EE676

ECONOMIC BURDEN AND UNMET NEEDS OF RETINITIS PIGMENTOSA: EXPERT PERSPECTIVE IN BRAZIL AND MEXICO

Yann Laurent Boixel[1], Tatiana Pineda[2], Francesca Albanese[3], Renato Watanabe de Oliveira[4], Carolina Morfín Thurmer[5], Francisco Javier Picó Guzmán[5]

1 – Janssen, Mexico City, Mexico; 2 – Janssen-Cilag Colombia, Bogota, Colombia; 3 – Janssen, Panama, Panama; 4 – Janssen-Cilag Brasil, São Paulo, Brazil; 5 – Lifesciences Consultants, Madrid, Spain

Key Takeaway

The results suggest a significant impact of RP from an economical perspective, with most of the economic burden relying on indirect costs. Patients, of which high proportion deal with vision loss and work impairment, also cope with high burden

Conclusions

Most of the total RP annual cost is indirect, specific related to productivity loss and secondary disease treatment

The RP diagnosis can take more than 7 years, which results from lack of awareness and healthcare system complexities

A significant proportion of RP patients are considered legally blind or present central vision loss, which impact other domains of their lives

Introduction

- Retinitis Pigmentosa (RP) is group of heterogeneous inherited retinal dystrophies, characterized by severe and progressive visual loss, finally leading to complete blindness[1]
- Currently, there is only one treatment option approved for a specific form of RP, however access to it remains a challenge. In general, the care of RP patients is limited, with supportive therapies and management of possible comorbid ocular conditions, such as cataracts, cystoid macular edema and glaucoma[2]
- As a result, the disease has a relevant burden to the patient and to the society, with increased mortality risk, impact in quality of life, productivity, economic impact etc[3]
- In Latin America, there is scarce evidence on the burden of RP from patient and society perspectives. This study estimates the economic burden of RP in Brazil and Mexico from a societal perspective.

Methods

Figure 1: study design

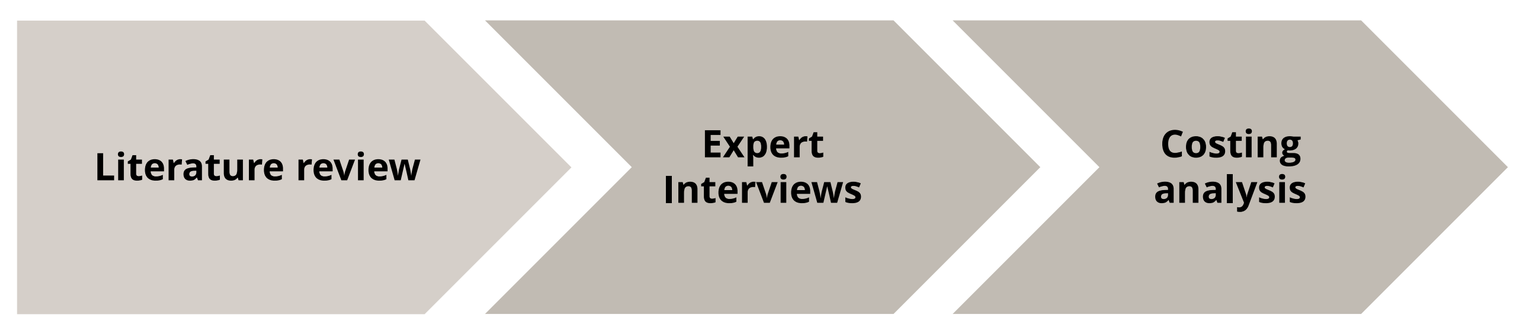
Literature review
Expert Interviews
Costing analysis

Literature review

- A systematic literature review was conducted on studies describing economic, clinical, and social burden for patients with RP, to define the variables needed in Cost Of Illness (COI) estimation. PubMed, Science Direct, Google Scholar, and Scielo databases were used. Research published between January 2018 and December 2023 in English, Spanish, or Portuguese were included.

Interviews

- Data on the clinical and social burden, and resources used, were collected through double-blinded, semi-structured, online interviews with medical experts and patients' associations groups, from Brazil and Mexico.

Costing

- Total direct, indirect and intangible costs were estimated in the study. Data from the literature review and the interviews were combined to calculate costs. Costs were calculated by multiplying the healthcare resources by cost of unit in each country in the estimated population. Unit costs were collected from official pricing and reimbursement lists, and secondary data of the respective healthcare setting and country. Local currencies were converted to 2022 USD adjusted by purchasing power parity (PPP) obtained from The World Bank database.

Results

Participants' profile

Table 1: Profile of participants

Variable	Brazil	Mexico
Profile		
Medical experts	4	4
Patient association groups	1	2

RP patient's profile

Table 2: Description of the demographic and clinical profile

Variable	Brazil	Mexico
Average number of RP patients followed-up by medical expert, per year, n (SD)	468 (112)	203 (82)
Age distribution, %		
<30 years-old	22	20
30-39 years-old	29	31
40-59 years-old	23	33
60-69 years-old	13	9
≥70 years-old	13	7
RP genetic subtype, %		
Autosomal dominant	11	11
Autosomal recessive	35	35
Sporadic	34	34
X-Linked	20	20

Diagnosis

- Participants reported that it took 5 to 7.6 years from first symptom to diagnosis of RP. The main reason include **lack of awareness from physicians and patients**, which may reflect in misdiagnosis, and **delayed referrals** across different specialties.

