ISPOR Europe 2024 at Barcelona
09:00 - 11:30, Wednesday, 20 November 2024

EE800

SHIONOGI

A budget Impact Analysis of Reducing Viral Transmission with Baloxavir Marboxil Treatment for Seasonal Influenza in Japan

Naoya Itsumura[1], Shogo Miyazawa[2], Satoki Fujita[2]

[1]Asia Value & Access, Shionogi & Co., Ltd., Osaka, Japan, [2]Data Science, Shionogi & Co., Ltd., Osaka, Japan

OBJECTIVES

- Baloxavir marboxil (baloxavir) is an oral antiviral that rapidly stops influenza viral shedding. However, the impact of reduced viral shedding on the number of infections and the resulting budget impact are unknown.
- This study aimed to investigate the reduction in the total number of infections and the budget impact of baloxavir compared to neuraminidase inhibitors (NAIs) using a SEIR (susceptible, exposed, infected, recovered) model.

METHODS

- The population-level SEIR model estimated the attack rates by comparing the impact of treatment ratio of baloxavir and NAIs to linking the time of infectious virus negativity, using the clinical trial data of baloxavir compared to oseltamivir for the influenza treatment.
- The population was stratified into four subgroups: 1) otherwise healthy children, 2) high-risk children, 3) otherwise healthy adolescents and non-elderly adults, and 4) high-risk adolescents, high-risk adults, and the elderly (Figure 1).
- The parameters were calculated using Japanese real-world data from the 2018/2019 season (Figure 1), because there were almost no influenza cases during the COVID-19 pandemic. The budget impact analysis was conducted at the total population level, incorporating the attack rates from the SEIR model.

