TFS HealthScience
Clinical Research Excellence

RWD24

A Target Trial Emulation design to seek EMA approval for a reformulated drug with a Non-Inferiority, Pivotal, Retrospective Study: A Success Story

Magda Bosch de Basea[1], Neus Valveny[2], Paula Casajust[1], María Luz Samaniego[3], Jordi Figuerola[4], Andrew Shala[1], Tomás O´Mahoney[5], Marta Mas[1], Marta Forcadell[6], Emili González-Pérez[6]

1. Medical Writing, TFS HealthScience, Barcelona, Spain; 2. RWE, TFS HealthScience, Barcelona, Spain; 3. Biostatistics, TFS HealthScience, Barcelona, Spain; 4. Data Management, TFS HealthScience, Barcelona, Spain; 5. RWE, TFS HealthScience, Newbridge, Ireland; 6. Clinical Research and Evidence-Generation Science Unit, Neuraxpharm Pharmaceuticals S.L., Barcelona, Spain.

Introduction

- For many patients with a chronic neurologic condition, the control of an acute event (sudden intensification of symptoms) remains a challenge. An important factor is drug compliance, which presents an inverse relationship with the number of daily doses.
- With the goal of providing patients with the convenience of a once-daily regimen, a specific drug was developed as an extended-release (XR) oral formulation, potentially improving compliance and the efficacy-tolerability ratio.
- In the US, the Food and Drug Administration approved both the immediate release (IR) and extended-release oral formulations, whereas in Europe, the XR formulation has not been approved by the European Medicines Agency (EMA) yet.
- A target trial emulation framework was applied to improve the quality of this observational study and due to its ability to prevent avoidable biases (i.e., immortal time bias or prevalent user selection bias).

Objective

- We aimed to estimate that the effectiveness, and safety of the **XR formulation** was not "considerably inferior" than the **IR formulation** in subjects suffering from a CNS disease who initiated treatment between September 2015 and April 2022, in a non-inferiority study.
- The results of the study were aimed to support the request for the XR formulation approval to the of EMA.

Data source

We used **retrospective real-world data** of subjects from a large national private claims and electronic medical records (EMR) database in the US. Natural language processing was used on unstructured notes, as well.

Methods

- The **Target trial emulation framework,** which mimics the target trial was used for designing and analyzing this observational study aimed to estimate the causal effectiveness and safety of using the XR formulation. This framework included the following components:

- **Eligibility criteria:** ✓ Aged ≥12 years old; ✓ CNS disease diagnosis prior to drug of interest; ✓ Initiating an oral XR or IR formulation of the study drug.
- **Treatment strategy:** New users of XR and IR formulations (i.e., no filled dispensation of XR and IR formulations during the preceding 12 months).
- **Treatment assignment to the IR or XR cohort:** consistent with first filled dispensation date for each subject. To emulate randomization XR patients were matched to IR patients using propensity scores (PS) based on 28 variables. Figure 3: Probability density distribution before and after Propensity Score matching from logit regression.
- **Outcomes:** Effectiveness (i.e., proportion of subjects free from acute events for at least 24 weeks) and safety-related outcomes (i.e., incidence rate of adverse events of special interest) identified during follow-up.
- **Causal contrasts of interest:** as-treated (analyses according to the treatment they received) and, for sensitivity analyses, intention-to-treat (analyses according to the initial treatment assigned, regardless of discontinuation) strategies were used (see Figure 1 below).

Figure 1. Illustration of the as-treated and intention-to-treat strategies.

