Quantifying health related quality of life benefits from patients treated with mavacamten for obstructive hypertrophic cardiomyopathy (HCM) in China based on the EXPLORER-CN trial

MSR120

Zoe Cheah,[1] Claire Simons,[1] Yue Zhong,[2] Yu Jia,[3] Michael Hurst[4]

[1]OPEN Health HEOR and Market Access UK; [2]Bristol Myers Squibb, Princeton, NJ, USA; [3]Bristol Myers Squibb (China) Investment Co. Ltd, Shanghai, China; [4]Bristol Myers Squibb, Uxbridge, UK

Background

- Hypertrophic cardiomyopathy (HCM) is characterised by primary left ventricular (LV) hypertrophy, with a proportion of patients having obstruction in the outflow tract of the left ventricle (categorised as obstructive HCM).[1-4]
- For patients with obstructive HCM, symptomatic burden can vary significantly (from asymptomatic, defined as a New York Heart Association [NYHA] class I, to breathlessness at rest, defined as NYHA class IV), and the disease is associated with an increased risk of atrial fibrillation, heart failure, and malignant ventricular arrythmias.
- As of 2024, there are no therapies that target the underlying pathophysiology of the disease that are reimbursed in China for symptomatic (NYHA class II/III) obstructive HCM.
- EXPLORER-CN (NCT05174416) is a phase III, multicentre, randomised, double-blind, placebo-controlled registration trial that evaluated the efficacy and safety of mavacamten (a first-in-class, small-molecule, selective allosteric inhibitor of cardiac myosin ATPase) in Chinese adults with symptomatic obstructive HCM.[5-6]
- Results from EXPLORER-CN showed that treatment with mavacamten + standard of care (SoC; including beta blockers or calcium channel blockers) was associated with a statistically significant and clinically meaningful improvement in Valsalva left ventricular outflow tract (LVOT) gradient from baseline to week 30 compared to placebo + SoC.
- Treatment with mavacamten was also associated with an improvement for all secondary endpoints, including change in NYHA class from baseline at week 30 and change from baseline to week 30 in Kansas City Cardiomyopathy Questionnaire (KCCQ) Clinical Summary Score (CSS).
- Generally, little has been studied about the quality of life (QoL) of patients with obstructive HCM in China.
- Thus, this analysis aims to explore the modelled QoL as assessed by EQ-5D of Chinese patients with symptomatic obstructive HCM by utilising data from EXPLORER-CN.

Objectives

- The aim of this work was to explore the modelled QoL of Chinese patients with symptomatic obstructive HCM through the utilisation of EXPLORER-CN trial data.

Methods

- Utility analyses were conducted on the ITT population of EXPLORER-CN, which comprised of 81 patients, of which 54 were randomised to mavacamten + SoC and 27 were randomised to placebo + SoC in a 2:1 ratio.
- The only QoL data collected in EXPLORER-CN was the KCCQ-CSS and KCCQ Overall Summary Score (OSS), which is derived from a validated patient reported outcome measure, KCCQ-23, for obstructive HCM patients.[8]
- Other trials studying mavacamten as an intervention namely EXPLORER-HCM (NCT03470545)[9], a trial conducted in non-Asian countries.
- EXPLORER-HCM additionally utilised EuroQol-five dimension five-level (EQ-5D-5L) scores which can be used to inform utilities for obstructive HCM patients.
- To enable an appropriate comparison between EXPLORER-HCM and EXPLORER-CN utility data, KCCQ-23 scores collected from EXPLORER-CN were converted to EQ-5D index scores using a mapping algorithm outlined in Thomas et al., (2021).[10]
- Linear mixed models (LMMs) were used as they account for the correlation between repeated measurements and provide the option to include fixed and random effect terms for time and interactions with baseline covariates, which were then fit to utility data to determine the impact of baseline covariates on utility.
- Relevant covariates were selected using a stepwise (forwards and backwards) regression model as shown in **Figure 1**.
- For each model, fixed effects, as well as random intercepts, random slopes, and both random intercepts and slopes were explored.
- The optimal model was defined as the model which minimised the AIC and BIC, while reflecting reality and generating plausible results.
- Once the optimal model was identified, least square mean estimates of the EQ-5D values along with the corresponding standard errors were calculated for each health state which were then compared to observed values to assess predictive accuracy (**Figure 1**).
- As EXPLORER-HCM collected EQ-5D in their trial, the results produced from mapping the KCCQ-23 to EQ-5D in EXPLORER-CN were compared against EXPLORER-HCM EQ-5D data that was mapped to the China Value Set11 to validate the results produced (**Figure 1**).

