A cost-effectiveness analysis of dapagliflozin in heart failure with preserved or mildly reduced ejection fraction in Portugal

André Andrade[1], Rita Sousa[1], Ana Lúcia Tavares[1]

AstraZeneca Produtos Farmacêuticos, Lda., Portugal[1]

EE14

Background and Objective

- Heart failure (HF) is a leading cause of hospitalization and mortality worldwide, affecting 1% to 3% of the general adult population. [1] HF is associated with reduced quality of life, high morbidity, mortality, and significant excess financial expenditure.
- Dapagliflozin is a sodium-glucose co-transporter-2 (SGLT2) inhibitor, which was originally developed as a glucose-lowering agent for the treatment of type 2 diabetes mellitus, that reduces the risk of death and other adverse outcomes among patients with chronic heart failure and a reduced ejection fraction (i.e., a left ventricular ejection fraction of ≤ 40%) and in those with chronic kidney disease, regardless of the presence or absence of type 2 diabetes mellitus. [2]
- The DELIVER clinical trial is an international, multicentre, double-blind, randomized placebo-controlled Phase III trial aimed to evaluate the effect of dapagliflozin versus placebo in reducing the composite of cardiovascular (CV) death or HF events [Hospitalization for HF (HHF) or Urgent HF visit (UHFV)] in patients with chronic HF and an ejection fraction >40%. The study concluded that dapagliflozin is associated with statistically significant reductions in the primary composite of CV death and HF events (HR: 0.82; 95% CI: 0.73-0.92; p<0.001) and for HHF individually (HR: 0.79; 95% CI: 0.69-0.91). [2]
- The aim of this analysis was to assess the cost-effectiveness of dapagliflozin added to standard-of-care therapy (SOC), versus SOC only, in patients with heart failure with preserved (>50%) or mildly reduced (40% - 50%) ejection fraction (HFpEF/HFmrEF), in the Portuguese setting.

Methods

Effectiveness model

- A lifetime Markov state-transition model was developed by HEOR Ltd., to quantify the long-term health economic impact of dapagliflozin plus SOC versus SOC in the treatment of patients with HF. **(Figure 1)**

