Avalere Health

Multi-disciplinary approach to develop an ultra-rare disease decision aid

Wahid N[1], Corcoran H[1], Zhang S[1], Harrington S[1], Housman L[1]
1Avalere, Washington, DC, USA

PCR42

Introduction

Paroxysmal nocturnal hemoglobinuria (PNH) is an ultra-rare, acquired, and potentially life-threatening hematologic disease characterized by complement-mediated hemolysis, thrombosis, and bone marrow dysfunction.[1, 2] PNH has been estimated to affect 10 to 15 individuals per million globally,[3] with a median onset age of 35.5 years.[4] Four United States Food and Drug Administration (US FDA)-approved pharmacological treatments exist for managing PNH: two complement C5 inhibitors, eculizumab[8] and ravulizumab[9], one C3 inhibitor, pegcetacoplan[10], and one targeted B factor inhibitor of the alternative complement pathway, Iptacopan.[11] Individuals newly diagnosed with an ultra-rare disease such as PNH can feel overwhelmed as limited information may be available, and they may be unable to work through the risks and benefits of their treatment options. Patient decision aids offer a valuable solution to support patients in understanding their treatment options that help facilitate patient engagement in a process called shared decision-making (SDM).

Objectives

- Despite multiple treatment options for PNH, an ultra-rare and potentially life-threatening hemolytic disease, no comprehensive patient decision aid existed to support treatment selection.
- This study aimed to design, test, validate, and finalize a comprehensive SDM tool ("decision aid" or "DA") for PNH. The decision aid was designed to help patients understand the causes of PNH, PNH symptoms, and the benefits and risks of different treatments, in addition to gathering their preferences and values, to facilitate a productive discussion with their physician.

Methods

A multi-disciplinary team designed, tested, and validated a web-based DA using primary and secondary research (targeted literature reviews, cognitive interviews (n=4 physicians; n=10 patients), and a decisional needs analysis (n=12 physicians; n=60 patients)). The study comprised of four phases: development, alpha testing, beta testing, and finalization (**Figure 1**).

A Steering Committee was formed to provide guidance to the PNH decision aid team on the development and use of the decision aid as well as provide oversight of the decision aid development process. Key considerations for an effective decision aid include accessibility, content-quality, information delivery, user experience, evaluation, implementation, and dissemination.[9]

