EE106
Lucie Kutikova,
LKutikova@Novavax.com

Cost-effectiveness of introducing Nuvaxovid to COVID-19 vaccination in the United Kingdom: Dynamic Transmission Model

Clive Pritchard[1], Lucie Kutikova[2]*, Richard Pitman[1], Kira Zhi Hua Lai[3], Hadi Beyhaghi[4], Ilana Gibbons[5], Amanda Erbe[6], Marija Živković-Gojović[7], Catherine Cosgrove[8], Mark Sculpher[9], David Salisbury[10]

[1]ICON Clinical Research, Reading, United Kingdom, [2]Novavax Europe, Zürich, Switzerland, [3]ICON Clinical Research, Toronto, Canada, [4]Novavax, Inc., Gaithersburg, MD, USA, [5]Novavax Europe, Reading, United Kingdom, [6]RTI Health Solutions, Research Triangle Park, NC, USA, [7]Formerly employed by ICON Clinical Research, Toronto, Canada, [8]St George's University of London, London, UK, [9]University of York, York, United Kingdom [10]Royal Institute of International Affairs, Chatham House, London, UK

BACKGROUND AND OBJECTIVES

- As of December 2023, approximately 24 million people had been infected by SARS-CoV-2 in the UK, leading to over 200,000 premature deaths.[1]
- Vaccination against SARS-CoV-2 remains a key preventive measure to control COVID-19. Nuvaxovid®, a recombinant Matrix-M®–adjuvanted protein, showed similar efficacy to mRNA vaccines in clinical trials and real-world studies, with lower rates of reactogenicity.[2,3,4,5]

To support decision-making on vaccine selection for the UK, a population-based compartmental **dynamic transmission model (DTM)** with a cost-utility component was developed to evaluate the cost-effectiveness of **Nuvaxovid** compared to **mRNA vaccines** in individuals aged 65 years or older and those aged 12-64 years with preconditions from a UK National Health Service perspective

METHODS

- A dynamic transmission approach was used to capture the indirect effects of vaccination on SARS-CoV-2 transmission and to calibrate multiple model parameters (**Figure 1**). The DTM was calibrated to official epidemiology statistics for mortality, incidence, and hospitalization.