Figure 1. Model Structure

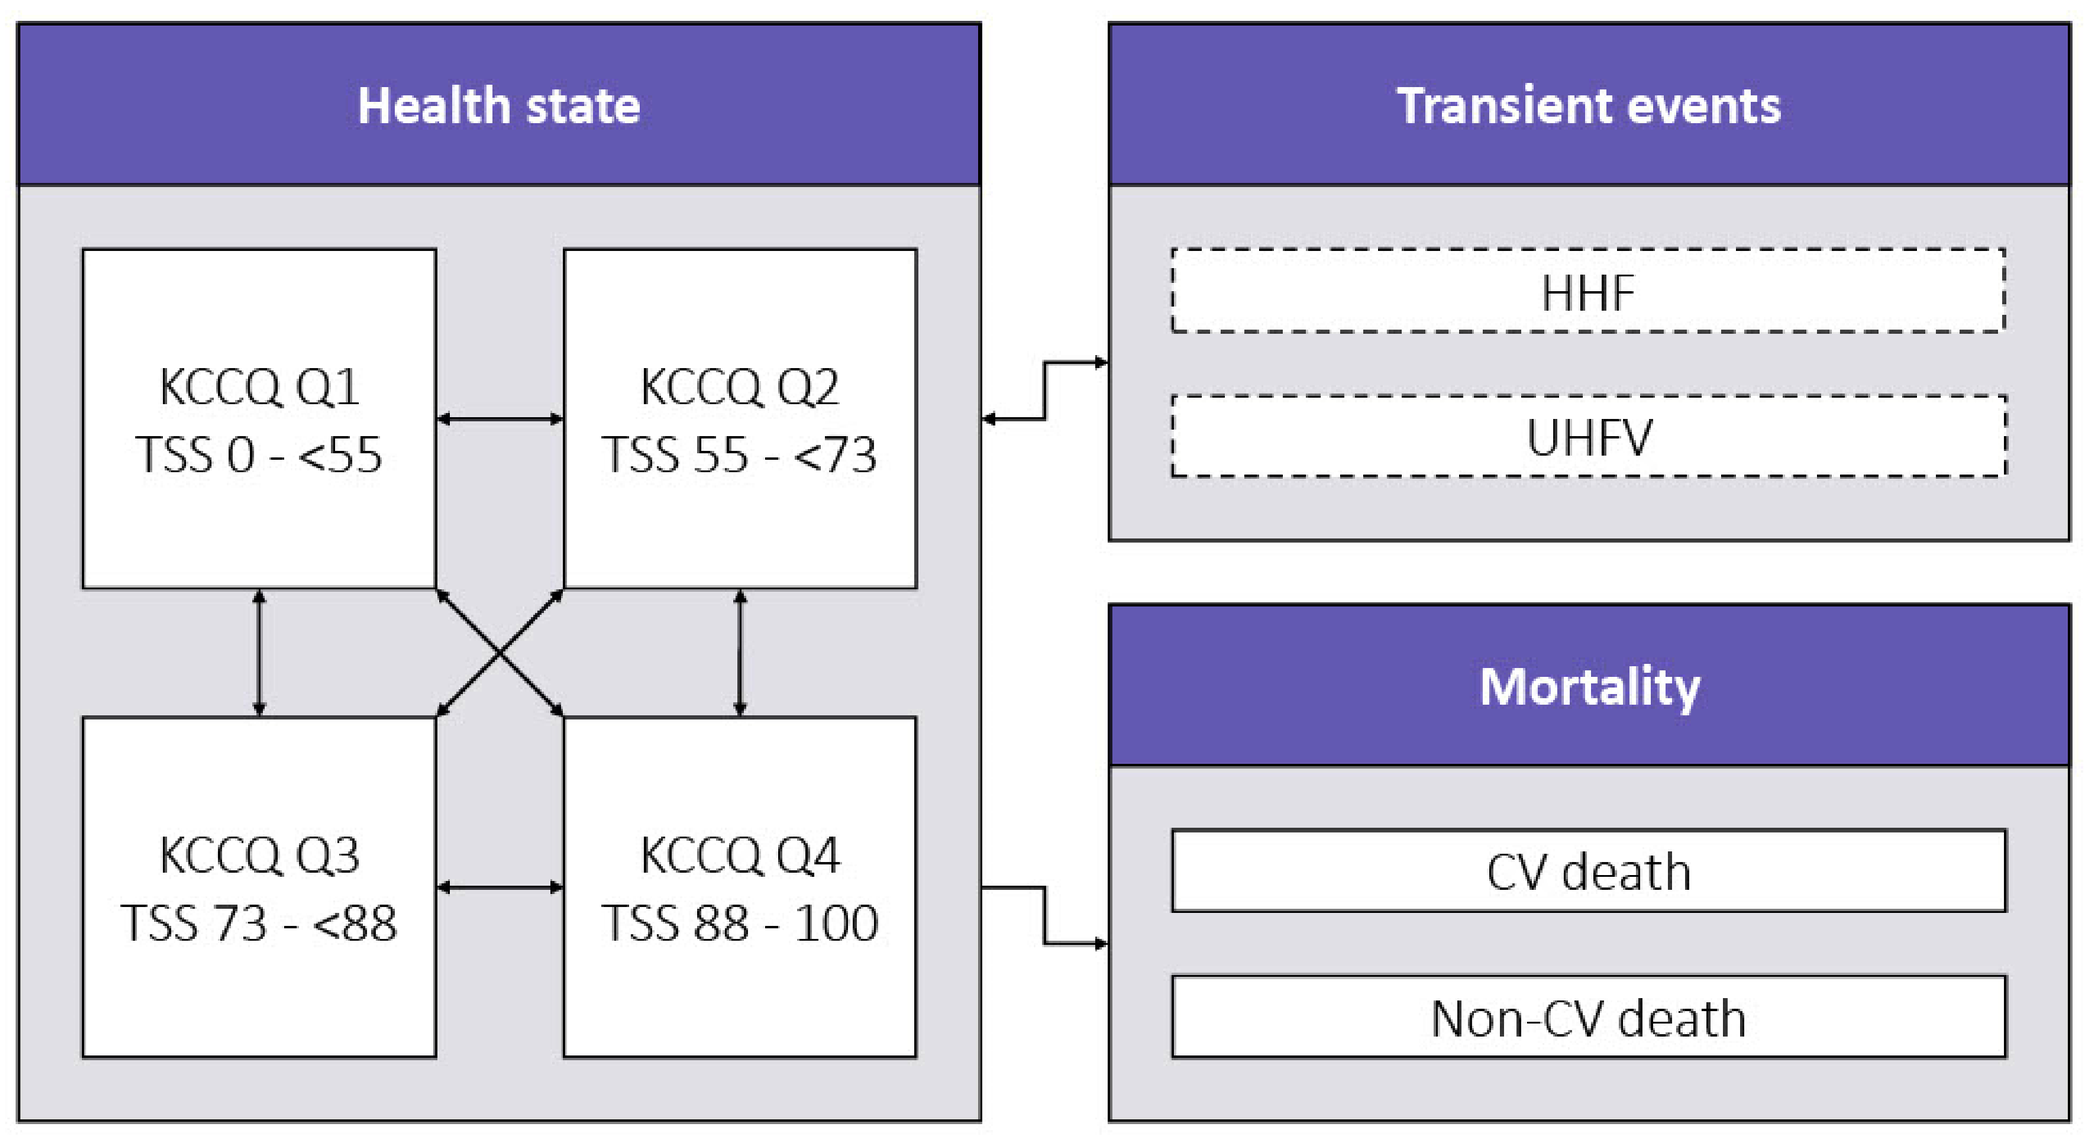
Health state
KCCQ Q1
TSS 0 - <55
KCCQ Q2
TSS 55 - <73
KCCQ Q3
TSS 73 - <88
KCCQ Q4
TSS 88 - 100
Transient events
HHF
UHFV
Mortality
CV death
Non-CV death

- Disease progression is captured through transitions between discrete health states characterized by the quartiles of the total symptom score (TSS) of the Kansas City Cardiomyopathy Questionnaire (KCCQ) calculated across all patients at baseline, independent of treatment arm. The KCCQ-TSS quantifies patients' symptom frequency and severity.
- In addition to the health states, the model captures the incidence of HHF and UHFV (transient events), where patients incur in additional event-specific cost and utility decrement only in the cycle of incidence.
- Patient mortality is captured through the application of parametric survival equations describing CV death and non-CV death, calculated as the difference between all-cause death and CV death.
- The baseline patient characteristics are derived from the DELIVER clinical trial and used to determine the cohort's initial health state distribution across KCCQ-TSS quartiles, as well as inform the adjusted risk equations and survival equations.
- The analysis was conducted from the Portuguese National Health Service perspective assuming a 4% discount rate for costs and consequences and a lifetime time horizon.