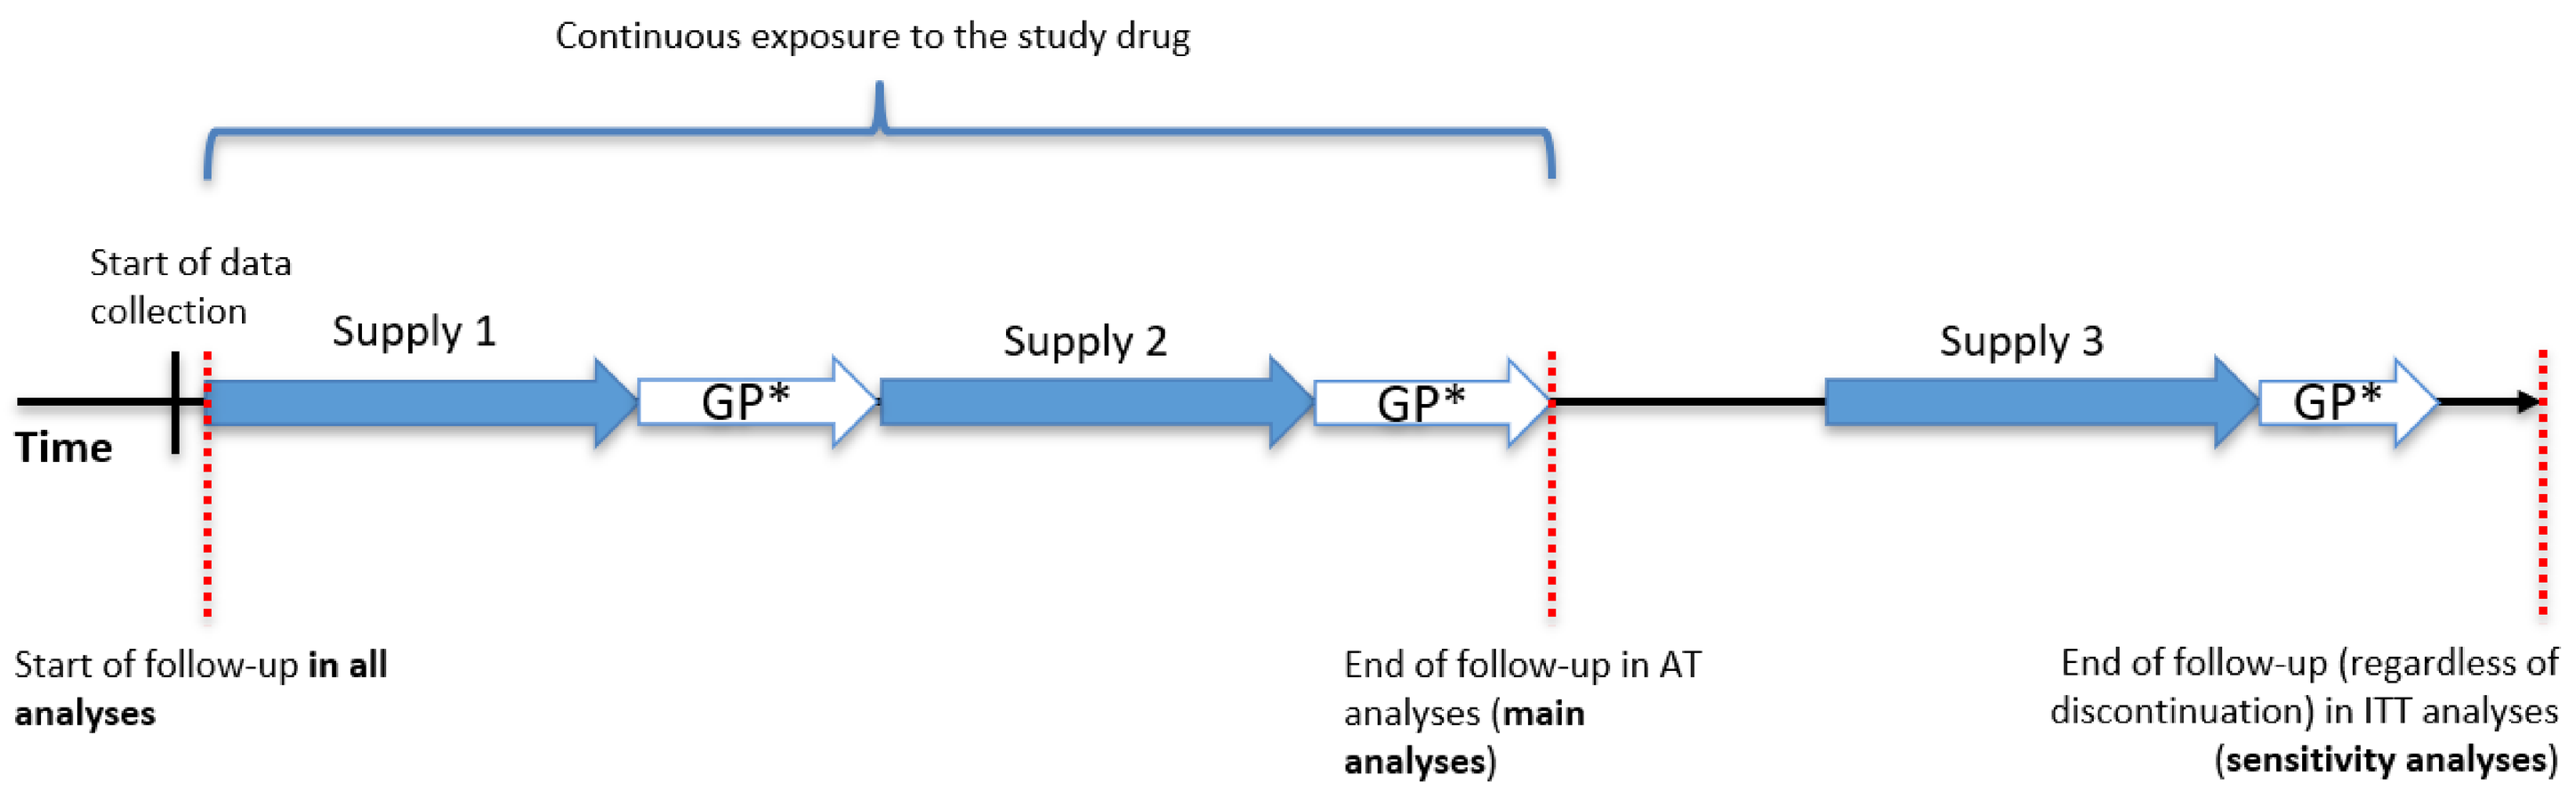
Continuous exposure to the study drug
Start of data collection
Supply 1
Supply 2
Supply 3
GP*
GP*
GP*
Time
Start of follow-up **in all analyses**
End of follow-up in AT analyses (**main analyses**)
End of follow-up (regardless of discontinuation) in ITT analyses (**sensitivity analyses**)

*AT: As-treated strategy; ITT: Intention-to-Treat strategy; GP: Grace period (permissible interval of 14 days between prescriptions during which an individual who appears to be unexposed is considered exposed).

