CARE PATHWAY OF LUNG CANCER AND MELANOMA PATIENTS TREATED WITH IMMUNE CHECKPOINT INHIBITORS (ICI) IN HOSPITAL AT HOME AND ADMINISTRATION COSTS IN FRANCE IN 2021 AND 2022

HSD46

Anne-Claire Toffart[1], Gaëtan Casanova[2], Morgane Pierre[3], Hervé Lemasson[3], Véronique Moreau-Mallet[3], Nicolas Pagès[4], Ronan Jolivel[4], Arnaud Panes[4], Mélanie Chartier[3], Maurice Pérol[5]

1 CHU Grenoble Alpes - CS 10217 - 38043 Grenoble Cedex 9, France, 2 APHP, 55 boulevard Diderot 75610 Paris Cedex 12, France, 3 Bristol Myers Squibb (BMS), 3 rue Joseph Monier 92500 Rueil-Malmaison, France
4 HEVA, département pharmaco-épidémiologie, 186 avenue Thiers, 69465 Lyon Cedex 06, France, 5 Centre de Lutte Contre le Cancer de Léon Bérard, 28 rue Laennec 69008 Lyon, France

Objectives

For almost 10 years, ICI are administered as short-term infusions every 2 to 6 weeks, mainly in Outpatient Hospitals (OH). As the use of ICIs increases, OH are facing an increase in the number of patients to be treated, making reception capacities difficult.

In France, Hospitalization At Home (HAH) system allows patients to receive their ICI in a familiar, less stressful atmosphere, without fatigue from traveling and waiting at the hospital.

For patients living far from hospitals or having experience minimal ICI side effects, in good general condition and therapeutic response, HAH is an opportunity to improve ICI patient access and ease patient burdens.

Guidelines from the French Society of Cancer Immunotherapy support ICI use in HAH settings[1].

In this context, the study objective was to describe the role and cost of HAH in the ICI administration to lung cancer and melanoma patients in France in 2021 and 2022.

Methods

This retrospective observational cohort study was conducted in France using data from French national hospital databases (Medical Information Systems Program (PMSI) related to Medicine-Surgery-Obstetrics (MCO) and Hospital-at-home (HAH) activities).

All adult patients who received at least one immunotherapy treatment (nivolumab alone or in combination with ipilimumab, ipilimumab alone, pembrolizumab, atezolizumab, or durvalumab) between January 1, 2021, and December 31, 2022, for lung cancer or melanoma patients and treated in HAH were included. Additionally, the identification of expensive molecules is considered through a specific database names the "FICHCOMP-HAH file".

Melanoma patients were further categorized, based on the "LES code*" of the treatment used, as metastatic, adjuvant, other melanoma or multi-melanoma (when several codes of melanoma identified for the same patient).

A descriptive analysis was conducted to characterize patients, HAH centers, care pathways and administration costs.

In France, a HAH stay is divided in sub-sequences which are characterized by 3 variables: main reason for admission, associated reason for admission and a dependency score (the Karnofsky score). All sub-sequences with the infusion of an ICI (identified by the codification of the LES Code during the subsequence) were identified. HAH stay cost were calculated in the public health insurance perspective with the day rate linked to the combination of the 3 variables.

All costs were described according to French National Health Insurance (NHI) perspective in €2022.

Patient care pathways were analyzed descriptively and by treatment sequences with the TAK® algorithm: Time sequence Analysis K-clustering

The TAK were performed with an alignment of patients to the first HAH stay in the study period. Multi-immunotherapy were excluded for this data-visualization.

**LES code*: Data code affiliated to assets enrolled in the list of expensive and innovative asset paid on top of the DRG-based financing

Results

In 2021-2022, 330 lung cancer patients and 148 melanoma patients received immunotherapy in HAH. Melanoma patient distribution were: 69 metastatic, 42 adjuvant, 34 multi-melanoma and 3 other melanoma.

In lung cancer, the mean age of patients was 66 (±10) years old and 64% (n=211) were men. The mean age of melanoma patients was 64 (±16) years old and 57% (n=84) were men.

In metastatic melanoma, the mean age of patients was 68 (±15) years old and 61% (n=42) were men. In adjuvant melanoma, the mean age of patients was 60 (±14) year old and 52% (n=22) were men (**Table 1**).

Table 1. Characteristics of patients by indication

	N	Mean age (± SD)	Median age [Q1;Q3]	Male N (%)
Lung cancer	330	66 (±10)	67 [59 ; 73]	211 (64)
Total melanoma	148	64 (±16)	65 [53 ; 76]	84 (57)
Metastatic melanoma	69	68 (±15)	70 [57 ; 81]	42 (61)
Adjuvant melanoma	42	60 (±14)	59 [52 ; 70]	22 (52)
Multi melanoma	34	58 (±16)	60 [48 ; 67]	19 (56)
Other melanoma	3	N<11	N<11	N<11

SD: Standard Deviation

Among patients with lung cancer (n=330), the mean Karnofsky score was of 64 (±13). For patients with melanoma (n=148) the mean Karnofsky score was of 64 (±10).

In the case of adjuvant (n=42) and metastatic melanoma (n=69), the mean Karnofsky score was respectively 61 (±9) and 64 (±10).(**Table 2**)

Table 2. Karnofksy score distribution by indication

	N	Mean (SD)	Min	Q1	Med	Q3	Max
Lung cancer	330	64 (13)	20	57	60	75	90
Total melanoma	148	64 (10)	28	60	60	70	90
Metastatic melanoma	69	64 (10)	40	60	60	70	90
Adjuvant melanoma	42	61 (9)	28	60	60	68	83
Multi melanoma	34	67 (9)	50	60	60	79	80
Other melanoma	3	N<11	N<11	N<11	N<11	N<11	N<11
Total	478	64 (12)	20	60	60	70	90

SD : standard Deviation; Med: Median

The mean Karnofsky score was 64 but varied by region.
It was lowest for patients treated in Île-de-France (mean score 58) and highest for patients in Auvergne Rhône-Alpes (mean score 73) (**Figure 1**). There were also regional differences by indication. For example, for lung cancer, the average Karnofsky score was higher in Auvergne-Rhône-Alpes (73) than in Nouvelle-Aquitaine (63) or Ile-de-France (57).