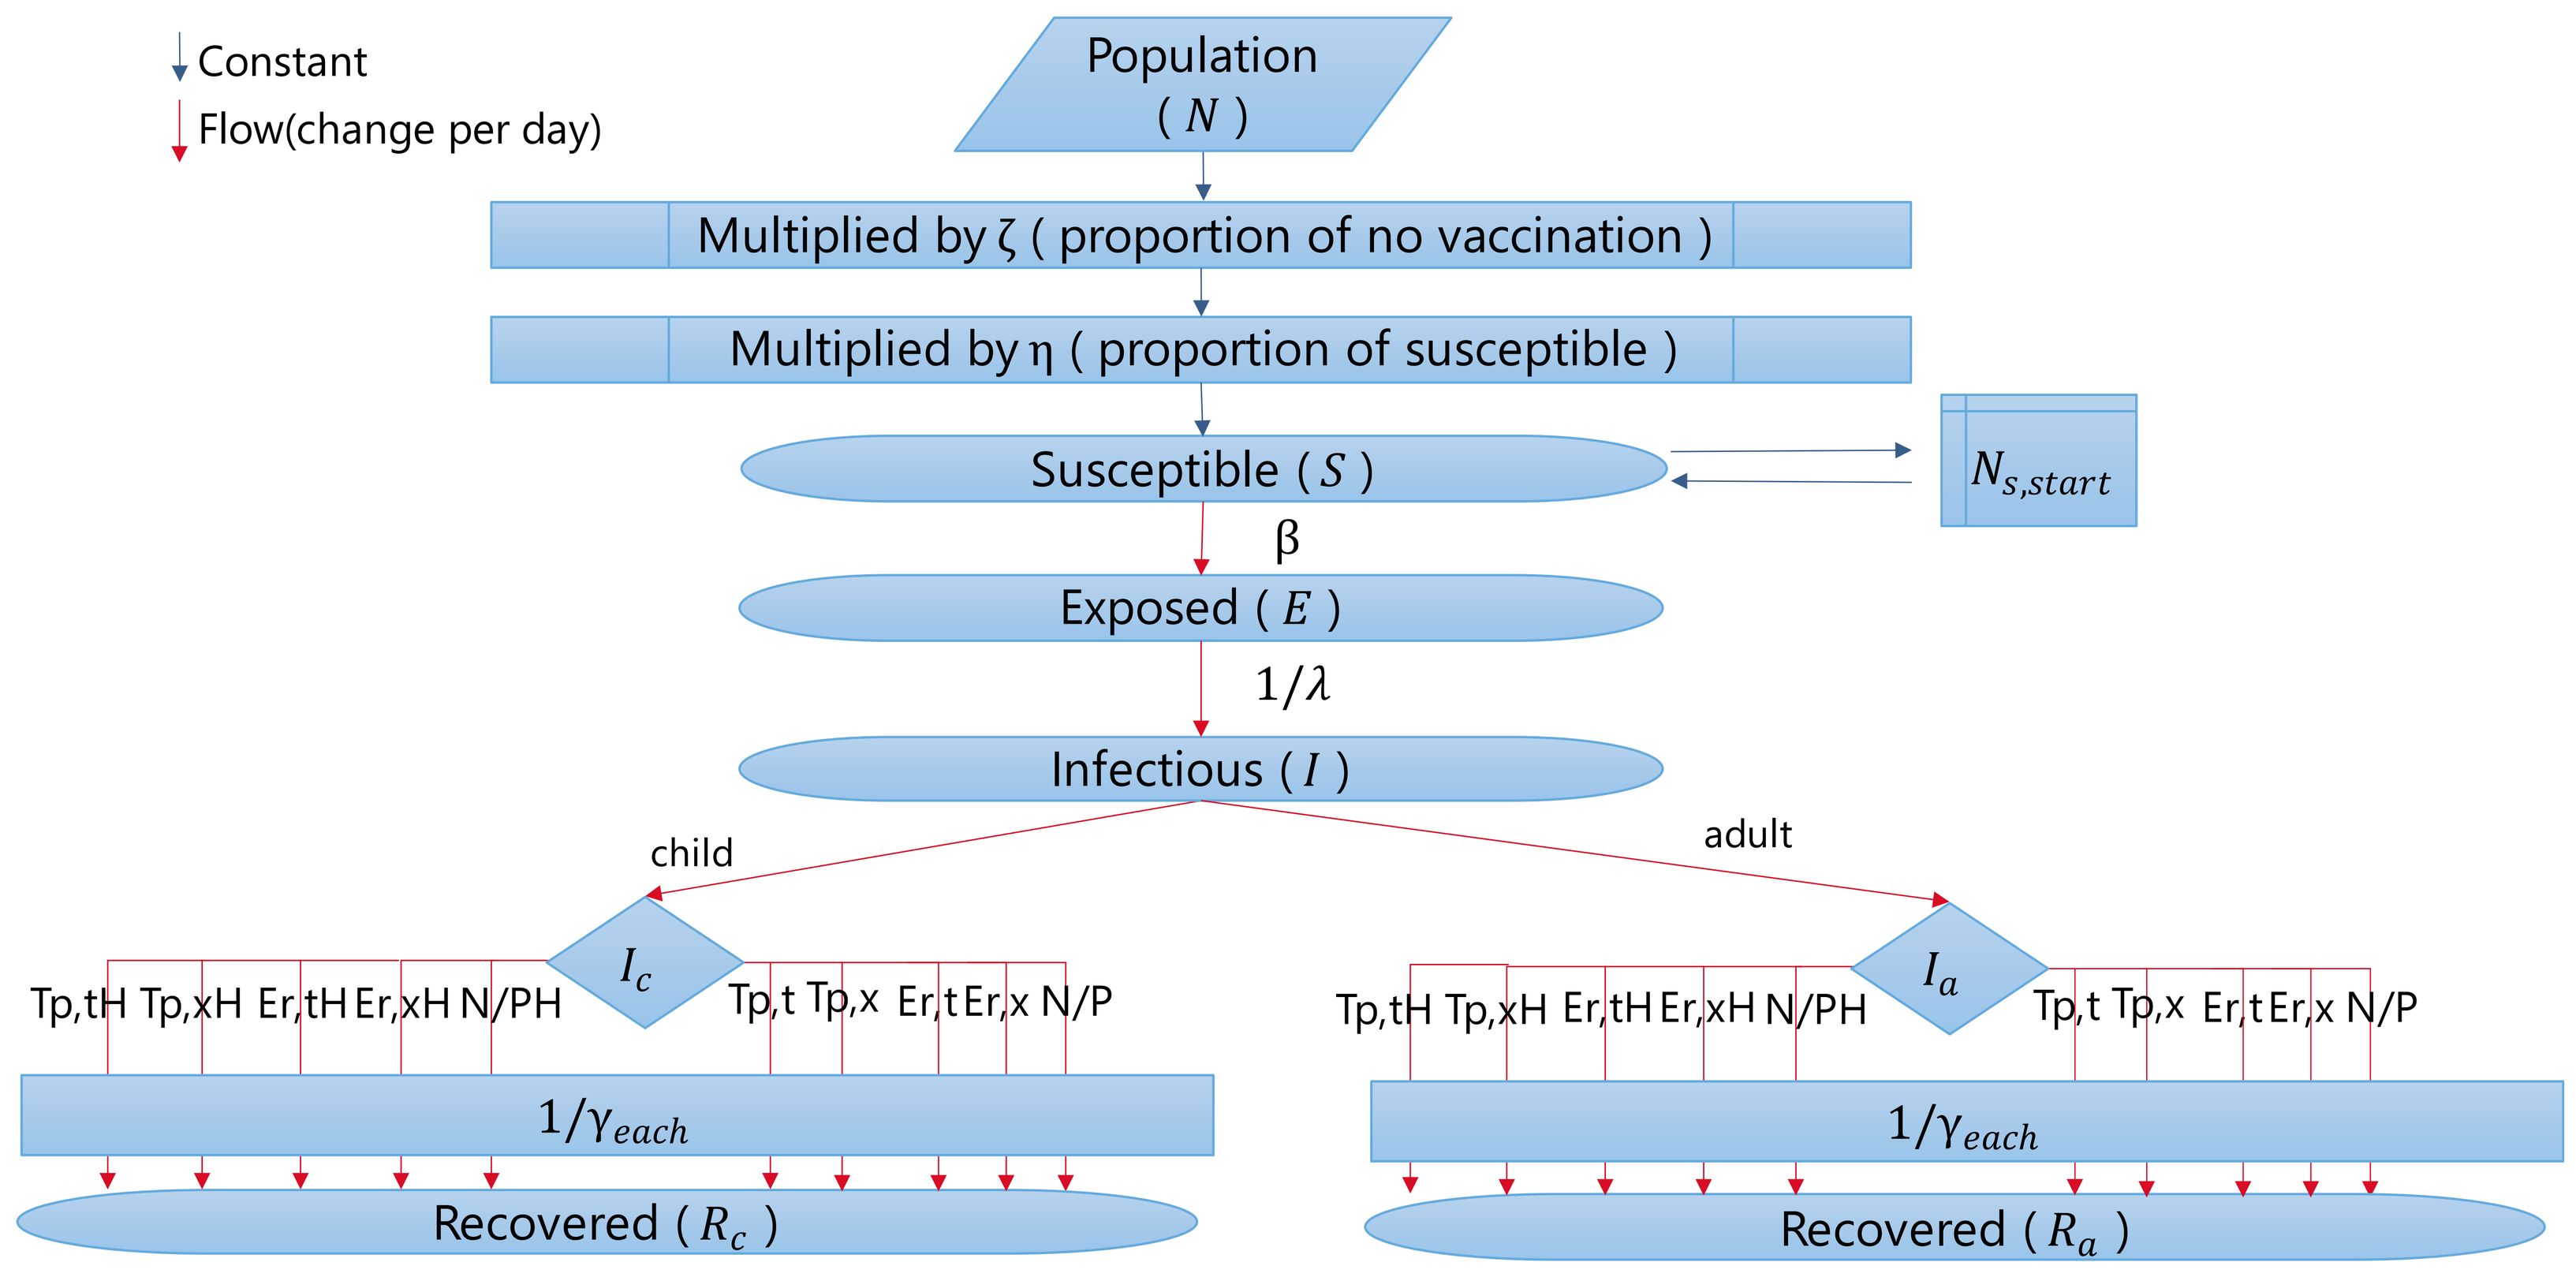
Constant
Flow(change per day)
Population (N)
Multiplied by ζ (proportion of no vaccination)
Multiplied by η (proportion of susceptible)
Susceptible (S)
N s,start
β
Exposed (E)
1/λ
Infectious (I)
child
adult
Ic
Ia
Tp,tH Tp,xH Er,tH Er,xH N/PH
Tp,t Tp,x Er,t Er,x N/P
1/γ each
Recovered (Rc)
Recovered (Ra)

