Understanding Patient Preferences of Patients with Type 2 Diabetes Mellitus in Second-Line Drug Intervention: A Discrete Choice Experiment

Ann-Kathrin Fischer[1 2], Andrew Sadler[2], Magdalene Bravo Guijarro[3], Elke Mathey[3], Axel Mühlbacher[1 2]

[1] Health Economics and Health Care Management, Hochschule Neubrandenburg, Germany
[2] Gesellschaft für empirische Beratung GmbH, Affiliated Institute of the University of Applied Sciences Neubrandenburg, Germany
[3] Novo Nordisk Pharma GmbH, Mainz, Germany

Poster Session 3: PCR143

Background

- Second-line interventions (escalation) in Type 2 Diabetes mellitus (T2D) are significant in healthcare due to rising global prevalence.
- Clinical guidelines such as the German National Care Guideline (NVL) offer various second-line options.
- There is a lack of clear understanding of patient preferences when choosing second-line options.

Study Objective

- Aim is to assess the relative importance of attributes associated with second-line intervention for T2D from the patient's perspective in Germany.

Method: Discrete Choice Experiment

- Adult T2D patients recruited in Germany (August-November 2023)
- Two populations: Population 1 (experienced in monotherapy) and Population 2 (experienced in second-line intervention).
- Mixed-methods approach employed for a Discrete Choice Experiment (DCE): literature review, formative quality interviews, pre-tests, and final quantitative data collection via online surveys.
- DCE involved 8 attributes, a partial profile design with 12 choice tasks, each offering 3 treatment alternatives which systematically varied in benefits, side effects, and administration attributes.
- Statistical analysis was conducted using a conditional logit model (CL) and a random parameter logit model (RPL) to examine both the main effects and heterogeneity in patients' preferences.

Results: Patient Preferences for Second-line Intervention

- 615 patients completed the survey with a final study population of 583 adult patients (292 in Population 1; 291 in Population 2) after data cleansing.
- Most important attributes influencing choice decision: risk of nausea, risk of nerve damage, and weight change. Followed by type and frequency of intake that was considered more important than risk of myocardial infarction, risk of stroke, and risk of severe hypoglycemia. Intake schedule had the least impact on choice decisions (**Table 1, Figure 1-2**).
- Both populations exhibited similar average preferences, but large standard deviations indicated considerable heterogeneity, especially regarding weight changes. Also notable is the attribute type and frequency of intake . (**Table 1**)
- Sample was divided by BMI for CL analysis: BMI <25 (underweight or normal weight), BMI = 25-29.9 (overweight), BMI >= 30 (obese) to analyze differences regarding weight change. (**Figure 3**)
 - BMI <25: Clear preference for lower weight loss (2 kg vs. 6 kg).
 - BMI 25-29.9: Indifference between 2 kg and 6 kg weight loss.
 - BMI >=30: Clear preference for higher weight loss (6 kg vs. 2 kg).

Discussion

- Individual respondents may react differently to specific attributes within both populations. Existence of specific subgroups of patients with varying preferences, especially regarding weight changes and type/frequency of intake, suggests potential for further subgroup analysis.
- Results support need for personalized treatment approaches and relevance of shared decision making considering individual preferences when switching patients from monotherapy to second line treatment.

