LOSS OF PRODUCTIVITY (LoP) AND QUALITY OF LIFE (QoL) OF HIGHLY CONSULTATIVE MIGRAINE PATIENTS IN COLOMBIA

PCR30

Arciniegas J[*], Bolaños-López J[§], Reyes Sanchez JM[*], Bello C[§], Gonzalez FJ[§], Perez LE[§], Garcia M[*], Mesa A[*], Gomez D[§]

[*]Pfizer, Colombia. [§]Biociencias-Sura

BACKGROUND

- Migraine imposes a high social, personal, and economic burden, not only because of its impact on life (QoL) and the consumption of direct medical resources by migraine patients, but because of its impact on work productivity, migraine is one of the of the most frequent causes of absenteeism at work[1,2]. However, there is currently limited data on QoL and virtually no information available on loss of productivity (LoP) of Latin American patients, especially in highly consultative migraine patients in Colombia.

OBJECTIVE

- To estimate the LoP costs and QoL associated with highly consultative migraine adult patients in an HMO in Colombia.

METHODS

- This is a cross-sectional study that integrated data from electronic health records and a probabilistic telephone survey of highly consultative migraine patients' who received medical attention with an HMO in Colombia. Most demographical and clinical characteristics came from the health records and the LoP and QoL came from the survey.
- Based on a previous study[3], high consultation is defined as two or more migraine-related diagnoses (ICD-10th code: G43) visits to health services in 2022. To estimate the loss of productivity the HALT-90 index was used[4] and to measure the QoL the WHOQoL-BREF was applied[5], both instruments were administered in their translated and authorized versions in Spanish. HALT-90 index measures migraine's impact on the patient's life and lifestyle (grading it between mild to severe impact) allowing to estimate productive time lost through the disabling effect of the disease using 5 questions[6]. WHOQoL-BREF measures 4 specific health domains and a general health domain, through 26 Likert questions, that provides 5 indexes ranging from 0 to 100 points, with 0 being the lowest and 100 the highest[7].
- The sampling method corresponded to one-stage probability sampling without replacement. The population size is 3,762, the design effect of the sample design (DEFF) was 1 corresponding to a simple random sampling design, the significance level was set at 5% and it aimed to reduce the estimated coefficient of variation. The survey was collected during the first semester of 2024.
- The exchange rate used was COP 3,845.22= 1 USD. Income imputation was conducted, using the mean salary by contribution base income according to the HMO records. The cost of LoP was estimated multiplying the absenteeism and presentism days per the daily income.

$LoPCost_{\iota} = (Absenteeism.days_{\iota} + Presentism.days_{\iota}) * Daily.income_{\iota}$

RESULTS

- The expanded results represent a total of 3,762 highly consultative migraine adult patients at the HMO which derived from the 504 random sample.
- Highly consultative migraine adult patients were female (89.9%, CI95% 87.2%- 92.5%), middle-aged (37.7 years, CI95% 36.6-38.9), lived in urban areas (97.0%, CI95% 95.5%-98.5%), were affiliated to the contributory regime* (86.1%, CI95% 83.1%-89.1%), and 51.6% were single (CI95% 47.2%-56.0%).
- This population tended to have an education beyond high school; 37.3% had a technical or technological degree (CI95% 33.1%-41.5%), 17.9% had a bachelor's degree (CI95% 14.5%-21.2%) and 6.0% had a postgraduate degree (CI95% 3.9%-8.0%) (**Figure 1**).

