HTA261

Preparing for the EU Regulation on HTA and joint clinical assessments of vaccines

Largeron N, Sanofi, Lyon, France - **D'Agostino P**, CSL SEQIRUS, Munich, Germany - **Danko D**, Ideas & Solutions, Budapest, Hungary - **Eskola J**, National Institute for Health and Wellfare, Helsinki, Finland - **Godfroid P**, Janssen pharmaceutical companies of Johnson & Johnson, Diegem, Belgium - **Hanley R**, Takeda Pharmaceuticals GMBH, Glattpark, Switzerland - **de Pouvourville G**, ESSEC Business School, Cergy Pontoise France - **Postma M**, University of Groningen, Groningen, The Netherlands; Universitas Padjadjaran, Bandung, Indonesia - **Puig-Barberà J**, FISABIO (Fundación para el Fomento de la Investigación Sanitaria y Biomédica de la Comunitat Valenciana), Valencia, Spain - **Schmitt J**, Global Health Press, Germany/Singapore - **de Waure C**, University of Perugia, Perugia, Italy - **Vicere' A**, Vaccines Europe, EFPIA, Ixelles, Belgium - **Beck E**, GSK, Wavre, Belgium

BACKGROUND INFORMATION

- The EU Regulation on Health Technology Assessment (HTA) entered into force in January 2022 and will apply as of January 2025. Its goal is to improve the availability of innovative health technologies, the resource efficiency, the quality of HTAs, and business predictability.
- Vaccines will be subject to joint scientific consultations (JSC) from 2025 and joint clinical assessments (JCA) from 2030 onwards.
- Consideration of vaccines-specificities in the implementation of HTA Regulation can improve the time to population access and bring benefits for public health, the economy, and society.

OBJECTIVE & METHOD

Standardise vaccine-specific evaluation for use in the EU Regulation on Health Technology Assessment (HTA) by developing guiding principles for the clinical assessment of vaccines.

- Systematic literature review and two targeted literature reviews conducted to i) capture the current guidelines and recommendations for vaccine assessment and appraisal, ii) identify methods used to assess evidence and iii) outline the differences in recommendations for four selected vaccines.
- Expert consultations to discuss the literature review findings, reflect on vaccine-specific considerations in HTAs and obtain insights into the EU's future joint clinical assessments (JCAs) of vaccines.
- Development of guiding principles for clinical assessment of vaccines.

RESULTS

Approaches to the clinical assessment and appraisal of vaccines across EU Member States:

- Formal National Immunisation Technical Advisory Groups (NITAGs) exist in all 27 EU member states but only 7 of them follow the guidance of a published decision-making framework.
- Health Technology Assessment Bodies (HTAb) are involved in vaccine appraisals in 14 countries and 4 of them apply vaccine-specific criteria.
- Substantial heterogeneity exists among EU members states including processes, timelines, scope of vaccine evaluation employed by HTABs and NITAGs.
- Significant differences exist in the number and type of vaccine-specific criteria currently being used to evaluate vaccines by NITAGs and HTABs.

