Anticipating the Real-World Impact of Therapeutic Innovations in France by Transporting Evidence from the United States - *A Case Study in Metastatic Triple-Negative Breast Cancer (mTNBC) based on the Flatiron Health Database*

MSR54

LB. Simomia Mbowen[1], V. Machuron[1], G. Machuron[1], R. Jegou[2], C. Esnault[1]

1. Roche SAS, Boulogne-Billancourt, France; 2. Keyrus Life Science, Nantes, France

Context & Objectives

Approval of innovative treatments is often earlier in the US than in France, making US data particularly valuable for predicting potential impacts in France and supporting decision-making, including for Health Technology Assessment. Taking the example of mTNBC, this study aims to:

1. Estimate the impact of access to innovative medicines on overall survival (OS) of mTNBC patients in the US (including immunotherapies and targeted therapies)
2. Evaluate the transportability of OS estimates from the US to France *before* access to innovative medicines in mTNBC (validation with French data)
3. Predict the impact of access to innovative medicines on OS in France by extrapolating US data in mTNBC

Methods

Populations (figure 1):

- **Flatiron US data** of female adult patients treated for mTNBC(*) (n = 2126) (1)
 - Pre_US: metastatic diagnosis between 01/07/13 and 31/12/2017 (n = 812)
 - Post_US: metastatic diagnosis between 01/04/19 and 30/09/2023 (n = 1052)
- **Aggregate French data** of adult female patients treated for metastatic TNBC from the national ESME mBC database (diagnosis < 2017) (2). OS IPD were reconstructed (3).

Statistical considerations (figure 2):

- **Adjustment methods:**
 1. 1:1 k-NN matching with replacement between Pre_US and Post_US
 2. MAIC between Pre_US and ESME data (4)
 3. MAIC between ESME and either Post_US (3a), or the Post_US_matched population (3b) using the same weights estimated in analysis 2 (**)
- **Variables used for all adjustments:**
 Age *(<50, 50-60, 60-70, >70)*, ECOG PS *(0, 1, 2+)*, De novo status (y/n), Sites of metastases (Lung *(y/n)*, Bone *(y/n)*, Nodes *(y/n)*), Number of metastatic sites at diagnosis *(1, 2+).*
 - Only for 2: 1L treatments use (Anthracyclines *(y/n)*, Taxanes *(y/n)*, Capecitabine *(y/n)*)
 - Missing values: single imputation randomly drawn from the distribution of missing data (FCS)
- **Overall Survival:** from 1L treatment initiation (index date). Censoring at extraction date.
- **Sensivity analyses (SA):**
 - for missing values: complete case analysis and missing values as a category
 - for the adjustment method: IPTW for 1
 - for the censoring rule in OS analyses: date of last known alive