Figure 1: Structure of the SEIR model

B = Exposed rate, 1/λ = Infection rate, 1/γ = recover rate, Tp = Treated patients, t = Tamiflu (oseltamivir) treatment, x = Xofluza (baloxavir) treatment, H = High-risk group, Er = Emerging resistant virus, N/P = No treated patients or Patients infected resistance virus, SEIR = Susceptible, Exposed, Infected, Recovered.

Variable	Inputs	Source
Population size	**10,000**	Assumption
Initial number of infected	**10**	Assumption
Susceptible (S)	**66.4%**	Calculation from RWD in Japan
Transmittability (β)	**0.25**	Calculation from RWD in Japan
Infected but not infectious (λ)	**1 day**	Miyazawa et al., (2022)
Antiviral treatment rate	**92.1%**	Calculation from RWD in Japan
Adults in the population	**47.4%**	Calculation from 2018/19 season data
HR proportion in children	**44.8%**	Calculation from 2018/19 season data
HR proportion in adults	**44.1%**	Dronova et al., (2021)
Simulation period	**365 days**	Influenza one season
Time to cessation of viral shedding (γ)		
No antiviral in OwH	**4.71 days**	Estimating from CAPSTONE-1 trial*
Oseltamivir in OwH	**3.83 days**	Estimating from CAPSTONE-1 trial*
Baloxavir in OwH	**2.11 days**	Estimating from CAPSTONE-1 trial*
No antiviral in HR	**4.53 days**	Estimating from CAPSTONE-2 trial**
Oseltamivir in HR	**3.90 days**	Estimating from CAPSTONE-2 trial**
Baloxavir in HR	**2.72 days**	Estimating from CAPSTONE-2 trial**

Table 1: Key Inputs and assumptions for the SEIR model

Due to lack of data, the time to cessation of viral shedding by NAIs (laninamivir, zanamivir, and oseltamivir) was assumed to be equal to oseltamivir. HR = High-risk, OwH = Otherwise health

RESULTS

- The SEIR model estimated greater reductions in infections with increased baloxavir treatment (Table 2). The baloxavir share for adults was varied in the model based on the share in 2018/2019 season (baloxavir vs NAIs for adults: 50% vs 50%; for children: 20% vs 80%).
- We estimated that influenza patients decreased from 11.7 million to 9.6 million (18.3% reduction) if 10% of NAIs for adults were replaced with baloxavir in the 2018/2019 season.
- The budget impact was calculated to be a benefit of 6.0 billion JPY in drug costs, 16.0 billion JPY in medical costs, and 20.7 billion JPY in labor productivity losses (Figure 2).

Baloxavir increase or decrease scenario	Share of antiviral treatment				Proportion of infected population
	Baloxavir for Adults	NAIs for Adults	Baloxavir for Children	NAIs for Children	
-20%	30%	70%			**14.1%**
-10%	40%	60%			**11.9%**
Base case	50%	50%	20%	80%	**9.9%**
+10%	60%	40%			**8.0%**
+20%	70%	30%			**6.5%**

Table 2: Proportion of infected population with varying shares of baloxavir or NAIs

The base case is the most similar estimation to the 2018/2019 season in Japan.