Adverse Events (AEs)

- The serious adverse events with a frequency > 1% of the trial population (acute kidney injury, fracture, urinary tract infection, and volume depletion) or of special clinical interest (amputation) were included in the model.

Utilities

- Modelled utility values were derived from the analysis of individual patient data from the DELIVER clinical trial assigned to KCCQ-TSS quartile health state, mapped using the Portuguese value sets [3] and used in the model.

Costs

- The model included costs associated with health state resources use, transient events, treatment, and adverse events, which were obtained from Portuguese databases and literature.

Results

Basecase scenario

- The modelled time in each health state by treatment arm are presented in **Figure 2**, and cost-effectiveness results for the base-case scenario are presented in **Table 1**.

Figure 2. Modelled time in each health state by treatment arm

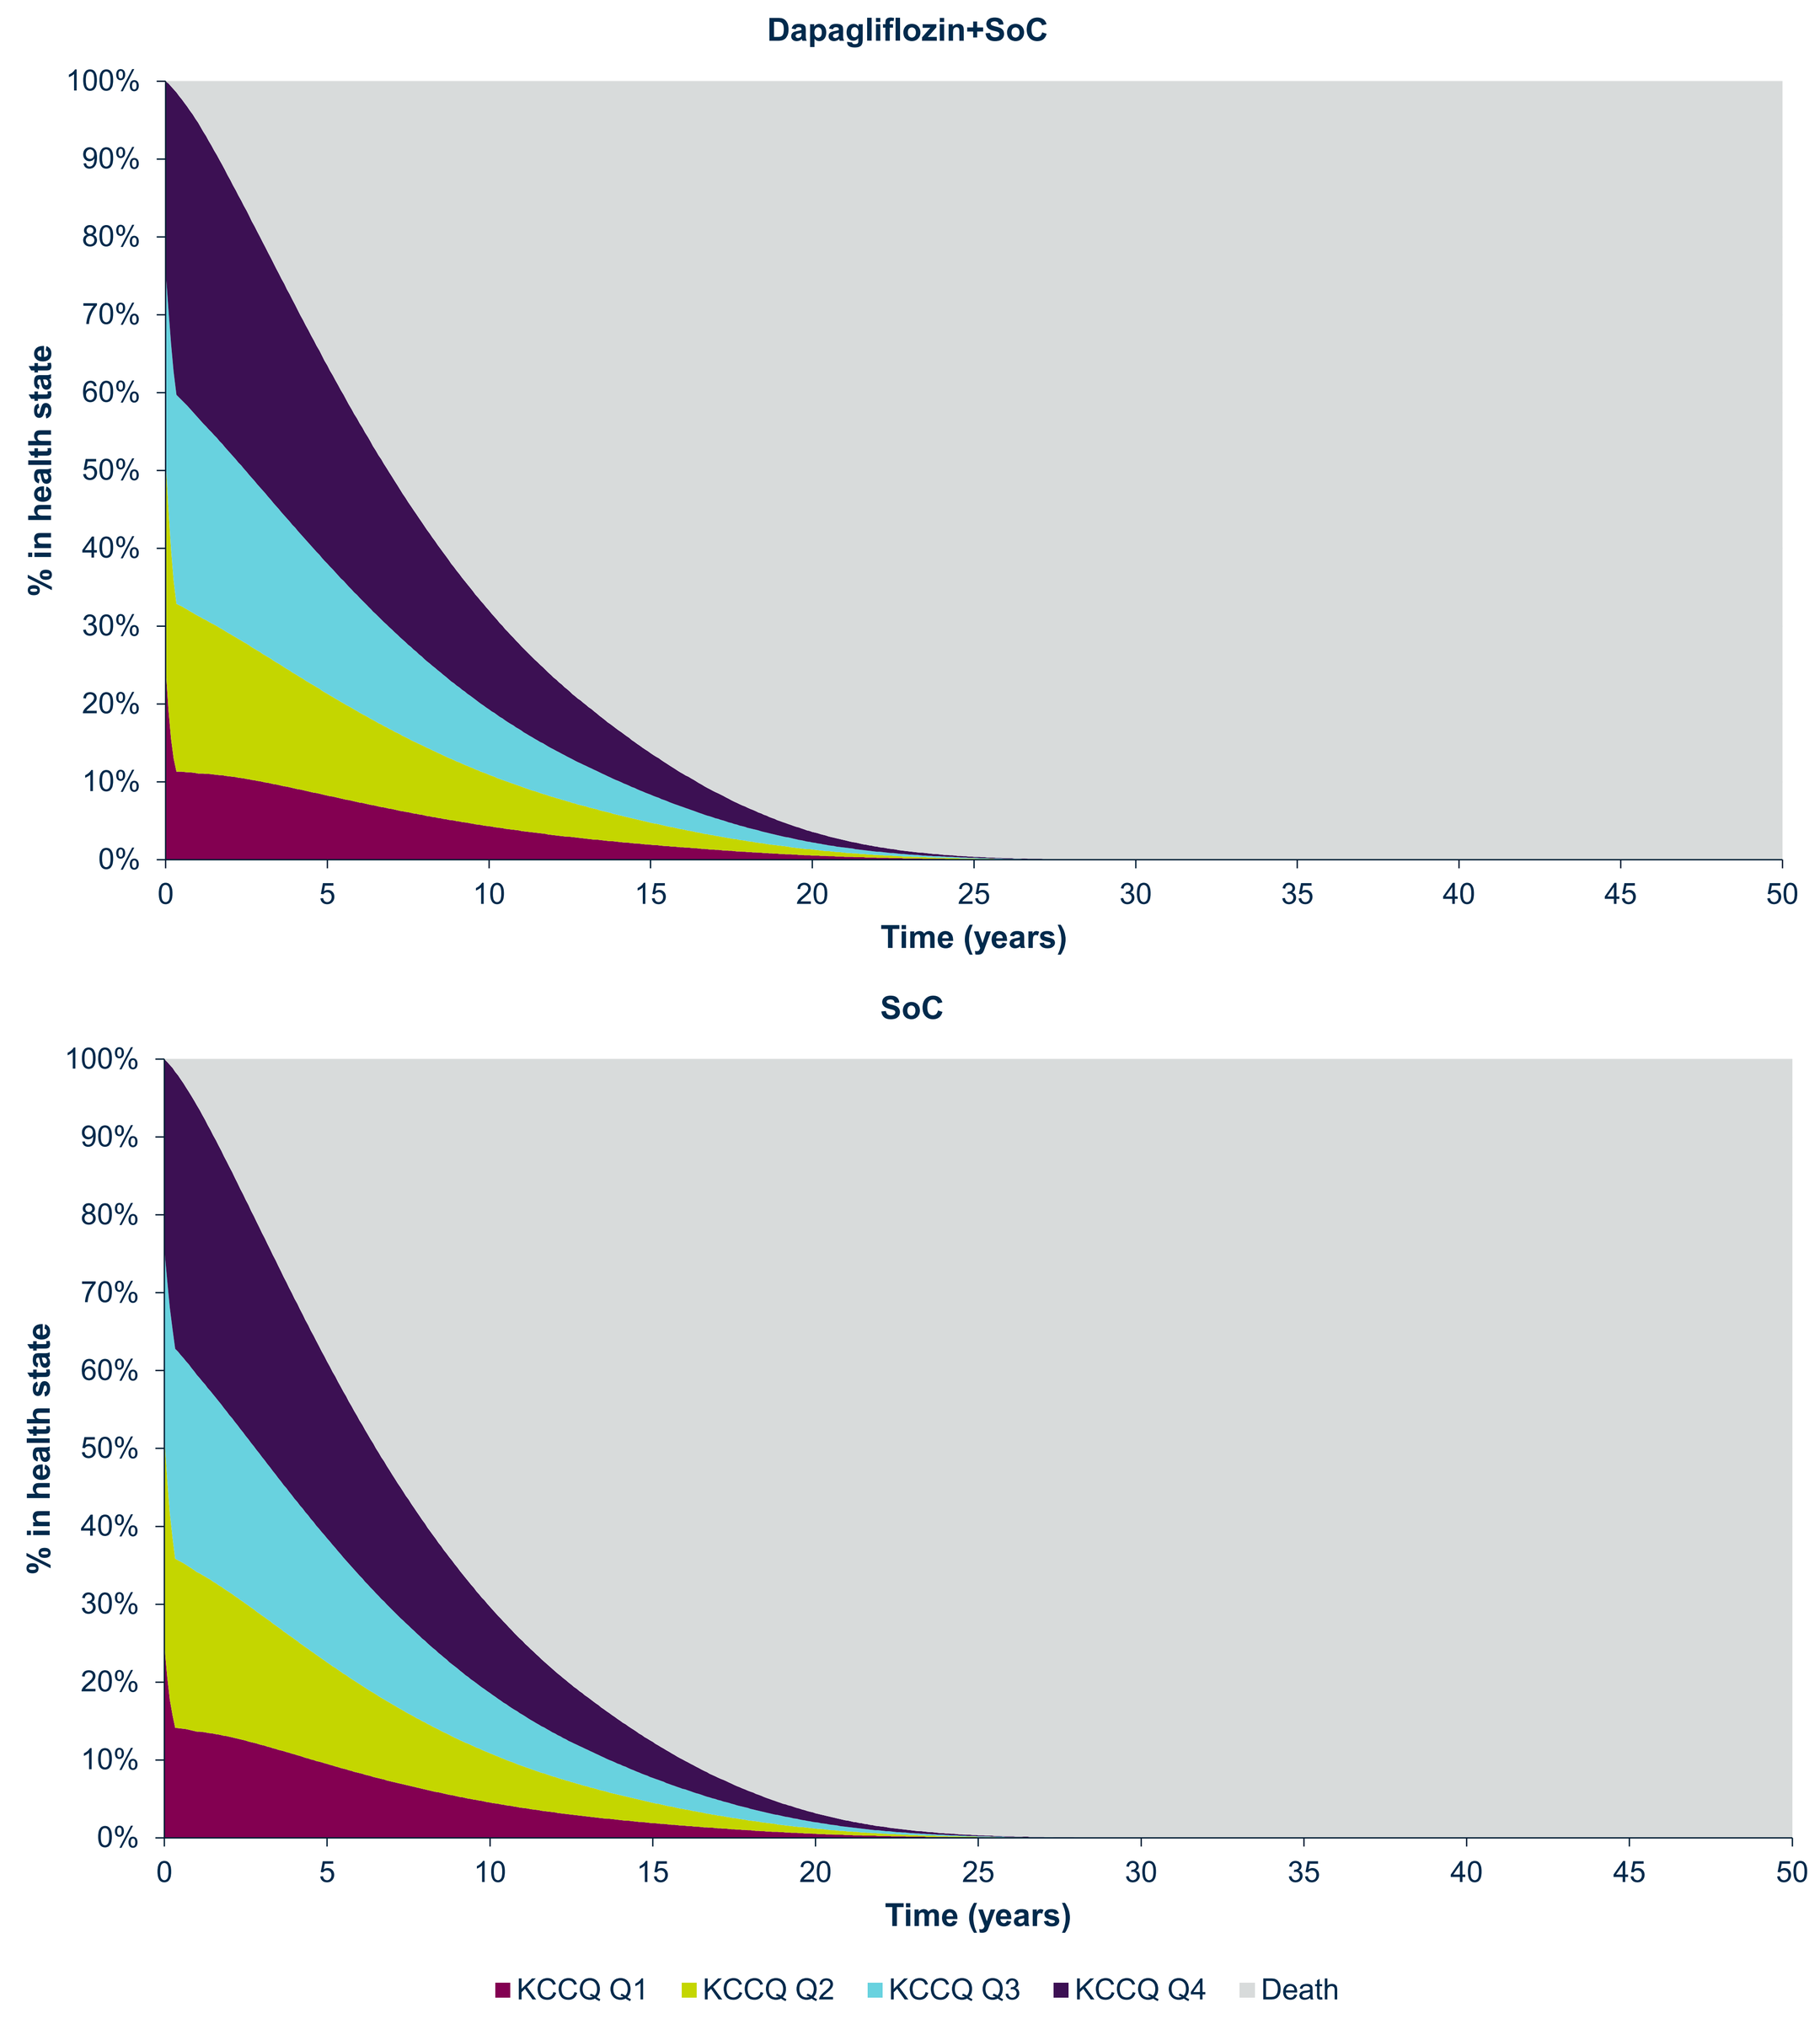
Dapagliflozin+SoC
SoC
% in health state
Time (years)
KCCQ Q1
KCCQ Q2
KCCQ Q3
KCCQ Q4
Death