Figure 1. Overview of methods used

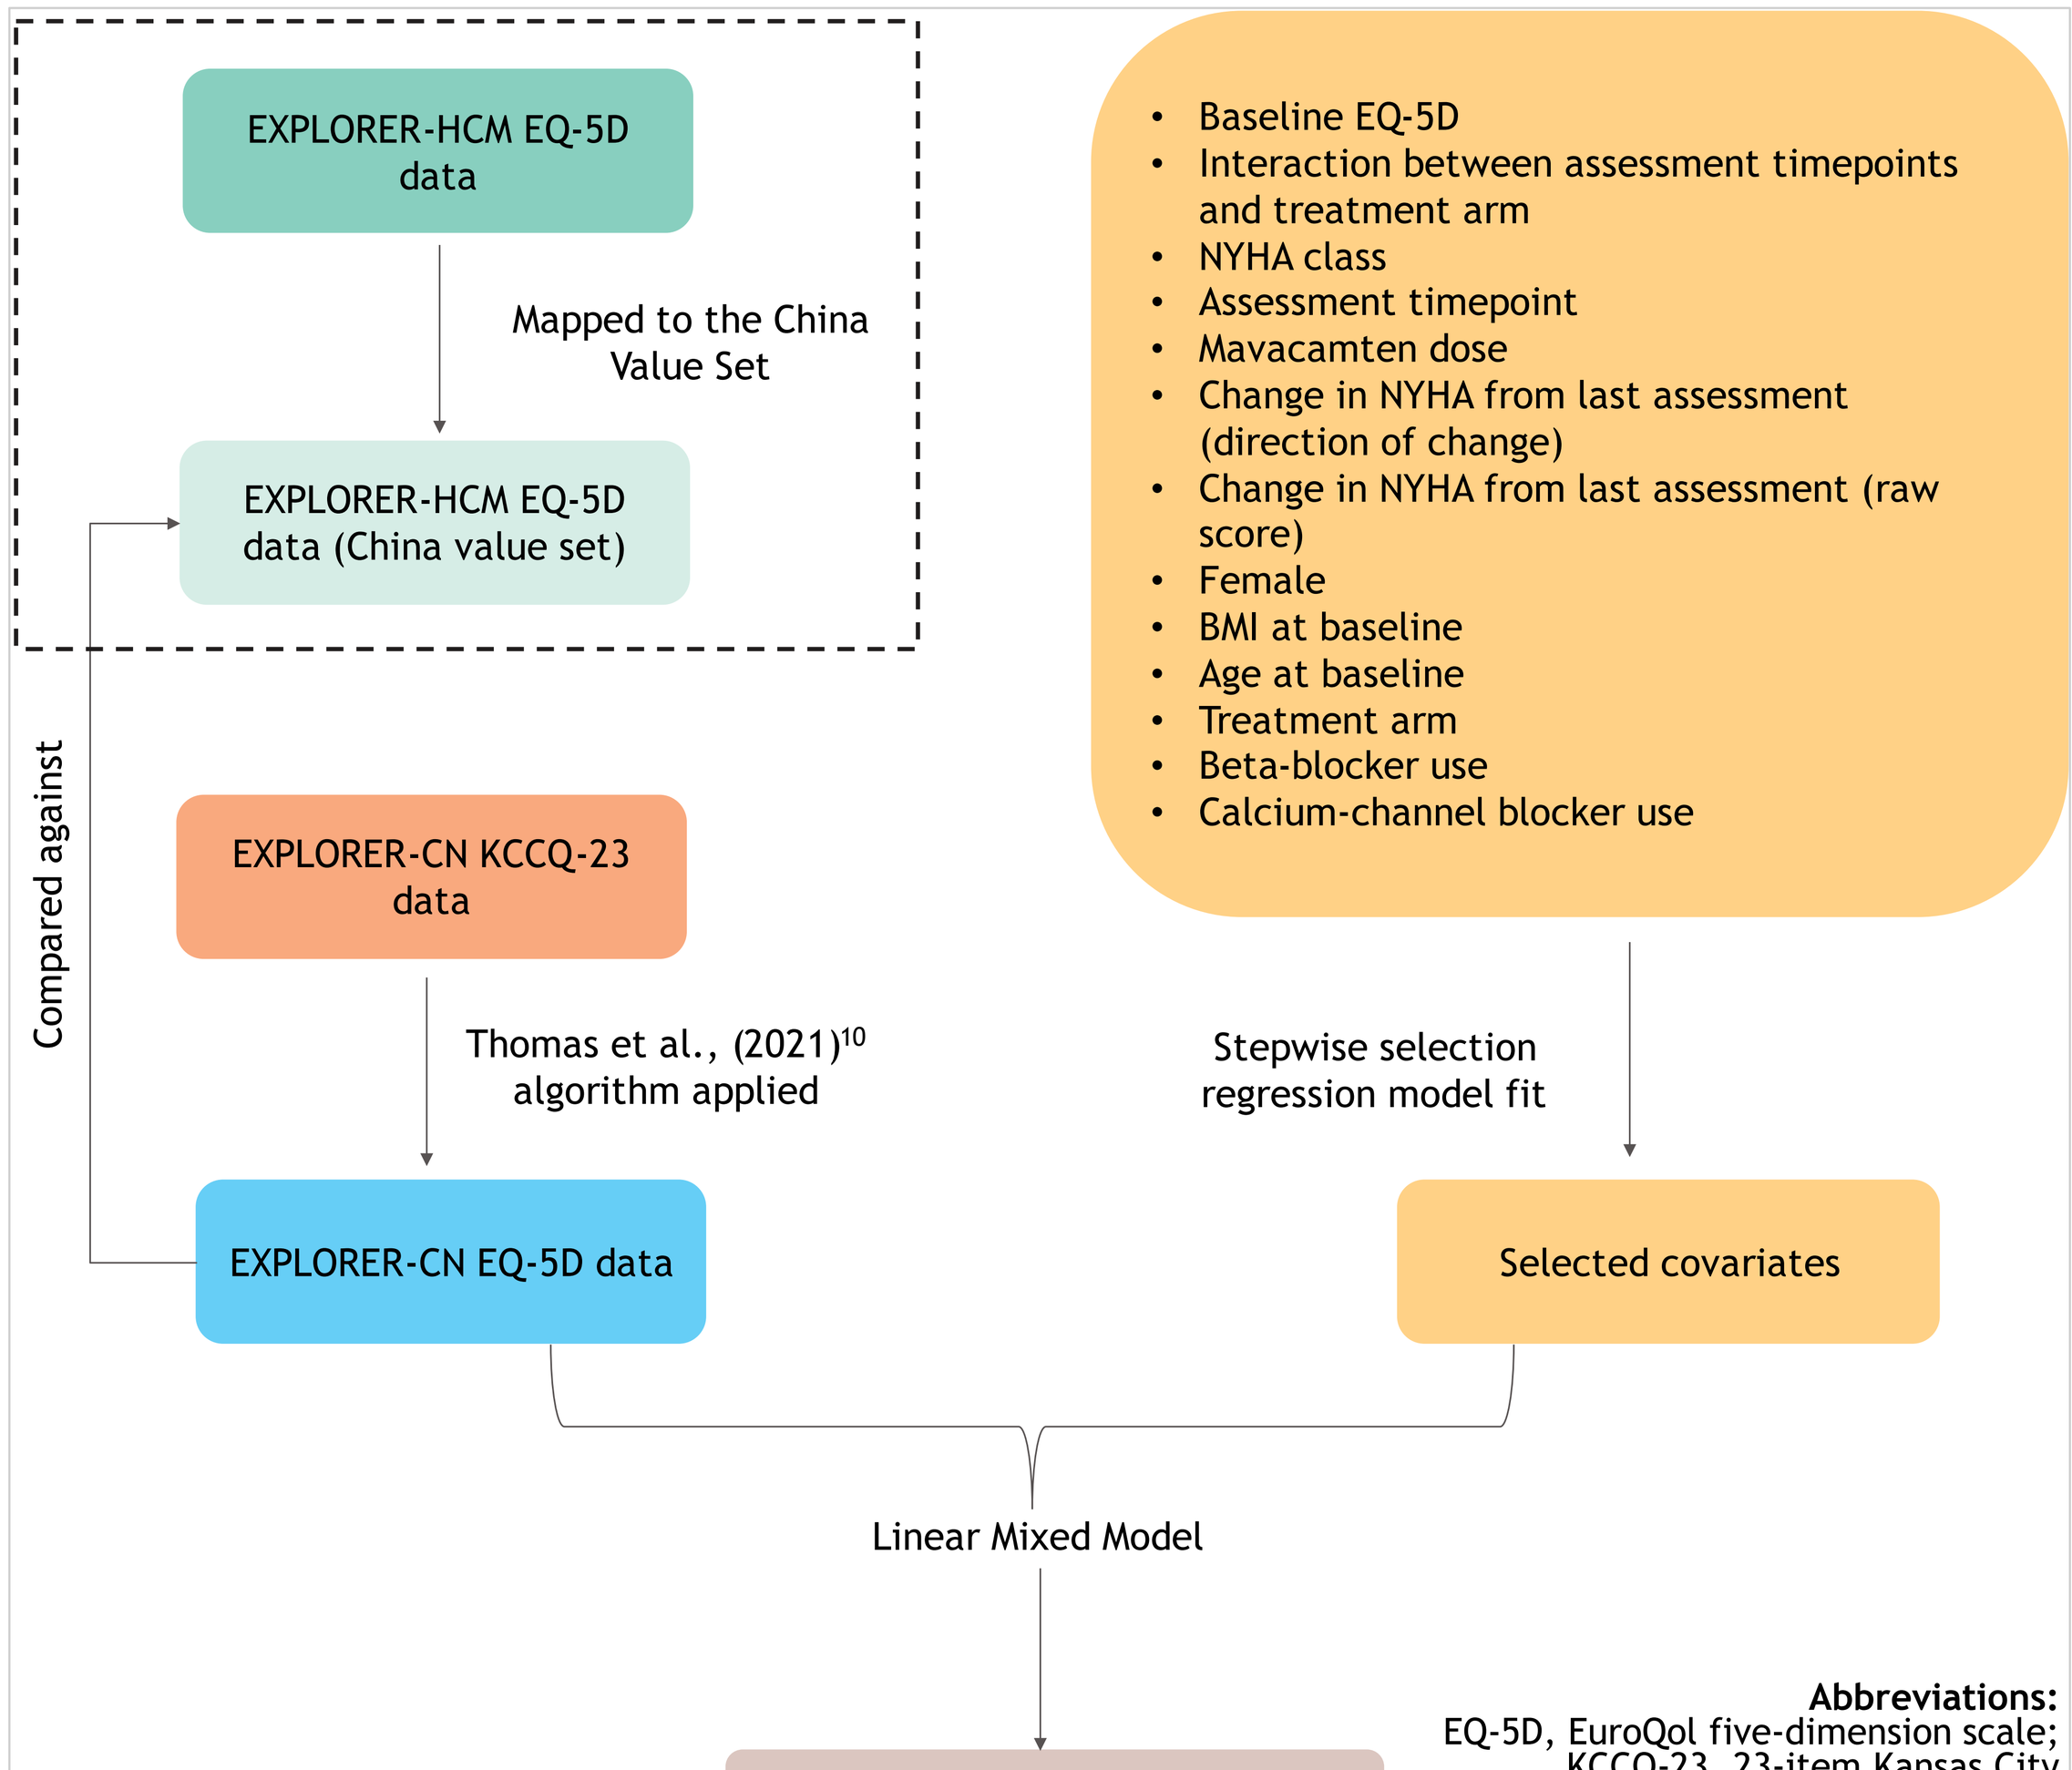

EXPLORER-HCM EQ-5D data
Mapped to the China Value Set
EXPLORER-HCM EQ-5D data (China value set)
Compared against
EXPLORER-CN KCCQ-23 data
Thomas et al., (2021)10 algorithm applied
EXPLORER-CN EQ-5D data
Baseline EQ-5D
Interaction between assessment timepoints and treatment arm
NYHA class
Assessment timepoint
Mavacamten dose
Change in NYHA from last assessment (direction of change)
Change in NYHA from last assessment (raw score)
Female
BMI at baseline
Age at baseline
Treatment arm
Beta-blocker use
Calcium-channel blocker use
Stepwise selection regression model fit
Selected covariates
Linear Mixed Model
EQ-5D least square mean estimates