Figure 1. Barchart of educational level of highly consultative migraine patients

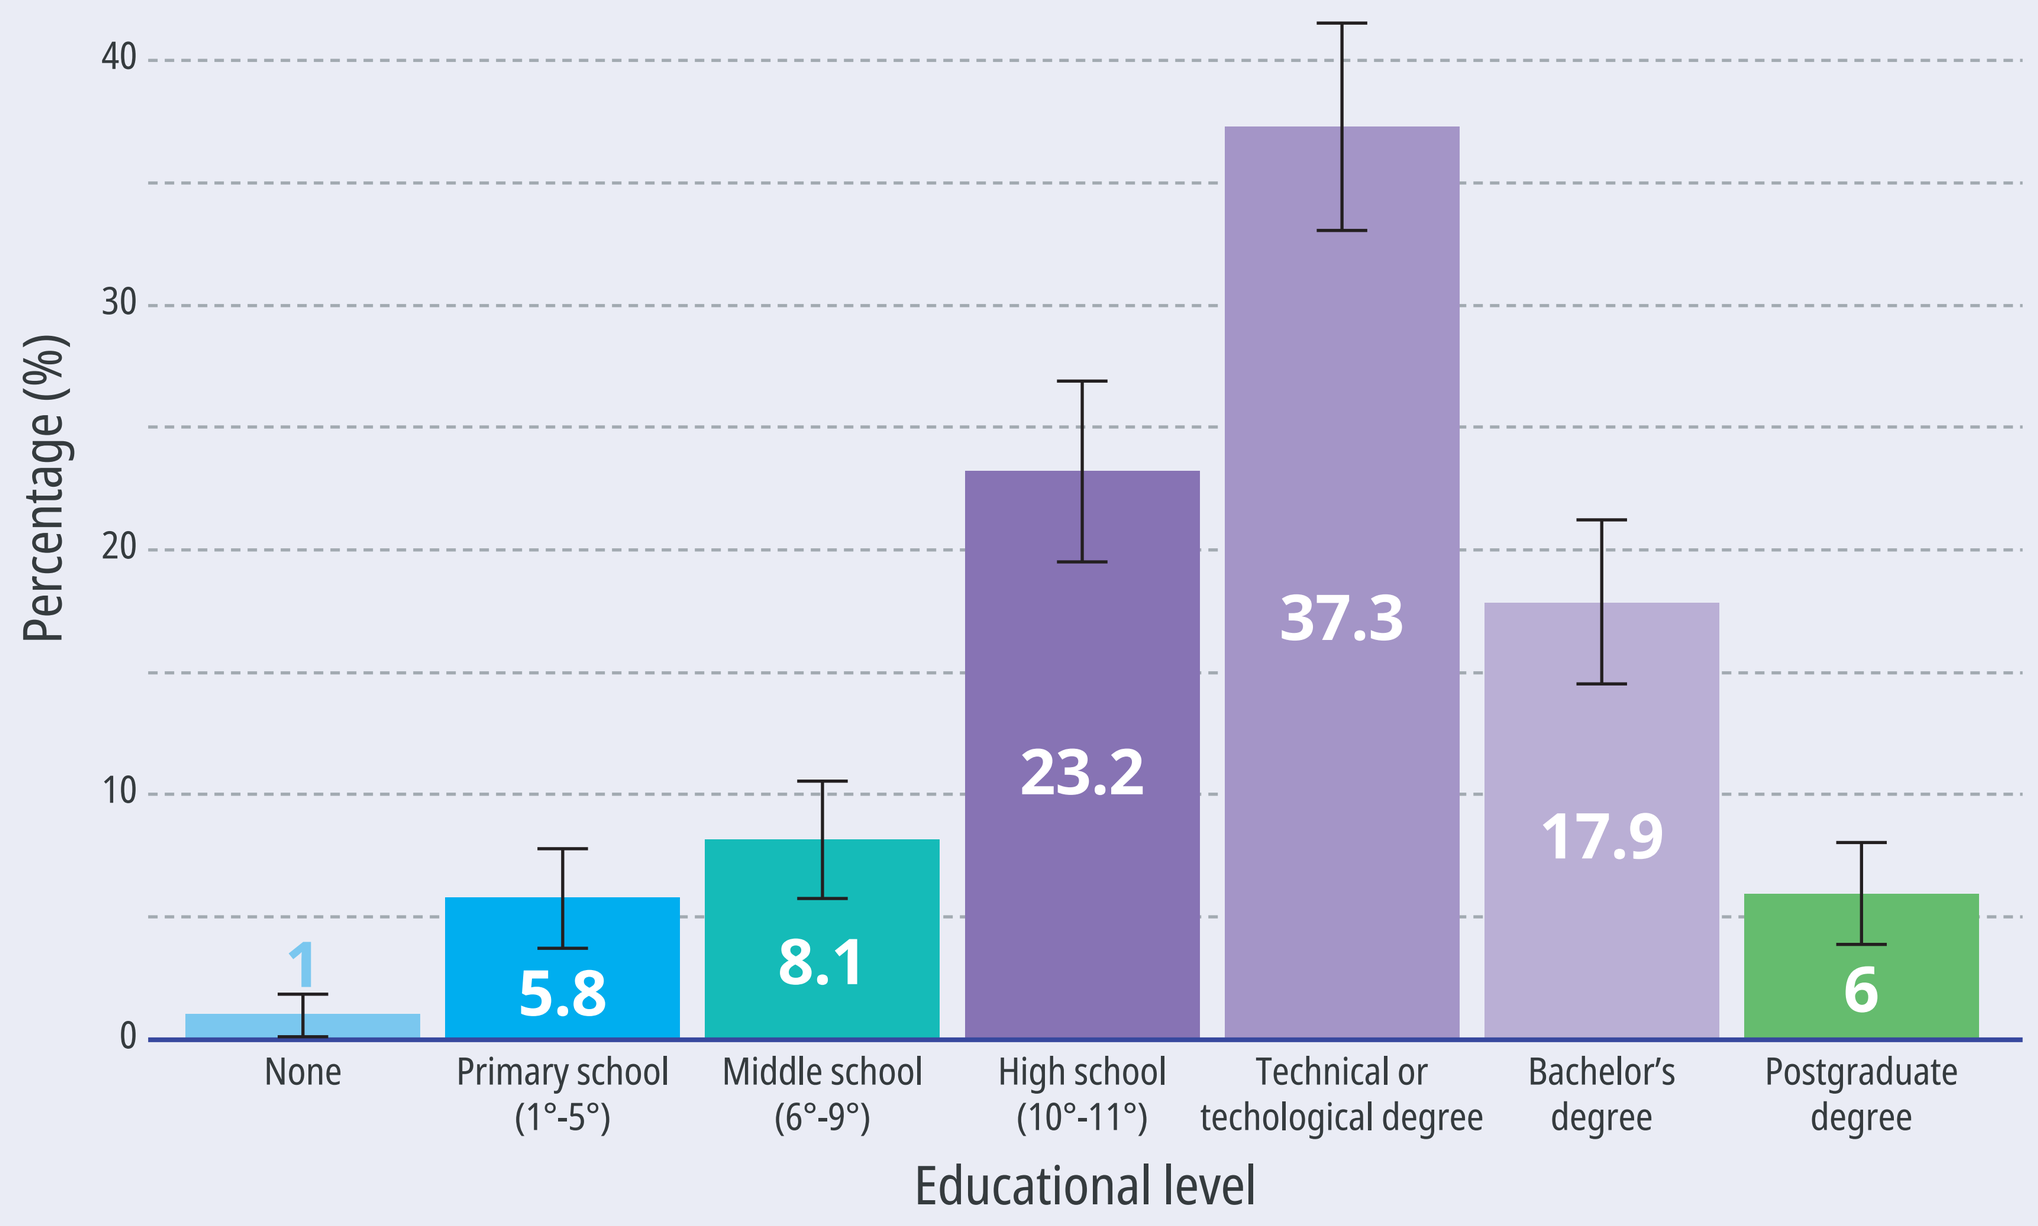
Percentage (%)
40
30
20
10
0
1
5.8
8.1
23.2
37.3
17.9
6
None
Primary school (1°-5°)
Middle school (6°-9°)
High school (10°-11°)
Technical or techological degree
Bachelor's degree
Postgraduate degree
Educational level

- For these patients indicated that working occupied most of their time in the past week (the proportion who said they spent most of their time working was 60.3%, CI95% 56.0%-64.6%), and most of them mentioned that their main source of income was their job (66.3%, CI95% 62.1%-70.4%). Their mean income was USD 414.5 (CI95% 386.1-442.9), which represents 1.1 times the current legal minimum wage, and in average these patients reported to have 1.14 economic dependents.
- The mean weight and height of these patients was159 cm (CI95% 158-160) and 67.6 kg (CI95% 66.4-68.8), respectively, and in average these patients presented 1.44 comorbidities, with mental health conditions (41.7%, CI95% 37.3%-46.0%), cancer (25.4%, CI95% 21.6%-29.2%) and hypertension (19.0%, CI95% 15.6%-22.5%) being the most frequent comorbidities.
- On average these patients were 47.2 months since initial diagnosis (CI95% 42.5-52.0), and they present 10.3 monthly migraine days [MMD] (CI95% 9.5 -11.2) and 7.6 episodes in the last 3 months (CI95% 6.7-8.4) (**Figure 2**).

