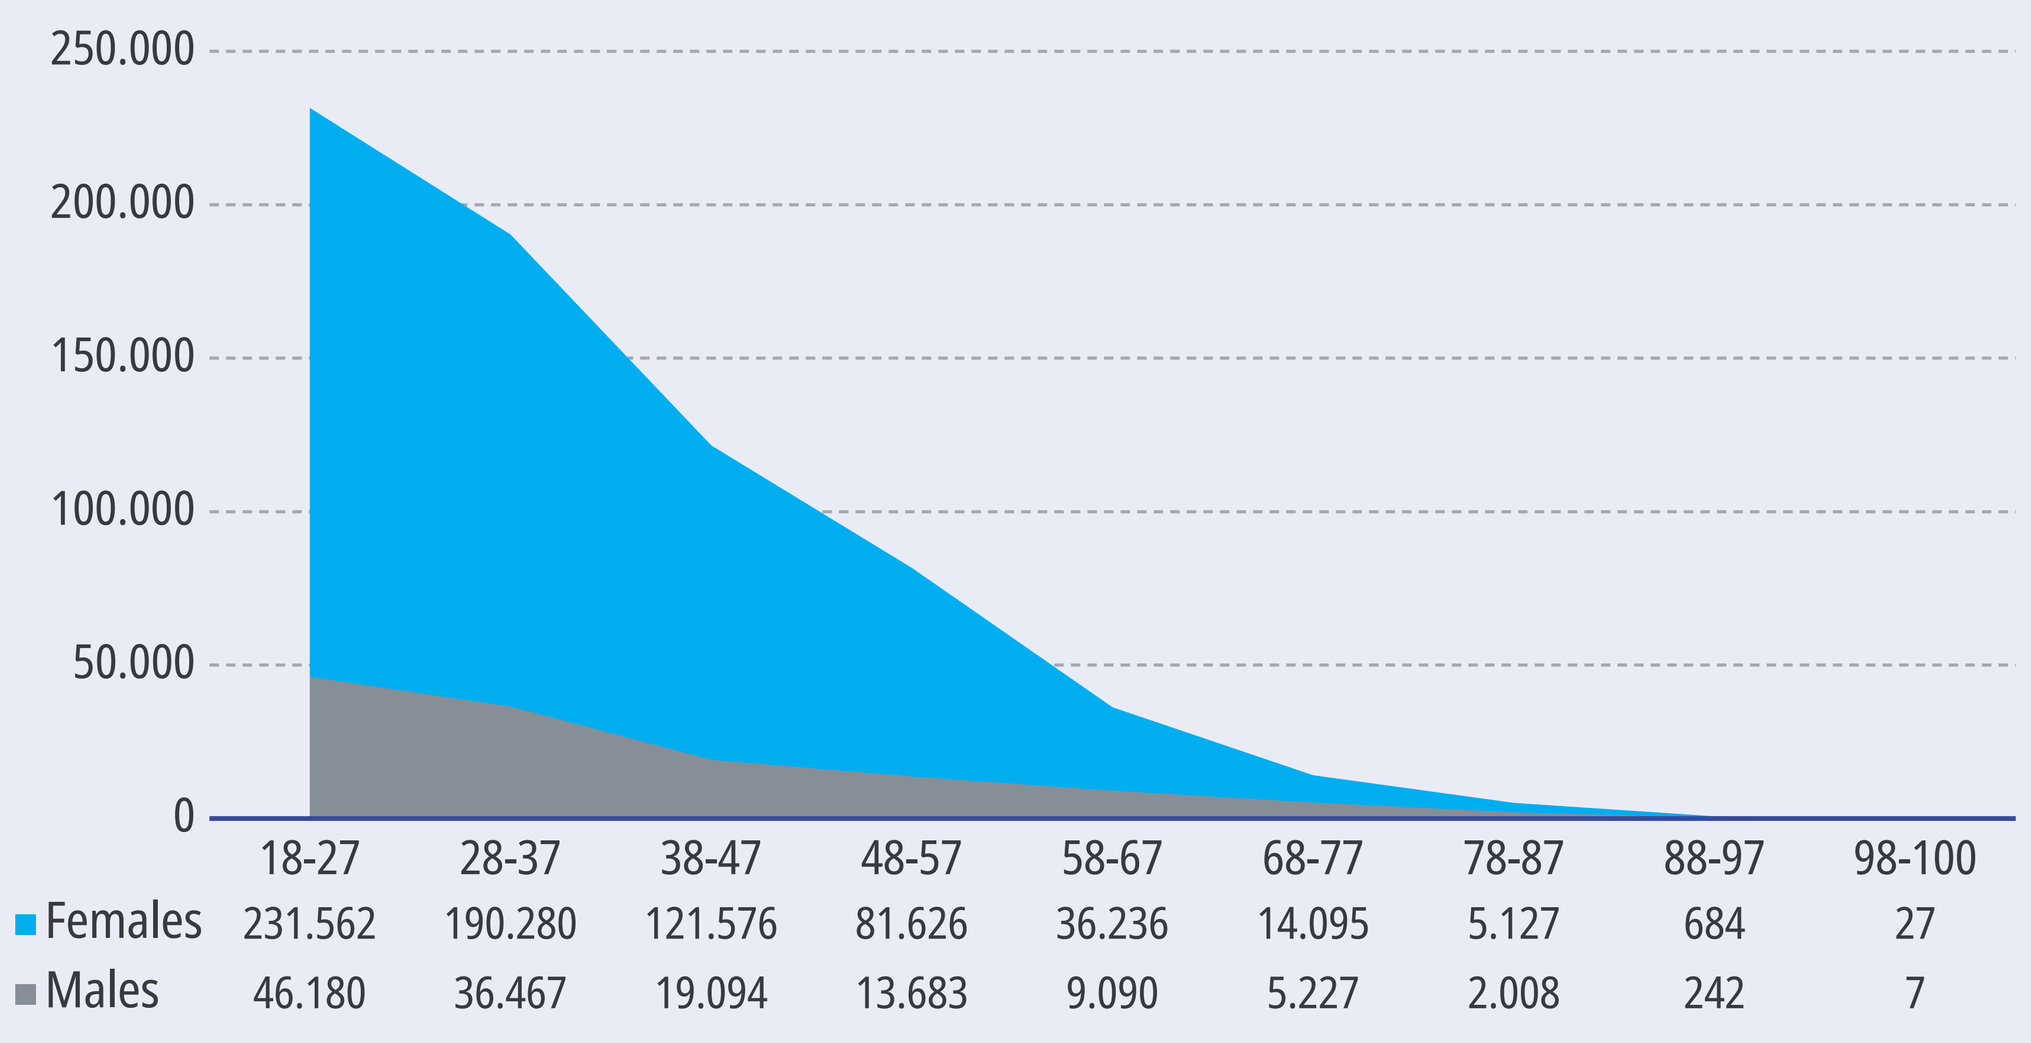
250.000
200.000
150.000
100.000
50.000
0
18-27 28-37 38-47 48-57 58-67 68-77 78-87 88-97 98-100
Females 231.562 190.280 121.576 81.626 36.236 14.095 5.127 684 27
Males 46.180 36.467 19.094 13.683 9.090 5.227 2.008 242 7