Table 1. Cost-effectiveness results for the base-case scenario.
LY, QALY and Costs break down by category

	Dapagliflozin	SOC	Incremental
Health outcomes - life years (LY)			
KCCQ Q1	0.830	0.949	-0.119
KCCQ Q2	1.350	1.336	0.014
KCCQ Q3	1.702	1.629	0.073
KCCQ Q4	2.549	2.282	0.267
Total	**6.431**	**6.197**	**0.234**
Health outcomes - quality-adjusted life years (QALY)			
KCCQ Q1	0.564	0.645	-0.081
KCCQ Q2	1.064	1.053	0.011
KCCQ Q3	1.345	1.287	0.057
KCCQ Q4	2.014	1.803	0.211
Transient events	0.009	0.011	-0.002
Adverse events	-0.022	-0.026	0.003
Total	**4.973**	**4.774**	**0.200**
Cost outcomes (€)			
Health states	2,719.30 €	2,620.25 €	99.05 €
Transient events	1,162.48 €	1,447.08 €	-284.60 €
Death	1,026.66 €	1,105.46 €	-78.80 €
Treatment	3,814.51 €	1,733.16 €	2,081.35 €
Adverse events	531.74 €	644.93 €	-113.19 €
Total	**€9,255**	**€7,551**	**€1,704**

- Dapagliflozin plus SOC was more effective than SOC alone, leading to a mean 0.234 additional LYs (6.431 versus 6.197) and a mean 0.200 additional QALY (4.973 versus 4.774), per patient.
- Dapagliflozin plus SOC versus SOC alone was associated with incremental cost-effectiveness ratios (ICER) of 7,273.39€/LY and 8,539.98€/QALY (**Figure 3**).

Sensitivity analysis

- Model robustness was demonstrated through scenario analyses.
- The probabilistic sensitivity analysis (PSA) also showed robust results with probabilistic ICERs of 7,220.18 €/LY and 8,495.03 €/QALY (**Figure 3**).
- The cost-effectiveness acceptability curve shows that, at a willingness-to-pay threshold of 20,000€/QALY, dapagliflozin has a probability of cost-effectiveness of approximately 87% (**Figure 4**).