Figure 1. Official bodies in charge of vaccine assessment in the EU

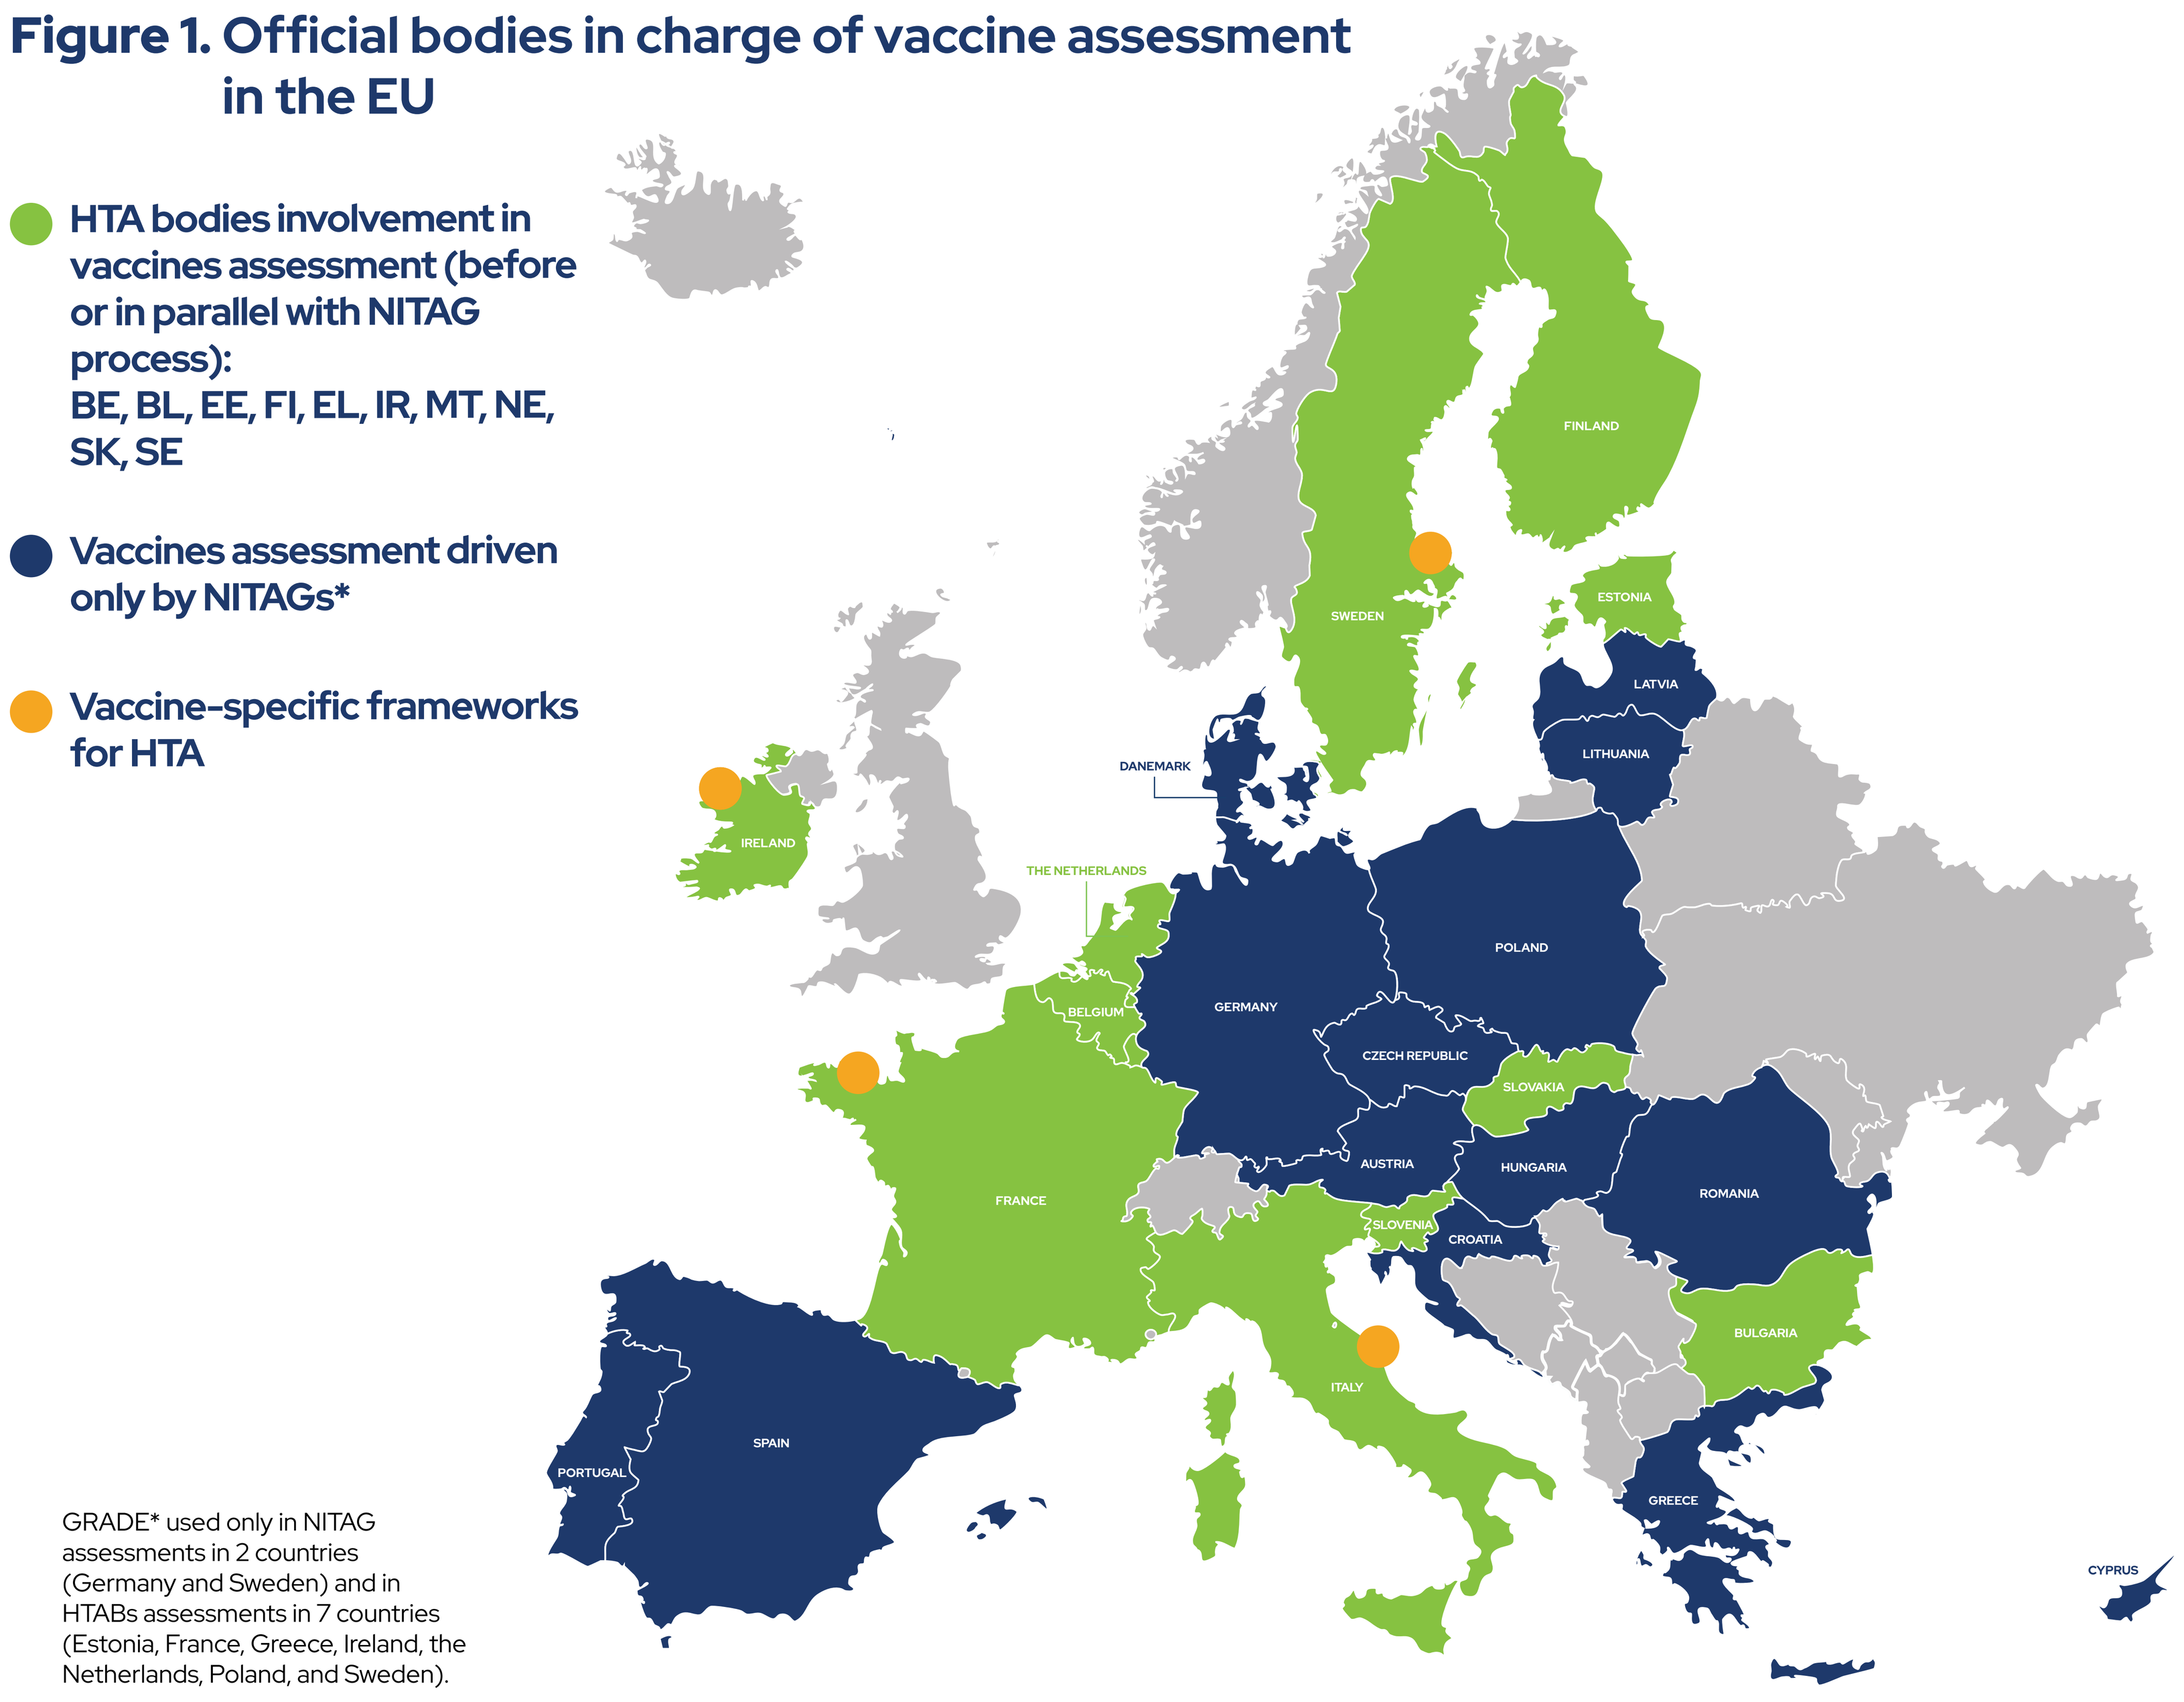
HTA bodies involvement in vaccines assessment (before or in parallel with NITAG process): BE, BL, EE, FI, EL, IR, MT, NE, SK, SE
Vaccines assessment driven only by NITAGs*
Vaccine-specific frameworks for HTA
FINLAND
SWEDEN
ESTONIA
LATVIA
LITHUANIA
DANEMARK
IRELAND
THE NETHERLANDS
BELGIUM
GERMANY
POLAND
CZECH REPUBLIC
SLOVAKIA
AUSTRIA
HUNGARIA
ROMANIA
FRANCE
SLOVENIA
CROATIA
BULGARIA
ITALY
SPAIN
PORTUGAL
GREECE
CYPRUS
GRADE* used only in NITAG assessments in 2 countries (Germany and Sweden) and in HTABs assessments in 7 countries (Estonia, France, Greece, Ireland, the Netherlands, Poland, and Sweden).

