EE782

The Landscape of Pre-Exposure Prophylaxis (PrEP) in Brazil: Challenges, Advances, and Opportunities as a Public Health Strategy in the Battle Against HIV/AIDS

Graziela Bernardino,[1] Gabriel Marasco,[2] Ian Jacob,[3] Larissa Rodrigues,[1] Marcella Alemar,[1] Roberto Zajdenverg,[1] Rodrigo Zilli,[1] Straus Tanaka[1]

[1]GSK, Rio de Janeiro, Brazil; [2]Origin Health, São Paulo, Brazil; [3]ViiV Healthcare, London, UK

Key Takeaways

- In transgender individuals and cisgender men who have sex with men who have a greater likelihood of HIV infection, scenarios in which oral PrEP was the only prevention strategy resulted in ~214,000-479,000 estimated new HIV infections and ~R$ 7.2-17.3 billion in costs over 10 years
- Emergency attendance or hospitalization, comorbidities, and monitoring accounted for 96% of total costs
- Increasing prevention options with new forms of PrEP may be essential to ending the HIV epidemic and enhancing the effectiveness of public health policies in Brazil

Introduction

- Despite the availability of pre-exposure prophylaxis (PrEP) with tenofovir and emtricitabine (TDF/FTC) in a single daily pill, the incidence of HIV remains high in Brazil[1,2]
- Data monitoring of oral PrEP in Brazil demonstrates ~32% of PrEP users are not persistent, and this rate can increase up to 60% among youths, highlighting the need for new PrEP options that promote higher persistence rates[3]

Aim

- This study estimated the number of new HIV infections and related costs over a 5- and 10-year period among transgender individuals and cisgender men who have sex with men in a hypothetical scenario in which oral PrEP is the only prevention strategy

Methods

- A prediction tool was developed to estimate the number of HIV infections over time and related costs and populations
 - Cost and population were modeled using variables described in the study design
- HIV infections were estimated by the rate of infection with oral PrEP (HTPN 083 pivotal study)[4] for primary and secondary HIV infections
 - The recent trend of new HIV infections was also considered to validate the plausibility of the results
- HIV costs were calculated considering a delphi panel conducted with local specialists in Brazil and data from the Ministry of Health (antiretroviral therapy [ART] drug costs)

Study Design

Definition of population:

Transgender individuals and cisgender men who have sex with men who have a greater likelihood of HIV infection:

Scenario 1: aged 15 to 30 years

Scenario 2: aged ≥ 15 years

HIV infection assessment:

- Primary HIV infection: proportion of individuals who are seroconverted
- Secondary HIV infection: resulting or onward infections from those of the primary population

Estimation based on:

- Clinical trial and literature data[4]
- HIV infection records in Brazil from 2012-2022[5]

Costs included:

- HIV management costs: diagnosis, HIV treatment, comorbidity/co-infection treatment monitoring, and emergency attendance or hospitalizations

Results

Scenario 1 (Aged 15-30 Years)

- Approximately 47,400 new HIV infections and R$ 1 billion in costs were estimated over a 5-year period, and approximately 214,000 new HIV infections and R$ 7.2 billion in costs were estimated over a 10-year period (Figure 1)

Figure 1. Cumulative HIV Cost and HIV Infections Over A 10-Year Time Period: Scenario 1 (15-30 Years)