Figure 3. Cost-effectiveness plan

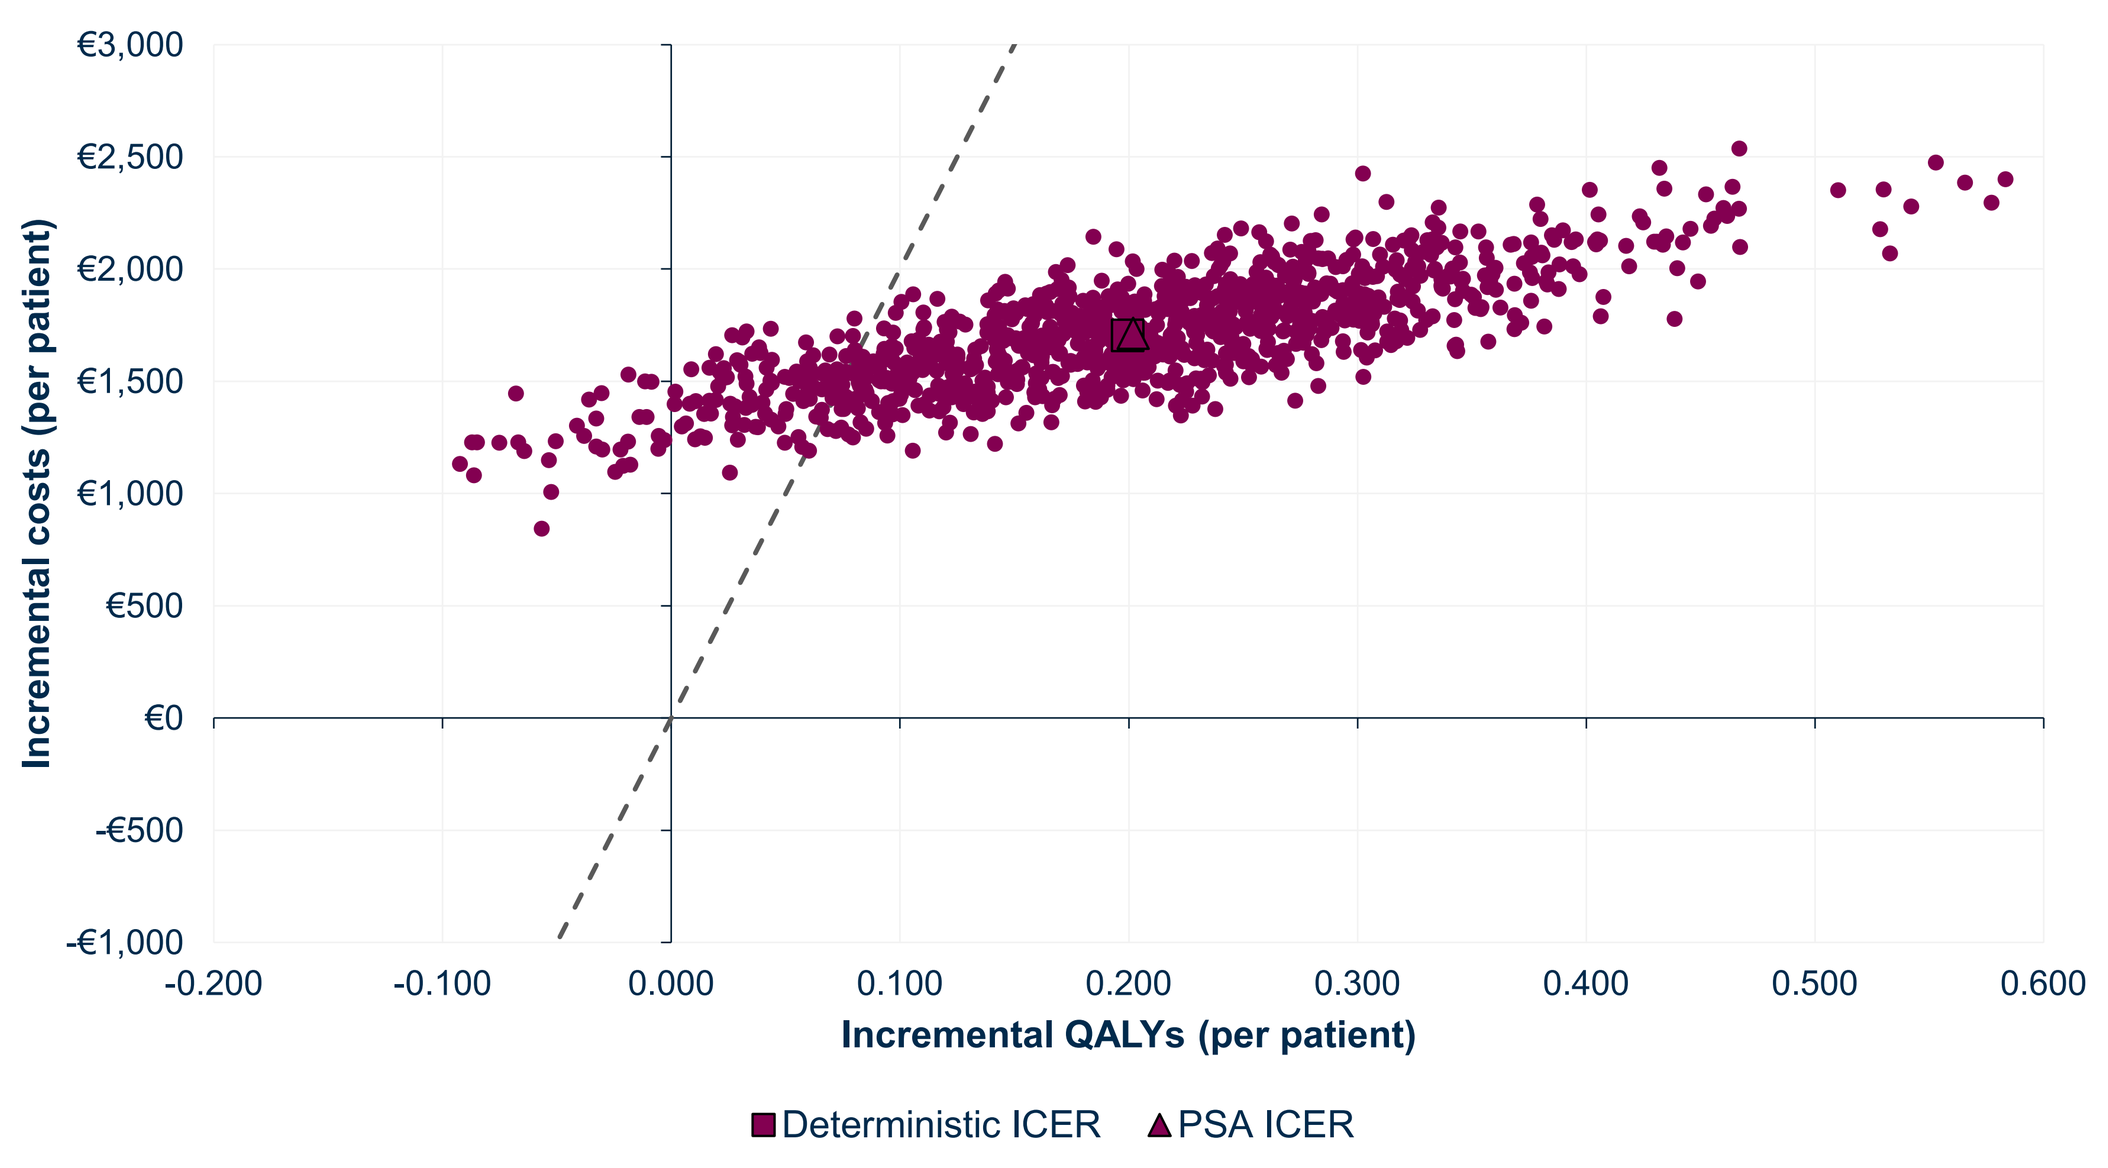
Incremental costs (per patient)
Incremental QALYs (per patient)
Deterministic ICER
PSA ICER