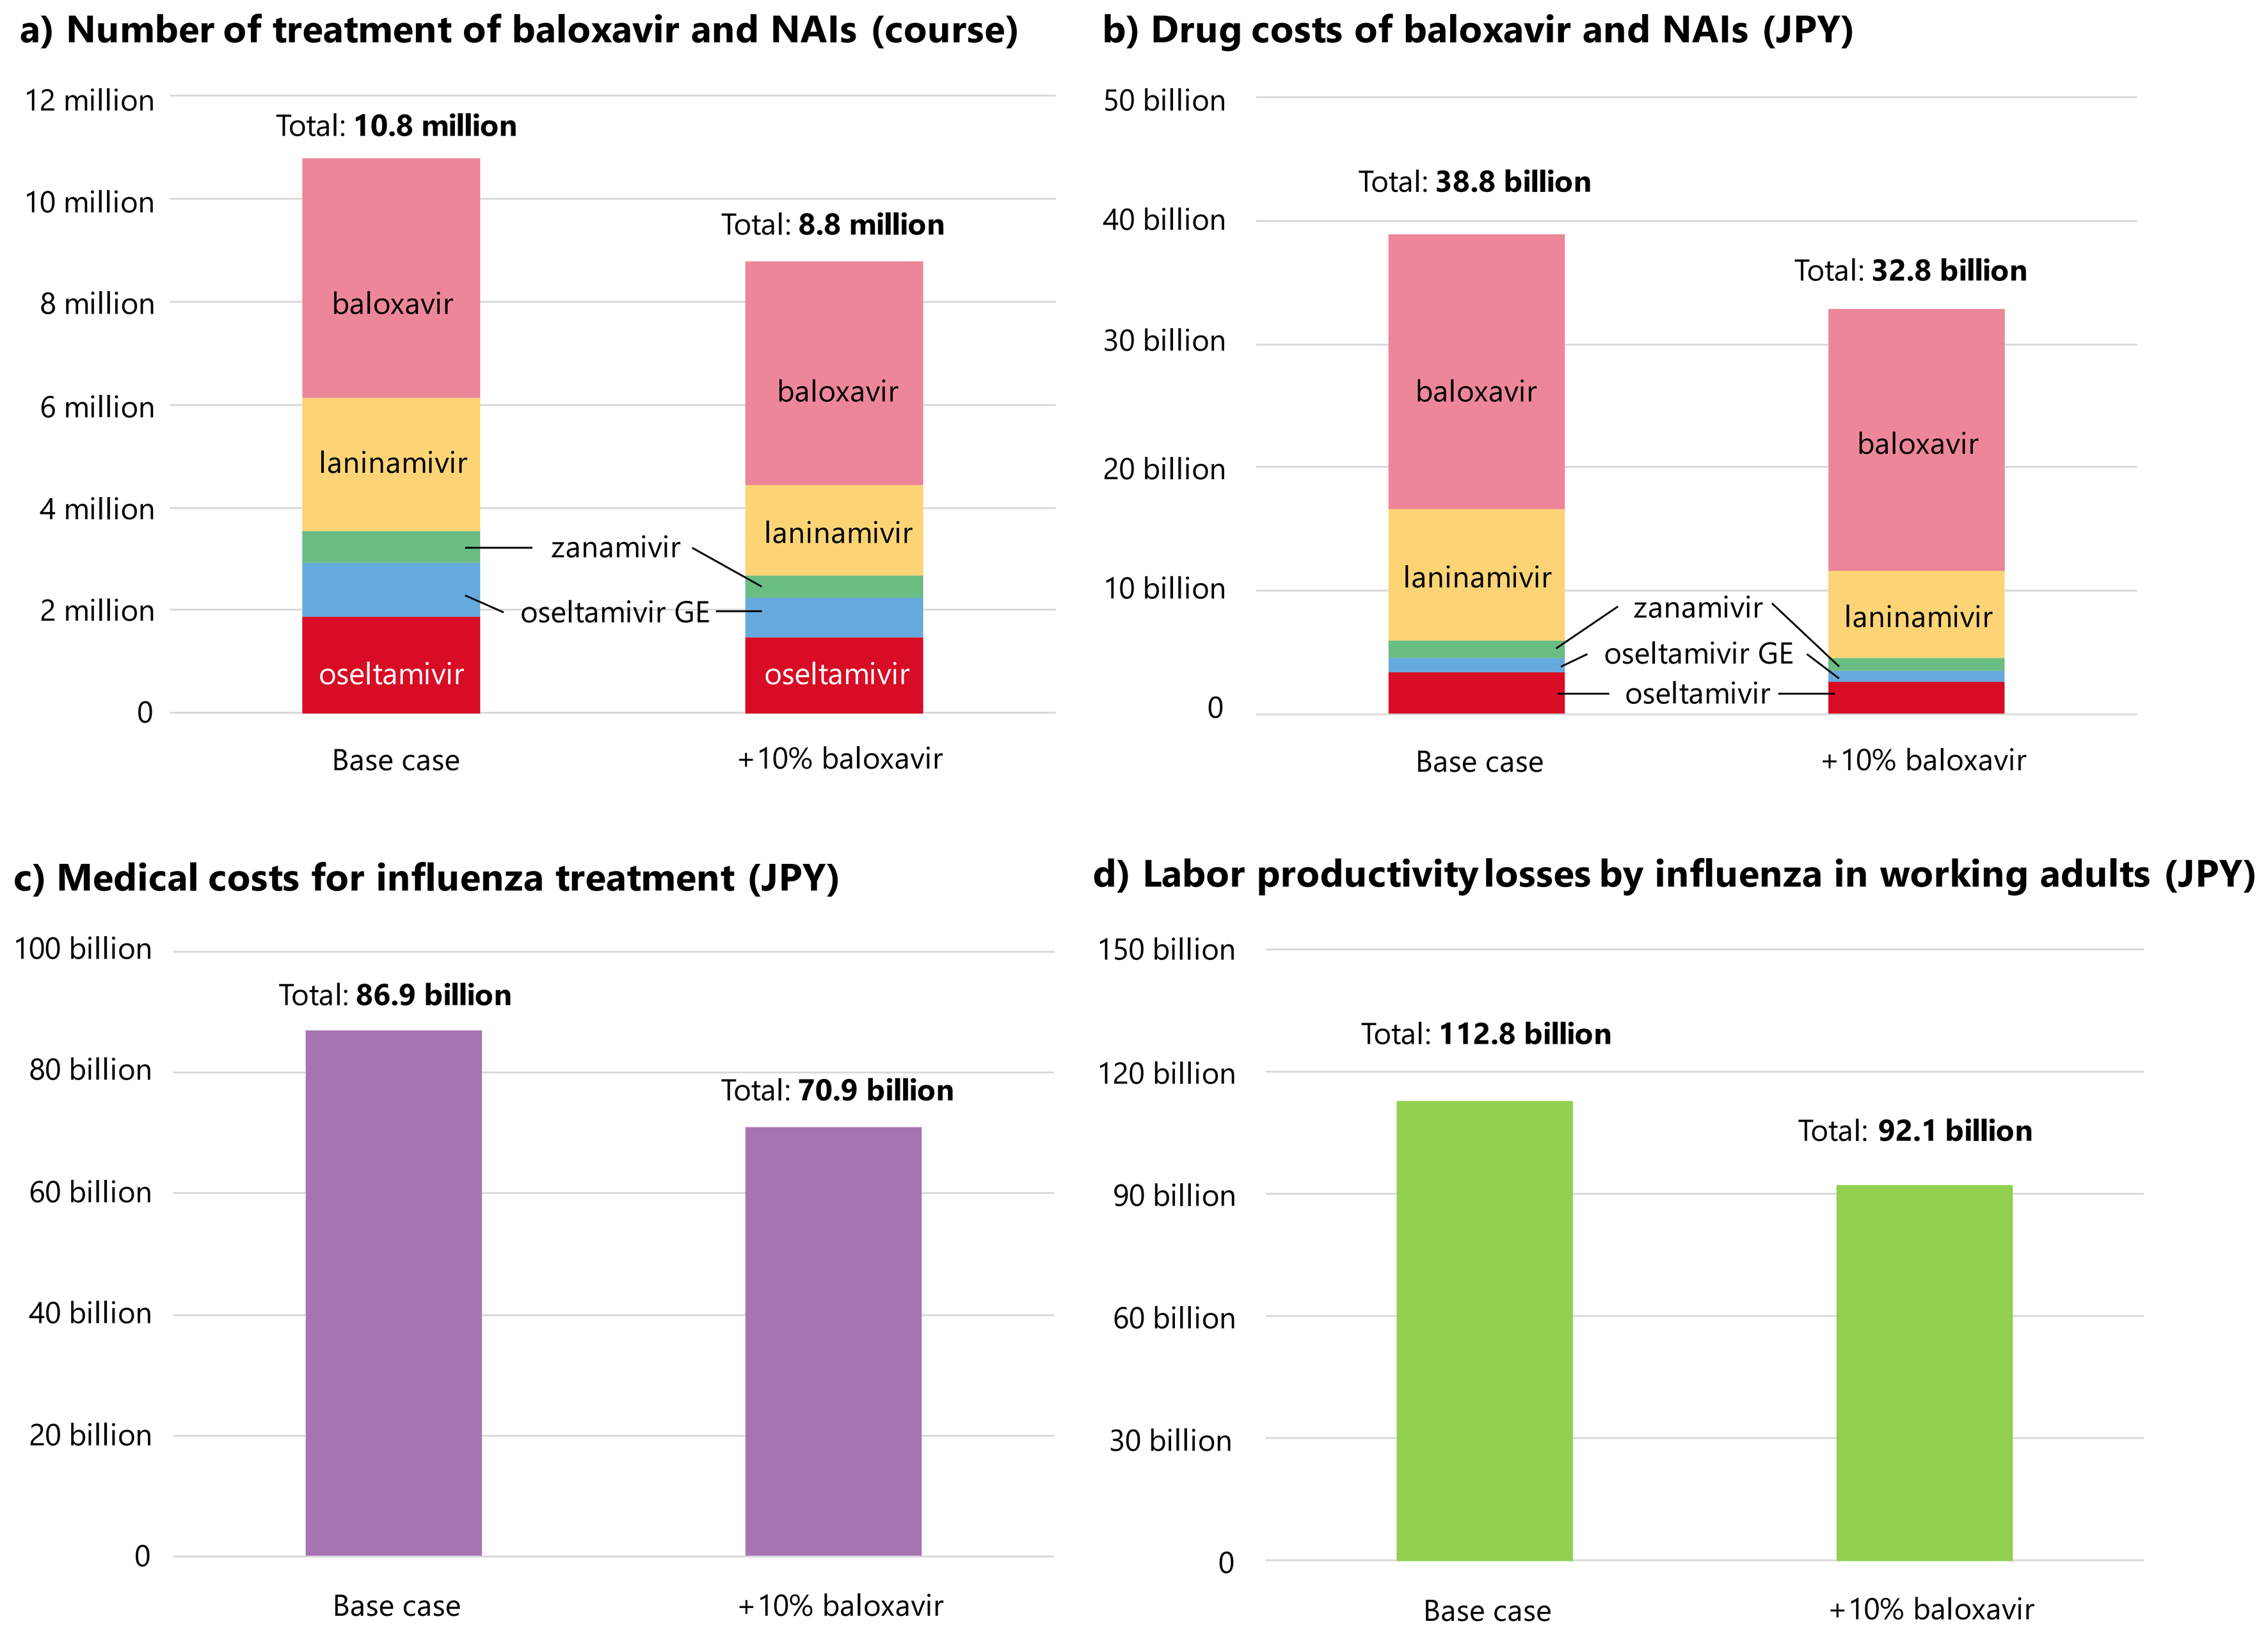
a) Number of treatment of baloxavir and NAIs (course)
12 million
10 million
8 million
6 million
4 million
2 million
0
Total: 10.8 million
Total: 8.8 million
baloxavir
laninamivir
zanamivir
oseltamivir GE
oseltamivir
Base case
+10% baloxavir
b) Drug costs of baloxavir and NAIs (JPY)
50 billion
40 billion
30 billion
20 billion
10 billion
0
Total: 38.8 billion
Total: 32.8 billion
c) Medical costs for influenza treatment (JPY)
100 billion
80 billion
60 billion
40 billion
20 billion
Total: 86.9 billion
Total: 70.9 billion
d) Labor productivity losses by influenza in working adults (JPY)
150 billion
120 billion
90 billion
60 billion
30 billion
Total: 112.8 billion
Total: 92.1 billion