Figure 2: Time to RP diagnosis in years

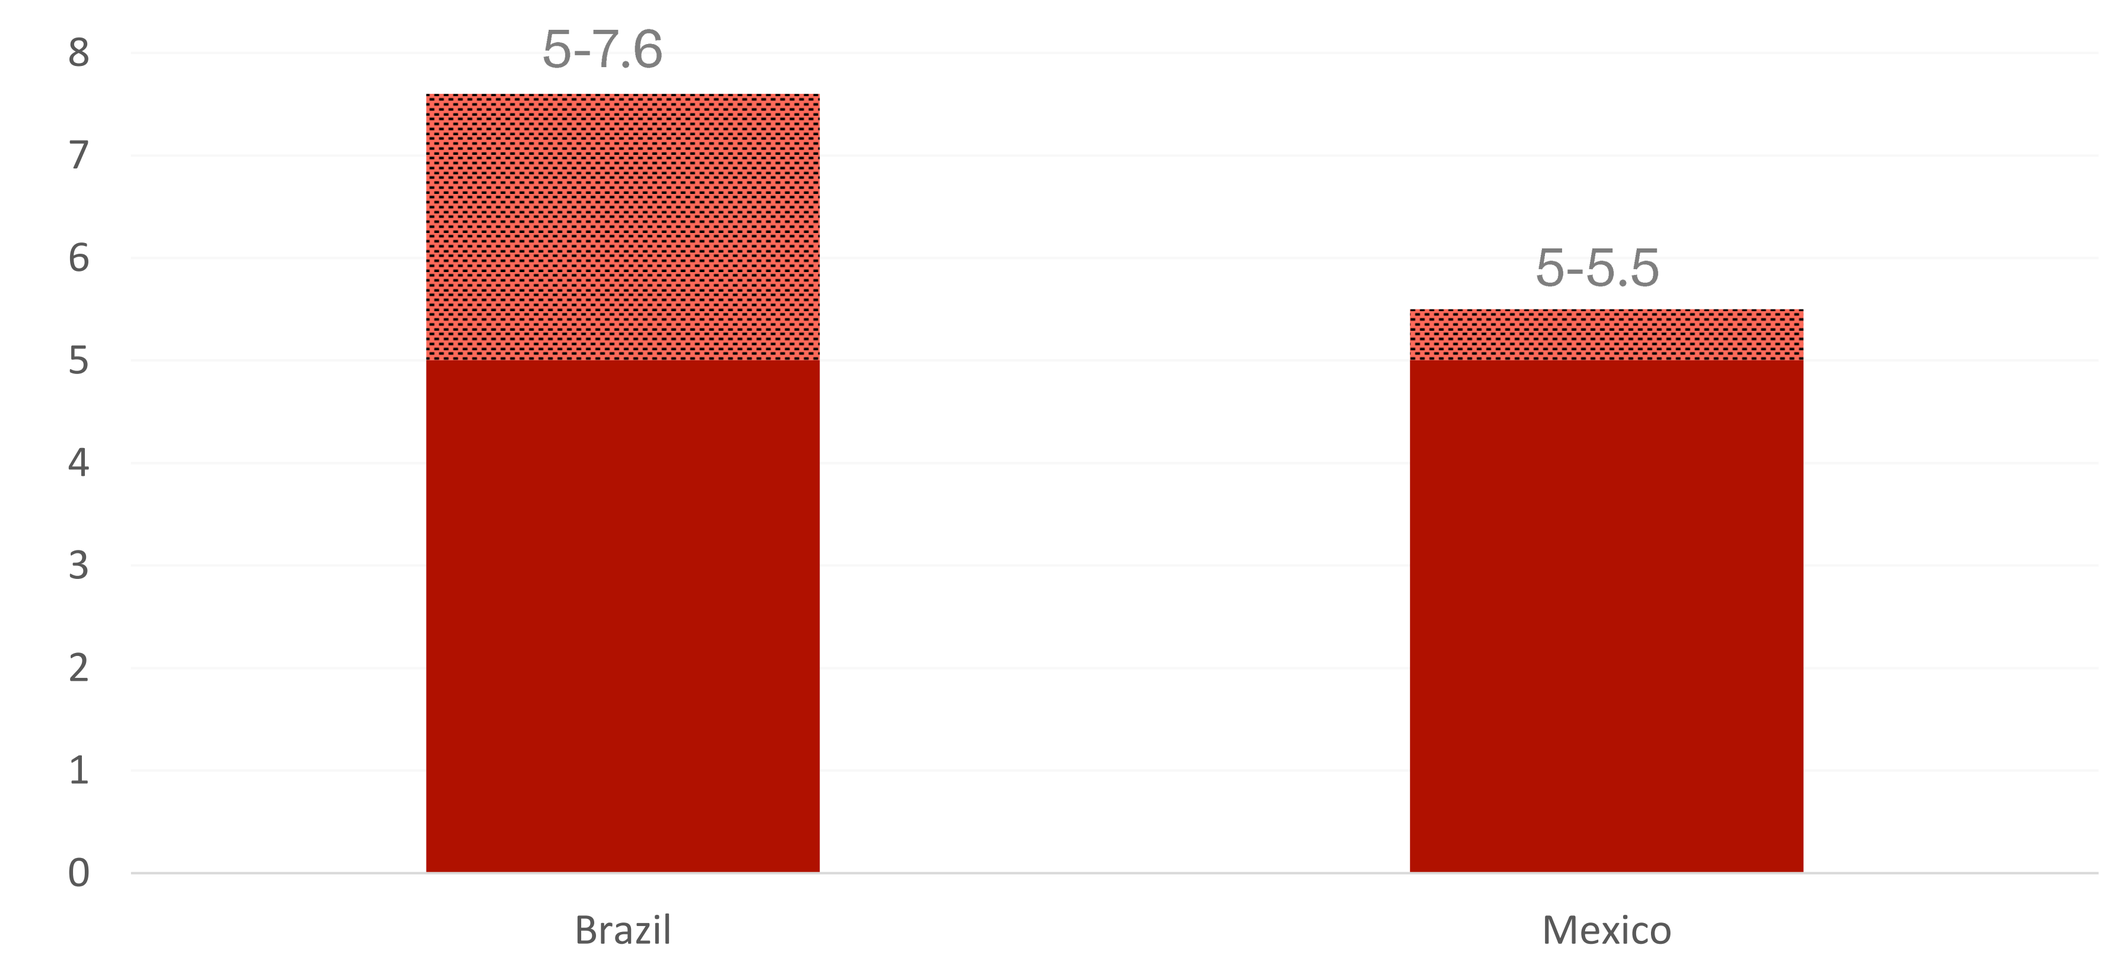
8
7
6
5
4
3
2
1
0
5-7.6
5-5.5
Brazil
Mexico

Clinical profile

- Most RP patients already had peripheral vision loss, and a relevant proportion were considered legally blind.