Figure 4. Cost-effectiveness acceptability curve

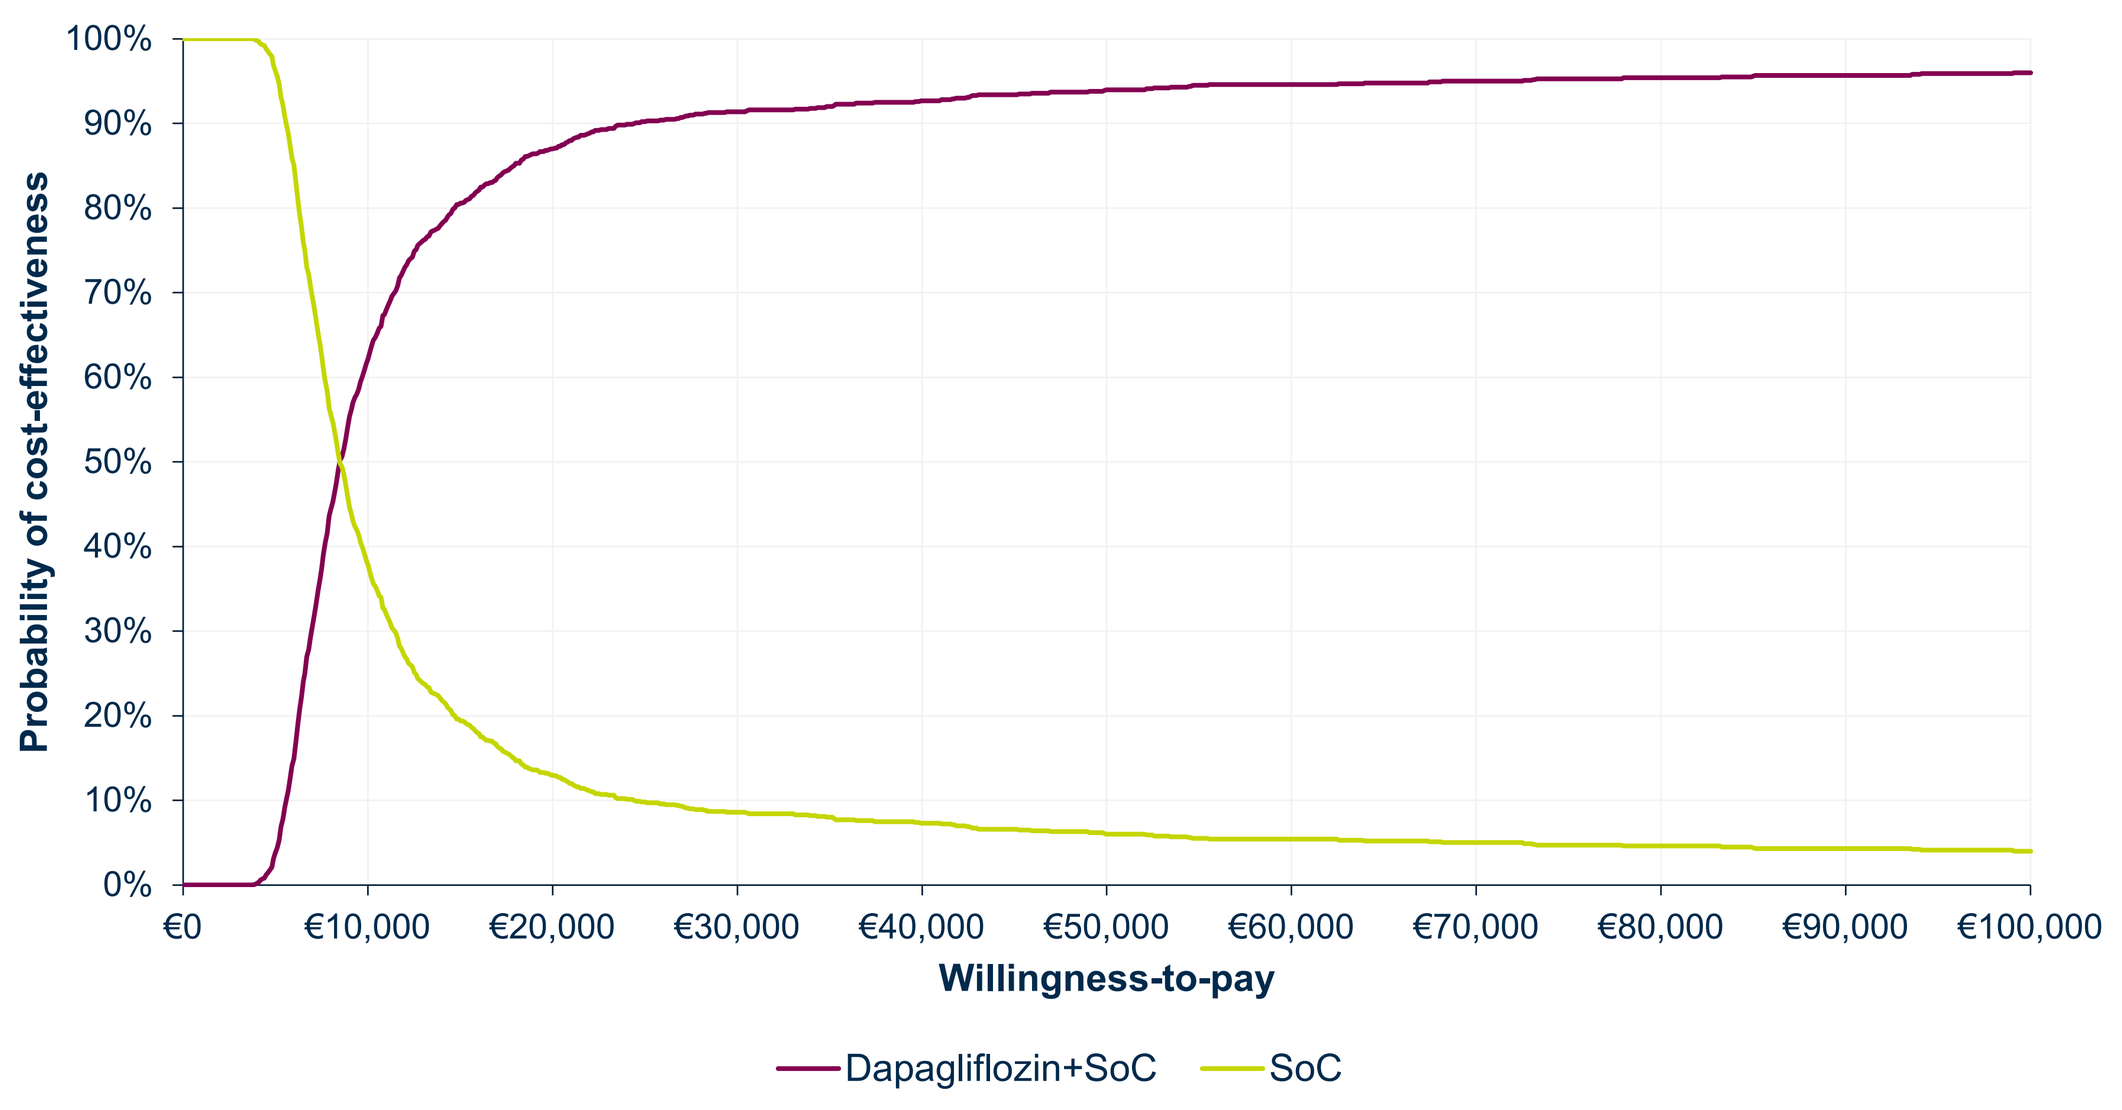
Probability of cost-effectiveness
Willingness-to-pay
Dapagliflozin+SoC
SoC

Conclusions

- Dapagliflozin, added to SOC, was considered cost-effective compared with SOC alone in patients with HFpEF/HFmrEF.
- This cost-effectiveness analysis was considered valid and supported the reimbursement decision in Portugal.

Disclosures

All authors disclose employment by AstraZeneca.

Corresponding author email address:
andre.andrade@astrazeneca.com.

References

1. World Heart Federation, https://world-heart-federation.org/what-we-do/heart-failure/
2. Solomon SD, McMurray JJV, Claggett B et al., Dapagliflozin in Heart Failure with Mildly Reduced or Preserved Ejection Fraction. N Engl J Med. 2022 Sep 22;387(12):1089-1098. doi: 10.1056/NEJMoa2206286. Epub 2022 Aug 27. PMID: 36027570.
3. Ferreira PL, Pereira LN, Antunes P, Ferreira LN. EQ-5D-5L Portuguese population norms. Eur J Health Econ. 2023 Dec;24(9):1411-1420. doi: 10.1007/s10198-022-01552-9. Epub 2023 Jan 11. PMID: 36630005; PMCID: PMC9838350.

Funded by AstraZeneca

Presented at the ISPOR Europe 2024, November 17-20, Barcelona, Spain.