Will We Have Access to Healthcare? Rising Burden of Chronic Disease Hospitalisations: Evidence From Routine Hospital Data

(1) Chair of Health Economics, Policy and Management, School of Medicine, University of St.Gallen, (2) AstraZeneca AG, Medical Affairs

Daria Bukanova-Berend (1), Thomas Campbell-James (2), Justus Vogel (1)

Universität St.Gallen School of Medicine

AstraZeneca

HPR 210

BACKGROUND & RESEARCH QUESTION

Background:

Chronic diseases currently impose a substantial health and financial burden on the Swiss health system. With an ageing population affecting both the demand (patients) and supply (primary care physicians) of healthcare services, this burden is bound to increase. Identifying the factors that can prevent the continuous growth of potentially avoidable hospitalisations is essential.

Research question:

How did the number of potentially avoidable hospitalisations (PAHs) and associated resources develop (beds, expenditures) from 2012 to 2022 and how will they evolve until 2032?

Chronic diseases: Asthma, Chronic obstructive pulmonary disease (COPD), Congestive heart failure (CHF), Hypertension, Type 2 diabetes.

DATA

OECD: Health Quality and Outcomes indicators (2022-2023)

Federal Statistical Office (FSO): Routine Hospital Data

- Gender & Age group (5-year ranges),
- Area of residence,
- Length of stay in days,
- Principal and secondary diagnoses,
- Treatments (up to 99),
- Swiss DRG code

FSO: population 2012-2022

Reference tariffs for 26 cantons

Scenario analyses:

- **FSO:** population projection 2023-2032
- **Swiss Medical Association**: Primary care physicians (PCP) 2012-2022

METHODS

1. Identify and calculate **PAHs** per chronic disease on a national level
2. Convert inpatient days into hospital **bed numbers**
3. Multiply the effective DRG cost weight (CW) by cantonal hospital reference tariff to calculate **expenditures**
4. Project **PAHs*** based on:
 a. demographic change → Derive factor for each forecasting year from FSO population dataset considering age group, gender and canton; Extrapolate each case with the factors for each forecasting year
 b. PCP supply (*combined with 1*) → Method: Random forest ML model; Prediction: PAHs rate per 100K inhabitants per canton; Features: PCP full-time equivalents (FTEs) per 100K inhabitants per age group per canton, 26 canton dummies
 c. GDMT implementation (*CHF and COPD cases, combined with 1&2*) → Literature search and economic modelling

* 2019 as a base year

RESULTS

Descriptive statistics

Base year 2019

	Asthma	COPD	CHF	Hypertension	Type 2 Diabetes
Total PAHs	**1 608 (100)**	**10 664 (100)**	**17 948 (100)**	**3 654 (100)**	**6 332 (100)**
Female, N (%)	1 121 (69.7)	5 187 (48.6)	9 093 (50.7)	2 601 (71.2)	2 240 (35.4)
Swiss, N (%)	1 498 (93.2)	10 465 (98.1)	17 638 (98.3)	3 549 (97.1)	6 197 (97.9)
Age Group, N (%)					
20-49	508 (31.6)	310 (2.9)	180 (1.0)	245 (6.7)	981 (15.5)
50-59	286 (17.8)	1 262 (11.8)	415 (2.3)	382 (10.5)	1 017 (16.1)
60-69	223 (13.9)	2 667 (25.0)	1 359 (7.8)	539 (14.8)	1 316 (20.8)
70-79	270 (16.8)	3 925 (36.8)	4 289 (23.9)	956 (26.2)	1 590 (25.1)
80-89	261 (16.2)	2 175 (20.4)	8 349 (46.5)	1 215 (33.3)	1 189 (18.8)
90+	60 (3.7)	325 (3.0)	3 356 (18.7)	317 (8.7)	239 (3.8)
Comorbidities					
Mean (SD)	4.53 (4.12)	7.74 (4.67)	11.38 (4.12)	4.28 (3.57)	8.59 (6.00)
Median (IQR)	3 (2 - 6)	7 (4 - 10)	11 (8 - 14)	4 (2 - 6)	8 (4 - 12)

COPD, CHF, Hypertension and Type 2 diabetes prevailed in 60+ age group in 2019. Patients with PAHs due to CHF had ~11 comorbidities on average