Figure 1: SDM development flowchart

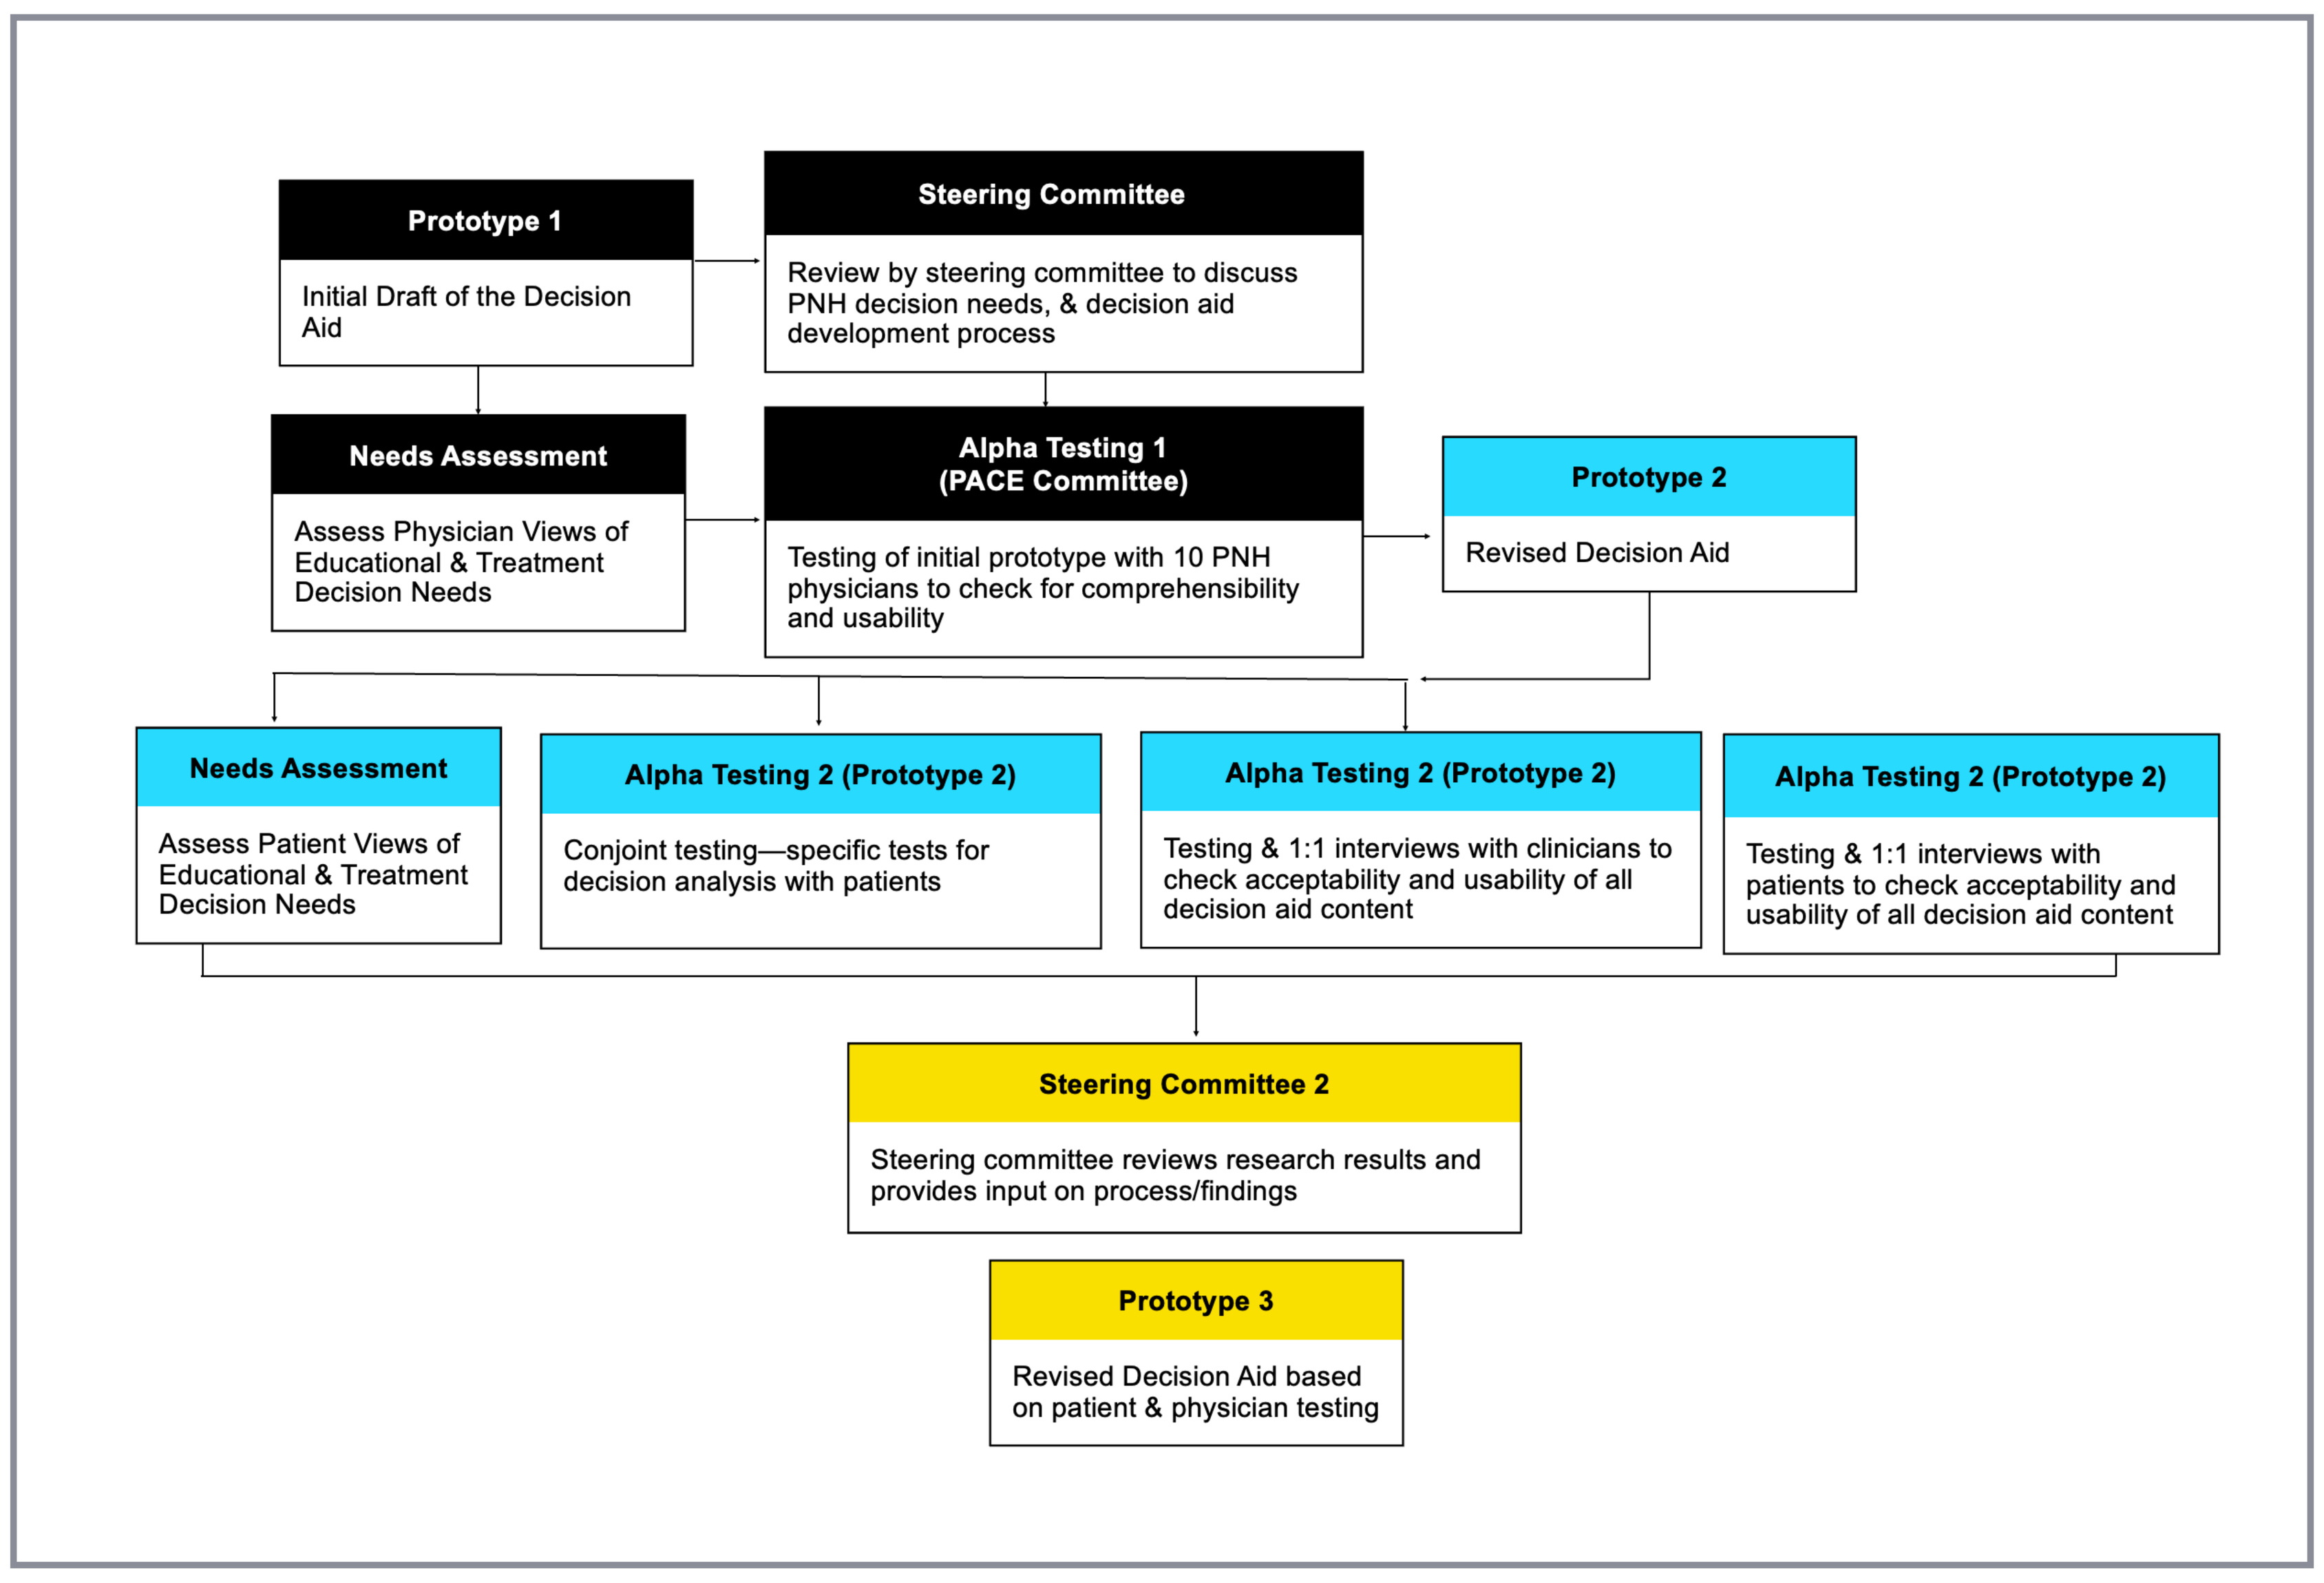
Prototype 1
Initial Draft of the Decision Aid
Steering Committee
Review by steering committee to discuss PNH decision needs, & decision aid development process
Needs Assessment
Assess Physician Views of Educational & Treatment Decision Needs
Alpha Testing 1 (PACE Committee)
Testing of initial prototype with 10 PNH physicians to check for comprehensibility and usability
Prototype 2
Revised Decision Aid
Needs Assessment
Assess Patient Views of Educational & Treatment Decision Needs
Alpha Testing 2 (Prototype 2)
Conjoint testing—specific tests for decision analysis with patients
Alpha Testing 2 (Prototype 2)
Testing & 1:1 interviews with clinicians to check acceptability and usability of all decision aid content
Alpha Testing 2 (Prototype 2)
Testing & 1:1 interviews with patients to check acceptability and usability of all decision aid content
Steering Committee 2
Steering committee reviews research results and provides input on process/findings
Prototype 3
Revised Decision Aid based on patient & physician testing