Figure 2. Violinplots of MMD and migraine episodes in the last quarter

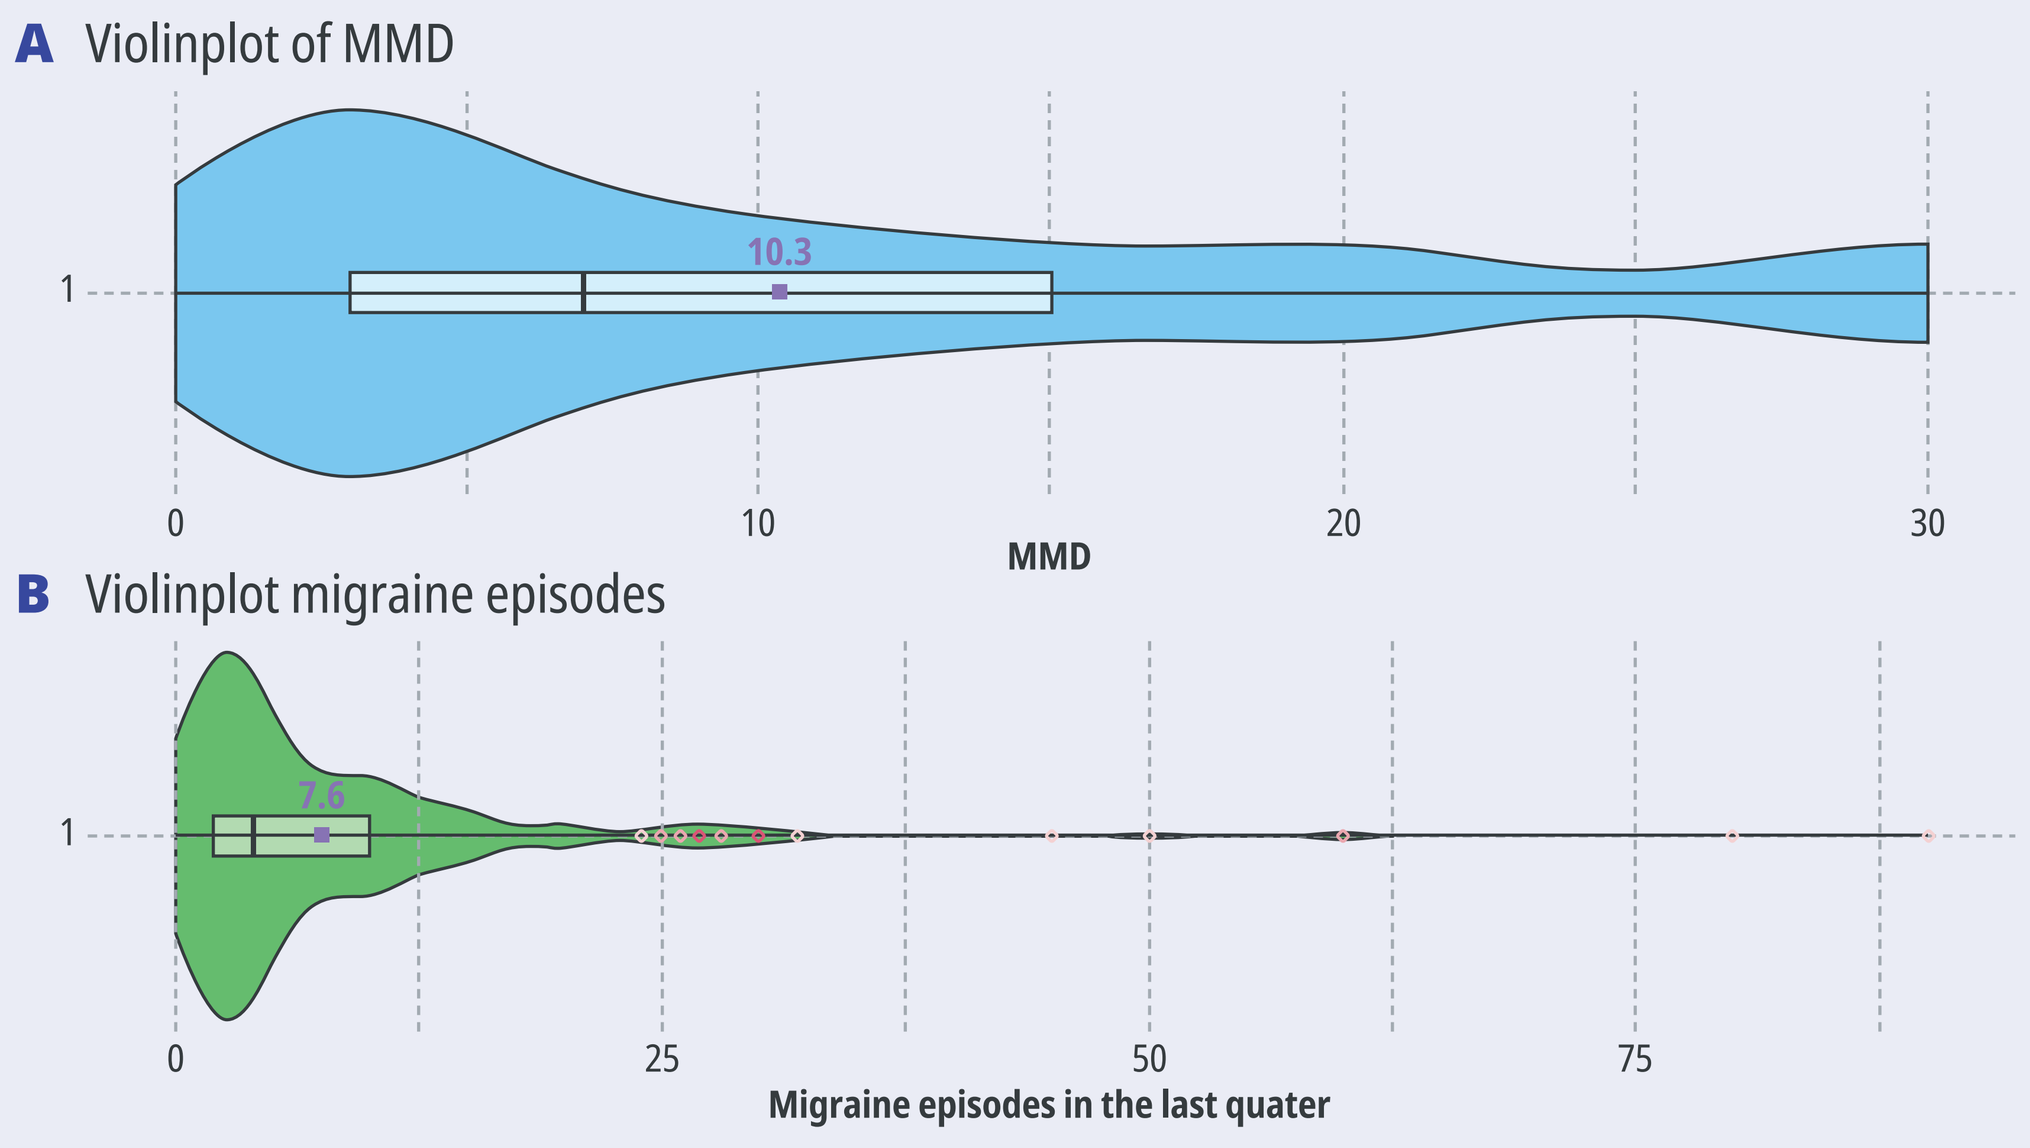
A Violinplot of MMD
1
10.3
0
10
20
30
MMD
B Violinplot migraine episodes
1
7.6
0
25
50
75
Migraine episodes in the last quater

- About 30.2% of this population (CI95% 26.1%-34.2%) mentioned to be very dissatisfied with their current acute drug while 29.0% considered that they were neither satisfied nor dissatisfied with their current preventive treatment (CI95% 25.0%-32.9%) (**Figure 3**).