Figure 2: Reductions in number of treatment, drug costs, medical costs, and labor productivity losses with increasing baloxavir share

a) We assumed that the market share of drugs other than baloxavir was not changed.

b) Drug costs were calculated based on age and weight indication of each drug.

c) Medical costs were calculated based on GP visit fee and diagnostic tests.

d) Labor productivity losses were calculated by assuming that influenza infection caused two days of absence from work and working adults lose the average wage each day. JPY = Japanese yen

CONCLUSIONS

- Compared to NAIs, baloxavir treatment may result in fewer influenza cases and could save costs of billions of JPY per influenza season from public healthcare payers' perspective and social perspective in Japan.
- As influenza epidemic have reappeared in Japan from the 2023/2024 season, the results of the transmission model and the budget impact analysis need to be updated to reflect the post COVID-19 pandemic reality.

Funding: This study was funded by Shionogi & Co., Ltd.

Conflict of interest: NI, SM, and SF are employees of Shionogi & Co., Ltd.

* Hayden FG, Sugaya N, Hirotsu N, et al. Baloxavir Marboxil for Uncomplicated Influenza in Adults and Adolescents. N Engl J Med. 2018;379(10):913-923.

** Ison MG, Portsmouth S, Yoshida Y, et al. Early treatment with baloxavir marboxil in high-risk adolescent and adult outpatients with uncomplicated influenza (CAPSTONE-2): a randomised, placebo-controlled, phase 3 trial. Lancet Infect Dis. 2020;20(10):1204-1214.