Figure 1. Schematic representation of the model concept

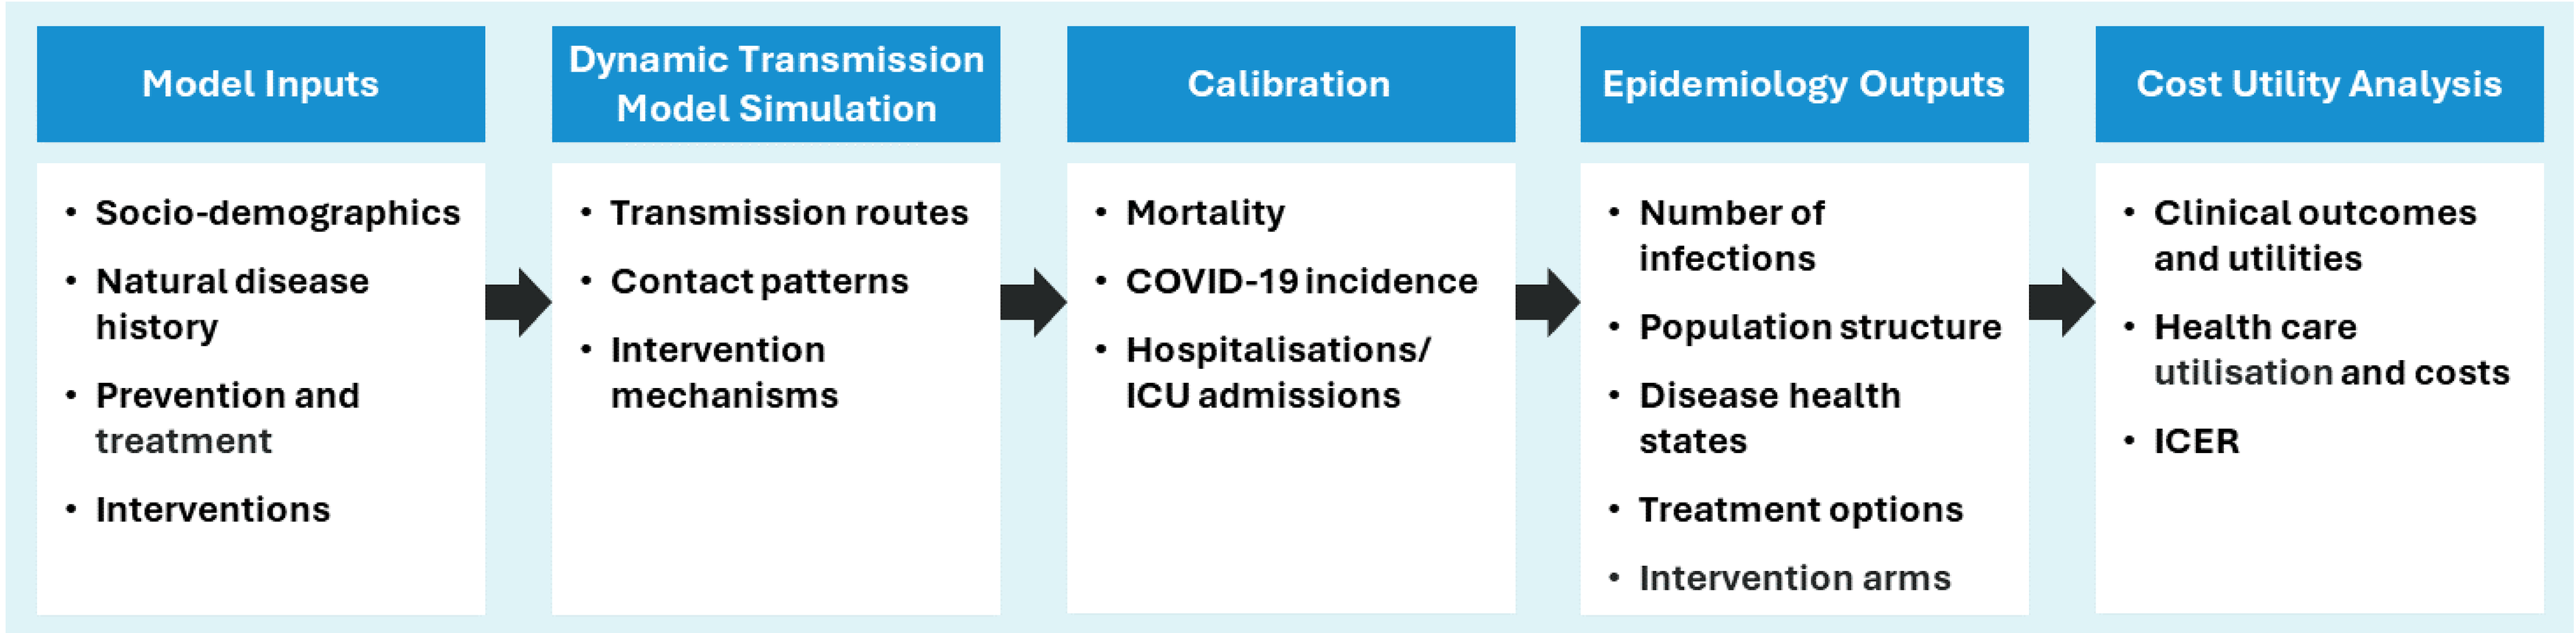
Model Inputs
Socio-demographics
Natural disease history
Prevention and treatment
Interventions
Dynamic Transmission Model Simulation
Transmission routes
Contact patterns
Intervention mechanisms
Calibration
Mortality
COVID-19 incidence
Hospitalisations/ ICU admissions
Epidemiology Outputs
Number of infections
Population structure
Disease health states
Treatment options
Intervention arms
Cost Utility Analysis
Clinical outcomes and utilities
Health care utilisation and costs
ICER

- The model's time horizon was 1 year, and vaccines were assumed to be administered once, at the beginning of the modelled 12-month period.
- The model's design and data inputs were informed by a targeted literature review. Clinical and utility model inputs are detailed in **Table 1** and cost and healthcare resource use model inputs are detailed in **Table 2.**
- Deterministic and probabilistic (10,000 iterations) sensitivity analyses were conducted, varying all key input parameters from **Tables 1** and **Table 2**. Scenario analyses were conducted assessing vaccine waning, Nuvaxovid market shares, and the vaccinated population.

