Patients with Hepatitis Delta Virus Have Higher Economic Burden than Hepatitis B Virus Mono-Infection: Results from a Large Healthcare Provider in Israel

Poster # 2009

R. HAZZAN [1,2], C. WEIL [3], M. HOSHEN [3], S. GAZIT [3], O. TASSA-LIANI [4], Y. GREEN [4], M. ROCK [5], C. KIM [5]

[1] Azrieli Faculty of Medicine, Bar-Ilan University, Safed, Israel; [2] Maccabi Healthcare Services, Tel Aviv, Israel; [3] Maccabi Research and Innovation Center, Maccabi Healthcare Services, Tel Aviv, Israel; [4] Gilead Sciences Israel, Ltd., Hod Hasharon, Israel; [5] Health Economics and Outcomes Research, Global Value and Access, Gilead Sciences, Inc., Foster City, CA, USA

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- Within a large healthcare provider database capturing ~27% of the population in Israel, patients with hepatitis delta virus (HDV) infection had greater healthcare resource utilization and total costs compared with patients with hepatitis B virus (HBV) mono-infection.
- The higher costs were driven primarily by a greater rate of hospitalization, inpatient length of stay and all-cause total healthcare costs compared with patients with HBV mono-infection.

Plain Language Summary

- This real-world study of a large healthcare provider database in Israel found that patients with HDV had higher healthcare costs compared to patients with HBV mono-infection.
- These findings highlight the need for effective HDV screening, diagnosis, and treatment, which may reduce its clinical and economic burden.

Introduction

- Hepatitis D virus (HDV) coinfects with hepatitis B virus (HBV), causing the most severe form of viral hepatitis.[1]
- Patients with HDV coinfection have a greater risk of cirrhosis, hepatocellular carcinoma, liver transplant, and liver-related mortality compared to those with HBV mono-infection (HBV-Mono).[1,2,3]
- Data regarding healthcare resource use and costs incurred by patients with HDV are limited.
- Understanding the clinical and economic burden of HDV is critical to support the development and adoption of new treatment strategies.

Objective

- This study compared healthcare resource utilization (HCRU) and costs among adult patients with HDV infection versus patients with HBV-Mono in Israel.

Methods

- **Design:** Retrospective cohort study.
- **Database:** Anonymized data from Maccabi Healthcare Services (MHS) covering 1.8 million patients.
- **Population:** Diagnosis and laboratory data were used to identify patients with HDV and HBV-Mono ≥18 years of age from 2004 to 2021. Adults with chronic HBV who had a diagnosis or positive HDV test were included in the HDV cohort (index date = HDV diagnosis). The HBV-Mono cohort comprised of HBV patients tested for HDV (antibody/PCR test) without positive result or HDV diagnosis (index date = first HDV-negative test result). Patients were continuously enrolled for ≥12 months pre- and post-index (**Figure 1**).
- **Analysis:** Patient characteristics were described at the index date. All-cause HCRU and costs (using Israel Ministry of Health unit costs) per person per year were evaluated based on outpatient, inpatient and emergency room visits, and pharmacy purchases 12-months post-index. Total costs were reported in 2022 US dollars PPPY and based on unit costs from the Israel Ministry of Health. Statistical comparison between HDV vs. HBV-Mono cohorts were conducted using Fisher's exact test, Chi-squared test, and Wilcoxon rank sum test.