- **Statistical analysis:** Multivariable models, adjusted by additional covariates (like concomitant drugs at index date), covariates with special relevance for the disease and a post-baseline confounder (i.e., non-adherence).
- **Additional counterfactual reasoning:** Negative controls (outcomes with no causal relationship with studied drugs or outcomes) were defined by central nervous system experts.

Negative Control Outcomes		
Acute atopic conjunctivitis	COPD	Asthma episode/attack
Allergic rhinitis due to pollen	GERD	Carbuncle and furuncle
Arthritis	Hypercholesterolemia	Tinnitus

Results

- **Figures 2 and 3** illustrate the process for achieving comparable XR and IR cohorts under a target trial emulation framework.

Figure 2. Disposition of subjects in the study

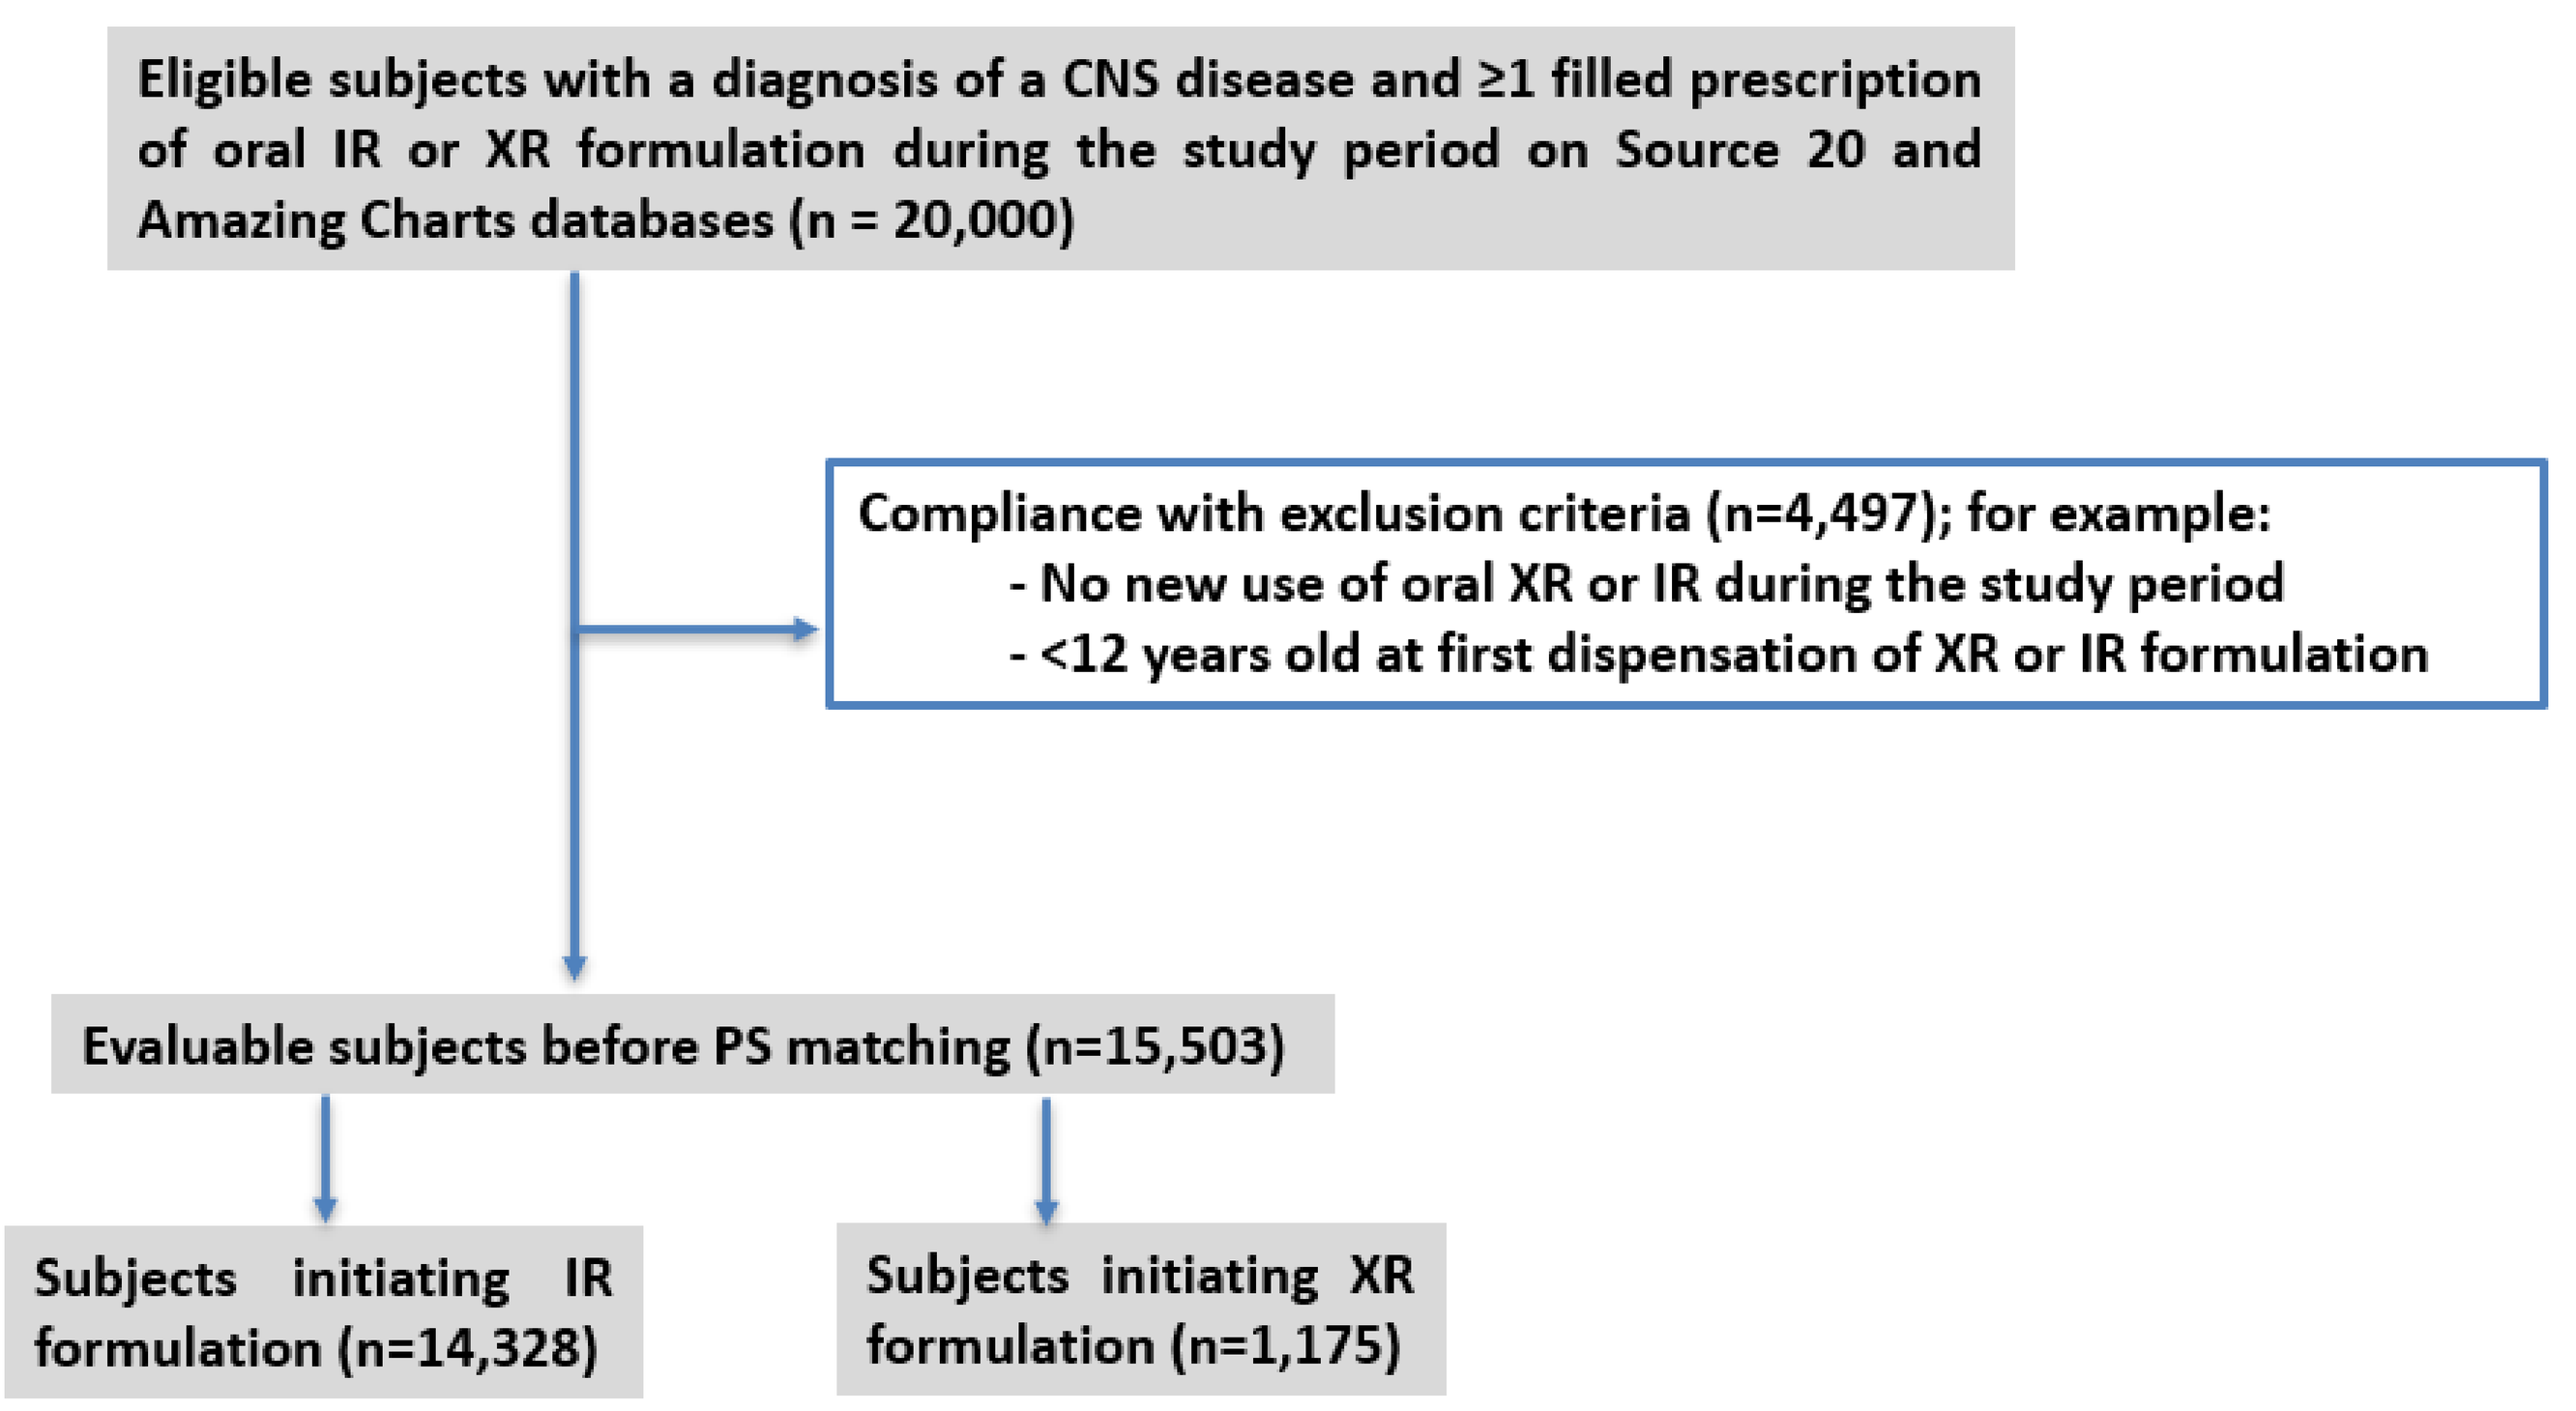
Eligible subjects with a diagnosis of a CNS disease and ≥1 filled prescription of oral IR or XR formulation during the study period on Source 20 and Amazing Charts databases (n = 20,000)
Compliance with exclusion criteria (n=4,497); for example:
- No new use of oral XR or IR during the study period
- <12 years old at first dispensation of XR or IR formulation
Evaluable subjects before PS matching (n=15,503)
Subjects initiating IR formulation (n=14,328)
Subjects initiating XR formulation (n=1,175)

