Qualitative Analysis of Value-based Contract Experience, Perception, and Future Adoption Across Geographies

HTA63

AUTHORS

> Sathi C[1], Dumoulin O[1], Bento G[1], Grandy A[1], Mestre-Ferrandiz J[2], Pascual-Argente N[2], Chouman S[3], Towle P[3], Bechara A[1]

[1] Global Market Access, AliraHealth | [2] Universitat Pompeu Fabra, Barcelona School of Management, Barcelona, Spain | [3] Takeda Pharmaceutical Company Limited, Zurich, Switzerland

OBJECTIVES

> Value-based healthcare has gained significant attention as a promising approach to improve patient outcomes and overcome reimbursement barriers while managing costs. This study aimed to explore the current state of Value-based Contracts (VBCs) implementation, by assessing payer experiences, perceptions, and willingness to adopt VBCs in the future. Both outcome base agreements (OBA) and financial-based agreements (FBA) were considered in the scope of VBCs in this study.

METHODOLOGY

> Insights gathered from in-depth interviews (N=20) conducted with payers (public, private, national, regional) from Europe (Germany, Italy, The Netherlands, Sweden, and Spain), Latin America (LATAM) (Argentina, Brazil, Colombia, and Mexico), Asia-Pacific (APAC) (Australia, China, South Korea, and Taiwan), and the US. These insights underwent a thematic analysis, where themes related to experience, perception, and future adoption were grouped and analyzed.

RESULTS

> In Europe, payers are familiar with VBCs and acknowledge their positive impact on patient outcomes and collaborations. However, they also highlight administrative burdens and scares resources, leading to fee for services approach or classic reimbursement processes. In LATAM and APAC, less mature markets use FBAs for access equity and cost containment. Limited data infrastructure constrains OBA adoption. US payers have mixed interest in VBCs, with concerns about necessity and financial benefits. Challenges such as data sharing, outcome assessment, trust, and financial flows impact VBC adoption. While FBA are more favored, OBA still have opportunities in high budget impact drugs and orphan drugs.

