Drug Utilization and Expenditure in an Insurance Company for Poor Population of the Colombian Caribbean, 2021-2023

RWD169

J. ALVIS **[1], *K. GAMERO*** **[1], *G. HEREDIA*** **[2], *R. TORRES*** **[2], *J. MARTINEZ*** **[2], *E. CONEO*** **[2], *G. VIANA*** **[2], *NJ. ALVIS*** **[3], *N. ALVIS*** **[3]**

1Distribuciones Pharmaser, Cartagena, Colombia, 2Mutualser EPS, Cartagena, Colombia, 3 Universidad de la Costa, Barranquilla, Atlántico, Colombia

Introduction

The Colombian health system covers 98% of the population and 98% of the existing medications.

Objective

The aim of this study was to assess the utilization and drug expenditure evolution in an insurance company for poor population of the Colombian Caribbean.

Methods

A cross-sectional descriptive study was conducted based on the individual records of outpatient dispense records from the insurance company. The number of insured population per year, the annual healthcare resource utilization, and the total drug expenditures were considered. Additionally, the average annual usage rate and the mean cost per patient attended, along with their confidence intervals, were estimated. RMR: $4.090.

Results

During 2021-2023, the number of insured population grew by 7.6% (2021: 2.43 million - 2023: 2.62 million), while the healthcare resource utilization increased by 21.8% (2021: 48.45 million - 2023: 59.02 million).
Drug utilization doubled in the analyzed period (2021: 6.8 million - 2023: 10.5 million), and its expenditure grew by 115% (2021: $71.7 million USD - 2023: $154.7 million USD).
The average drug expenditure per affiliated person was $62 in 2021 and $104 USD in 2023. The number of patients attended increased 28.8% (2021: 1,151,069 patients - 2023: 1,482,283 patients).

Conclusions

This study provides key inputs for decision-making to allocate resources in the health insurance system of Colombia. Our results depicted the increasing high economic burden that the Colombian health system bears to care for the sick population.
Resources allocated to the insurance companies only grew by 35% in the last two years, showing a disproportion between this allocation and the current healthcare resource utilization and expenditure.

Table 1. Estimation of Drug utilization and Expenditure in an insurance company in Colombia. 2021-2023

Variable	2021	2023	Percentage change
Number of insured users	2,435,138	2,625,529	7.8%
Number of drug dispensations	48,450,000	59,027,894	21.8%
Drug consumption	6,838,329	10,509,381	53.7%
Drug utilization rate	14%	18%	
Expenditure (Million US Dollar)	$71.7	$154.7	115.8%
Average Drug Expenditure per Affiliated Person (US Dollar)	$62	$104	67.7%
Patients treated	1,151,069	1,482,283	28.8%

Contact information

¡Escanéeme! - Scan Me!

www.pharmaser.com.co www.mutualser.org