Figure 3: Proportion of RP patients presenting the respective symptom

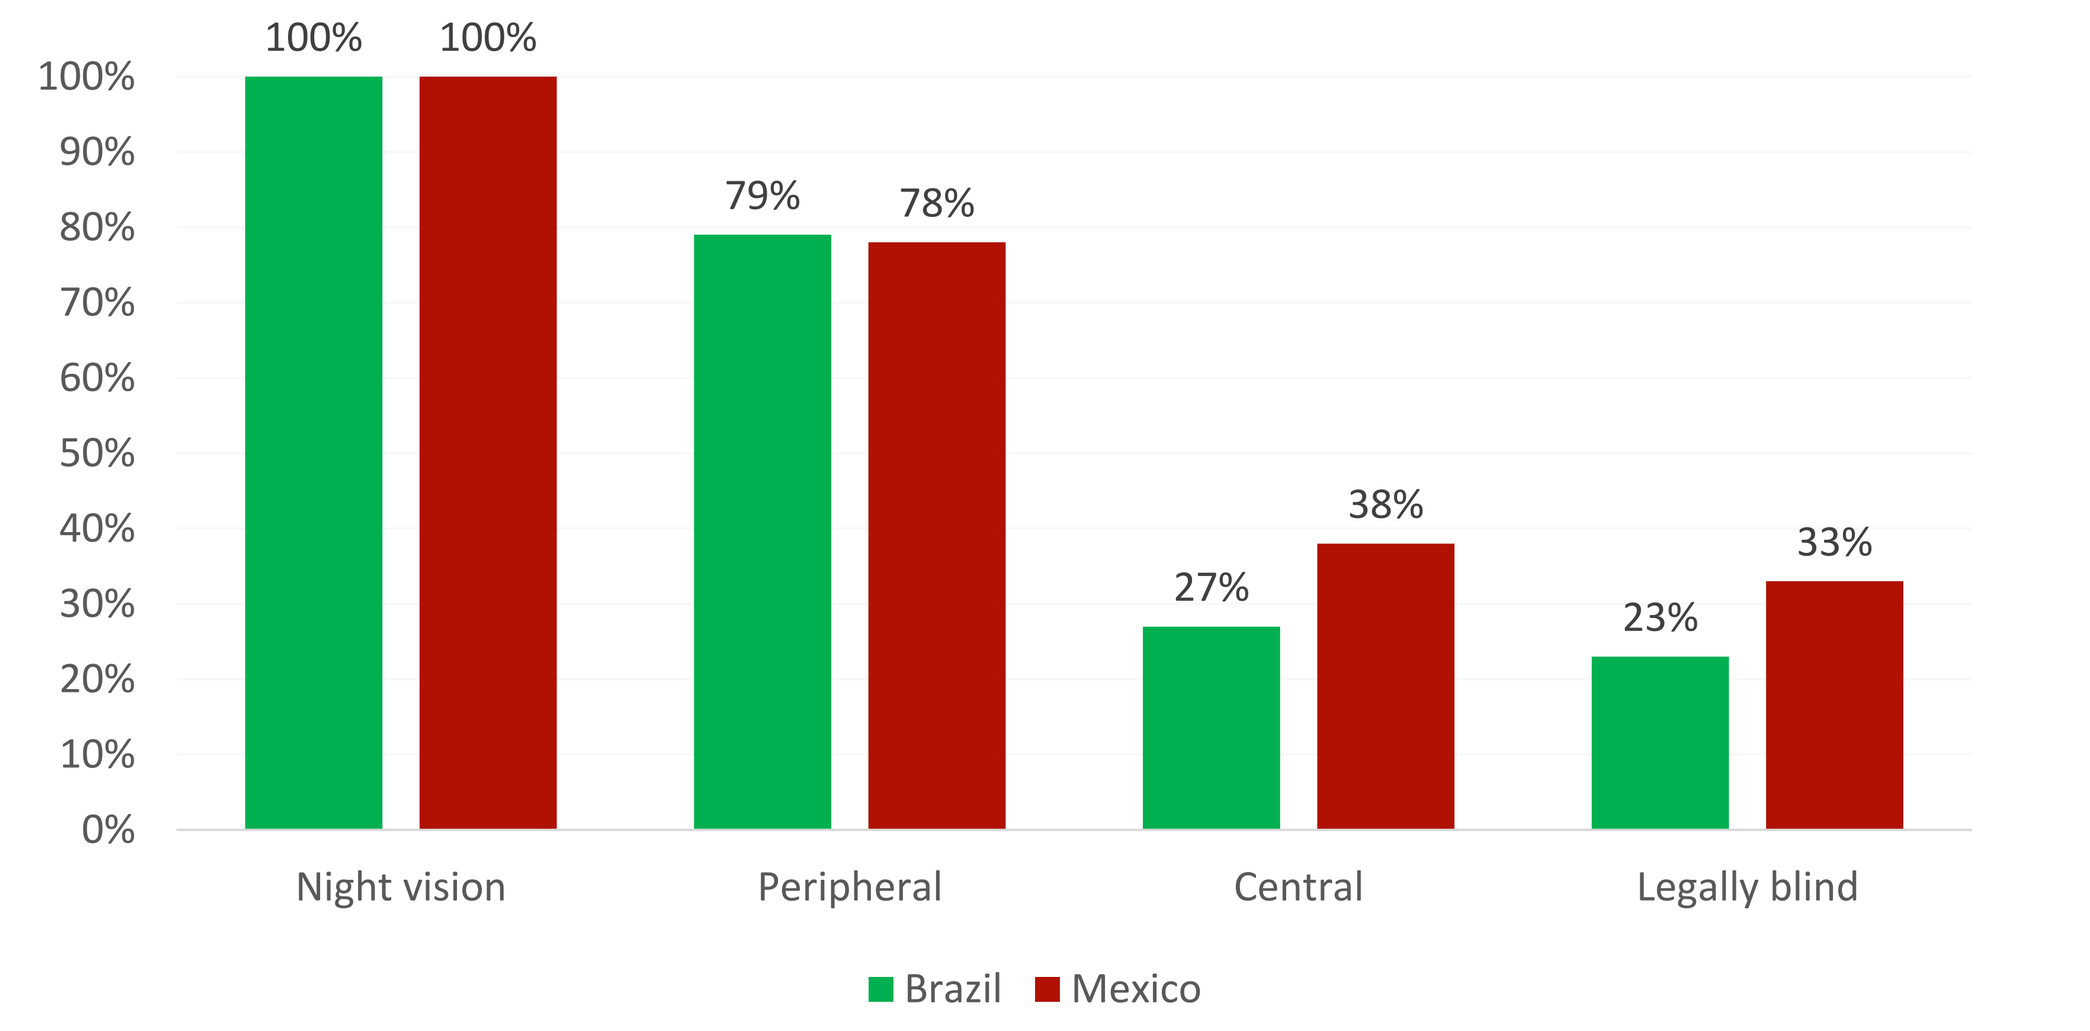
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
100% 100%
79% 78%
27% 38%
23% 33%
Night vision
Peripheral
Central
Legally blind
Brazil
Mexico

Costs

- The average of total annual cost per RP patient is around 1.9k USD and 2.4k USD in Mexico and Brazil, respectively, with a total annual cost of **50M USD** and **102M USD** per year, considering estimated prevalent patients in both countries (26k and 43k)

Figure 4: Distribution of type of costs of the total annual cost per patient, on both countries