Figure 3. Barchart of the satisfaction level of current migraine treatments

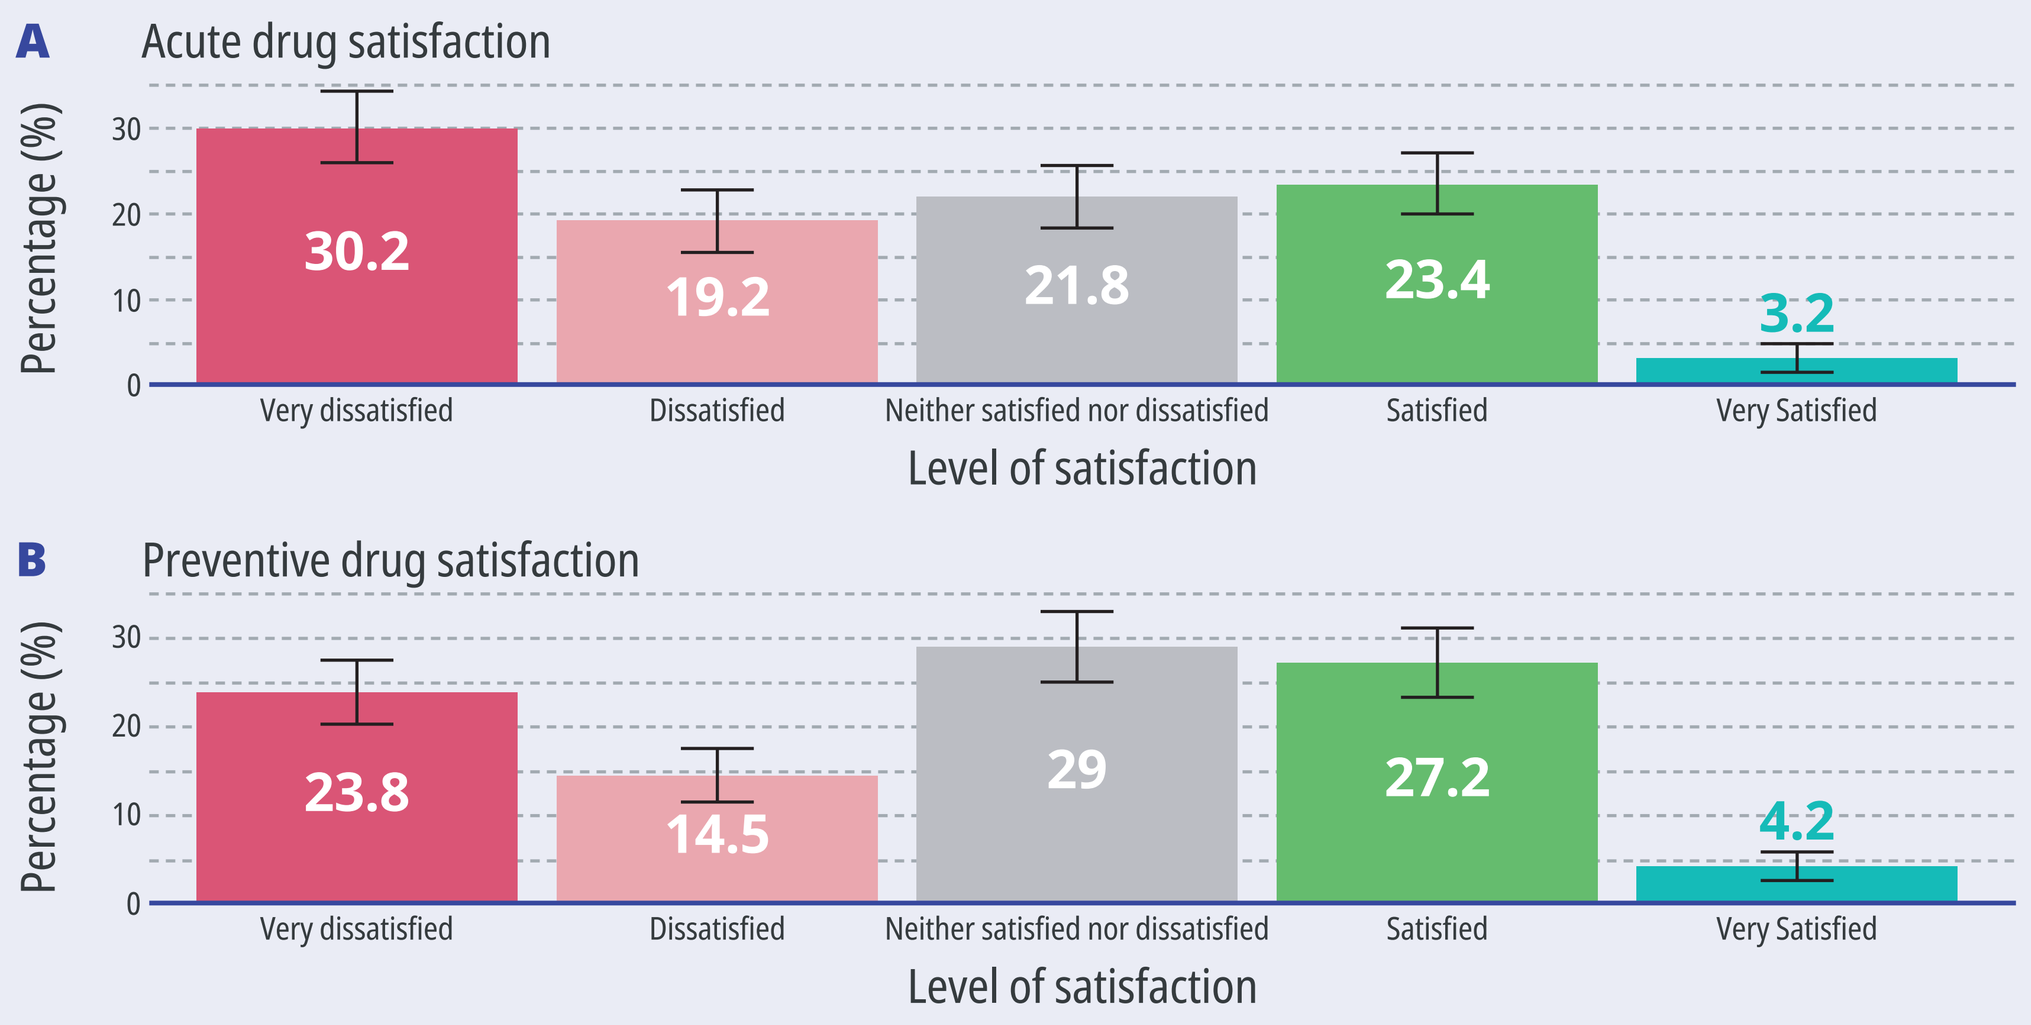
A Acute drug satisfaction
Percentage (%)
30
20
10
0
30.2
19.2
21.8
23.4
3.2
Very dissatisfied
Dissatisfied
Neither satisfied nor dissatisfied
Satisfied
Very Satisfied
Level of satisfaction
B Preventive drug satisfaction
Percentage (%)
30
20
10
0
23.8
14.5
29
27.2
4.2
Very dissatisfied
Dissatisfied
Neither satisfied nor dissatisfied
Satisfied
Very Satisfied
Level of satisfaction

LoP

- The HALT-90 index showed that 40.5% were grade IV or had severe impact on the patient's life and lifestyle indicating a high need for care (CI95% 36.2%-44.8%) (**Figure 4**).