Table 1. Clinical and utility inputs

Parameter	Base case
Vaccine uptake, efficacy, waning, and tolerability[a] parameters	
Vaccine uptake by age[6]	5.34% - 75.7%
Initial vaccine efficacy against all infections[7]	82%
Start of waning post vaccination[8]	1 month
Vaccine efficacy waning per month[8]	0.08
Efficacy against severe disease[9]	50%
Nuvaxovid average AEs per recipient[10]	1.572
mRNAs average AEs per recipient[10]	2.497
Utility decrements	
Symptomatic case[11]	0.008
Non-fatal hospitalisation[12]	0.0201
Non-fatal ICU[13]	0.15
Long COVID[14]	0.13
Nuvaxovid tolerability[a,b,10,15]	0.00022
mRNA tolerability[a,b,10,15]	0.00034

[a]Tolerability for Nuvaxovid and mRNA vaccines was based on a meta-analysis of reactogenicity events in clinical trials and was a key driver of QALY gains with a Nuvaxovid-only versus mRNA-only vaccination strategy. However, head-to-head clinical trials were not conducted to date; [b]vaccine-related adverse event (AE) was assumed to last 1 day.

Table 2. Cost and healthcare resource use inputs

Parameter	Base case value	
	Frequency	Cost per case
Vaccine-related costs		
Cost of vaccines[16]	–	£71.00
Vaccine administration costs[17]	–	£7.54
Cold chain transportation wastage[18]	Nuvaxovid: 0.02% mRNA: 0.10%	–
Freeze-related costs (thawing, handling, monitoring of thawed vials)[a,19,20]	–	Nuvaxovid: £0 mRNA: £0.14
Healthcare resource use costs		
General practitioner visit[13,21]	15.5%	£44.20
Accident and emergency (A&E) visits[13,22]	2.7%	£260.88
Hospitalization[22,23,24]	0.09% – 11.89%	£3,533.68
ICU hospitalization[6,22,23,25]	5.54% – 0.58%	£24,494.10 ($2,449.41/day, 10 days)
Post-hospitalization (ICU and non-ICU) care[14]	–	£413.95
Long COVID care[14]	10%	£2,515.46

[a]time spent multiplied by the average salary of the nurse and pharmacist

RESULTS

- In the probabilistic base case, a Nuvaxovid-only strategy provided total incremental cost savings of £1,388,323 and 1,558 additional QALYs compared to an mRNA-only strategy (**Table 3**). Cost savings were driven by the elimination of freeze-related operational costs, while QALY gains were driven by vaccine tolerability.

Table 3. Probabilistic results

	Nuvaxovid	mRNA vaccines	Incremental
Costs	£4,979,904,295	£4,981,292,618	−£1,388,323 (95% CI: −£225,070,810, £221,780,060)
QALY losses	−483,644	−485,201	1,558 QALYs (95% CI: −38,215 QALYs, 41,067 QALYs)
ICER			Dominant

- The Nuvaxovid only strategy had a 70% probability of being cost-effective at a £20,000/QALY threshold (**Figure 2**). Across most cost-effectiveness thresholds (up to £200,000/QALY), the acceptability curve remained relatively flat as the threshold was varied, because most simulations produce a dominant result.