PCP and corresponding full-time equivalents (FTE) per 100k inhabitants

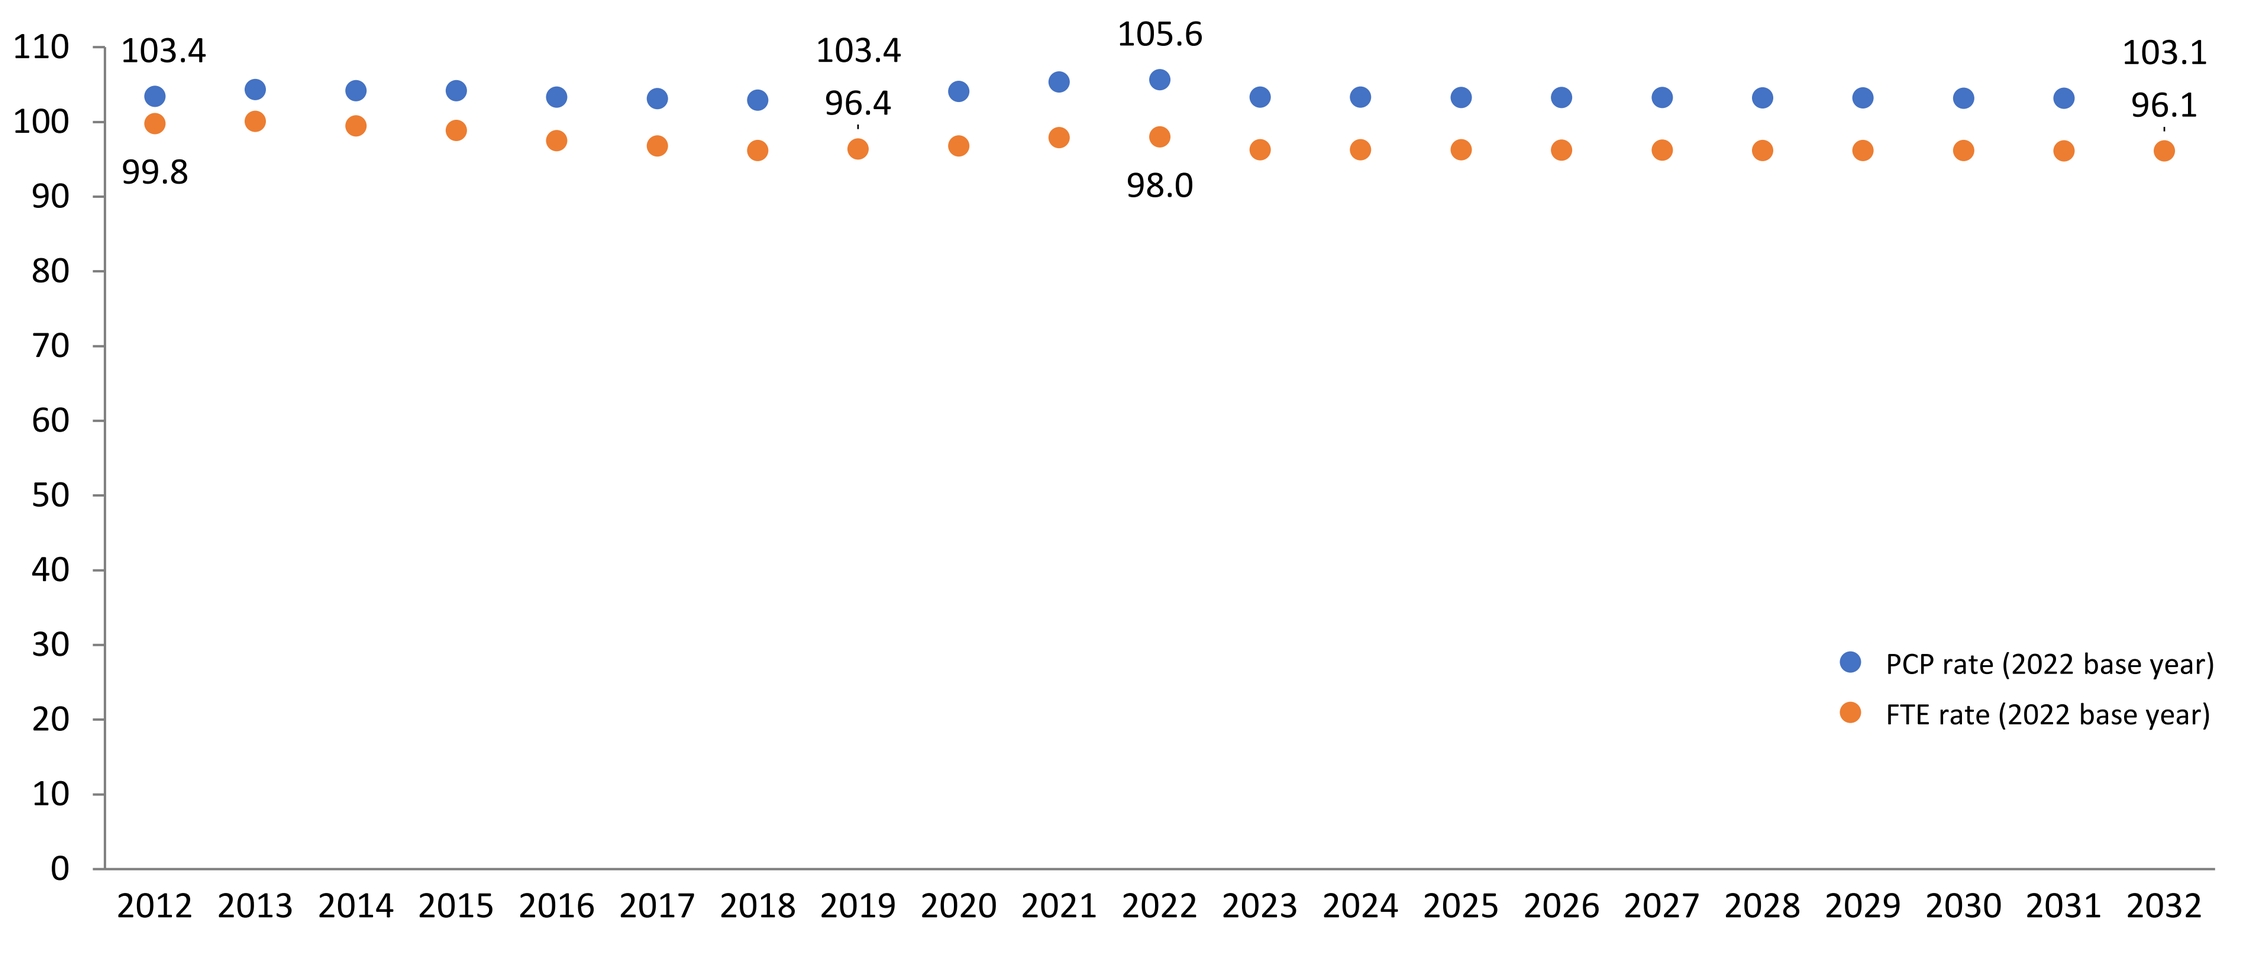
103.4
99.8
103.4
96.4
105.6
98.0
103.1
96.1
PCP rate (2022 base year)
FTE rate (2022 base year)
2012 2013 2014 2015 2016 2017 2018 2019 2020 2021 2022 2023 2024 2025 2026 2027 2028 2029 2030 2031 2032

Number of PCP per 100K inhabitants has been and will stagnate, FTEs per 100K inhabitants will decrease

Demographic change of the demand (base scenario)