Figure 4. Barchart of HALT-90 index classification

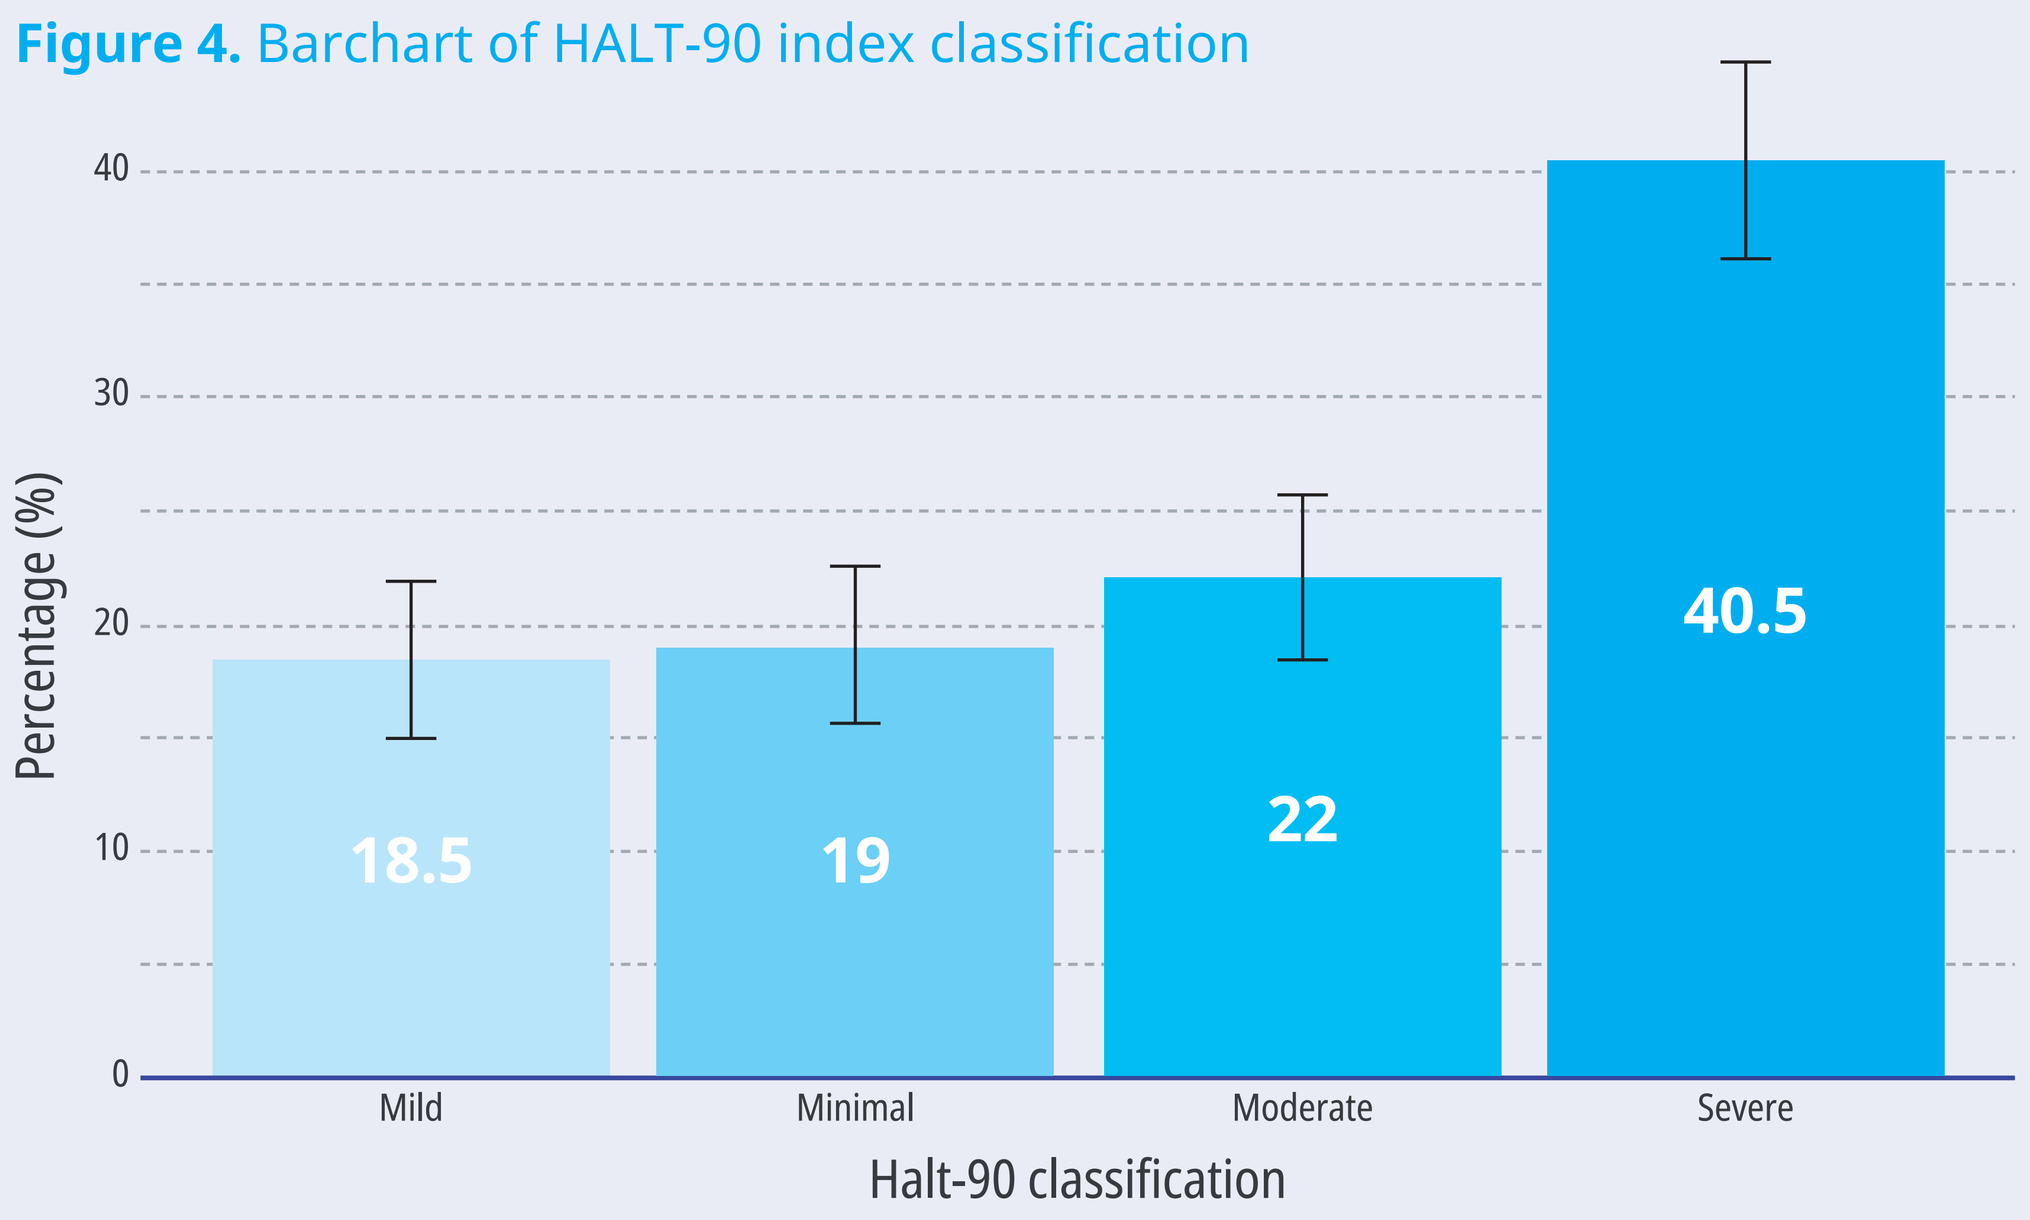
Percentage (%)
40
30
20
10
0
18.5
19
22
40.5
Mild
Minimal
Moderate
Severe
Halt-90 classification