Results

Development of the DA relied on a team comprised of qualitative researchers to capture decisional needs and facilitate cognitive interviews; market research experts to identify individualized, optimal treatment plans based on conjoint analysis; and a Steering Committee to provide clinical input and represent the patient voice. Harmonization across these different perspectives proved invaluable for designing a DA that was acceptable and relevant for different stakeholders by raising questions and surfacing solutions that work within the PNH workflow. Development of the DA closely followed the International Patient Decision Aids Standards (IPDAS) (**Figure 2**) and the Standards for UNiversal reporting of patient Decision Aid Evaluation (SUNDAE) Checklist.

Figure 2: IPDAS collaboration patient decision aid development model[10]

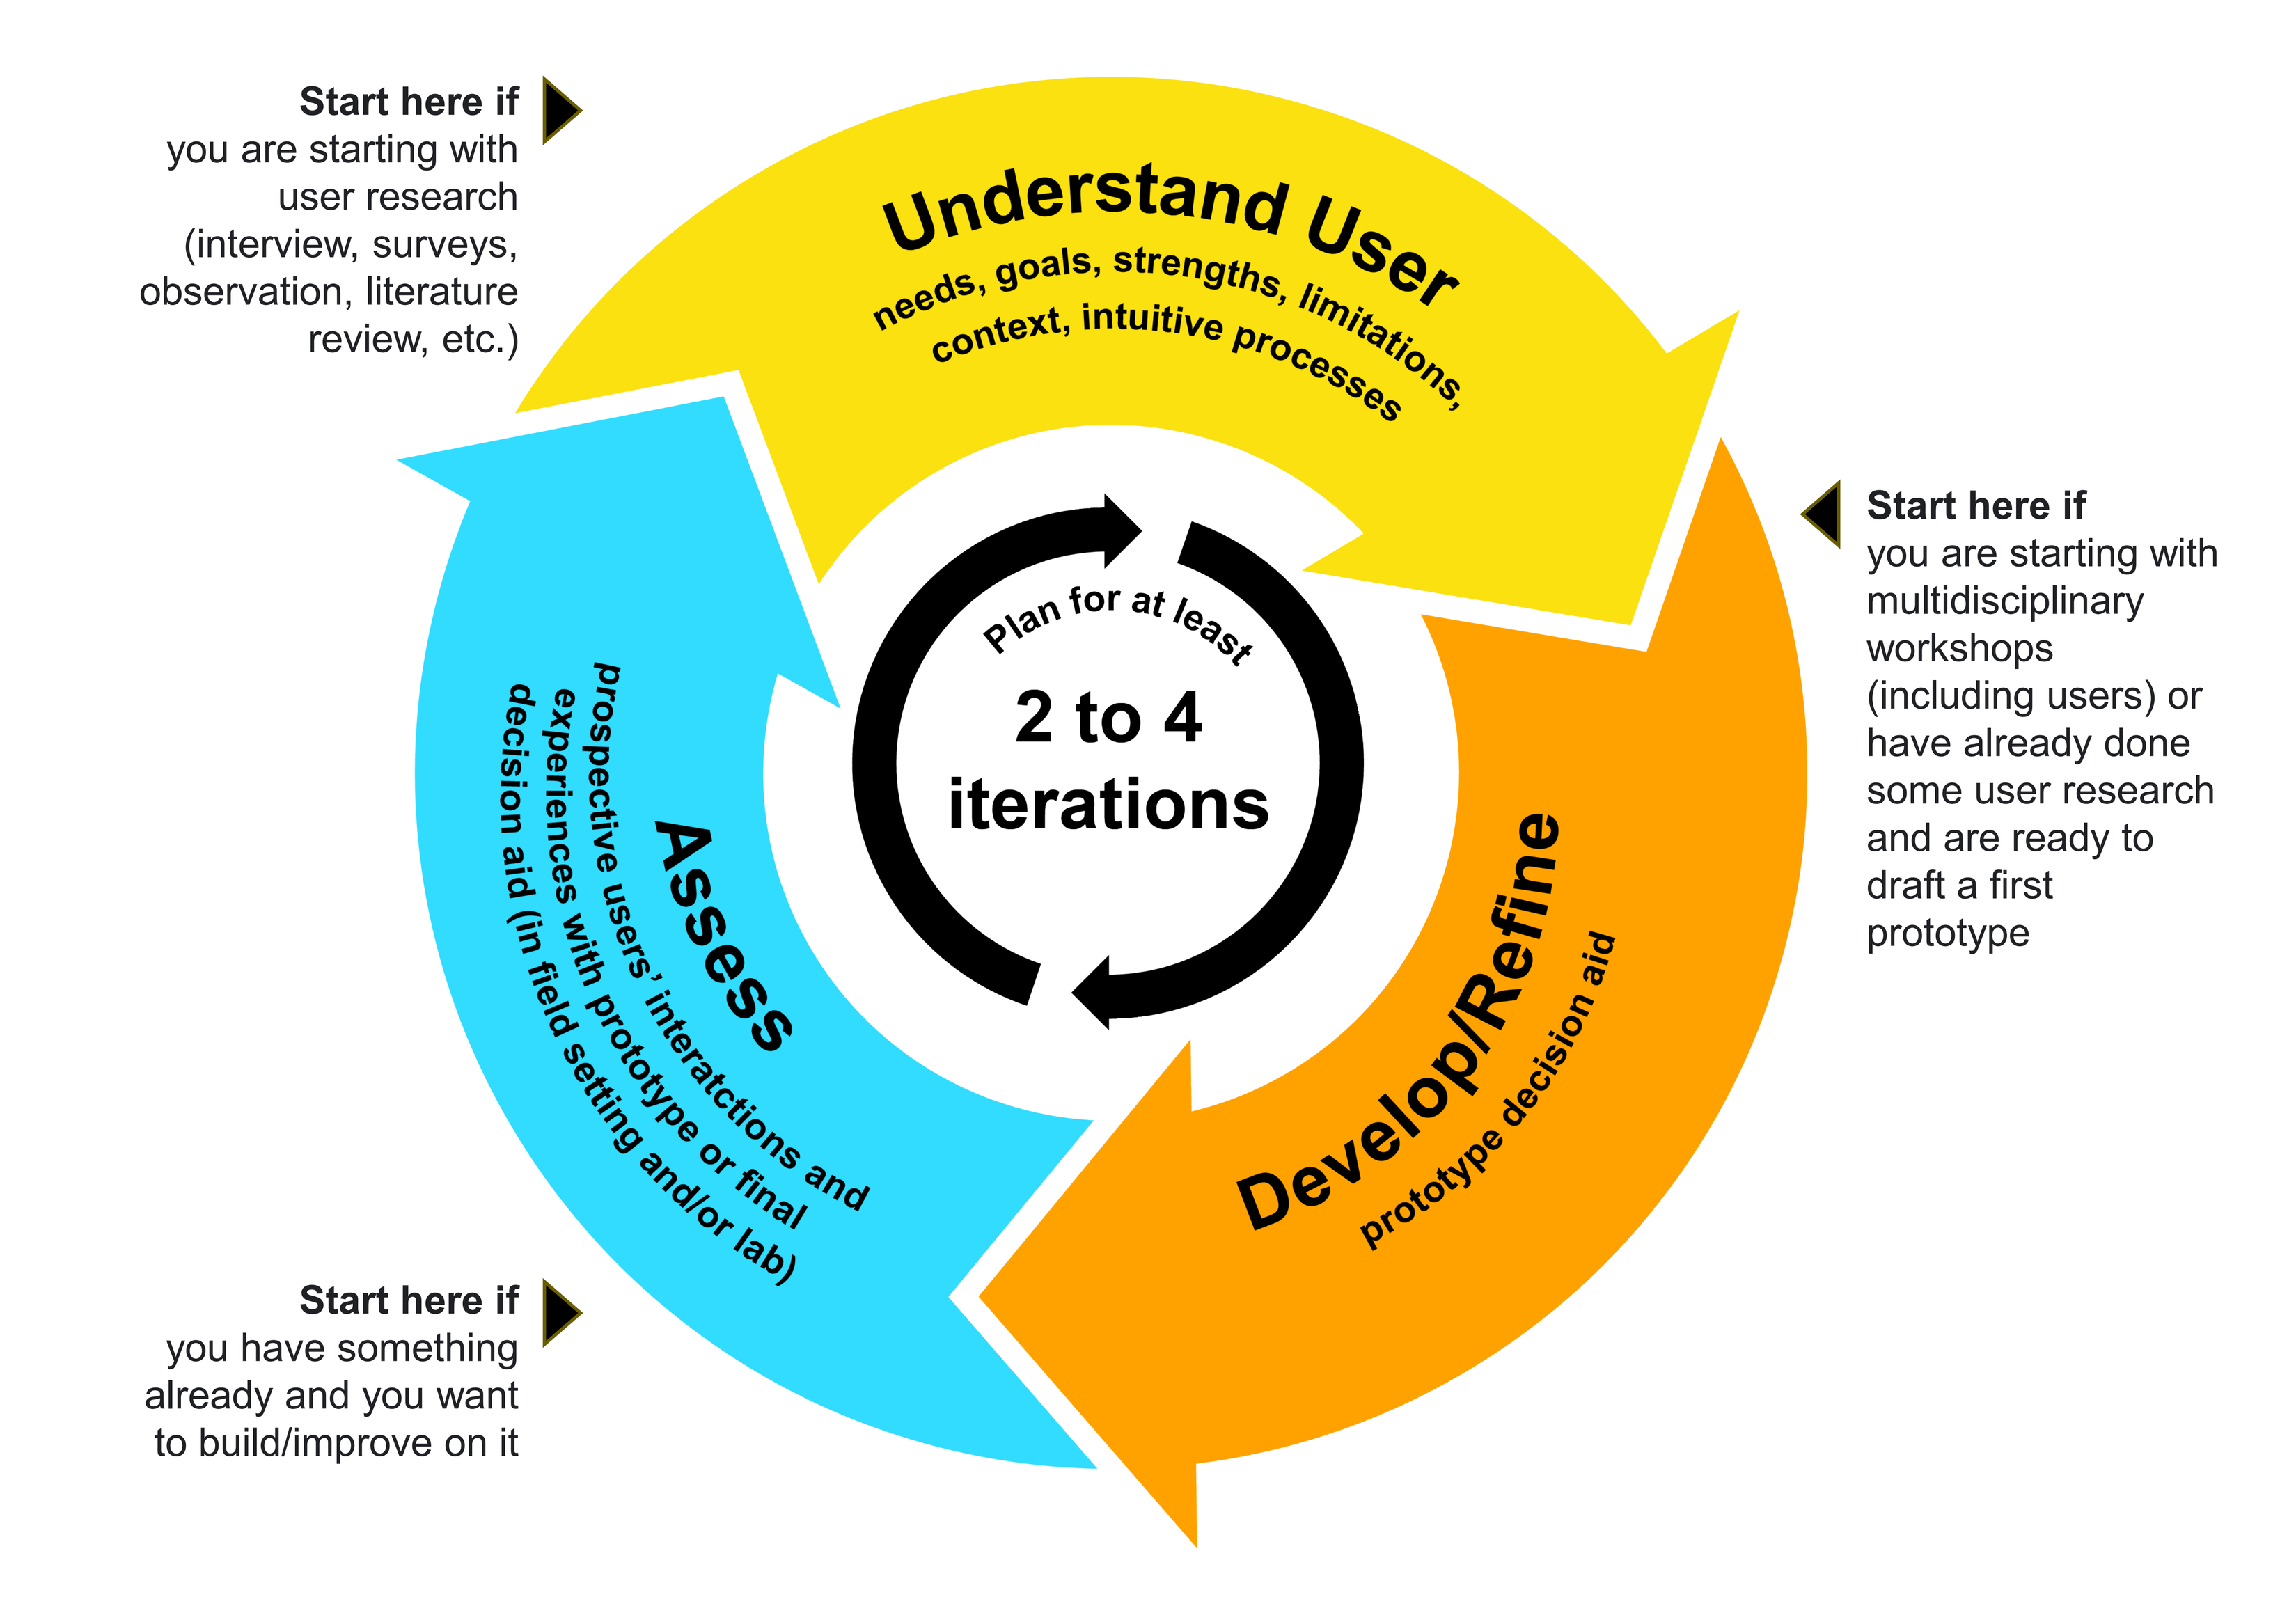
Start here if you are starting with user research (interview, surveys, observation, literature review, etc.)
Understand User
needs, goals, strengths, limitations, context, intuitive processes
Start here if you are starting with multidisciplinary workshops (including users) or have already done some user research and are ready to draft a first prototype
Plan for at least
2 to 4 iterations
Assess
prospective users' interatctions and experiences with prototype or final decision aid (in field setting and/or lab)
Develop/Refine
prototype decision aid
Start here if you have something already and you want to build/improve on it

