Poster EE230 presented at ISPOR Europe 2024
17–20 November
Barcelona

Budget Impact Analysis of Aztreonam-Avibactam (ATM-AVI) for the Treatment of Infections Caused by Confirmed and Suspected Metallo-β-Lactamase (MBL)-producing Enterobacterales in Italy

EE230

Falcone M[1], Bao X[2], Woodcock F[2], Di Virgilio R[3], Vidal Pereira MA[3], Kantecki M[4], Gheorghe M[5]

1. University of Pisa, Italy 2. Source HE, London, United Kingdom 3. Pfizer Italy, Rome, Italy 4. Pfizer International Operations, Paris, France 5. Pfizer Inc., Bucharest, Romania

Background

- Antimicrobial resistance is one of the major threats to human health around the world, with a substantial public health burden and global economic damage.[1] **MBLs are a type of carbapenemase produced by bacteria that confer resistance to a wide range of antibiotics**.[2-4]
- **ATM-AVI (EMBLAVEO®) is approved in Europe in adult patients for the treatment of complicated intra-abdominal infection (cIAI); hospital-acquired pneumonia (HAP), including ventilator-associated pneumonia (VAP); complicated urinary tract infection (cUTI), including pyelonephritis and for infections due to aerobic Gram-negative organisms with limited treatment options (LTO).**

Objective

- **This analysis evaluates the budget impact of introducing ATM-AVI ± metronidazole (MTZ) for the treatment of infections caused by confirmed and suspected MBL-producing Enterobacterales (MBL-CREs) including cIAI, HAP/VAP, cUTI and those with LTO in the Italian setting.**
- Data from REVISIT were used to cover the indications cIAI and HAP/VAP and the combined results for cIAI and HAP/VAP from the REVISIT trial were extrapolated for the indications cUTI and infections with LTO to cover all the label indications.[5,6]

Methods

- **The model compared two scenarios (world "with" and world "without" ATM-AVI ± MTZ)** to assess the budget impact of introducing ATM-AVI ± MTZ for the treatment of confirmed and suspected MBL-CRE infections over a 3-year time horizon.
- A Phase 3 randomised clinical trial (REVISIT) investigated ATM-AVI ± MTZ for the treatment of cIAI and HAP/VAP.[5, 6]
- The number of patients receiving each treatment is based on the estimated market share of each treatment and total eligible patients.[7, 8]
- **The model accounts for the burden of antimicrobial resistance to the treated individual**. Compared to susceptible infections, resistant infections were associated with higher length of stay (LoS) and treatment costs, higher probability of death and higher probability of infection recurrence.[9]
- Subsequent treatments are not modelled explicitly but are assumed to be captured in the extended hospital days. **The net budget impact is calculated as the difference in total cost between the worlds "with" and "without" ATM-AVI ± MTZ.**

Results

- In the world "with ATM-AVI", there were:
 - **780 additional cures**
 - **281 fewer deaths**
 - **4,056 fewer bed days.**
- Overall, the budget impact of infections suspected or documented to be caused by MBL-CRE including cIAI, HAP/VAP, cUTI and infections with LTO is **€6,837,527 over 3 years**, equivalent to **€356 per patient**.
- Deterministic sensitivity analysis indicates that results are sensitive to probability of cure, proportion of MBL-CRE, incidence of nephrotoxicity (NTX), hospitalisation cost, and LoS in intensive care unit (ICU).