- On average in the last 3 months, the HALT-90 instrument showed that the labor absenteeism was 2.7 days (CI95% 2.2-3.2) per person per quarter and labor presenteeism was 8.7 days (CI95% 7.5-9.9) representing about 12.1% of the 72 working days.
- This LoP represents an indirect cost 101.1 USD (CI95% 84.0-118.2) per quarter per person. In addition, by quantifying non-labor losses (leisure, housework and household chores) it can be argued that USD 152.4 (CI95% 123.2- 181.7) could be lost (**Figure 5**).

Figure 5. Barchart of indirect cost by type

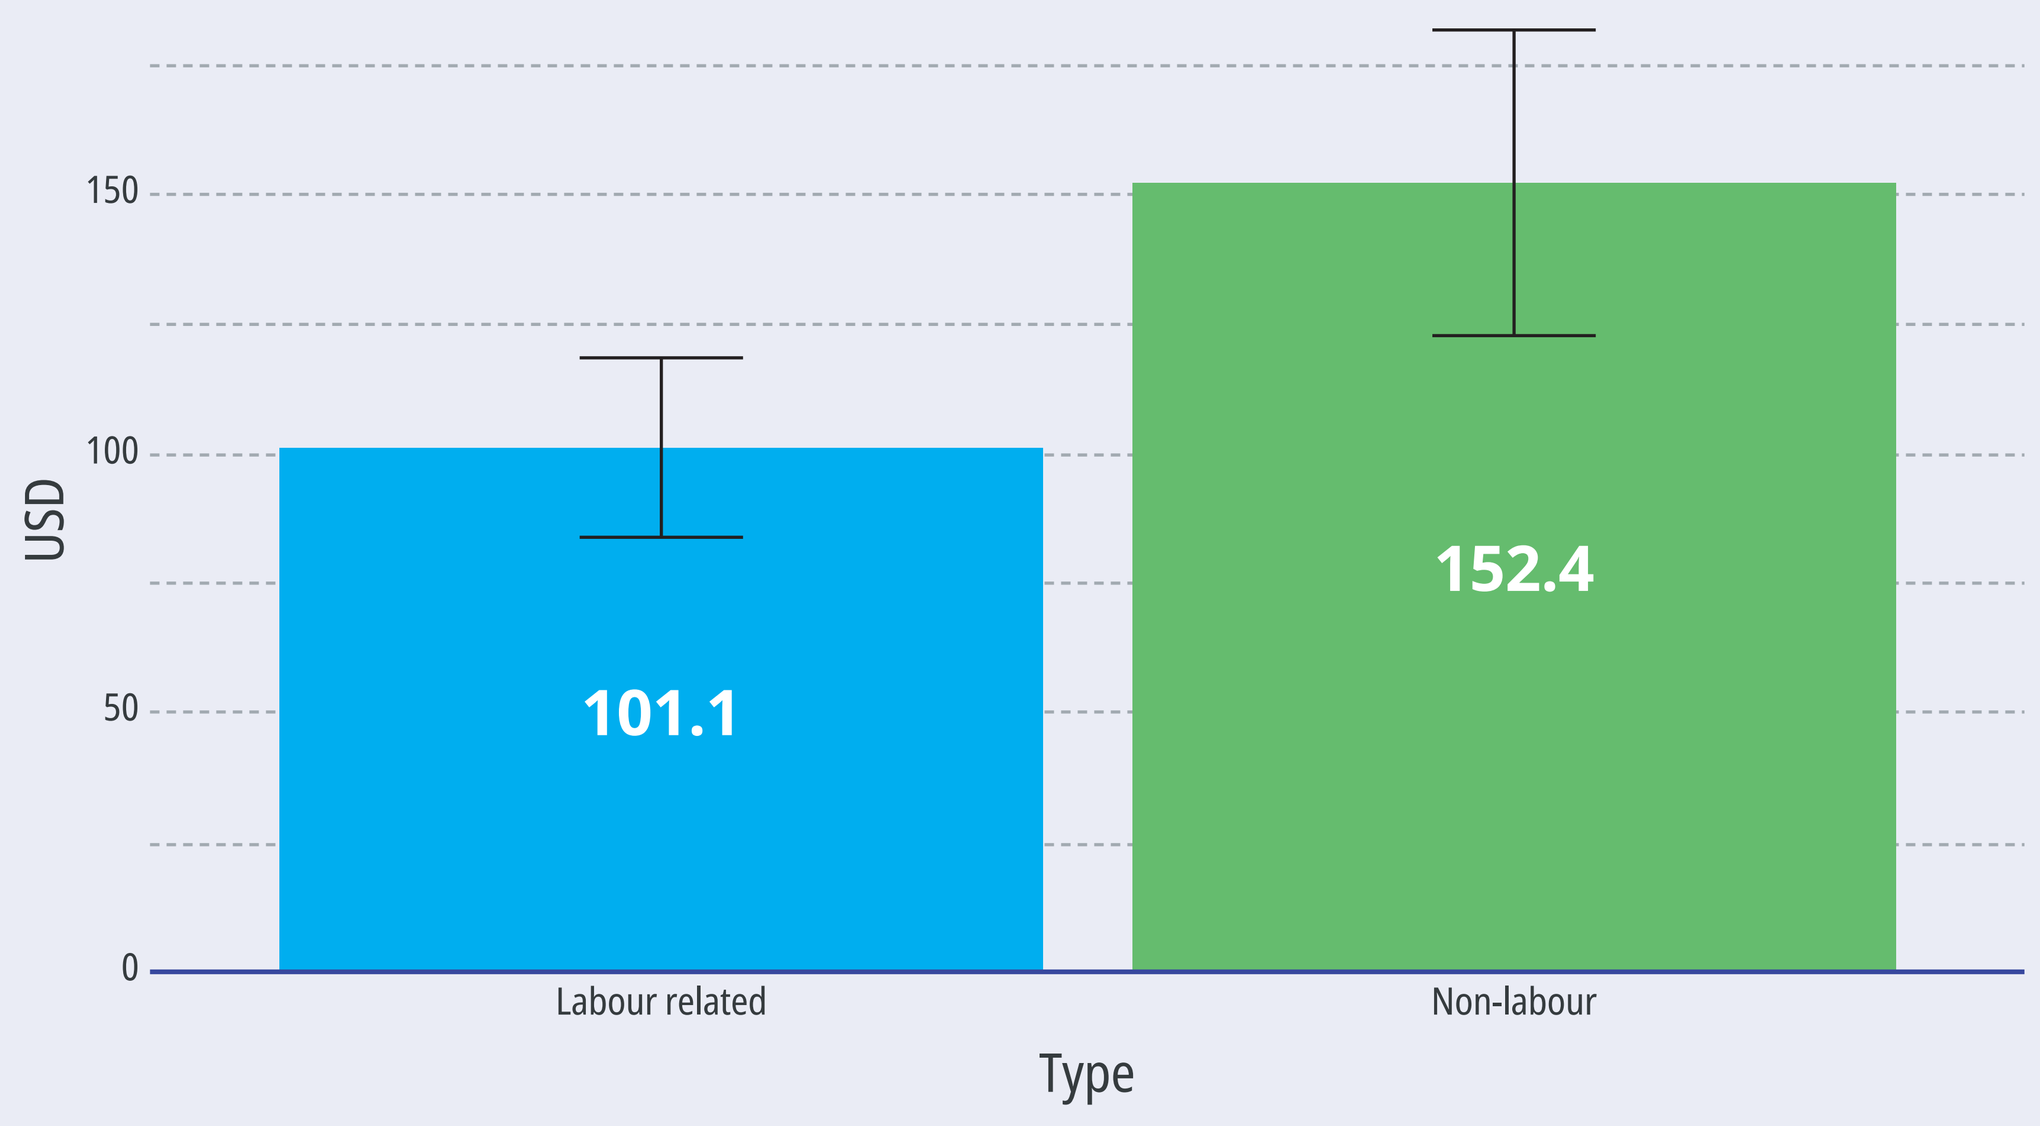
USD
150
100
50
0
101.1
152.4
Labour related
Non-labour
Type

