Acceptance code: **HSD11**

PATIENT JOURNEY MAPPING OF THE GENE THERAPY IN HAEMOPHILIA B IN SPAIN – THE BHEMOGEN PROJECT

Pablo Q[1], Montoro JB[2], Poveda JL[3], Gilabert Perramon A[4], Herrera Arroyo C[5], Álvarez MT[6], Bonanad S[3], Rodríguez López M[7], García Barcenilla S[6], García-Diego DA[8], Gómez de Agüero Sánchez I[9], Gil A[10], Blanco Gayoso M[9]

[1]Virgen del Rocío University Hospital, Sevilla, Spain, [2]Hospital Universitario de la Vall d´Hebrón, Barcelona, Spain, [3]Hospital Universitari i Politècnic La Fe, Valencia, Spain, [4]Consortium of Health and Social Care of Catalonia, Barcelona, Spain, [5]Reina Sofía General University Hospital, Cordoba, Andalusia, Spain, [6]La Paz University Hospital-IdiPaz, Madrid, Spain, [7]Álvaro Cunqueiro University Hospital, Vigo, Spain, [8]FedHemo, Madrid, Spain, [9]CSL Behring, Barcelona, Spain, [10]Omakase Consulting S.L., Barcelona, B, Spain.

BACKGROUND AND OBJECTIVES

- **Haemophilia B** is a rare, inherited bleeding disorder characterized by an increased bleeding tendency due to a partial or complete deficiency of clotting factor IX (FIX).
- **The approval of the gene therapy for the treatment of Haemophilia B** represents a **paradigm shift in the management of this disease**. This disruptive innovation presents challenges and opportunities for healthcare systems, professionals and patients that must be addressed in a practical and holistic way to ensure that this therapeutic option is accessible to candidate patients with Haemophilia B in Spain. Despite the considerable experience in treating Haemophilia B and the recent introduction of gene therapy in a different indication, the literature shows notable absences of patient care roadmaps for neither Haemophilia B nor treatment with gene therapy in any indication.
- The objective of this project is to **develop a patient journey for the first gene therapy in Haemophilia B in Spain** from a multidisciplinary point of view and to **generate knowledge on practical and contextual aspects** affecting its introduction and practical management by identifying challenges and proposing solutions.

METHODS

Literature review to identify current care pathway, challenges and practical experiences derived from the incorporation of gene therapy for other indications. **Constitution of a multidisciplinary group of ten experts** (haematologists, nurses, hospital pharmacists, regional specialists in advanced therapies, hospital managers, representatives of regional health services and representatives of patient associations) from five Autonomous Communities with experience in the management of Haemophilia B and gene therapy, including CSUR centres and reference centres for this pathology. **Design of the patient care roadmap** based on individual interviews, group discussion and consensus, including differences and actions required *versus* the current care pathway for Haemophilia B. Obtaining **scientific endorsements** from Scientific Societies and Patient Associations.

RESULTS

PROPOSED PATIENT CARE ROADMAP - GENE THERAPY IN HAEMOPHILIA B IN SPAIN

- The patient care roadmap for gene therapy in Haemophilia B designed and agreed by the group of experts covers all different stages from diagnosis to long-term follow-up of the patient (Figure 1).
- Its design is based on the current Haemophilia B patient care roadmap, identifying the necessary organizational, structural and care changes specifically related to this new therapeutic option (Figure 1).