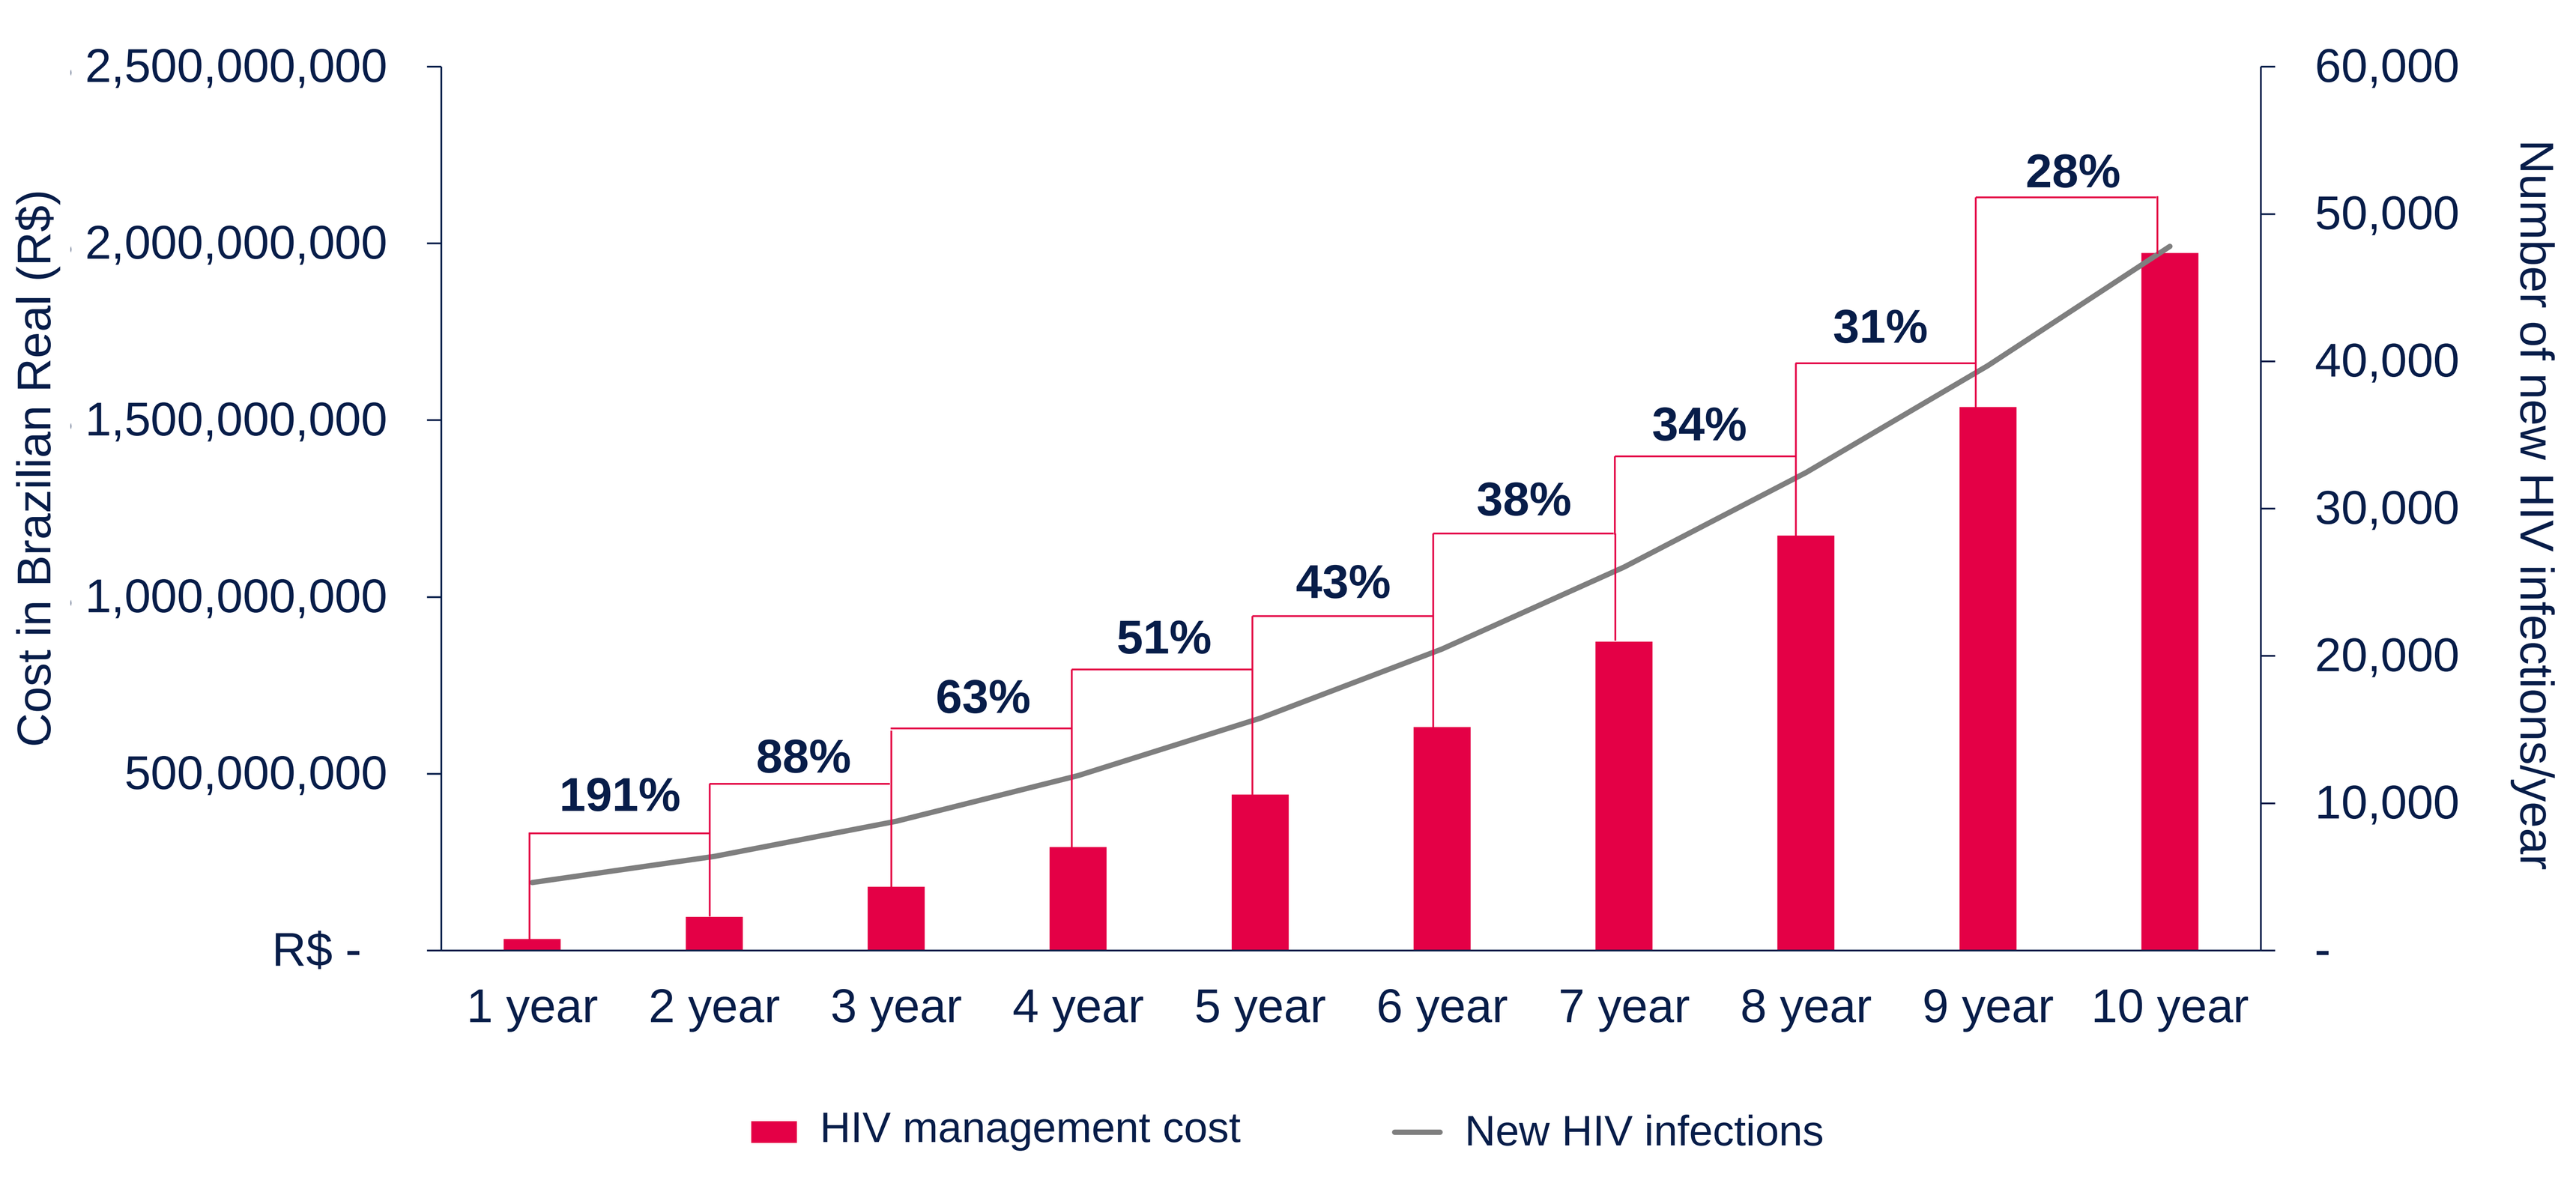
HIV management cost and number of new HIV infections per year
Cost in Brazilian Real (R$)
2,500,000,000
2,000,000,000
1,500,000,000
1,000,000,000
500,000,000
R$ -
Number of new HIV infections/year
60,000
50,000
40,000
30,000
20,000
10,000
-
191%
88%
63%
51%
43%
38%
34%
31%
28%
1 year
2 year
3 year
4 year
5 year
6 year
7 year
8 year
9 year
10 year
HIV management cost
New HIV infections