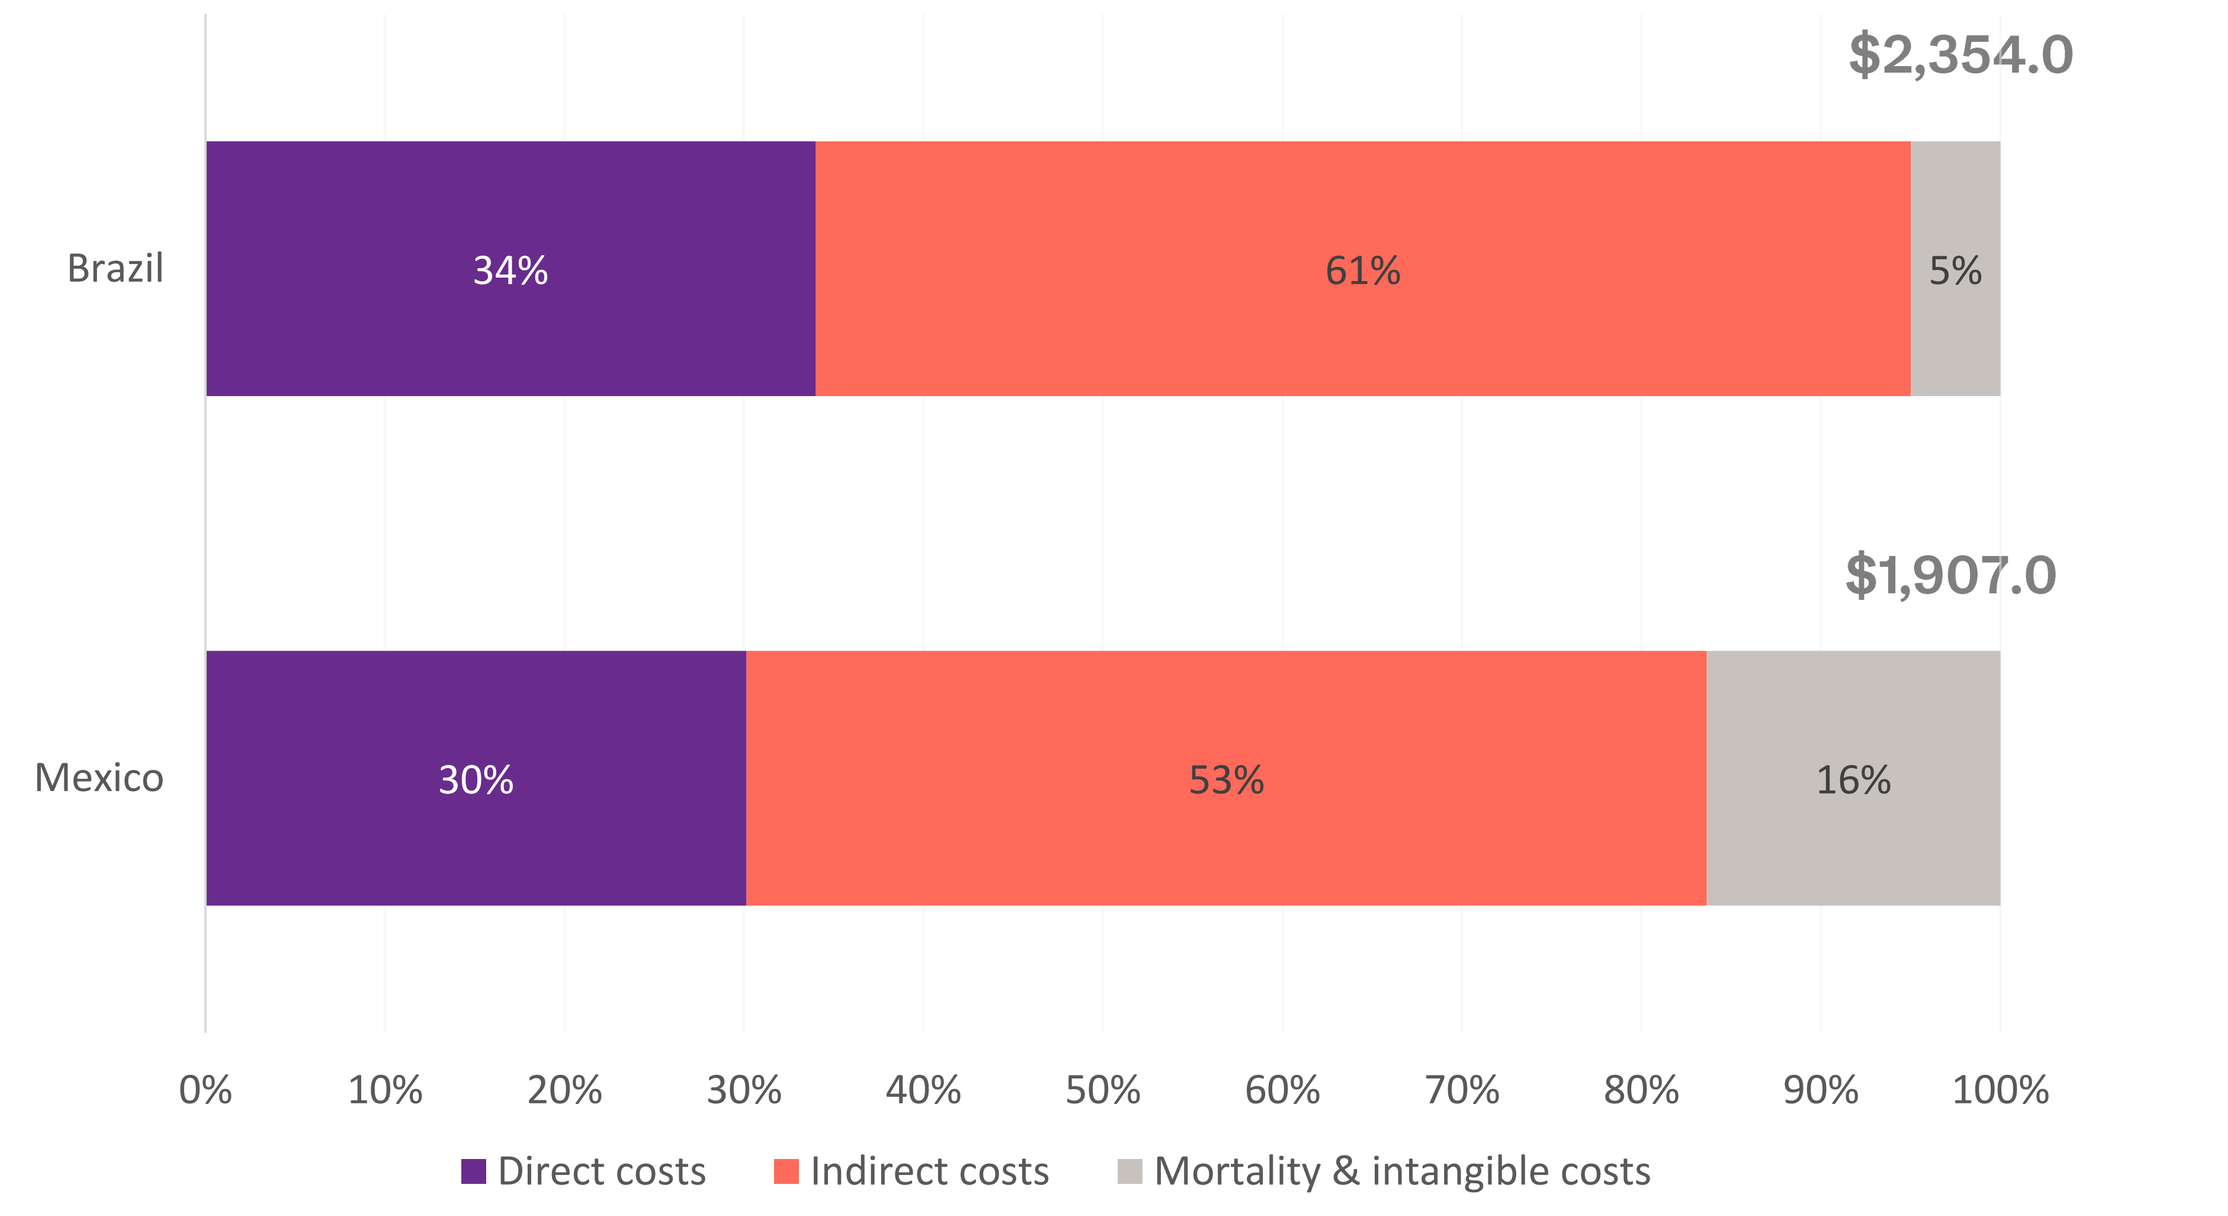
$2,354.0
Brazil
34%
61%
5%
$1,907.0
Mexico
30%
53%
16%
0% 10% 20% 30% 40% 50% 60% 70% 80% 90% 100%
Direct costs
Indirect costs
Mortality & intangible costs

