CARDIOVASCULAR AND METABOLIC RISK IN PATIENTS WITH CLASSIC CONGENITAL ADRENAL HYPERPLASIA: RECENT FINDINGS FROM A SYSTEMATIC LITERATURE REVIEW

SA118

M. HARDING[1], E. LACHARME[2], P. REEVES[1], R. SOHAIL[1] and J. FERRER[1]

[1]Decisive Consulting, London, United Kingdom; [2]Neurocrine Biosciences, Basel, Switzerland

INTRODUCTION

- **Classic congenital adrenal hyperplasia** (CAH) is a group of genetic disorders characterised by **impaired cortisol synthesis**, **requiring lifelong glucocorticoid (GC) therapy**, which, while essential, often involves supraphysiologic doses **that may contribute to long-term health complications**.[1,2]
- Among such long-term health complications, **cardiovascular and metabolic (CV/M) complications** pose a **notable risk** to patients.[3]

OBJECTIVE

- The objective of this poster is to **present findings from a targeted update** of a 2021 Global Value Dossier (GVD) **discussing the burden of CAH**. The update, conducted as an SLR in June 2024, **aimed to gather recent evidence across multiple aspects of CAH**, including its epidemiology, clinical and economic burden, treatment landscape, and emerging therapies.
- While the update covered various topics, **this poster focuses specifically on evidence regarding CV/M health complications in 21-hydroxylase deficiency (21-OHD) CAH patients**, which comprise 95% of cases of CAH. This emphasis reflects the identification of studies investigating links between GC treatment and CV/M outcomes in this population.[4–8] This update was not intended to be a comprehensive SLR focusing on one single objective, but rather a focused effort to identify relevant publications from the specified time period below to supplement the existing evidence base. The findings presented here should be considered in conjunction with a broader body of published literature on CAH.

METHODS

A targeted literature update was performed, searching PubMed and the Directory of Open Access Journals (DOAJ) for studies published **between November 2022 and May 2024**. This 18-month timeframe was chosen to capture recent evidence, with the aim of updating the 2021 GVD with current data.

The search strategy was designed to identify literature addressing multiple aspects of CAH, including:

1. Impact of chronic supraphysiological GC dosing
2. Epidemiology of CAH
2. Clinical burden and prognosis
3. Economic burden and cost-effectiveness
5. Current treatment landscape
6. Surrogate endpoints and biomarkers
7. Emerging therapies

While the search strategy encompassed all these areas, for this presentation, **data specifically related to CV/M complications in CAH patients were extracted and analysed**. The update followed the Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) guidelines. Inclusion criteria encompassed observational studies, randomized controlled trials, systematic reviews, and meta-analyses. Two independent reviewers screened titles, abstracts, and full texts based on predefined criteria.

RESULTS

- **Searches yielded 1064 articles, of which 123 proceeded to full-text review and 92 to inclusion in the final review.**
- After a comprehensive review of the study objectives and key outcomes of those 93 studies, **five main categories emerged:**

1. Clinical Management and Treatment Outcomes (n = 27)
2. GC Usage, Treatment Burden, and Associated Morbidities in CAH (n = 24)
3. Diagnosis, Screening, and Genetic Aspects (n = 13)
4. Quality of Life and Psychosocial Outcomes (n = 18)
5. Epidemiology and Healthcare Utilisation (n = 10)

- Filtering for study titles evaluating "cardiovascular", "cardiometabolic" or "cardiac" outcomes in the wider category of "GC Usage, Treatment Burden, and Associated Morbidities in CAH" revealed 7 studies, **of which 5 exclusively focused on CV/M complications and comprised a cohort of 21-OHD CAH patients.**
- Of the two studies excluded, one was excluded as it did not exclusively focus on CV/M complications and one was exclusively focused on a non-classical (NC-CAH) population, which whilst relevant for the overall SLR update, was not analysed here because NC-CAH cohort does not require lifelong GC therapy.[1,2]