PAHs development due to demographic change

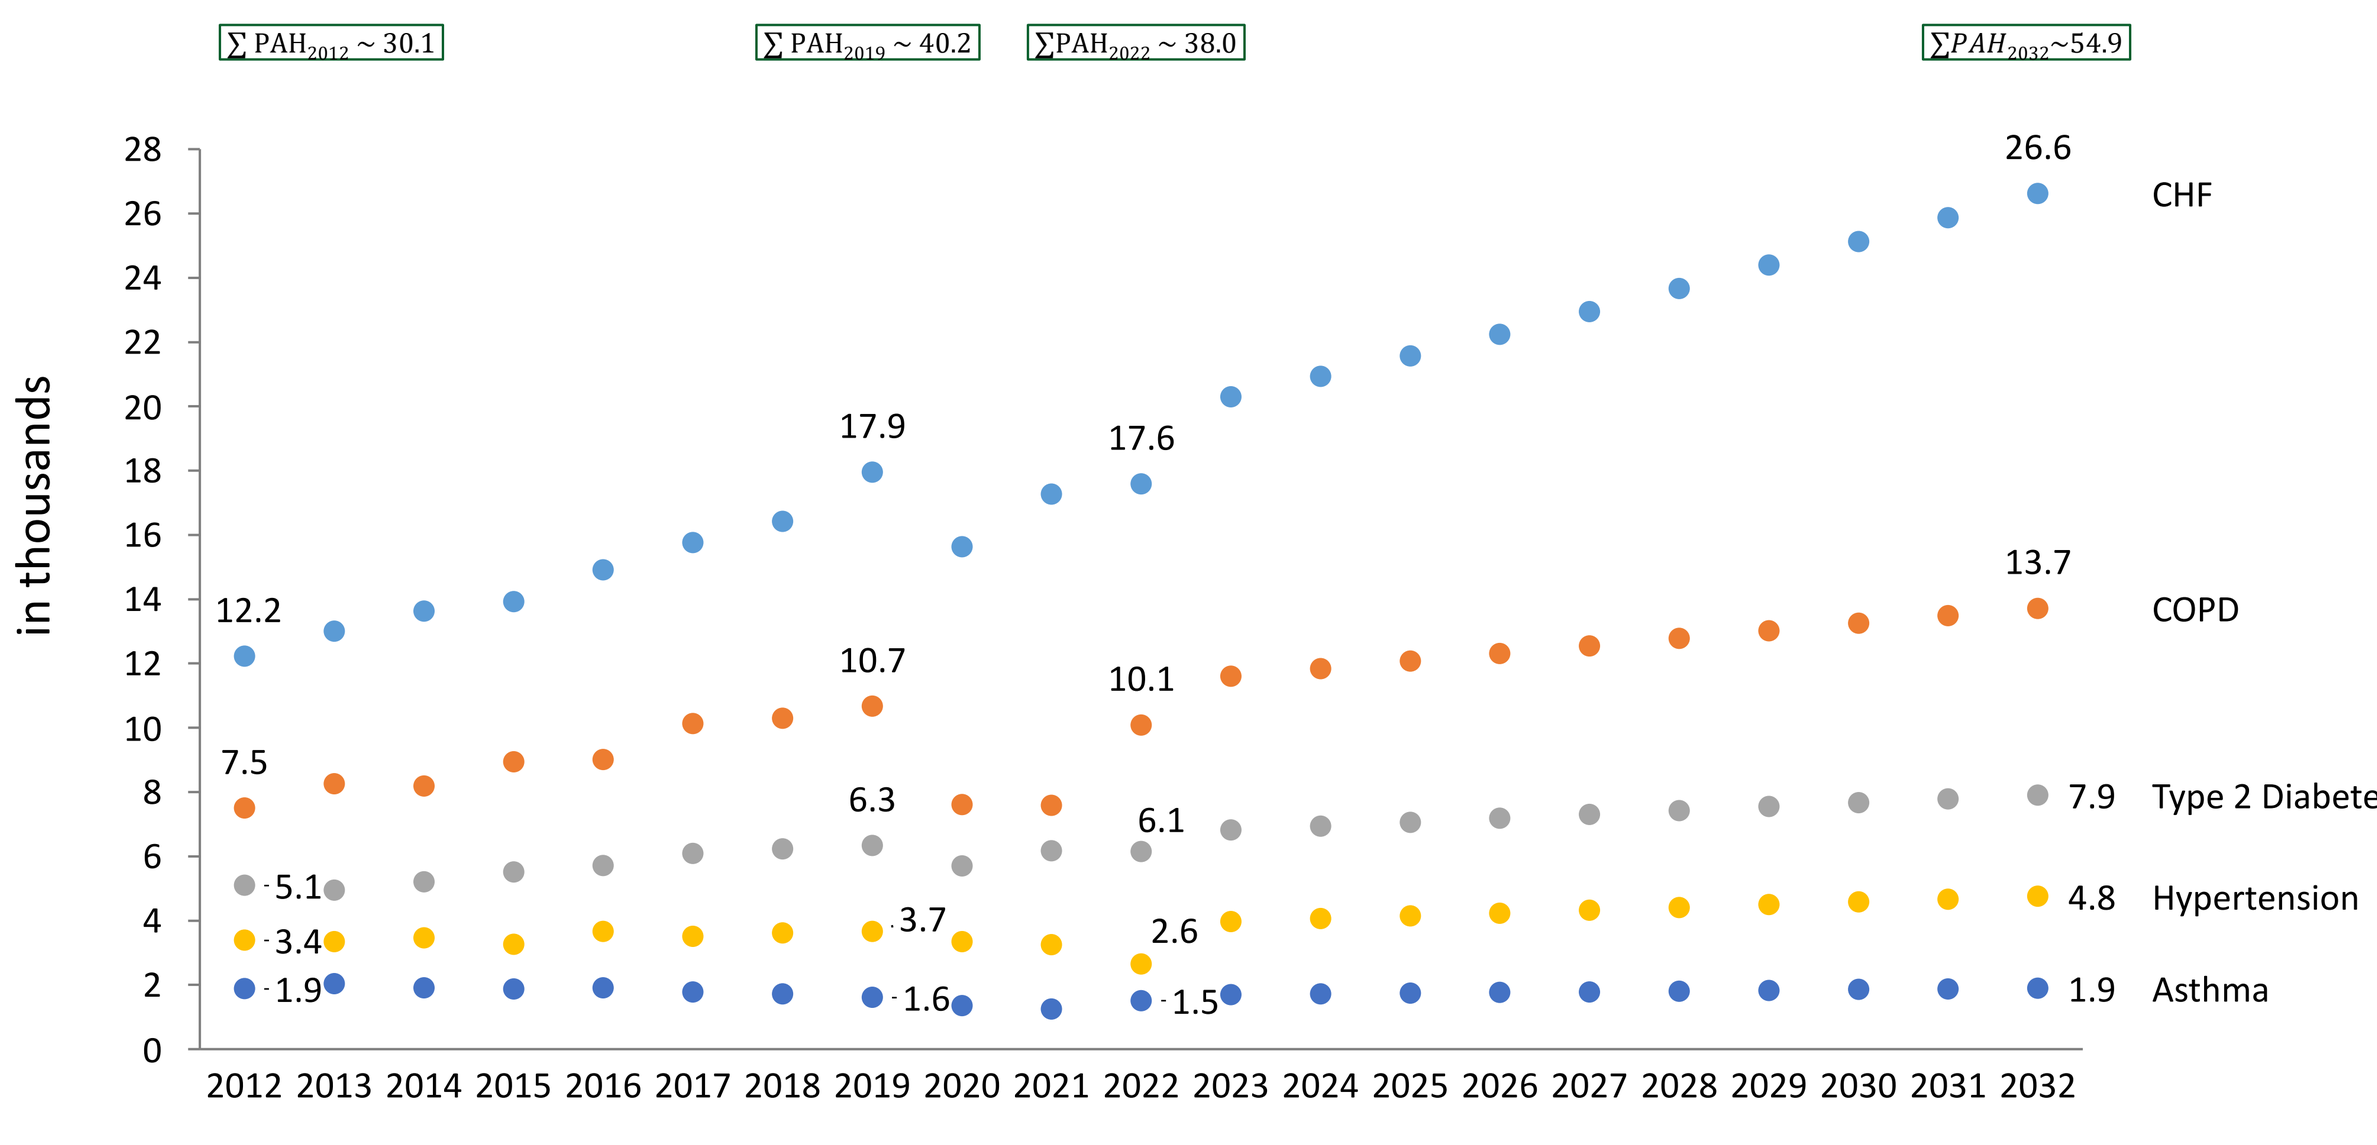
∑ PAH2012 ~ 30.1
∑ PAH2019 ~ 40.2
∑PAH2022 ~ 38.0
∑PAH2032~54.9
in thousands
CHF
COPD
Type 2 Diabetes
Hypertension
Asthma
12.2
17.9
17.6
26.6
7.5
10.7
10.1
13.7
5.1
6.3
6.1
7.9
3.4
3.7
2.6
4.8
1.9
1.6
1.5
1.9

