Physical Therapy in Patients with Pulmonary Arterial Hypertension (PAH): Results from a German Claims Data Analysis

Acceptance Code: HSD44

Barthuber L, Nill I, Schmid T
MSD Sharp & Dohme GmbH, Munich, Germany

Jacob C
Xcenda GmbH, part of Cencora Inc., Hannover, NI, Germany

Objectives

- Pulmonary arterial hypertension (PAH) is a rare, life-threatening, and progressive disease characterized by remodeling and progressive narrowing of the pulmonary vessels.[1]
- Medical guidelines recommend supervised physical therapy (PT) for clinically stable patients on PAH-specific treatment.[2]
- To date, data on the utilization of PT in PAH patients in Germany are not available. By conducting a claims data analysis, we aimed at closing this gap.

Methods

Study design

- A retrospective claims database analysis using anonymized German Statutory Health Insurance (SHI) data was conducted.
- The study covered a time period from 2016 to 2021 to include the most recent available years in the database at the time of study initiation.

Data source

- Data from the Institute for Applied Health Research Berlin GmbH (InGef) research database was used.
- The database contains information on insured individuals from more than half of the German SHIs and is representative for the German population with respect to age, gender and region.[3]

Study population

- The study population comprised PAH patients identified by ≥ 1 inpatient (primary or secondary discharge diagnosis) and/or ≥ 2 outpatient (verified) diagnosis codes for PAH (ICD-10-GM I27.0) in 2021.
- Thereof, patients with a prescription of pre-defined PAH-specific treatment in 2021 were assessed.[4]
- To determine patients on PT, a list of all aids and remedies according to the Medical Aids[5] and Remedies[6] Directive of Federal Joint Committee (G-BA) (identified by aids position numbers on a ten-digit level and by remedies position number on five-digit level) recorded between 2016- 2021 was generated.
- Thereafter, PAH-related aids and remedies (incl. supervised PT such as breathing exercises) were selected.
- Specific inhalation and respiratory devices were defined as PAH-related aids.
- Patients with PT were identified by the remedy codes shown in **Table 1**.

Table 1: PAH-related remedies

Remedies position number	Description "Service provider" (digit 1)	Description "Service type" (digits 2-3)	Description "Service" (digits 2-5)
20501	Physiotherapist	Physiotherapy, normal, individual treatment	Physiotherapy, including respiratory gymnastics, also on a neurophysiological basis
20702	Physiotherapist	Physiotherapy, special, individual treatment	Physiotherapy, also respiratory therapy for mucoviscidosis and severe bronchial diseases, 60 min
33103	Speech therapist/ breathing, speech and voice teacher	Individual treatment (price incl. preparation and follow-up)	45 minutes (therapy time with the patient)

- Number of identified PAH patients was extrapolated to the German population based on data extracted from the Federal Office of Statistics (DESTATIS)[7] for the year 2021.

Results

- Among approximately 3.3 million individuals continuously observable in the InGef research database from 2016 to 2021, our study identified 3,270 patients (97.9 per 100,000 individuals) with at least one inpatient or two outpatient diagnoses for PAH in 2021 **(see Figure 1)**. This corresponds to approximately 81,500 PAH patients in Germany.
- Thereof, 403 patients (12.1 per 100,000 individuals) received PAH-specific treatment in 2021 **(see Figure 1)**. Extrapolated to the German population, this corresponds to approximately 10,030 patients with PAH-specific treatment.