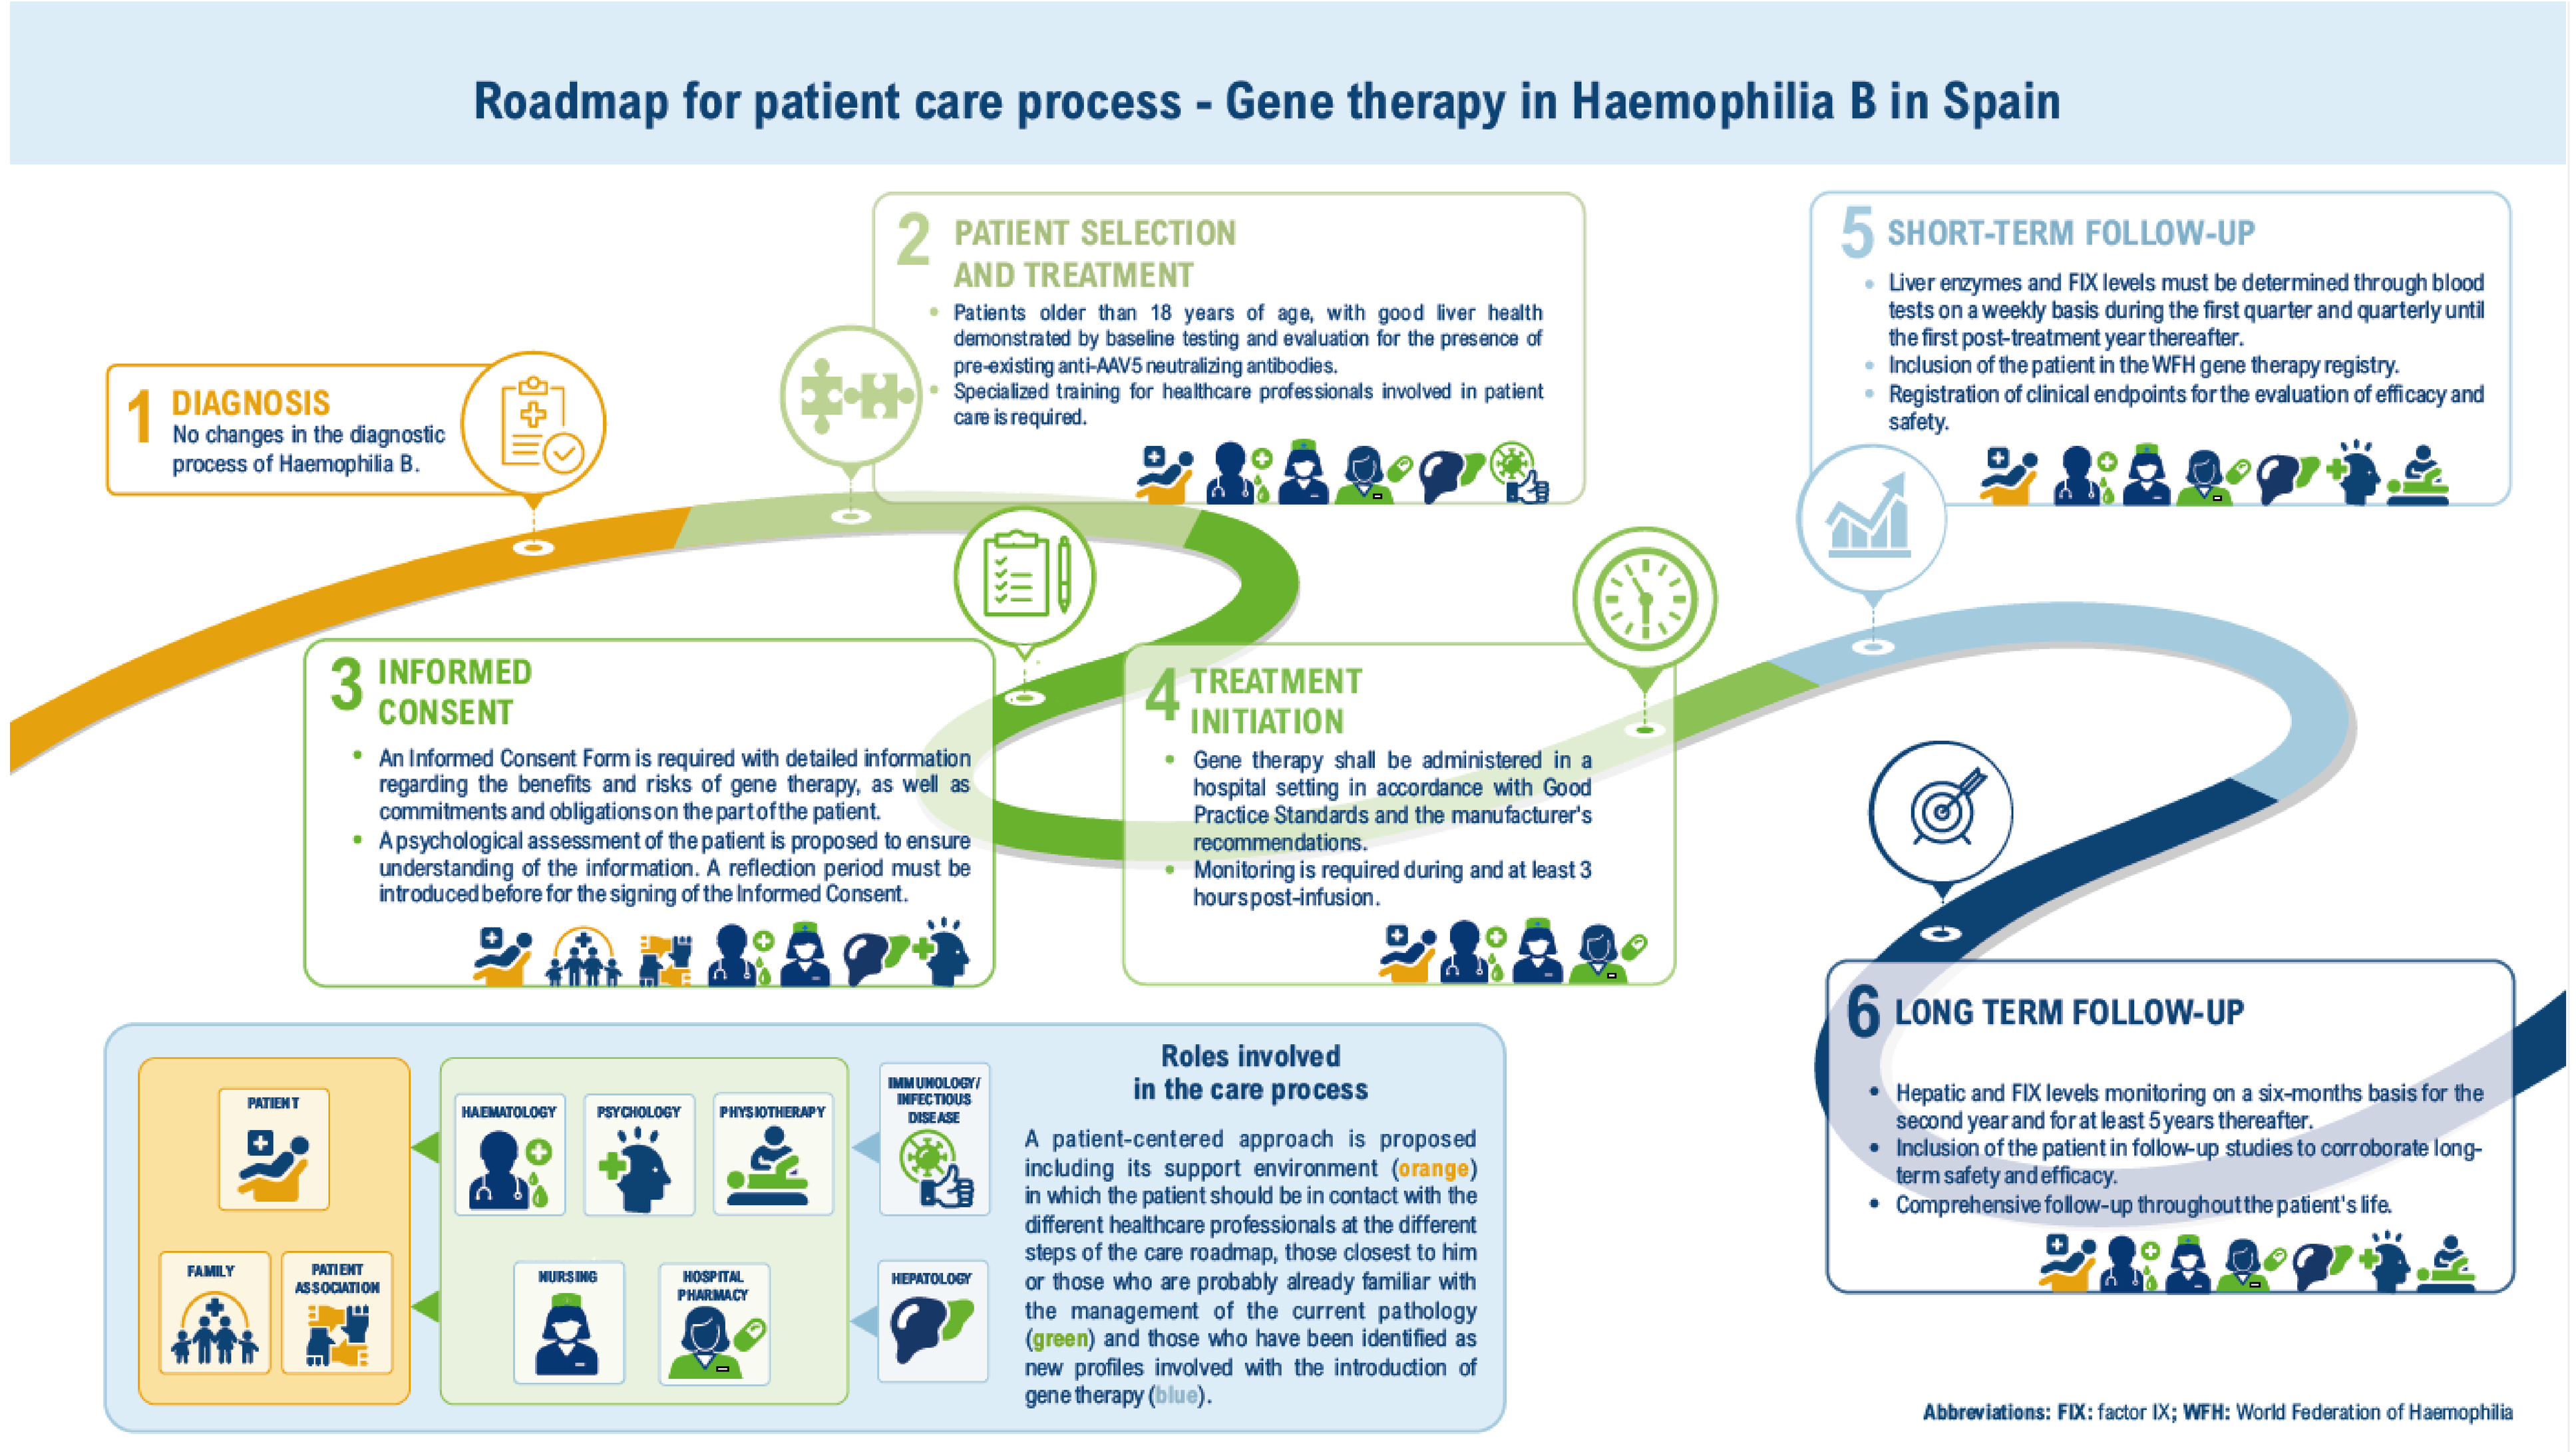

Roadmap for patient care process - Gene therapy in Haemophilia B in Spain
1 DIAGNOSIS
No changes in the diagnostic process of Haemophilia B.
2 PATIENT SELECTION AND TREATMENT
Patients older than 18 years of age, with good liver health demonstrated by baseline testing and evaluation for the presence of pre-existing anti-AAV5 neutralizing antibodies.
Specialized training for healthcare professionals involved in patient care is required.
3 INFORMED CONSENT
An Informed Consent Form is required with detailed information regarding the benefits and risks of gene therapy, as well as commitments and obligations on the part of the patient.
A psychological assessment of the patient is proposed to ensure understanding of the information. A reflection period must be introduced before for the signing of the Informed Consent.