- The fiscal burden of migraine in the entire population was estimated to cost the Colombian government 245,667 M COP in 2022. The burden was mostly due to forgone taxes (62.0%), while 19.2% was due to productivity losses in the public sector and 14.8% was due to public healthcare costs (Table 1).

Table 1. Fiscal perspective: incremental consequences of migraine in the adult population with prevalent migraine (COP, millions).

	MIGRAINE POPULATION	GENERAL POPULATION	INCREMENTAL
Public sector absenteeism	-68,315	-21,032	-47,283
Direct taxes from employment	484,778	509,920	-25,142
Indirect taxes from employment	1,048,645	1,103,032	-54,387
Foregone corporation taxes	-105,068	-32,348	-72,720
Unemployment allowances	-108,766	-96,365	-12,400
Disability pension	-5,459	-5,147	-312
Taxes from transfers	584,601	581,547	3,055
Healthcare costs	–36,478	0	–36,478
TOTAL	**1,793,939**	**2,039,606**	**-245,667**

* Negative values express costs to the government, and positive values express benefits to the government's accounts.

- Chronic migraine accounts for 7.6% of all cases, with a total of 61,463 cases. The financial impact of the chronic migraine cohort is estimated to be approximately 65,401 M COP, an average of 1,064,074 COP per chronic migraine patient.
- Total annual gross income losses to the cohort with prevalent migraine was estimated to be 311,388M COP, an average of 382,913 COP per person with migraine. Total healthcare costs (public and private) attributed to the population with prevalent migraine 47,102M COP (Table 2).
- The overall fiscal burden was equivalent to 0.02% of Colombian gross domestic product[15] and 0.3% of the annual public healthcare spending[15].

Table 2. Societal perspective: incremental consequences of migraine in the adult population with prevalent migraine (COP, millions).

	MIGRAINE POPULATION	GENERAL POPULATION	INCREMENTAL
Gross income from any employment	**6,003,964**	**6,315,352**	**-311,388**
Private employees	2,997,603	3,063,842	-66,239
Public employees	682,167	697,233	-15,066
Self-employed	2,324,194	2,554,278	-230,083
Total healthcare costs	**47,102**	**0**	**-47,102**
Government/ compulsory schemes	-36,478	0	-36,478
Private Insurance	-3,792	0	-3,792
Out-of-pocket	-6,833	0	-6,833
TOTAL	**5,956,862**	**6,315,352**	**-358,490**

* Negative values express societal losses, and positive values express societal benefit

CONCLUSION

- This fiscal analysis monetizes the direct and indirect economic burden of migraine to the Colombian government, providing a valuable insight to decision-makers. The findings highlight important consequences of migraine to the labour market, especially in terms of reduced productivity and personal income losses due to foregone employment.