CHF and COPD accounted for ~71% of PAH volume in 2019, increase is observed for all diseases 2012-2019/22 and until 2032

Healthcare expenditures

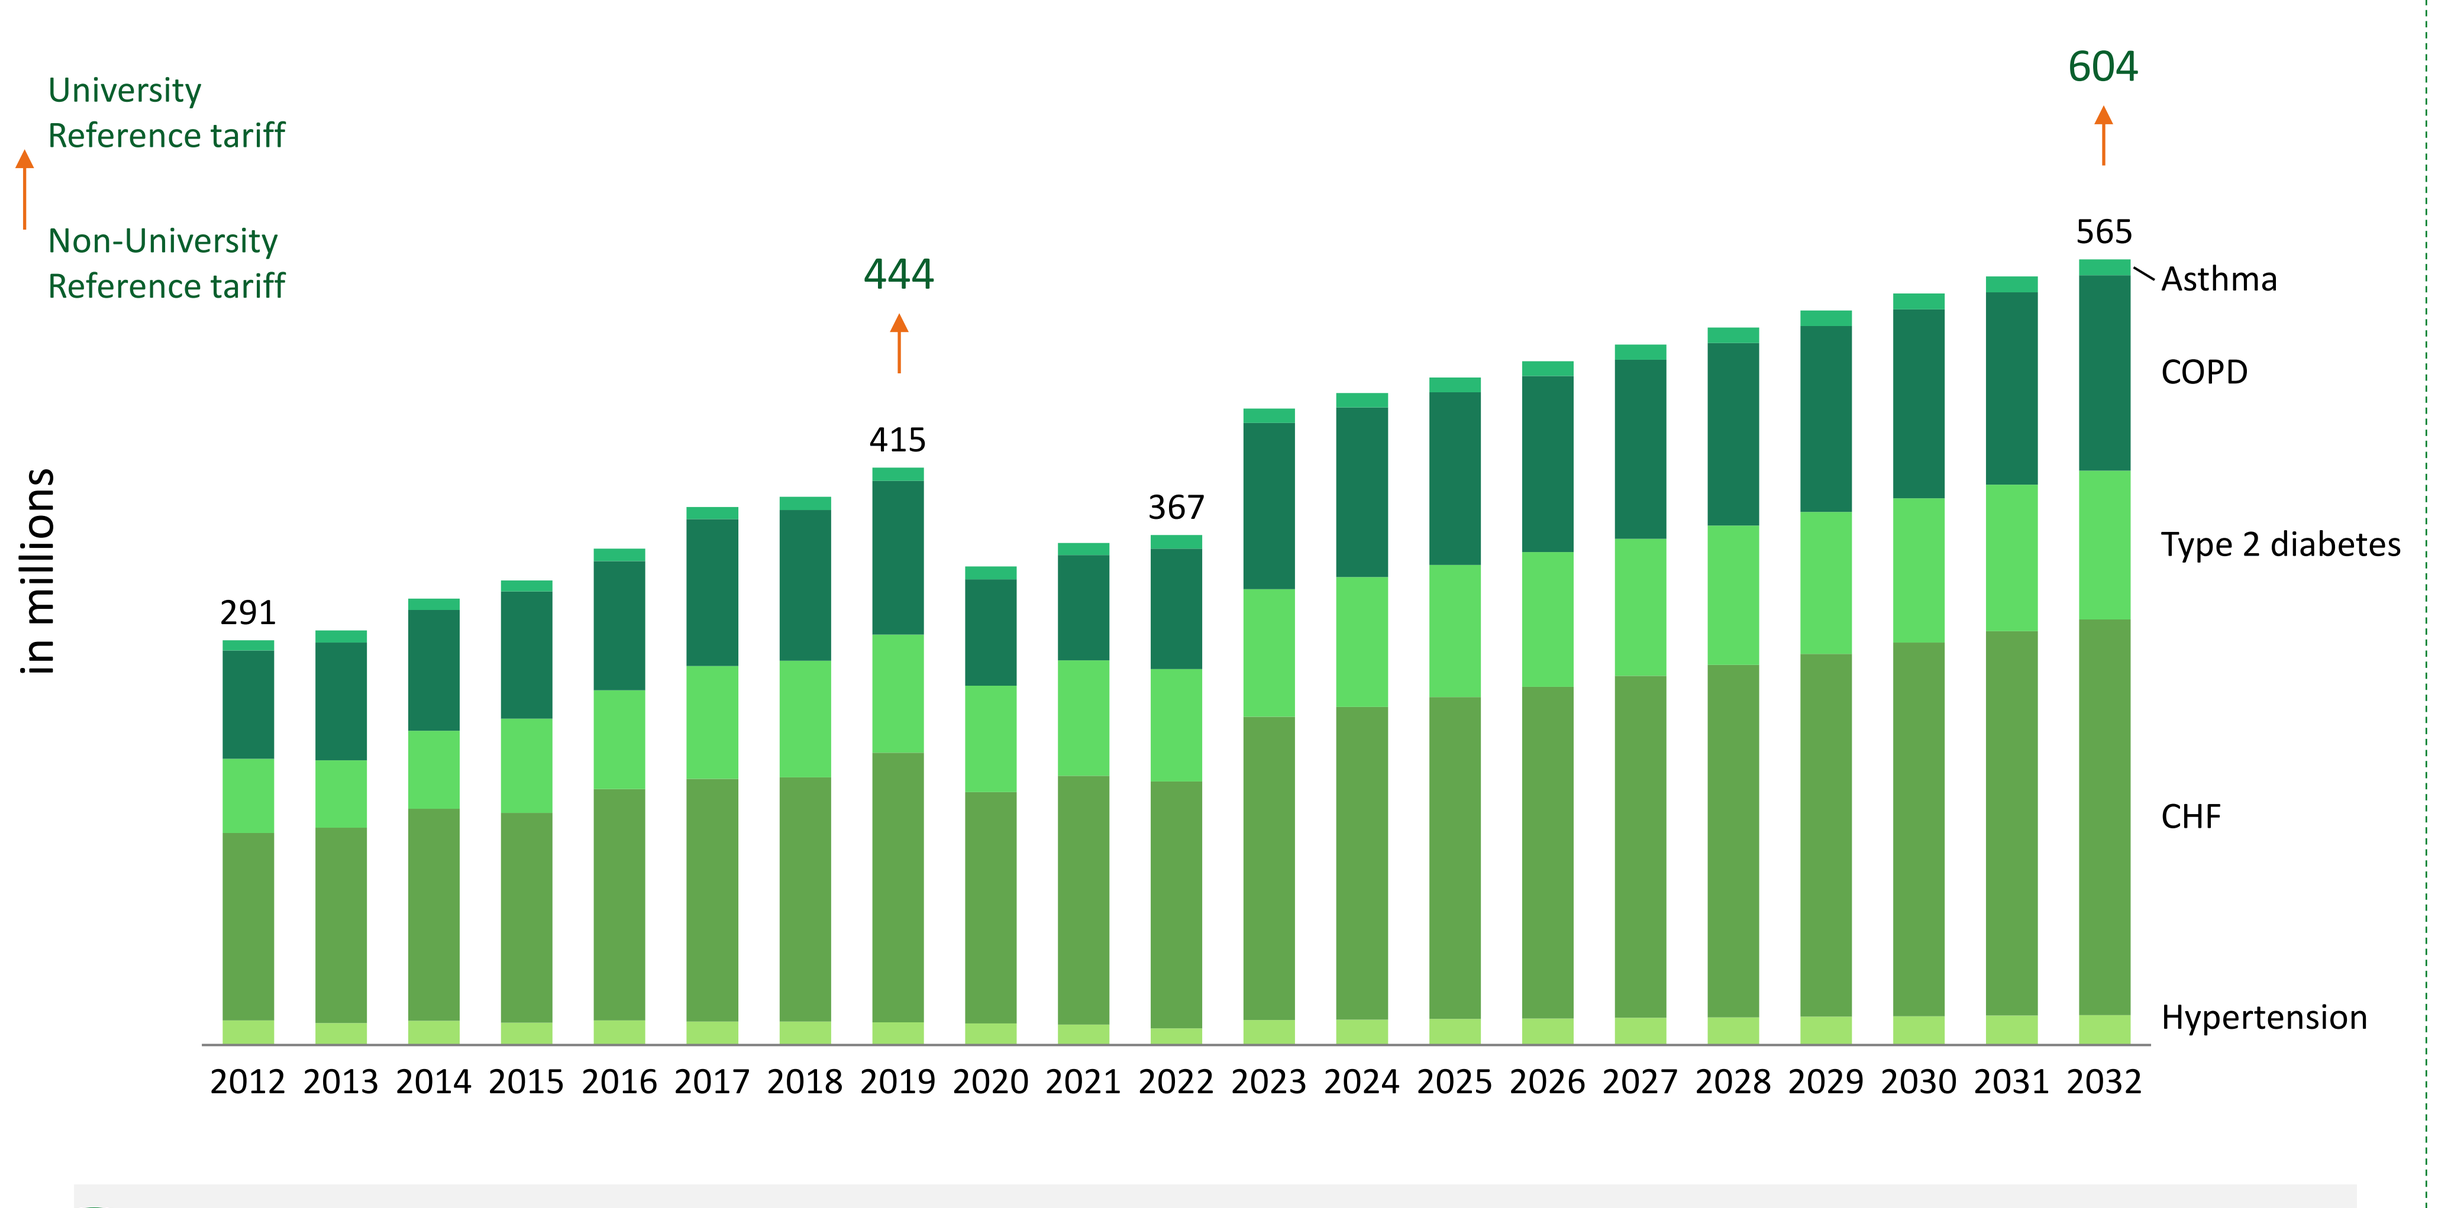
University Reference tariff
Non-University Reference tariff
in millions
291
415
444
367
565
604
Asthma
COPD
Type 2 diabetes
CHF
Hypertension