Scenario 2 (Aged ≥15 Years)

Figure 2. Cumulative HIV Cost and HIV Infections Over A 10-Year Time Period: Scenario 2 (≥15 Years)

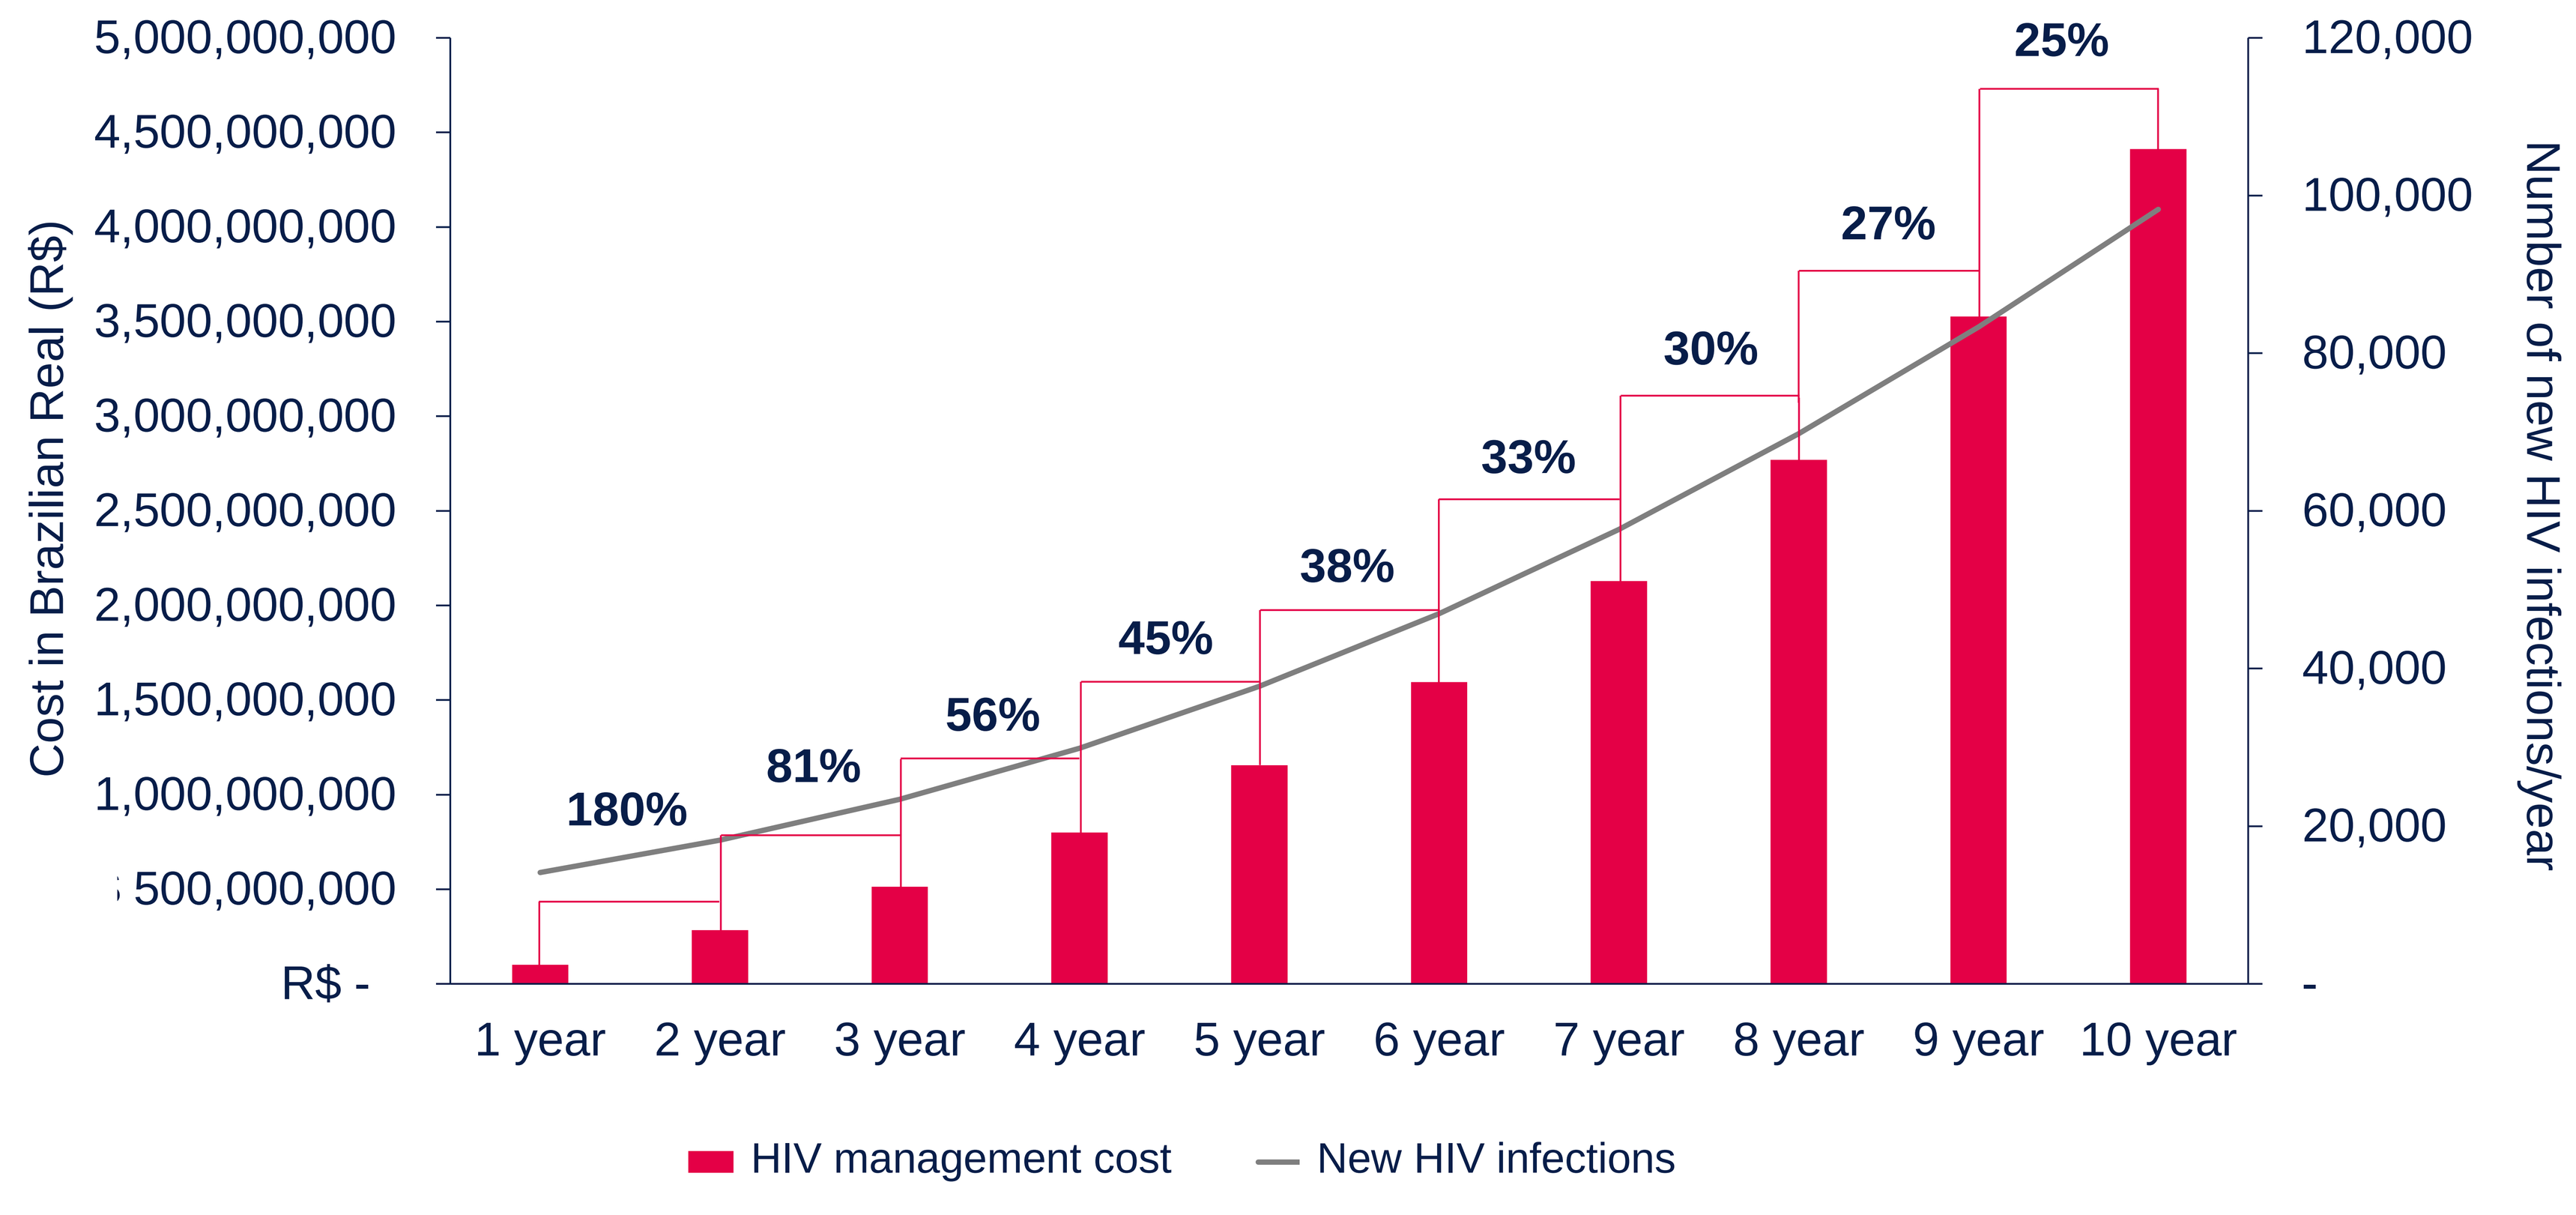
HIV management cost and number of new HIV infections per year
Cost in Brazilian Real (R$)
5,000,000,000
4,500,000,000
4,000,000,000
3,500,000,000
3,000,000,000
2,500,000,000
2,000,000,000
1,500,000,000
1,000,000,000
500,000,000
R$ -
Number of new HIV infections/year
120,000
100,000
80,000
60,000
40,000
20,000
-
180%
81%
56%
45%
38%
33%
30%
27%
25%
1 year
2 year
3 year
4 year
5 year
6 year
7 year
8 year
9 year
10 year
HIV management cost
New HIV infections