Fig. 1

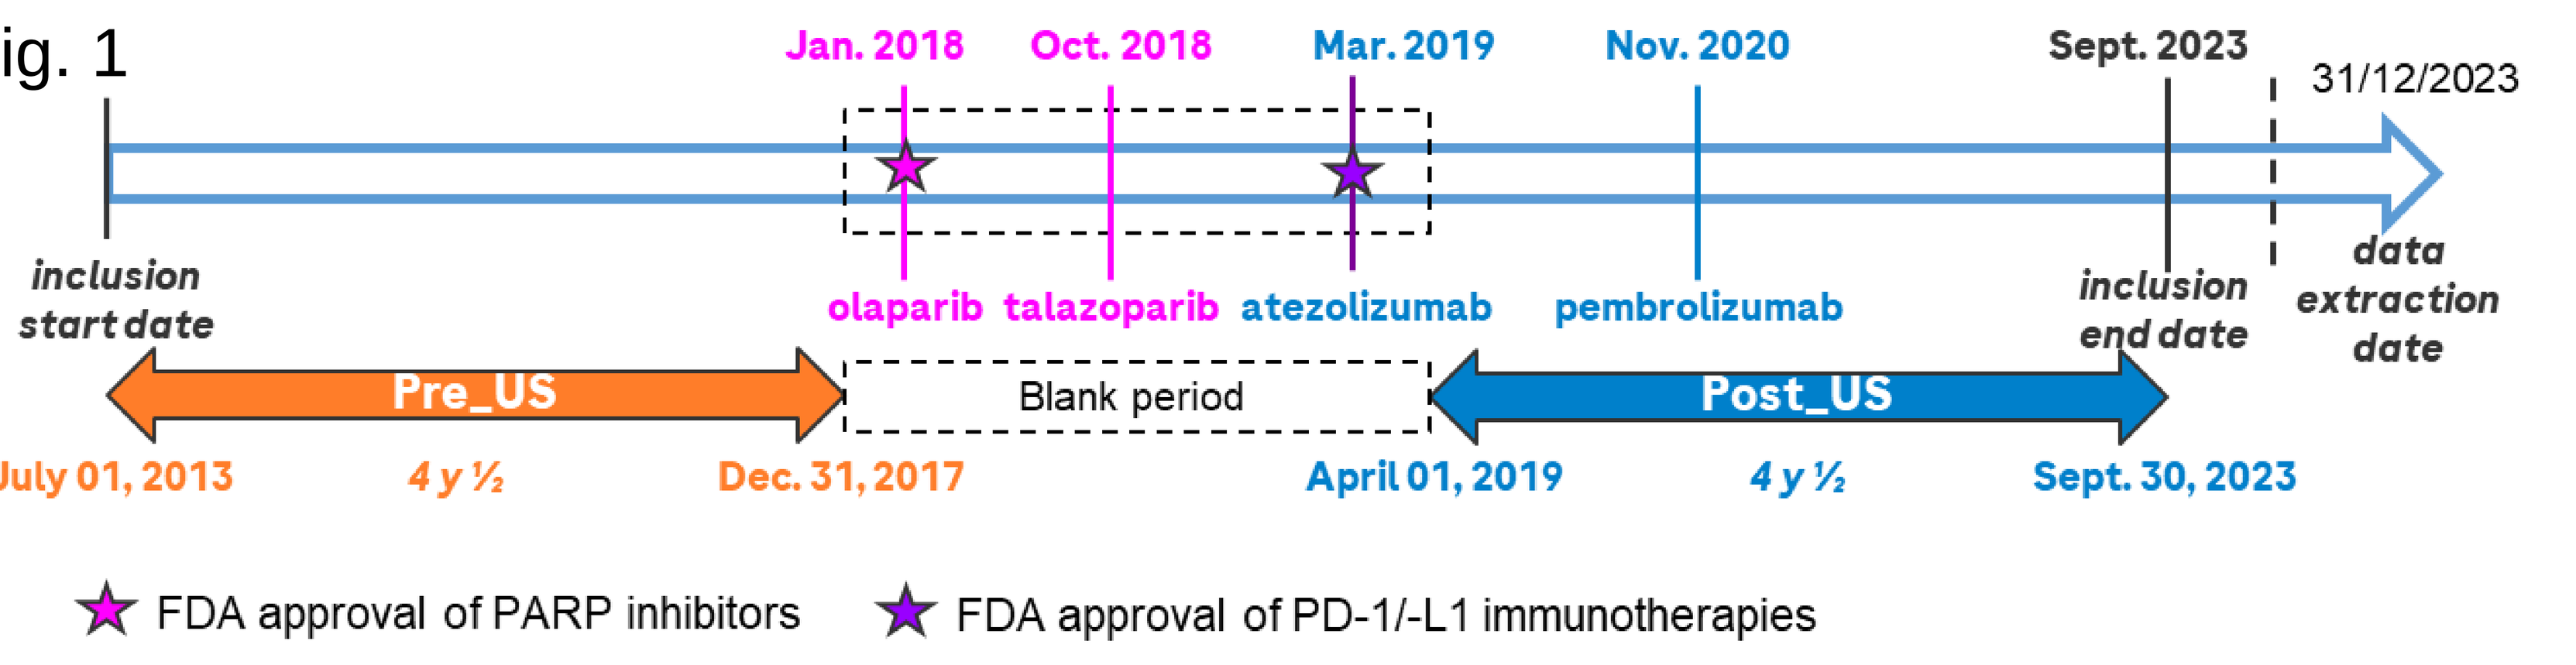
Jan. 2018
Oct. 2018
Mar. 2019
Nov. 2020
Sept. 2023
31/12/2023
inclusion start date
olaparib
talazoparib
atezolizumab
pembrolizumab
inclusion end date
data extraction date
Pre_US
Blank period
Post_US
July 01, 2013
4 y ½
Dec. 31, 2017
April 01, 2019
4 y ½
Sept. 30, 2023
FDA approval of PARP inhibitors
FDA approval of PD-1/-L1 immunotherapies