Abbreviations: EQ-5D, EuroQol five-dimension scale; KCCQ-23, 23-item Kansas City cardiomyopathy questionnaire; NYHA, New York Heart Association

Results

Descriptive analysis of results

- **Figure 2** displays the observed, raw means and 95% confidence intervals (CIs) of the KCCQ-23 OSS (L), as well as the KCCQ-23 mapped to EQ-5D using the Thomas et al., (2021)[10] algorithm (R) respectively at each EXPLORER-CN assessment visit, by treatment arm.
- Trends across both measures of utility were similar, suggesting that the mapping process did not result in large deviations of utility between the KCCQ-23 OSS and the mapped EQ-5D values.
- Across both patient reported outcomes (PROs), there is no evidence of statistically significant differences in health-related QoL at baseline between treatment arms.
- Patients on mavacamten experience greater improvements across all PRO measures compared to placebo, with the largest improvement observed at week 30.

Differences in utility observed by treatment arm

- For the EXPLORER-CN EQ-5D estimates, the estimated mean change in utility from baseline to week 30 was 0.04 (95% confidence interval [CI] 0.005, 0.07) and -0.04 (95% CI -0.07, -0.01) for mavacamten and placebo patients respectively as shown in **Figure 3**.

Figure 2. Mean KCCQ-23 OSS (left) and EQ-5D (right) by treatment arm and assessment timepoint in EXPLORER-CN