Figure 3. Propensity score matching emulating target trial randomization

- PS variables assessed and included in the model

Covariate category	Number of covariates in category	
	Assessed for PS matching (Total n=65)	Finally included in the model (Total n=28)
Sociodemographic characteristics	5	2
Covariates related to lifestyle	5	5
Epilepsy history and complications	1	1
Other comorbidities	6	2
Laboratory values	3	0
Prior/concomitant use of same class drugs	4	2
Prior use of other drugs	40	15
HCRU covariates	1	1

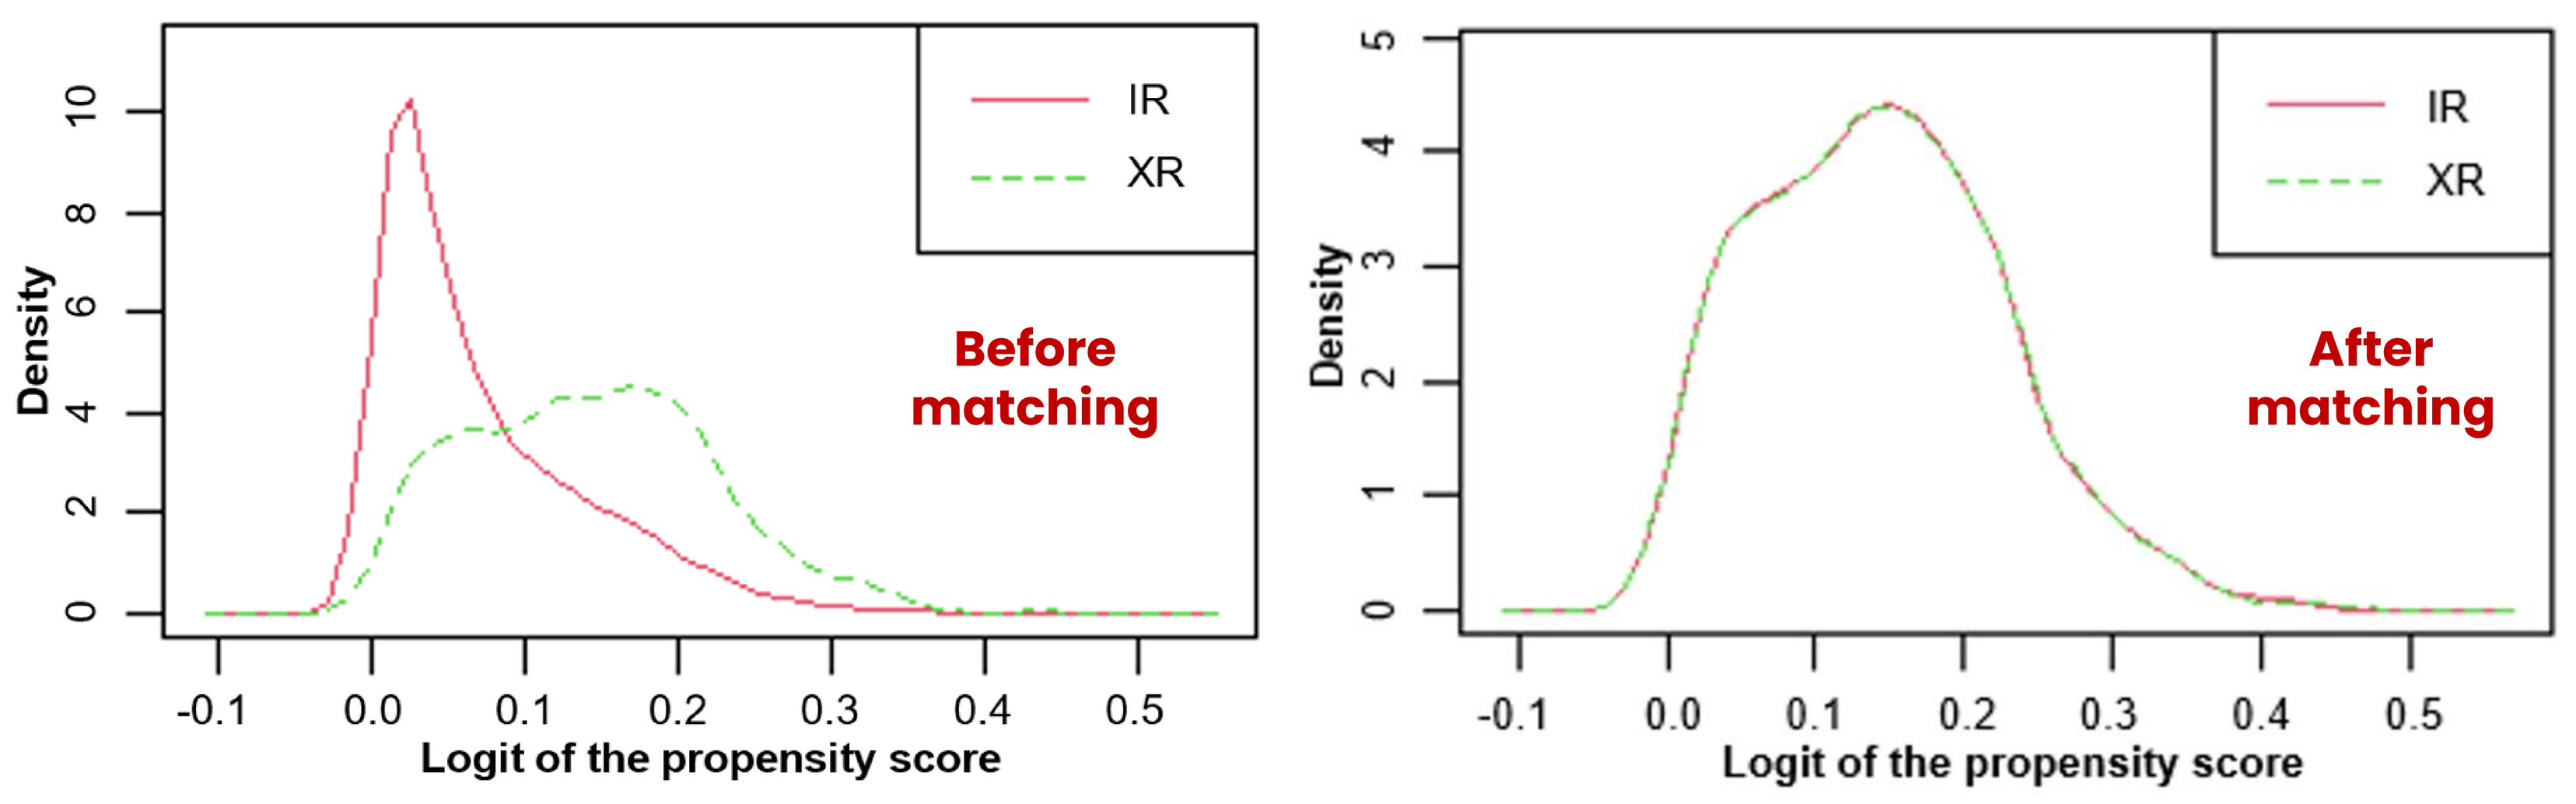
Density
IR
XR
Before matching
After matching
Logit of the propensity score