Figure 1. Study design

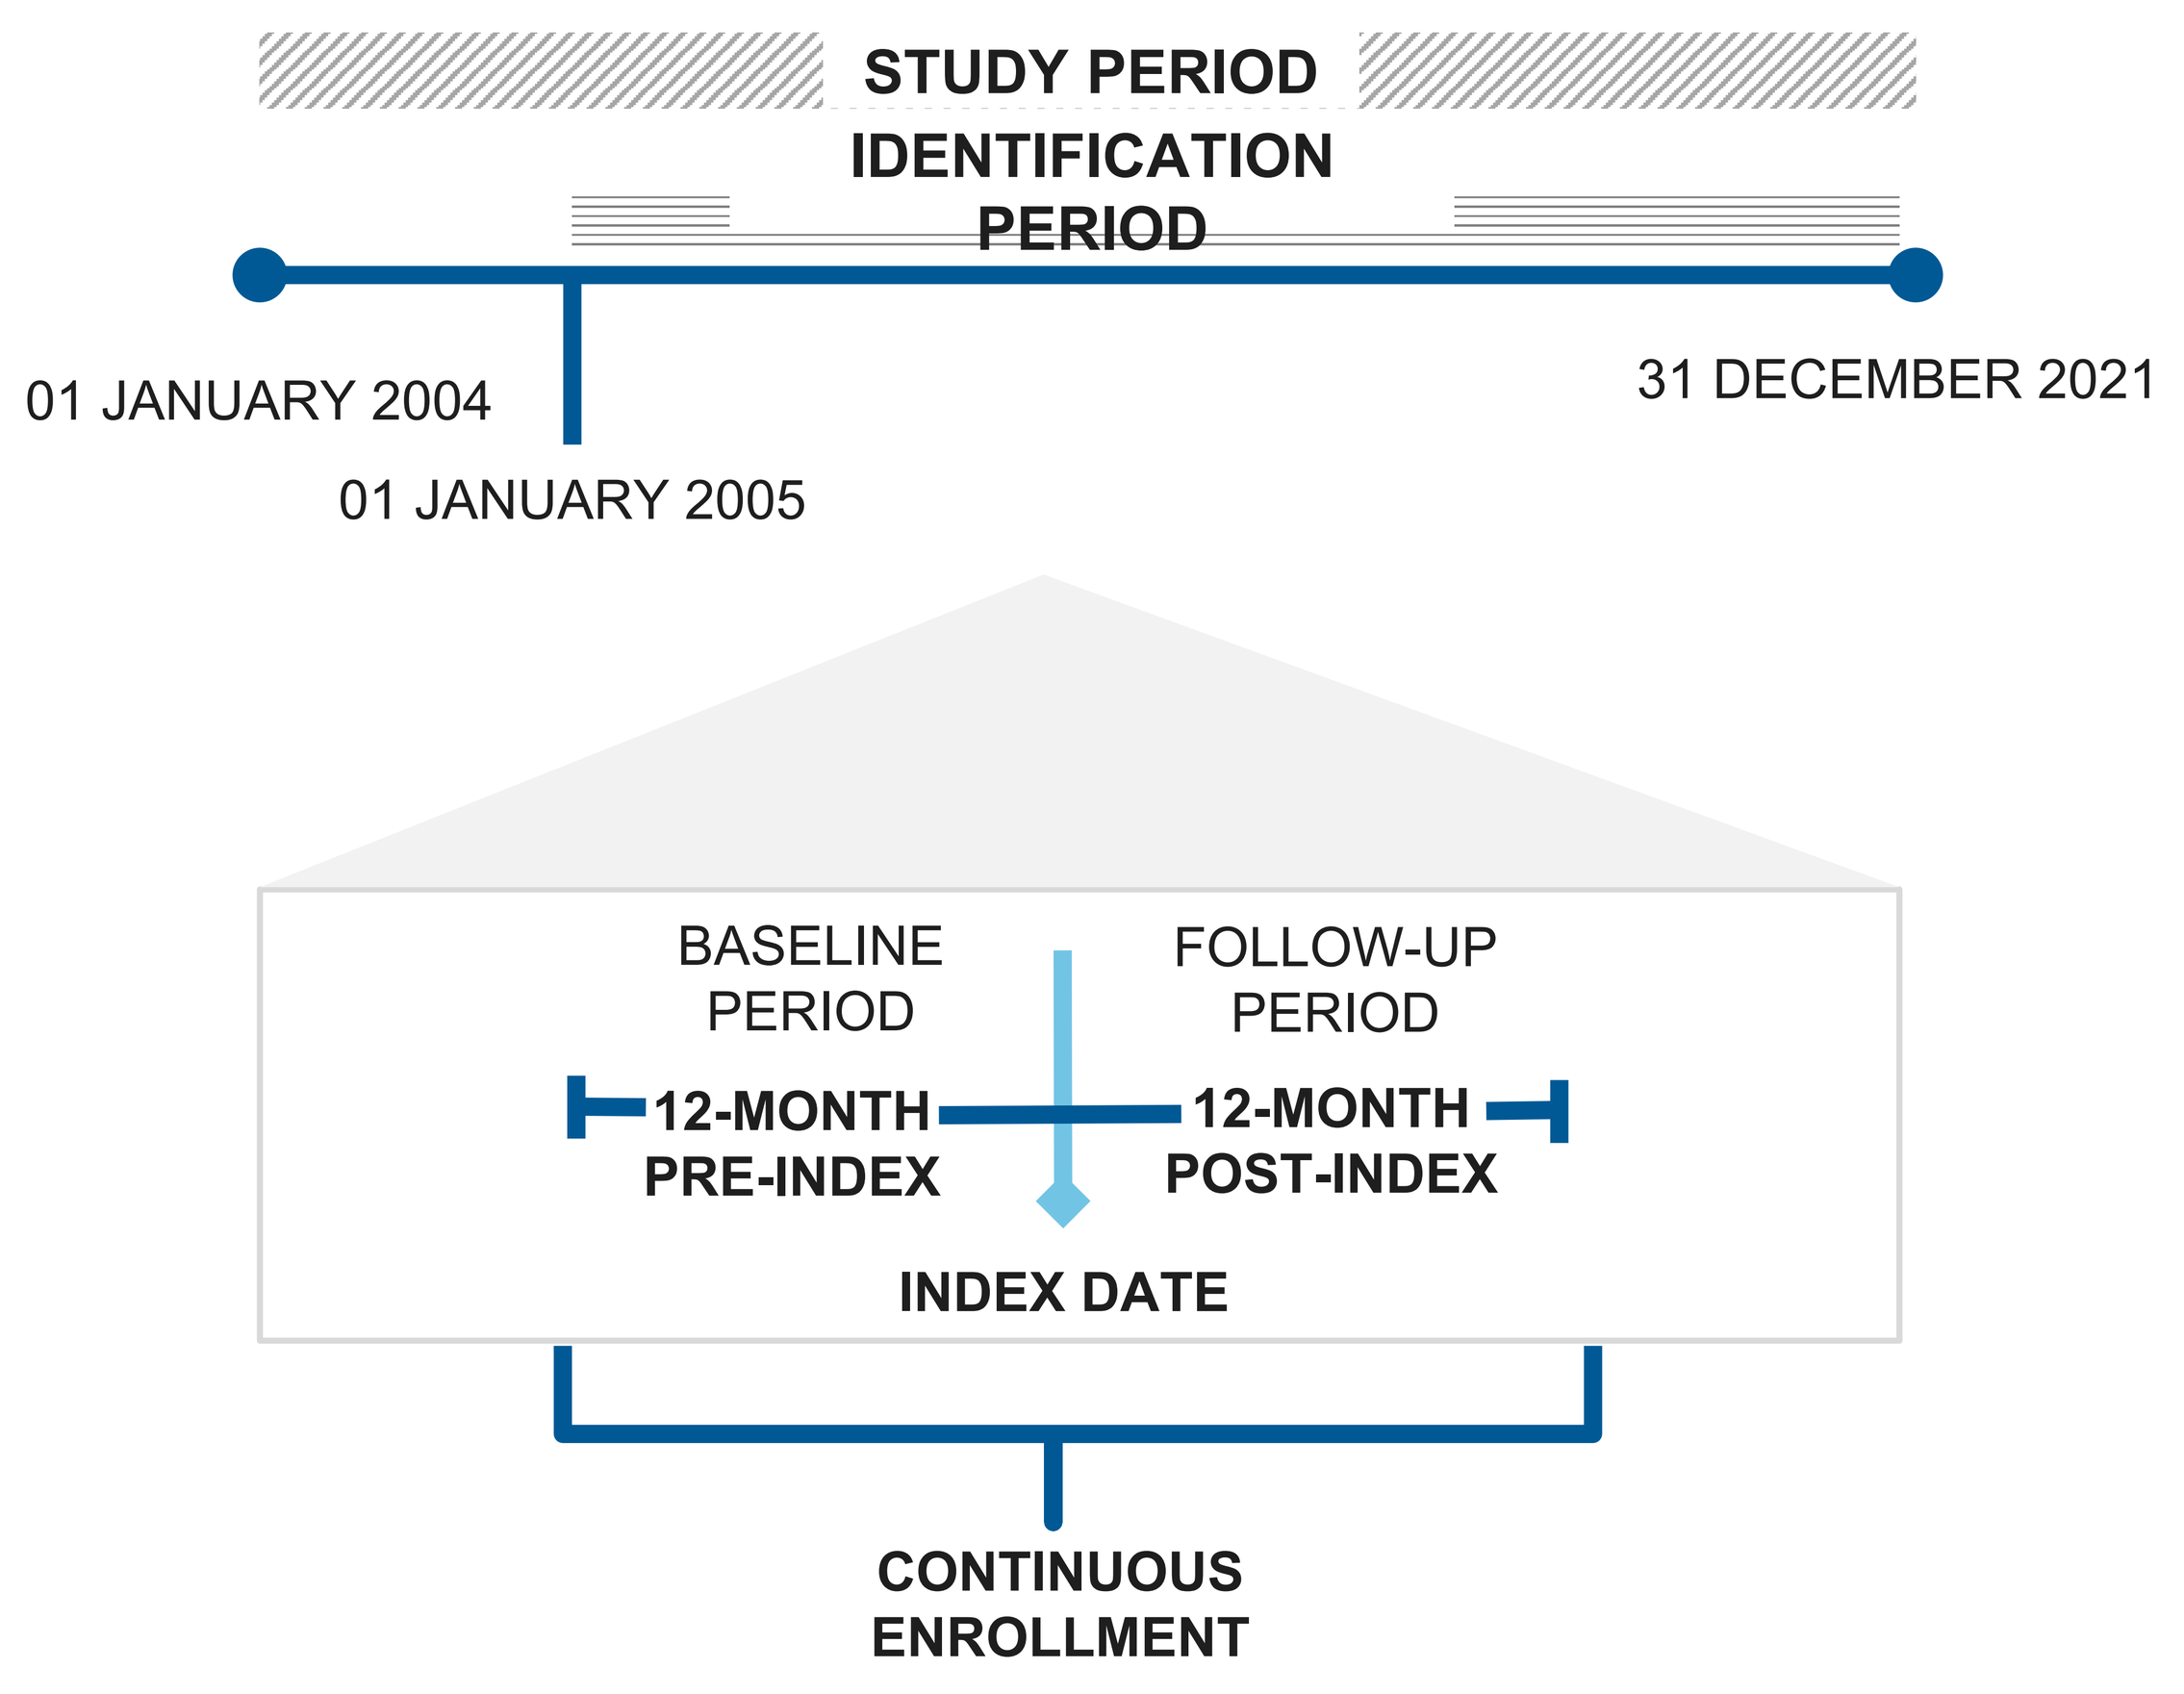

STUDY PERIOD
IDENTIFICATION PERIOD
01 JANUARY 2004
01 JANUARY 2005
31 DECEMBER 2021
BASELINE PERIOD
FOLLOW-UP PERIOD
12-MONTH PRE-INDEX
12-MONTH POST-INDEX
INDEX DATE
CONTINUOUS ENROLLMENT

Results

Patient Attrition and Patient Characteristics

- Of the 1.8 million adult patients,155 patients diagnosed with HDV and 2,420 with HBV-Mono were eligible for the economic burden analysis (**Figure 2**).
- Compared to patients with HBV-Mono, patients with HDV had lower socioeconomic status, and higher baseline prevalence rates for HIV (4.5% vs 0.8%; *P*<0.001), HCV RNA+ (5.8% vs 1.7%; *P*=0.002), and drug dependence (6.5% vs 1.7%; *P*<0.001) (**Table 1**).