		Population 1 (N=292)				Population 2 (N=291)			
Attributes	Levels	Mean	p	SD	p	Mean	P	SD	p
Risk of myocardial infarction	0 out of 100 patients (0%)	0.84	0.00	0.25	0.10	0.92	0.00	0.26	0.36
	2 out of 100 patients (2%)	0.22	0.00	-0.10	0.50	0.21	0.00	0.23	0.10
	4 out of 100 patients (4%)	-0.21	0.00	0.00	0.98	-0.36	0.00	-0.03	0.85
	7 out of 100 patients (7%)	-0.86	0.00	-0.15	0.53	-0.78	0.00	-0.46	0.17
Risk of stroke	0 out of 100 patients (0%)	0.64	0.00	.	.	0.73	0.00	.	.
	1 in 100 patients (1%)	0.16	0.03	.	.	0.25	0.00	.	.
	2 out of 100 patients (2%)	-0.12	0.08	.	.	-0.29	0.00	.	.
	4 out of 100 patients (4%)	-0.67	0.00	.	.	-0.69	0.00	.	.
Risk of nerve damage	0 out of 100 patients (0%)	1.76	0.00	-0.46	0.00	2.01	0.00	-0.26	0.08
	5 out of 100 patients (5%)	0.36	0.00	-0.16	0.33	0.38	0.00	-0.34	0.02
	10 out of 100 patients (10%)	-0.28	0.00	-0.24	0.18	-0.19	0.01	-0.19	0.16
	15 out of 100 patients (15%)	-1.85	0.00	0.86	0.00	-2.20	0.00	0.79	0.00
Risk of nausea	0 out of 100 patients (0%)	3.17	0.00	0.49	0.00	3.19	0.00	0.63	0.00
	10 out of 100 patients (10%)	1.34	0.00	0.25	0.20	1.30	0.00	0.42	0.00
	30 out of 100 patients (30%)	-1.40	0.00	-0.74	0.00	-1.61	0.00	0.67	0.00
	50 out of 100 patients (50%)	-3.11	0.00	0.00	0.99	-2.88	0.00	-1.73	0.00
Risk of severe hypoglycemia	0 out of 100 patients (0%)	0.69	0.00	.	.	0.77	0.00	.	.
	1 in 100 patients (1%)	0.21	0.00	.	.	0.12	0.08	.	.
	2 out of 100 patients (2%)	-0.26	0.00	.	.	-0.20	0.01	.	.
	4 out of 100 patients (4%)	-0.63	0.00	.	.	-0.69	0.00	.	.
Weight change	Decrease of -6kg	1.05	0.00	0.94	0.00	1.46	0.00	0.98	0.00
	Decrease of -2kg	1.31	0.00	0.86	0.00	1.41	0.00	0.59	0.00
	Increase of +2kg	-0.36	0.00	0.36	0.05	-0.51	0.00	0.43	0.00
	Increase of +6kg	-2.00	0.00	-2.16	0.00	-2.35	0.00	-2.00	0.00
Type and frequency of intake	Oral 1x per week	0.80	0.00	0.68	0.00	0.68	0.00	0.45	0.00
	Oral 7 times a week	-0.18	0.00	0.23	0.07	-0.27	0.00	0.25	0.00
	Injection 1x a week	0.49	0.00	0.18	0.24	0.44	0.00	0.27	0.01
	Injection 7x a week	-1.11	0.00	-1.09	0.00	-0.85	0.00	-0.97	0.00
Schedule of intake	Independent of meals in the morning	0.07	0.17	-0.04	0.85	0.04	0.41	.	.
	Dependent on meals in the morning	-0.10	0.06	0.03	0.87	-0.17	0.00	.	.
	Independent of meals in the evening	0.03	0.60	-0.04	0.79	0.07	0.17	.	.
	Dependent on meals in the evening	0.00	0.98	0.05	0.86	0.05	0.31	.	.
	Log likelihood (model)	*1963.12*				*-1882.74*			
	Degrees of freedom	*42*				*39*			
	AIC	*4010.25*				*3843.48*			
	BIC	*4315.18*				*4126.49*			

Mean= mean coefficients; p= p-value; SD= standard deviations
The sign of the estimated standard deviations is irrelevant: interpret them as being positive.

Table 1: Parameter estimates of the random parameter logit model

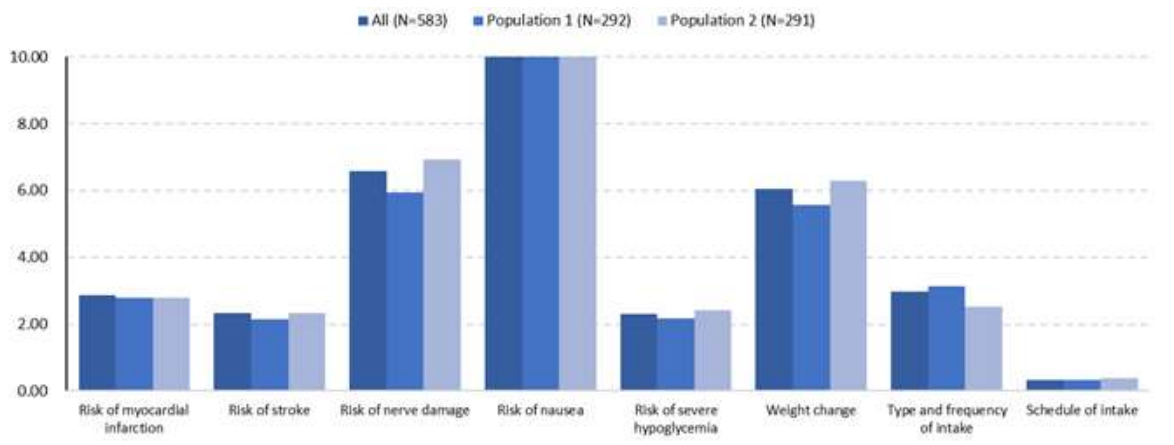

All (N=583)
Population 1 (N=292)
Population 2 (N=291)
Risk of myocardial infarction
Risk of stroke
Risk of nerve damage
Risk of nausea
Risk of severe hypoglycemia
Weight change
Type and frequency of intake
Schedule of intake