Figure 1: Patient selection steps

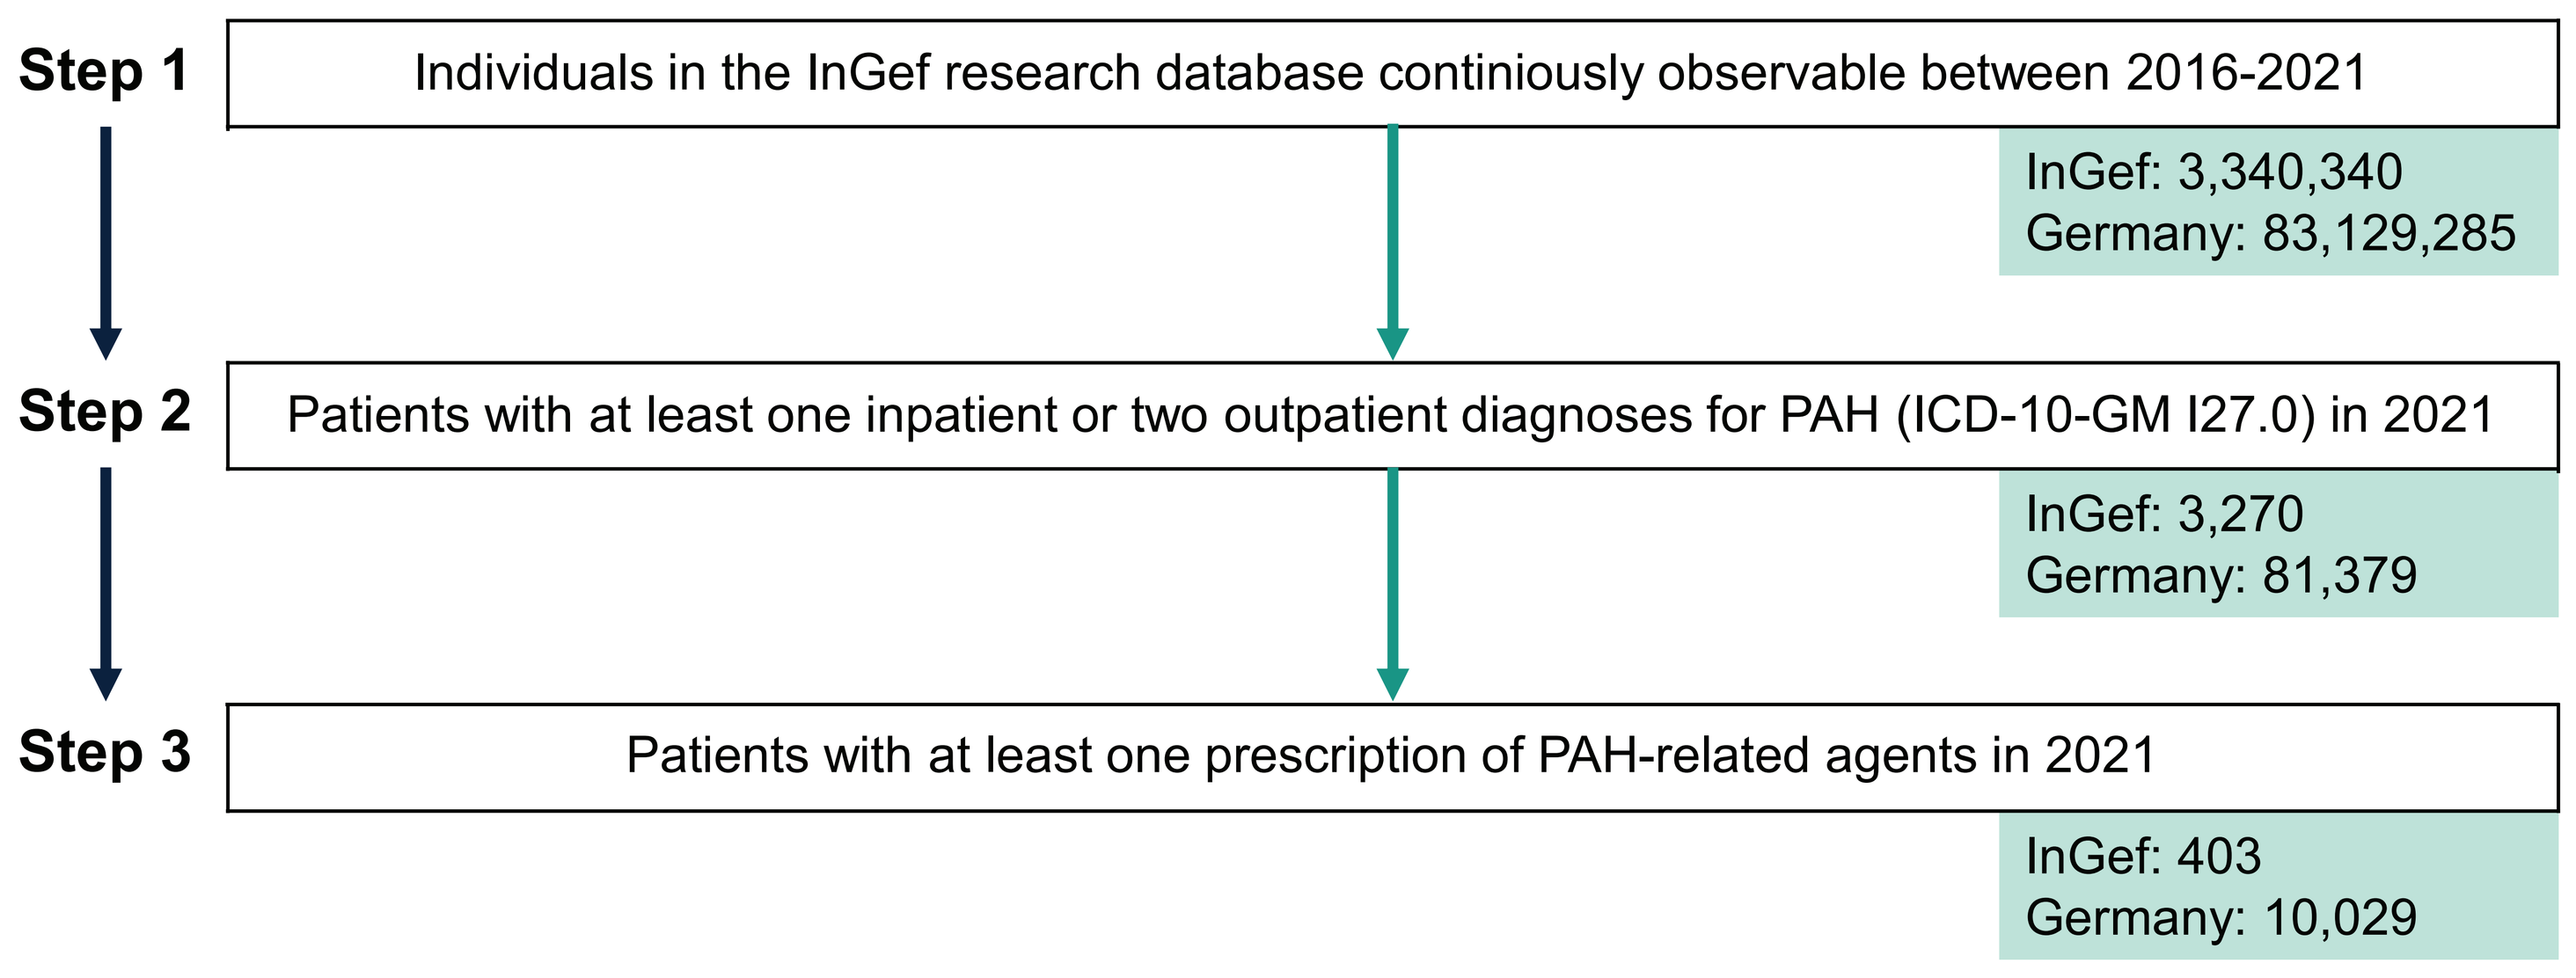
Step 1
Individuals in the InGef research database continiously observable between 2016-2021
InGef: 3,340,340
Germany: 83,129,285
Step 2
Patients with at least one inpatient or two outpatient diagnoses for PAH (ICD-10-GM I27.0) in 2021
InGef: 3,270
Germany: 81,379
Step 3
Patients with at least one prescription of PAH-related agents in 2021
InGef: 403
Germany: 10,029