Figure 2. Patient Attrition Flow Chart

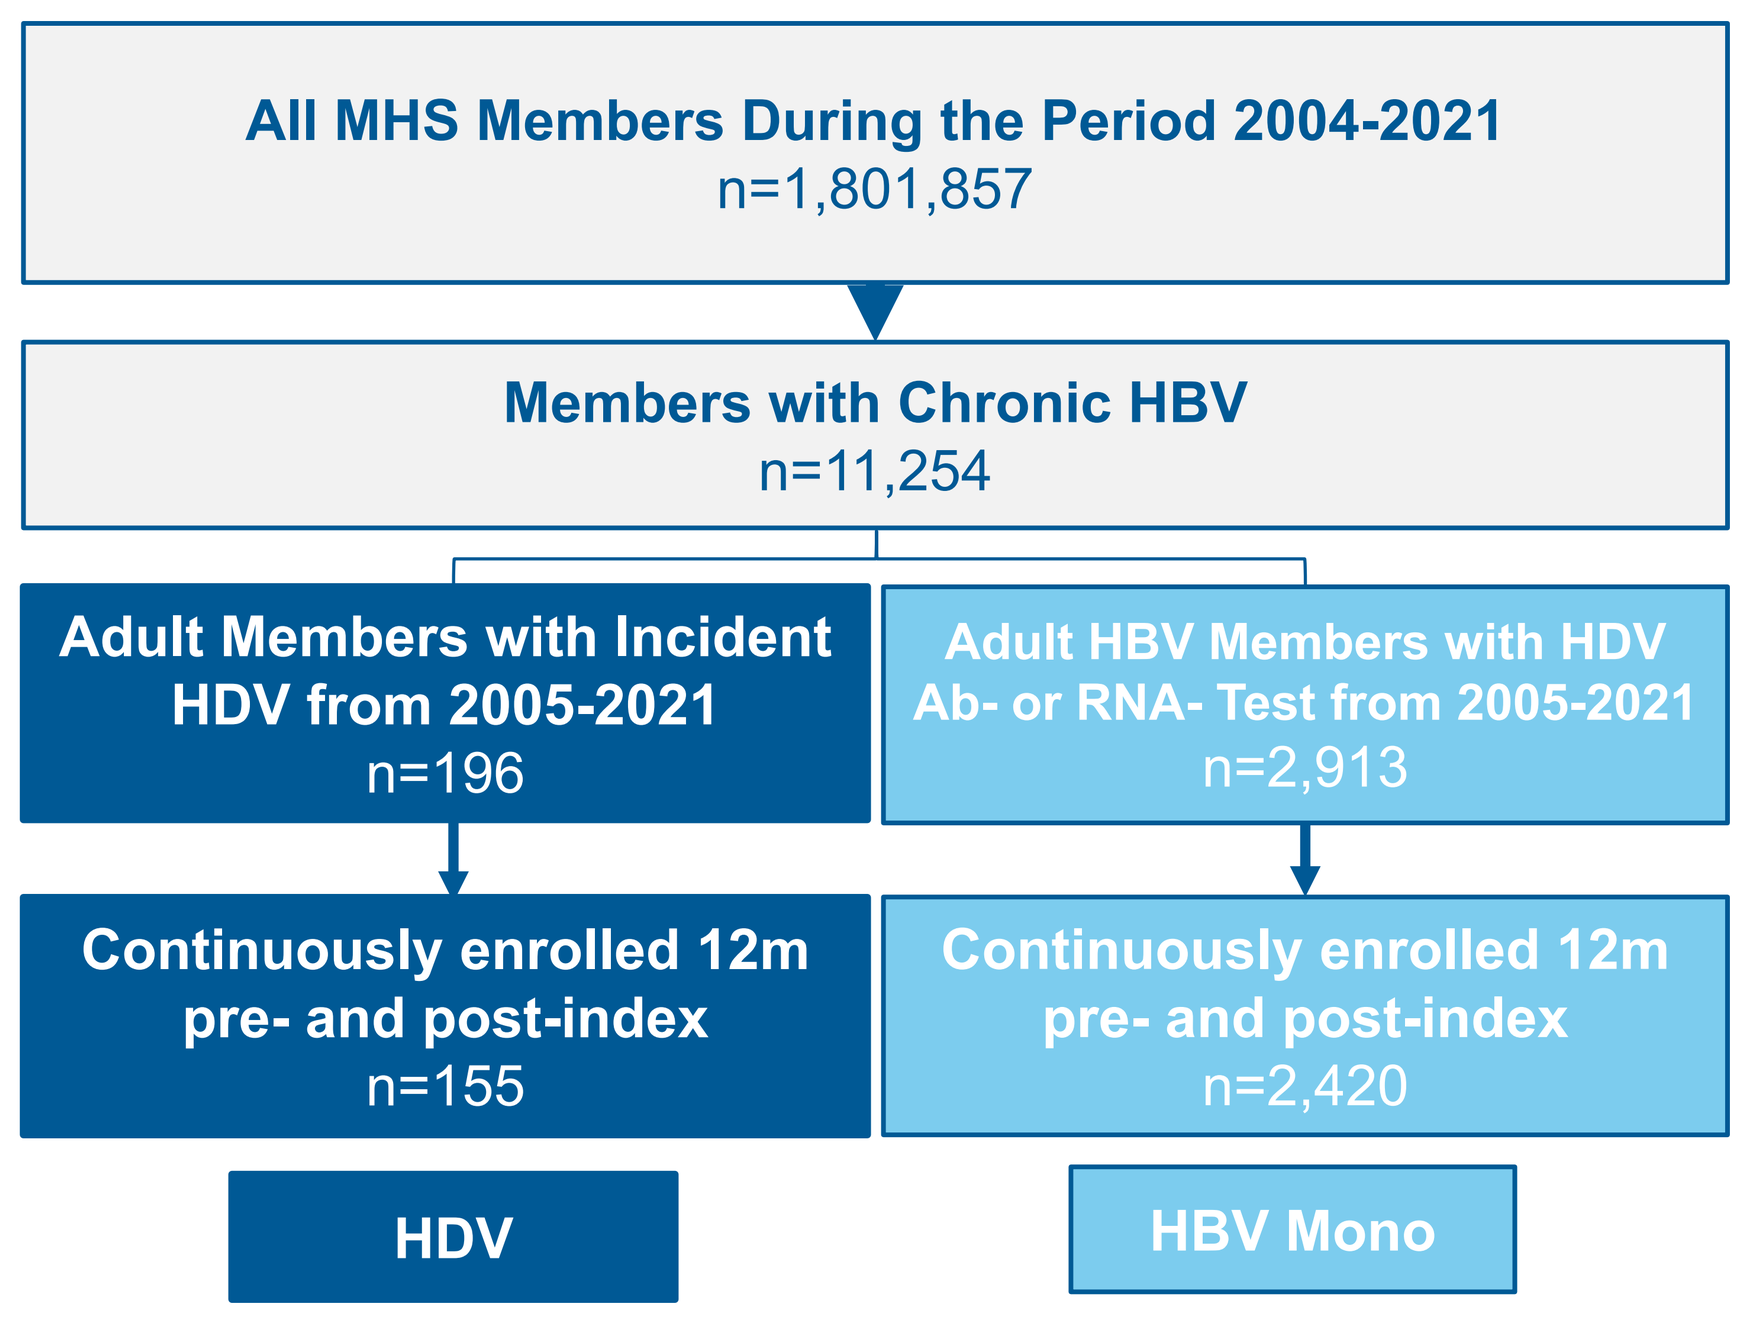

All MHS Members During the Period 2004-2021
n=1,801,857
Members with Chronic HBV
n=11,254
Adult Members with Incident HDV from 2005-2021
n=196
Adult HBV Members with HDV Ab- or RNA- Test from 2005-2021
n=2,913
Continuously enrolled 12m pre- and post-index
n=155
Continuously enrolled 12m pre- and post-index
n=2,420
HDV
HBV Mono

Table 1. Patient Characteristics

Baseline Characteristics	HDV (N=155)	HBV-Mono (N=2,420)	*P*-value
Age, years; median (IQR)	42.5 (33.1, 53.1)	45.0 (34.2, 56.1)	0.13
Male sex, n (%)	81 (52.3%)	1,420 (58.7%)	0.12
Socioeconomic status, n (%)			**0.002**
Low (1-3)	15 (9.7%)	299 (12.4%)	
Mid (4-6)	111 (71.6%)	1,403 (58.0%)	
High (7-10)	28 (18.1%)	715 (29.5%)	
Born in Eastern Europe, n (%)	77 (49.7%)	1,019 (42.1%)	**0.023**
Disease state (most severe state at index), n (%)			**<0.001**
Non-cirrhosis	136 (87.7%)	2,337 (96.6%)	
Compensated cirrhosis	10 (6.5%)	45 (1.9%)	
Decompensated cirrhosis	8 (5.2%)	24 (1.0%)	
Hepatocellular carcinoma	1 (0.6%)	8 (0.3%)	
Liver transplant	0	6 (0.2%)	
Comorbidities, n (%)			
Cancer	9 (5.8%)	155 (6.4%)	0.8
Cardiovascular disease	10 (6.5%)	163 (6.7%)	0.9
Diabetes	11 (7.1%)	197 (8.1%)	0.6
Chronic kidney disease	14 (9.0%)	254 (10.5%)	0.6
Hypertension	28 (18.1%)	534 (22.1%)	0.2
Human immunodeficiency virus	7 (4.5%)	10 (0.8%)	**<0.001**
Hepatitis C virus RNA+	9 (5.8%)	41 (1.7%)	**0.002**
Drug dependence	10 (6.5%)	41 (1.7%)	**<0.001**
Charlson Comorbidity Index, mean (SD)	1.1 (1.8)	1.0 (1.5)	0.8
0	79 (51.0%)	1,250 (51.7%)	0.6
1	42 (27.1%)	627 (25.9%)	
2	11 (7.1%)	253 (10.5%)	
3	10 (6.5%)	135 (5.6%)	
4+	13 (8.4%)	155 (6.4%)	

Economic Burden

- Patients with HDV had more inpatient (IP) visits (20.6% vs 12.0%; *P*=0.002), and a longer median length of stay for IP visits compared to patients with HBV-Mono (7.0 vs 3.0 days; *P*=0.0001) (**Figure 3**).
- Patients with HDV patients had significantly higher mean IP costs PPPY ($2,240 vs $718; *P*<0.001) and mean total costs PPPY ($5,293 vs $3,859; *P*=0.048) compared to patients with HBV-Mono, respectively (**Figure 4**).