Figure 1: Relative attribute importance standardized to size 10 based on the most important attribute); Type and frequency of intake: Distinction between oral intake and injection in relation to frequency per day or per week.; Schedule of intake: Instructions on medication intake regarding timing in relation to meals and time of day.

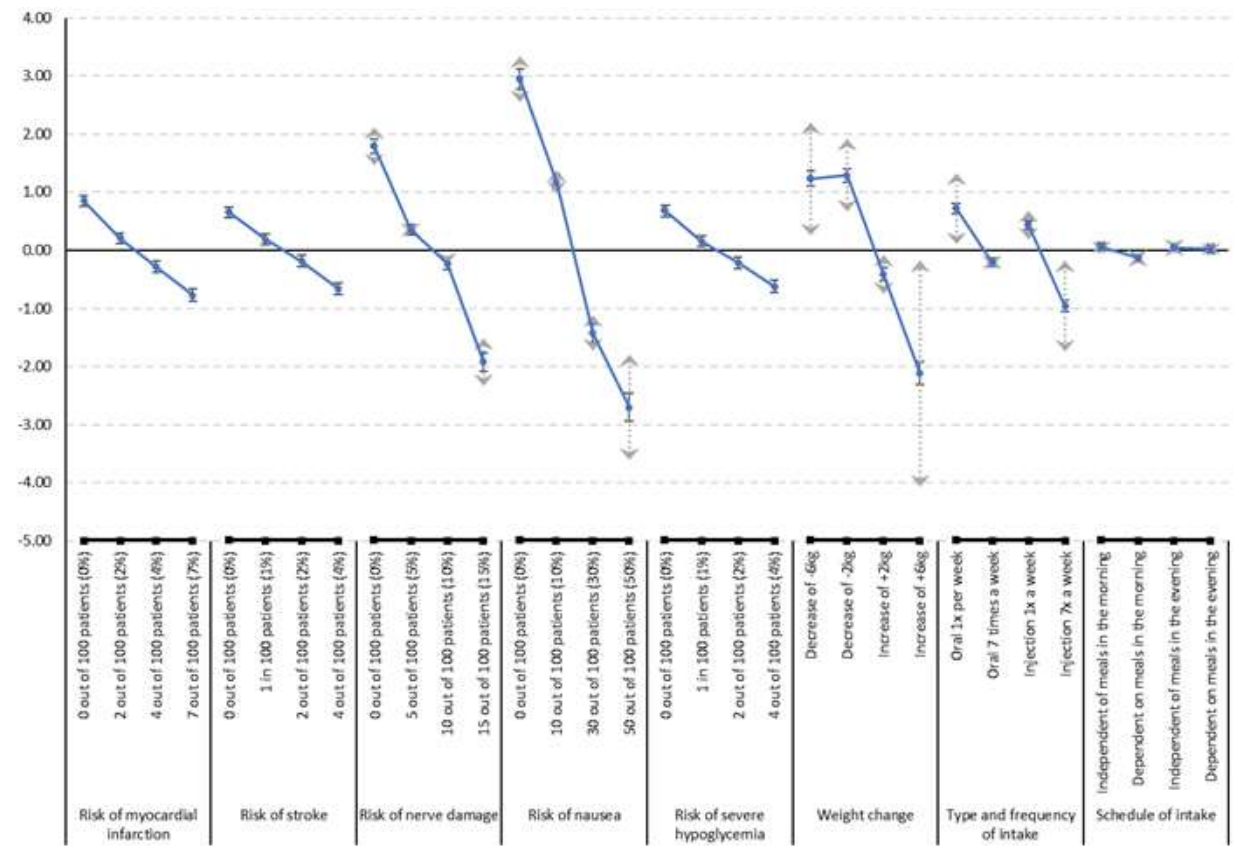

Risk of myocardial infarction
Risk of stroke
Risk of nerve damage
Risk of nausea
Risk of severe hypoglycemia
Weight change
Type and frequency of intake
Schedule of intake