- Of the total costs, the largest proportion is related to indirect costs (53% and 61% in Mexico and Brazil, respectively).
- Of the indirect costs, productivity loss and treatments of comorbidities were the most impactful factors on both countries. For direct costs, formal care, ophthalmologist appointment and mental health support were the main points in Brazil and Mexico, respectively.

Figure 5: Proportion of type of costs in the total annual cost per patient, on both countries

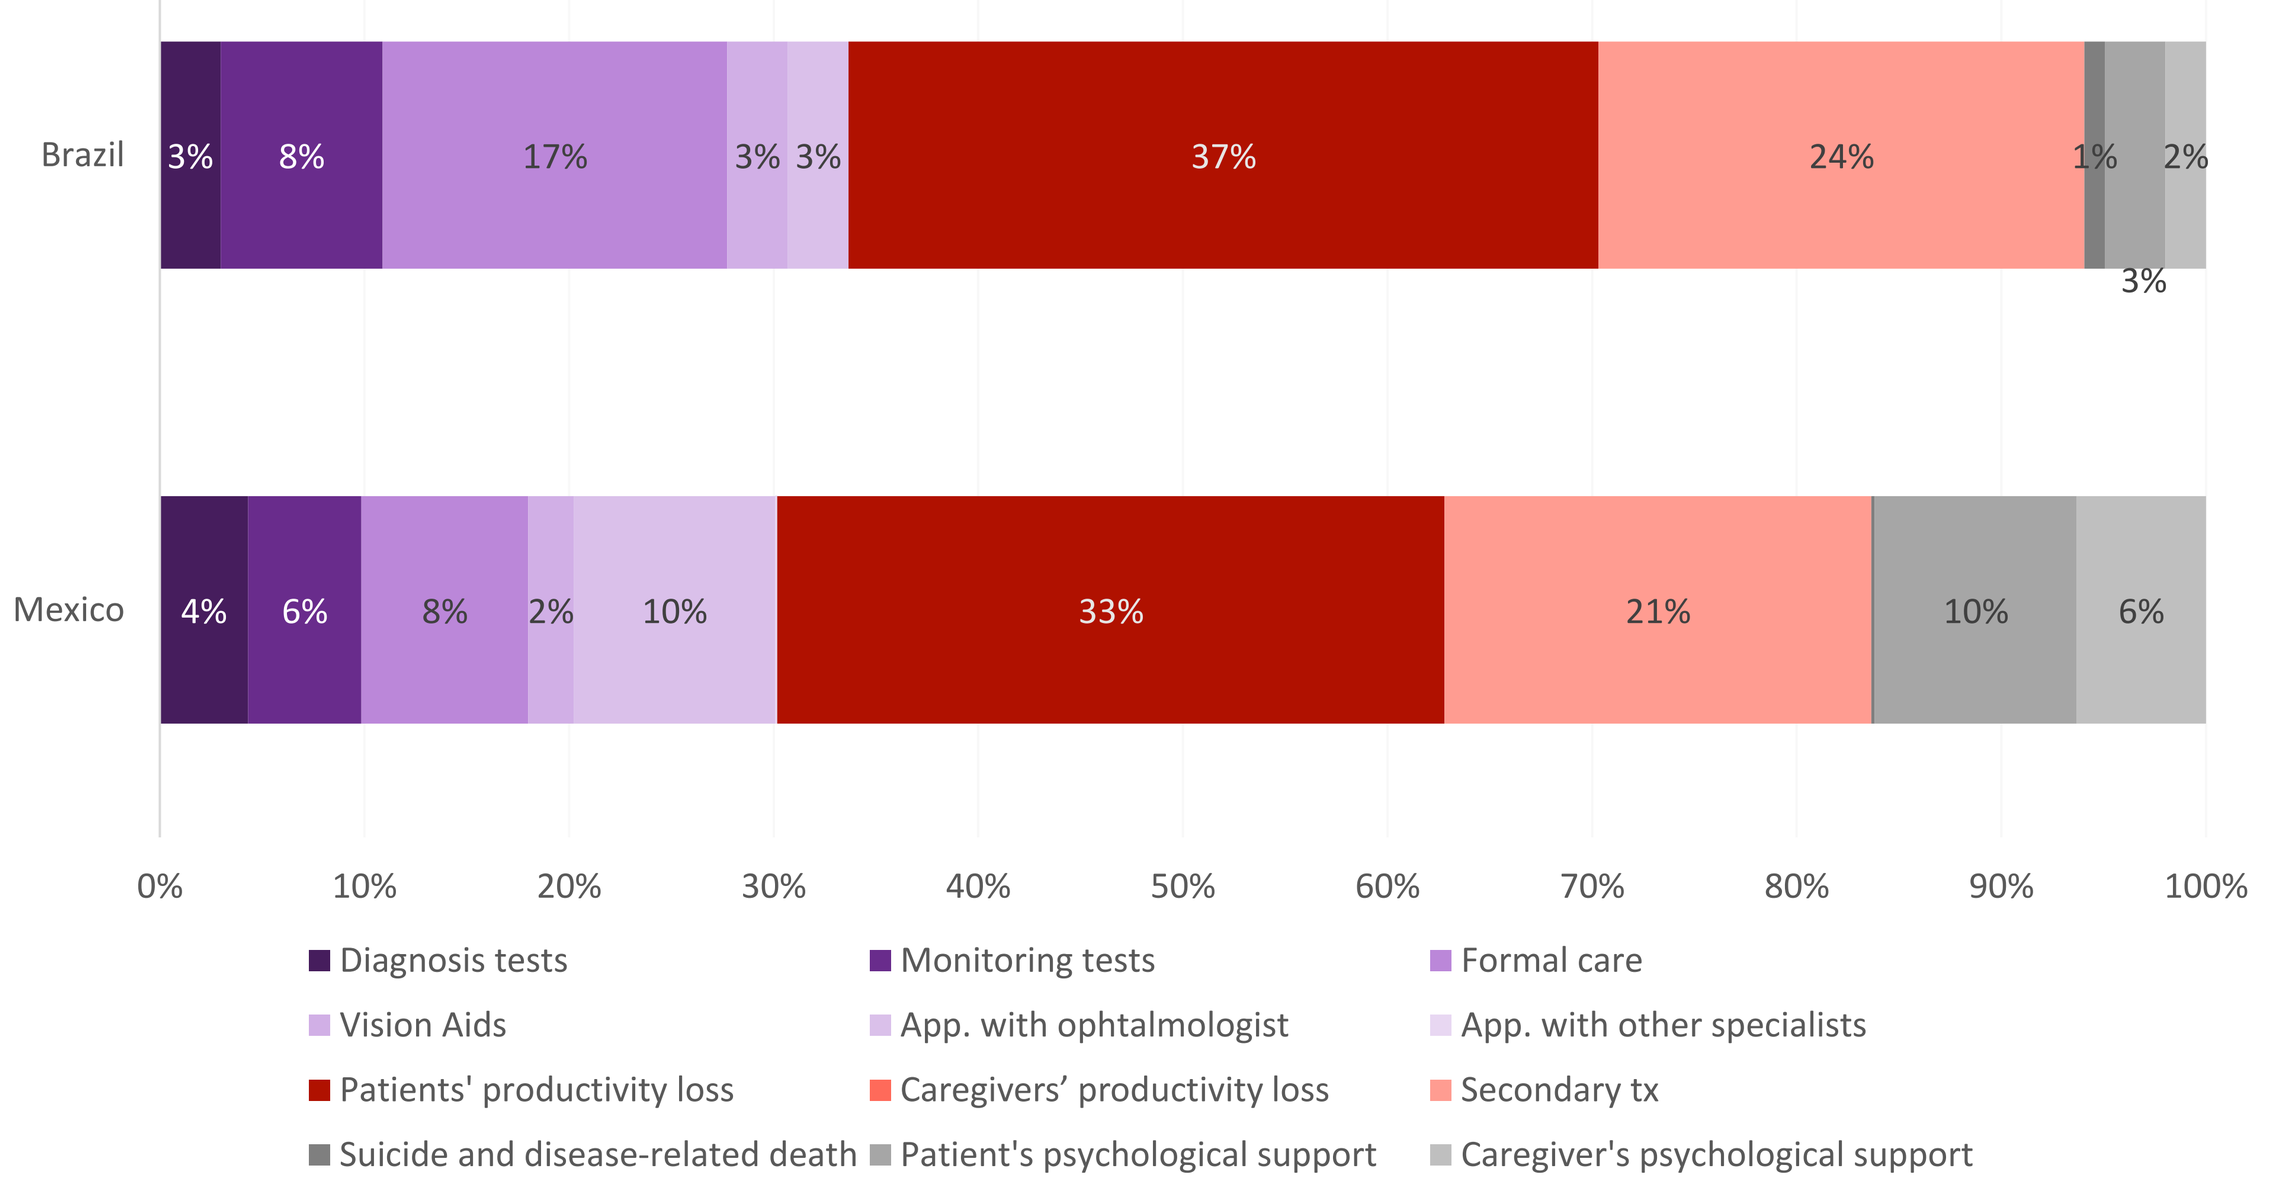
Brazil
3% 8% 17% 3% 3% 37% 24% 1% 2%
3%
Mexico
4% 6% 8% 2% 10% 33% 21% 10% 6%
0% 10% 20% 30% 40% 50% 60% 70% 80% 90% 100%
Diagnosis tests
Monitoring tests
Formal care
Vision Aids
App. with ophtalmologist
App. with other specialists
Patients' productivity loss
Caregivers' productivity loss
Secondary tx
Suicide and disease-related death
Patient's psychological support
Caregiver's psychological support

- In both countries, a relevant proportion of patients had some level of working impairment.
- Age-related macular edema and diabetic retinopathy were the comorbidities that most affected the cost of treatment.