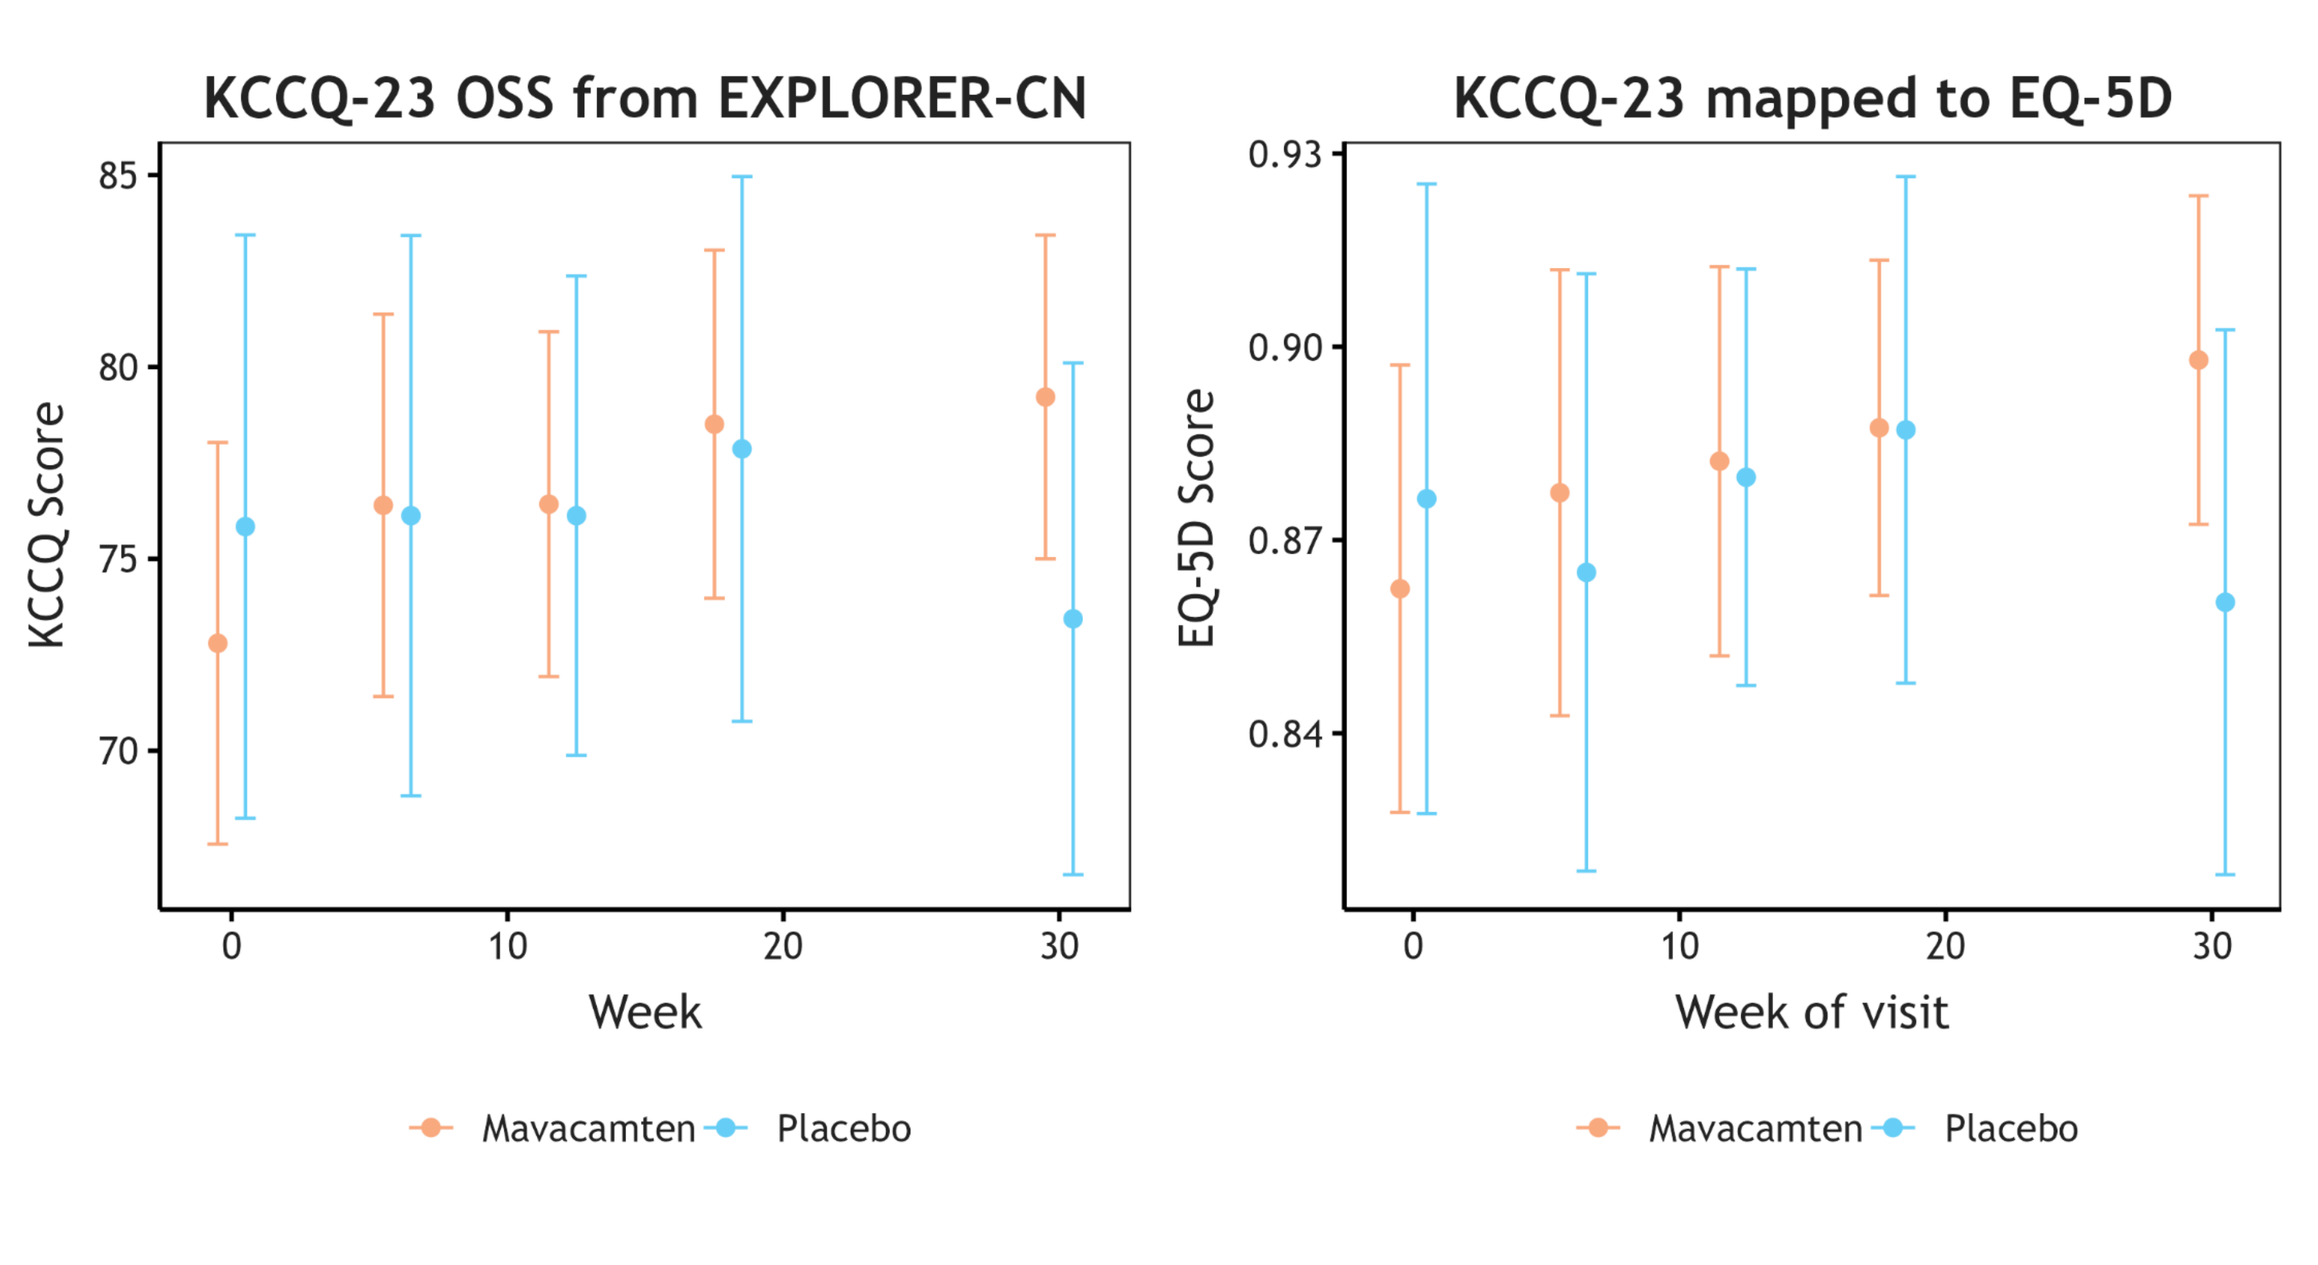

KCCQ-23 OSS from EXPLORER-CN
KCCQ Score
85
80
75
70
0
10
20
30
Week
KCCQ-23 mapped to EQ-5D
EQ-5D Score
0.93
0.90
0.87
0.84
Week of visit
Mavacamten
Placebo

The mean KCCQ-23 OSS (L) and mean KCCQ-23 mapped EQ-5D (R) by treatment arm are displayed above, along with their 95% confidence intervals. The "jitter" of markers for the treatment arms was done for visualization purposes only and the data was collected at the same point in time for both treatment arms.
Source: own calculations using data from the EXPLORER-CN trial.
Abbreviations: EQ-5D, EuroQol five-dimension scale; KCCQ-23, 23-item Kansas City cardiomyopathy questionnaire.