Figure 1: BIA model structure

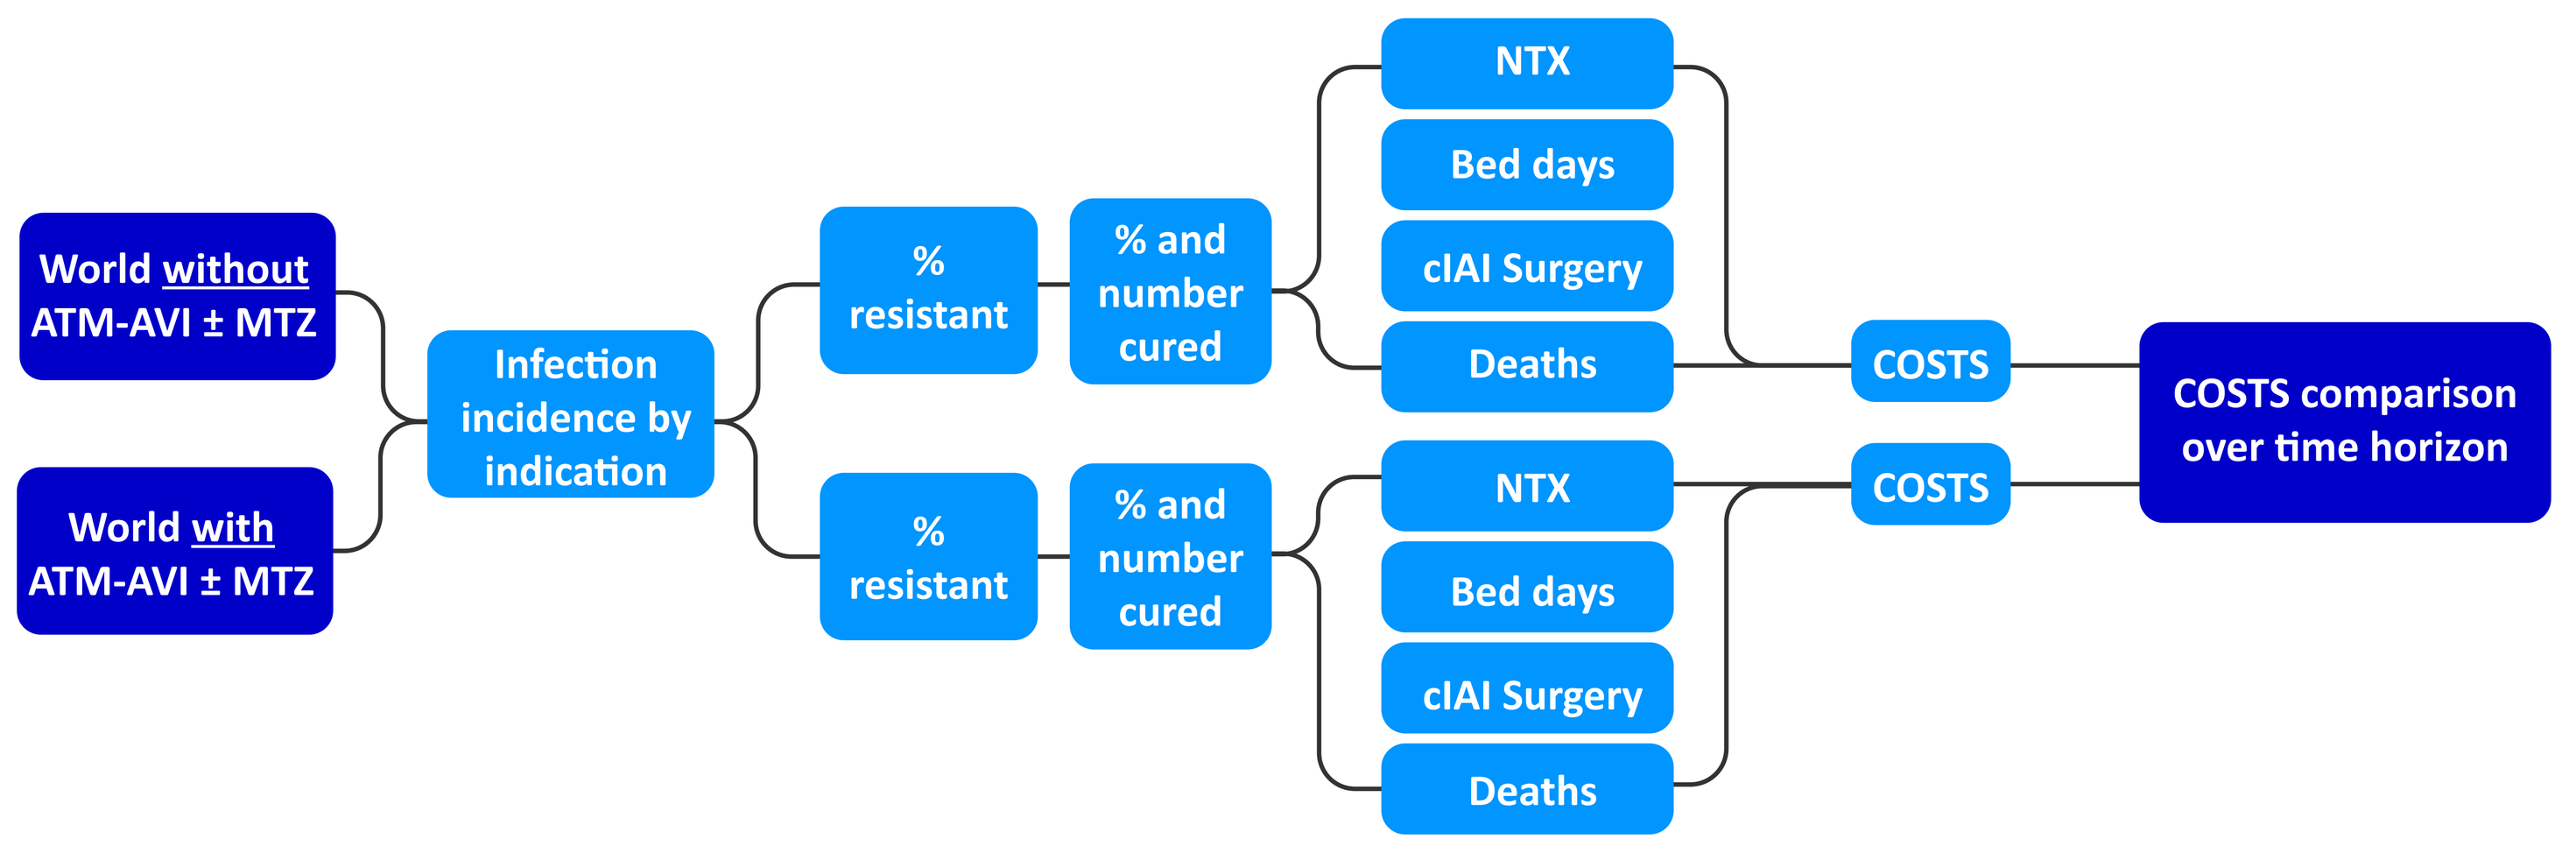

World without ATM-AVI ± MTZ
World with ATM-AVI ± MTZ
Infection incidence by indication
% resistant
% and number cured
NTX
Bed days
cIAI Surgery
Deaths
% resistant
% and number cured
NTX
Bed days
cIAI Surgery
Deaths
COSTS
COSTS
COSTS comparison over time horizon

Abbreviations: ATM-AVI, aztreonam-avibactam; BIA, budget impact analysis; cIAI, complicated intra-abdominal infections; MTZ, metronidazole; NTX, nephrotoxicity.

Table 1: Number of patients with MBL-CREs

Indication	Year 1	Year 2	Year 3
cIAI	88	91	94
cUTI	1,430	1,480	1,531
HAP/VAP	825	854	884
LTO	443	458	474
Total targeted	2,786	2,883	2,984
Total number of patients targeted & suspected (1 targeted + 1.2 suspected)	6,191	6,407	6,632

Abbreviations: cIAI, complicated intra-abdominal infections; CRE, carbapenem-resistant Enterobacterales; cUTI, complicated urinary tract infections; HAP, hospital-acquired pneumonia; LTO, limited or no treatment options; MBL, metallo-β-lactamase; VAP, ventilator-associated pneumonia.

Table 2: Market share

Treatment	World without ATM-AVI ± MTZ	World with ATM-AVI ± MTZ Year 1	World with ATM-AVI ± MTZ Year 2	World with ATM-AVI ± MTZ Year 3
ATM-AVI ± MTZ	0.00%	6.27	10.98%	15.09%
COL + MER	70.00%	66.73%	64.02%	62.91%
Cefiderocol	30.00%	27.00%	25.00%	22.00%

Abbreviations: ATM-AVI, aztreonam-avibactam; COL + MER, colistin + meropenem; MTZ, metronidazole.