Figure 1: PRISMA diagram – new studies identified via databases and registries

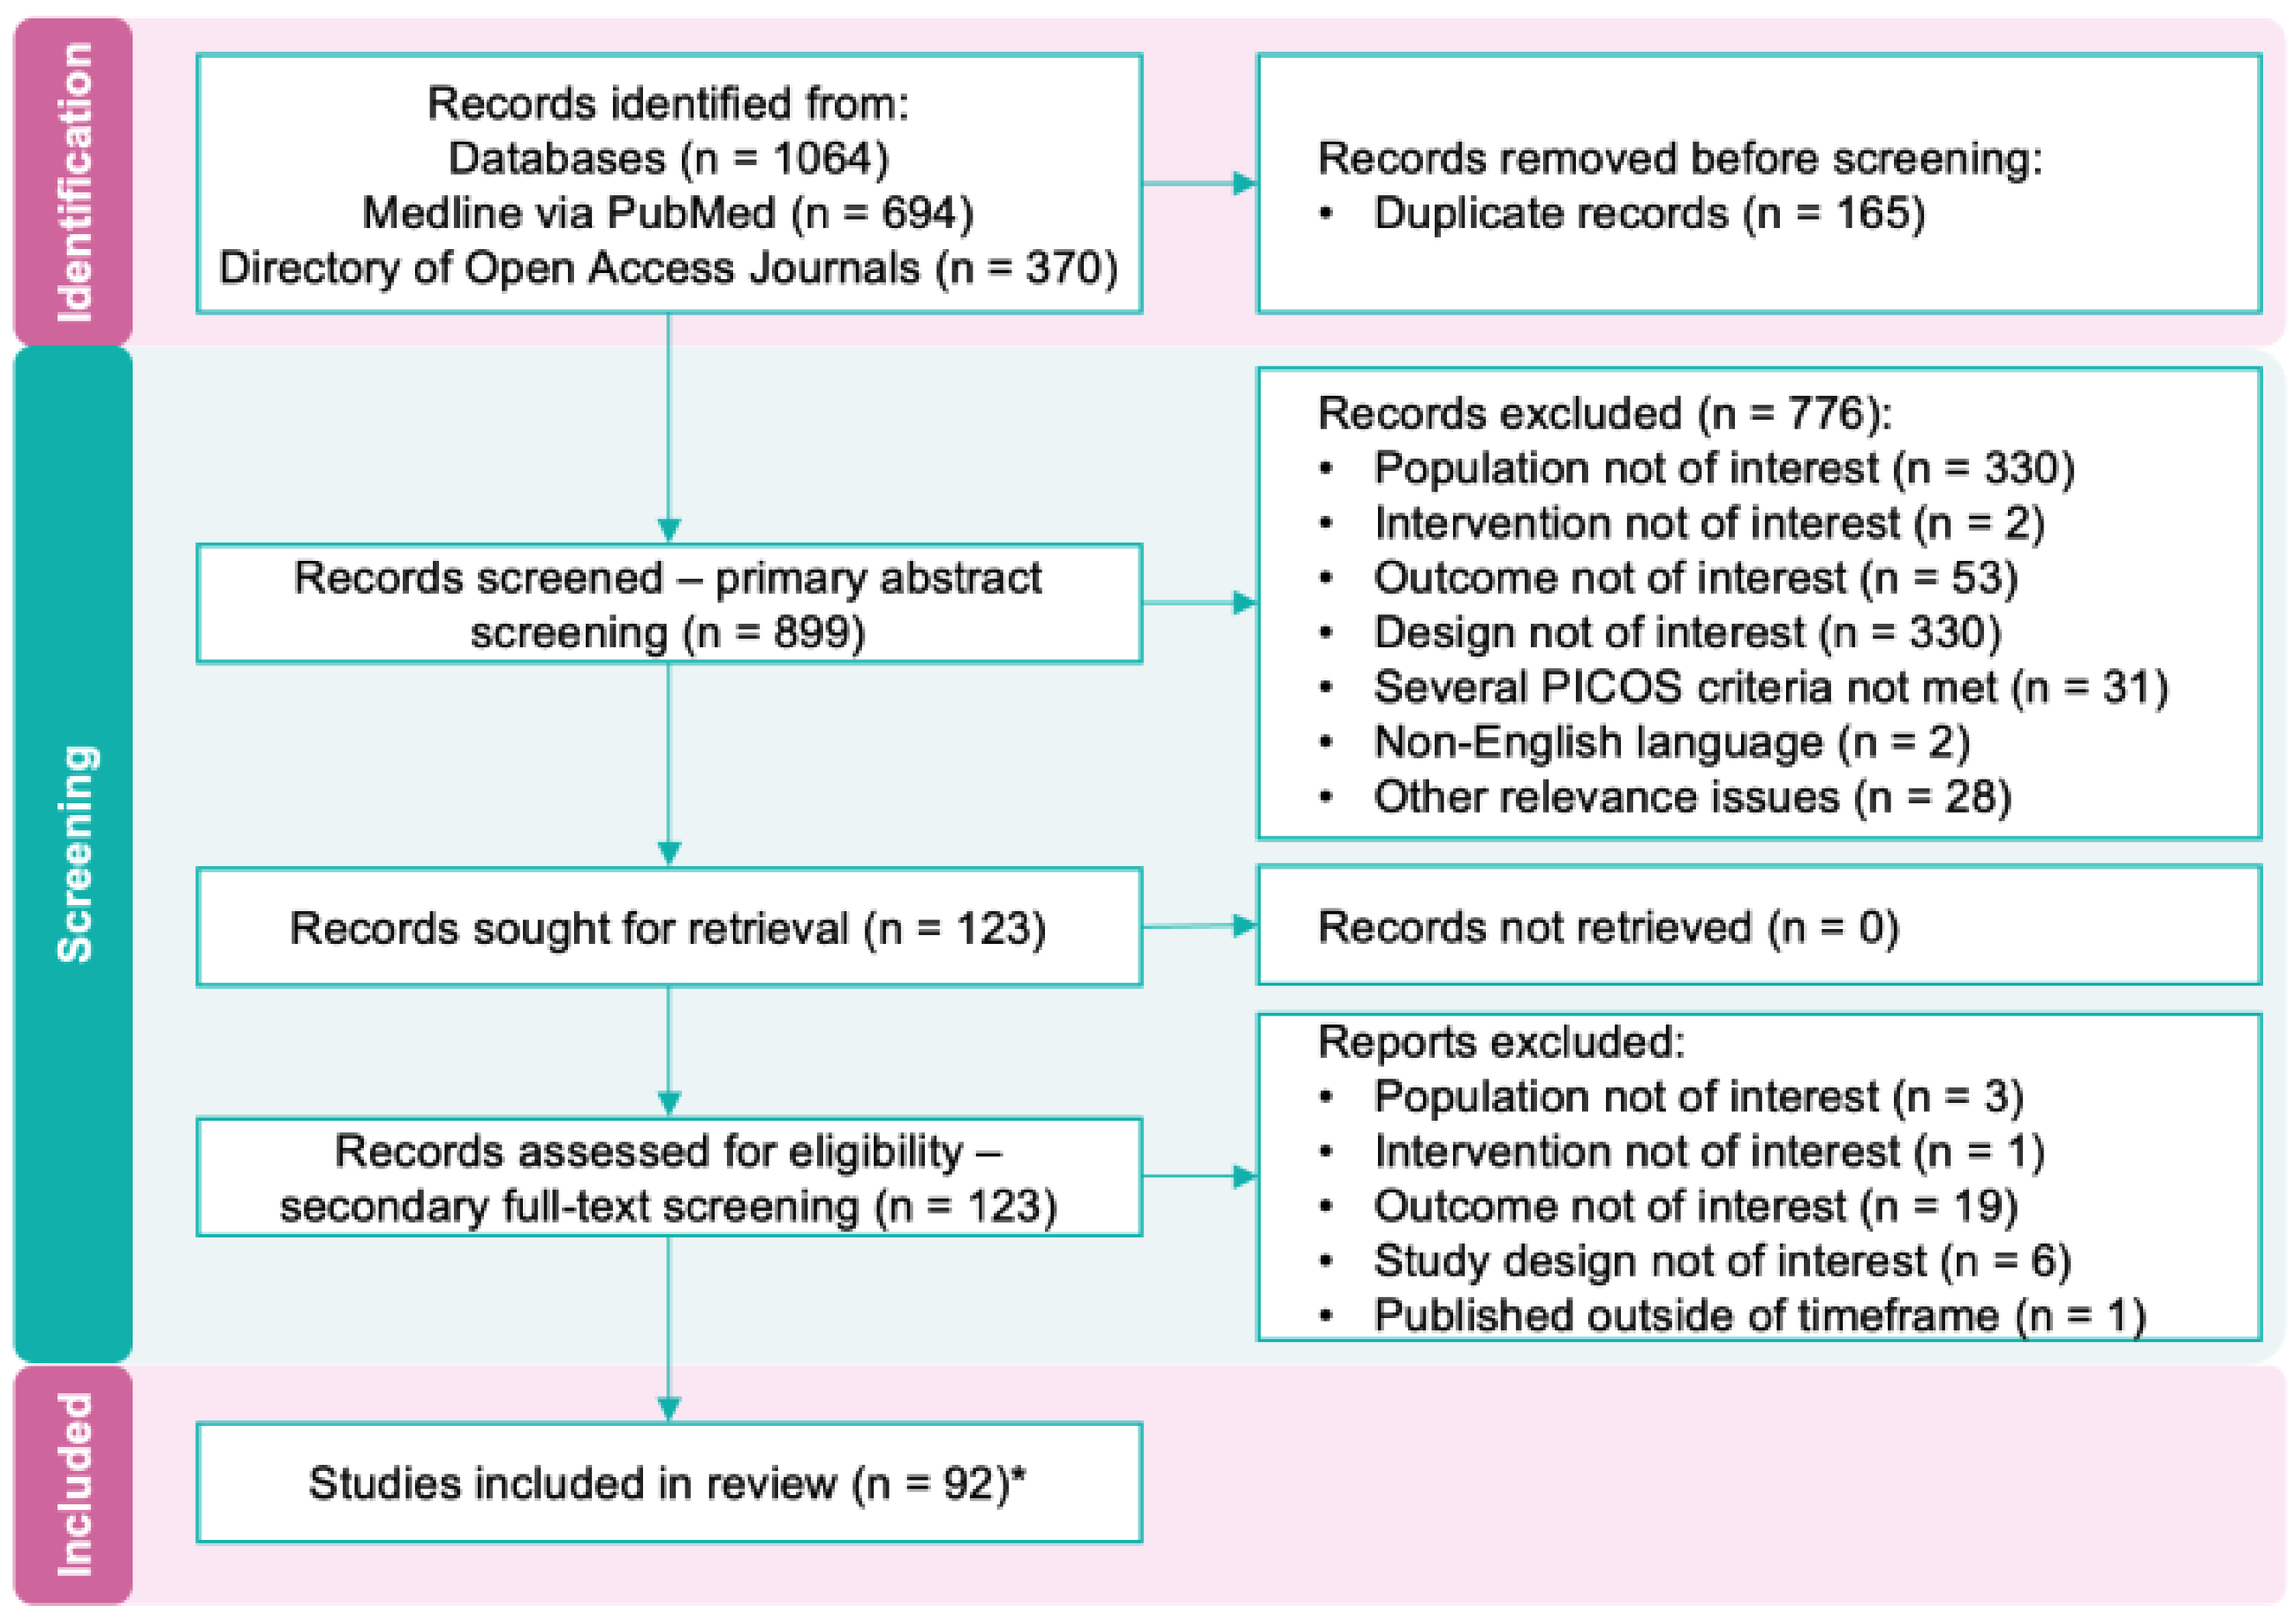
Identification
Records identified from:
Databases (n = 1064)
Medline via PubMed (n = 694)
Directory of Open Access Journals (n = 370)
Records removed before screening:
• Duplicate records (n = 165)
Screening
Records screened – primary abstract screening (n = 899)
Records excluded (n = 776):
• Population not of interest (n = 330)
• Intervention not of interest (n = 2)
• Outcome not of interest (n = 53)
• Design not of interest (n = 330)
• Several PICOS criteria not met (n = 31)
• Non-English language (n = 2)
• Other relevance issues (n = 28)
Records sought for retrieval (n = 123)
Records not retrieved (n = 0)
Records assessed for eligibility – secondary full-text screening (n = 123)
Reports excluded:
• Population not of interest (n = 3)
• Intervention not of interest (n = 1)
• Outcome not of interest (n = 19)
• Study design not of interest (n = 6)
• Published outside of timeframe (n = 1)
Included
Studies included in review (n = 92)*