Figure 3. Median HCRU PPPY in the 12-Month Period Post-index Date Among Patients with HDV and HBV-Mono

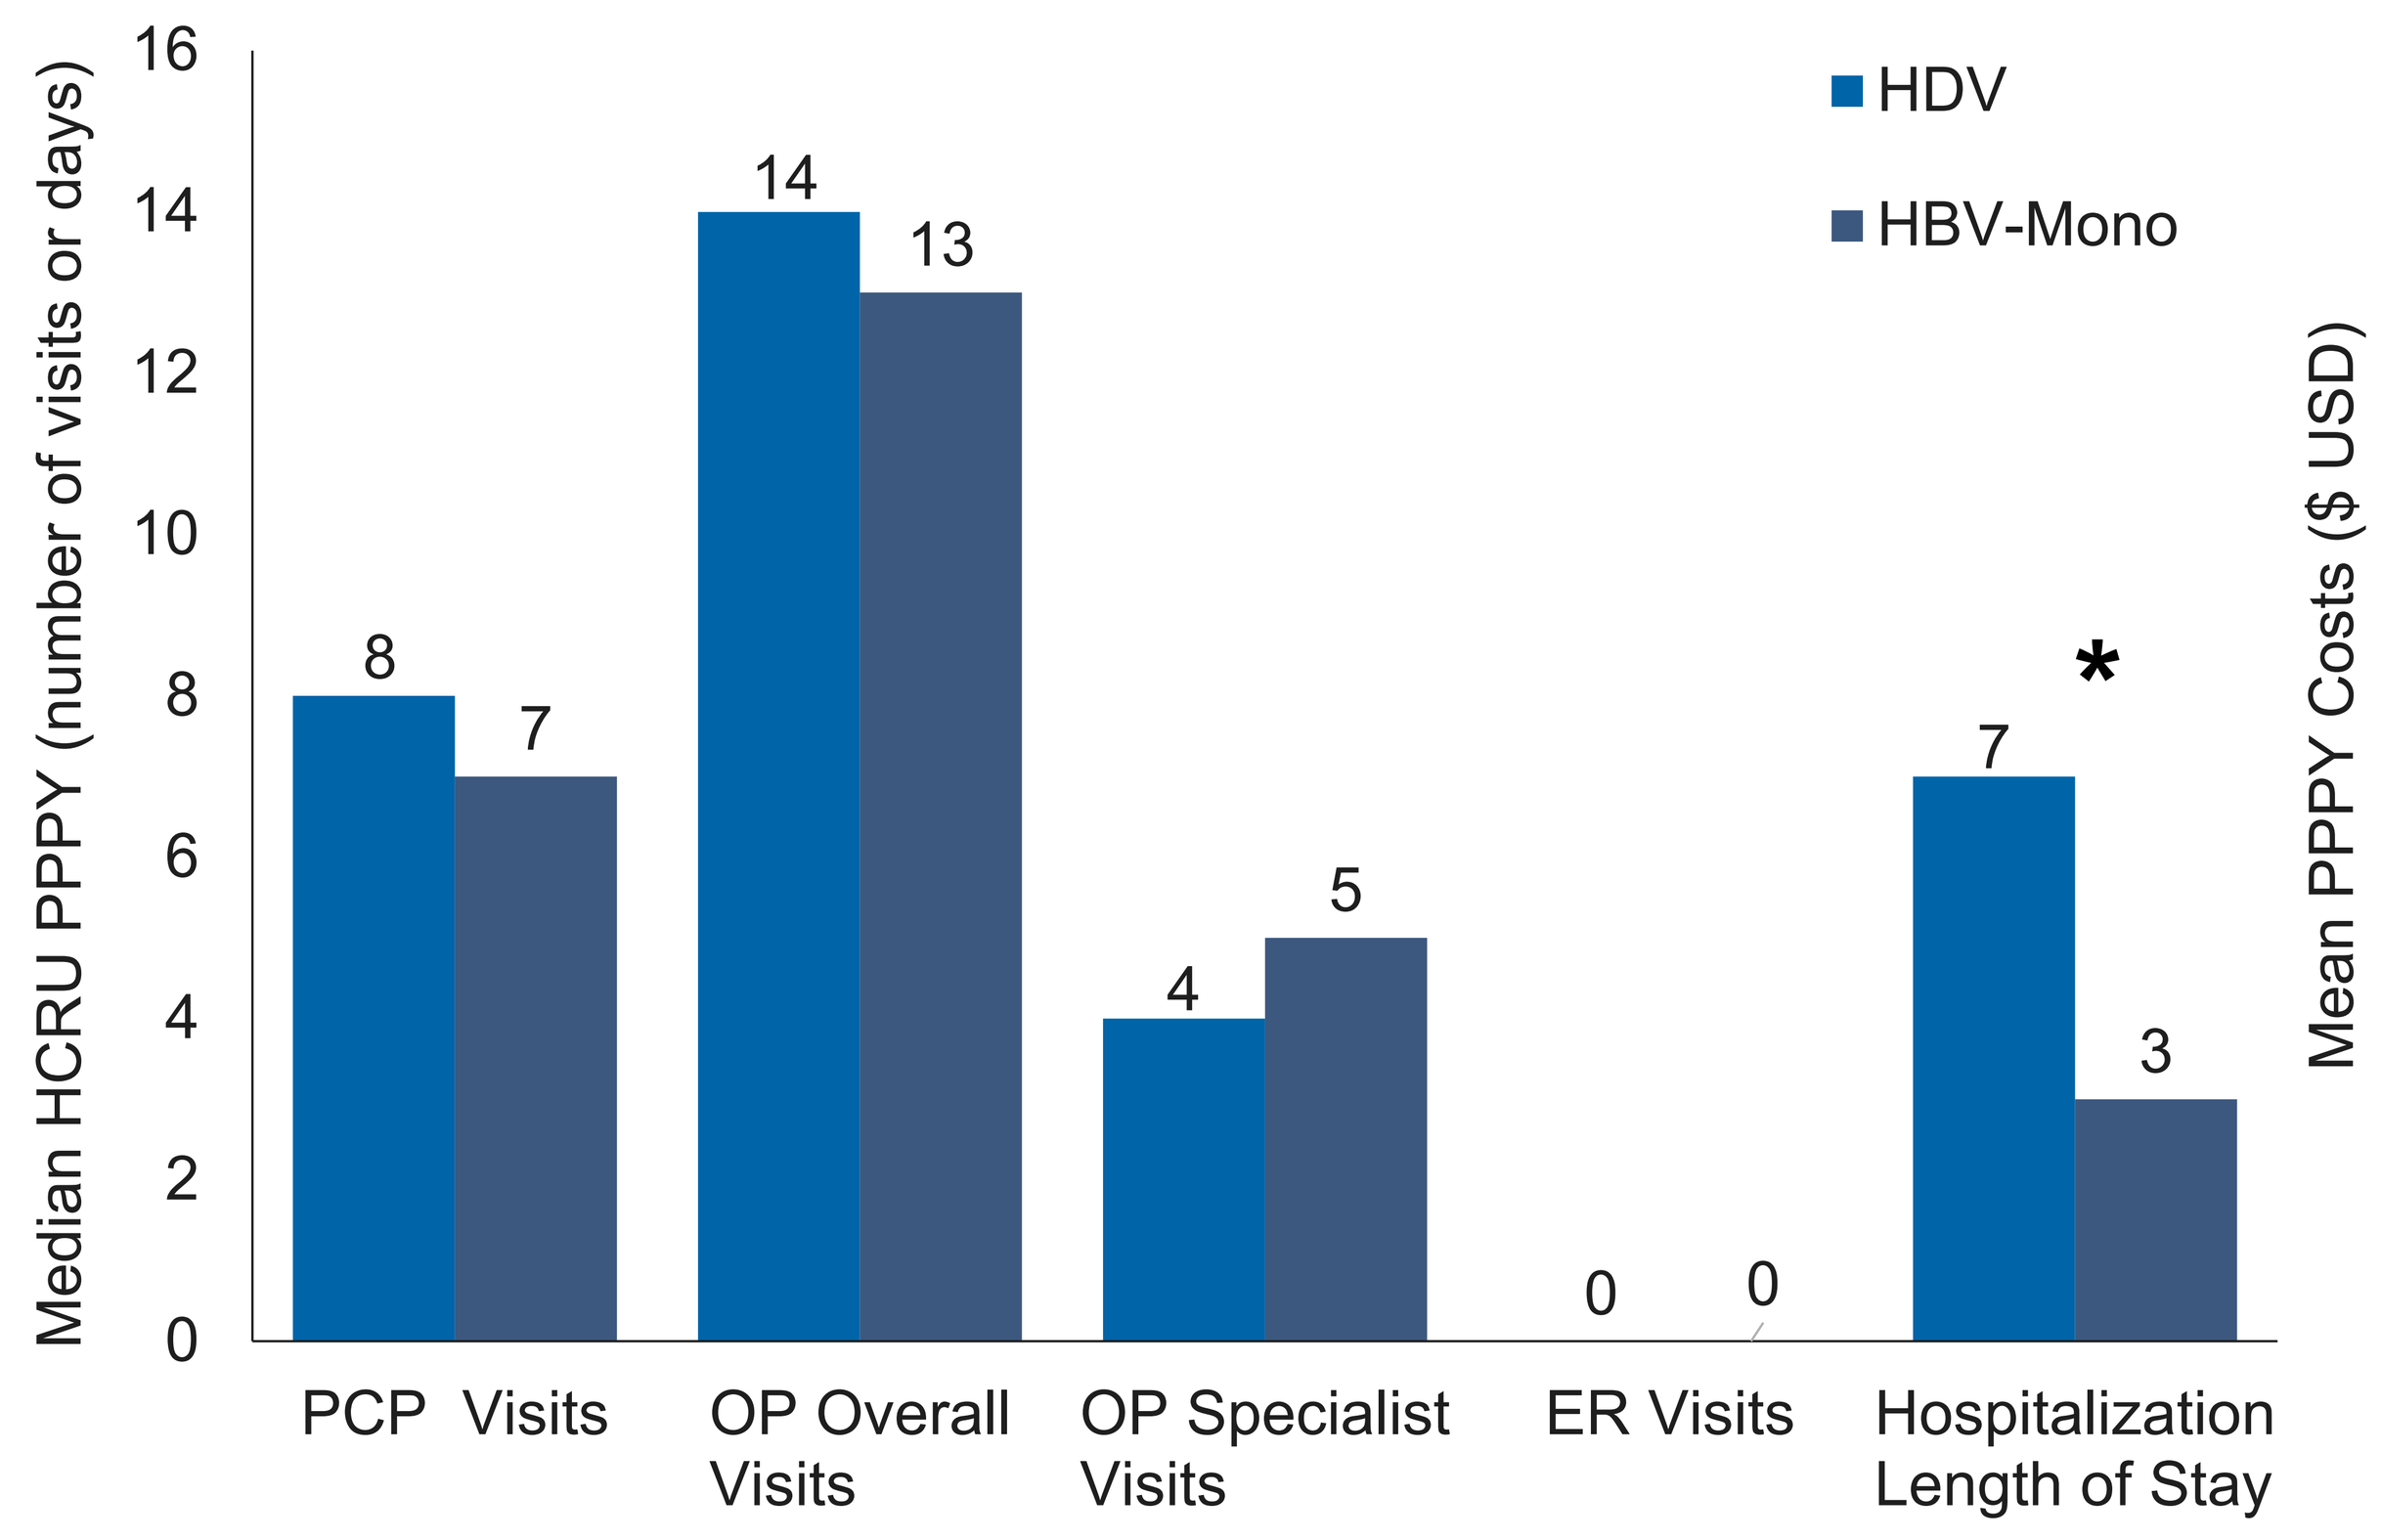

Median HCRU PPPY (number of visits or days)
0
2
4
6
8
10
12
14
16
HDV
HBV-Mono
8
7
14
13
4
5
0
0
7
3
*
PCP Visits
OP Overall Visits
OP Specialist Visits
ER Visits
Hospitalization Length of Stay

*p-value < 0.05 for comparisons.