Fig. 2

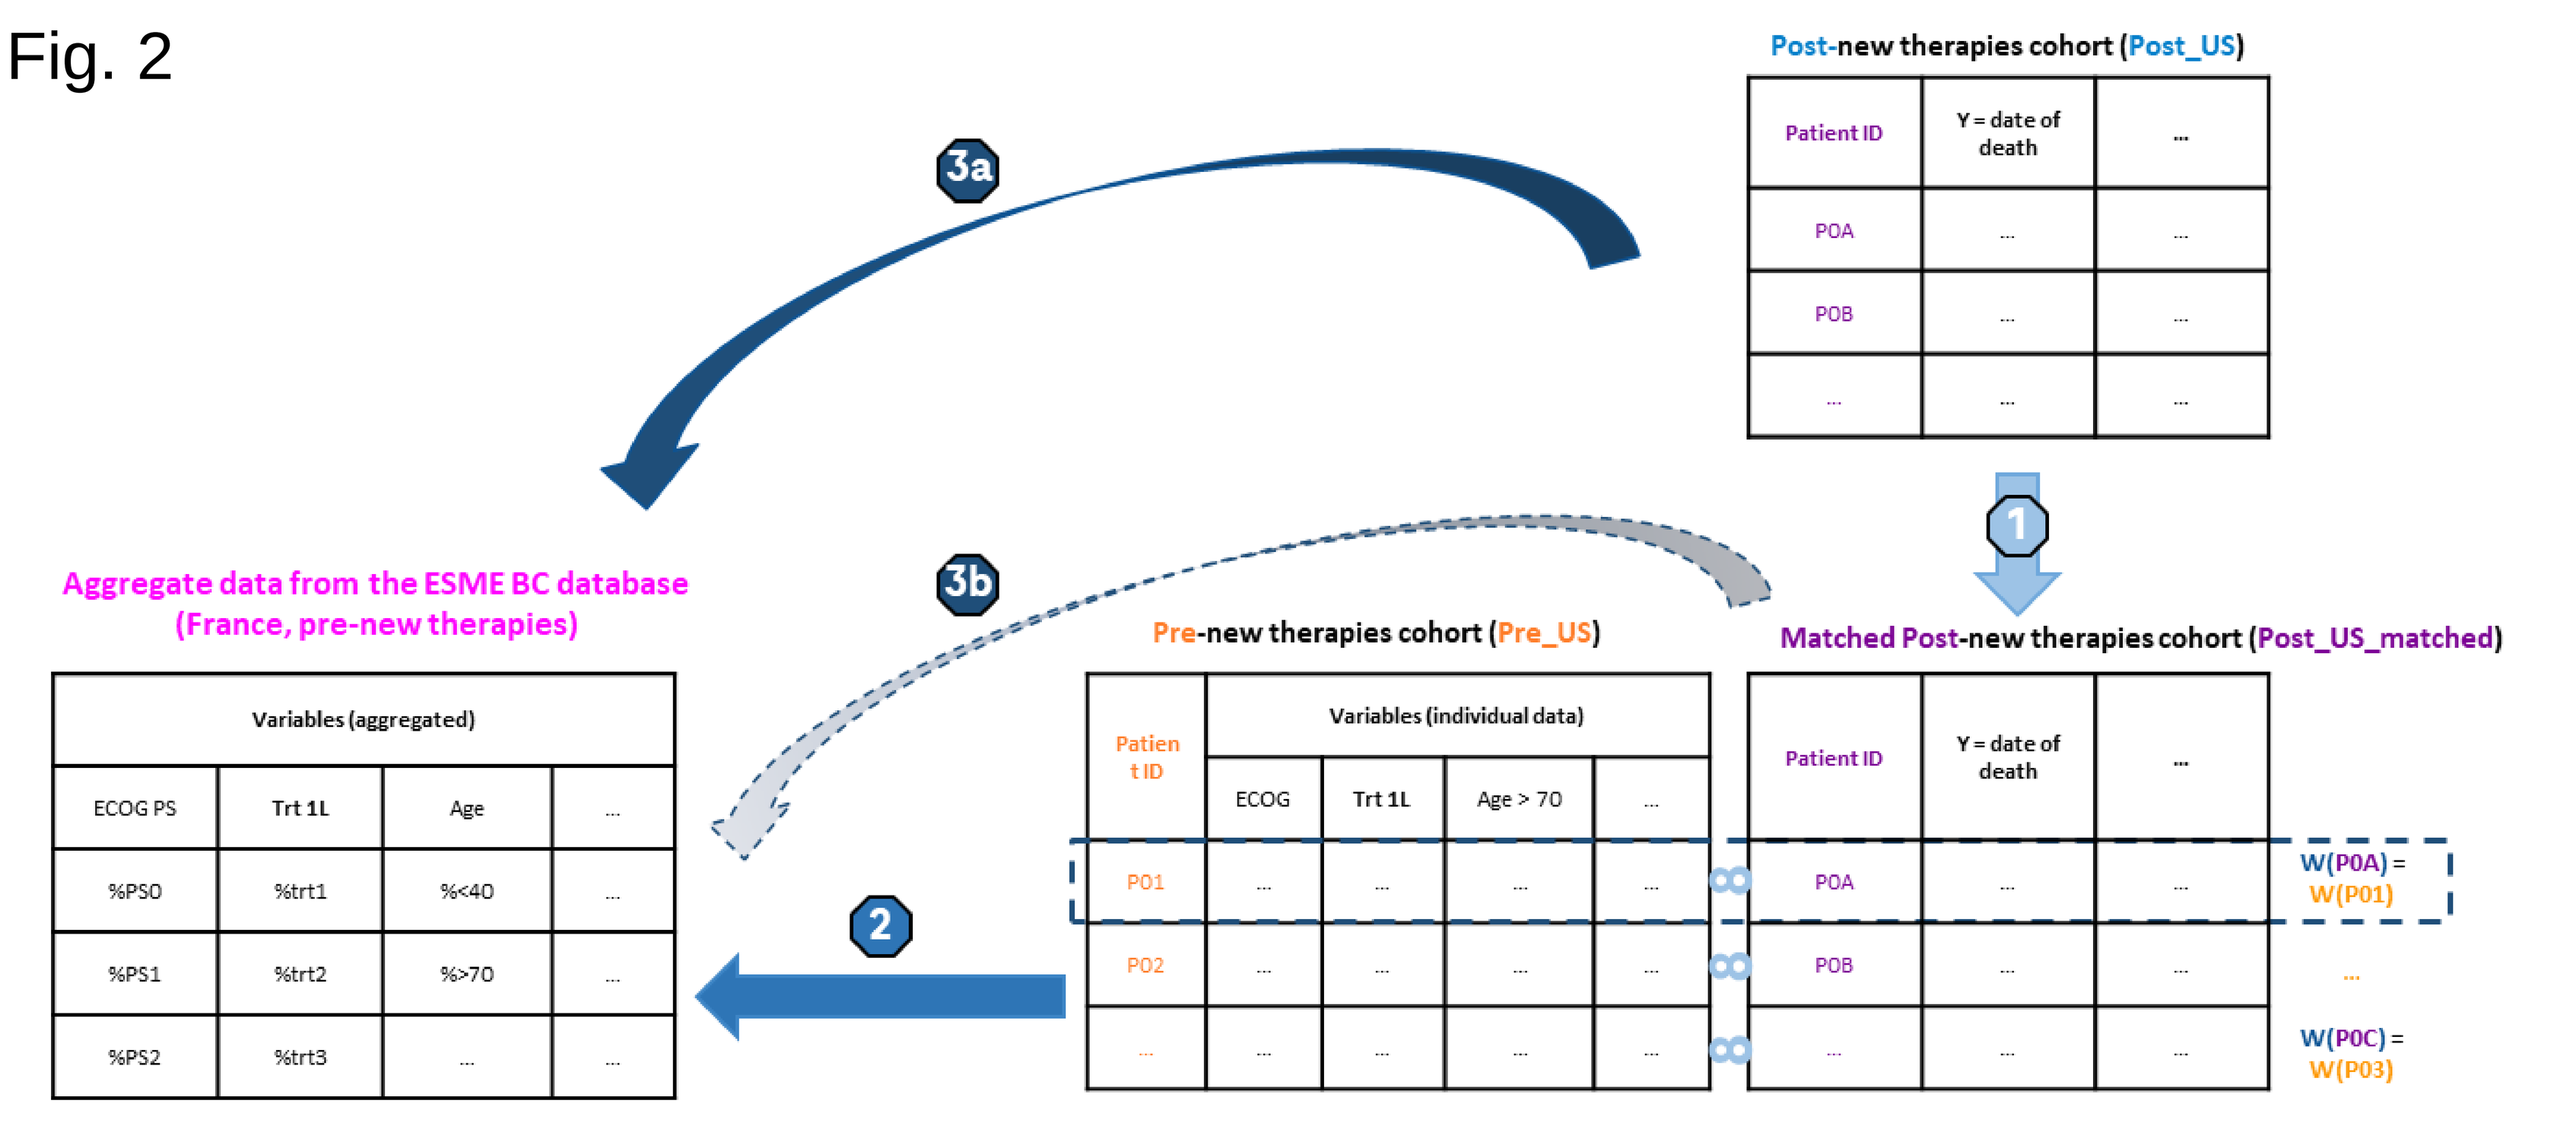
Post-new therapies cohort (Post_US)
Patient ID
Y = date of death
P0A
P0B
3a
3b
1
2
Aggregate data from the ESME BC database (France, pre-new therapies)
Variables (aggregated)
ECOG PS
Trt 1L
Age
%PS0
%trt1
%<40
%PS1
%trt2
%>70
%PS2
%trt3
Pre-new therapies cohort (Pre_US)
Patient ID
Variables (individual data)
ECOG
Trt 1L
Age > 70
P01
P02
Matched Post-new therapies cohort (Post_US_matched)
W(P0A) = W(P01)
W(P0C) = W(P03)