* Note that the number of studies included in the final review is 92 and not 93 because two of the records concerned the same study.

Table 1: Details of the 5 identified studies containing CV/M risk data for 21-OHD CAH

Study	Population	Design	Key outcomes assessed
Charoensri et al. 2023[4]	254 US adults with CAH (186 females, 68 males; 147 classic [57.9%], 107 NC-CAH [42.1%]; median age 35 years [IQR 28.25, 46]). No controls were included in the study	Retrospective cross-sectional	Prevalence of cardiovascular diseases and risk factors
Chen et al. 2024[5]	1,647 US youth with CAH (547 classic [33.2%], remainder non-classic/unspecified; median age 12.9 years [7.3, 17.6]) and 6,588 propensity-matched controls	Retrospective cross-sectional	Odds of CV/M diagnoses compared to controls
Espinosa Reyes et al. 2023[6]	22 Cuban adolescents/young adults with CAH (13 classic, 9 NC-CAH; mean age 17.1 ± 5.5 years) and 22 sex-, age, BMI- and Tanner staged matched controls	Cross-sectional case-control	Endothelial dysfunction, carotid intima media thickness (CIMT), epicardial fat
Kara et al. 2023[7]	41 Turkish adults with classic CAH (mean age 30 ± 8 years) and 38 sex-, age-, BMI- matched controls	Cross-sectional case-control	Visceral adiposity index (VAI), Framingham risk score
Righi et al. 2023[8]	244 young adults with classic CAH from 13 centres (mean age 33 years [19, 94]). No controls were included in the study	Cross-sectional survey	Prevalence of CV/M comorbidities

Prevalence of CV/M risk factors in CAH patients

Chen et al. (2024)[5]

- **Prevalence findings (CAH patients vs. controls):**
 - Overweight/obesity: 63.6% vs. 32.5%
 - Hypertension: 16.7% vs. 6.1%
 - Dysglycaemia: 2.7% vs. 1.4%
 - Dyslipidaemia: 7.5% vs. 3.4%
 - Liver dysfunction: 12.5% vs. 5.9%
- **Compared to matched controls, the study found higher odds of:**
 - Overweight/obesity: OR (odds ratio) 3.63 (95% CI 3.24-4.07, p<0.0001)
 - Hypertension: OR 3.07 (95% CI 2.60-3.64, p<0.0001)
 - Dysglycaemia: OR 1.95 (95% CI 1.35-2.82, p<0.0001)
 - Dyslipidaemia: OR 2.28 (95% CI 1.79-2.91)
 - Liver dysfunction: OR 2.30 (95% 1.91-2.76, p<0.0001)

Charoensri et al. (2023)[4]