Figure 3. Mean EQ-5D difference at week 30 (vs baseline) by treatment arm

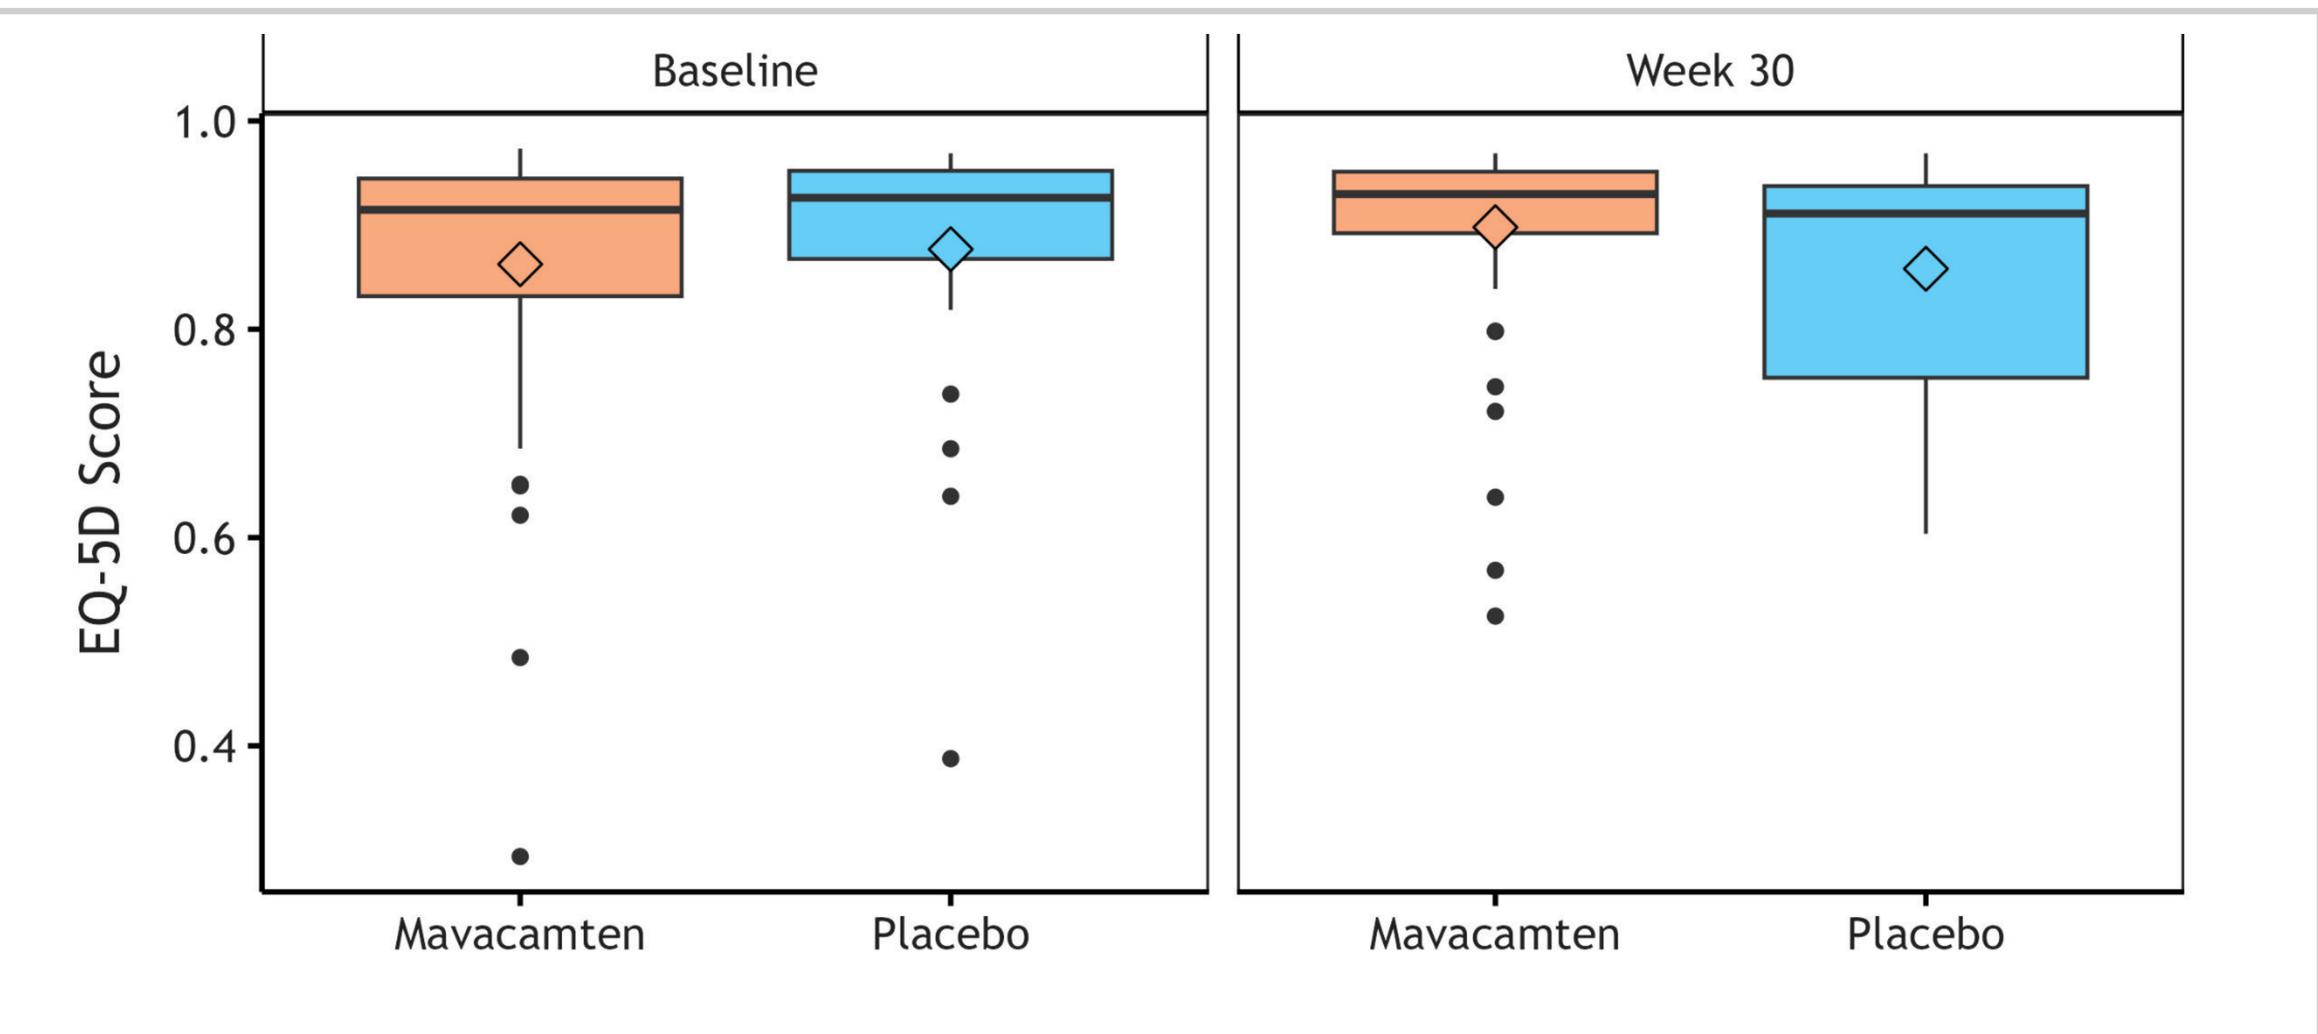

Baseline
Week 30
EQ-5D Score
1.0
0.8
0.6
0.4
Mavacamten
Placebo

Abbreviations: EQ-5D, EuroQol five-dimension scale; NYHA, New York Heart Association.

Linear Mixed Model results

- The preferred model specification featured a random intercept at the patient level as opposed to one at the treatment arm level including baseline EQ-5D value as its preferred covariate.
- Despite not being selected as a variable of interest through stepwise regression, NYHA class as a categorical variable was also included in the preferred random intercept model, as there is a correlation between higher EQ-5D scores and lower NYHA class (and vice versa).
- When incremental change in EQ-5D was included as a dependent variable, baseline EQ-5D was not included as a variable of interest, suggesting that it added little explanatory power to the model, and as such baseline EQ-5D was removed as a variable retaining NYHA class only.
- All other potential covariates added were statistically insignificant and did not improve the explanatory power of the model.

Derivation of an inflation factor

- To compare EQ-5D data from EXPLORER-CN with EQ-5D data collected in EXPLORER-HCM, the same LMM was fit to EXPLORER-HCM data mapped to the China value set.[11]
- The estimated decrement between NYHA II and NYHA III was smaller for the mapped EQ-5D values generated from EXPLORER-CN data when compared to the EXPLORER-HCM data mapped to the China value set.[11]
- When KCCQ-23 data from EXPLORER-HCM was mapped to EQ-5D[10] and compared against original EQ-5D data collected in the trial, the same reduction in decrement that occurred **(Figure 4)** was observed in EXPLORER-CN data (**Table 1**).
- This reduction in estimated decrements is a known issue with mapping algorithms, with an inflation factor calculated using the EXPLORER-HCM data mapped to the Chinese value set to combat this loss of granularity.[11]
- The relative increase in decrement was calculated for each estimated health state by dividing the health-state estimates from the original EXPLORER-HCM EQ-5D data by the estimates derived from the KCCQ-mapped EQ-5D data as outlined in the equation below and in **Figure 4.**
- As a result, EQ-5D decrements corresponding to NYHA II and III estimated from EXPLORER-CN were inflated by 2.32 and 3.58 respectively as shown in **Figure 4.**
- Both trials were assumed to be comparable for this inflation factor to be applied.

$$Inflation\ factor = \frac{Observed\ EQ-5D\ estimate}{KCCQ-mapped\ EQ-5D\ estimate}$$

- Additionally, standard errors for the inflated EXPLORER-CN estimates were inflated by 1.30 as recommended by Thomas et al., (2021).[10] due to observations that the variance decreases by a magnitude of 1.7 when KCCQ is mapped to EQ-5D.
- The original EXPLORER-CN decrement estimates obtained from the preferred LMM previously outlined as well as the inflated estimates can be found in **Table 1**.
- Notably, the main limitation of this method is that the inflation factor is estimated from EXPLORER-HCM data.
- Thus, applying this to the EXPLORER-CN KCCQ-23 mapped EQ-5D may not accurately reflect the EQ-5D that would have been collected in the trial in reality.