Main drivers for healthcare expenditures are CHF and COPD, also Type 2 Diabetes has a considerable share

Scenario analyses

PAHs development due to PCP supply change

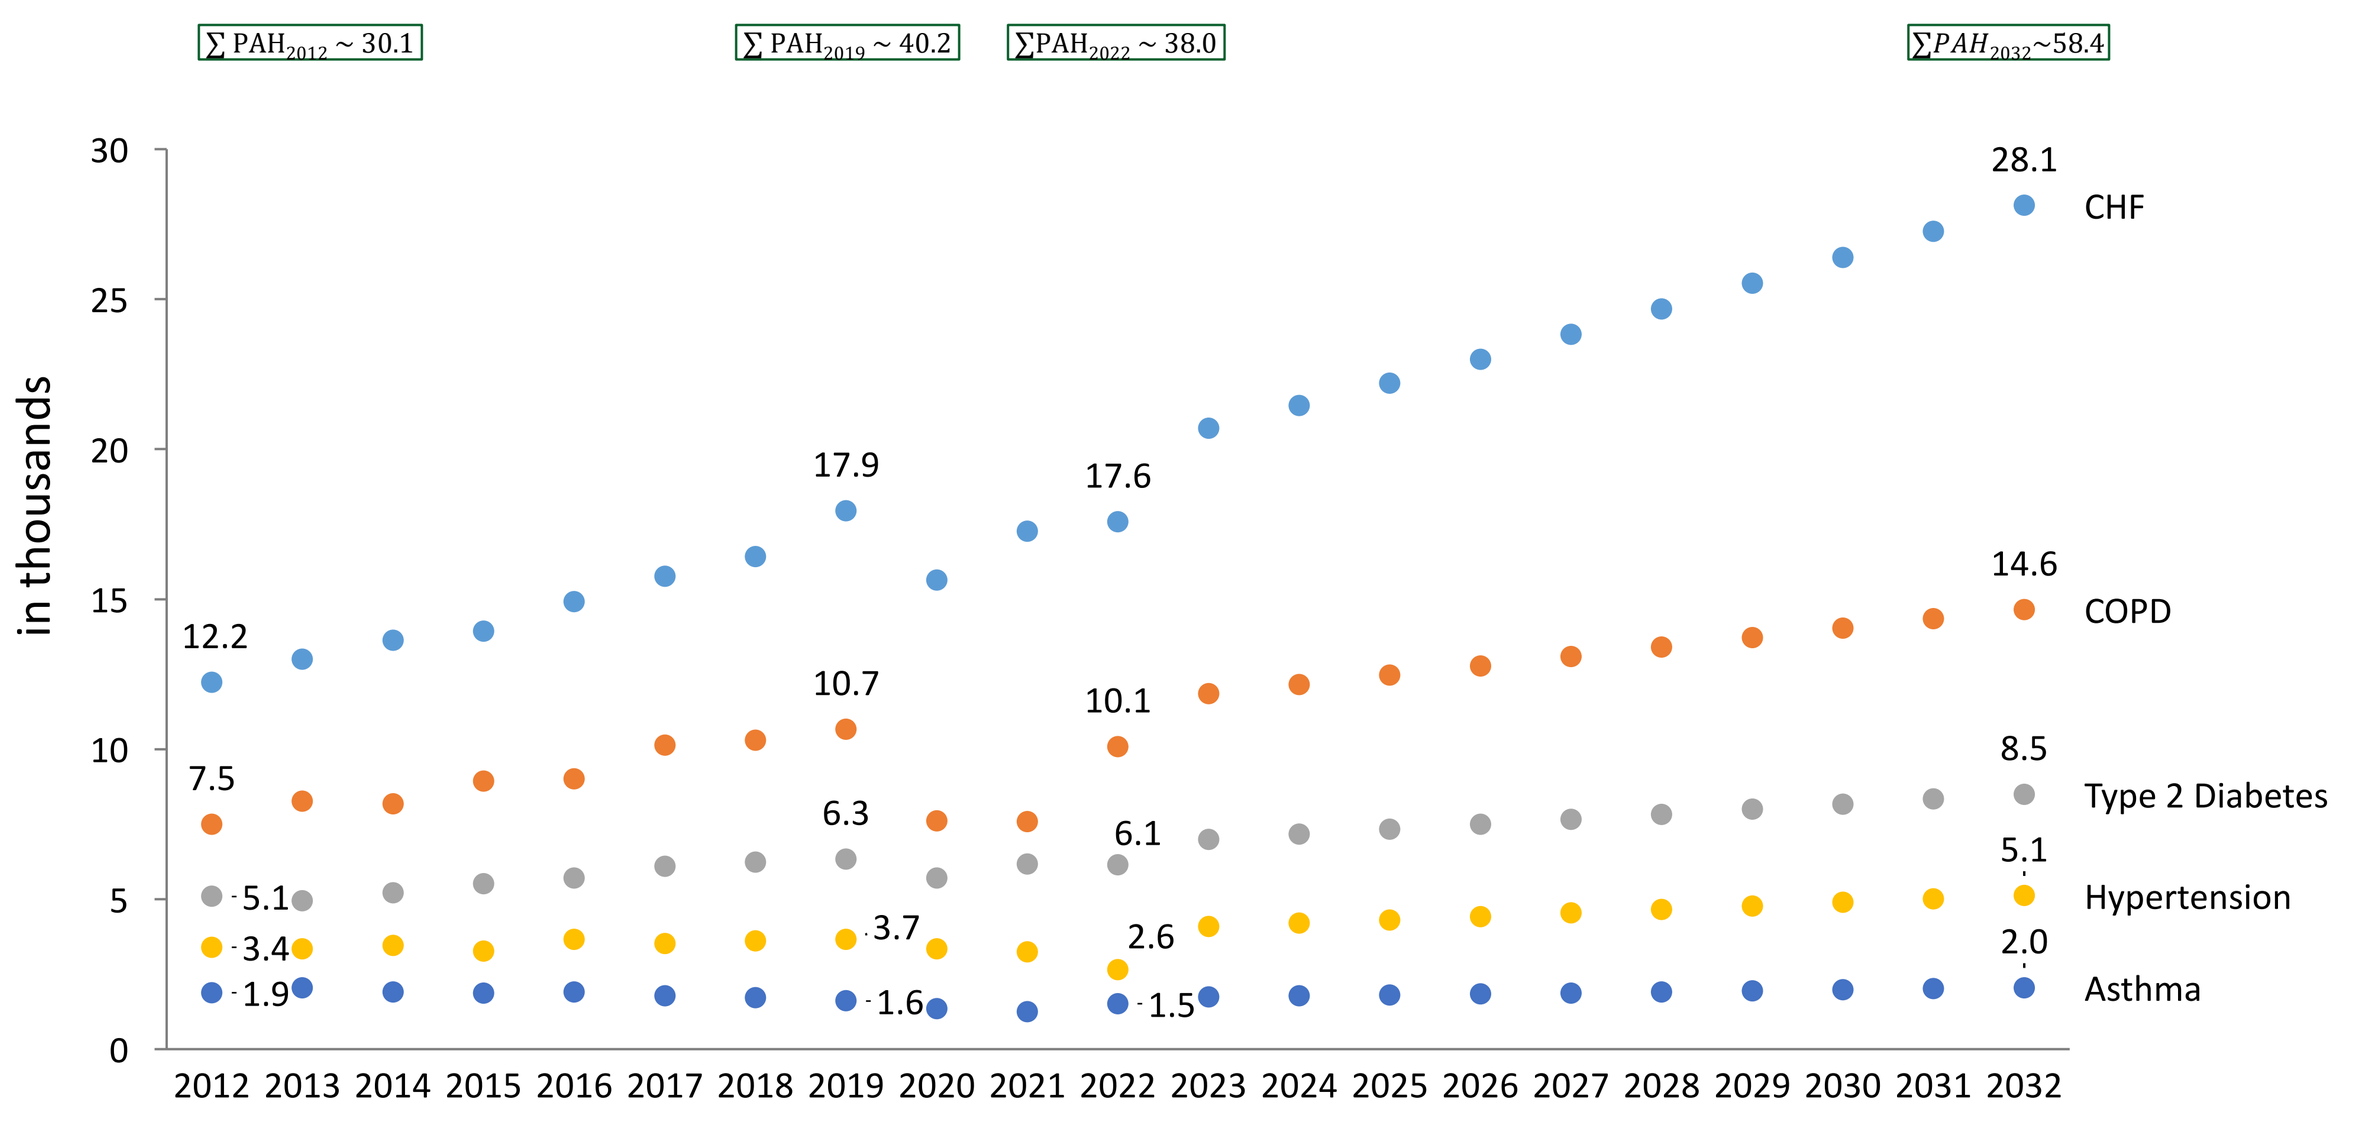
∑ PAH2012 ~ 30.1
∑ PAH2019 ~ 40.2
∑PAH2022 ~ 38.0
∑PAH2032~58.4
in thousands
CHF
COPD
Type 2 Diabetes
Hypertension
Asthma
12.2
17.9
17.6
28.1
7.5
10.7
10.1
14.6
5.1
6.3
6.1
8.5
3.4
3.7
2.6
5.1
1.9
1.6
1.5
2.0