- Obesity is the most prevalent comorbidity in CAH patients at 42.1%, comparable to the CDC's reported 40% in adults 20-39 years of age. **Classic CAH patients showed significantly higher rates (54.4% male patients, 46.7% female patients) versus non-classic CAH (18.2% male patients, 33.3% female patients), with one likely factor being GC overtreatment (≥20 mg/day hydrocortisone-equivalent [Hce]).**

Surrogate markers, adiposity indices, and risk scores for cardiovascular risk in CAH patients

Espinosa Reyes et al. (2023)[6]

- Carotid intima media thickness (CIMT) is a surrogate marker for atherosclerosis progression. Mean CIMT was found to be:
 - 0.45mm in CAH vs. 0.42mm in controls (p=0.07)
 - 52% of CAH patients had increased CIMT vs. 27% of controls (p=0.09)
- **CIMT positively correlated with 17-hydroxyprogesterone levels (r=0.510, p=0.018), diastolic blood pressure (r=0.444, p=0.04), homeostatic model assessment of insulin resistance (HOMA-IR) index (r=0.507, p=0.01).**
- The authors note that CIMT is a well-established surrogate marker but not an outcome measure for cardiovascular events.
- Epicardial fat, measured by echocardiography, correlated with: total cholesterol levels (r=0.679, p<0.01), time since CAH diagnosis (r=0.462, p=0.03), GC dose (r=0.499, p=0.04).
- **The authors state that adolescents and young adults with CAH should be regularly assessed for cardiovascular risk and non-invasive methods such as measurement of epicardial fat or CIMT are useful as independent predictors of future cardiovascular events**.

Kara et al. (2023)[7]

- The Visceral Adiposity Index (VAI) is a model used to estimate visceral fat, which is a significant risk factor for metabolic and cardiovascular diseases (CVD(s)).
- Median VAI was found to be: 3.7 in CAH vs. 2.5 in controls (p=0.02).
- **GC dose (mean GC dose was 17±9 mg/day HCe) independently associated with VAI (β=0.17, p=0.018) and Framingham Risk Score (β=0.04, p=0.03).**

Long-term implications and comorbidities in CAH patients

Righi et al. (2023)[8]

- Of 244 adults with CAH:
 - 73 (30%) were treated for at least one CV/M (or bone) comorbidity
 - 9 (12%) treated for 2 comorbidities
 - 3 (4%) for 3 comorbidities
 - 3 (4%) for 4 comorbidities

Charoensri et al. (2023)[4]

- **CVD prevalence: Overall: 7.5% (vs. a 1% prevalence for CVD in U.S. adults). In CAH patients >60 years of age the CVD prevalence was found to be 25%.** CVD is defined in this study to include ischemic heart disease, heart failure, atrial fibrillation/flutter, pulmonary embolism, stroke, peripheral vascular disease, abdominal aortic aneurysm, acquired aortic stenosis and idiopathic pulmonary hypertension.
- Factors found to be independently associated with CVD:
 - Increasing age (adjusted OR 1.05 [95% CI 1.01-1.09], p=0.02),
 - Hypertension (adjusted OR 4.27 [95% CI 1.41-12.92], p=0.01),
 - **Higher GC doses (adjusted OR 1.51 [95% CI 1.11-2.06], p=0.01, per 10 mg/day HCe), with classic CAH patients receiving median doses of 30 mg/day (IQR 20,40) in males and 20 mg/day (IQR 15,30) in females..** The study found no significant relationship between cardiovascular events and biochemical markers of poorly controlled disease (17-hydroxyprogesterone, androstenedione, testosterone, and adrenocorticotropin hormone levels), hence the study author suggested that cardiovascular complications in these patients are **more strongly associated with higher GC exposure.**

DISCUSSION

LIMITATIONS:

