EPH160

A Targeted Literature Review to Assess the Impact of Environmental and Socio-demographic Factors on the Onset of Type 1 Diabetes

Allali N[1], Petrie JL[2], Créquer-Grandhomme A[1], Vataire AL[1], Miry B[1], Bahloul A[1]
[1]Sanofi France, Gentilly; [2]Putnam, United Kingdom

INTRODUCTION

In 2021, approximately 8.4 million people worldwide were diagnosed with type 1 diabetes[1]. The incidence of type 1 diabetes is increasing at an average annual rate of 2-5% in paediatric populations[2], suggesting it is a public health concern

Identifying and understanding the influence of external factors on the aetiology of type 1 diabetes is essential for effective screening and prevention of the disease

OBJECTIVE

- To provide an overview of the published literature on environmental and socio-demographic factors associated with onset of type 1 diabetes

METHODS

- An electronic search strategy was run in May 2024 in Medline and Embase to capture narrative and systematic reviews reporting on environmental or socio-demographic factors associated with onset of type 1 diabetes
- 669 reviews were independently screened by 2 reviewers against PICOS criteria, and the reference lists of relevant reviews were searched to identify and prioritise the most robust observational studies published in the last 10 years reporting environmental and/or socio-demographic factors associated with onset of type 1 diabetes
- Supplemental hand searching was also performed

Results of the literature search

- Twenty-two publications, reporting 18 studies using several national and international databases, were included in this targeted review. These studies covered countries located in Europe, North America, the Middle East, Africa, and Australasia (Figure 1)