4 TREATMENT INITIATION
Gene therapy shall be administered in a hospital setting in accordance with Good Practice Standards and the manufacturer's recommendations.
Monitoring is required during and at least 3 hours post-infusion.
5 SHORT-TERM FOLLOW-UP
Liver enzymes and FIX levels must be determined through blood tests on a weekly basis during the first quarter and quarterly until the first post-treatment year thereafter.
Inclusion of the patient in the WFH gene therapy registry.
Registration of clinical endpoints for the evaluation of efficacy and safety.
6 LONG TERM FOLLOW-UP
Hepatic and FIX levels monitoring on a six-months basis for the second year and for at least 5 years thereafter.
Inclusion of the patient in follow-up studies to corroborate long-term safety and efficacy.
Comprehensive follow-up throughout the patient's life.
Roles involved in the care process
PATIENT
FAMILY
PATIENT ASSOCIATION
HAEMATOLOGY
PSYCHOLOGY
PHYSIOTHERAPY
NURSING
HOSPITAL PHARMACY
IMMUNOLOGY/ INFECTIOUS DISEASE
HEPATOLOGY
A patient-centered approach is proposed including its support environment (orange) in which the patient should be in contact with the different healthcare professionals at the different steps of the care roadmap, those closest to him or those who are probably already familiar with the management of the current pathology (green) and those who have been identified as new profiles involved with the introduction of gene therapy (blue).
Abbreviations: FIX: factor IX; WFH: World Federation of Haemophilia

Figure 1: Proposed patient care roadmap for gene therapy in Haemophilia B in Spain.