- The average age was 71.0 years (SD: 14.8 years), 63.8% were female **(see Table 2)**.

Table 2 Age and gender distribution of PAH patients treated with PAH-related agents in 2021*

Demographics	Male (n=146) n, summary statistic	Male %	Female (n=257) n, summary statistic	Female %	Total (n=403) n, summary statistic	Total %
Age distribution						
Mean	70.1		71.6		71.0	
SD	15.3		14.4		14.8	
Min	1		4		1	
Q1	63		63		63	
Median	74		76		75	
Q3	80		82		81	
Max	95		93		95	
Age groups						
0-17 years	<5	-	<5	-	5	1.2%
18-29 years	0	0.0%	<5	-	<5	-
30-39 years	<5	-	11	4.3%	13	3.2%
40-49 years	<5	-	9	3.5%	10	2.5%
50-59 years	18	12.3%	23	8.9%	41	10.2%
60-69 years	31	21.2%	43	16.7%	74	18.4%
70-79 years	45	30.8%	71	27.6%	116	28.8%
≥80 years	45	30.8%	98	38.1%	143	35.5%
Total	**146**	**36.2%**	**257**	**63.8%**	**403**	**100.0%**

** Due to data protection restrictions, the number of patients <5 cannot be reported*

- Valid aids and remedies data from 2016 to 2021 were available for 383 patients (extrapolated to the German population: ~9,500 patients) **(see Figure 2)**.
- Thereof, 45.4% (n=174; extrapolated to the German population: ~4,300 patients) had a prescription for PAH-related aids and remedies in 2021 **(see Figure 2A)** and 67.4% (n=258; extrapolated to German population: ~6,400 patients) between 2016-2021 **(see Figure 2B)**.
- PT was prescribed to 25.6% (n=98; extrapolated to the German population: ~2,450 patients) of patients with valid aids and remedies data in 2021 **(see Figure 2A)** and 47.8% (n=183; extrapolated to the German population: ~4,560 patients) of patients between 2016-2021 **(see Figure 2B)**.