Figure 1. Distribution of included publications (n=22) by country

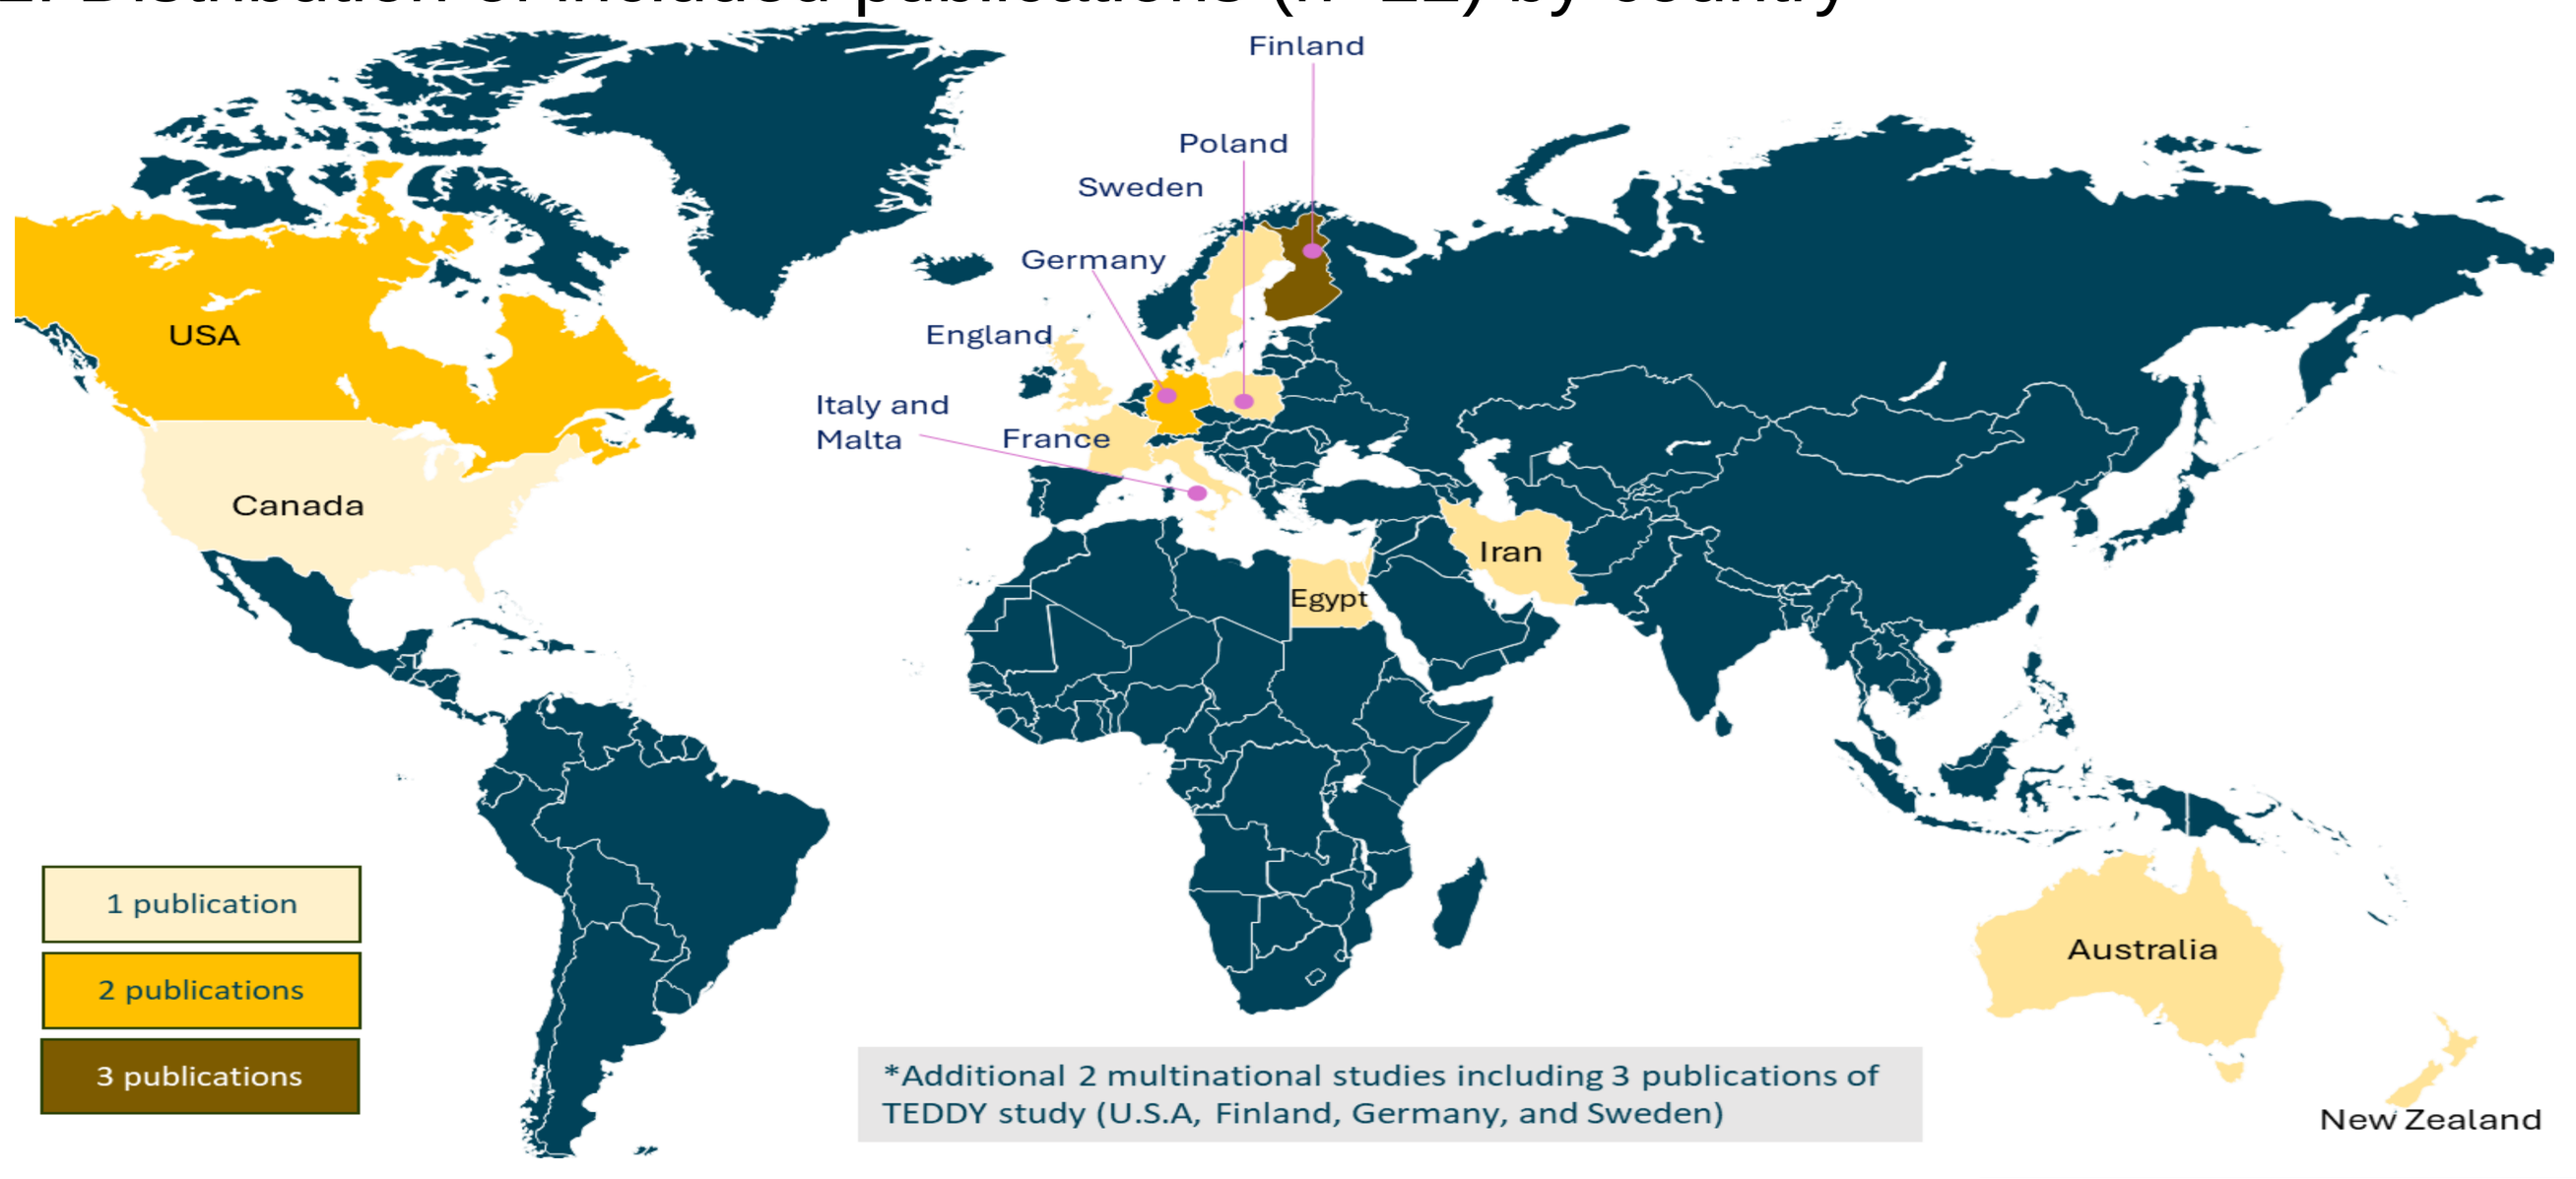
Finland
Poland
Sweden
Germany
England
Italy and Malta
France
USA
Canada
Iran
Egypt
Australia
New Zealand
1 publication
2 publications
3 publications
*Additional 2 multinational studies including 3 publications of TEDDY study (U.S.A, Finland, Germany, and Sweden)