- Emergency attendance or hospitalization accounted for 40% of total costs, whereas costs related to comorbidities and monitoring accounted for 56%; the remaining 4% was divided between diagnosis and initial approach, tests for switching ART, patients on first-line ART, and patients on second-line ART+ (Table)
- The first 5 years accounted for 14% of total costs, while the remaining 86% was spent in the last 5 years analyzed

Table. HIV Management Costs Estimated in 5 and 10 Years: Scenario 1 (15-30 Years)

Costs per category, R$	In 5 years	In 10 years
Diagnosis and initial approach	49,967,840.30	225,353,974.61
Monitoring	211,841,406.53	1,504,633,437.99
Tests for switching ART	30,766.50	154,457.59
Patients on 1L ART	15,276,429.61	68,896,396.38
Patients on 2L ART+	1,443,987.33	10,435,523.17
Comorbidities	353,975,931.16	2,514,163,925.75
Emergency attendance or hospitalization	409,452,394.72	2,908,193,324.74

1L, first-line; 2L, second-line; ART, antiretroviral therapy; R$, Brazilian Real.

- Approximately 123,300 new HIV infections and R$ 2.9 billion in costs were estimated over a 5-year period, and approximately 479,000 new HIV infections and R$ 17.3 billion in costs were estimated over a 10-year period (Figure 2)
- The proportions in scenario 2 for HIV management are similar to those in scenario 1; approximately 17% of costs are spent in the first 5 years of the analysis, while the remaining 83% are spent in the last 5 years

Conclusions

- Current literature data and the PrEP monitoring panel highlight significant issues with adherence and persistence, which are critical gaps in the current PrEP landscape in Brazil
- This study assessed consequences in terms of new HIV diagnoses and associated costs
 - Over a 10-year period, scenario 1 estimated ~214,000 new HIV diagnoses and R$ 7.2 billion in costs, whereas scenario 2 estimated ~479,000 new HIV diagnoses and R$ 17.3 billion in costs
- Expanding prevention options with new forms of PrEP and prevention strategy choices are essential to more effectively achieve the goals of ending the HIV epidemic and enhancing the effectiveness of public health policies in Brazil

Acknowledgments: This study was funded by GSK (study 144950). Editorial assistance and graphic design support for this poster were provided under the direction of the authors by MedThink SciCom and funded by ViiV Healthcare.

References: 1. Luz et al. *BMC Public Health*. 2023;23:1128. **2.** National Commission for Incorporating Technologies Into the SUS. https://www.gov.br/conitec/pt-br/midias/relatorios/2017/resoc44_tenofovir_entricitabina_prep_hiv_decisao_final.pdf. Accessed September 16, 2024. **3.** Department of HIV, AIDS, Tuberculosis, Viral Hepatitis and Sexually Transmitted Infections: PrEP Panel. https://www.gov.br/aids/pt-br/assuntos/prevencao-combinada/prep-profilaxia-pre-exposicao/painel-prep. Accessed September 16, 2024. **4.** ClinicalTrials.gov. https://clinicaltrials.gov/study/NCT02720094. Accessed September 16, 2024. **5.** UNAIDS. https://www.unaids.org/en/regionscountries/countries/brazil. Accessed September 16, 2024.

ISPOR Europe 2024; November 17-20, 2024; Barcelona, Spain

Presenting author: Marcella Alemar; marcella.r.alemar@gsk.com