1. **DIAGNOSIS**. Introduction of gene therapy does not impact this step since all patients are already diagnosed.
2. **PATIENT SELECTION AND TREATMENT**. **Hepatology and infectology/immunology profiles are added** as experts involved in this process. The **clinical eligibility criteria, evaluation and selection** of the patient candidate for gene therapy are determined by the Summary of Product Characteristics and will be individualized on a case-by-case basis.
3. **INFORMED CONSENT** (IC). The IC is crucial in this therapeutic modality, and differential to the currently used or the one that it is used in clinical trials with gene therapy. **Introduction of a specific IC generation process and documentation** for gene therapy including detailed information, as well as commitments and obligations for the patient is required. The **psychology and hepatology profiles are included** as advisory roles. The **psychological evaluation** of the patient prior to signing the IC document is critical.
4. **INITIATION OF TREATMENT**: It contemplates **adaptation of specific areas for the preparation and administration of gene therapy** and the **implementation of a detailed and specific post-infusion follow-up protocol** for this therapeutic modality. Gene therapy treatment is complex and requires **specialized and continuous training** for every healthcare professional involved.
4. **SHORT- AND LONG-TERM FOLLOW-UP**: The figure of **hepatology** is added given the need for continuous monitoring of the patient's liver health in the short and long term.

ECONOMIC ASPECTS RELATED TO CURRENT STRUCTURING AND FUNCTIONING OF THE SPANISH NATIONAL HEALTHCARE SYSTEM

Adapting Centres' structures and ensuring adequate qualification for gene therapy infusion

It will be important to find the right formula to balance key aspects such as the structural and organizational adequacy of hospital centres where patients already receive their usual therapy and are followed up by their Haematology Unit, and the training and qualification of healthcare professionals ensuring treatment availability and equity of access by patients who are candidates for such therapy in the different Spanish Autonomous Regions.

Cost compensation logistics for patients from other Autonomous Communities

Collaboration between hospitals that have already established administrative processes for the management of advanced therapies and cross-payments between Autonomous Regiones, with those centres that do not yet have this experience, in order to facilitate procedures and improve efficiency in the economic management of gene therapy will contribute to access and equity.

Operability of registry systems for the follow-up of clinical variables

Patient registries represent a basic tool both for clinical research and for short- and long-term evaluation of gene therapy effectiveness and safety. A strategy of continuous adaptation and improvement of existing patient registry systems and clinical variables is proposed, in order to effectively addressed the changing dynamics of medical care and the incorporation of disruptive therapeutic innovations such as gene therapy.

PENTALOGUE OF ACTIONS AND RECOMMENDATIONS FOR THE PRACTICAL IMPLEMENTATION OF THE PROPOSED PATIENT CARE ROADMAP

- **Review of the organisational structure of reference centres treating Haemophilia B patients, guaranteeing availability and equity in patient access across the different Spanish regions.**
- **Review and evaluation of existing multidisciplinary committees to ensure a coordinated and multidisciplinary approach.**
- **Development of a comprehensive and detailed patient informed consent process.**
- **Inclusion of patients in international registry systems for patient follow-up and obtaining robust data on clinical outcome variables.**
- **Comprehensive and specific training for healthcare professionals, tailored to their participation in each stage of the patient journey.**

CONCLUSIONS

This project represents the **first patient care roadmap for gene therapy for Haemophilia B in Spain**. Its implementation in Spanish hospitals could contribute to **guide effective incorporation of this disruptive therapy within the healthcare system and delivery the expected health outcomes**. It does also represent the first pathway for any gene therapy in Spain. The identified challenges and the proposals to address them set a precedent for current and future gene therapies. Incorporation of the proposed steps in Haemophilia B-treating centres must be accompanied by a process flow that includes a guarantee of timelines and governance to achieve the necessary quality of care and the desired clinical objectives.

With the scientific endorsement of the following Scientific Societies and Patient Associations:

Developed by

Sponsored by

ISPOR EUROPE 2024