Results – factors associated with T1D and/or islet autoimmunity

Environmental factors

Factor	Country/Region			
	North America	Europe	Scandinavia	Middle East
Diet				
High calorie, high protein, or high carbohydrate diets		↑		
Gut microbiota				
Higher levels of Firmicutes, or lower levels of Bacteroidetes		↑*		
Higher Bifidobacterium spp levels		↓*		
Viruses/infections				
Early CMV infection			↓*	
CVB1			↑* ⇡*	
Enterovirus-B (consecutive positive samples)	⇡*	⇡*	⇡*	
Rotavirus vaccination	↓*			
COVID-19 infection	↑*	↑↓		
Gastrointestinal infection before 1 year of age - IAA age 2-4 years	⇡	⇡	⇡*	
Gastrointestinal infection 1 to <2 years of age - IAA up to 10 years	⇣*	⇣	⇣*	
Air pollution				
NO_x exposure		-	↑*[a]	-
SO_2 exposure		↑*		↑
O_3 exposure	↑*		↑	↑
PM_{10} exposure		↑*		-
Other factors				
Living in a built-up area		↓	⇡*	
Urinary concentrations of the pesticide β-BHC				↑*
Solar radiation exposure		↑*		↑*

Socio-demographic factors

Factor	Country/Region					
	North America	Europe	Scandinavia	Middle East	Australasia	Egypt
Sex, race, and ethnicity						
Sex	-	-	-	-		
Asian ethnicity		↓*				
Black ethnicity		↓*				
White ethnicity		↑*				
Socio-economic factors						
Lower parental education		↑*		-		
Parental occupation				-		
Higher deprivation		↑			↑*	
Other socio-demographic factors						
High population density		↓*				
Rural residency		↑*				↑*
Agricultural environment			⇣*			

Abbreviations: β-BHC, beta-hexachlorocyclohexane; CMV, cytomegalovirus; CVB1, coxsackie virus B1; IAA, insulin autoantibody; NOx, nitric oxide; O_3, ozone; PM_{10}, particulate matter less than 10 micrometres in diameter; SO_2, sulphur dioxide; T1D, type 1 diabetes

↑ Factor associated with increased risk of type 1 diabetes
⇡ Factor associated with increased islet autoimmunity risk
↓ Factor associated with decreased risk of type 1 diabetes
⇣ Factor associated with decreased islet autoimmunity risk
↑↓ Factor associated with both increased and decreased risk of type 1 diabetes depending on individual countries within region
- Factor not associated with increased or decreased risk of type 1 diabetes
[a] Significantly increased risk of T1D in offspring of those exposed to concentrations of >22.7 μg/m^3 during the third trimester of pregnancy
*Statistically significant association
Autoimmunity refers to islet autoimmunity (IA) that progresses to T1D
Bold font highlights factors that are robust across different geographies

CONCLUSIONS

- **Overall, this targeted review identified a multitude of different environmental and socio-demographic factors associated with the onset of type 1 diabetes across a range of geographical locations**
- **Some identified risk factors, such as UV exposure and social deprivation, were associated with increased risk of type 1 diabetes across different countries whereas the risk of type 1 diabetes associated with other factors, such as No_x and PM_{10} air pollutant exposure, COVID-19 infection, and parental education, differed between countries. This suggests that individual countries may have distinct patterns of risk factors associated with onset of type 1 diabetes**
- **Therefore, robust country-specific studies involving a large number of patients that assess multiple environmental and socio-demographic factors would be necessary to gain accurate insight into specific risk factors relevant to each country**

REFERENCES
1. Gregory et al. The Lancet Diabetes & Endocrinology. 2022;10(10):741-60
2. Stene et al. Diabetes in America 2023. https://www.ncbi.nlm.nih.gov/books/NBK597412/

A full reference list of all studies included in the review is available in the supplemental material PDF

CONFLICTS OF INTEREST
Noémie Allali, Amandine Créquer-Grandhomme, Benoit Miry, Amar Bahloul, and Anne-Lise Vataire are Sanofi employees and may hold shares and/or stock options in the company. John Petrie has nothing to declare.

ACKNOWLEDGEMENTS
Assistance in conduction of the review and the design of this poster were provided by Hamza Bensalah and Steven Banks (Putnam)

ISPOR Europe 2024 | Barcelona, Spain | 17-20 November