PCR20

ColdU

Content Validity of Patient Reported Outcome Measures Evaluating Symptoms and Impact in Patients with Chronic Inducible Cold Urticaria

Ashna Alladin[1], Isabelle Guillemin[2], Rebeca Oliván[3], Jingdong Chao[4], Jérôme Msihid[5], Renata Martincova[6], Efstathios Zikos[5], Lydia Braham-Chaouche[5], Marieke Krol[7], Chien-chia Chuang[8], Ella Brookes[9]

[1]IQVIA, Patient Centered Solutions, Munich, Germany; [2]IQVIA, Patient Centered Solutions, Lyon, France; [3]IQVIA, Patient Centered Solutions, Barcelona, Spain; [4]Regeneron Pharmaceuticals, Inc, Tarrytown, NY, USA; [5]Sanofi, Gentilly, France; [6]Sanofi, Prague, Czech Republic; [7]IQVIA, Amsterdam, Netherlands; [8]Sanofi, Health Economics and Value Assessment, Cambridge, MA, USA; [9]Sanofi, Reading, UK

Introduction

- Chronic inducible cold urticaria (ColdU) is characterized by itchy wheals (hives), angioedema, burning, and pain localized in response to cold exposure and has severe impact on patients' health related quality of life (HRQoL).[1-3]
- In most patients with ColdU, the standard dose of antihistamine (first-line treatment), is insufficient for complete protection indicating the necessity for a more effective therapy.[1]
- A Phase 3 clinical study (LIBERTY-CINDU CUrIADS - NCT04681729) evaluated the efficacy and safety of dupilumab in patients with ColdU.
- The following patient-reported outcomes measures (PROMs) were collected: Cold Urticaria Activity Score (ColdUAS), Urticaria Control Test (UCT), Dermatology Life Quality Index (DLQI), Children DLQI (CDLQI), and Infant's Dermatitis Quality of Life (IDQOL).

Methods

Study Design and Population

- This qualitative interview study included adults aged 18-80 years, adolescents aged 12-17 years, and children aged 4-11 years (dyad/caregiver only) from the United States (US) with a confirmed diagnosis of ColdU.
- The included patients must have a documented medical history of cold exposure-triggered urticaria within past 6 months, necessitating a visit to a healthcare provider or the use of prescription medication for ColdU symptoms during the episode.
- One-to-one cognitive debriefing (CD) interviews were conducted by telephone with the participants about their understanding of instruction/item, response options, and recall period of following PROMs: ColdUAS, UCT, DLQI, CDLQI, and IDQOL.

Objective

- **We determined the content validity of PROMs used in the Phase 3 study using a qualitative interview study.**

Conclusions

- **Content validity of ColdUAS, UCT, and DLQI/CDLQI was demonstrated, indicating that they could effectively inform key ColdU symptoms, disease activity and impact- related efficacy endpoints in clinical trials.**
- **Caregiver of a 2-year-old child had difficulty in understanding and relating to IDQOL which needs further investigation to assess its content validity.**

Results

Demographic and clinical characteristics

- Eight adults, five adolescents, six children with their caregivers, and one caregiver of a 2-year-old child were interviewed.
 - Participants were predominantly female (60-100%) and White (60-100%).
 - Mean time since diagnosis of ColdU was longer among adults (93 months) and adolescents (108 months) versus children (10 months) **(Table 1)**.
 - H1-antihistamines was the most common current (adults, n=1; adolescents, n=1; children, n=3) and previous treatment (adults, n=4; adolescents, n=3; children, n=2). Other previous treatments were corticosteroids, biologics, and bronchodilators.
 - Participants had comorbidities such as asthma, allergic rhinitis, food allergies, conjunctivitis, and anxiety.

Table 1. Patient demographic and clinical characteristics

Characteristics	Adults (n=8)	Adolescent (n=5)	Children (n=6)	Caregiver (n=1)
Age (years), mean (SD)	45 (9.1)	15 (0.8)	9 (2.4)	-
Female, n (%)	8 (100)	3 (60)	4 (66.7)	1 (100)
Race, n (%)				
White	8 (100)	3 (60)	6 (100.0)	-
Multi-racial	-	2 (40)	-	1 (100)
Time since diagnosis of ColdU (months), mean (SD)	93.3 (37.5)	108 (40)	10 (6.8)	-
Other skin conditions, n (%)				
Mild eczema	1 (12.5)	1 (20)	-	-
Systemic lupus erythematosus	1 (12.5)	-	-	-
Dermatitis due to ingestion of food	1 (12.5)	-	-	
None	2 (25.0)	2 (40)	4 (66.7)	-
Missing	3 (37.5)	2 (40)	2 (33.3)	1 (100)

