PCR220

Patient-Reported Outcomes in LGMD2E/R4, 2D/R3, and 2C/R5: Descriptive Analysis of Baseline PROMIS Data From the JOURNEY Natural History Study

Ivana Audhya[1], Alise Nacson[1], Piper Fromy[2], Katherine Gooch[1], Herb Stevenson[1]

[1]Sarepta Therapeutics, Inc., Cambridge, MA, USA; [2]Seeing Theta, Saumur, France

Presented at ISPOR Europe
17–20 November 2024
Barcelona, Spain

Methods (cont)

Patient-Reported Outcomes Measurement Information System (PROMIS) measures

Caregivers of patients 4–7 years of age

- PROMIS Parent Proxy Item Bank Version 2.0 – Physical Function Mobility (23 items)
- PROMIS Parent Proxy Item Bank Version 2.0 – Physical Function Upper Extremity – Short Form 8a (8 items)
- PROMIS Parent Proxy Item Bank Version 1.0 – Strength Impact – Short Form 8a (8 items)
- PROMIS Parent Proxy Item Bank Version 1.0 – Sleep Disturbance – Short Form 8a (8 items)
- PROMIS Parent Proxy Item Bank Version 2.0 – Fatigue – Short Form 10a (10 items)
- PROMIS Parent Proxy Item Bank Version 1.0 – Positive Affect – Short Form 8a (8 items)

Patients 8–17 years of age

- PROMIS Pediatric Item Bank Version 2.0 – Physical Function Mobility (24 items, for ambulatory subjects)
- PROMIS Pediatric Item Bank Version 2.0 – Physical Function Upper Extremity – Short Form 8a (8 items)
- PROMIS Pediatric Short Form Version 1.0 – Strength Impact – 8a (8 items)
- PROMIS Pediatric Item Bank Version 1.0 – Sleep-Related Disturbance – Short Form 8a (8 items)
- PROMIS Pediatric Item Bank Version 2.0 – Fatigue – Short Form 10a (10 items)
- PROMIS Pediatric Item Bank Version 1.0 – Positive Affect – Short Form 8a (8 items)

Patients ≥18 years of age

- PROMIS Adult Item Bank Version 2.0 – Physical Function Mobility (15 items, for ambulatory patients)
- PROMIS Adult Item Bank Version 2.0 – Physical Function Upper Extremity (46 items, for all patients regardless of ambulatory status)
- PROMIS Adult Item Bank Version 1.0 – Sleep Disturbance – Short Form 8a (8 items)
- PROMIS Adult Item Bank Version 1.0 – Fatigue – Short Form 7a (8 items)
- PROMIS Adult Item Bank Version 1.0 – Positive Affect – Short Form 15a (15 items)

PROMIS measures interpretation

- Higher PROMIS scores equal more of the concept being measured

Results (cont)

Supplemental Table 1 *Baseline Race and Ethnicity Characteristics Stratified by LGMD Subtypes*

Characteristic	2C/R5 (n=34)	2D/R3 (n=32)	2E/R4 (n=25)	Overall (N=91)
Race[a], n (%)				
American Indian/Alaska Native	0 (0)	1 (3.0)	0 (0)	1 (1.1)
Asian	3 (8.8)	1 (3.0)	1 (3.4)	5 (5.5)
Black/African American	0 (0)	0 (0)	3 (10.3)	3 (3.3)
White	27 (79.4)	30 (90.9)	19 (65.5)	73 (80.2)
Other[b]	4 (11.8)	1 (3.0)	6 (20.7)	9 (9.9)
Ethnicity, n (%)				
Hispanic/Latino	2 (5.9)	3 (9.4)	0 (0)	5 (5.5)
Not Hispanic/Latino	31 (91.2)	29 (90.6)	25 (100.0)	85 (93.4)
Unknown	1 (2.9)	0 (0)	0 (0)	1 (1.1)

[a]3 participants selected more than 1 race category. [b]Excludes Native Hawaiian, other Pacific Islander. LGMD=limb-girdle muscular dystrophy.

Supplemental Table 2 *Baseline PROMIS Scores by LGMD Subtype and Ambulatory Status*

		2C/R5 (n=34)	2D/R3 (n=32)	2E/R4 (n=25)	Ambulatory (n=45)	Nonambulatory (n=46)
A PROMIS Strength Impact						
4–7 y	n	2	3	3	8	NA
	Mean (SD)	4.6 (0.18)	2.4 (1.32)	4.2 (0.76)	3.6 (1.32)	NA
8–12 y	n	7	4	3	11	3
	Mean (SD)	3.2 (1.24)	3.9 (0.62)	3.4 (2.10)	3.7 (1.31)	2.7 (0.58)
13–17 y	n	13	7	5	10	15
	Mean (SD)	2.3 (1.25)	2.9 (1.86)	2.9 (1.92)	3.8 (1.44)	1.8 (1.03)
≥18 y	n	NA	NA	NA	NA	NA
	Mean (SD)	NA	NA	NA	NA	NA
B PROMIS Positive Affect						
4–7 y	n	2	3	3	8	NA
	Mean (SD)	4.8 (0.27)	3.0 (1.84)	4.5 (0.51)	4.0 (1.33)	NA
8–12 y	n	7	4	3	11	3
	Mean (SD)	4.2 (0.44)	3.8 (0.91)	4.4 (0.80)	4.2 (0.71)	3.9 (0.50)
13–17 y	n	13	7	5	10	15
	Mean (SD)	3.7 (0.62)	3.4 (1.03)	4.0 (0.82)	3.9 (0.63)	3.5 (0.86)
≥18 y	n	12	18	14	16	28
	Mean (SD)	3.4 (0.77)	3.5 (1.05)	3.8 (0.75)	3.5 (0.88)	3.6 (0.90)
C PROMIS Sleep Disturbance						
4–7 y	n	2	3	3	8	NA
	Mean (SD)	1.6 (0.44)	1.6 (0.52)	1.7 (0.63)	1.6 (0.47)	NA
8–12 y	n	7	4	3	11	3
	Mean (SD)	1.9 (0.86)	2.8 (0.89)	1.8 (0.43)	2.1 (0.86)	2.2 (1.05)
13–17 y	n	13	7	5	10	15
	Mean (SD)	2.3 (1.00)	2.2 (0.91)	1.7 (0.60)	2.2 (0.77)	2.1 (1.01)
≥18 y	n	12	18	14	16	28
	Mean (SD)	2.5 (0.94)	2.8 (1.08)	2.8 (0.96)	2.7 (0.89)	2.8 (1.06)

LGMD=limb-girdle muscular dystrophy; PROMIS=Patient-Reported Outcome Measurement Information System; y=years of age.