CO191

Application of Causal Inference Methods to Assess Efficacy in Secondary Outcomes in Ophthalmology Clinical Trials for Use in Economic Models

Michele Intorcia[1], Matthew Davis[2], Sujata Sarda[1], Warren Stevens[2]

[1]Apellis Pharmaceuticals, Waltham, MA, USA; [2]Medicus Economics, LLC

Introduction

- Geographic atrophy (GA), an advanced form of age-related macular degeneration (AMD), is a chronic progressive degenerative disease with an estimated prevalence of 5 million patients globally and 1 million in the United States (US).[1] Treatments for GA aim to slow lesion growth so that visual acuity (VA) is maintained and the negative impact of GA on patient quality of life is minimized[2]
- *Pegcetacoplan* (PEG), a novel C3 inhibitor, is the first drug approved by the US Food and Drug Administration for treatment of GA secondary to AMD.[3] This approval was based on the results of two phase 3 trials (NCT03525600 [DERBY], NCT03525613 [OAKS]) demonstrating that both monthly and every-other-month (EOM) dosing of PEG slowed GA lesion growth, with effects increasing over the 24-month follow-up period. No statistically meaningful differences were found on the secondary functional endpoints[4]
- Recent evidence has shown GA lesion size on its own has poor correlation with VA, unless lesion location and distance from the fovea are taken into account.[5,6] In particular, percentage of foveal involvement (ie, central macular occupancy [CMO]) has been found to be most predictive of VA loss.[7] As such, evaluations of the relationship between GA lesion growth and VA decline need to adjust for foveal involvement; however, these data have historically been lacking from clinical assessments as evidence of their importance in predicting VA decline has only emerged recently

Objective

- With randomized controlled trials (RCTs) designed to assess clinical primary endpoints, such as lesion growth reduction in GA, sometimes the secondary outcomes that are used for economic modeling purposes, such as VA, may not have been adequately powered or randomized. This analysis applied causal inference methods to better reflect the complex relationship between lesion growth and vision loss, via key VA risk factors like CMO that were not accounted for in the trial design

Methods

- **Patient sample:** The trial sample (n=1211) was subset to those with complete baseline measures (n=888) and stratified by lesion distance from the foveal center (≥250 vs <250 µm). Monthly and EOM dosing groups were combined into pooled "treated" and "sham" groups (**Figure 1**)
- **Inclusion/exclusion:** Patients evaluated with Cirrus (Zeiss) spectral-domain optical coherence tomography (SD-OCT) imaging were not included in the analysis as OCT-derived CMO data were unavailable for these devices. All other eligibility restrictions were as per the study design of the trials[4]
- **Outcome:** VA was evaluated as change from baseline up to 24 months compared to sham
- **Statistical analysis:**
 - Model specification and baseline covariate selection were done a priori based on clinical rationale[5,6]
 - The adjusted impact of treatment was estimated using inverse probability of treatment weighted regression models, with propensity scores estimated from baseline CMO; lesion size; VA; age; sex; geographic region; and multifocal, bilateral, pseudophakic, and better- vs worse-seeing eye status (**Table 1**)
 - CMO was defined as the proportion of the total area of photoreceptor or retinal pigment epithelium (RPE) loss in Early Treatment Diabetic Retinopathy Study (ETDRS) regions 1–5 divided by the maximum possible area of regions 1–5