Payer Experience With VBCs

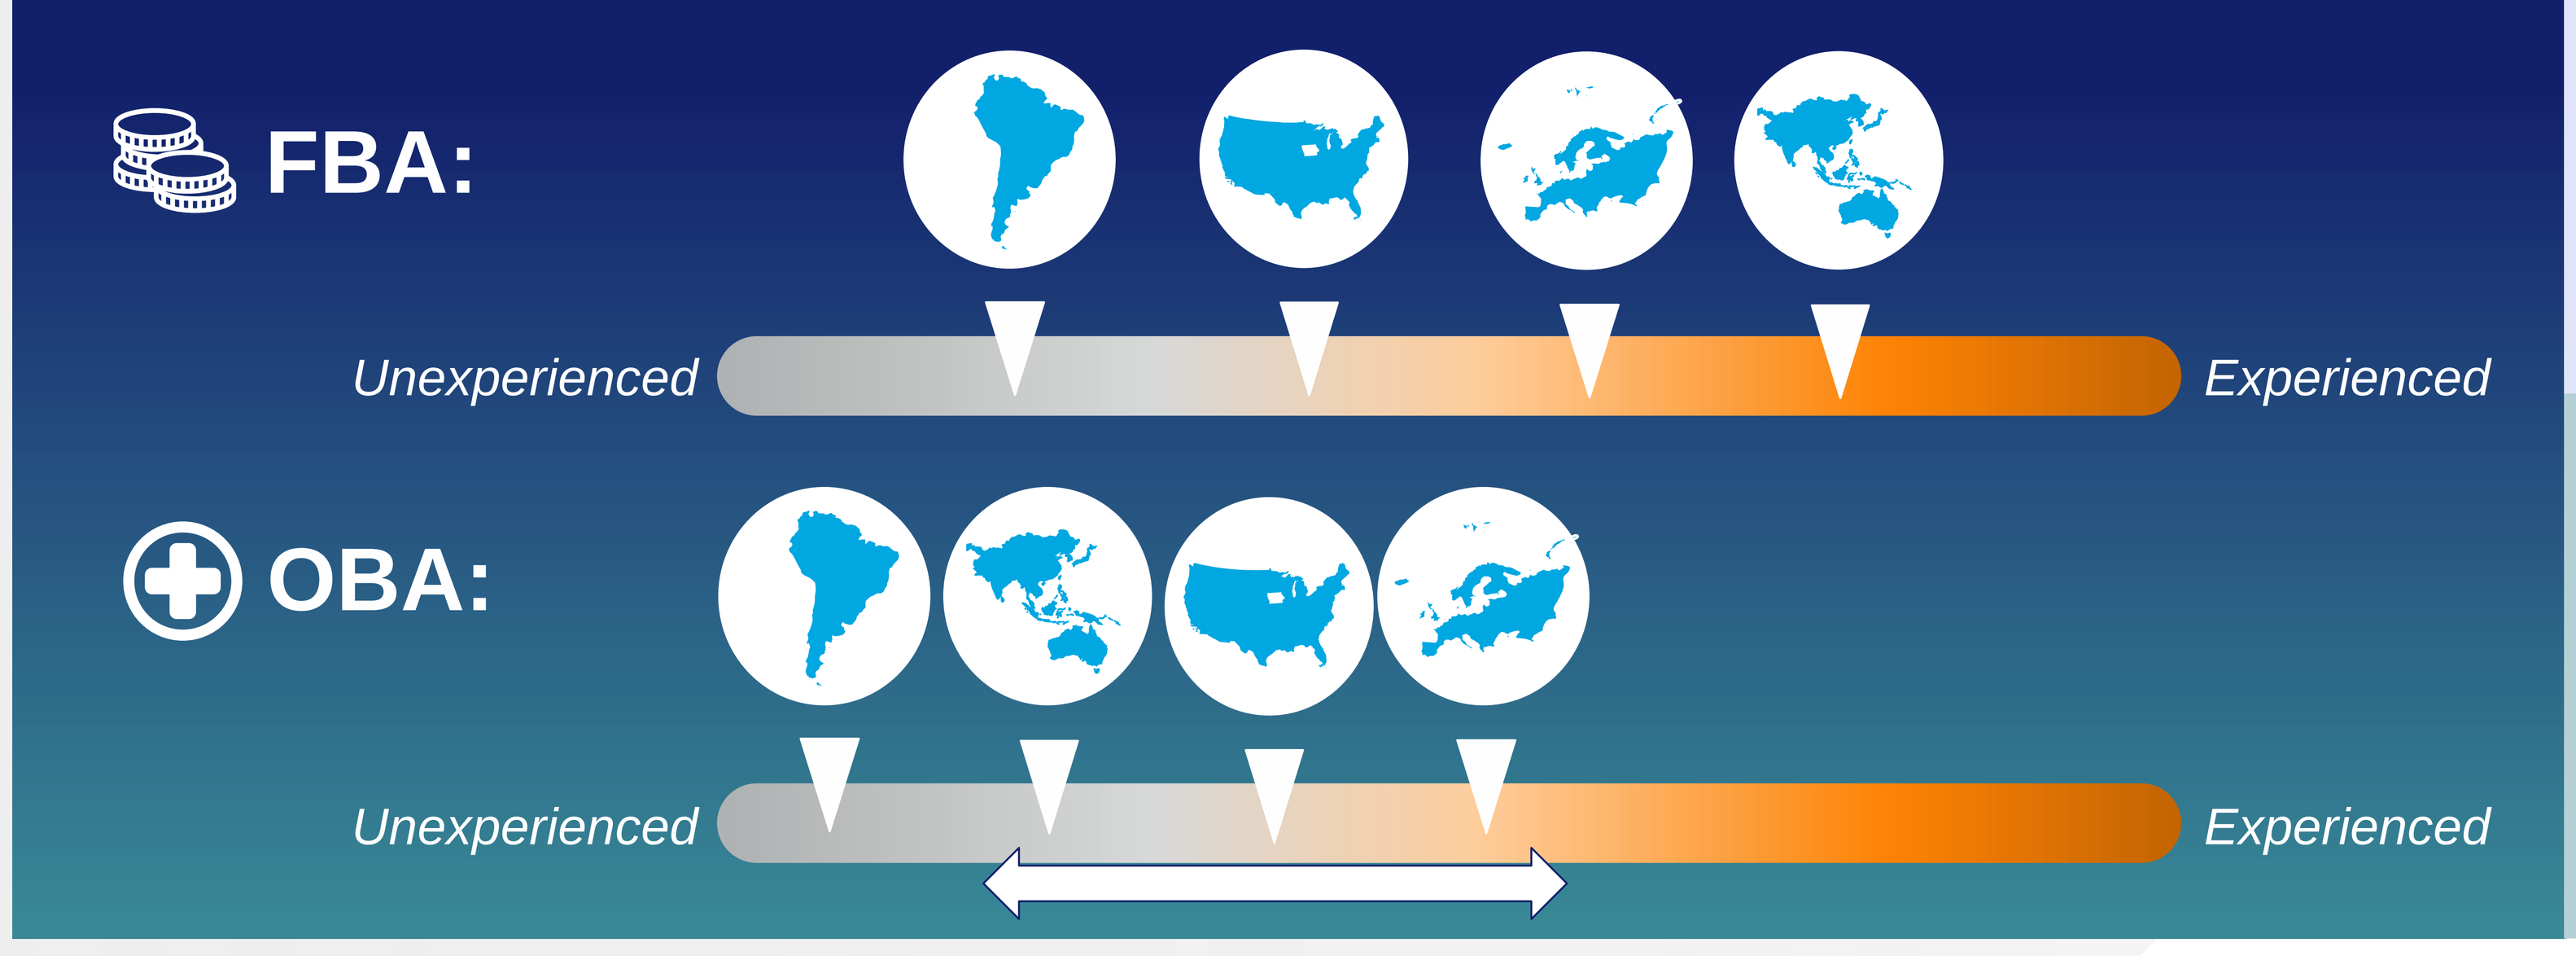
FBA:
Unexperienced
Experienced
OBA:
Unexperienced
Experienced