Figure 1. Mean Karnofsky score by region

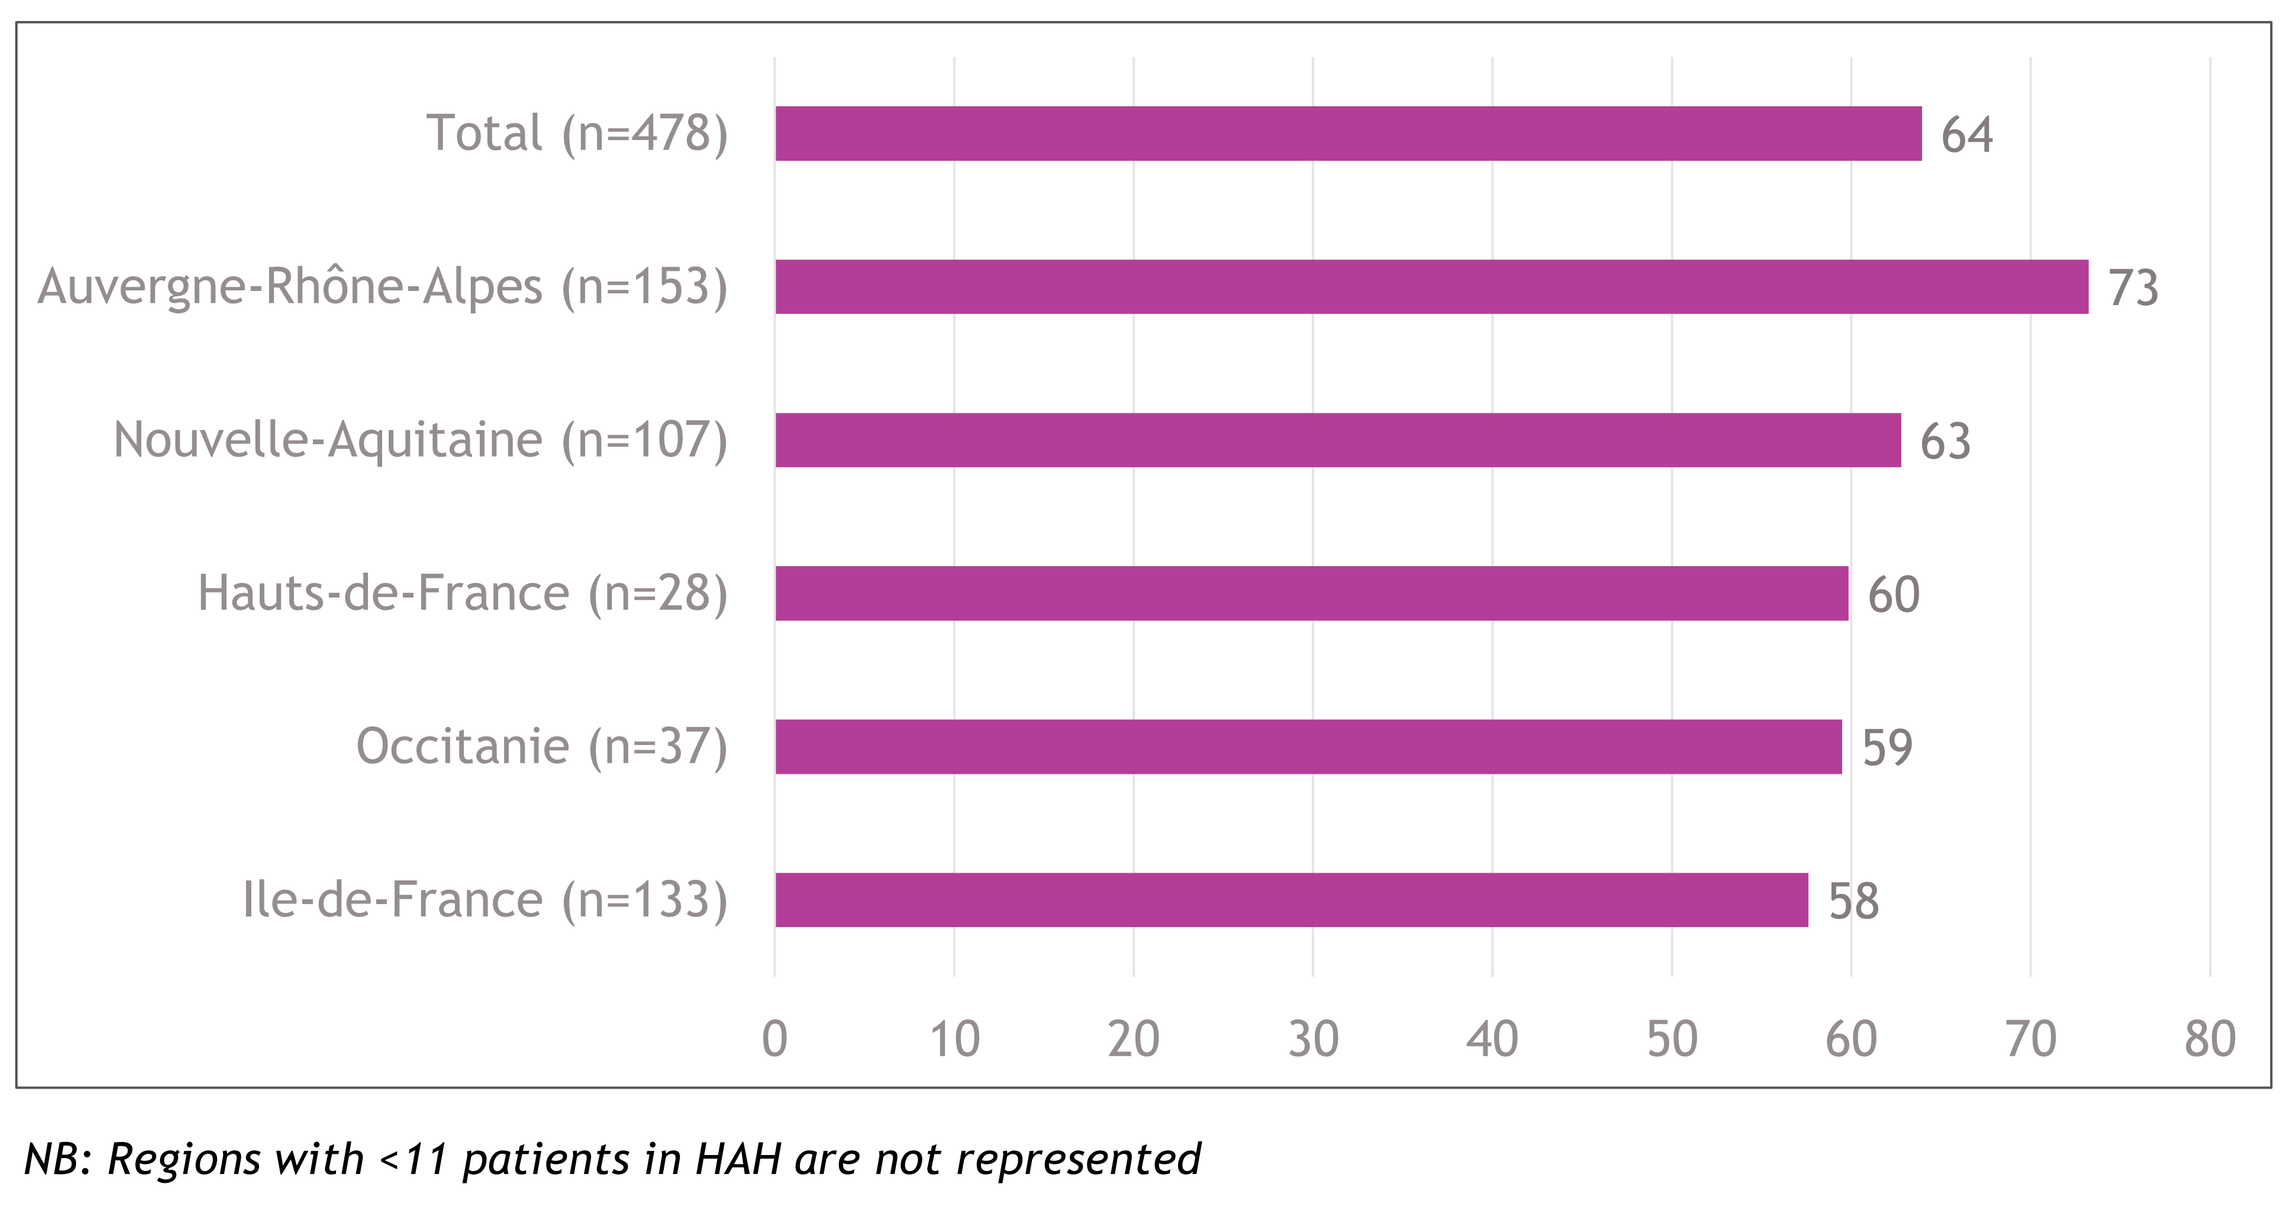
Total (n=478) 64
Auvergne-Rhône-Alpes (n=153) 73
Nouvelle-Aquitaine (n=107) 63
Hauts-de-France (n=28) 60
Occitanie (n=37) 59
Ile-de-France (n=133) 58
0 10 20 30 40 50 60 70 80