Figure 2: RPL N = 583, Coefficients in 95% confidence interval and standard deviations (grey arrows

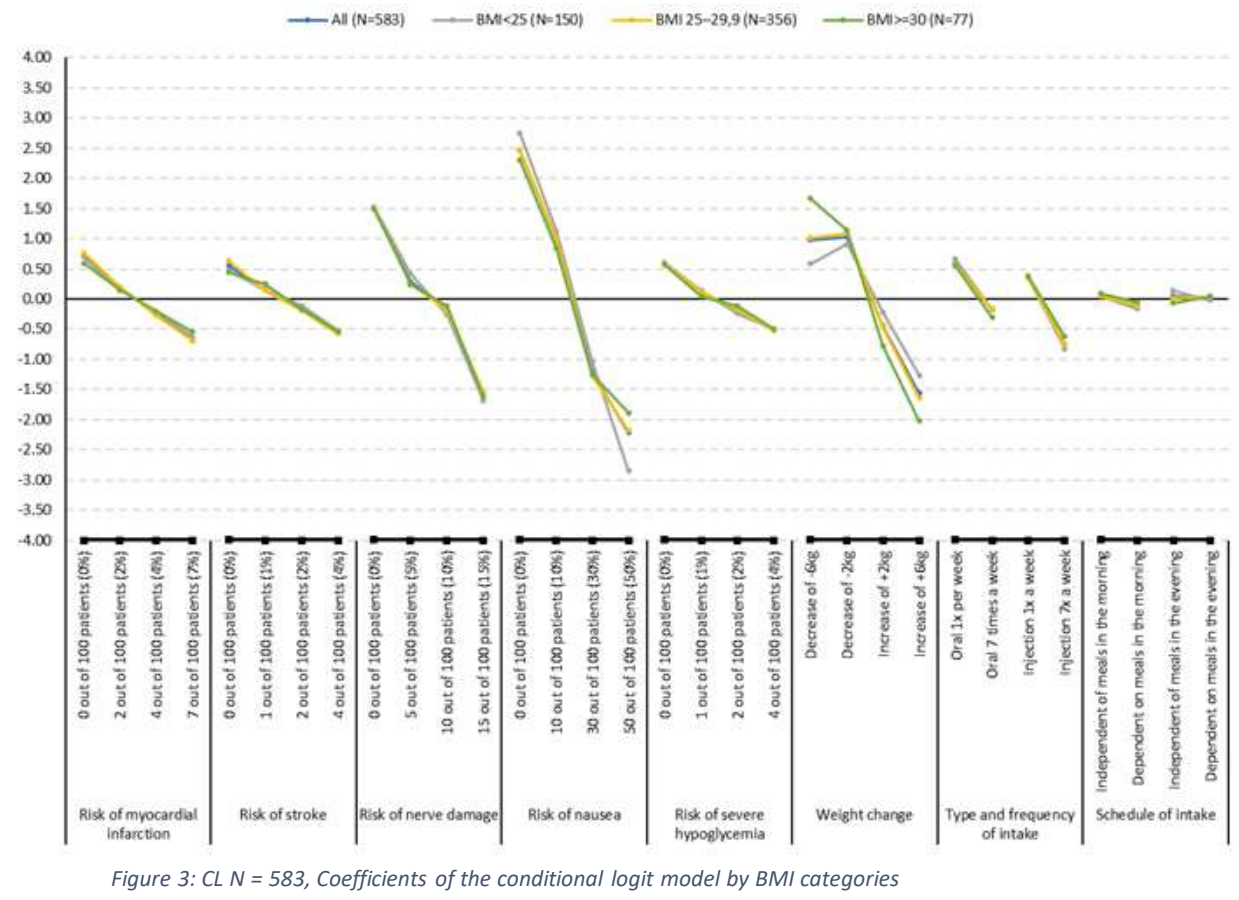

All (N=583)
BMI<25 (N=150)
BMI 25–29,9 (N=356)
BMI>=30 (N=77)
Risk of myocardial infarction
Risk of stroke
Risk of nerve damage
Risk of nausea
Risk of severe hypoglycemia
Weight change
Type and frequency of intake
Schedule of intake

Figure 3: CL N = 583, Coefficients of the conditional logit model by BMI categories

References: Available upon request**; Disclosure:** The research project is funded by Novo Nordisk Pharma GmbH; **Institutional Review Board:** Ethics Committee at Hochschule Neubrandenburg (HSNB/195/22); **Correspondence**: fischer@empirische-beratung.de