Conclusions

Qualitative and quantitative data from feasibility and usability testing indicated that the SDM tool was appropriate for the target population and accepted by end-users. This outcome was achieved by following the IPDAS and SUNDAE checklist with ongoing communications and collaboration across a multi-stakeholder team following an iterative testing process. Orientation towards the Steering Committee charter and bi-directional communication across all parties was essential for data collection, analysis, and iterative updates to the web-based DA. Incorporating both patient and provider feedback through the Steering Committee, interviews, and physician expert panels provided ample end-user feedback to guide development. **Figure 3** illustrates how the tool addresses decision-making and clinical challenges faced by individuals affected by PNH and their care teams. This multi-disciplinary approach can be applied as a model for DA development for other health conditions to ensure tool validity and reliability.

Figure 3: Shared decision-making tools address commonly faced care challenges[11]

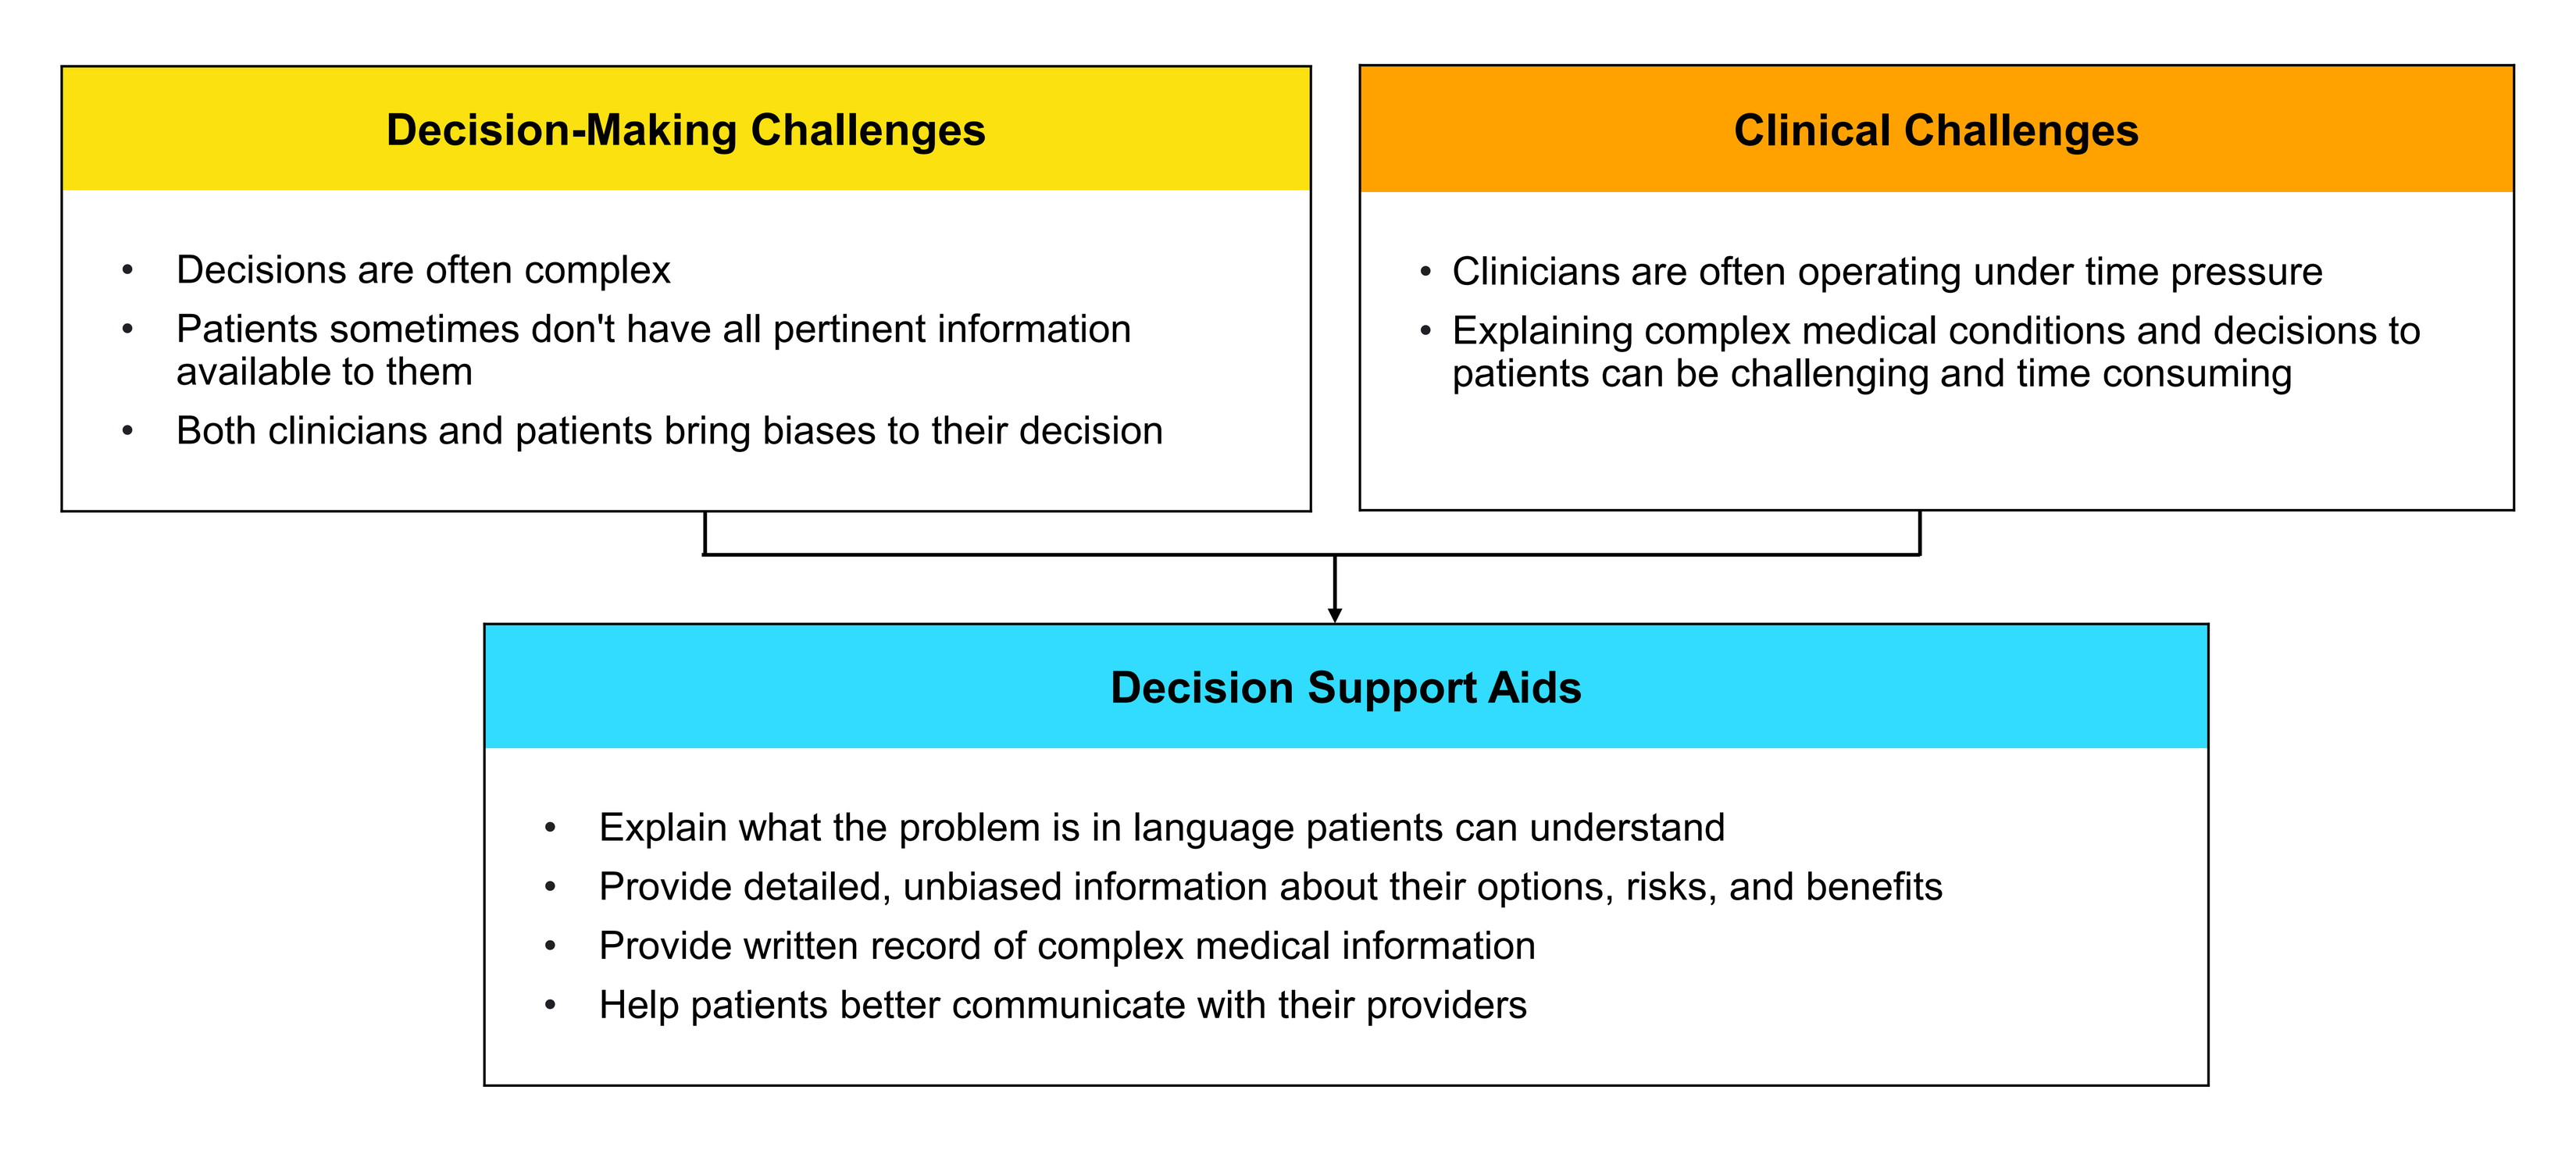
Decision-Making Challenges
Decisions are often complex
Patients sometimes don't have all pertinent information available to them
Both clinicians and patients bring biases to their decision
Clinical Challenges
Clinicians are often operating under time pressure
Explaining complex medical conditions and decisions to patients can be challenging and time consuming
Decision Support Aids
Explain what the problem is in language patients can understand
Provide detailed, unbiased information about their options, risks, and benefits
Provide written record of complex medical information
Help patients better communicate with their providers