NB: Regions with <11 patients in HAH are not represented

HAH and MCO Treatment sequences duration

- **Duration (days) before switching to HAH**

For all patients (n=469), the mean delay between the first MCO and the first HAH administration was 302 days (±400) (approximately 10 months on average).

When considering indications, our results emphasized that patients treated in a context of adjuvant melanoma were transferred faster in HAH (107 days (±79) in average; 3.5 months) than patients treated for a metastatic melanoma (381 days (±433); 12.5 months) or lung cancer (304 days (±342) ; 10.0 months).The mean number of administrations per patient was 14 (±9) in the context of lung cancer. This mean number was 15 (±9) for metastatic melanoma and 9 (±4) for adjuvant melanomas.

Regional variations in HAH delays (2 to 22 months on average) were observed, with Hauts-De-France as the minimum and Auvergne-Rhône-Alpes the maximum.

- **Duration spent in HAH**

For all patients (n=330), the mean delay between the first and last HAH administration was 242 days (±242) (approximately 8 months on average).

Regions were relatively homogeneous with around 206 (±171) to 263 days (±253) (7 to 9 months).

The average Karnofsky score was different between regions for the same indication. For instance, in lung cancer, it ranged from 55 in Occitanie to 73 in Auvergne-Rhône-Alpes, suggesting a different profile of patients treated in HAH (**Figure 2**).

The average length of stay did not seem to be correlated with the Karnofsky index of patients.

Figure 2 presents the distribution of treatment sequences, delays between first MCO and first HAH administration, and between first and last HAH administration, by indications and regions alongside with mean Karnofsky score.

Figure 2. Average duration (in days) before and after the first administration of immunotherapy in HAH by region and by indication over the study period.