The decrease in PCP FTEs per 100k inhabitants might increase PAHs by ~6.4% compared to demographic change scenario

PAHs development due to GDMT implementation

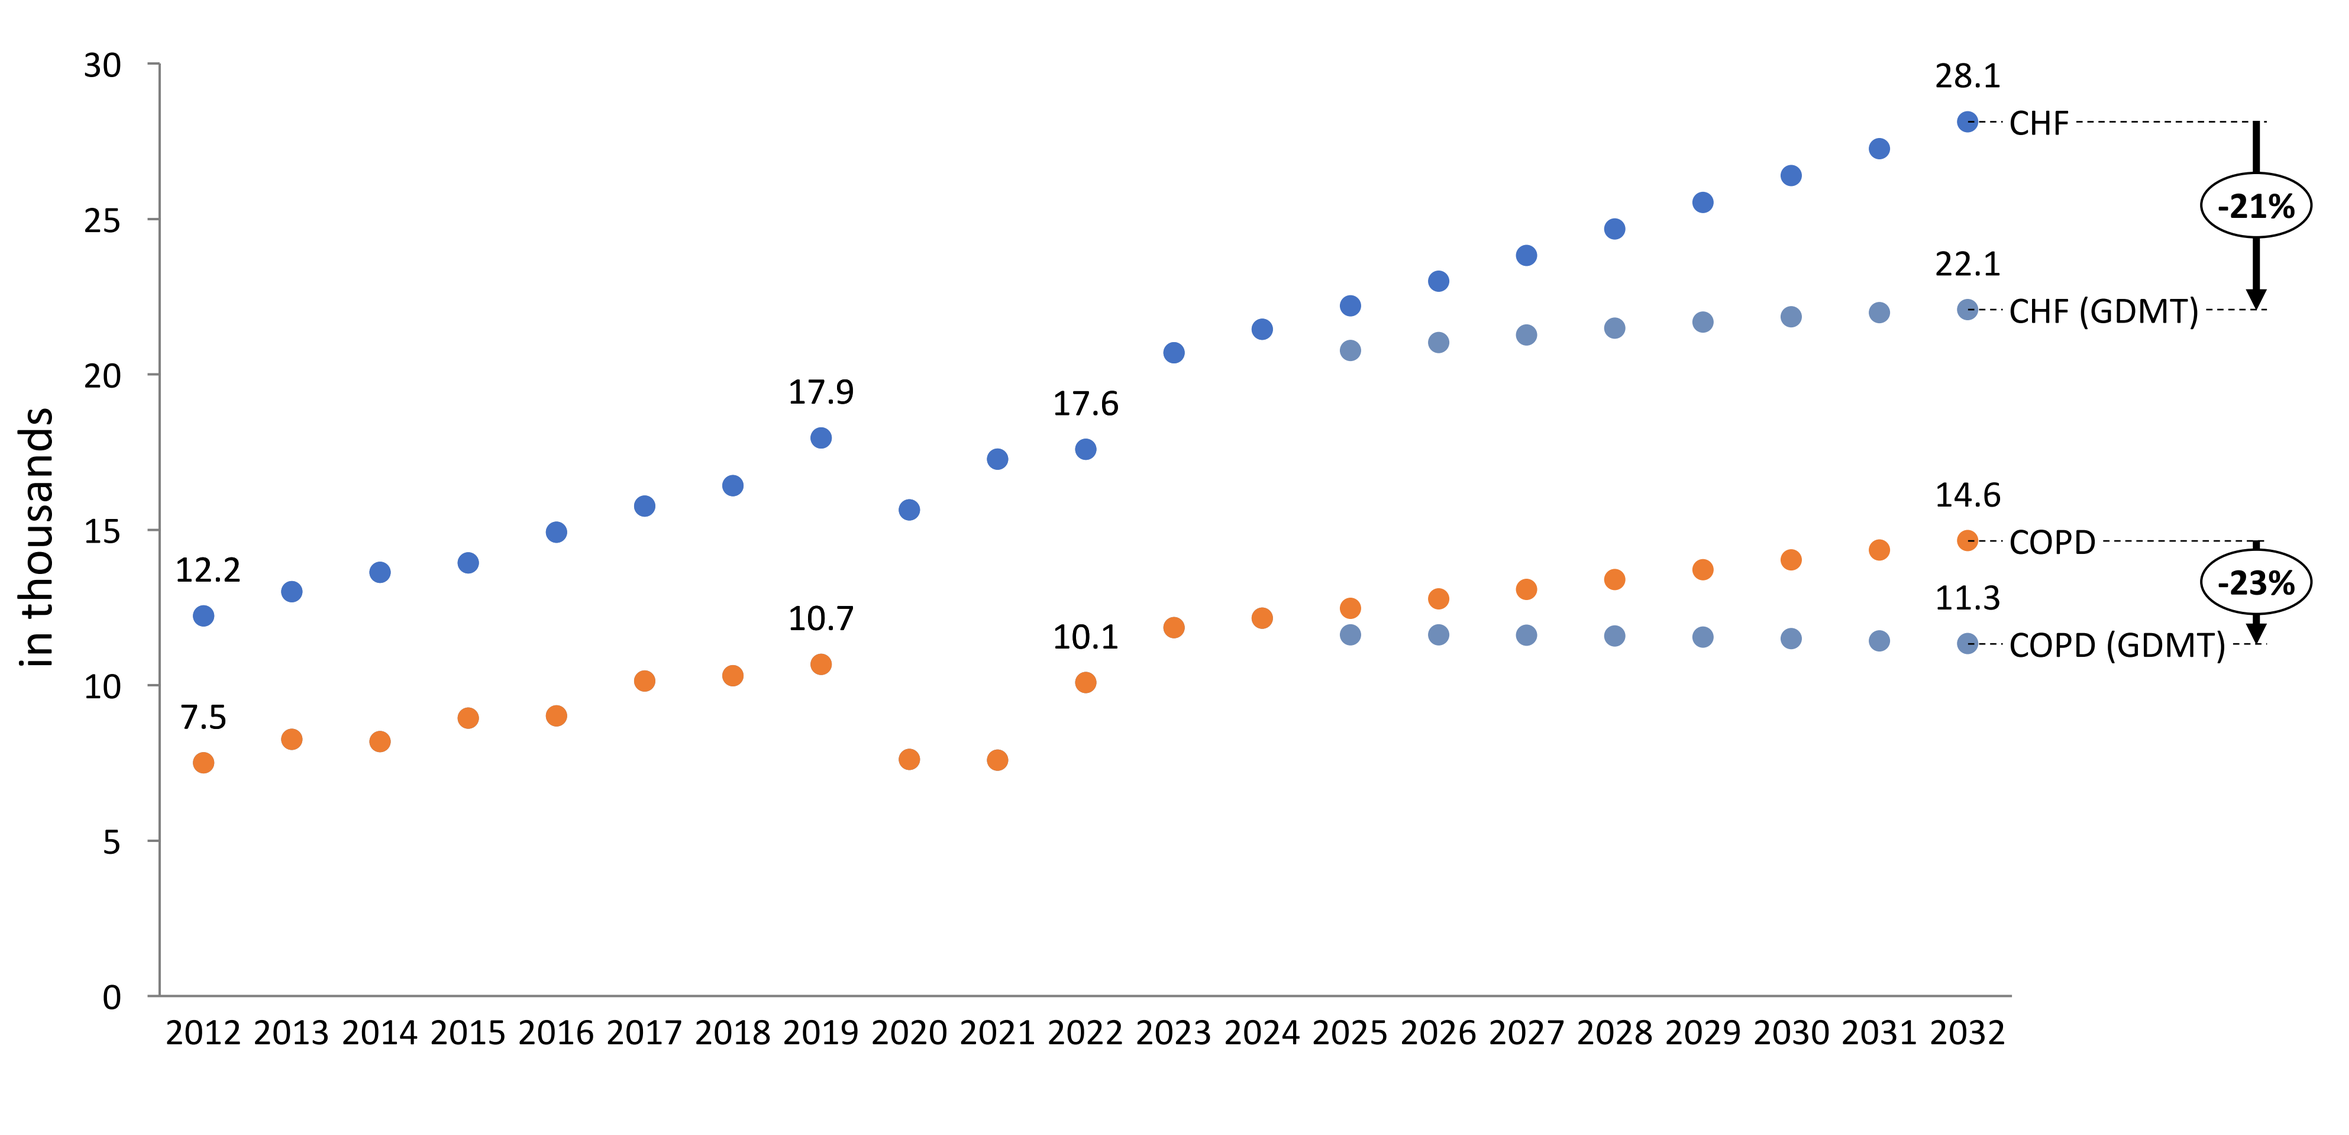
in thousands
28.1
CHF
-21%
22.1
CHF (GDMT)
14.6
COPD
-23%
11.3
COPD (GDMT)
12.2
17.9
17.6
7.5
10.7
10.1