DISCUSSION

- It is recommended that when planning public health programs and setting priorities for people living with migraine, timely and accurate diagnosis, and appropriate treatment are implemented. Focus should be given to people living with chronic migraine, who experience the higher disease burden.

REFERENCES

1. Steiner TJ, Stovner LJ, Jensen R, Uluduz D, Katsarava Z. 2020. Migraine remains second among the world's causes of disability, and first among young women: findings from GBD2019. J Headache Pain. 2020;21(137). 2. Hansson-Hedblom, A., Axelsson, I., Jacobson, L., Tedroff, J., & Borgström, F. 2020. Economic consequences of migraine in Sweden and implications for the cost-effectiveness of onabotulinumtoxinA (Botox) for chronic migraine in Sweden and Norway. The journal of headache and pain, 21, 1-15. 3. Martins, R., Jakobsson, M., Surmay, G., & Connolly, M. 2023. EE659 Estimating the Economic Consequences of Migraine in Sweden: A Fiscal Model Using the Government Perspective of Costs. Value in Health, 26(12), S180. 4. Martins, R., Large, S., Russell, R., Surmay, G., & Connolly, M. P. 2023. The Hidden Economic Consequences of Migraine to the UK Government: Burden-of-Disease Analysis Using a Fiscal Framework. Journal of Health Economics and Outcomes Research, 10(2), 72. 5. Bolaños-López, Arciniegas JA, Rubio A, Gonzalez FJ. et al. 2024. Treatment patterns and healthcare resource utilization in Colombian migraine patients from 2018 to 2022. Value in health. 27 (6), S1 (June 2024). 6. National Administrative Department of Statistics, DANE. Demografía y población. [Internet]. Available from: https://www.dane.gov.co/index.php/estadisticas-por-tema/demografia-y-poblacion 7. ILOSTAT, Employment by gender, age and working time arrangement, 2022. [Internet]. Available from: https://rshiny.ilo.org/dataexplorer7/?lang=en&id=SDG_0552_NOC_RT_A. 8. Ministry of Health and Social Protection. 2024. Sala situacional de las Personas con Discapacidad, Ministerio de Salud y Protección Social. [Internet]. Available from:https://www.sispro.gov.co/observatorios/ondiscapacidad/Paginas/Sala-Situacional-Discapacidad-en-Colombia.aspx. 9. Ministry of Health and Social Protection. 2024. Technical Note of the sufficiency study: 2023. [Internet]. Available from: Unidad de pago por capitación (UPC): Estudios de suficiencia. Available from: https://www.minsalud.gov.co/salud/POS/Paginas/unidad-de-pago-por-capitacion-upc.aspx. 10. Ministry of Health and Social Protection. SISMED - Sistema de Información de Precios de Medicamentos [Internet]. Available from: https://www.sispro.gov.co/centralprestadores-de-servicios/Pages/SISMED-Sistema-de-Informacion-de-Precios-de-Medicamentos.aspx. 11. Stewart WF, Wood GC, Manack A, Varon SF, Buse DC, Lipton RB. 2010. Employment and work impact of chronic migraine and episodic migraine. J Occup Environ Med. 52(1):8-14. 12. Buse, DC, Reed, ML, Fanning, KM, Bostic, R, Dodick, DW, Schwedt, TJ, Munjal, S., Singh, P and Lipton, RB (2020). Comorbid and co-occurring conditions in migraine and associated risk of increasing headache pain intensity and headache frequency: results of the migraine in America symptoms and treatment (MAST) study. The Journal of Headache and Pain, 21, 23. 13. Buse DC, Manack A, Serrano D, Turkel C, Lipton RB. 2010. Sociodemographic and comorbidity profiles of chronic migraine and episodic migraine sufferers. J Neurol Neurosurg Psychiatry.81(4):428-432. 14. Vo, P, Fang, J, Bilitou, A, Laflamme, AK, & Gupta, S (2018). Patients' perspective on the burden of migraine in Europe: a cross-sectional analysis of survey data in France, Germany, Italy, Spain, and the United Kingdom. The journal of headache and pain, 19, 1-11. 15. Organization for Economic Co-operation and Development. 2024. Annual GDP and components - expenditure approach [Internet]. Available from: https://data-explorer.oecd.org.

Study funding statement: This study was sponsored by Pfizer · Poster presented at ISPOR Europe, Barcelona Spain, 17-20 November, 2024.