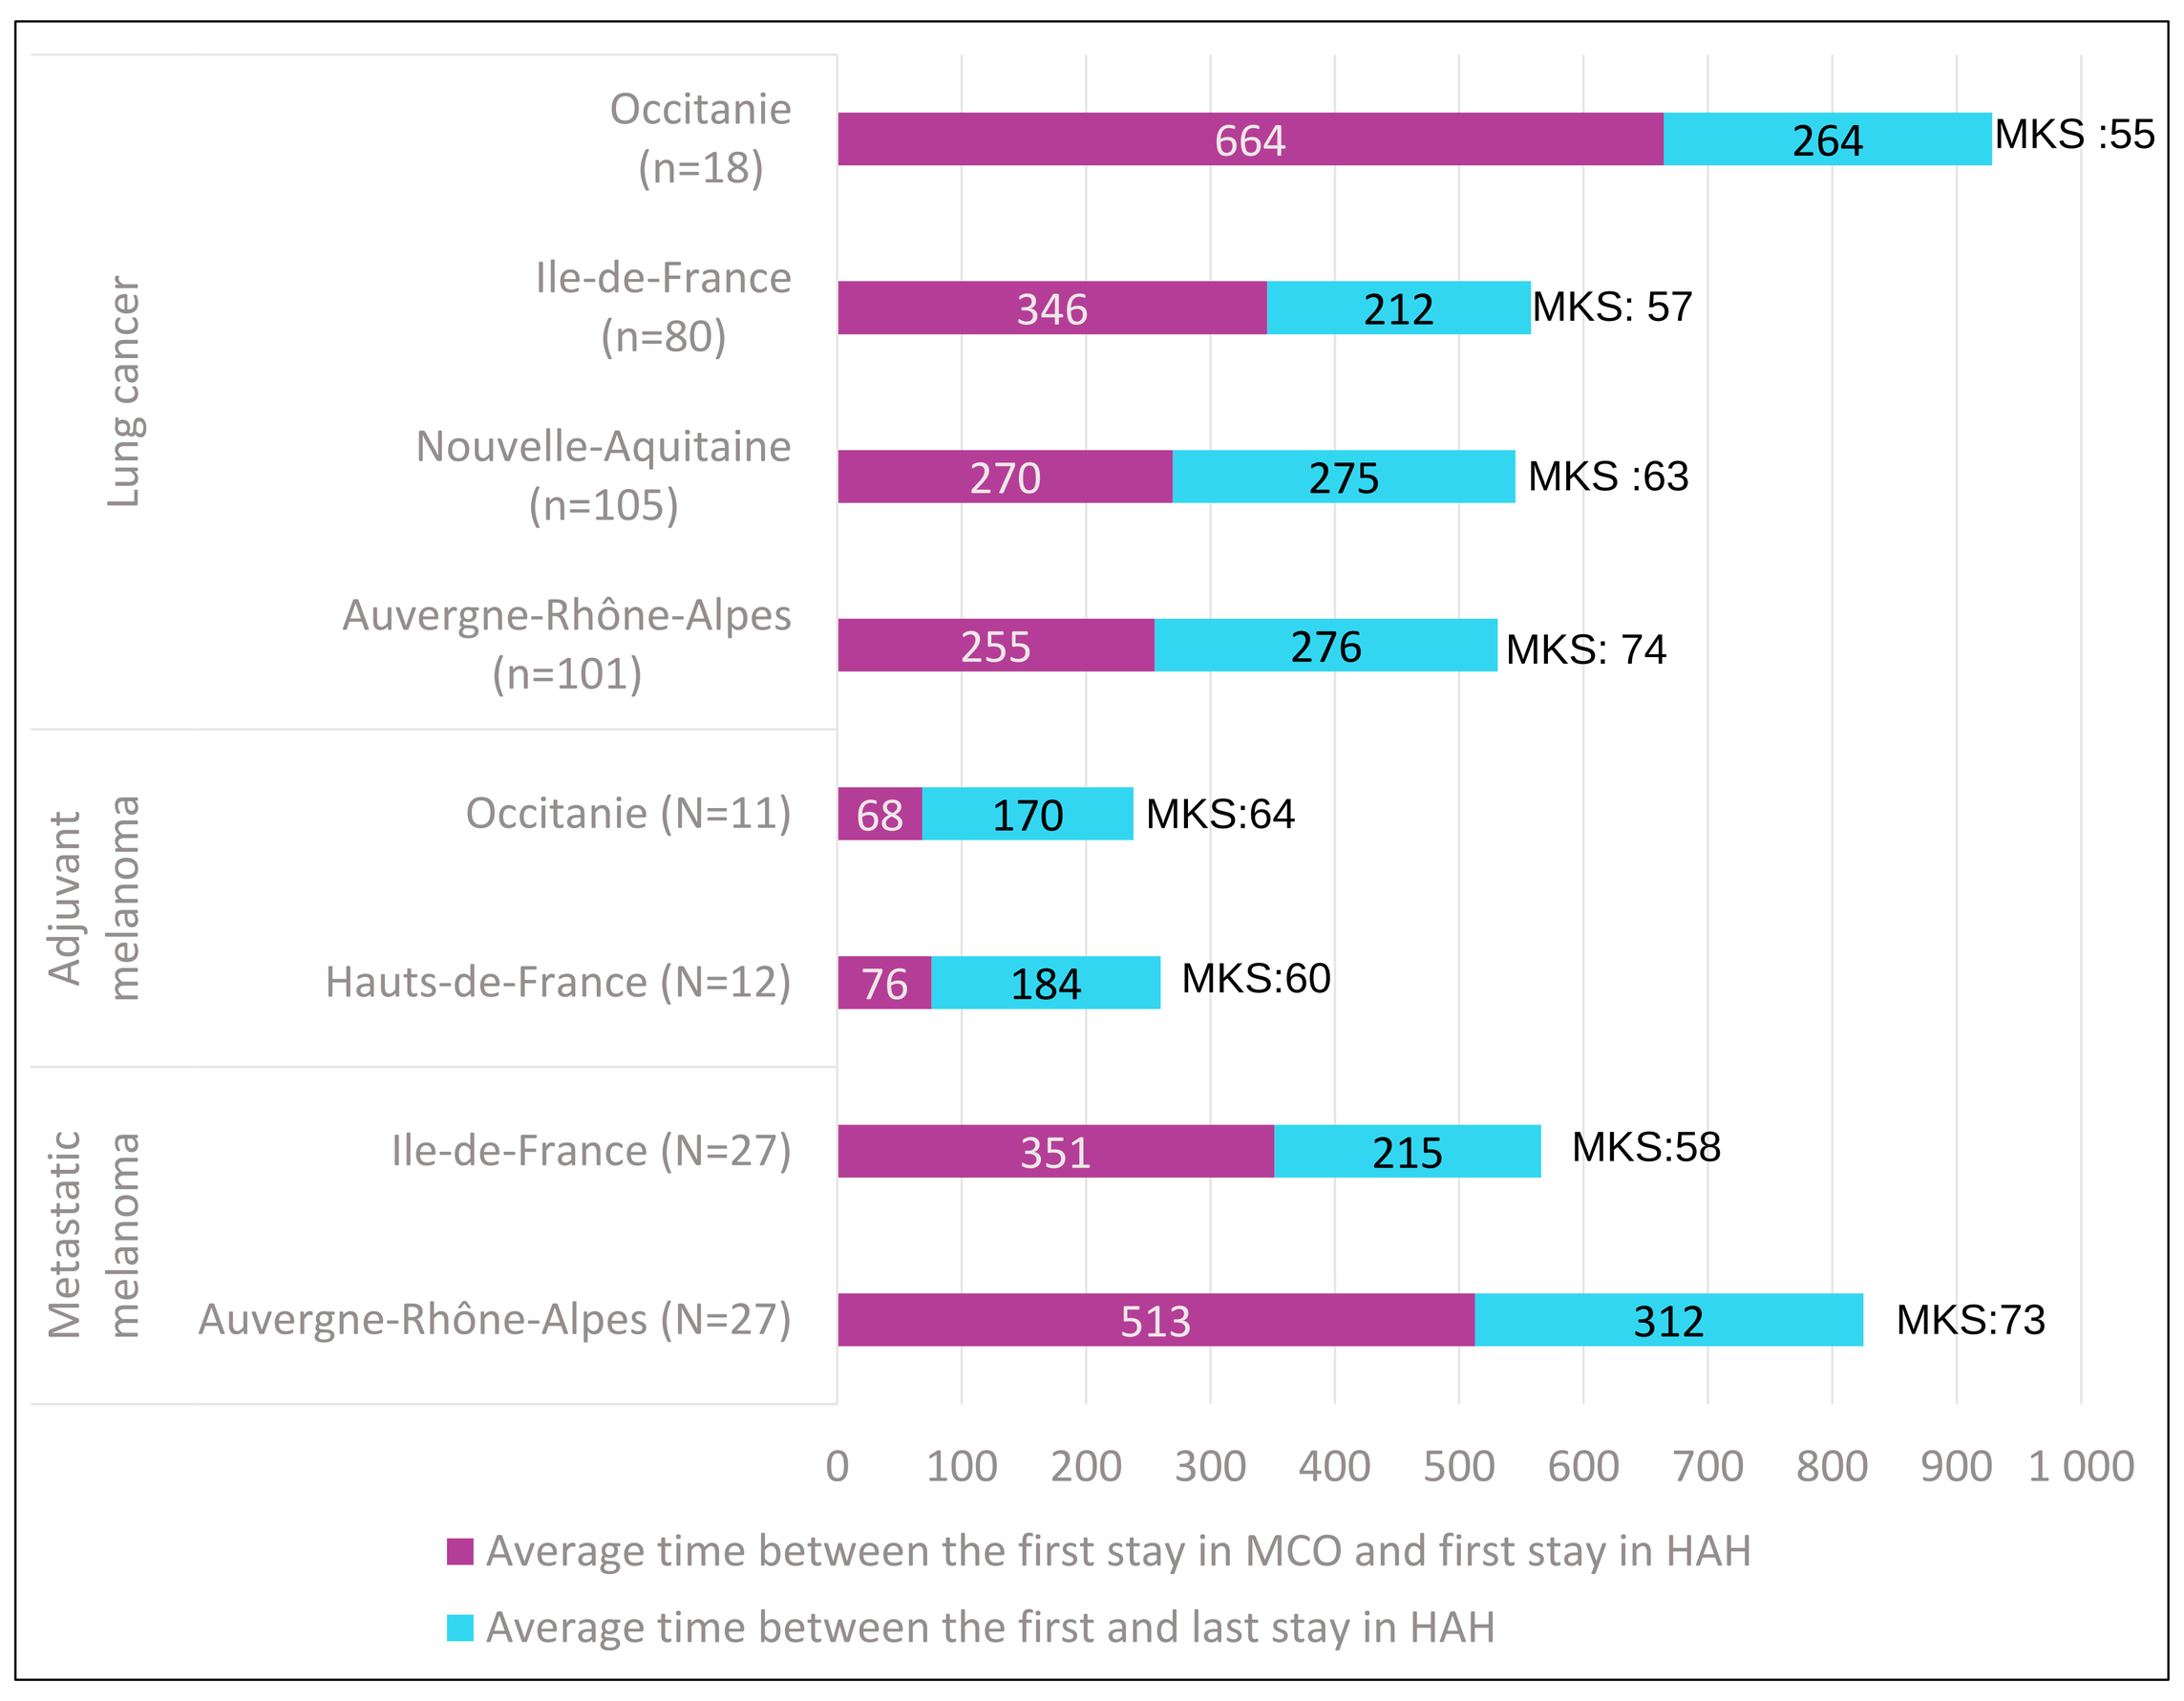
Lung cancer
Occitanie (n=18) 664 264 MKS :55
Ile-de-France (n=80) 346 212 MKS: 57
Nouvelle-Aquitaine (n=105) 270 275 MKS :63
Auvergne-Rhône-Alpes (n=101) 255 276 MKS: 74
Adjuvant melanoma
Occitanie (N=11) 68 170 MKS:64
Hauts-de-France (N=12) 76 184 MKS:60
Metastatic melanoma
Ile-de-France (N=27) 351 215 MKS:58
Auvergne-Rhône-Alpes (N=27) 513 312 MKS:73
0 100 200 300 400 500 600 700 800 900 1 000
Average time between the first stay in MCO and first stay in HAH
Average time between the first and last stay in HAH