Figure 4. Derivation of the inflation factors using EXPLORER-HCM data

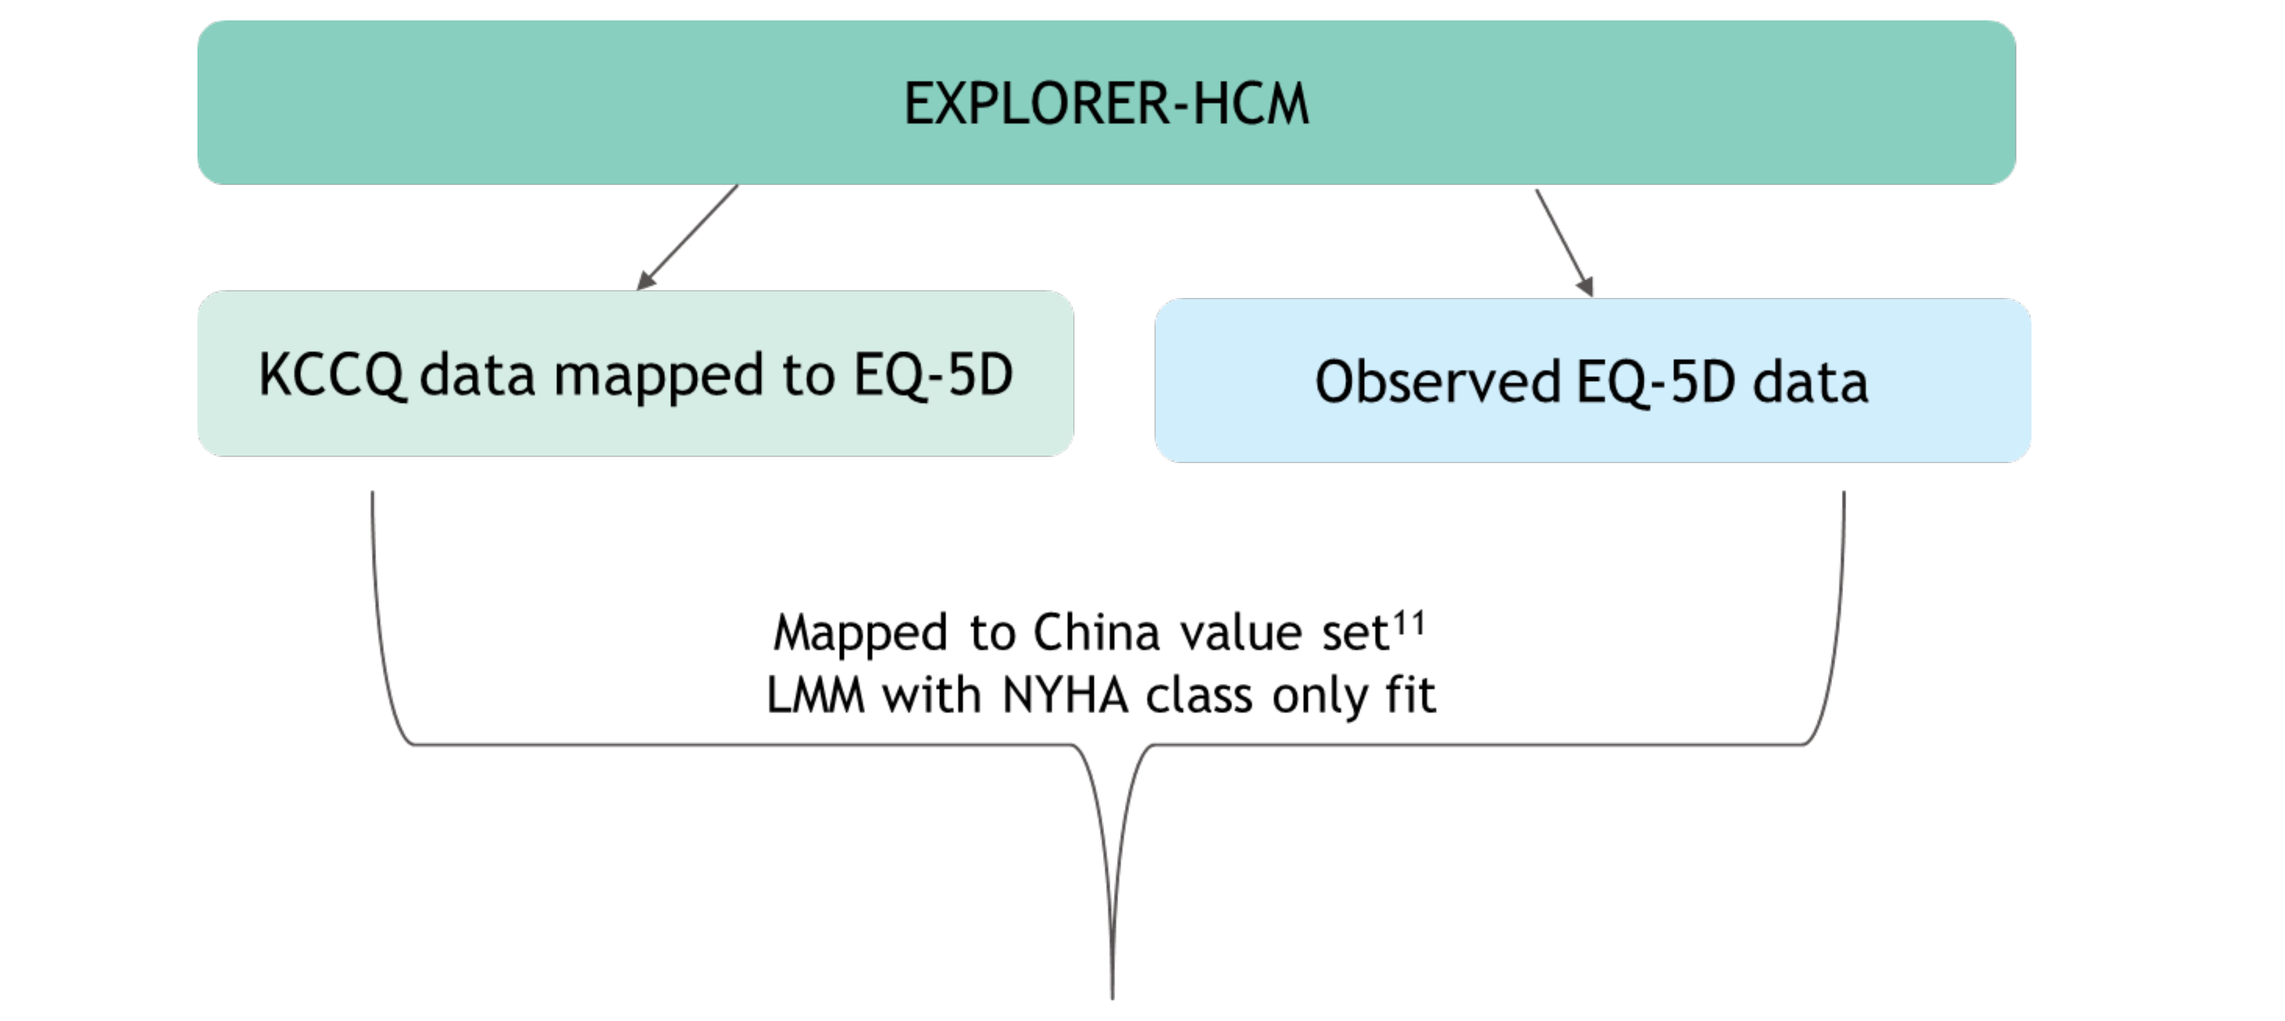

EXPLORER-HCM
KCCQ data mapped to EQ-5D
Observed EQ-5D data
Mapped to China value set11
LMM with NYHA class only fit

Dependent variable	Parameter	Estimate
Observed EQ-5D data (EXPLORER-HCM)	Intercept	0.933
	NYHA II	-0.051
	NYHA III	-0.179
KCCQ-23 data mapped to EQ-5D (EXPLORER-HCM)	Intercept	0.896
	NYHA II	-0.022
	NYHA III	-0.050
Calculated inflation factor		
NYHA II versus I	2.32	
NYHA III versus II	3.58	

Source: own calculations using data from the EXPLORER-HCM trial.
Abbreviations: LMM, linear mixed model; NYHA, New York Heart Association Functional Classification; EQ-5D, EuroQol five dimension scale; KCCQ-23, 23-item Kansas City cardiomyopathy questionnaire

Table 1. Observed and inflated EXPLORER-CN EQ-5D decrements

Dependent variable	Parameter	Estimate	Standard error
KCCQ data mapped to EQ-5D (EXPLORER-CN)	Intercept	0.905	0.015
	NYHA II	-0.026	0.012
	NYHA III	-0.061	0.018
• Inflated KCCQ data mapped to EQ-5D (EXPLORER-CN)	Intercept	0.905	0.020
	NYHA II	-0.060	0.016
	NYHA III	-0.218	0.023

Source: own calculations using data from the EXPLORER-HCM trial.
Abbreviations: NYHA, New York Heart Association Functional Classification; EQ-5D, EuroQol five dimension scale; KCCQ, 23-item Kansas City cardiomyopathy questionnaire

Validation of results

- The predicted EQ-5D by NYHA class estimated for both EXPLORER-CN and EXPLORER-HCM from the preferred LMM closely aligns to the observed values.
- **Figure 5** also shows that prediction accuracy across all NYHA classes is acceptable, however, the model underpredicts at higher values for patients in NYHA I, whereas the converse is observed regarding NYHA II and III.
- For NYHA III, the model predicts a bimodal distribution towards higher EQ-5D scores where none is observed. This could be due to the small sample size of patients in NYHA III in EXPLORER-CN, which increases the uncertainty.
- When EXPLORER-CN values were inflated using the estimated inflation factors, the decrement estimates produced aligned with those produced from the EXPLORER-HCM EQ-5D data mapped to the China Value Set.[11]
- Thus, the observations above validate the approach undertaken to estimate EQ-5D data for EXPLORER-CN.