Cognitive Debriefing

ColdUAS

- Generally, the instructions and items were well understood and found relevant by adults and adolescents except the terms 'disease activity' and 'wheals' **(Figure 1)**.
- Response options and recall period were well understood and generally considered to be appropriate. One participant reported that it was not appropriate to combine symptom severity and life impairment in the response options used in Item 5.

Figure 1. Overview of understanding and relevance of ColdUAS (adult and adolescent participants)

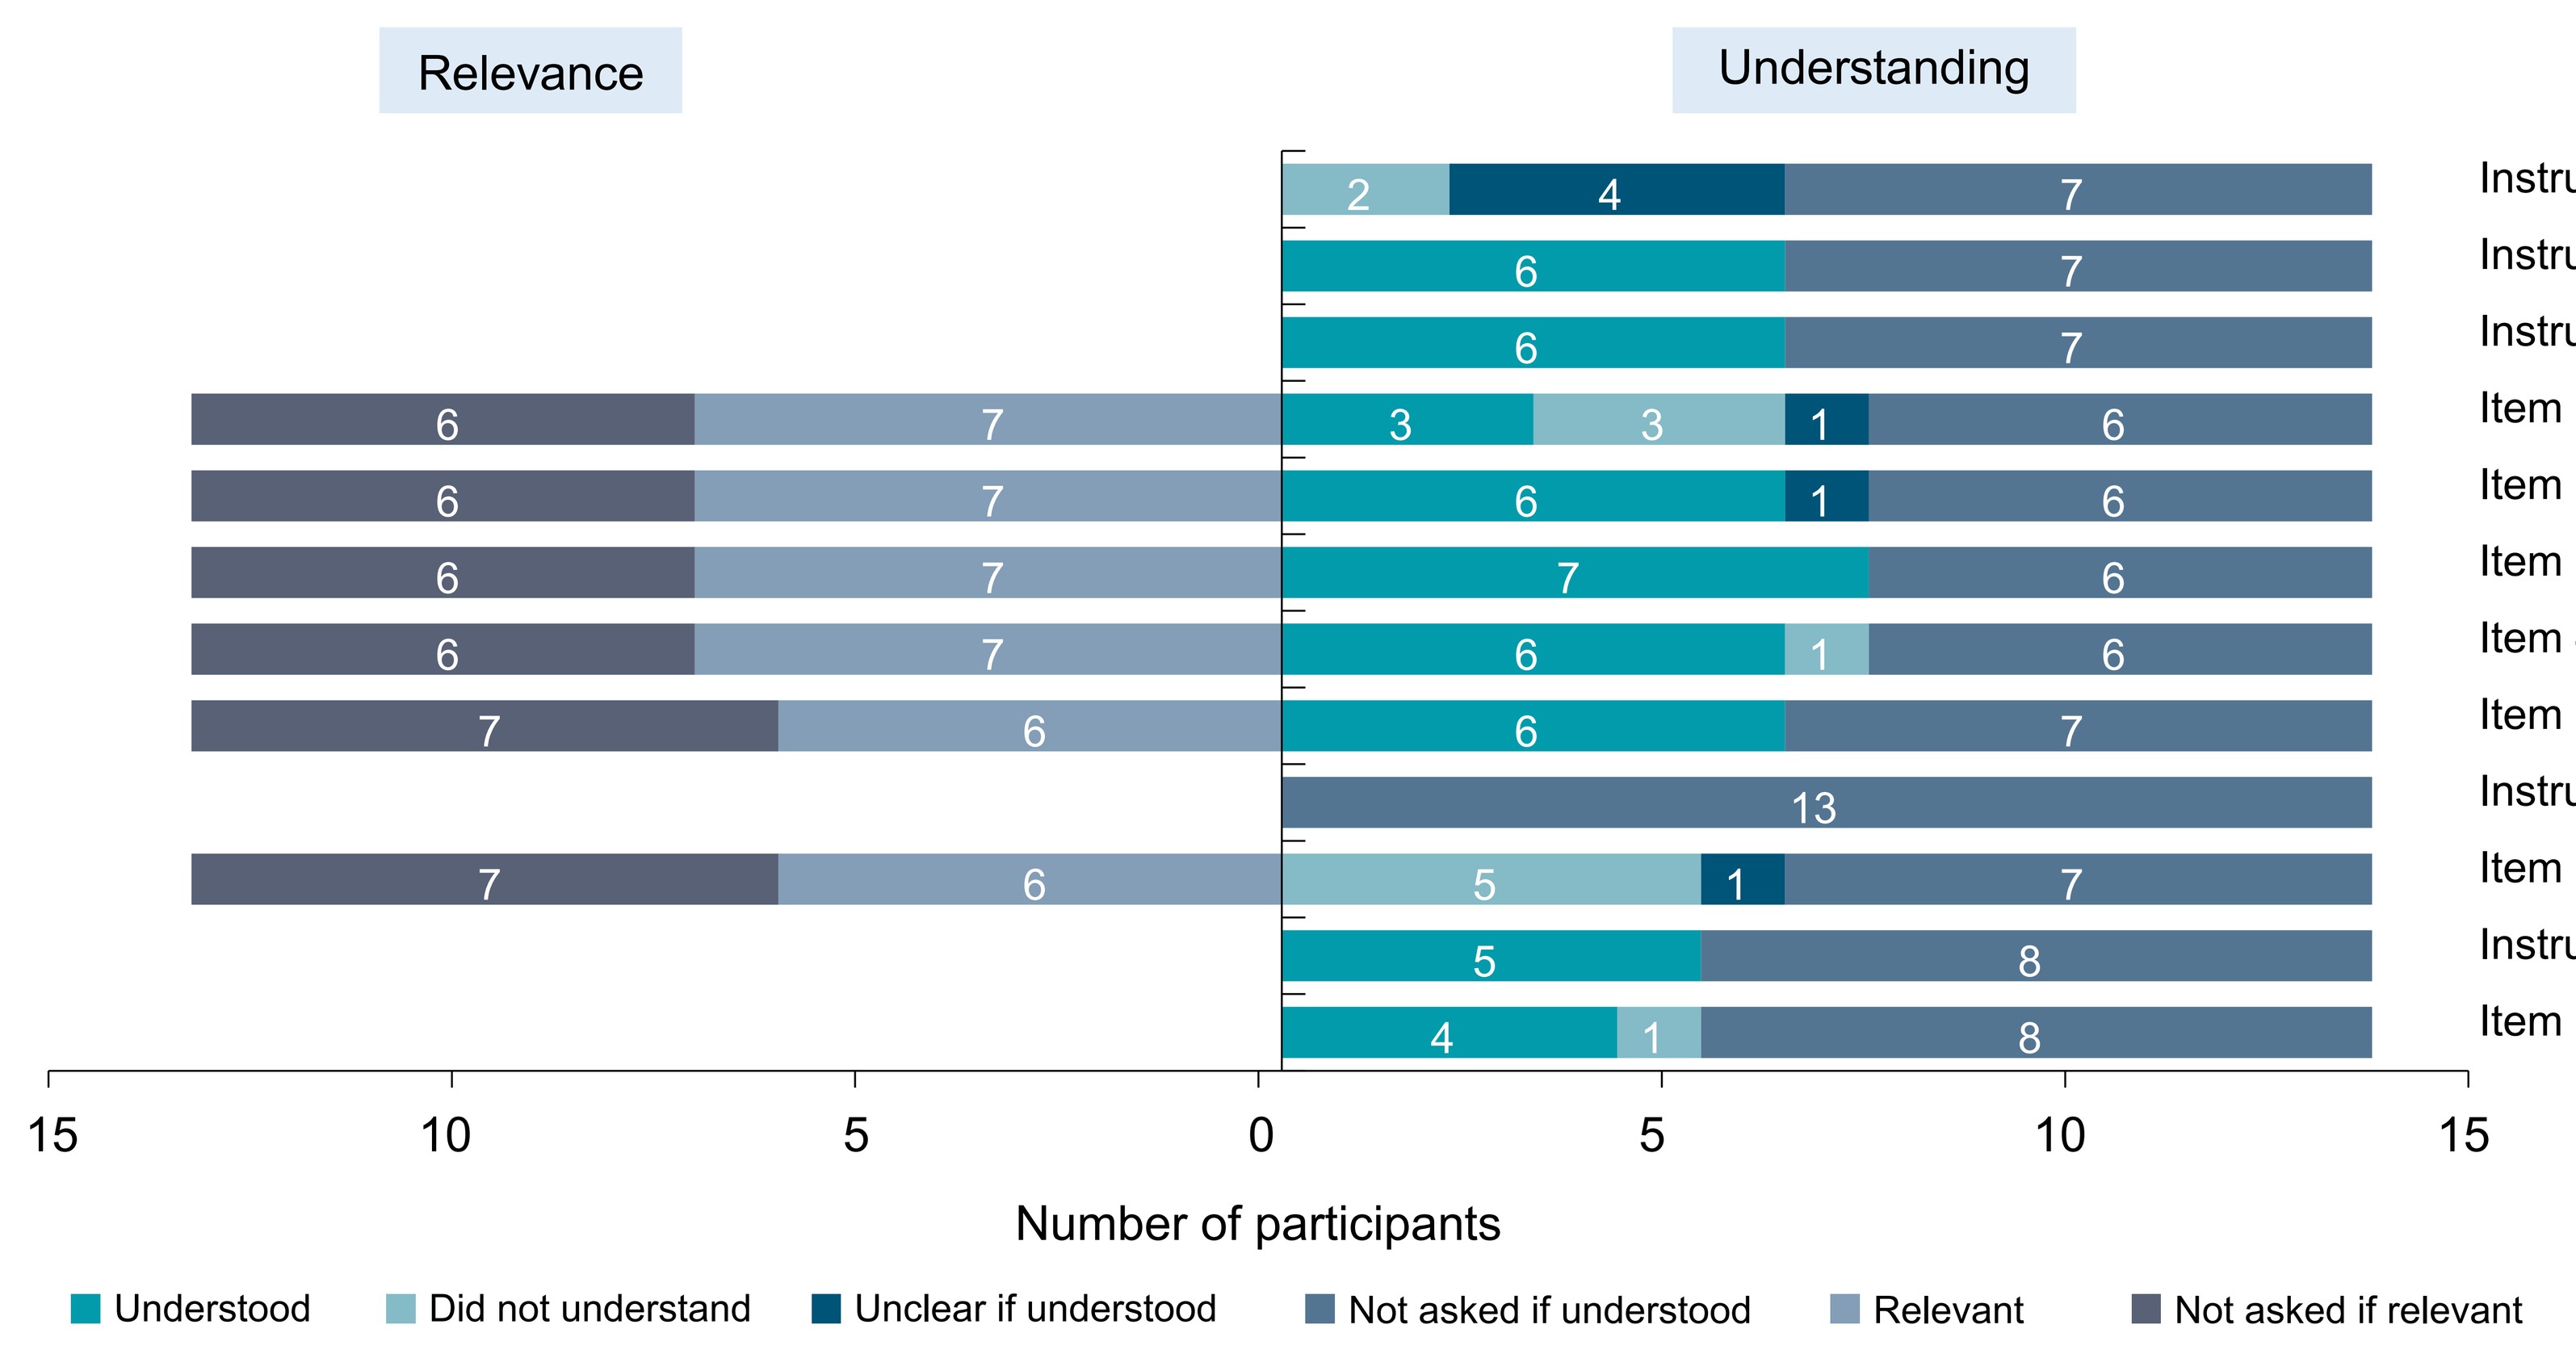
Relevance
Understanding
Instruction 1
Instruction 2
Instruction 3
Item 1
Item 2
Item 3
Item 4
Item 5
Instruction 4
Item 6
Instruction 5
Item 7
15 10 5 0 5 10 15
Number of participants
Understood
Did not understand
Unclear if understood
Not asked if understood
Relevant
Not asked if relevant