*defined as no positive and at least 1 negative test results for PR, ER, HER2 between +/-90 days around metastatic diagnosis. ** The use in analysis 3b of the same weights as in analysis 2 attempts to give more importance to 'Post' patients whose theoretical management before the arrival of the new therapies corresponds to that of French patients.

Results

First Line Therapy Class	Pre_US, n=812	Post_US, n=1052
Immuno/PD1/PDL1	2 (0.2%)	311 (30%)
Anthracyclines	124 (15%)	78 (7.4%)
Parp Inhibitor	4 (0.5%)	16 (1.5%)
Anti-Trop2	0 (0%)	43 (4.1%)
Capecitabine	163 (20%)	155 (15%)
Taxanes	291 (36%)	461 (44%)
Platines	205 (25%)	245 (23%)

Table of 1L treatment use per class of therapies before (Pre_US) and after (Post_US) the arrival of new therapies in the US (multiple choices). Blue boxes are new therapies (immunotherapies and targeted therapies)

OS	Post_US	Post_US_matched	Pre_US	ESME	Pre_Frenchlike	Post_Frenchlike
1-year rate	0.55 (0.52 ; 0.59)	0.60 (0.56 ; 0.63)	0.54 (0.51 ; 0.58)	0.55 (0.53 ; 0.57)	0.53 (0.49 ; 0.57)	0.54 (0.50 ; 0.58)
2-year rate	0.37 (0.34 ; 0.41)	0.39 (0.36 ; 0.43)	0.31 (0.28 ; 0.34)	0.30 (0.29 ; 0.32)	0.29 (0.25 ; 0.33)	0.37 (0.33 ; 0.41)
3-year rate	0.30 (0.27 ; 0.34)	0.32 (0.29 ; 0.36)	0.21 (0.19 ; 0.24)	0.18 (0.17 ; 0.20)	0.20 (0.17 ; 0.23)	0.29 (0.25 ; 0.33)
median	14.9 (13.1 ; 16.4)	16.0 (13.9 ; 18.6)	13.8 (12.6 ; 15)	13.8 (13.2 ; 14.6)	13.2 (11.6 ; 14.6)	13.8 (12.1 ; 16.2)

Table of OS medians and rate estimates at fixed time points (95% CI).