MKS: Mean Karnofsky score/ NB: Regions with <11 staff are not represented

Cost of immunotherapies administration in HAH

For all sub-sequences related to an ICI infusion the average cost was 331€ (±785€) for an administration. The mean cost was higher for other melanomas (444€; ±98€) and lung cancers (346€; ±901€). Median costs were very similar across indications, at 263€ for lung cancer, €262 for melanoma and 262€ overall (**Table 3**).

Table 3. Cost of administration in HAH setting, by indication, in euros

	Nb sub-sequences	Mean	STD	Min	Q1	Med	Q3	Max
Lung cancer	2,594	**346**	901	134	226	263	317	35,167
Total melanoma	836	**284**	79	153	226	262	263	904
Metastatic melanoma	490	**290**	86	153	226	262	296	904
Multi melanoma	168	**262**	49	226	226	262	263	487
Adjuvant melanoma	166	**278**	65	226	262	262	263	731
Other melanoma	12	**444**	98	306	342	451	489	685
Total	3,430	**331**	785	134	226	262	317	35,167

An analysis of sub-sequences number according to their distribution were also performed. The duration of a subsequence was mostly of one day (80.3%), sometimes two days (in 17.6% of cases) and rarely 3 days (1.0%) or more (1.1%).

The average cost of sub-sequences lasting 1 day was €247 (± €48), €446 (± €60) for those lasting 2 days, €798 (± €159) for those lasting 3 days, and finally €4080 (± €6246) when they lasted more than 3 days. The average cost of administering an ICI increased as the number of coded days increased. Therefore, coding a sub-sequence more than a day has a direct impact on the cost. (**Table 4**).

Table 4. Duration of Sub-Sequences (SS) and corresponding costs (€)

	SS with 1 day duration		SS with 2 days duration		SS with 3 days duration		SS > 3 days duration	
	Nb of SS N(%)	Mean Costs (€) (±SD)	Nb of SS N(%)	Mean Costs (€) (±SD)	Nb of SS N(%)	Mean Costs (€) (±SD)	Nb of SS N(%)	Mean Costs (€) (±SD)
Total (N=3,430)	2,753 (80.3)	247 (±48)	605 (17.6)	446 (±60)	33 (1.0)	798 (±159)	39 (1.1)	4,080 (±6,246)
By indications								
Lung cancer (N=2,594)	2,044 (78.8)	244 (±53)	482 (18.6)	450 (±58)	31 (1.2)	801 (±164)	37 (1.4)	4,258 (±6,364)
Total Melanoma (N=836)	709 (84.8)	255 (±24)	123 (14.7)	432 (±66)	2 (0.0)	N<11	2 (0.0)	N<11
• Metastatic melanoma (N=490)	394 (80.4)	254 (±26)	94 (19.2)	427 (±68)	1 (0.0)	N<11	1(0.0)	N<11
• Multi melanoma (N=168)	160 (95.2)	252 (±21)	8 (4.8)	N<11	0	-	0	-
• Adjuvant melanoma (N=166)	155 (93.3)	262 (±23)	10 (6.0)	N<11	1 (0.0)	N<11	0	-
• Other melanoma (N=12)	0	-	11 (91.7)	422 (±69)	0	-	1 (8.3)	N<11

Treatment sequences in lung cancer

Treatment sequences are only presented for lung cancer due to the small number of patient with melanoma indications.