Figure 5. Observed and predicted distributions of the EXPLORER-CN (top) and the EXPLORER-HCM (bottom) EQ-5D score by NYHA class

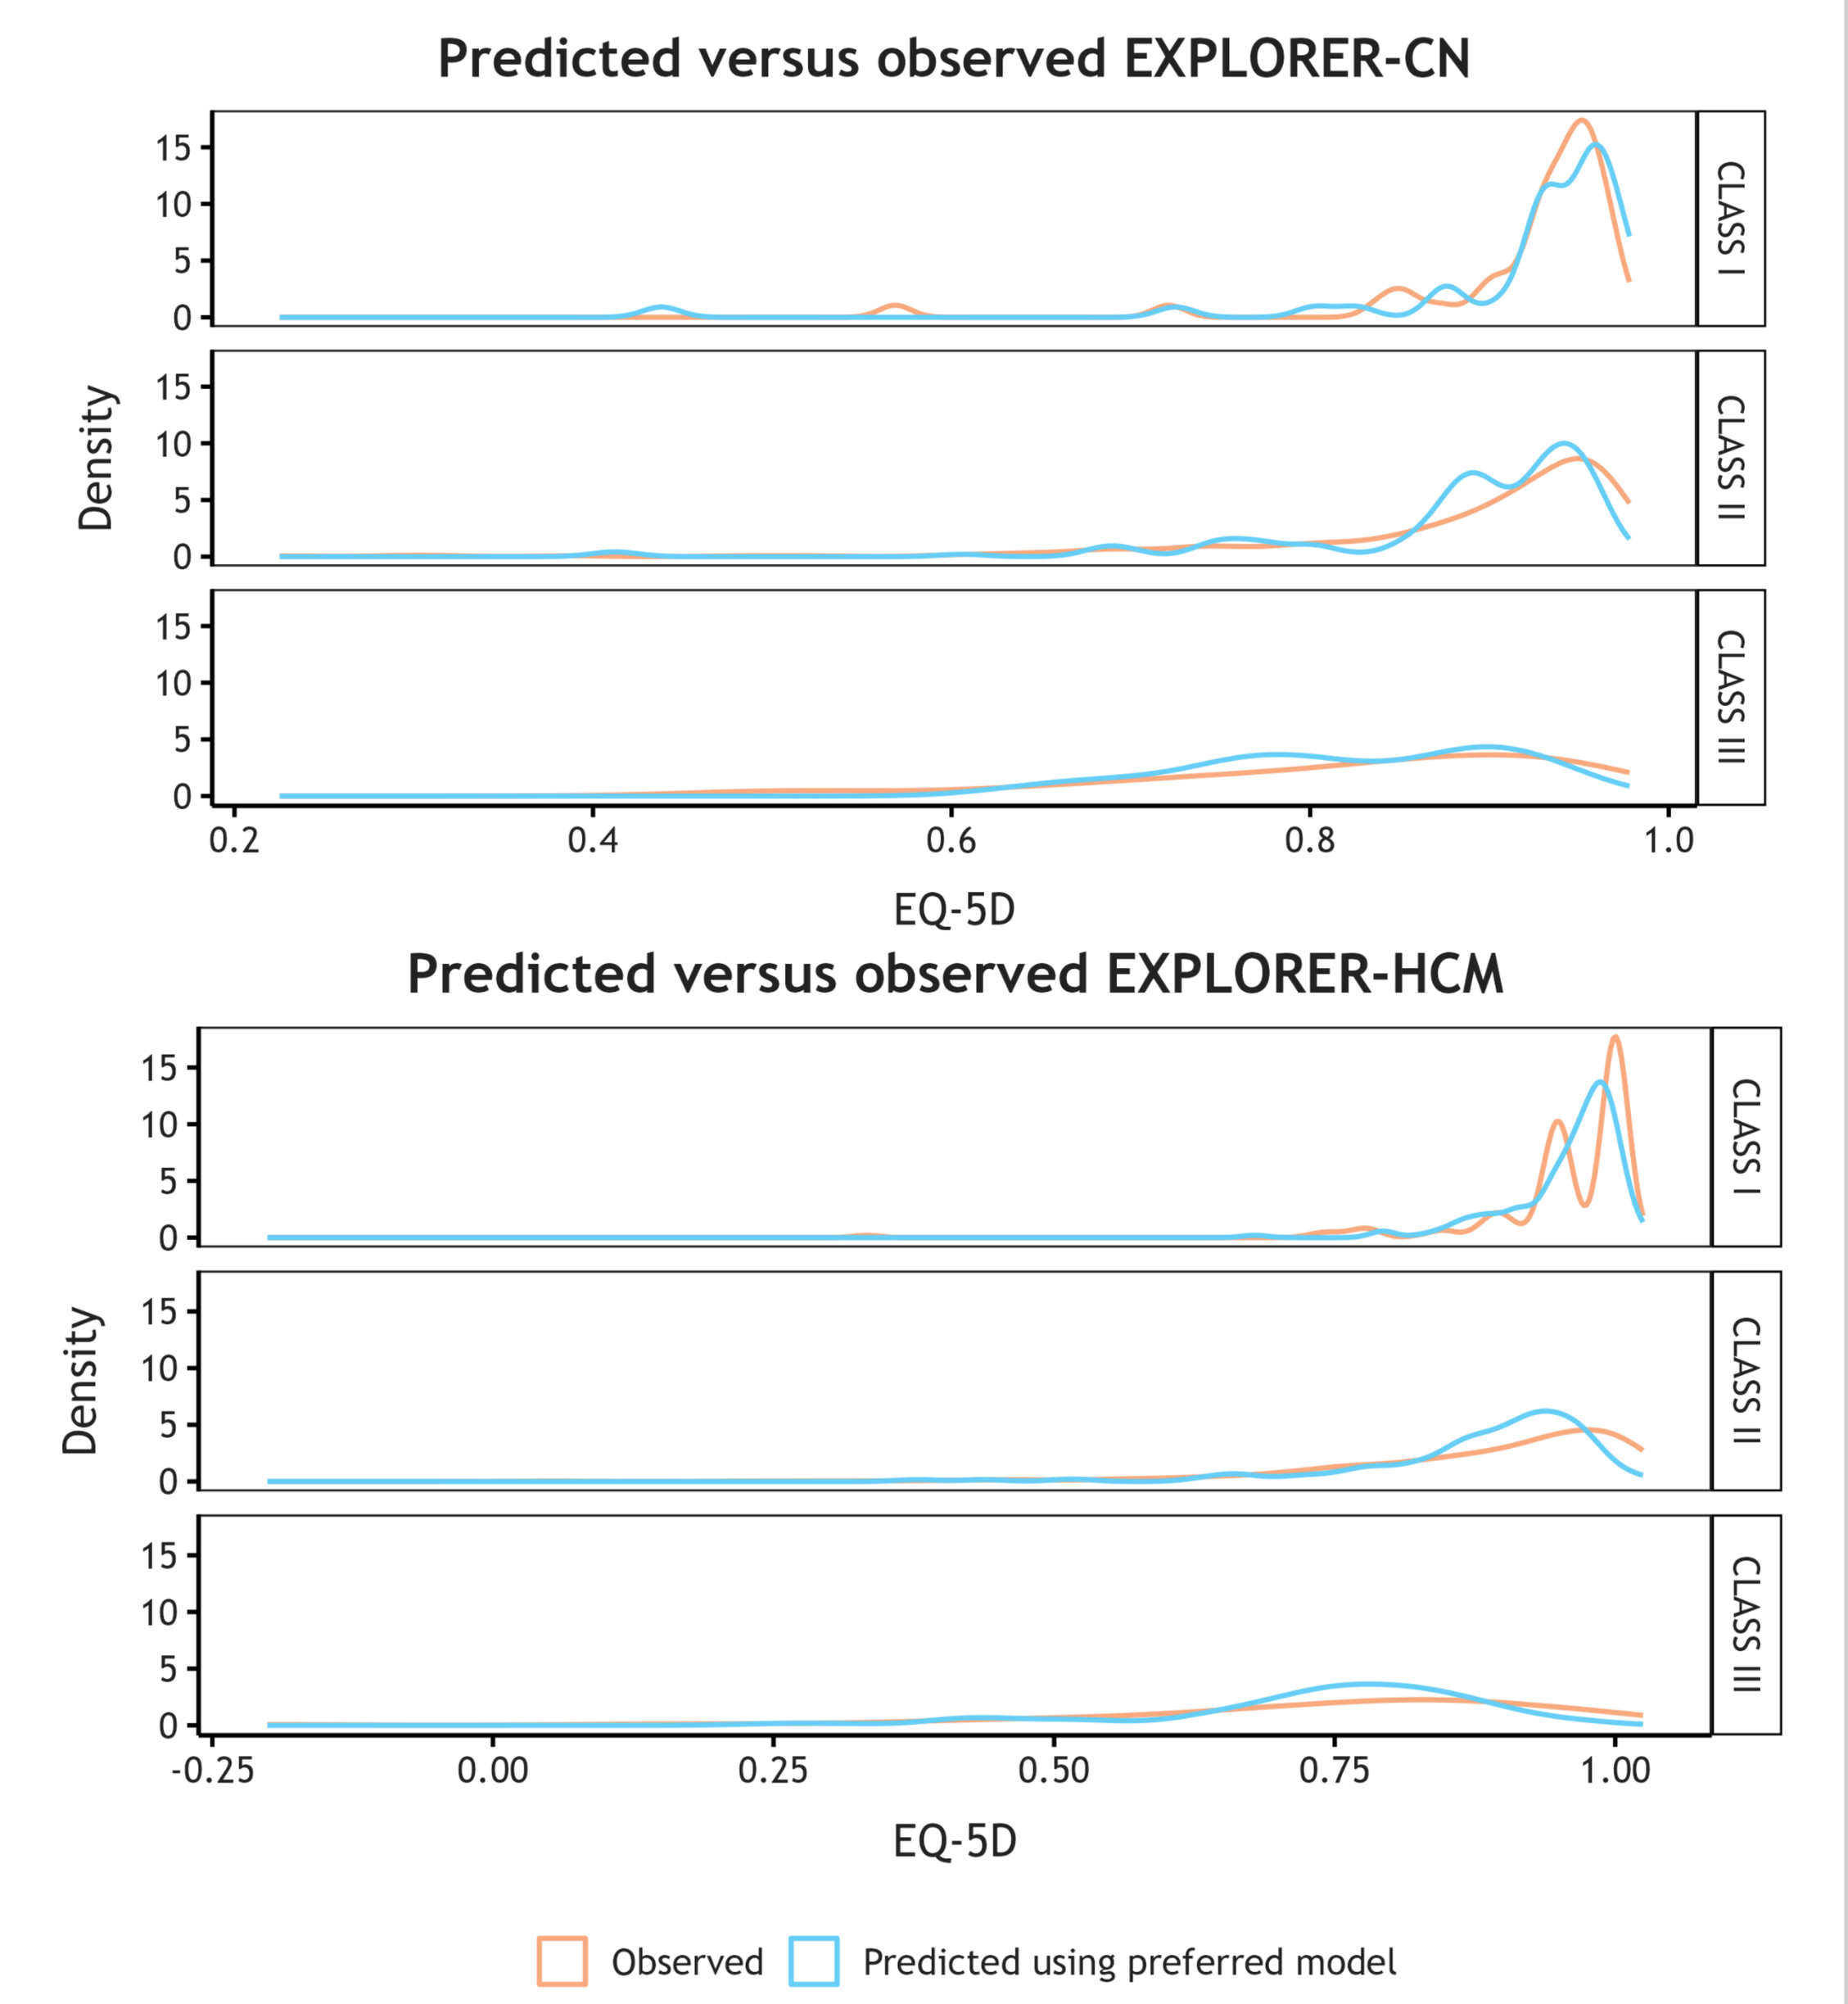

Predicted versus observed EXPLORER-CN
Density
CLASS I
CLASS II
CLASS III
EQ-5D
Predicted versus observed EXPLORER-HCM
Density
CLASS I
CLASS II
CLASS III
EQ-5D
Observed
Predicted using preferred model

Source: own calculations using data from the EXPLORER-CN (top) and EXPLORER-HCM (bottom) trial.
Abbreviations: EQ-5D, EuroQol five-dimension scale

Conclusions

- In the absence of EQ-5D data in the EXPLOER-CN trial, KCCQ data can be mapped to EQ-5D via the Thomas et al., (2021)[10] algorithm to highlight the QoL burden of patients with symptomatic obstructive HCM.
- Smaller decrements are observed between the NYHA II and III classes when LMMs were fit to KCCQ-mapped EQ-5D data in EXPLORER-CN compared to when the same LMM was fit to observed EQ-5D data collected in the EXPLORER-HCM trial.
- Deriving an inflation factor from EXPLORER-HCM data and applying it to the KCCQ-mapped EQ-5D data from EXPLORER-CN was able to account for the reduction in decrement observed.
- This study has several limitations, primarily that the inflated EXPLORER-CN EQ-5D may not be an accurate reflection of what the actual EQ-5D values would have been if they were collected in the trial.
- Moreover, since the inflation factor was calculated with EXPLORER-HCM data, the difference in decrements between NYHA classes may not be equal to what is observed.
- With regards to the KCCQ-mapped EQ-5D data from EXPLORER-CN, patients in NYHA I have a higher mean utility compared to patients in NYHA II and III.
- This implies that mavacamten has the potential to improve patients' QoL through improvements in symptomatic status, as 44.4% the mavacamten arm are in NYHA I at week 30 of EXPLORER-CN versus 4.0% in the placebo arm.