Fig. 3

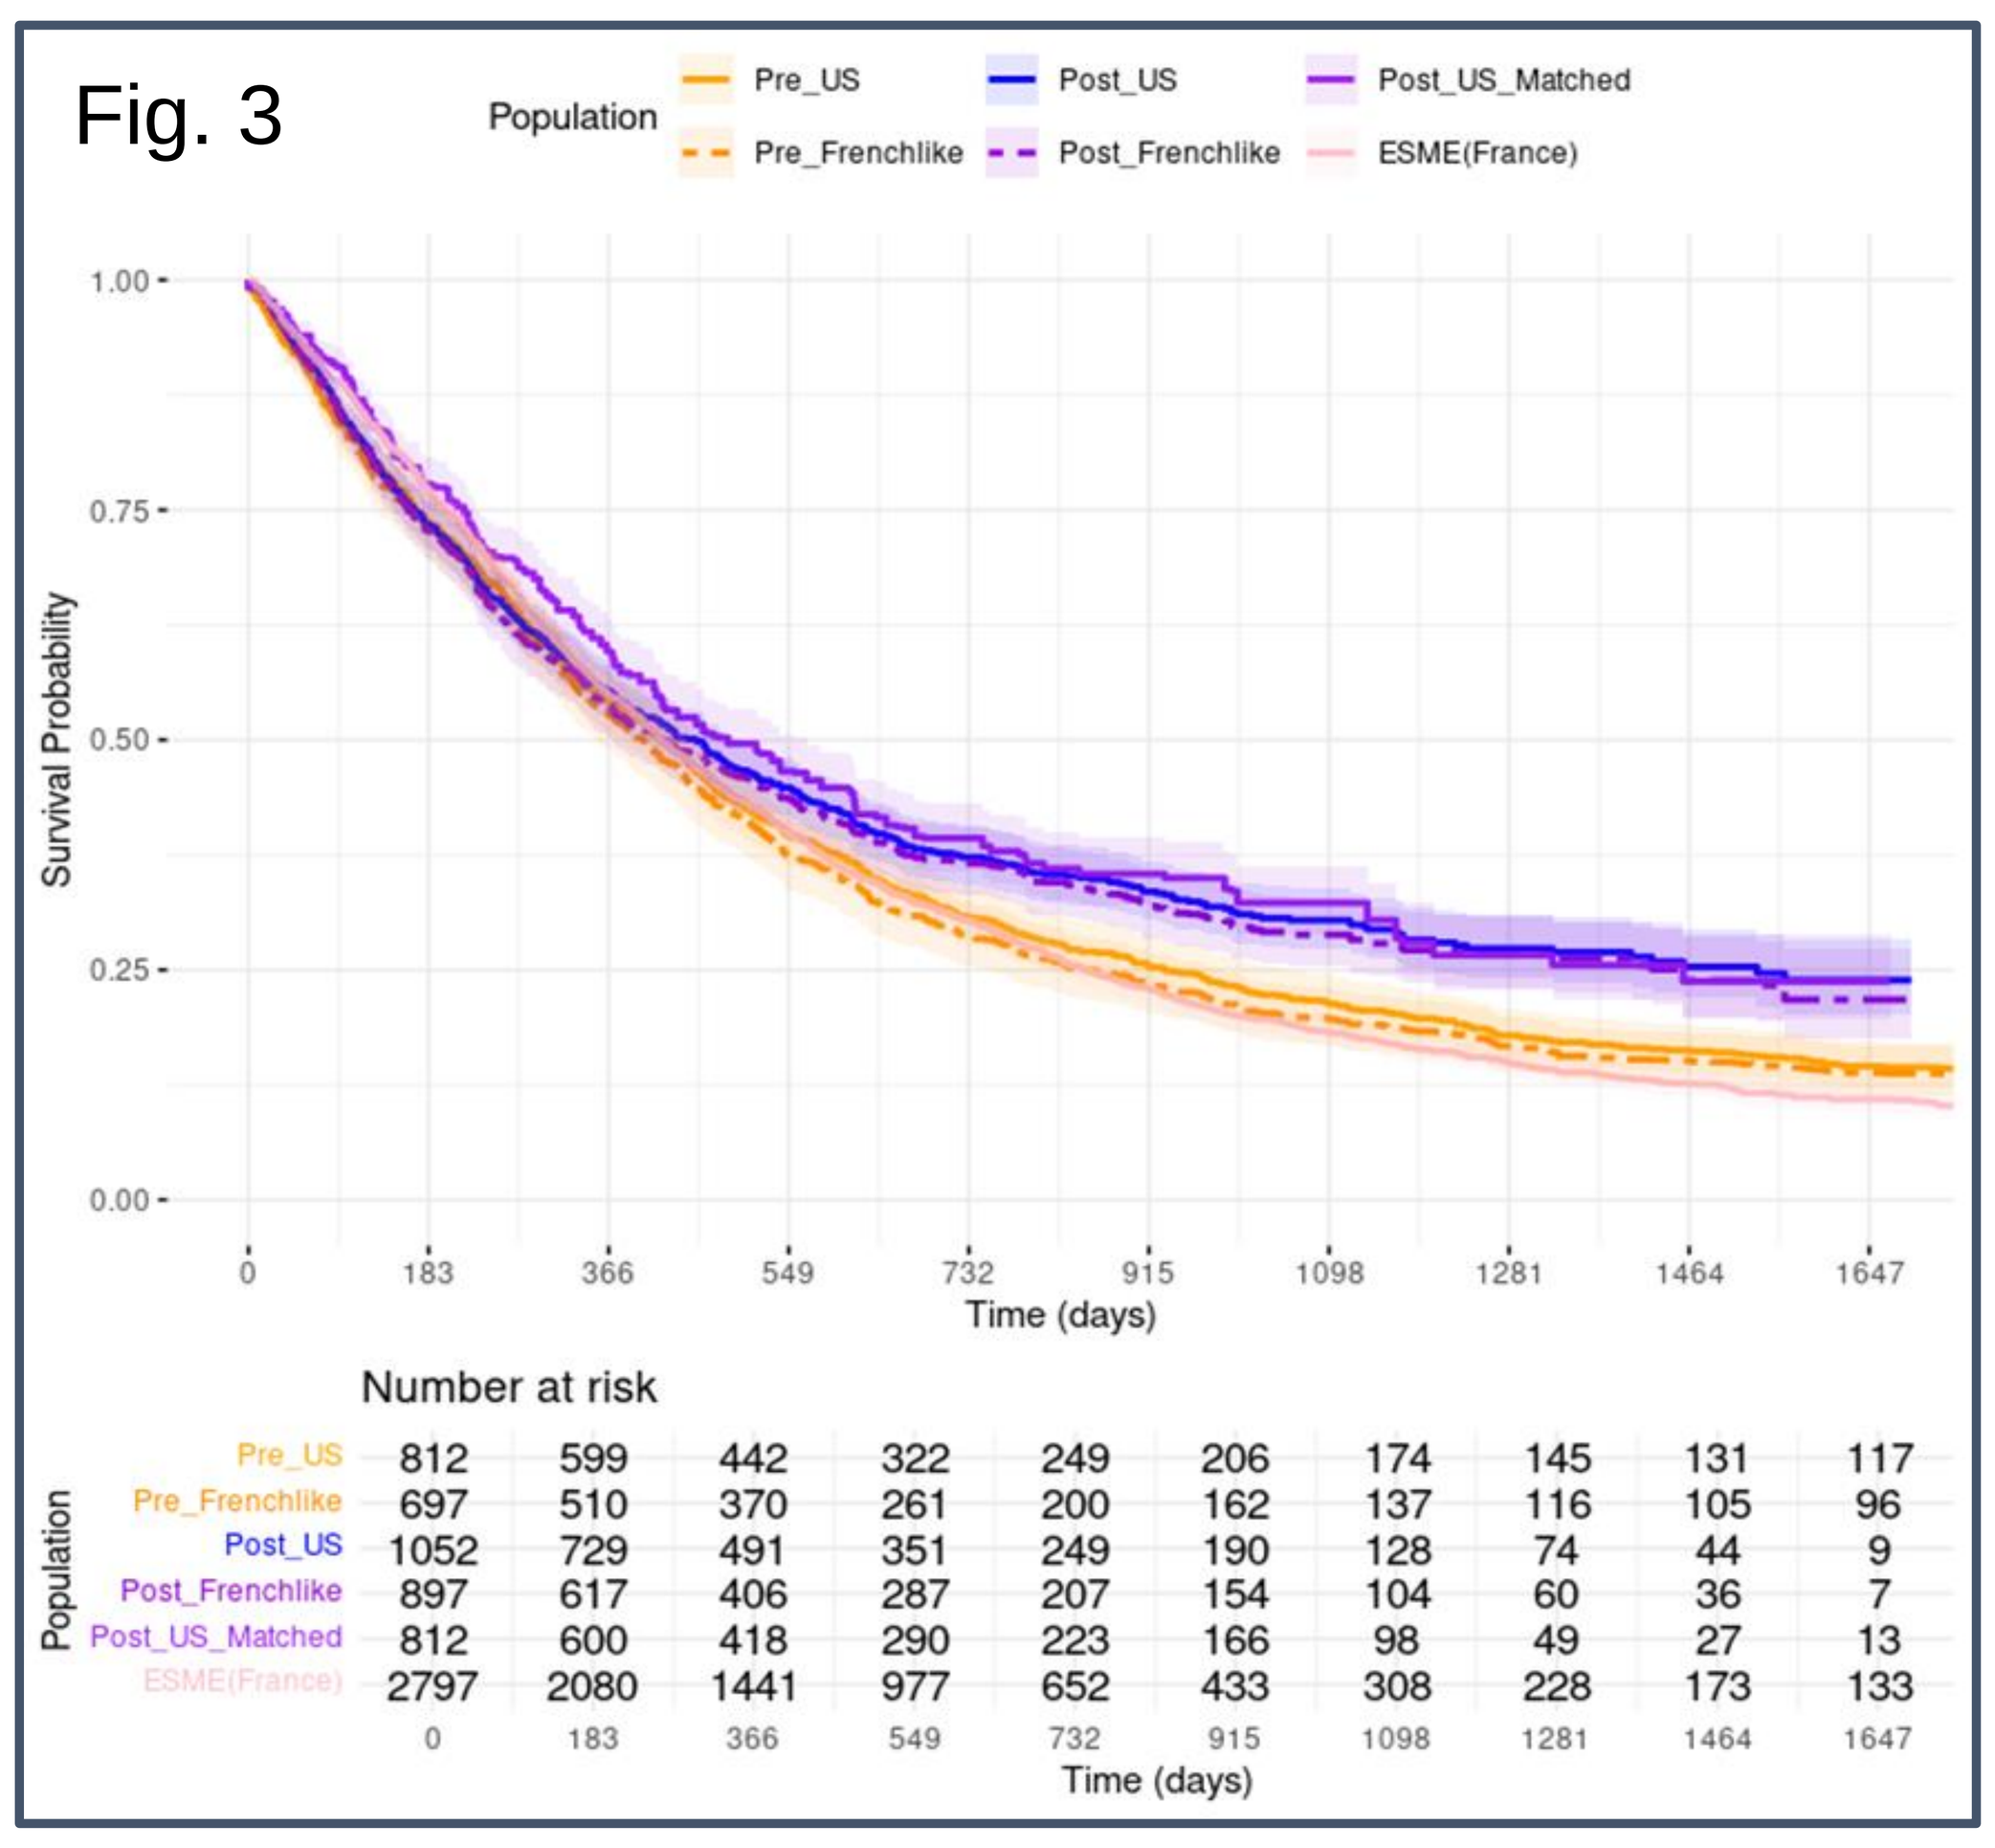
Population
Pre_US
Post_US
Post_US_Matched
Pre_Frenchlike
Post_Frenchlike
ESME(France)
Survival Probability
1.00
0.75
0.50
0.25
0.00
0 183 366 549 732 915 1098 1281 1464 1647
Time (days)
Number at risk
Population
Pre_US 812 599 442 322 249 206 174 145 131 117
Pre_Frenchlike 697 510 370 261 200 162 137 116 105 96
Post_US 1052 729 491 351 249 190 128 74 44 9
Post_Frenchlike 897 617 406 287 207 154 104 60 36 7
Post_US_Matched 812 600 418 290 223 166 98 49 27 13
ESME(France) 2797 2080 1441 977 652 433 308 228 173 133
0 183 366 549 732 915 1098 1281 1464 1647
Time (days)