Table 3: Treatment costs

	Cost per vial	Vial size	Info on dosing	Average Daily Dose	Daily cost
ATM-AVI	€200.41	1.5 g ATM 0.5 g AVI	- Doses are infused over 3 hours, q6h, requiring loading on Day 1 of the treatment cycle. - Total dose on Day 1 is 6.5 g of ATM and 2.17 g of AVI. On subsequent days the total dose is 6.0 g of ATM and 2.0 mg of AVI.[5]	6 g ATM 0.5 g AVI	€801.64
MTZ	€0.13	500 mg	500 mg MTZ IV infusion q8h[5, 10]	1,500 mg	€0.39
Meropenem	€18.44	1,000 mg	1,000 mg meropenem by 180 min IV infusion q8h[5, 11]	3,000 mg	€55.32
Colistin	€11.65	1 million IU	18 million IU on Day 1; 9 million IU daily on subsequent days[5, 12]	9million IU	€104.85
Cefiderocol	€135.38	1,000 mg	2 g q8h[13]	6,000 mg	€812.28

Abbreviations: ATM-AVI, aztreonam-avibactam; hr, hour; IU, international units; IV, intravenous; MTZ, metronidazole; q6h, every 6 hours; q8h, every 8 hours.

Table 4: Treatment duration (days)

Treatment	cIAI	cUTI	HAP/VAP	With LTO	Source
ATM-AVI	7.80	7.80	10.40	10.40	EMBLAVEO® SmPC[14]
MTZ	7.50	7.50	-	-	
COL + MER	8.50	8.50	10.30	10.30	
Cefiderocol	7.50	7.50	10.50	10.50	FETCROJA® SmPC[13]

Abbreviations: ATM-AVI, aztreonam-avibactam; cIAI, complicated intra-abdominal infections; COL + MER, colistin + meropenem; CSR, clinical study report; cUTI, complicated urinary tract infections; HAP, hospital-acquired pneumonia; MTZ, metronidazole; SmPC, summary of product characteristics; VAP, ventilator-associated pneumonia.

Table 5: Resource use inputs

	Value	Source
ICU cost per day	€1,590.17	Tan et al. 2012[15]
General ward cost per day	€867.83	Ministero dell'Economia e delle Finanze 2007[16]
Non-RRT additional LoS (days)	2.3	Simon et al. 2019[17]
RRT additional LoS (days)	5.2	
Number of in-hospital dialysis sessions	2.6	Assumes dialysis every other day per duration of additional LoS
Cost per instance of in-hospital dialysis	€147.25	Tariffe assistenza specialistica ambulatoriale[18]
Annual cost of long-term dialysis	€26,518	Roggeri et al. 2017[19]
Length of receiving EoL care (weeks)	1	Assumption
% time in ICU	50%	Assumption

Abbreviations: AKI, acute kidney injury; BSC, best supportive care; EoL, end of life; ICU intensive care unit; LoS, length of stay; RRT, renal replacement therapy.

Table 6: Results

	World without ATM-AVI ± MTZ	World with ATM-AVI ± MTZ	Difference	% change
Number of cured patients				
cIAI	330	364	35	10.5%
cUTI	4,378	4,835	457	10.4%
HAP/VAP	1,826	1,974	147	8.1%
With LTO	1,357	1,498	141	10.4%
Across all indications	7,891	8,671	780	9.9%
Number of deaths				
cIAI	68	61	-7	-10.4%
cUTI	1,479	1,345	-134	-9.1%
HAP/VAP	1,152	1,054	-98	-8.5%
With LTO	458	417	-42	-9.1%
Across all indications	3,158	2,876	-281	-8.9%
Number of hospital bed days				
cIAI	6,935	6,799	-136	-2.0%
cUTI	140,844	138,977	-1,867	-1.3%
HAP/VAP	133,861	132,386	-1,474	-1.1%
With LTO	43,639	43,061	-578	-1.3%
Across all indications	325,279	321,223	-4,056	-1.2%
Total budget impact				
cIAI	**€10,056,211**	**€10,170,563**	**€114,352**	**1.1%**
cUTI	**€196,062,828**	**€198,689,933**	**€2,627,105**	**1.3%**
HAP/VAP	**€201,354,091**	**€203,817,344**	**€2,463,253**	**1.2%**
With LTO	**€62,629,650**	**€64,262,466**	**€1,632,816**	**2.6%**
Across all indications	**€470,102,779**	**€476,940,307**	**€6,837,527**	**1.5%**
Budget impact per patient				
cIAI	**€16,544**	**€16,732**	**€188**	**1.1%**
cUTI	**€19,868**	**€20,135**	**€266**	**1.3%**
HAP/VAP	**€35,347**	**€35,779**	**€432**	**1.2%**
With LTO	**€20,484**	**€21,018**	**€534**	**2.6%**
Across all indications	**€24,446**	**€24,802**	**€356**	**1.5%**