- The predominance of cross-sectional studies limits causal inferences.
- **Variability in GC regimens** and adherence may confound the relationship between treatment and outcomes.
- **The impact of mineralocorticoid replacement on cardiovascular risk was often not specifically analysed**, representing a gap in understanding the full picture of treatment effects.
- The inadequacy **of cross-sectional androgen data to infer long-term disease control** further complicates the interpretation of cardiovascular risk in CAH patients.
- **The absence of long-term prospective data** impedes differentiation between risks attributable to CAH and its treatment versus other factors over time.
- **The limited number of studies (n = 5)** from the SLR focusing on CV/M risk restricts the generalisability of these findings and underscores the need for additional research in this area.
- The 18-month timeframe of the SLR, starting from 2022, **excluded relevant studies published prior to this period**. Notably, the National Institutes of Health (NIH) cohort study by Torky et al. (2021), which follows CAH patients in both adulthood and childhood, was not captured.[9] Studies such as this provide valuable insights that were outside the scope of this SLR.

COMPARATIVE PERSPECTIVE AND FUTURE DIRECTIONS:

- Our findings **align with an existing body of evidence on cardiovascular and metabolic outcomes in CAH patients:**
 - A 2018 meta-analysis by Tamhane et al. reported increased blood pressure, insulin resistance, and CIMT in CAH patients compared to controls, consistent with the results presented here.[3]
 - A longitudinal study by Torky et al. provides insights into the onset and progression of CV/M comorbidities from childhood to adulthood, finding that obesity, hypertension, fasting hyperglycaemia, and hypertriglyceridemia often began prior to age 10.[9] Torky et al. also noted associations between metabolic outcomes and treatment factors, such as mineralocorticoid dose and suppressed androstenedione levels.[9]
- In this brief evidence synthesis, **the interplay between CAH treatment, hormonal imbalances, and metabolic outcomes was highlighted** and we call for continued long-term prospective longitudinal research evaluating this.
- **Long-term longitudinal prospective studies would be highly valuable for inclusion in a later, more comprehensive evidence synthesis** to better elucidate the temporal relationships between treatment approaches and CV/M outcomes.

CH EU-CH-0001

CONCLUSIONS

The evidence presented here, whilst limited, **adds to an existing body of evidence on an elevated CV/M risk profile in CAH patients, including higher prevalence of traditional risk factors** and elevated surrogate markers, adiposity indices, and other risk scores.

The observed association between GC dosing (with doses above 15-20 mg/day Hce, and study doses ranging up to 40 mg/day) and CV/M risk factors highlights the need to appropriately consider GC treatment regimens, balancing effective disease control with minimisation of potential CV/M complications; this is in line with the widely recognised Speiser 2018 Endocrine Society guidelines.[10]

The broad spectrum of CV/M issues identified reinforces the importance of a multidisciplinary approach to CAH management, including early and ongoing CV/M monitoring; which is also well supported by the Speiser 2018 Endocrine Society guidelines.[10]

CONTACT INFORMATION

Emmanuel Lacharme, PharmD
Associate Director, Market Access, Europe
Neurocrine Biosciences, Hegenheimermattweg 167A, 4123 Allschwil, Basel, Switzerland
ELacharme@neurocrine.com, +33680416874

REFERENCES

1. El-Maouche D, et al. Lancet. 2017;390:2194-210.
2. Mayo Clinic [Internet]. Congenital adrenal hyperplasia; 2024 [cited 2024 Mar 20].
3. Tamhane S, et al. J Clin Endocrinol Metab. 2018;103:4097-103.
4. Charoensri S, Auchus RJ. J Clin Endocrinol Metab. 2024;109:e1133-42.
5. Chen LM, et al. J Clin Endocrinol Metab. 2024;109:362-70.
6. Espinosa Reyes TM, et al. BMC Endocr Disord. 2023;23:241.
7. Kara Z, et al. Metab Syndr Relat Disord. 2023;21:35-40.
8. Righi B, et al. Endocrine. 2023;80:630-8.
9. Torky A, et al. J Clin Endocrinol Metab. 2021;106:e2046-57.
10. Speiser PW, et al. J Clin Endocrinol Metab. 2018;95:4133-60.

Iconography made by Freepik, zero_wing from www.flaticon.com