QoL

- The QoL perception were in average 49.1 out of 100 (CI95% 47.4-50.8) for the general health score. The main domains related to this QoL were the physical domain 51.4 (CI95% 50.0-52.9), and the social relationships' domain 55.7 (CI95% 53.9-57.5) (**Figure 6**). These results indicate a low QoL when compared to findings by other authors who applied this instrument to the general population, where scores tend to exceed 61 out of 100[8,9].

Figure 6. QoL of highly consultative migraine patients

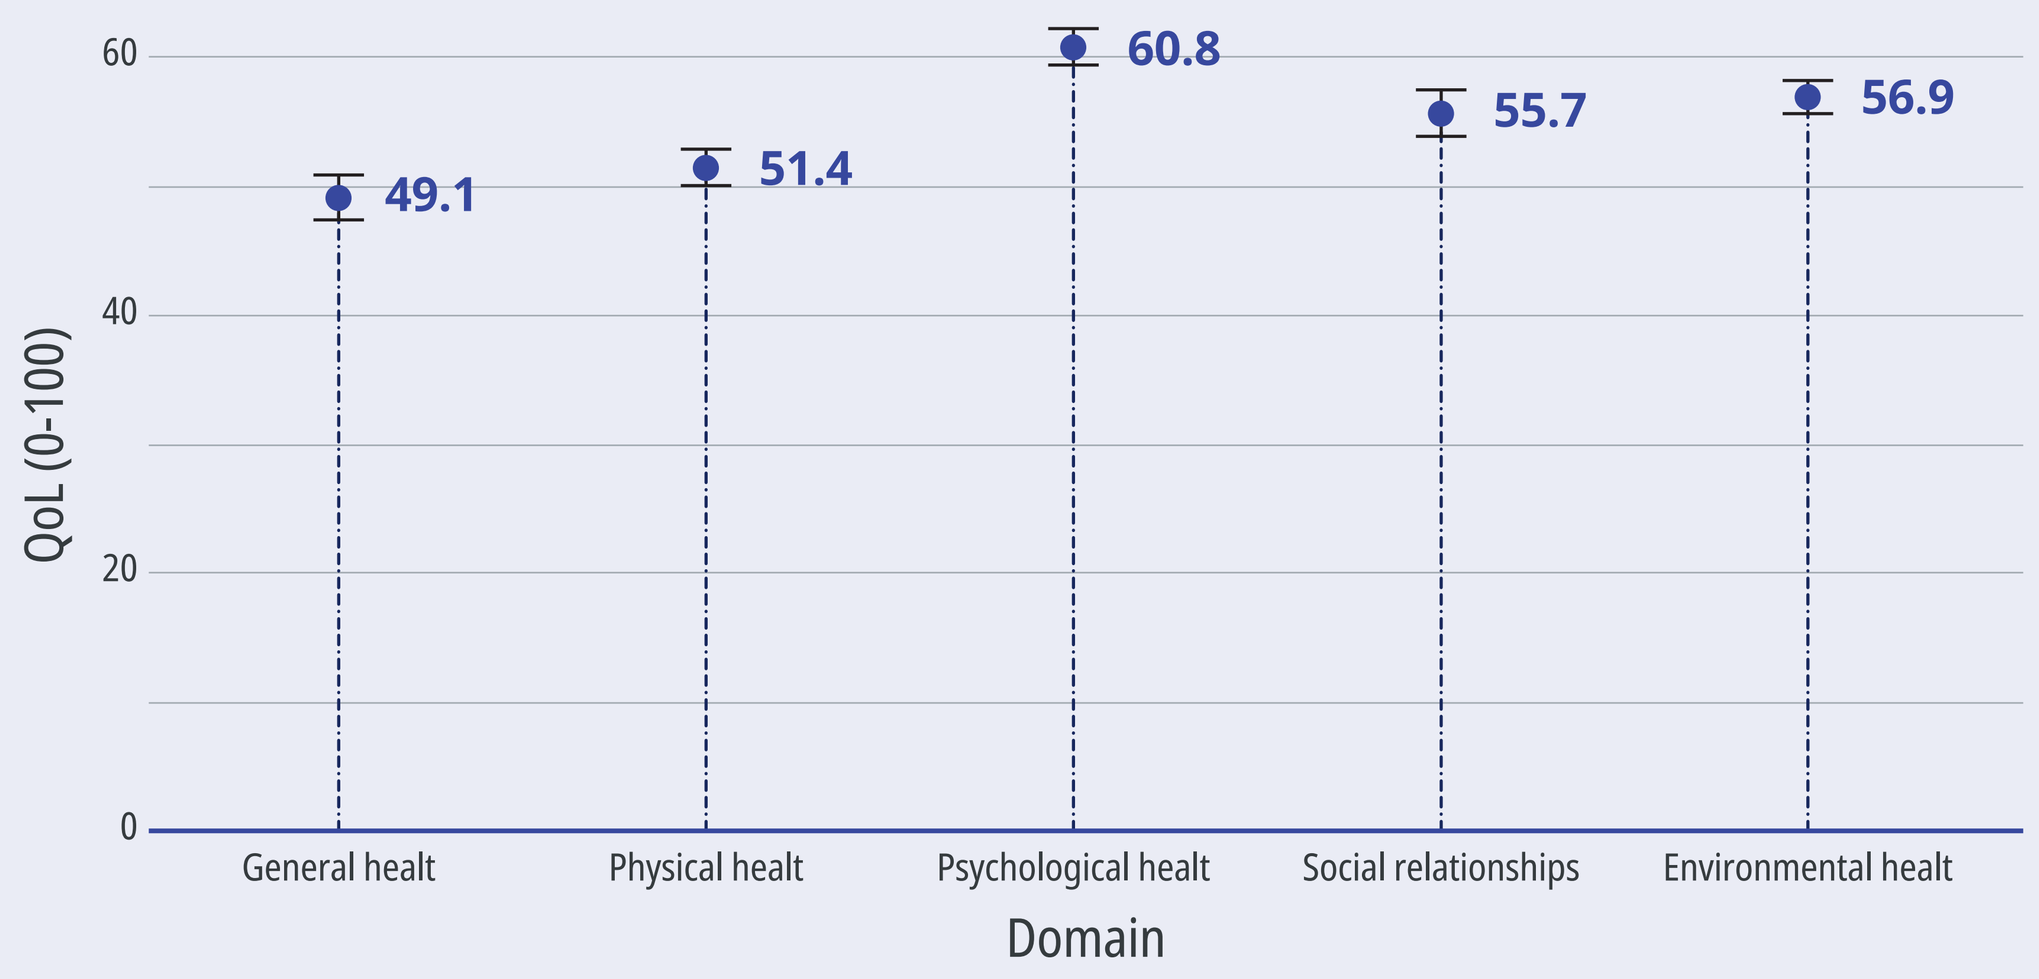
QoL (0-100)
60
40
20
0
49.1
51.4
60.8
55.7
56.9
General healt
Physical healt
Psychological healt
Social relationships
Environmental healt
Domain