PAHs due to CHF (HFrEF) and COPD may drop by 21 and 23% respectively due to optimal guideline adherence

DISCUSSION

- The number of PAHs increased significantly in the past (34%) and is project to rise by 45% by 2032, underscoring the rising burden of chronic diseases. When considering changes in the PCP supply structure, the situation becomes even more critical (rise by 58% by 2032)
- Improving adherence to clinical guidelines among both primary care physicians (PCPs) and patients holds significant potential to reduce the number of PAHs and the associated healthcare expenditures
- Encouraging medical students to specialize in general practice through financial incentives and mentorship programs might increase the supply of PCPs. Additionally, integrating telemedicine and digital health solutions may further decrease PAHs

SELECTED REFERENCES

(1) Jacques Spycher, Kevin Morisod, Karine Moschetti, Marie-Annick Le Pogam, Isabelle Peytremann-Bridevaux, Patrick Bodenmann, Richard Cookson, Victor Rodwin, Joachim Marti, Potentially avoidable hospitalizations and socioeconomic status in Switzerland: A small area-level analysis, Health Policy, Volume 139, 2024, 104948, ISSN 0168-8510, https://doi.org/10.1016/j.healthpol.2023.104948

(2) Berlin, C., Busato, A., Rosemann, T. et al. Avoidable hospitalizations in Switzerland: a small area analysis on regional variation, density of physicians, hospital supply and rurality. BMC Health Serv Res 14, 289 (2014). https://doi.org/10.1186/1472-6963-14-289

(3) Mannino DM, Yu T-C, Zhou H, Higuchi K. Effects of GOLD-Adherent Prescribing on COPD Symptom Burden, Exacerbations, and Health Care Utilization in a Real-World Setting. Chronic Obstr Pulm Dis. 2015;2:223–35.

(4) Palli SR, Zhou S, Shaikh A, Willey VJ. Effect of compliance with GOLD treatment recommendations on COPD health care resource utilization, cost, and exacerbations among patients with COPD on maintenance therapy. J Manag Care Spec Pharm. 2021;27:625–37.

(5) Mangold V, Boesing M, Berset C, Bridevaux P-O, Geiser T, Joos Zellweger L, et al. Adherence to the GOLD Guidelines in Primary Care: Data from the Swiss COPD Cohort. J Clin Med. 2023;12:6636.

(6) Huber M, Busch AK, Stalder-Ochsner I, Flammer AJ, Schmid-Mohler G. Medication adherence in adults after hospitalization for heart failure: A cross-sectional study. International Journal of Cardiology Cardiovascular Risk and Prevention. 2024;20:200234.

(7) Vaduganathan M, Claggett BL, Jhund PS, Cunningham JW, Pedro Ferreira J, Zannad F, et al. Estimating lifetime benefits of comprehensive disease-modifying pharmacological therapies in patients with heart failure with reduced ejection fraction: a comparative analysis of three randomised controlled trials. The Lancet. 2020;396:121–8.

CONTACT

Daria Bukanova-Berend, University of St.Gallen

- School of Medicine, Chair of Health Economics, Policy and Management, St. Jakob-Strasse 21, St. Gallen
- daria.bukanova-berend@unisg.ch

Disclosures: This project was financially supported by the AstraZeneca AG. Thomas Campbell-James is employed by the AstraZeneca AG. Justus Vogel has done a scientific workshop on clinical practice guideline adherence funded by the AstraZeneca AG.