Item 1: Wheals/swelling; Item 2: Itching/burning/pain/feeling hot; Item 3: Cold temperature exposure; Item 4: Cold temperature avoidance; Item 5: Symptoms rating; Item 6: Overall disease activity; Item 7: Change in disease activity.

Note: Due to time constrains, instruction 4 was not debriefed.

UCT

- Most items were well understood and found relevant by all participants (n=6/6) **(Figure 2)**.
 - One participant each did not clearly understand item 7b and if the item was referring to treatments that they do not use to treat their ColdU.
 - Items 2 and 4 were not relevant to experience of an adolescent..
- Response options and recall period were generally well understood and considered appropriate. One participant each had difficulty understanding the option 'seldom' used in item 3 and recalling.

Figure 2. Overview of understanding and relevance of UCT (adult and adolescent participants)

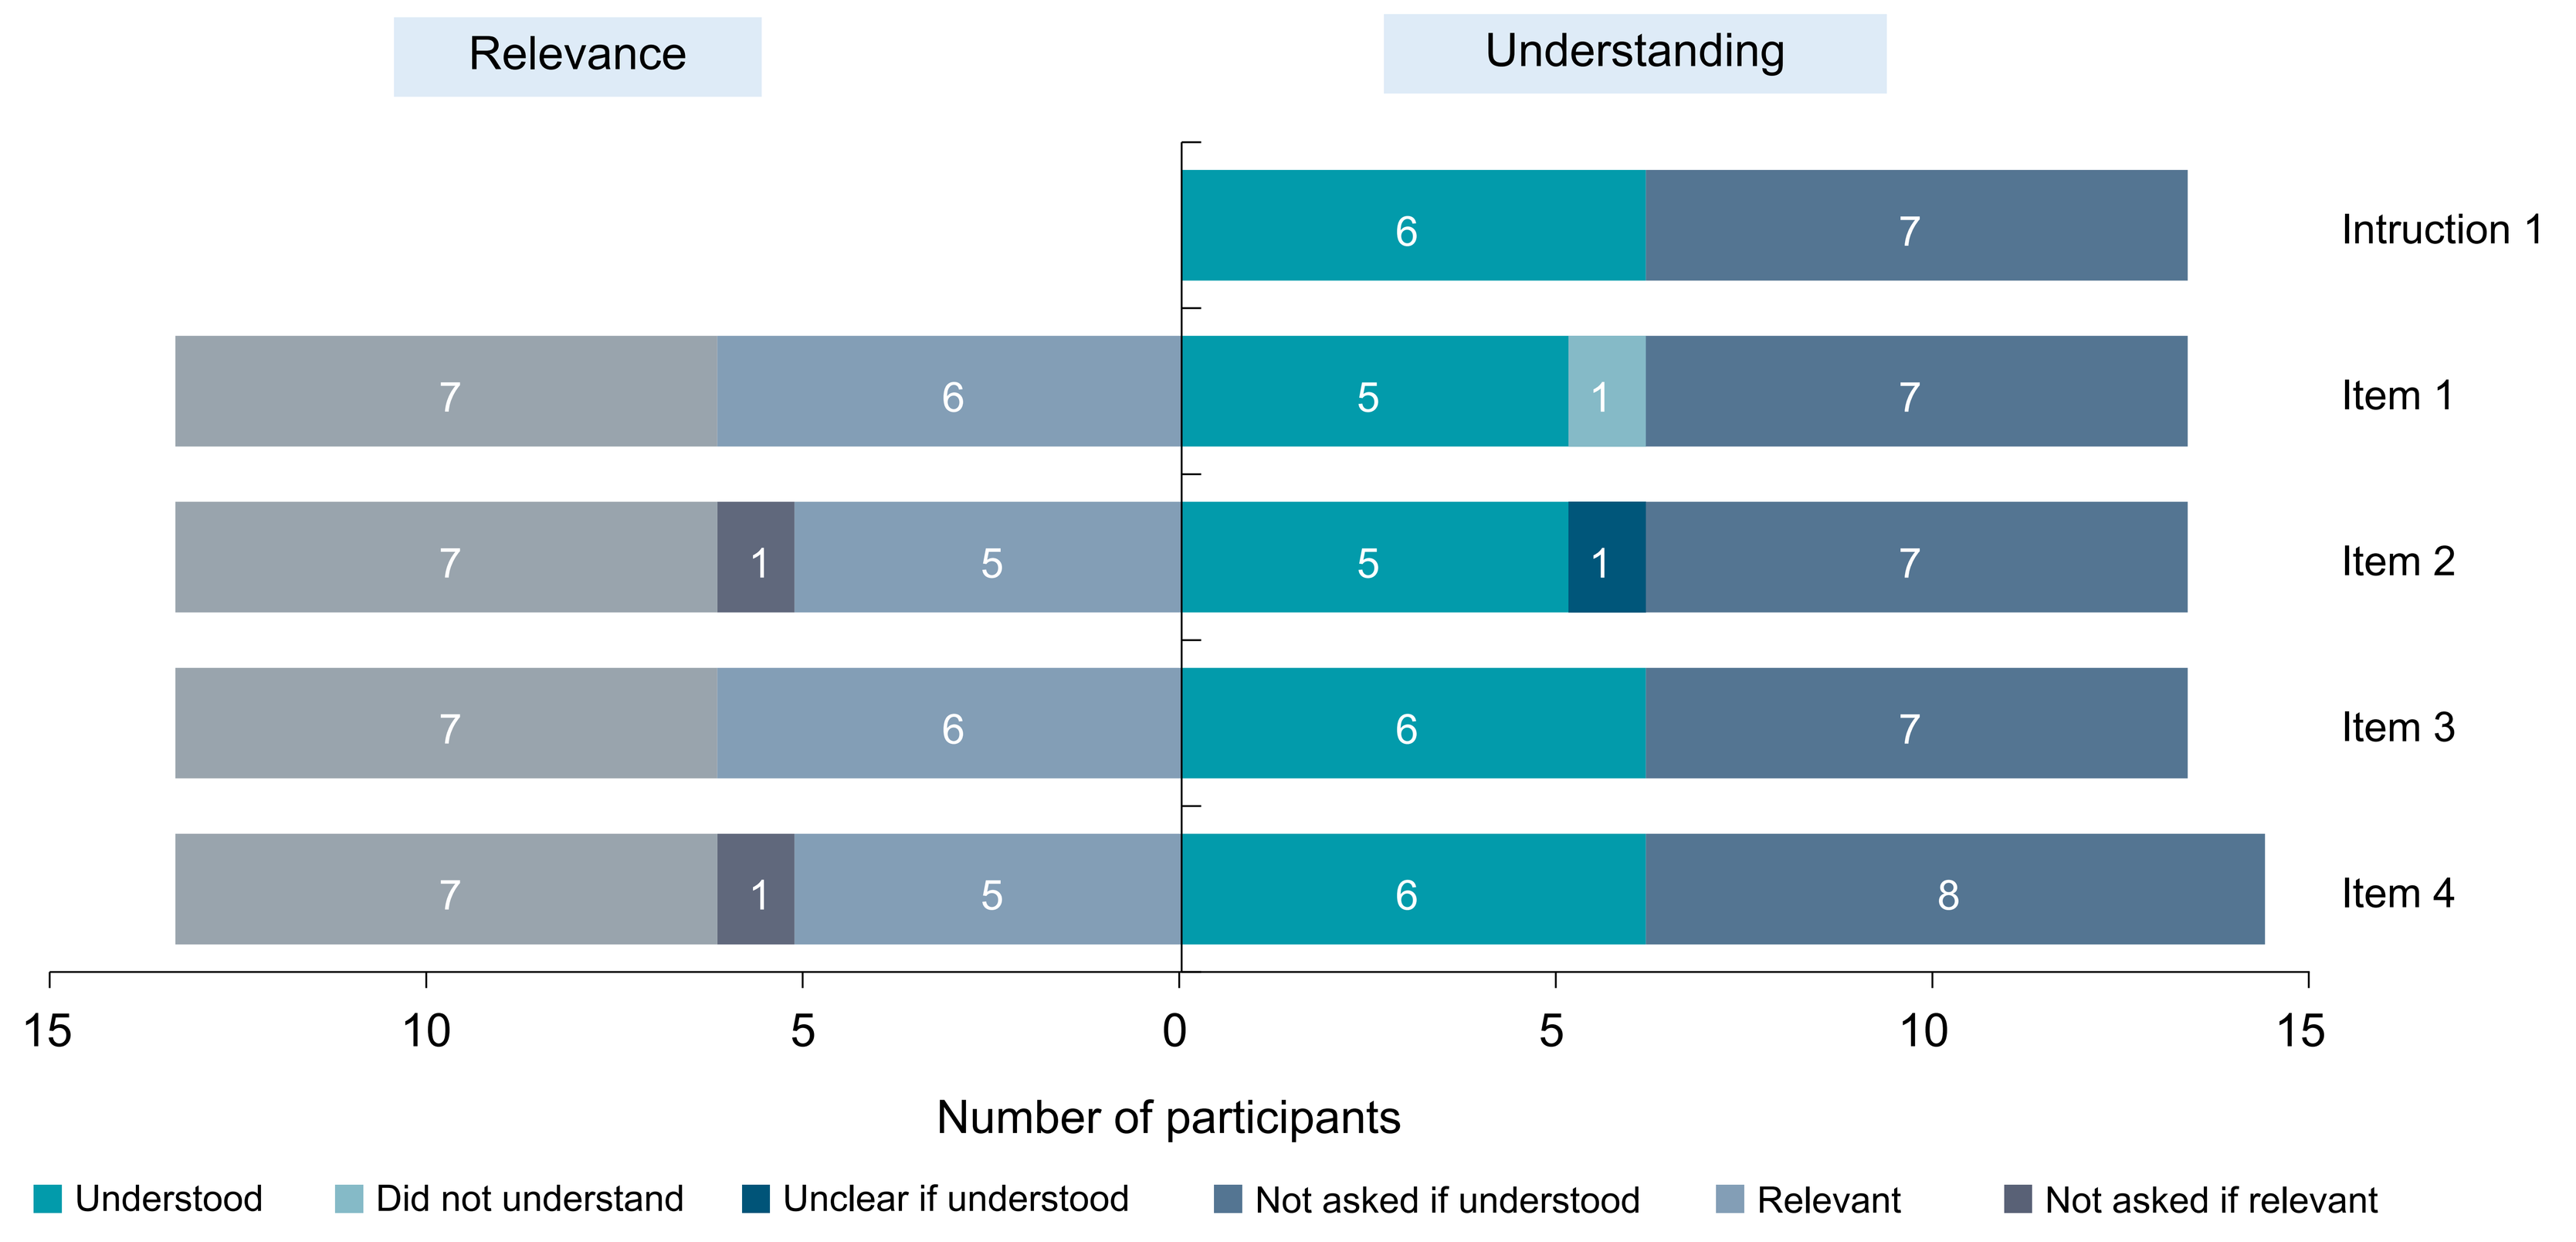
Relevance
Understanding
Intruction 1
Item 1
Item 2
Item 3
Item 4
15 10 5 0 5 10 15
Number of participants
Understood
Did not understand
Unclear if understood
Not asked if understood
Relevant
Not asked if relevant