Figure 4. Mean PPPY Costs in the 12-Month Period Post-Index Among Patients with HDV and HBV-Mono ($ USD)

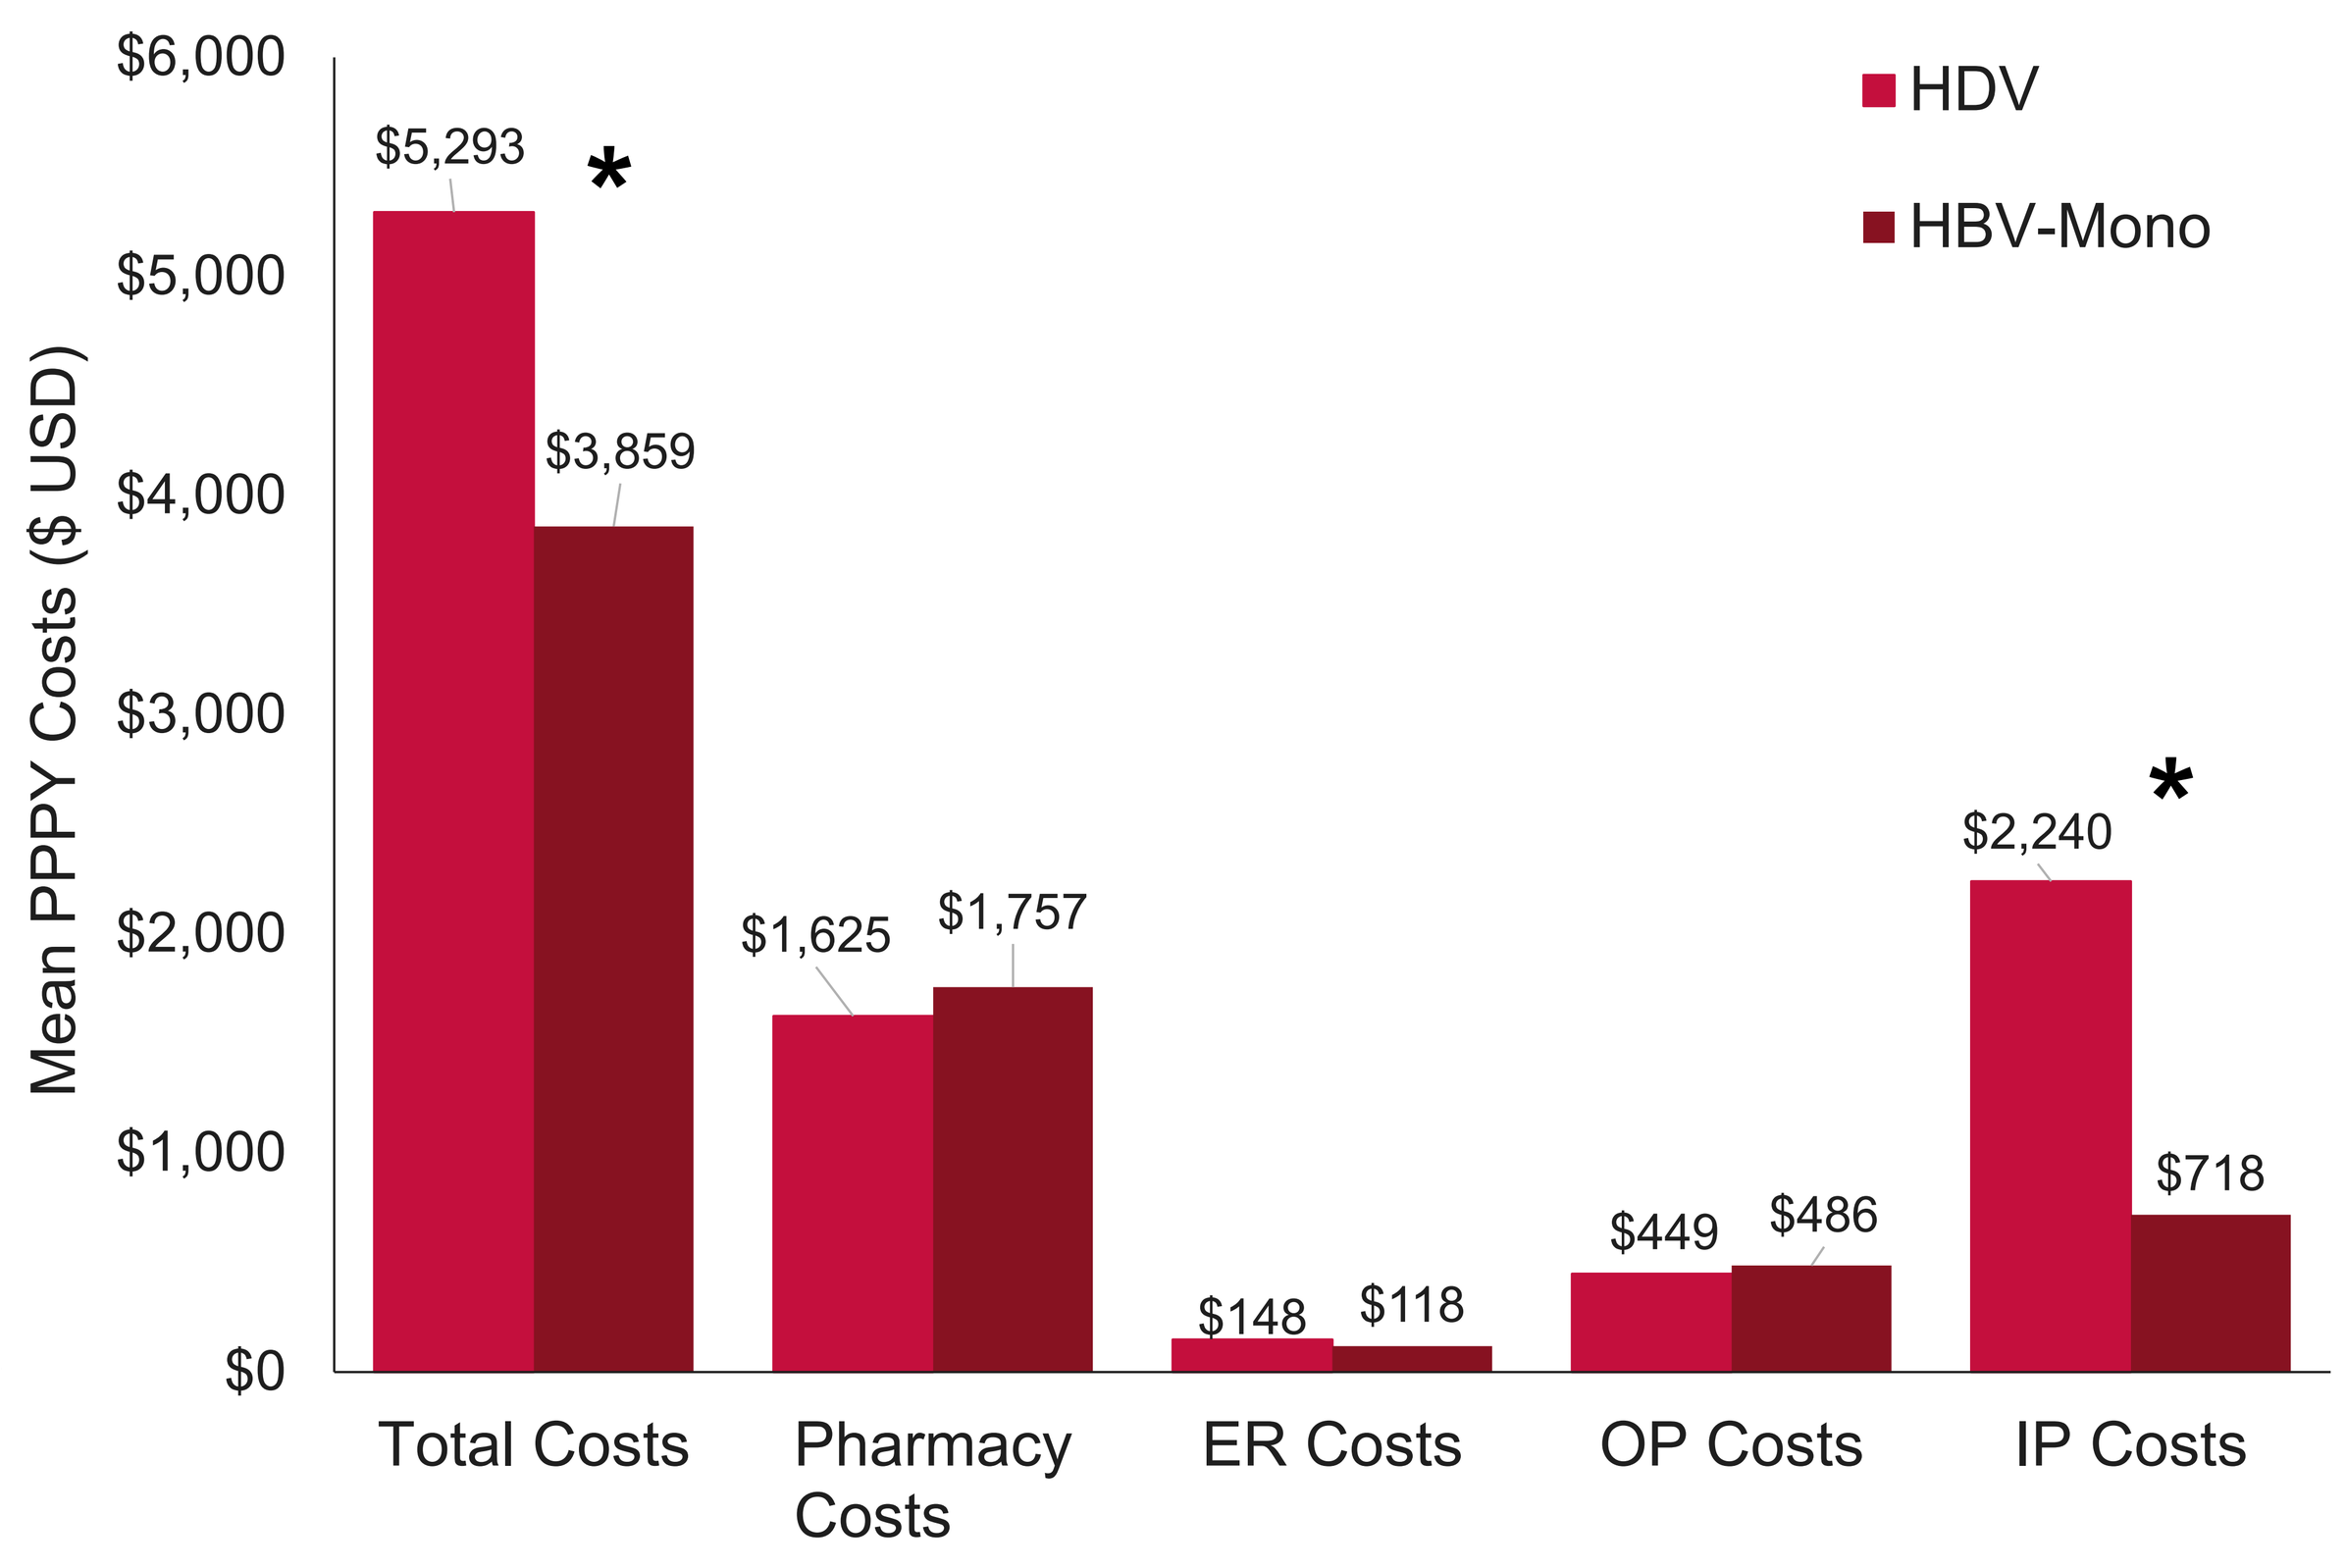

Mean PPPY Costs ($ USD)
$0
$1,000
$2,000
$3,000
$4,000
$5,000
$6,000
HDV
HBV-Mono
$5,293
$3,859
*
$1,625
$1,757
$148
$118
$449
$486
$2,240
$718
*
Total Costs
Pharmacy Costs
ER Costs
OP Costs
IP Costs

*p-value < 0.05 for comparisons.

Limitations

- The limitations of any retrospective claims study apply. Diagnoses made via ICD-9-CM codes are subject to miscoding and can lead to misclassification bias, and time of diagnosis may not correspond to the time of infection; therefore, results may reflect delayed HCRU and costs.
- Hospitalization costs were calculated using the Israeli Ministry of Health's standard per diem unit costs at public hospitals; this approach may not account for patient-level differences in intensity of care; and costs may not be generalizable to other countries and care settings.