- Therefore, the joint analysis of EXPLORER-CN and EXPLORER-HCM utility data provides useful insights into the QoL burden of patients with symptomatic obstructive HCM in China.
- Future work could investigate whether these trends hold in the long-term.

References

1. Maron BJ, et al. J Am Coll Cardiol 2014;64:83-99
2. Priori SG, et al. Eur Heart J 2015;36:2793-2867
3. Semsarian C, et al. J Am Coll Cardiol 2015;65:1249-1254.
4. RB&HH Specialist Care. https://www.rbhhspecialistcare.co.uk/news/our-new hypertrophiccardiomyopathy-service. Accessed September 9, 2024
5. Lim GB. Nat Rev Cardiol 2020;17:677.
6. Tian, Z., et al., BMJ open. 2023; 13:6
7. LianBio. https://investors.lianbio.com/news-releases/news-release-details/lianbio-announces-positive-topline-results-phase-3-explorer-cn. Accessed September 9, 2024
8. Green C.P., et al. Journal of the American College of Cardiology 2000; 35(5): 1245-55
9. Olivotto I. et al. Lancet Lond Engl 2020;396: 59-769
10. Thomas, M et al. European Heart Journal 2021; 7(4):388-396
11. Luo, N. Value in Health 2017; 20(4): 662-669

Acknowledgments

- This study was supported by Bristol Myers Squibb. All authors contributed to and approved the poster.
- ZC, CS, and MH contributed to the development of the poster by providing medical writing and editorial assistance.

Disclosures

- YZ, YJ, and MH are employees of Bristol Myers Squibb and may hold Bristol Myers Squibb stock options.
- ZC and CS are employees of OPEN Health, which has received consultancy fees from BMS.
- The authors declare having no competing interests beyond those reported.

Presented at ISPOR Europe 2024; November 17-20, 2024; Barcelona, Spain, and virtual

Email: <michael.hurst@bms.com>

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.