Figure 2. Probabilistic sensitivity analysis results

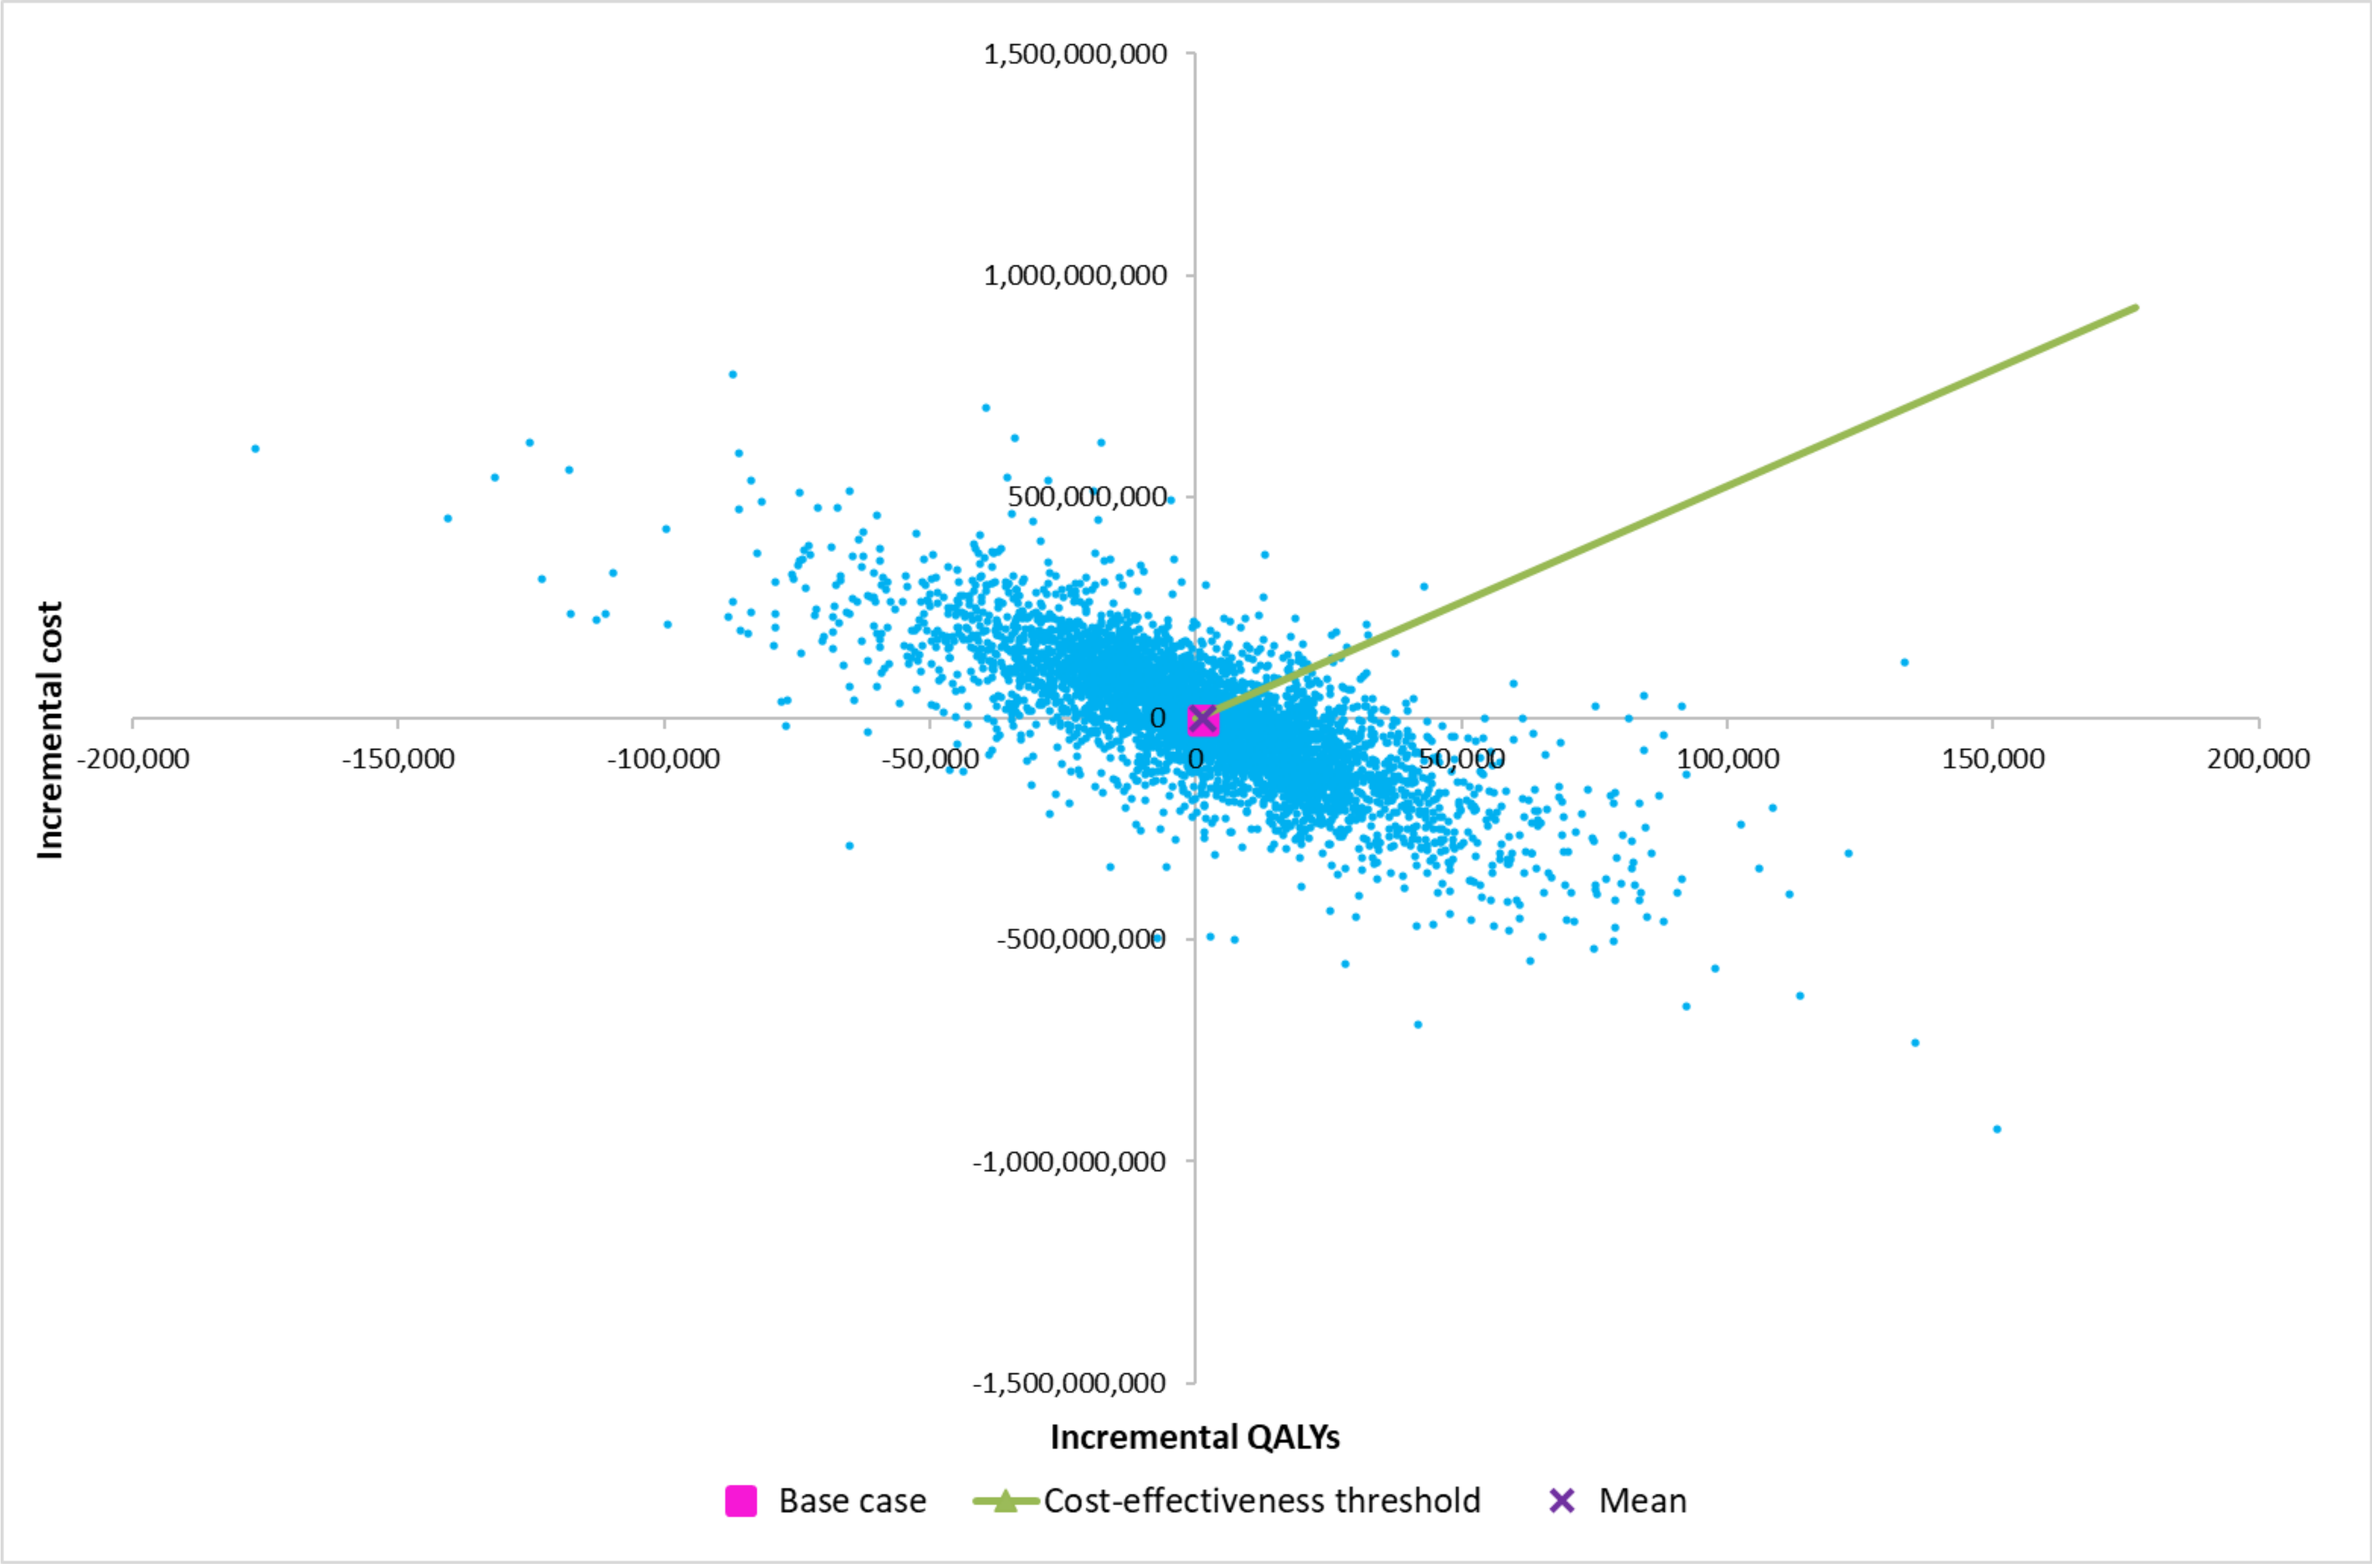
1,500,000,000
1,000,000,000
500,000,000
0
-500,000,000
-1,000,000,000
-1,500,000,000
Incremental cost
-200,000
-150,000
-100,000
-50,000
0
50,000
100,000
150,000
200,000
Incremental QALYs
Base case
Cost-effectiveness threshold
Mean