- The lower bound of the 95%CI of the adj. percentage difference in the proportion of subjects under the XR formulation free from acute events for at least 24 weeks was above the non-inferiority margin, and also supported the superiority of XR.

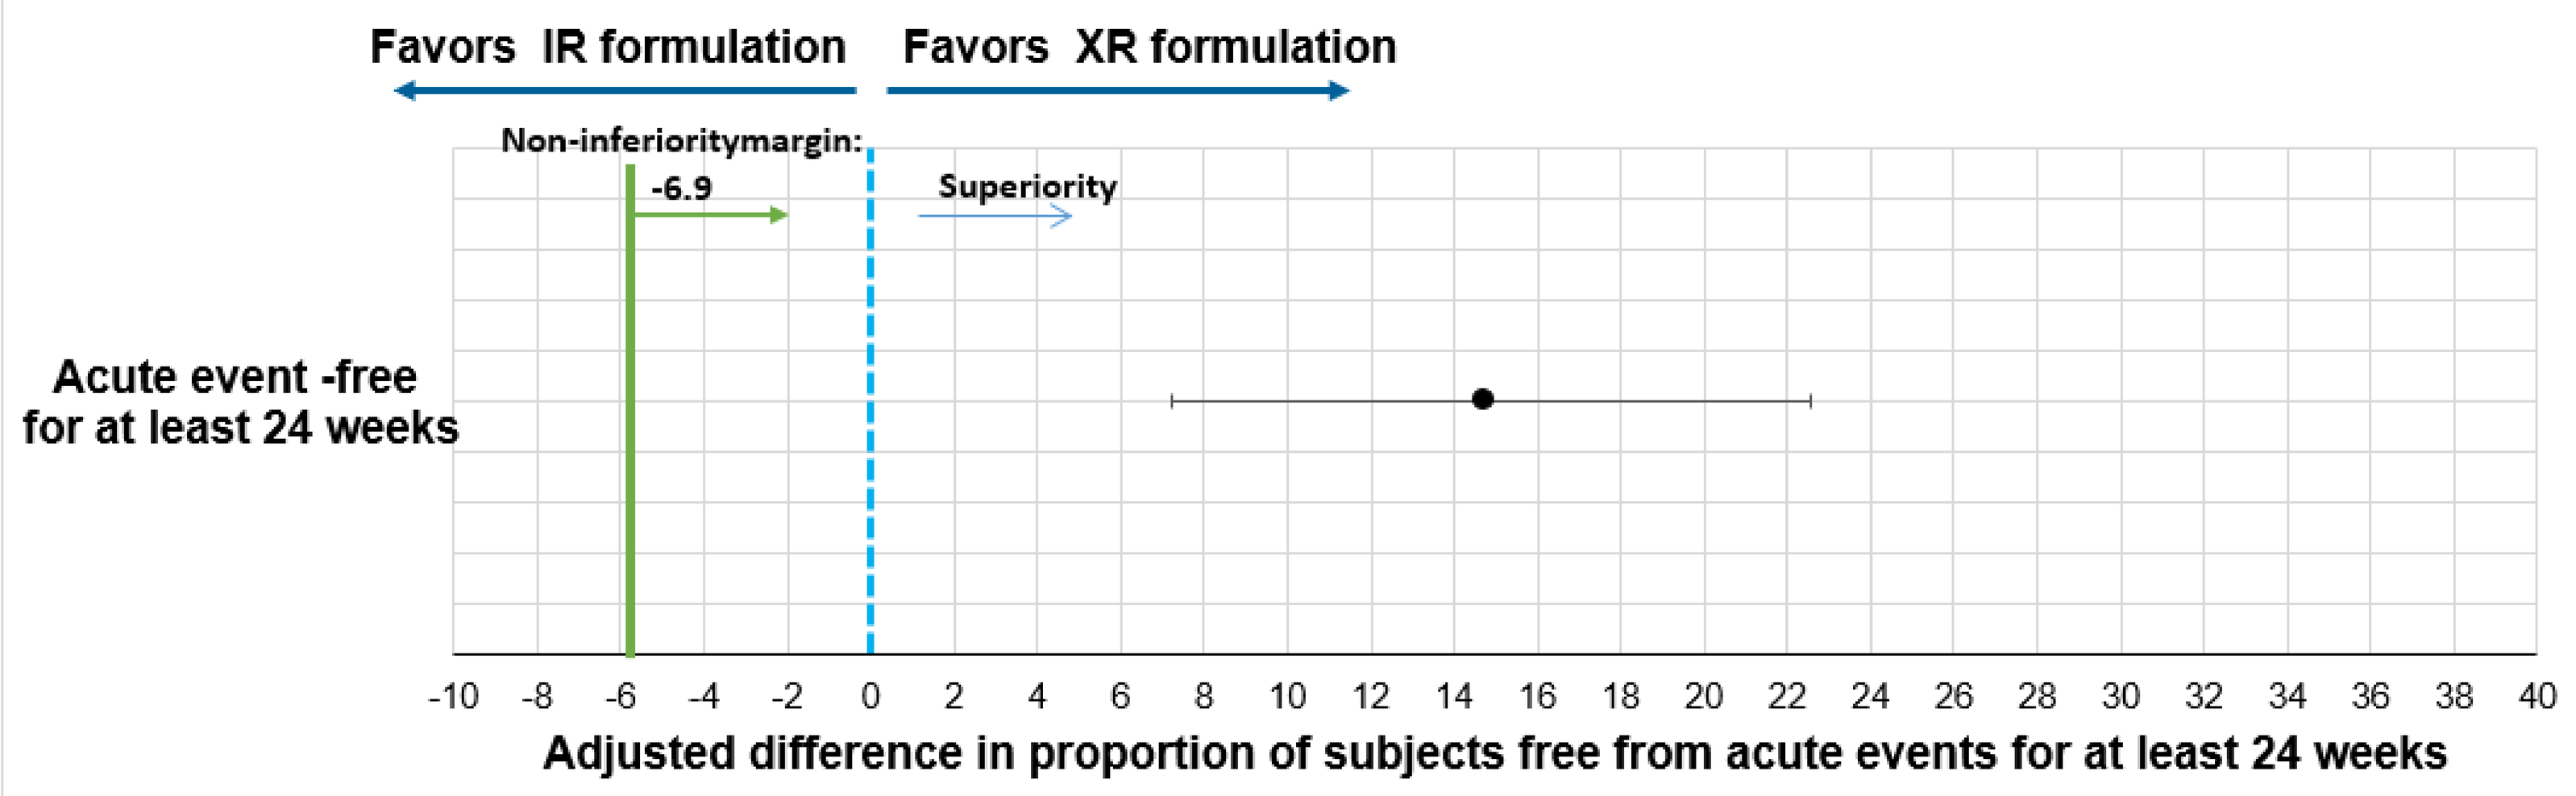
Favors IR formulation
Favors XR formulation
Non-inferioritymargin: -6.9
Superiority
Acute event -free for at least 24 weeks
Adjusted difference in proportion of subjects free from acute events for at least 24 weeks