Figure 1. Patient disposition

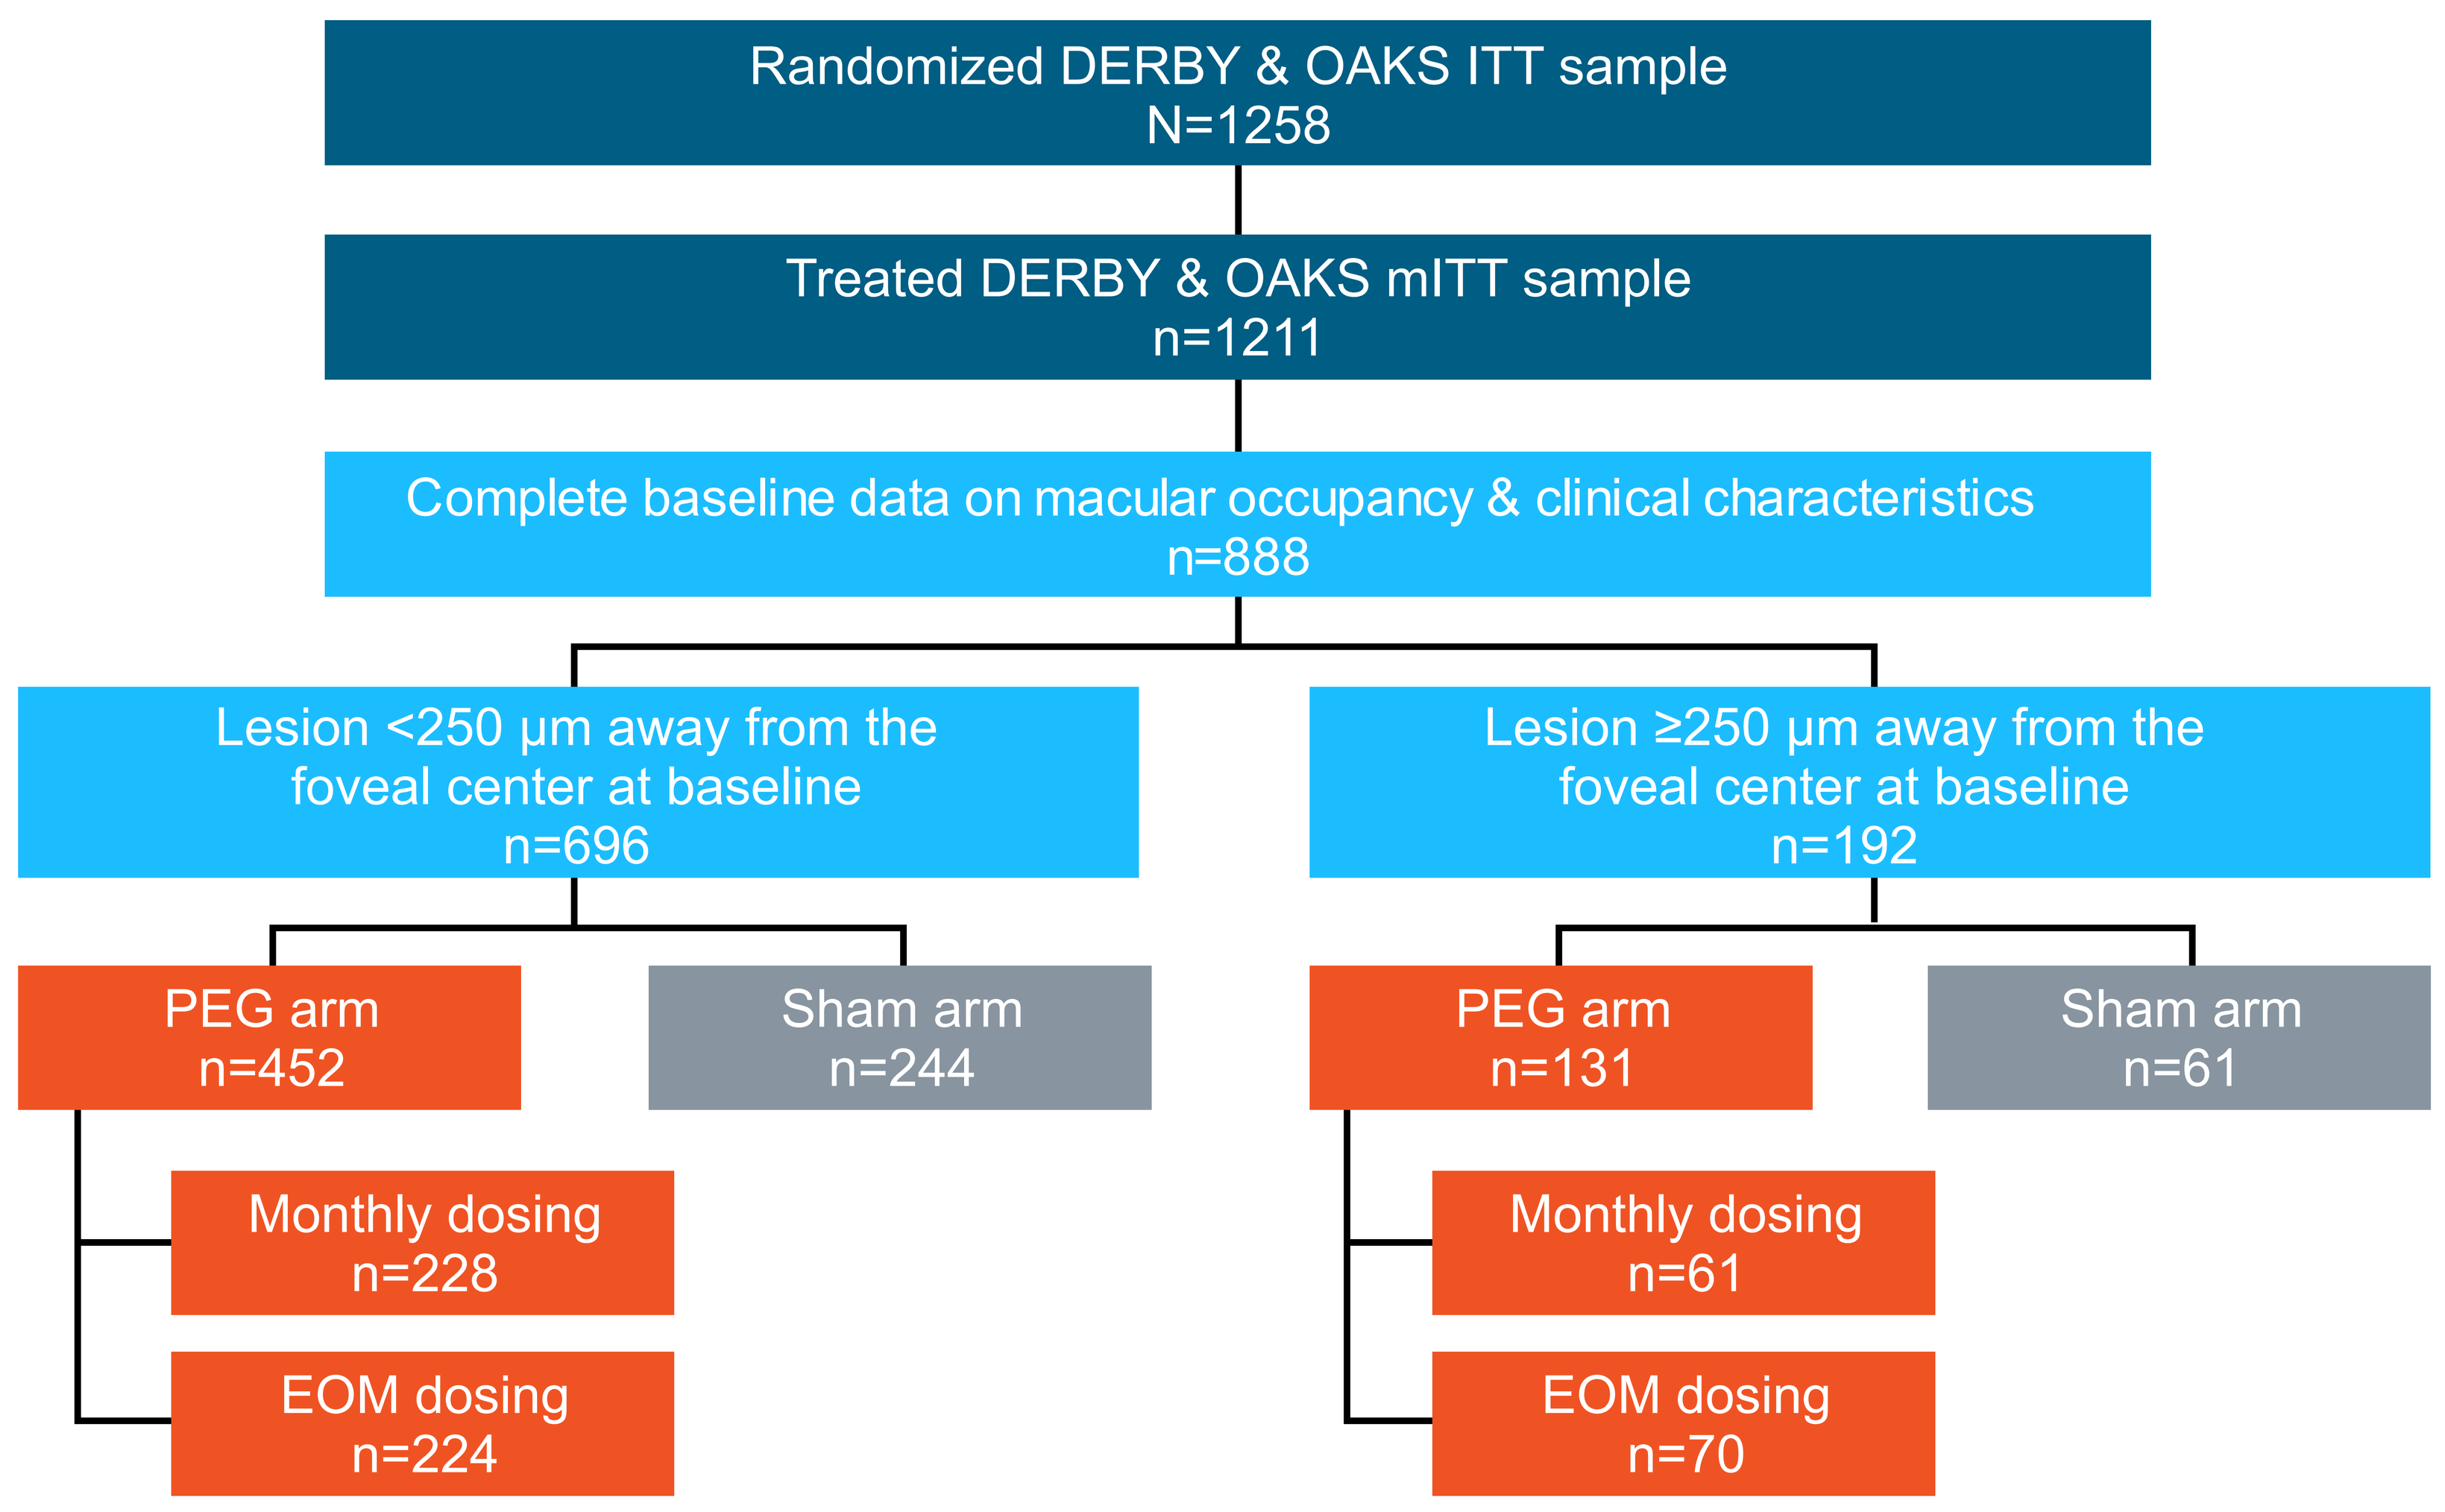
Randomized DERBY & OAKS ITT sample
N=1258
Treated DERBY & OAKS mITT sample
n=1211
Complete baseline data on macular occupancy & clinical characteristics
n=888
Lesion <250 µm away from the foveal center at baseline
n=696
Lesion ≥250 µm away from the foveal center at baseline
n=192
PEG arm
n=452
Sham arm
n=244
PEG arm
n=131
Sham arm
n=61
Monthly dosing
n=228
EOM dosing
n=224
Monthly dosing
n=61
EOM dosing
n=70