*It refers to the regime in which individuals with the capacity to make contributions to the health system in Colombia are included.

CONCLUSION

- Highly consultative migraine adult patients at the HMO were middle-aged, female, highly educated workers, with economic dependents and high comorbidities, particularly mental health conditions. This population is associated with severe headache impact, with substantial incidence on productivity losses and impact on quality of life.

REFERENCES

1. Nguyen JL, Munshi K, Peasah SK, Swart ECS, Kohli M, Henderson R, et al. Trends in utilization and costs of migraine medications, 2017–2020. J Headache Pain. 2022 Dec;23(1):111. **2.** Baigi K, Stewart WF. Chapter 25 - Headache and migraine: a leading cause of absenteeism. In: Lotti M, Bleecker ML, editors. Handbook of Clinical Neurology [Internet]. Elsevier; 2015 [cited 2023 Apr 26]. p. 447–63. (Occupational Neurology; vol. 131). Available from: https://www.sciencedirect.com/science/article/pii/B9780444626271000251. **3.** Bolaños J, Arciniegas J, Rubio A, Gonzalez F, Mesa A, Bello C, et al. Treatment patterns and healthcare resource utilization in Colombian migraine patients from 2018 to 2022 [Internet]. ISPOR international 2024; 2024 [cited 2024 Sep 1]; Atlanta. Available from: https://www.ispor.org/docs/default-source/intl2024/ispor24bolanoshsd11poster139860-pdf.pdf?sfvrsn=b1fede83_0. **4.** Lifting The Burden: The Global Campaign against Headache (n.d.). Headache-Attributed Lost Time, HALT 90 Index. Accessed on July 25: https://www.l-t-b.org/assets/30/A8830A81-035C-48C7-08E2AD8D70603F82_document/HALT.pdf. Lifting The Burden does not endorse any specific companies, products or services. **5.** Reproduced with permission from WHOQOL: Measuring Quality of Life, Geneva, World Health Organization (WHO), (2020), https://www.who.int/tools/whoqol/whoqol-bref/docs/default-source/publishing-policies/whoqol-bref/spanish-colombian-whoqol-bref, accessed: 17/08/2023. WHO does not endorse any specific companies, products or services. Translated into spanish from WHOQOL: Measuring Quality of Life, Geneva, World Health Organization (WHO), (2020), https://www.who.int/tools/whoqol/whoqol-bref/docs/default-source/publishing-policies/whoqol-bref/spanish-colombian-whoqol-bref, accessed: 17/08/2023. WHO is not responsible for the content or accuracy of this translation/adaptation. In the event of any inconsistency between the English and the insert language translation, the original English version shall be the binding and authentic version. Product Delivery and updates: The Licensed Materials and validated updates are available on WHO web site. WHO does not undertake to supply. **6.** Lifting The Burden. The Headache-Attributed Lost Time (HALT) Indices: measures of burden for headache management in primary care [Internet]. [cited 2024 Sep 28]. Available from: https://migraine-acute-treatment.ime.springerhealthcare.com/wp-content/uploads/EN-Guide.pdf. **7.** Skevington SM, Lotfy M, O'Connell KA. The World Health Organization's WHOQOL-BREF quality of life assessment: Psychometric properties and results of the international field trial. A Report from the WHOQOL Group. Qual Life Res. 2004 Mar;13(2):299–310. **8.** Bat-Erdene E, Hiramoto T, Tumurbaatar E, Tumur-Ochir G, Jamiyandorj O, Yamamoto E, et al. Quality of life in the general population of Mongolia: Normative data on WHOQOL-BREF. Nakhostin-Ansari A, editor. PLOS ONE. 2023 Sep 29;18(9):e0291427. **9.** Wong FY, Yang L, Yuen JWM, Chang KKP, Wong FKY. Assessing quality of life using WHOQOL-BREF: a cross-sectional study on the association between quality of life and neighborhood environmental satisfaction, and the mediating effect of health-related behaviors. BMC Public Health. 2018 Dec;18(1):1113.

Financial statement: This study was funded by Pfizer. Poster presented at ISPOR Europe 2024, Barcelona, Spain, 17-20 November 2024