- XR formulation showed a non-inferior and a more acceptable safety profile than the IR one. The incidence rate and the proportion of subjects experiencing at least one of some adverse events of special interest (AESI) were statistically significant lower in subjects being treated with the XR formulation. Other AESI were similar between both formulations.
- Users of XR formulation had a significantly lower rate of all analyzed all-cause HCRU outcomes during the follow-up period, including outpatient visits, ambulance use, emergency department visits, hospitalizations, filled prescriptions/dispensations and procedures.

Conclusions

- **EMA scientific advice corroborated the target trial emulation design to support the application of the new formulation.**
- **These comprehensive set of advanced pharmacoepidemiologic and statistical methodologies led to highly consistent results, showing non-inferiority of XR formulation vs IR in both primary and secondary endpoints, including effectiveness, safety and HCRU.**
- **Furthermore, a set of 9 negative control outcomes (not linked to study endpoints or exposure) reassured that the observed results were not due to residual or unmeasured confounding, thus validating the adequacy of the RWE study as a target (randomized) trial emulation (TTE).**

References

Hernán, M. A., Alonso, A., Logan, R., Grodstein, F., Michels, K. B., Willett, W. C., Manson, J. E., & Robins, J. M. (2008). Observational studies analyzed like randomized experiments: an application to postmenopausal hormone therapy and coronary heart disease. Epidemiology (Cambridge, Mass.), 19(6), 766–779.
Hernán, M. A., Wang, W., & Leaf, D. E. (2022). Target Trial Emulation: A Framework for Causal Inference From Observational Data. JAMA, 328(24), 2446–2447. https://doi.org/10.1001/jama.2022.21383.
Fu E. L. (2023). Target Trial Emulation to Improve Causal Inference from Observational Data: What, Why, and How?. Journal of the American Society of Nephrology : JASN, 34(8), 1305–1314.

Conflict of interests

All authors are/were employees of TFS HealthScience and Neuraxpharm Pharmaceuticals S.L. at the time of the abstract submission.