EOM, every other month; ITT, intent-to-treat; mITT, modified intent-to-treat; PEG, pegcetacoplan.

Results

- Differences in baseline characteristics were observed when stratifying patients by GA lesion distance from the foveal center. A greater proportion of patients with GA lesions <250 µm from the foveal center were male, had their worse-seeing eye as their study eye, and had a greater percentage of CMO compared with patients with GA lesions ≥250 µm from the foveal center (**Table 1**)

Table 1. Baseline patient demographics and disease characteristics

Baseline Measure	GA Lesion <250 µm From the Foveal Center (n=696)	GA Lesion ≥250 µm From the Foveal Center (n=192)	Difference	*P*-Value
United States, n (%)	440 (63)	136 (71)	−8%	NS
Age ≥75 years, n (%)	489 (70)	135 (70)	0%	NS
Female, n (%)	409 (59)	130 (68)	−9%	<0.05
GA lesion area, mean mm²	8.25	8.17	+0.08 mm²	NS
Study eye				
Worse-seeing eye, n (%)	369 (53)	65 (34)	+19%	<0.0001
Multifocal, n (%)	464 (67)	162 (84)	−18%	<0.0001
Pseudophakic, n (%)	491 (71)	132 (69)	+2%	NS
Bilateral GA, n (%)	569 (82)	150 (78)	+4%	NS
Macular occupancy, mean %	63	46	+17%	<0.0001
BCVA, mean ETDRS letters	55	74	−18	<0.0001

BCVA, best-corrected visual acuity; ETDRS, Early Treatment Diabetic Retinopathy Study; GA, geographic atrophy; NS, not significant.

- Average best-corrected visual acuity (BCVA) was 18 letters lower for patients with a lesion within 250 µm of the foveal center, and, for all patients at baseline, lower BCVA scores were observed as CMO increased (**Figure 2**)