- ICERs generated in the deterministic sensitivity analyses were most sensitive to the mRNA and Nuvaxovid vaccine efficacy parameters, including the monthly vaccine efficacy waning rate, initial vaccine efficacy, efficacy against severe disease, and the start of waning post vaccination.
- Nuvaxovid remained dominant over mRNA vaccines in scenarios assessing vaccine waning, Nuvaxovid market shares, and the vaccinated population.
 - For the waning scenario, waning for Nuvaxovid started at 2 months after vaccination (vs. 1 month for mRNA), based on a real-world cohort study in Italy[26]. Compared to the base case, this scenario resulted in increased incremental cost savings and increased incremental QALY gains due to lower hospitalizations, ICU admissions, long COVID management and other associated COVID-19 events (**Table 4**).

Table 4. Onset of waning scenario analysis results

	Nuvaxovid	mRNA vaccines	Incremental
Costs	£4,830,427,740	£4,918,318,902	−£87,891,163 (95% CI: −£438,548,577, £56,404,242)
QALY losses	−464,813	−477,568	12,755 QALYs (95% CI: −12,324 QALYs, 68,979 QALYs)
ICER			Dominant

CONCLUSIONS

- To our knowledge, this is the first dynamic transmission model to estimate the impact of vaccine selection on the vaccination strategy in the UK from an NHS perspective.
- The evolving dynamics of the COVID-19 epidemic and shifting public health surveillance around COVID-19 testing leads to several limitations of this analysis, therefore the parameters selected in this model may not accurately reflect the current COVID-19 landscape.
- Our study's findings indicate that a vaccine strategy incorporating Nuvaxovid into the existing COVID-19 vaccination program in the UK may yield advantages over an mRNA-only vaccine strategy, driven mainly by suggested improvements in **tolerability and operational efficiency of Nuvaxovid**. Cost savings and QALY gains increased when examining a scenario with a 2-month onset of waning for Nuvaxovid.

REFERENCES

1. WHO. https://tinyurl.com/Dashboard-4-COVID-19 Accessed Apr 4, 2023
2. Alves, K., et al. *Vaccine* **2023** doi:10.1016/j.vaccine.2023.05.051
3. Heath, P.T., et al. *Clin Infect Dis* **2023** doi:10.1093/cid/ciac803
4. MHRA, **2024** https://tinyurl.com/Comirnaty-Omicron-XBB-1-5
5. Heath, P.T., et al. N Engl J Med 2021 doi:10.1056/NEJMoa2107659
6. UKHSA, **2024** https://tinyurl.com/COVID-Flu-Surveillance
7. El Sahly, H.M., et al. *N Engl J Med* **2021** doi:10.1056/NEJMoa2113017
8. Menegale, F., et al. *JAMA Netw Open* **2023** doi:10.1001/jamanetworkopen.2023.10650
9. ECDC, **2023** https://tinyurl.com/COVID-VE-30Nov23
10. Sutton, N., et al. *Expert Rev Vaccines* **2022** doi:10.1080/14760584.2022.2098719
11. Van Hoek, A.J., et al. *PloS One* **2011** doi:10.1371/journal.pone.0017030
12. Halpin, S.J., et al. *J Med Virol* **2021** doi:10.1002/jmv.26368
13. Sandmann, F.G., et al. *Lancet ID* **2021** doi:10.1016/s1473-3099(21)00079-7
14. Metry, A., et al. *Health Technol Assess* **2023** doi:10.3310/NAFW3527
15. Leung, M.K. & J.H. You. Vaccine 2016 doi:10.1016/j.vaccine.2016.04.008
16. NHSBSA dm+d, **2024** https://dmd-browser.nhsbsa.nhs.uk/
17. NHS, **2024** https://tinyurl.com/ya2fzjem
18. Hajibabai L, et al. *Transp Science* **2022** https://doi.org/10.1287/trsc.2022.1134.
19. Yarnoff, B., et al., *HV&I* **2021** doi:10.1080/21645515.2021.1974289
20. NHS Employers. https://www.nhsemployers.org/articles/pay-scales-202324
21. Jones, K.C.W., et al. **2022** https://kar.kent.ac.uk/100519/
22. NHS, **2023** https://tinyurl.com/2021-2022-costs
23. Knock, E.S., et al. *Sci Transl Med* **2021** doi:10.1126/scitranslmed.abg4262
24. UKHSA, **2024** https://tinyurl.com/COVID-19-archive
25. Davies, N.G., et al. *Lancet Public Health* **2020** doi:10.1016/s2468-2667(20)30133-x
26. Mateo-Urdiales, A., et al. *JAMA Netw Open* **2023** doi:10.1001/jamanetworkopen.2023.36854

Disclosures and Acknowledgments: This study was funded by Novavax Inc. and conducted by ICON plc.
C.P., RP., KZHL and MZG are employees of ICON Clinical Research. L.K., H.B. and I.G. are employees of Novavax, Inc., and may hold stock in Novavax, Inc; A.E. is an employee of RTI Health Solutions who receives funding from pharmaceutical, biotechnology, and medical devices companies to conduct health economics and outcomes research. CC has received payments and grants to her institution from GSK, Moderna and Novavax for studies and work performed. She has not received any personal financial rewards. MS is a paid consultant for Novavax. DMS has undertaken consultancies for GSK, Sanofi, Cansino, Clover, Pfizer and Novavax. Editorial support and poster layout were provided by Anar Murphy, PhD, of Novavax, Inc.