Item 1: Suffered from itch, hives (welts), and/or swelling; Item 2: Quality of life; Item 3: Inadequate treatment; Item 4: Urticaria under control.

DLQI

- Instruction and items were well understood by either all participants (n=4/4) or almost all participants (n=3/4).
 - One participant each did not clearly understand item 7b and if the item was referring to treatments that they do not use to treat their ColdU.
- The response options were generally well understood. One participant each did not understand how to use the 'not relevant' option and considered response options to be too subjective, suggesting using frequency response options instead.
- One participant considered recall periods to be inappropriate as the past week was not fully representative of their experience of ColdU.

CDLQI

- Instructions and items were well understood (n=5/6) and found relevant (n=6/11) by children.
 - Some children reported that CDLQI items were not relevant to the experience of ColdU.
- Due to time constraints, understanding and relevance of the response options and recall period was not debriefed for all items, however feedback generally suggested that these were well understood.

IDQOL

- The instruction was not understood by the participant.
- According to the caregiver of the infant, the majority of IDQOL items (n=6/11) were not relevant to their child's experience of CICU.
- Due to time constraints, understanding and relevance of the response options and recall period were not debriefed for all items, however feedback generally suggested that these were well understood.

LIMITATIONS

- Recruitment for the study took place largely during summer months in which milder weather may had
- As this research was conducted with patients from the US only, results of the study may not be generalized to other countries/cultures without further research.

REFERENCES
1. M. Magerl et al., Allergy 2016;71:780
2. C. Dressler et al. J. Allergy Clin. Immunol. 2018;141(5):1726–34.
3. M. Maurer et al. J. Allergy Clin. Immunol. 2018;141(2):638–49.

FUNDING
This study was funded by Sanofi and Regeneron pharmaceuticals Inc.

ACKNOWLEDGEMENTS
The medical writing support was provided by Rahul Nikam and Deepti Sharda from Sanofi.

CONFLICTS of INTEREST
AA, IG, RO, and **MK**: Employees of IQVIA.
JC:Employee of Regeneron.
JM, RM, EZ, LBC, CCC, and **EB**: Employees of Sanofi.

Contact Information:
Ella Brookes │ ella.brookes@sanofi.com

The poster was presented at ISPOR EU 2024, Nov 17-20, 2024, Barcelona, Spain