Figure 6: Distribution of RP patients according to working impairment

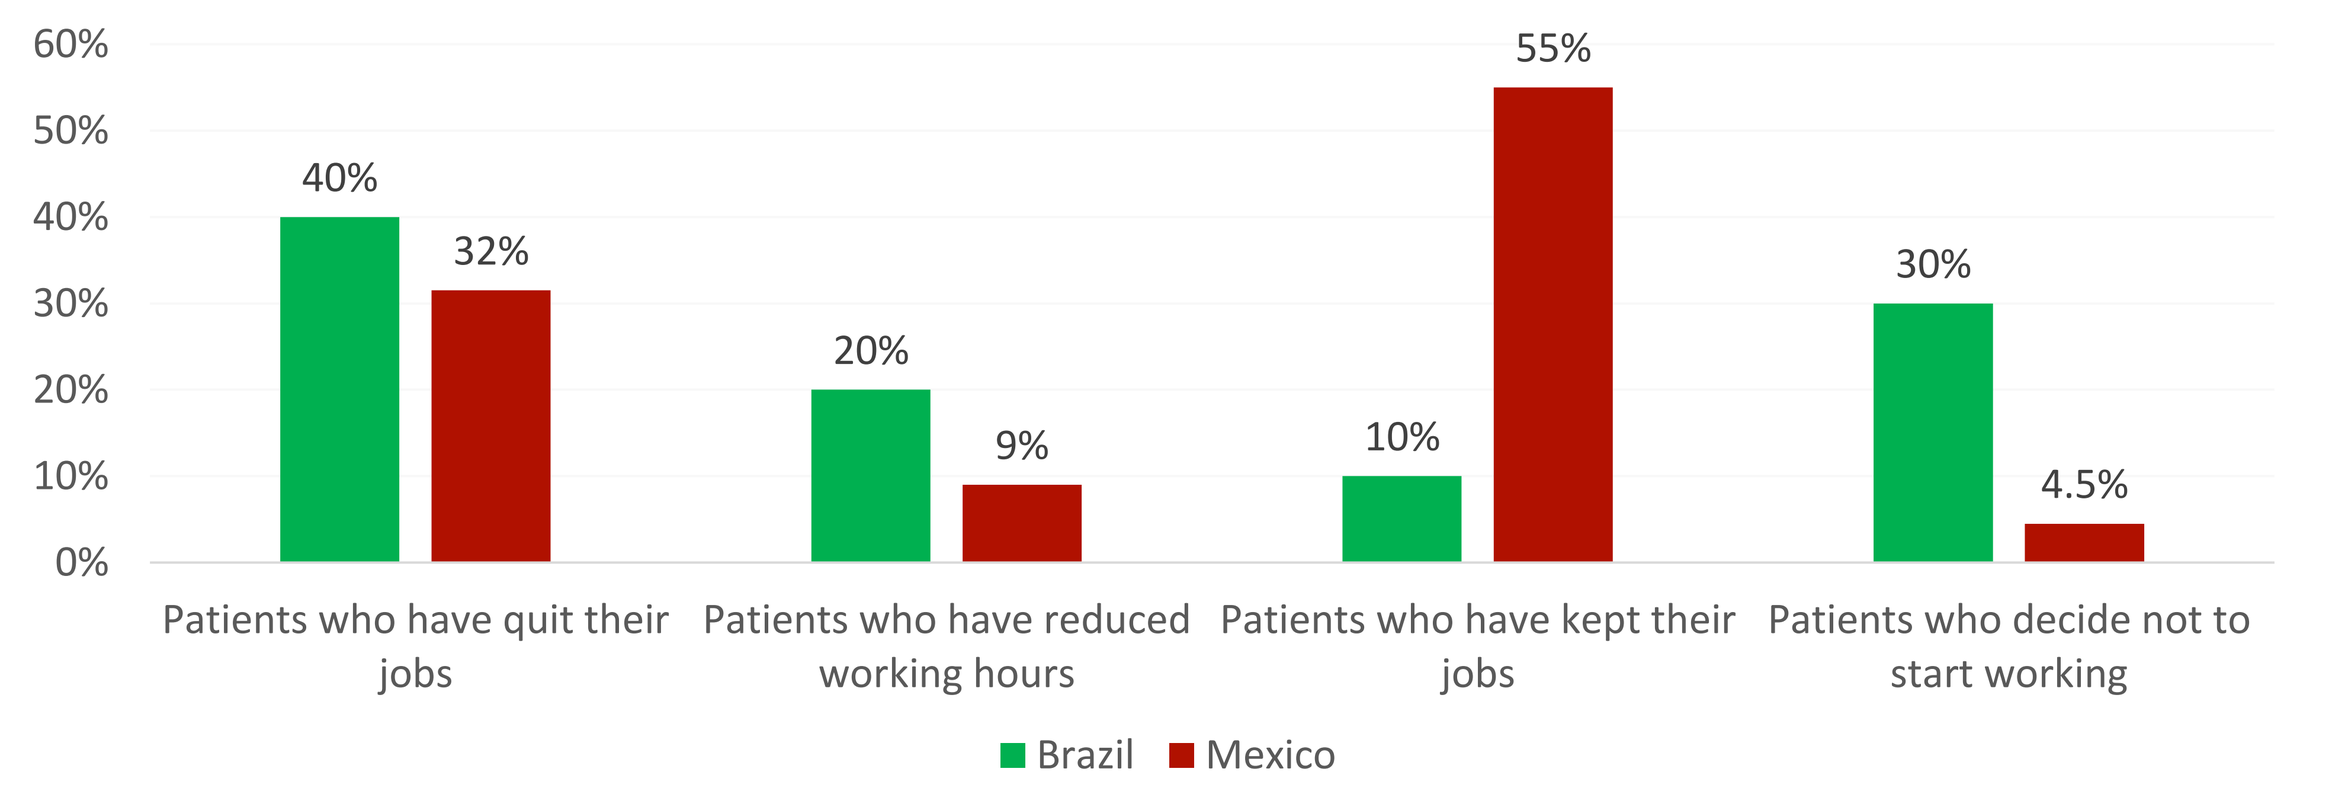
60%
50%
40%
30%
20%
10%
0%
40% 32%
20% 9%
10% 55%
30% 4.5%
Patients who have quit their jobs
Patients who have reduced working hours
Patients who have kept their jobs
Patients who decide not to start working
Brazil
Mexico

Figure 7: Impact of treatment cost of RP comorbidities in the total secondary disease costs