Figure 2. BCVA and central macular occupancy

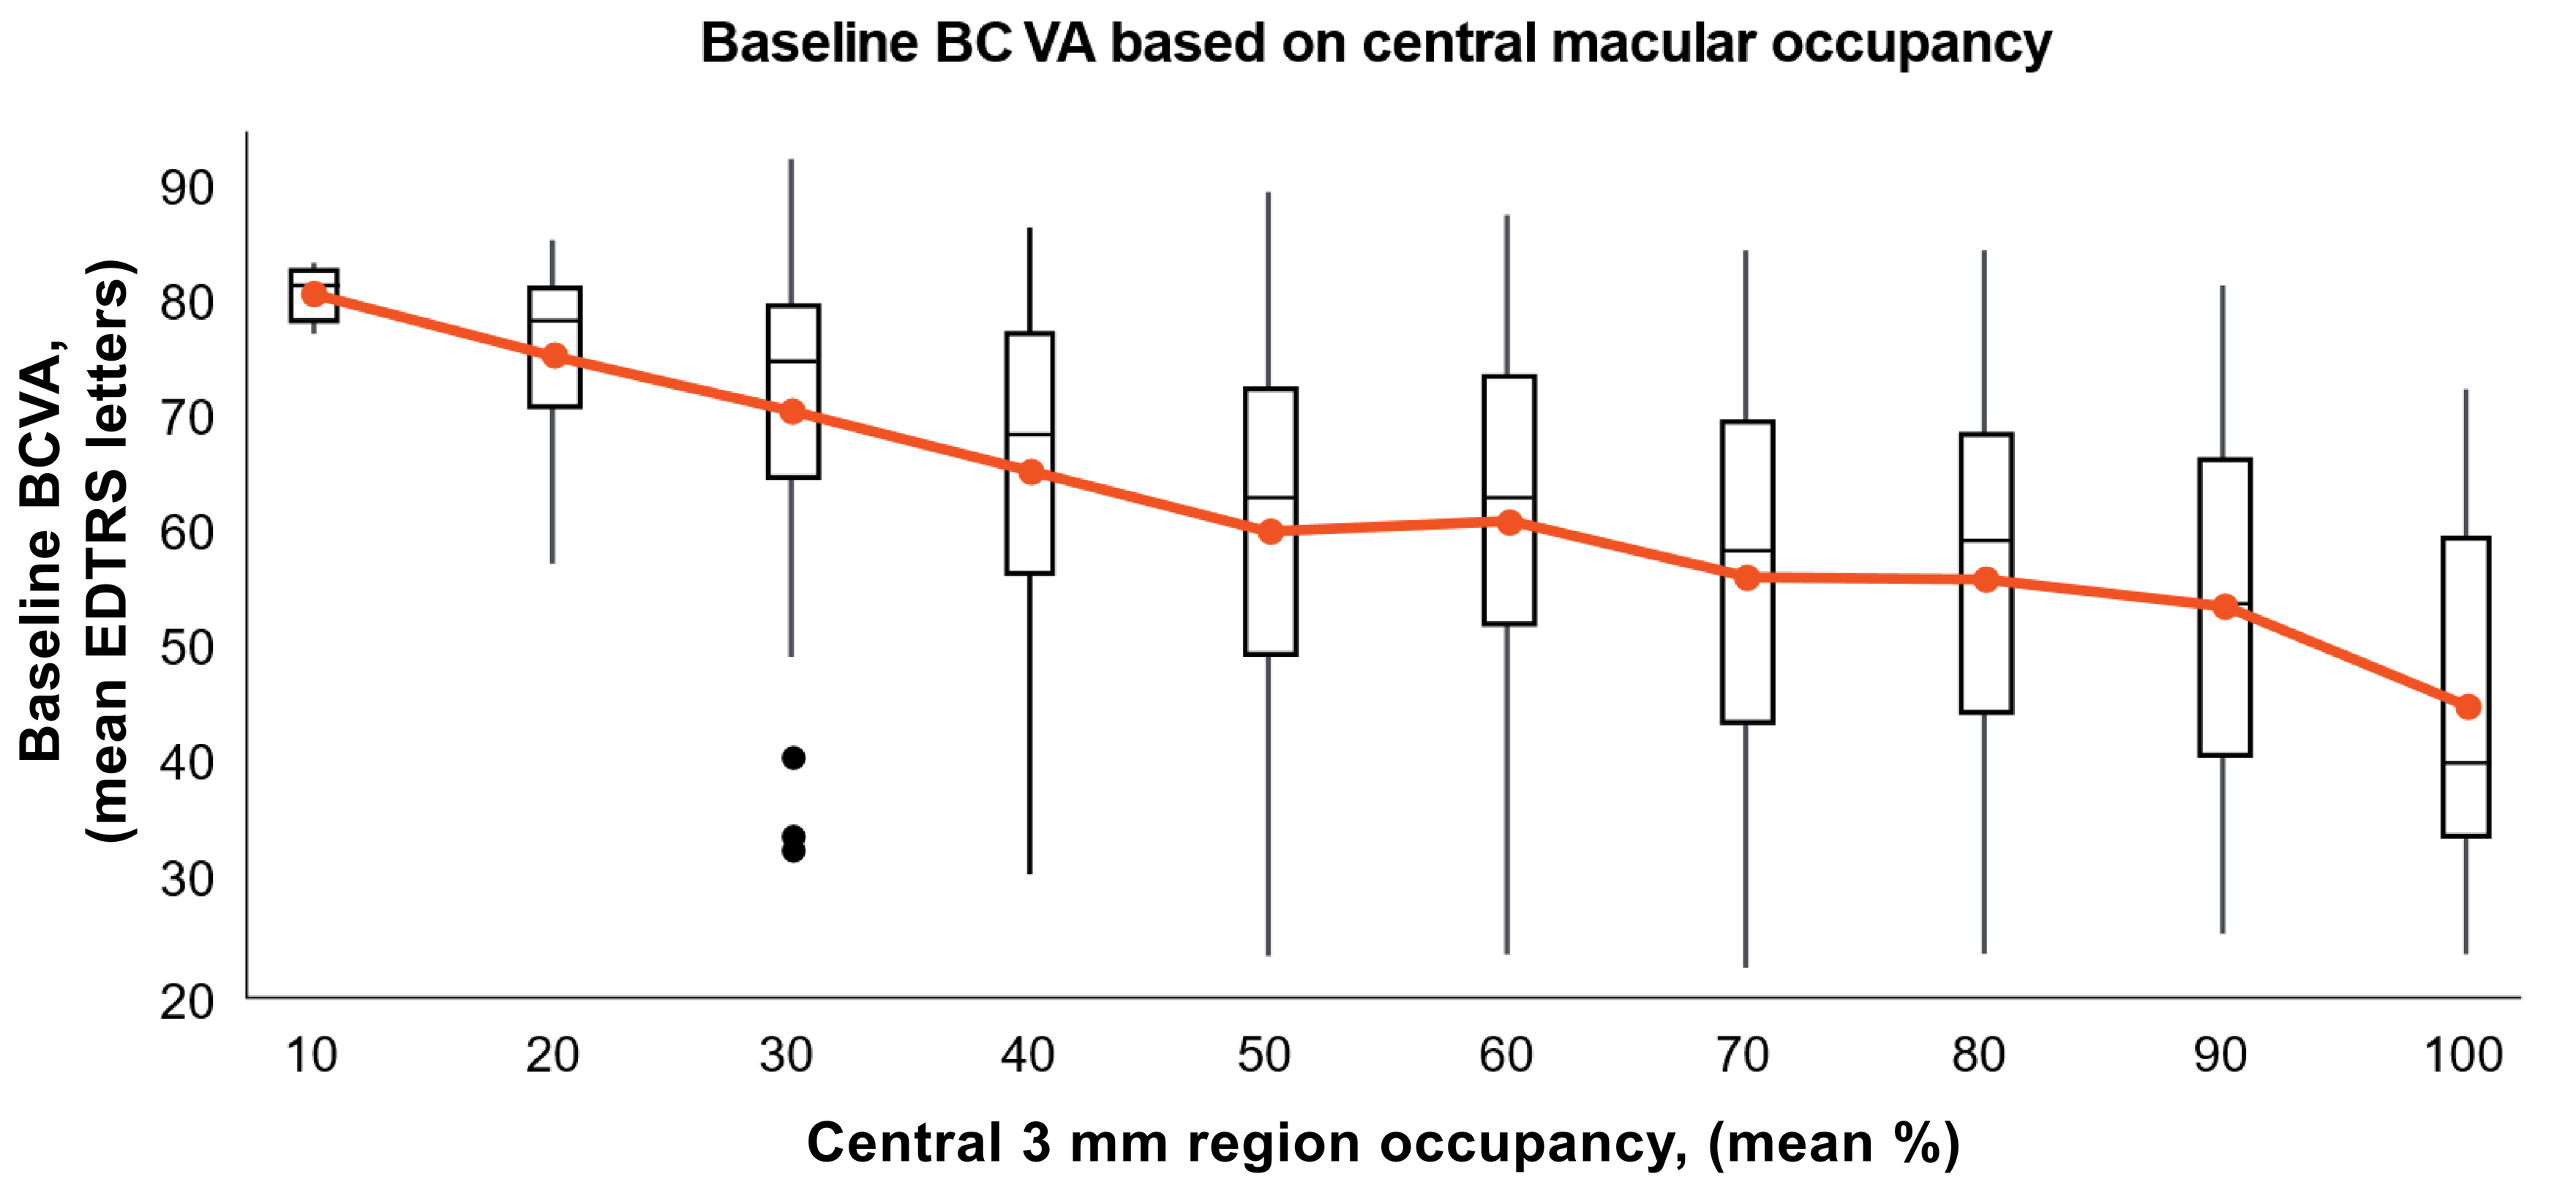
Baseline BC VA based on central macular occupancy
Baseline BCVA, (mean EDTRS letters)
90
80
70
60
50
40
30
20
10
20
30
40
50
60
70
80
90
100
Central 3 mm region occupancy, (mean %)