Figure 2: Number of patients with physical therapy

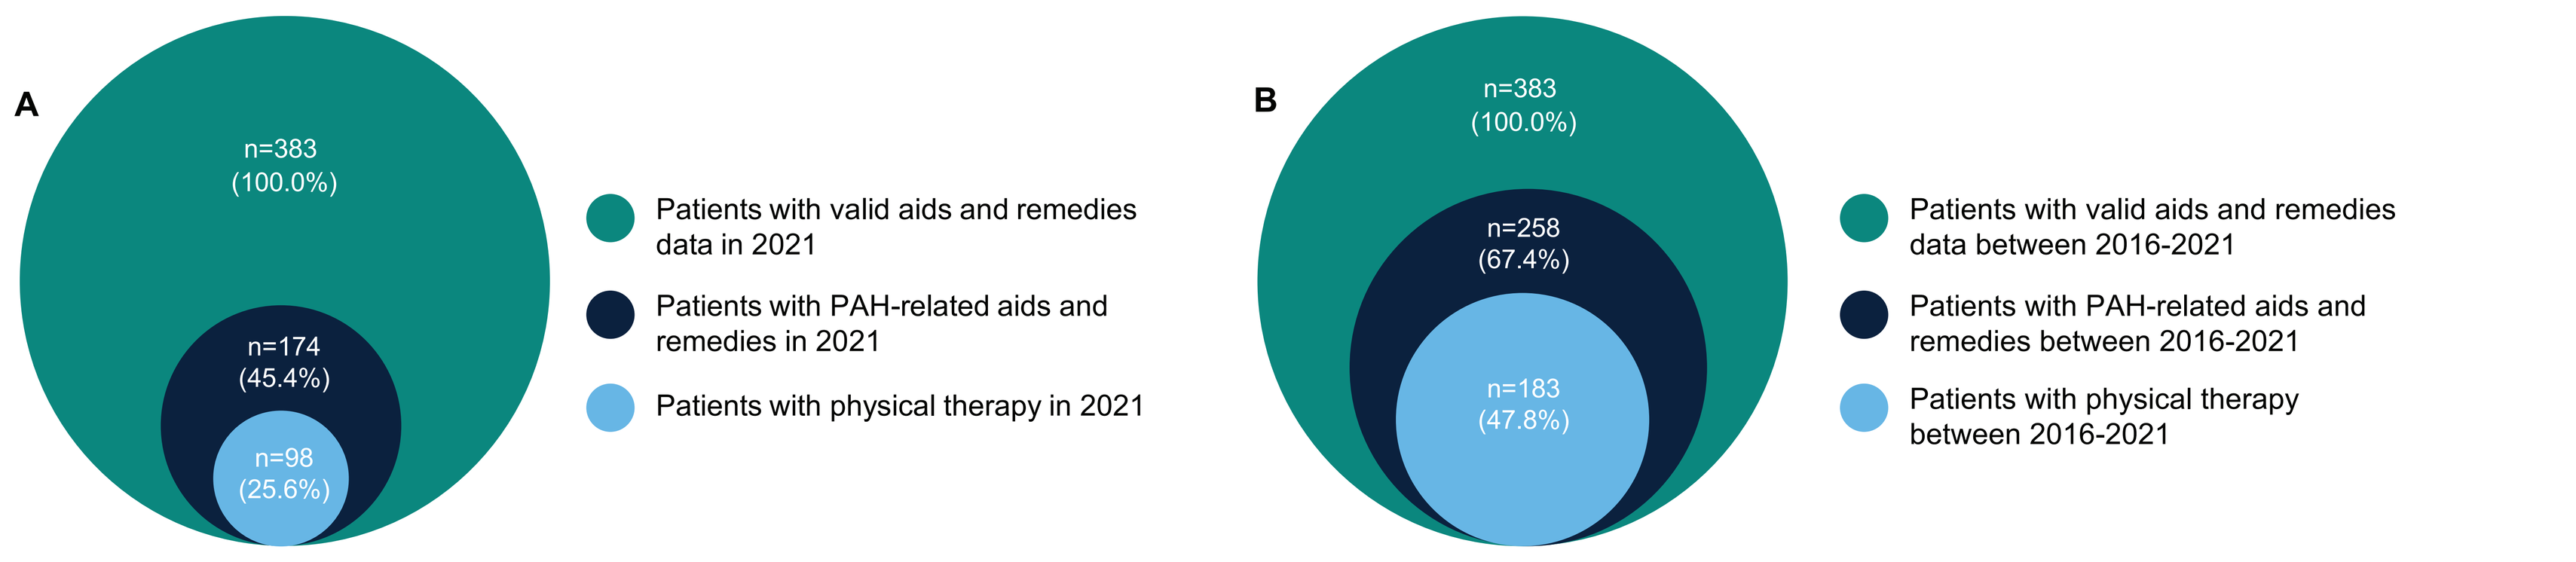
A
n=383
(100.0%)
n=174
(45.4%)
n=98
(25.6%)
Patients with valid aids and remedies data in 2021
Patients with PAH-related aids and remedies in 2021
Patients with physical therapy in 2021
B
n=383
(100.0%)
n=258
(67.4%)
n=183
(47.8%)
Patients with valid aids and remedies data between 2016-2021
Patients with PAH-related aids and remedies between 2016-2021
Patients with physical therapy between 2016-2021

Discussion

- As there is no specific diagnosis code for PAH, ICD-10-GM code I27.0 "Primary pulmonary hypertension" was used in the analysis. This code might be not specific enough and could have caused an overestimation of the number of PAH patients, especially compared to prevalences reported in published literature[8,9].
- Claims data may contain coding errors, including the treating physician's coding behavior (e.g., due to lack of precise diagnosis codes) leading to overestimation of patient counts.
- Data on a highly supervised rehabilitation program in expert centers which is available for PAH patients in Germany, is not included in the database. This may have caused an underestimation of the proportion of patients with PT.
- Nevertheless, to the best of our knowledge this is the first study to investigate the prescription of PT in PAH patients in Germany. The results contribute to closing the knowledge gap and providing initial figures which are a considerable basis for further analyses.