When considering all the immunotherapy drugs collectively, the time before the first HAH does not seem to impact HAH patterns. 59.1% (195/329) of patients experienced HAH over the entire follow-up period, while 40.9% (135/329) had at least one switch to MCO.

Overall, patients on durvalumab and pembrolizumab mostly stayed in HAH over the entire HAH follow-up. Patients on nivolumab more often switched to MCO than other treatments (**Table 5**). Treatment pattern after the first HAH dose differed a lot between patients, even under the same therapy (**Figure 3**).

Table 5. Health care pathway in MCO and HAH of lung cancer patients

	HAH only over the entire follow-up N (%)	At least one switch to MCO N (%)	All
Pembrolizumab	130 (62,5)	78 (37,5)	208
Durvalumab	33 (80,5)	8 (19,5)	41
Nivolumab	11 (27,5)	29 (72,5)	40
Atezolizumab	18 (50)	18 (50)	36
All	192 (59,1)	133 (40,9)	325

Figure 3. Treatment sequences in lung cancer (N=325)

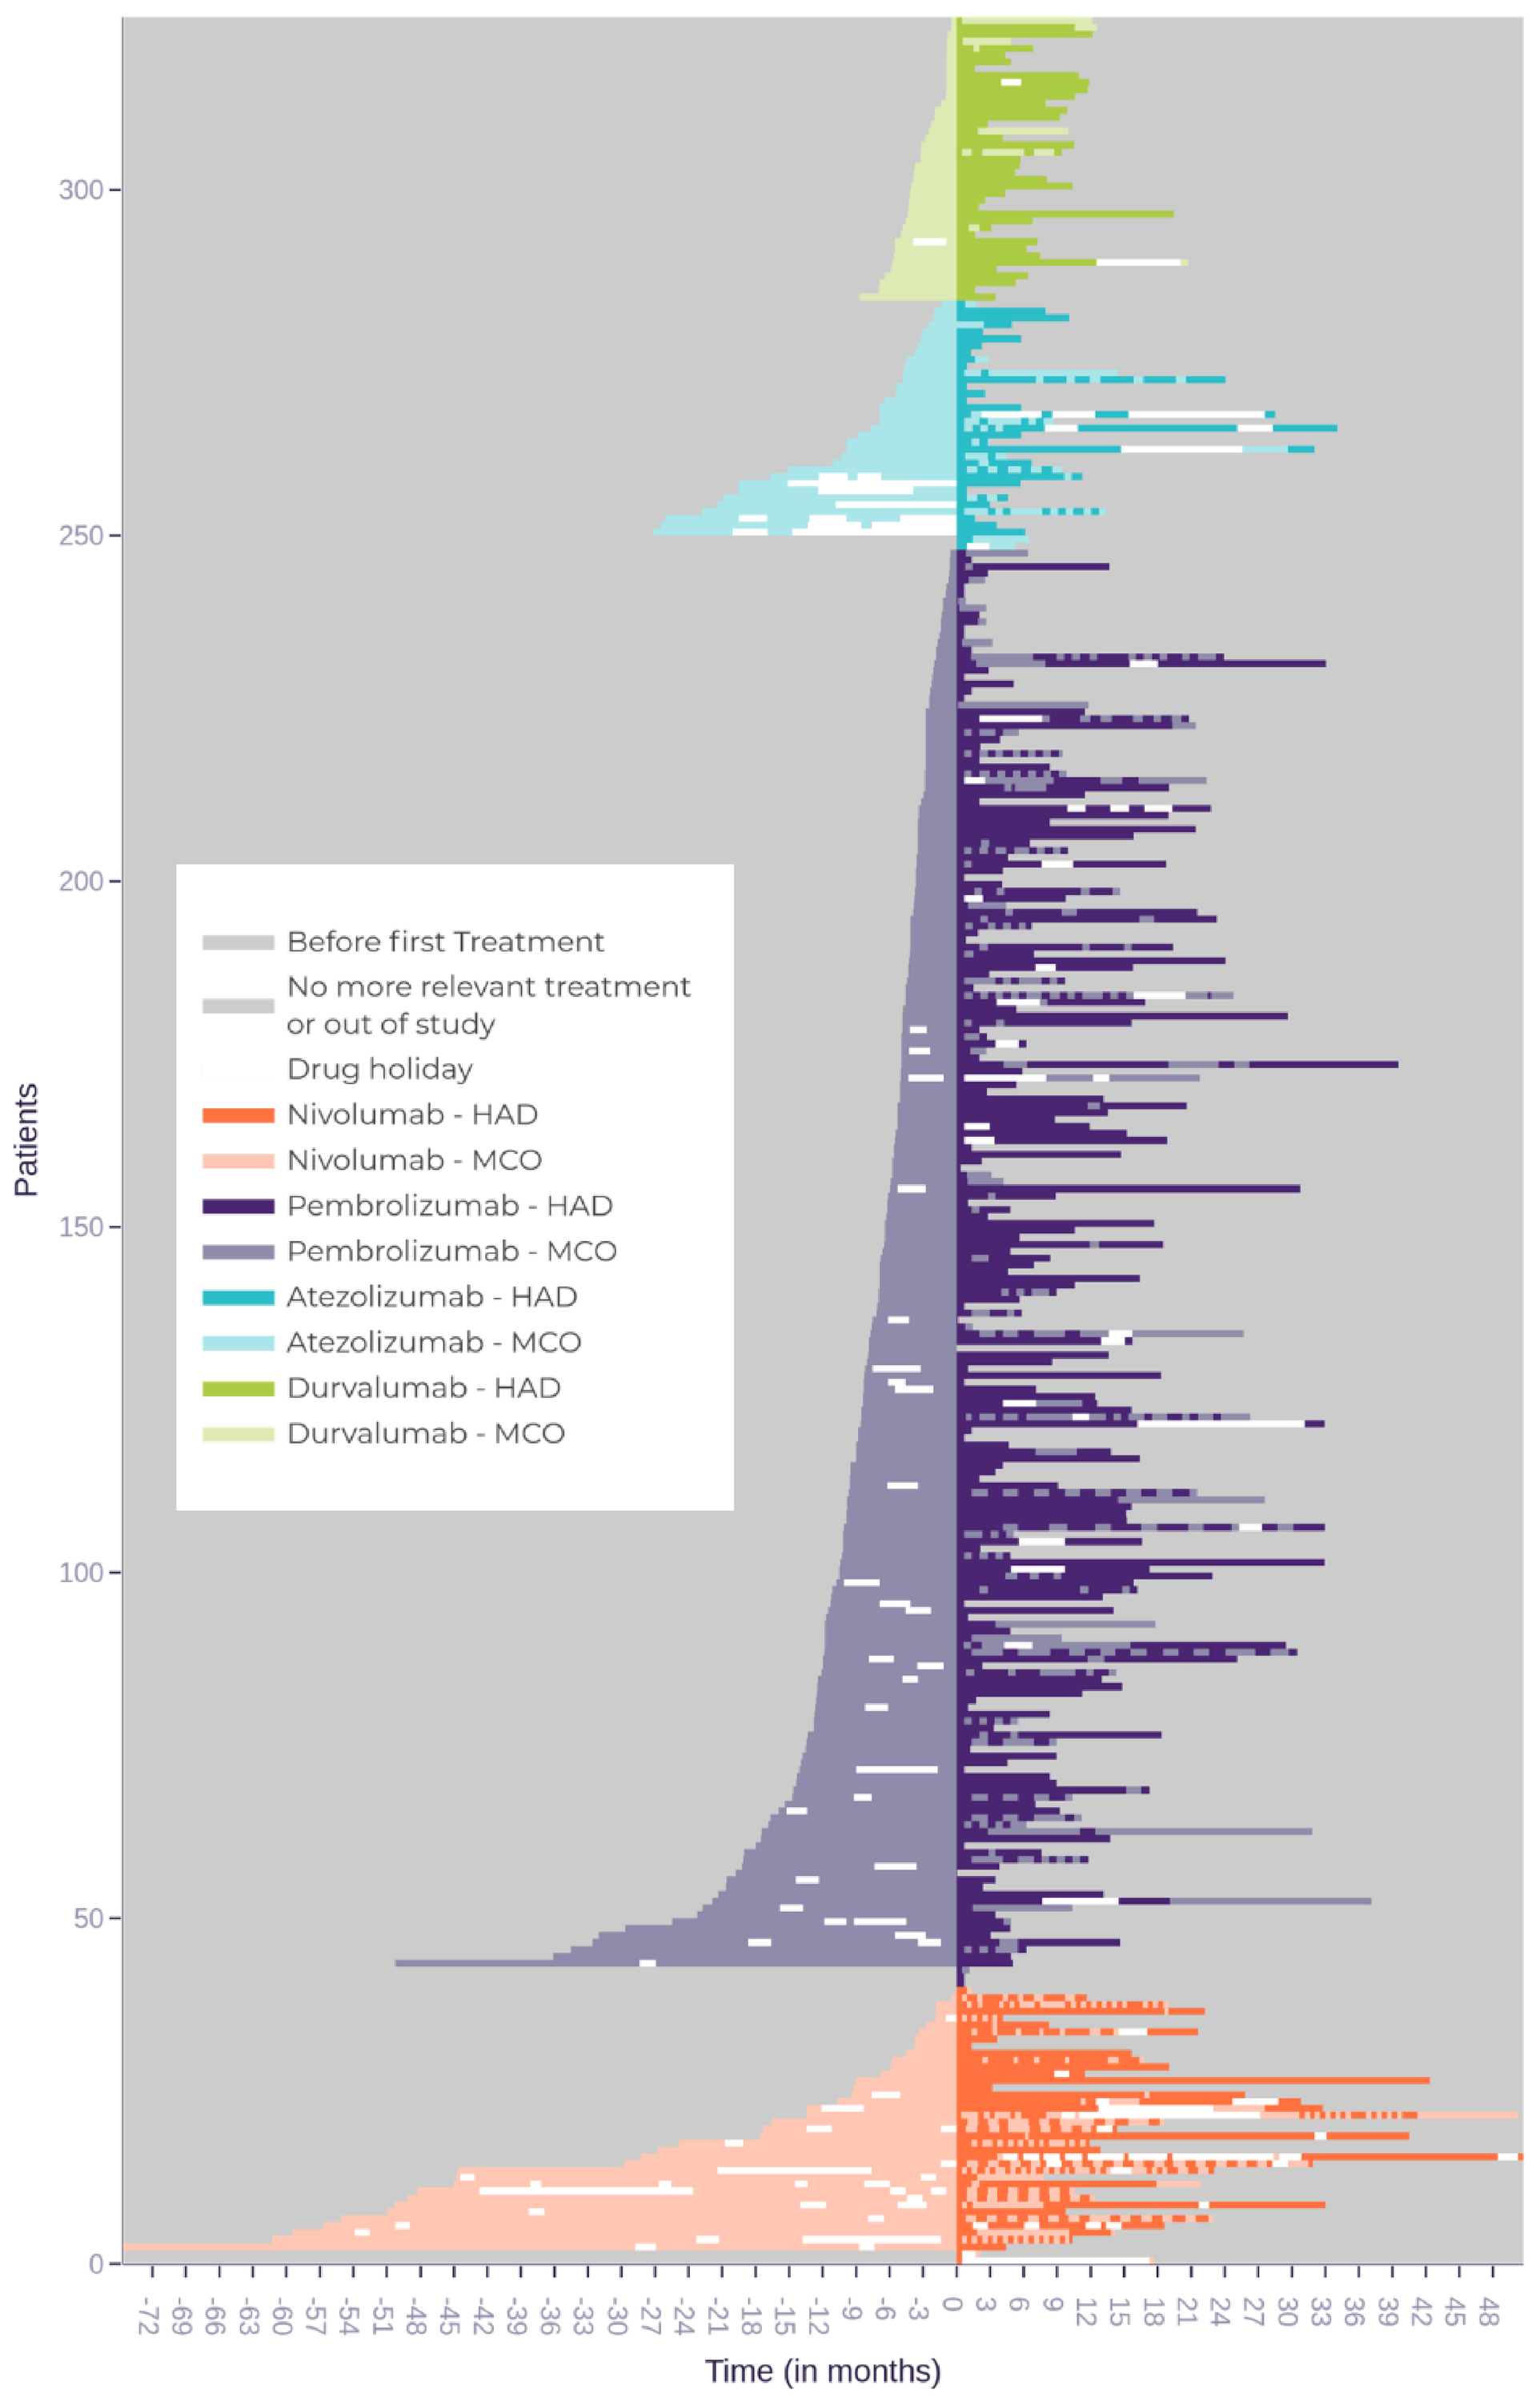
Patients
300
250
200
150
100
50
0
Before first Treatment
No more relevant treatment or out of study
Drug holiday
Nivolumab - HAD
Nivolumab - MCO
Pembrolizumab - HAD
Pembrolizumab - MCO
Atezolizumab - HAD
Atezolizumab - MCO
Durvalumab - HAD
Durvalumab - MCO
-72 -69 -66 -63 -60 -57 -54 -51 -48 -45 -42 -39 -36 -33 -30 -27 -24 -21 -18 -15 -12 -9 -6 -3 0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48
Time (in months)