Abbreviations: ATM-AVI, aztreonam-avibactam; cIAI, complicated intra-abdominal infections; cUTI, complicated urinary tract infections; HAP, hospital-acquired pneumonia; LTO, limited or no treatment options; MTZ, metronidazole; VAP, ventilator-associated pneumonia.

Conclusion

- This study is **the first budget impact analysis** of ATM-AVI ± MTZ.
- The **probability of cure, proportion of MBL-CRE, incidence of NTX, hospitalisation cost, and LoS in ICU are key drivers of model outcomes**.
- A key strength of this analysis is that it gives an estimate of the impact of introducing ATM-AVI ± MTZ on the hospital formulary, using inputs and assumptions that have been validated with leading clinicians.
- A key limitation of the study is that results in the model are driven by resistance, however resistance rates are constantly evolving.
- **This analysis includes the value of ATM-AVI considering the impact of resistant infections to the treated individuals.** However, no considerations on the broader population value or the value from a public health perspective were included in the model. For example, **adding a novel Gram-negative antibacterial agent such as ATM-AVI to the treatment landscape provides more treatment diversity and may reduce the transmission of resistant pathogens and prevent infections in the wider population**.
- **These results show that, in the Italian setting, while the introduction of ATM-AVI ± MTZ is associated with higher drug acquisition costs, there is also an increase in the number of cured patients and a decrease in mortality and hospital LoS. This leads to cost savings, which largely offset the increased drug costs**.

References: 1.Antimicrobial Resistance Collaborators. Lancet. 2022;399(10325):629-55. 2.Zhu et al. Int J Antimicrob Agents. 2021;58(5):106430. 3.Tacconelli et al. Lancet Infect Dis. 2018;18(3):318-27. 4. Control ECfDPa. 2018. Available at :https://www.ecdc.europa.eu/sites/default/files/documents/RRA-Enterobacteriaceae-Carbapenems-European-Union-countries.pdf.5. Pfizer. 2023. Data on file. REVIST clinical study report. 6.Carmeli et al. Infect Dis. 2023;10(Suppl 2). 7.Pfizer. Data on file. Clarivate epidemiology data. 8.Pfizer. Data on file. Market share data. 9.Poudel et al. PLoS One. 2023;18(5):e0285170. 10.Electronic Medicines Compendium (EMC). 2019. Available at: https://www.medicines.org.uk/emc/product/11952/smpc/print. 11.Quattrocolo et al. 2022. Available at: https://www.epicentro.iss.it/sorveglianza-ica/pdf/Protocollo%20studio%20pilota%20v1.01.pdf. 12.European Medicines Agency (EMA). Available at: https://www.medicines.org.uk/emc/product/1094/smpc/print. 13.European Medicines Agency (EMA). Available at: https://www.ema.europa.eu/en/documents/product-information/fetcroja-epar-product-information_en.pdf. 14.European Medicines Agency (EMA). Available at: https://ec.europa.eu/health/documents/community-register/2024/20240422162367/anx_162367_en.pdf. 15.Tan et al. Value Health. 2012;15(1):81-6. 16. Ministero dell'Economia e delle Finanze. 2007. Available at: https://www.anpo.net/wp-content/uploads/file-manager/OCSE/libro_verde_spesa_sanitaria2007.pdf#:~:text=Ministero%20dell%E2%80%99Economia%20e%20delle%20Finanze%20Commissione%20Tecnica%20per,prime%20indicazioni%20Doc.%202007%2F6%20Roma%2C%206%20settembre%202007 17.Simon et al. Antimicrob Agents Chemother. 2019;63(12). 18. Ministero Della Salute. https://www.salute.gov.it/imgs/C_17_pagineAree_1767_listaFile_itemName_0_file.pdf. 19.Roggeri et al. J Nephrol. 2017;30(2):263-9.