Funding support was provided by Apellis Pharmaceuticals.

References

1. Hill A, DeZern AE, Kinoshita T, et al. Paroxysmal nocturnal haemoglobinuria. Nat Rev Dis Primers 2017; 3: 17028. 20170518. DOI: 10.1038/nrdp.2017.28.
2. Wong RSM. Safety and efficacy of pegcetacoplan in paroxysmal nocturnal hemoglobinuria. Ther Adv Hematol 2022; 13: 20406207221114673. 20220728. DOI: 10.1177/20406207221114673.
3. Roth A, Maciejewski J, Nishimura JI, et al. Screening and diagnostic clinical algorithm for paroxysmal nocturnal hemoglobinuria: Expert consensus. Eur J Haematol 2018; 101: 3-11. 20180516. DOI: 10.1111/ejh.13059.
4. Schrezenmeier H, Roth A, Araten DJ, et al. Baseline clinical characteristics and disease burden in patients with paroxysmal nocturnal hemoglobinuria (PNH): updated analysis from the International PNH Registry. Ann Hematol 2020; 99: 1505-1514. 20200510. DOI: 10.1007/s00277-020-04052-z
5. Soliris® (eculizumab) injection, for intravenous use (Prescribing information). Alexion Pharmaceuticals, 2007.
6. Ultomiris® (ravulizumab-cwvz) injection, for intravenous or subcutaneous use (Prescribing information). Alexion Pharmaceuticals, 2018.
7. Empaveli® (pegcetacoplan) injection, for subcutaneous use (Prescribing information). Apellis Pharmaceuticals, 2021.
8. Novartis Phase III APPOINT-PNH trial shows investigational oral monotherapy iptacopan improves hemoglobin to near-normal levels, leading to transfusion independence in all treatment-naïve PNH patients. Novartis: Novartis, 2023.
9. Nunes JC, Baykaner T, Pundi K, DeSutter K, True Hills M, Mahaffey KW, Sears SF, Morin DP, Lin B, Wang PJ, Stafford RS. Design and development of a digital shared decision-making tool for stroke prevention in atrial fibrillation. JAMIA Open. 2023 Feb 2;6(1):ooad003. doi: 10.1093/jamiaopen/ooad003. PMID: 36751465; PMCID: PMC9893868.
10. Witteman HO, Maki KG, Vaisson G, et al. Systematic Development of Patient Decision Aids: An Update from the IPDAS Collaboration. Medical Decision Making. 2021;41(7):736-754.
11. Association of Community Cancer Centers, 2020. Engaging Patients in Healthcare Choices: An Overview of Patient Decision Aids.

Poster presented at ISPOR EU, 17–20 November 2024, Barcelona, Spain