How to read the TAK :

Each patient is represented by a line, their follow-up is represented from left to right, so that their initialization at the HAH takes place at t=0. Patients are ordered according to their treatment sequence after their first HAH infusion.
Hospital stays administrations that occur prior to the first HAH infusion are displayed for informational purposes but not considered for alignment. The colors represent the treatment (molecule and infusion method) received by the patient.

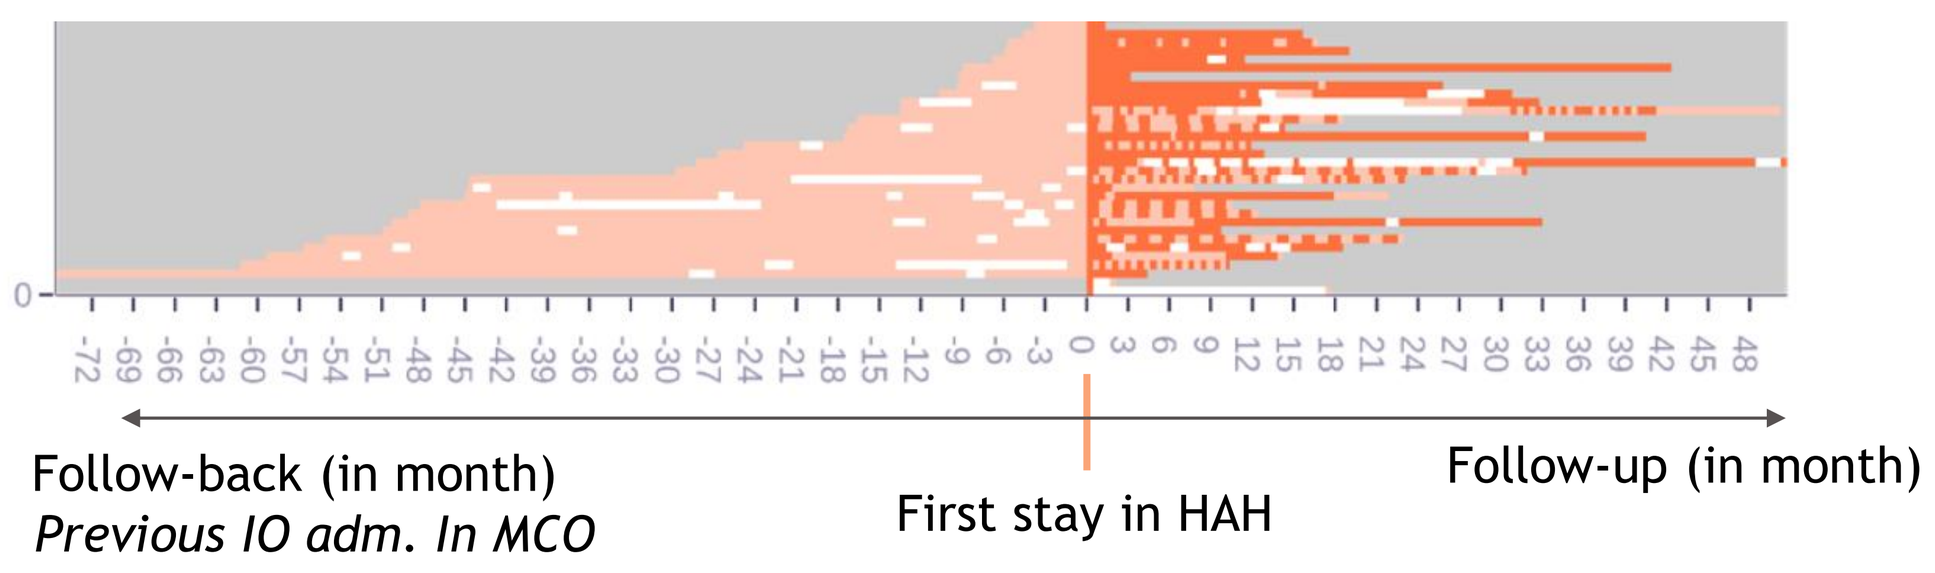
Follow-back (in month)
Previous IO adm. In MCO
First stay in HAH
Follow-up (in month)

Conclusion

- In 2021-2022, HAH ICI administration consumption shows a variety of patients care pathways.
- Time to HAH varied between 2 to 22 months and the average time spent in HAH was 7 to 9 months and no apparent correlation with indication or molecule.
- The possibilities for developing HAH must be explored in order to meet the government's objective of developing home administrations.
- Finally, there is a need to structure and harmonize the ICI infusion care pathway in HAH.

References

1. https://www.fitcancer.fr/wpcontent/uploads/2020/12/FITC_Recommandations_immunotherapies_a_domicile.pdf

Acknowledgments

This study was funded by Bristol Myers Squibb. MP, HL, MC and VMM are employed by Bristol Myers Squibb. NP, AN and RJ are employed by HEVA and received funding from Bristol Myers Squibb for the conduct of this study. ACT, MP and GC were compensated by Bristol Myers Squibb for their contribution to this study.

All authors contributed to and approved this poster.

Presented at ISPOR Europe 2024; November 17-20, 2024; Barcelona, Spain

Email: <morgane.pierre@bms.com>

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.