BCVA, best-corrected visual acuity.

- At 24 months, PEG was associated with 5.6 fewer ETDRS letters (~1 line on Snellen VA chart) lost (**Figure 3**) in patients with GA lesions ≥250 µm from the foveal center, with the difference increasing with time, meaning where lesion location is critical to VA impact, covariate adjustment increased the estimated treatment effect from an observed +2.2 letters to an adjusted +5.6 letters (*P*=0.078). In data not shown, this slower vision loss was seen in both every month and EOM PEG-treated patients

Figure 3. BCVA change from baseline in patients with GA lesions ≥250 µm from the foveal center

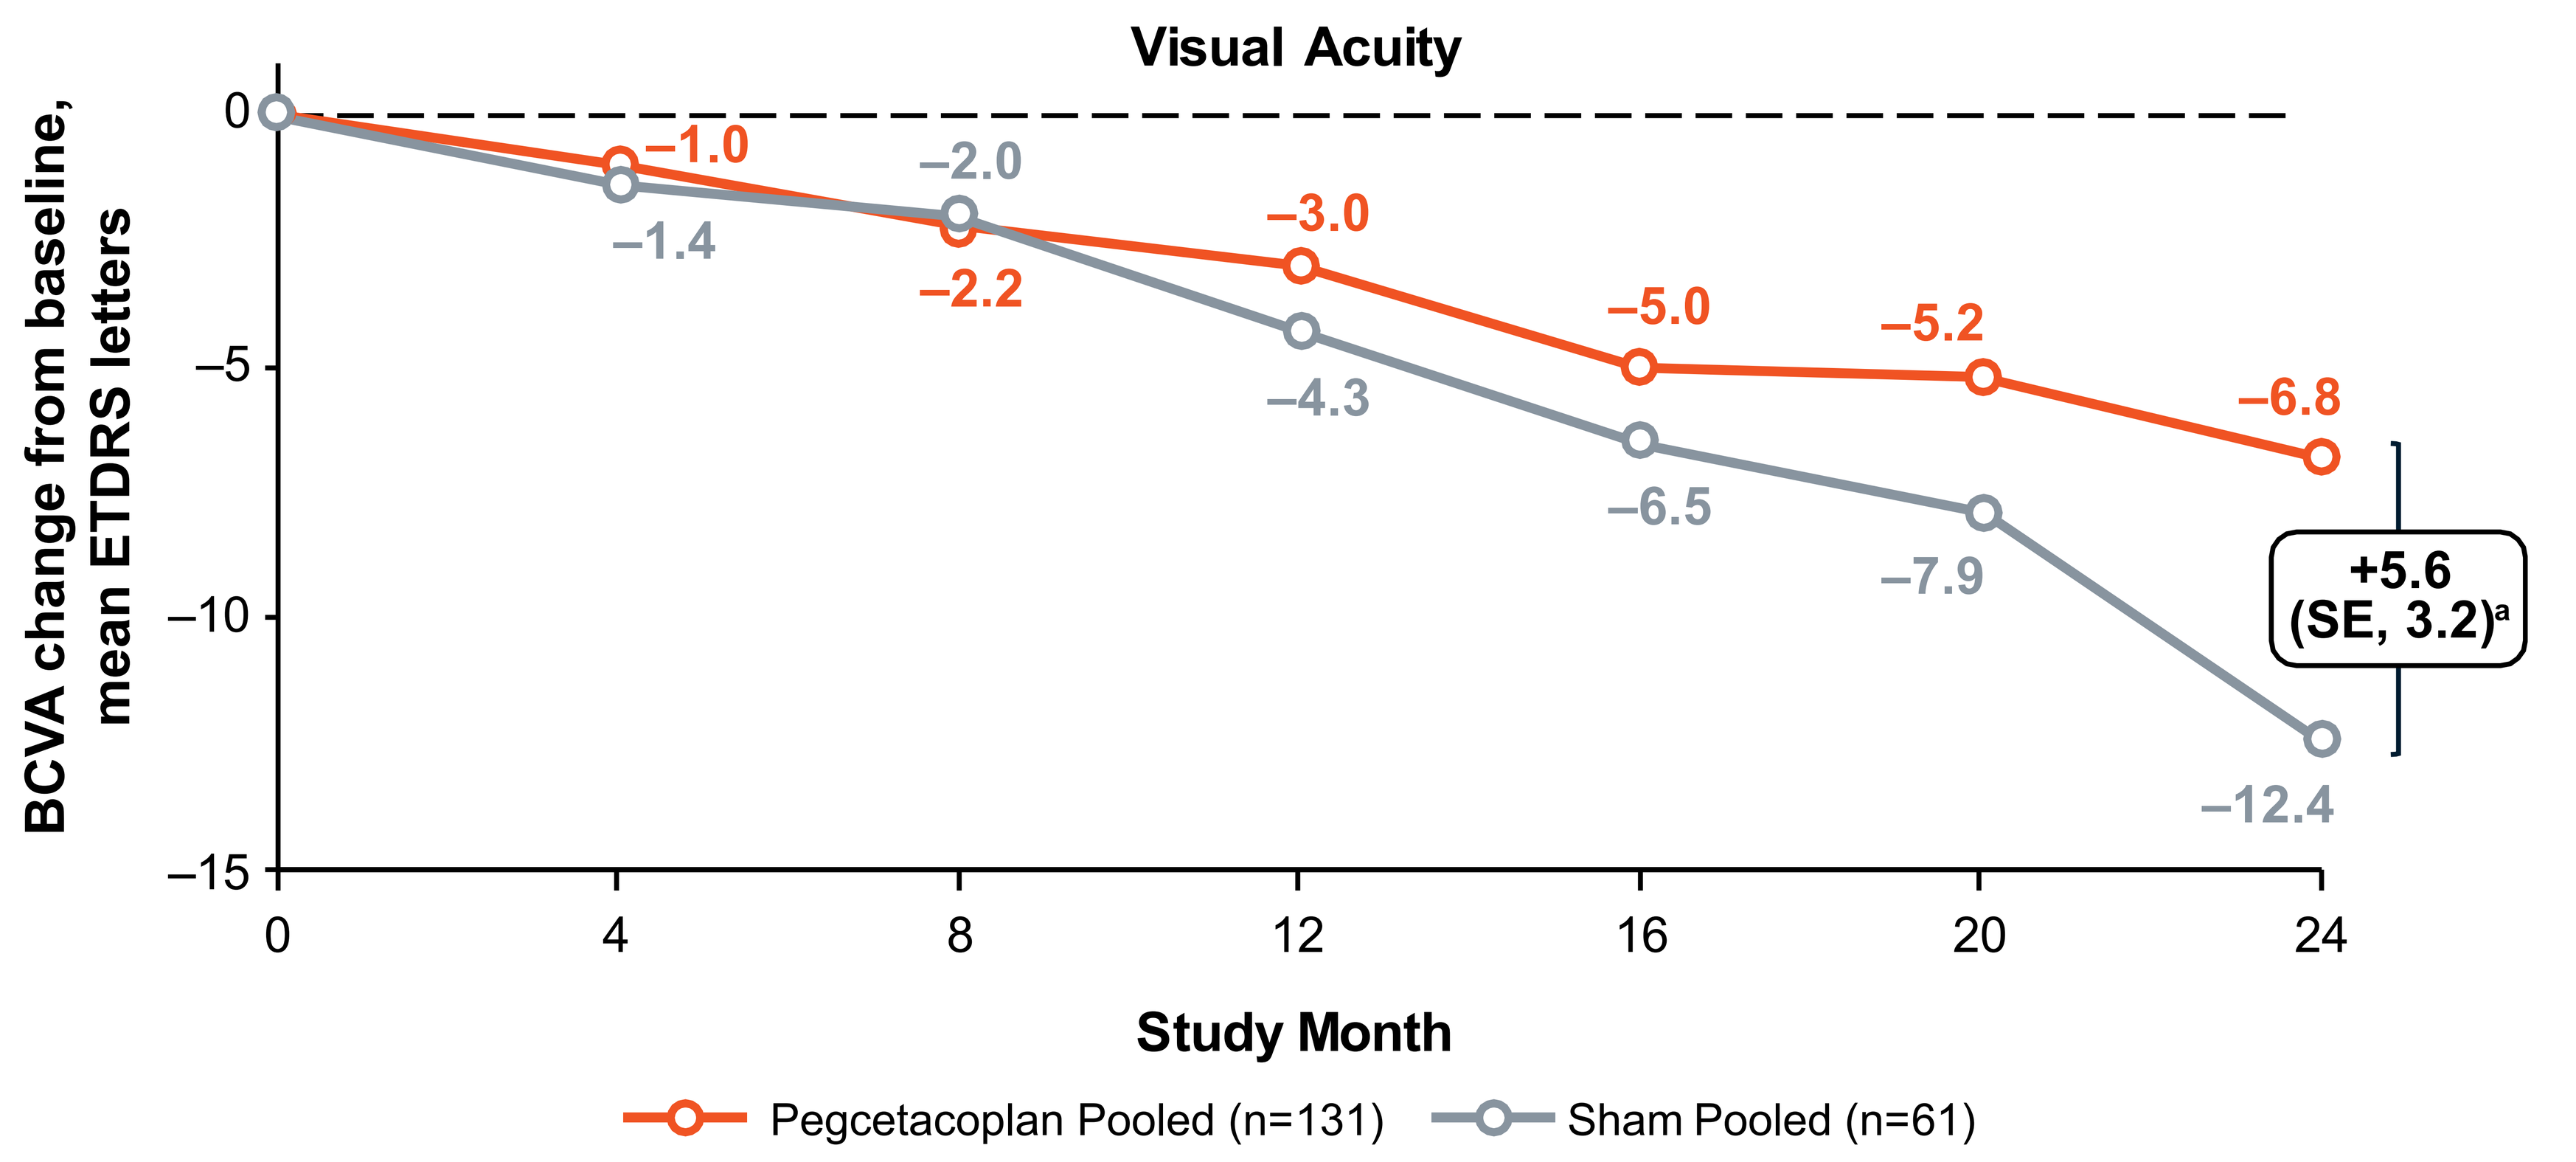
Visual Acuity
BCVA change from baseline, mean ETDRS letters
0
–5
–10
–15
–1.0
–1.4
–2.0
–2.2
–3.0
–4.3
–5.0
–6.5
–5.2
–7.9
–6.8
–12.4
+5.6 (SE, 3.2)a
0
4
8
12
16
20
24
Study Month
Pegcetacoplan Pooled (n=131)
Sham Pooled (n=61)