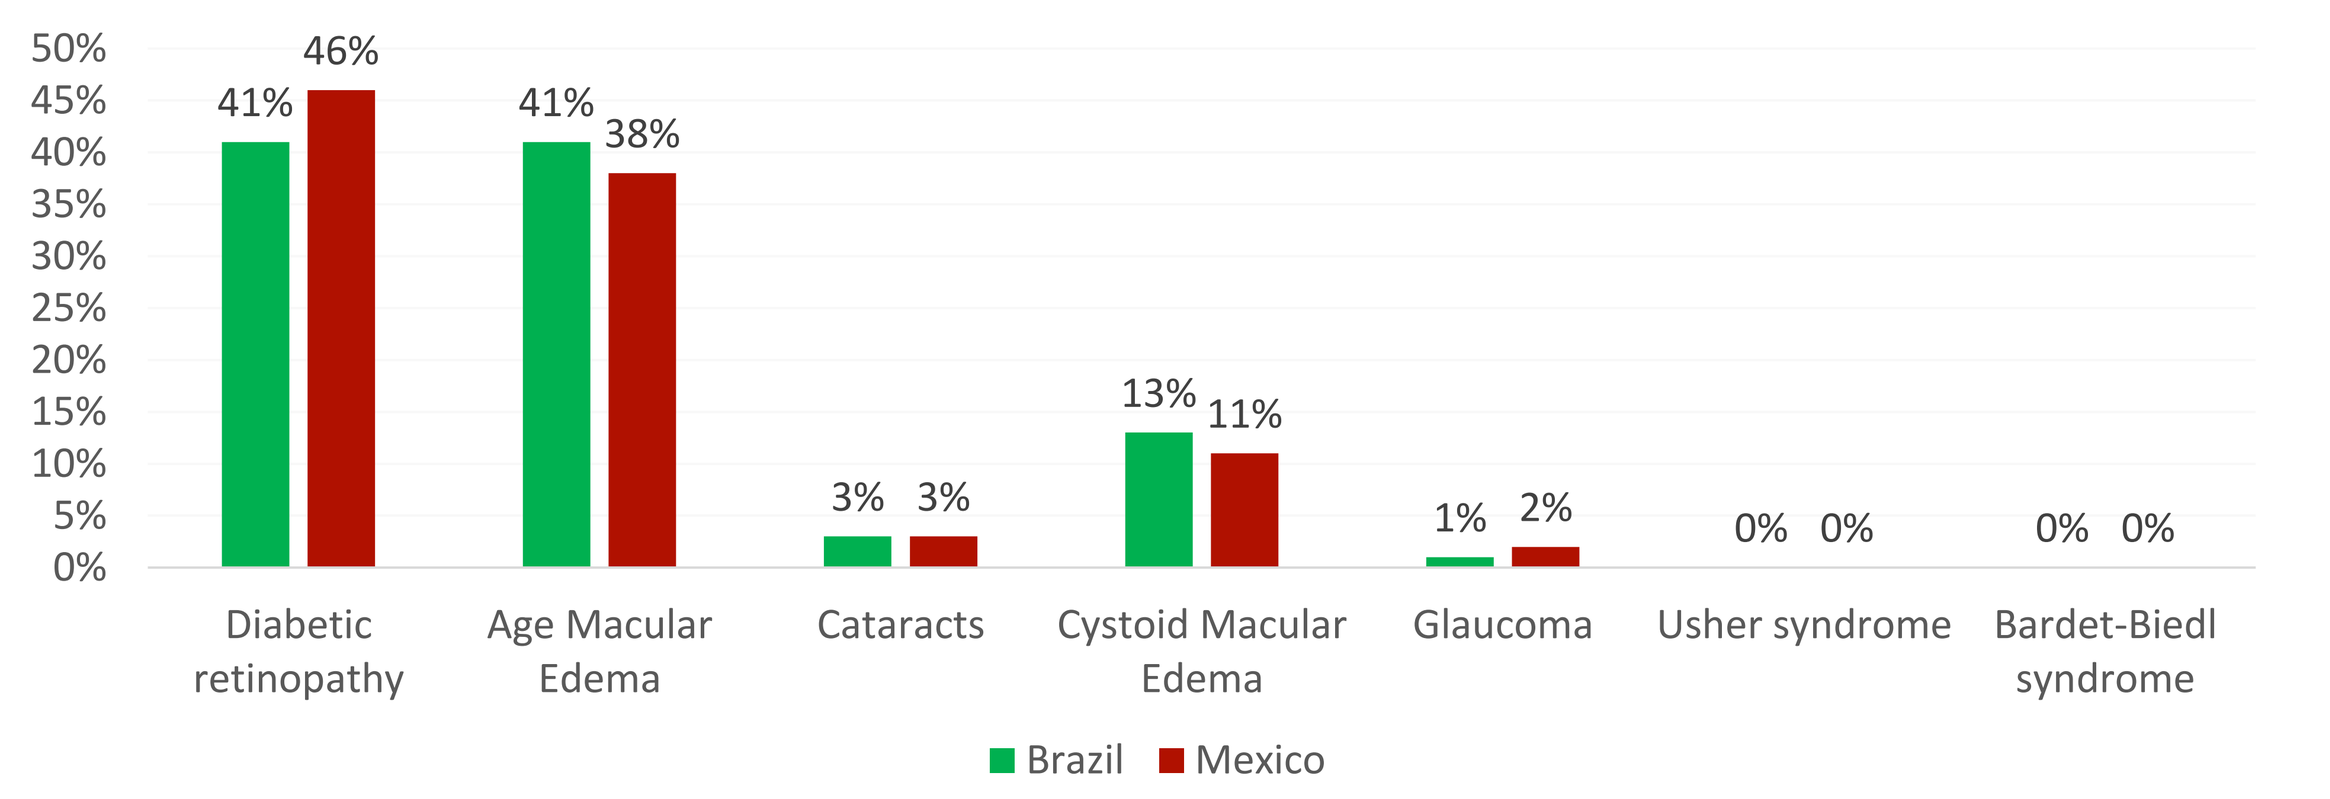
50%
45%
40%
35%
30%
25%
20%
15%
10%
5%
0%
41% 46%
41% 38%
3% 3%
13% 11%
1% 2%
0% 0%
0% 0%
Diabetic retinopathy
Age Macular Edema
Cataracts
Cystoid Macular Edema
Glaucoma
Usher syndrome
Bardet-Biedl syndrome
Brazil
Mexico

Acknowledgments

Not applicable.

Disclosures

Yann Laurent Boixel, Tatiana Pineda, Francesca Albanese and Renato Watanabe de Oliveira are employees of Janssen-Cilag. Carolina Morfín Thurmer and Francisco Javier Picó Guzmán are employees of Lifesciences Consultants, who received funding from Janssen-Cilag

References

1. Liu W, Liu S, Li P, Yao K. Retinitis Pigmentosa: Progress in Molecular Pathology and Biotherapeutical Strategies. Int J Mol Sci. 2022;23(9).
2. Nguyen XT, Moekotte L, Plomp AS, Bergen AA, van Genderen MM, Boon CJF. Retinitis Pigmentosa: Current Clinical Management and Emerging Therapies. Int J Mol Sci. 2023;24(8).
3. Chivers M, Li N, Pan F, Wieffer H, Slowik R, Leartsakulpanitch J. The Burden of X-Linked Retinitis Pigmentosa on Patients and Society: A Narrative Literature Review. Clinicoecon Outcomes Res. 2021;13:565-72.

Presented by Yann Laurent Boixel at ISPOR Europe; November 19th, 2024 ; Barcelona, Spain