Conclusion

- We found that one in four of the diagnosed PAH patients treated with PAH-specific treatment received PT as part of their aids and remedies prescription in 2021. When considering a more extended timeframe from 2016-2021, nearly half of the identified patients had a PT prescription.
- Further research is needed to evaluate the patterns of PT prescriptions in PAH patients in Germany.

References:

1. Boucly A, Gerges C, Savale L, Jaïs X, Jevnikar M, Montani D, Sitbon O, Humbert M. Pulmonary arterial hypertension. *Presse Med.* Sep 2023;52(3):104168. doi:10.1016/j.lpm.2023.104168
2. Humbert M, Kovacs G, Hoeper MM, Badagliacca R, Berger RMF, Brida M, Carlsen J, Coats AJS, Escribano-Subias P, Ferrari P, Ferreira DS, Ghofrani HA, Giannakoulas G, Kiely DG, Mayer E, Meszaros G, Nagavci B, Olsson KM, Pepke-Zaba J, Quint JK, Rådegran G, Simonneau G, Sitbon O, Tonia T, Toshner M, Vachiery JL, Vonk Noordegraaf A, Delcroix M, Rosenkranz S. 2022 ESC/ERS Guidelines for the diagnosis and treatment of pulmonary hypertension. *Eur Respir J.* Jan 2023;61(1)doi:10.1183/13993003.00879-2022
3. Ludwig M, Enders D, Basedow F, Walker J, Jacob J. Sampling strategy, characteristics and representativeness of the InGef research database. *Public Health.* May 2022;206:57-62. doi:10.1016/j.puhe.2022.02.013
4. Deutsche Gesellschaft für Pädiatrische Kardiologie und Angeborene Herzfehler e.V. (DGPK). S2k Leitlinie Pulmonale Hypertonie. Version 3.0. Stand 29.04.2020. https://register.awmf.org/de/leitlinien/detail/023-038
5. Gemeinsamer Bundesausschuss (G-BA). Richtlinie über die Verordnung von Hilfsmitteln in der vertragsärztlichen Versorgung – HilfsM-RL. https://www.g-ba.de/richtlinien/13/
6. Gemeinsamer Bundesausschuss (G-BA). Richtlinie über die Verordnung von Heilmitteln in der vertragsärztlichen Versorgung – HeilM-RL. https://www.g-ba.de/richtlinien/12/
7. Statistisches Bundesamt DESTATIS. Ergebnisse der Bevölkerungsfortschreibung auf Grundlage des Zensus 2011. Bevölkerung nach Nationalität und Geschlecht 2021. https://www.destatis.de/DE/Themen/Gesellschaft-Umwelt/Bevoelkerung/Bevoelkerungsstand/Tabellen/liste-zensus-geschlechtstaatsangehoerigkeit.html
8. Leber L, Beaudet A, Muller A. Epidemiology of pulmonary arterial hypertension and chronic thromboembolic pulmonary hypertension: identification of the most accurate estimates from a systematic literature review. *Pulm Circ.* Jan-Mar 2021;11(1):2045894020977300. doi:10.1177/2045894020977300
9. Hoeper MM, Huscher D, Pittrow D. Incidence and prevalence of pulmonary arterial hypertension in Germany. *Int J Cardiol.* Jan 15 2016;203:612-3. doi:10.1016/j.ijcard.2015.11.001