Figure 2. Criteria and methods for vaccine assessment

	Burden of Disease	Unmet Needs	Public Health Impact	Efficacy	Effectiveness	Transmission Models	Safety
1. NITAG	30	28	26	30	25	18	30
2. HTA	14	12	14	13	11	4	14

	Public Perception	Acceptability	Ethical Issues	Societal Impact	Herd Immunity	Equity	Technical Characterisitics
1. NITAG	20	2	12	5	1	18	1
2. HTA	4	0	4	4	0	1	0

HTA KEY: 12+ 6-11 ≤ 5 NITAG KEY: 20+ 16-19 ≤ 15

GUIDING PRINCIPLES FOR CLINICAL HTAS OF VACCINES

Three guiding principles consisting of thirteen recommendations were identified:

PRINCIPLE I:
Support the creation of appropriate terminology and measurements for clinical assessments of vaccines.

1: Develop a glossary defining the key concepts for the clinical assessments of vaccines

2: Define vaccine-relevant clinical endpoints, including immunogenicity, efficacy/ effectiveness, and long-term endpoints, by disease and vaccine type.

3: Define standardized and clearly determined measures and timelines for the assessment of safety events.

4: Develop feasible vaccine efficacy/effectiveness and safety comparison methods capturing the complexities of vaccines, vaccination programs, and implementation across the EU.

5: Recognise the role of epidemiological modelling in informing JCAs of the full clinical benefit of vaccines, herd immunity, and long-term disease-specific outcomes.

PRINCIPLE II:
Develop inclusive, timely, and transparent vaccine assessment processes to support stronger evidence generation for vaccine authorization and assessment.

6: Ensure appropriate vaccine-specific expertise, tools, and resources in the HTA Coordination Group and its subgroups.

7: Involve all relevant stakeholders, including NITAGs, national HTABs, the EMA, the ECDC, the vaccine industry and civil society representatives, in both Joint Scientific Consultations (JSC) and JCAs.

8: JSC process to be ready in 2025 for timely information of vaccine clinical development and JCA.

9: Encourage EU member states and relevant national agencies to consider the best and most timely use of JCAs of vaccines.

PRINCIPLE III:
Improve the collection and interoperability of Real-World Data (RWD), including robust surveillance, to foster evidence generation and support the standardization of vaccine clinical assessments.

10: Develop a repository of good practices in data collection, generation, and analysis and sources of evidence for vaccine-preventable diseases.

11: Establish EU-wide guidance regarding the collection of vaccine coverage data and post-licensure follow-up data.

12: Develop data collection registries to generate Real-World Evidence (RWE) beyond phase 3 and encourage collaboration between different stakeholders.

13: Develop EU guidance concerning the use and grading of RWE studies also including considerations of use of RWE for specific populations not included in pivotal trials

CONCLUSIONS

→ **There is an urgency to develop standardised and vaccine-specific methodologies and processes for use in the upcoming EU Regulation on HTA.**
→ **Substantial variation exists in the methods and timelines for vaccine assessment and appraisal across EU Member States.**
→ **The lack of consistency and inadequate vaccine-specific guidance is currently a hindrance to the submission and appraisal of new vaccine technologies.**
→ **A more unified process is therefore needed to ensure consistent, transparent, and timely access to vaccines in the EU.**

Rue du Trône 108/ Troonstraat 108
1050 Brussels - BELGIUM

www.vaccineseurope.eu VaccinesEurope VaccinesEurope VaccinesEurope