Abbreviations ER, emergency room; HBV, hepatitis B virus; HCRU, healthcare resource utilization; HDV, hepatitis delta virus; IP, inpatient; MHS, Maccabi Healthcare Services; OP, outpatient; PCP, primary care practitioner; PCR, polymerase chain reaction; PPPY, per patient per year; Q, quartile

References **1. Menta N et al.** *J Adv Res* 2019;17:3-15. **2. Maio Z et al**. *J Infect Dis* 2020;221:1677-1687. **3. Stockdale AJ et al.** *J Hepatol* 2020;73:523-532.

Acknowledgments: This study was funded by Gilead Sciences, Inc., Foster City, CA, USA. This study was sponsored by Gilead Sciences, Inc. Medical writing and editorial support were provided by Izabela Aleksanderek, PhD, of Maple Health, and funded by Gilead Sciences, Inc.

Disclosures: **RH** reports consulting fees from Gilead Sciences, Inc. **MR** and **CK** are employees of Gilead Sciences, Inc., and may own stock in Gilead Sciences, Inc. **OTL** and **YG** are employees of Gilead Sciences Israel, Ltd., and may own stock in Gilead Sciences, Inc. **CW**, **MH**, and **SG** report no conflicts of interest..

ISPOR EU: November 17-20, 2024; Barcelona, Spain