Adjusted HR(*) = 0.79 (0.65; 0.95), **p < 0.05**
(Post_US_matched vs Pre_US)

- **Around 36% of mTNBC Post_US patients are treated with immuno- or targeted therapies.**
- **Predicted impact of new therapies in the US:**
 - **OS: -21% of hazard rate**
 - *SA: HRs ranged from 0.78 to 0.85*
 - **2- and 3-year OS rates: +8% and +11%**
 - *SA at 2 years: from +6% to +7%*
 - *SA at 3 years: from +8% to +10%*

1

Very good transportability of Overall Survival data from the US to France.

- ***Pre_US, Pre_Frenchlike and ESME OS curves overlap strongly***

2

Adjusted HR(*) = 0.84 (0.74; 0.95), **p < 0.05**
(Post_Frenchlike vs Pre_Frenchlike)

Predicted impact of new therapies in France:

- **OS: -16% of hazard rate** *(SA: HRs = 0.80 to 0.89)*
- **2- and 3-year OS rates: +8% and +9%**
- ***Analysis 3b: HR = 0.79** (0.69; 0.90) **(p < 0.05)***

3a

Post_US_matched: Post_US patients are matched to resemble the Pre_US patients (SMDs < 0.05)

Pre_Frenchlike: Pre_US patients are weighted to resemble the ESME (France) patients (SMDs ≃ 0)

Post_Frenchlike: Post_US patients are weighted to resemble the ESME (France) patients (SMDs ≃ 0)

(*) Hazard ratios are estimated from Cox models using robust standard error estimation and, specifically for analysis 1, the procedure described by Austin and Cafri (2020) for valid inference with matching by replacement (5).

Conclusion

2 **This study provides evidence that the US Flatiron OS estimates in 1L mTNBC are transportable to French patients.**

1 3 **The predicted overall impact of new therapies on OS in mTNBC in France should be similar to that in the US: -15 to -20% in the OS hazard rate.**

Limitations

- The estimated increase in OS in the US may be due to introduction of new treatments in second and further lines (and not only in 1L).
- The estimated increase in OS in the mTNBC may be underestimated due to the impact of COVID-19 from 2020.
- Other differences between the US and France may affect the transportability of OS estimates, such as access to others treatments, local care management, centers expertise, etc.

FCS: Fully conditional specification
HR: Hazard Ratio
IPD: Individual Patient Data
IPTW: Inverse Probability of Treatment Weighting
k-NN: k-nearest neighbors
mTNBC: metastatic Triple-Negative Breast Cancer
MAIC: Matching Adjusted Indirect Comparison
OS: Overall Survival
SA: Sensitivity Analyses
SMDs: standardized mean differences
US: United States of America

References

1. Flatiron Health EHR database (https://flatiron.com/real-world-evidence/)
2. UNICANCER. Epidemiological Strategy and Medical Economic (ESME) Research Program / Academic Real World Data Platform: Evolution of the Therapeutic Care in Metastatic Breast Cancer From 2008 (ESME-MBC). clinicaltrials.gov; Available on: https://clinicaltrials.gov/study/NCT03275311
3. Guyot, P., Ades, A., Ouwens, M.J. et al. Enhanced secondary analysis of survival data: reconstructing the data from published Kaplan-Meier survival curves. BMC Med Res Methodol 12, 9 (2012). https://doi.org/10.1186/1471-2288-12-9
4. Phillippo D, Ades T, Dias S, Palmer S, Abrams KR, Welton NJ. NICE DSU Technical Support Document 18: Methods for population-adjusted indirect comparisons in submissions to NICE. Published online December 7, 2016. https://research-information.bris.ac.uk/en/publications/nice-dsu-technical-support-document-18-methods-for-population-adj
5. Austin PC, Cafri G. Variance estimation when using propensity-score matching with replacement with survival or time-to-event outcomes. Stat Med. 2020 May 20;39(11):1623-1640. doi: 10.1002/sim.8502. Epub 2020 Feb 28. PMID: 32109319; PMCID: PMC7217182

Funding:
This study was supported by Roche SAS, as part of an M2 internship.