[a]Model-adjusted for the covariates: Baseline study eye characteristics: BCVA, proportion of the central 3-mm region (ETDRS grid areas 1–5) occupied by the GA lesion as defined by RPE loss (foveal occupancy), ≥7.5 mm² lesion size, pseudophakia, subfoveal location, multifocal configuration; baseline fellow eye characteristics: bilateral GA, better- vs worse-seeing eye, CNV, pseudophakia; patient demographics: age ≥75 years, female, location.
BCVA, best-corrected visual acuity; CNV, choroidal neovascularization; ETDRS, Early Treatment Diabetic Retinopathy Study; GA, geographic atrophy; RPE, retinal pigment epithelium; SE, standard error.

Conclusion

- Adjustment methods, widely accepted in observational research, are now increasingly being applied to secondary outcomes and subgroup analyses of RCTs. In these phase 3 trials, adjustment for baseline imbalances allowed for estimation of treatment effect on functional measures, revealing a VA treatment benefit once methods were used to adjust for location and CMO of the GA lesion

References

1. Rahimy E, Khan MA, Ho AC, et al. Progression of geographic atrophy: retrospective analysis of patients from the IRIS® registry (Intelligent Research in Sight). *Ophthalmol Sci*. 2023;3(4):100318.(3). **2.** Bakri SJ, Bektas M, Sharp D, Luo R, Sarda SP, Khan S. Geographic atrophy: mechanism of disease, pathophysiology, and role of the complement system. *J Manag Care Spec Pharm*. 2023;29(5-a Suppl):S2-S11. **3.** SYFOVRE™ (Pegcetacoplan Injection), for Intravitreal Use [Prescribing Information]. Apellis Pharmaceuticals; February 2023. https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/217171s000lbl.pdf. Accessed August 20, 2024. **4.** Heier JS, Lad EM, Holz FG, Rosenfeld PJ, Guymer RH, Boyer D, Grossi F, Baumal CR, Korobelnik JF, Slakter JS, Waheed NK. Pegcetacoplan for the treatment of geographic atrophy secondary to age-related macular degeneration (OAKS and DERBY): two multicentre, randomised, double-masked, sham-controlled, phase 3 trials. *The Lancet*. 2023 Oct 21;402(10411):1434-48. **5.** Brader HS, Ying GS, Martin ER, Maguire MG. Characteristics of incident geographic atrophy in the complications of age-related macular degeneration prevention trial. *Ophthalmology*. 2013;120(9):1871-1879. **6.** Sayegh RG, Sacu S, Dunavölgyi R, et al. Geographic atrophy and foveal-sparing changes related to visual acuity in patients with dry age-related macular degeneration over time. *Am J Ophthalmol*. 2017;179:118-128. **7.** Bagheri S, Lains I, Silverman RF, Kim I, Eliott D, Silva R, Miller J, Husain D, Miller JW, Saad L, Vavvas DG. Percentage of foveal vs total macular geographic atrophy as a predictor of visual acuity in age-related macular degeneration. *J Vitreoretin Dis*. 2019 Sep;3(5):278-82.

Acknowledgments

The authors and Apellis thank the patients, caregivers, investigators, and staff who participated in this study. Professional medical writing support was provided by Andrea R. Waksmunski, PhD, and Lisa Feder, PhD (OPEN Health Communications), and funded by Apellis Pharmaceuticals.

Contact information:
michele.intorcia@apellis.com

Presented at the EU ISPOR 2024; November 17–20, 2024; Barcelona, Spain