> APAC: FBAs are widely implemented
> Europe: Most of European countries are experienced in FBAs
> LATAM: FBAs are recognized to be a valuable tool to address financial limitations, LATAM countries are adopting FBAs and gaining experience
> US: FBAs are widely implemented

> APAC: Little, yet increasing experience (moderate experience in Australia)
> Europe: Moderate to high experience (outside Germany and France)
> LATAM: Little, yet increasing experience despite limited data collection capabilities
> US: Heterogenous experience between different payers

Payer Perception of VBC Attractiveness

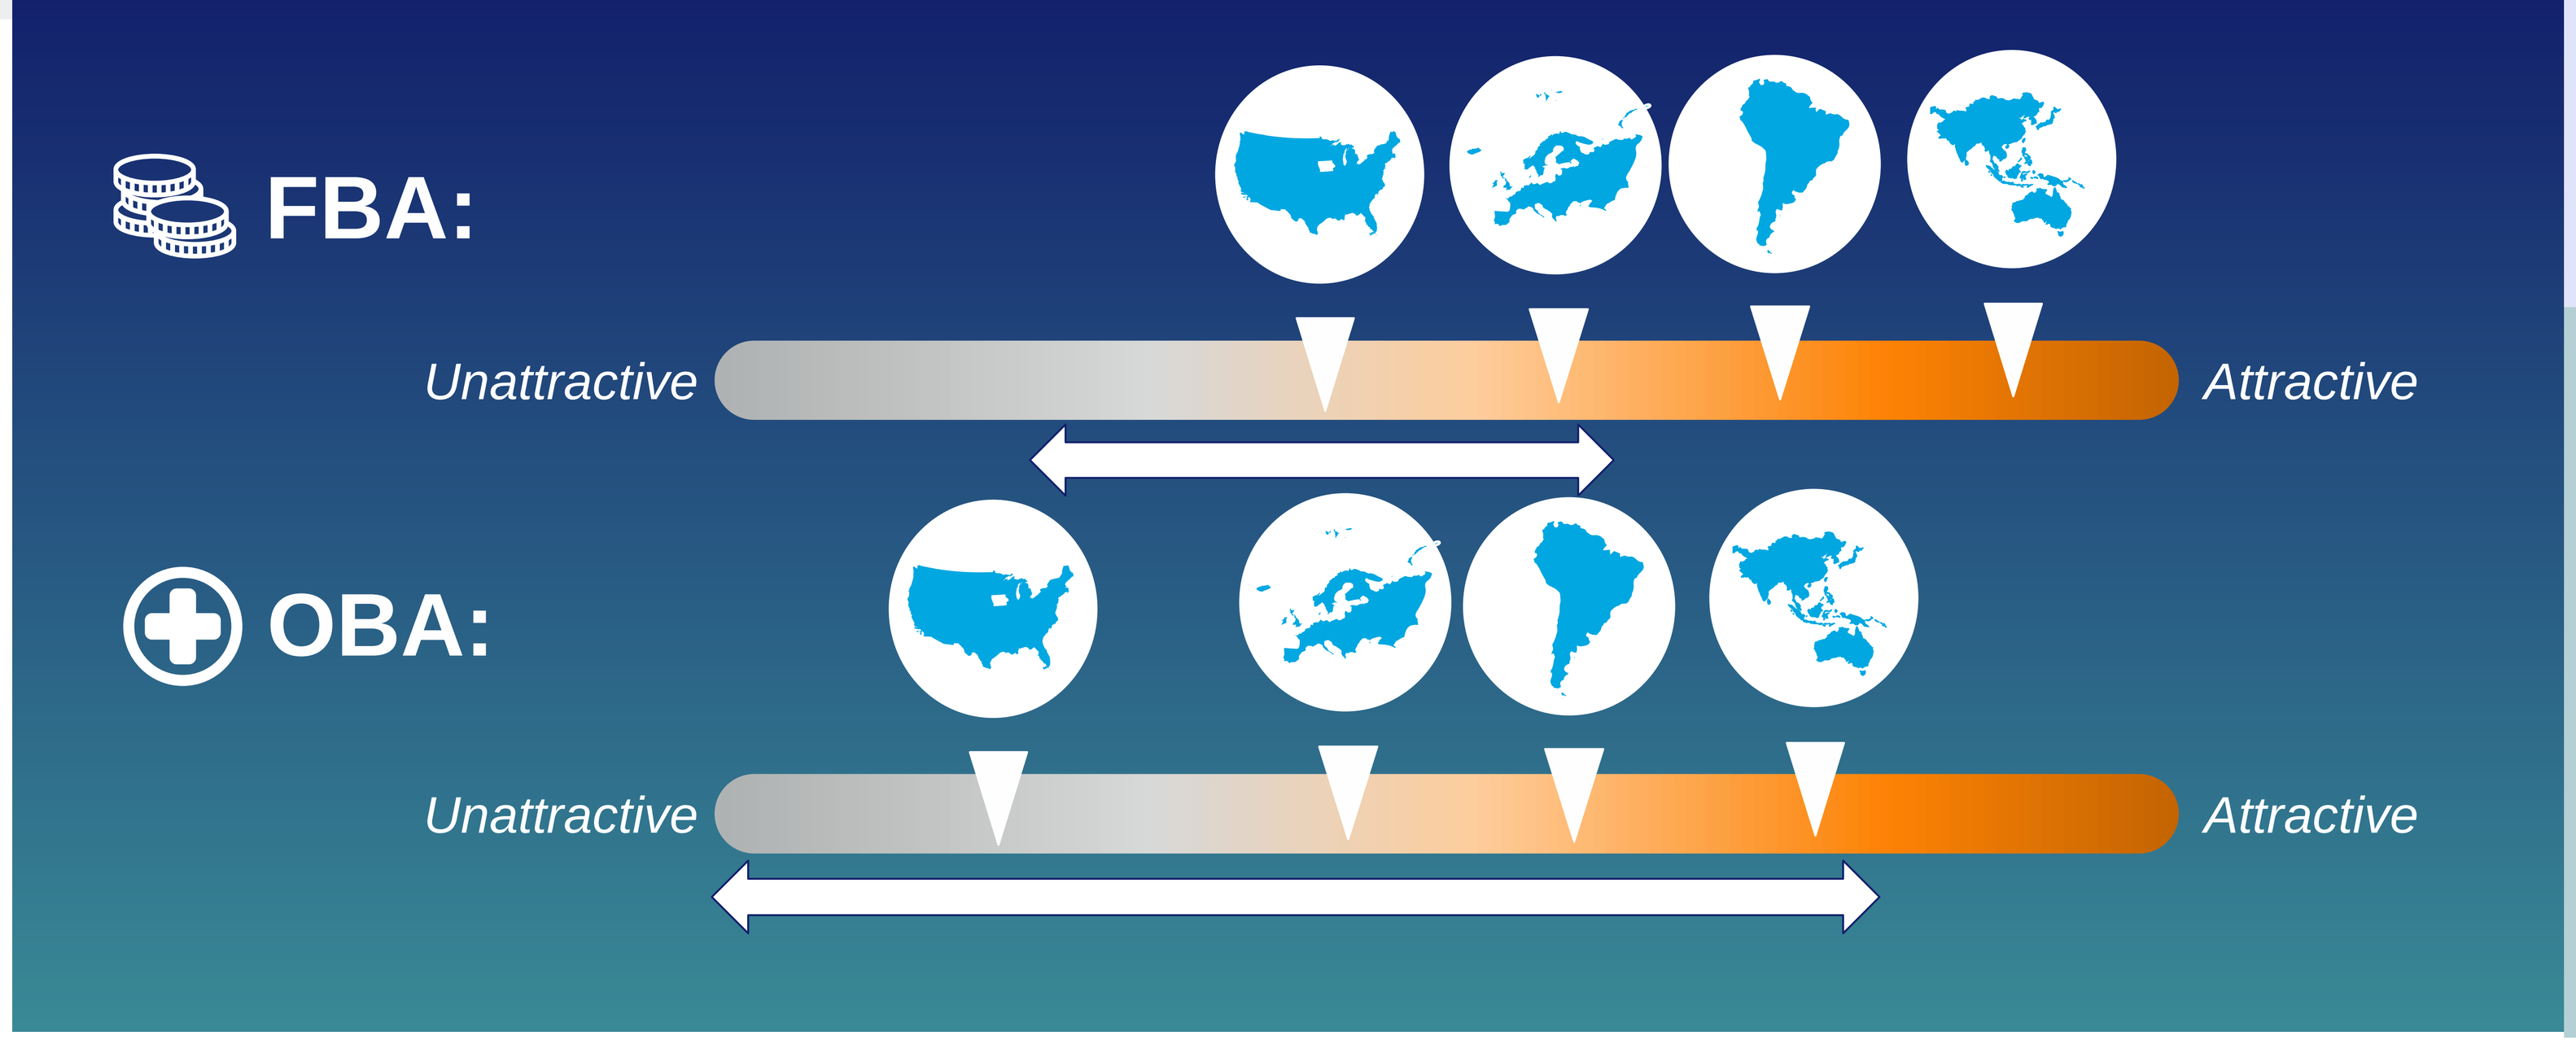
FBA:
Unattractive
Attractive
OBA:
Unattractive
Attractive

> APAC: Budget certainty and ease of implementation are recognized benefits
> Europe: FBAs are accepted and popular contracting solution
> LATAM: Budget certainty and ease of implementation are recognized benefits
> US: FBAs are accepted and popular contracting solution

> APAC: The benefits are acknowledged, however implementation is limited due to immature data infrastructures
> Europe: Potential is recognized despite the associated administrative burden
> LATAM: The benefits are acknowledged, however implementation is limited due to immature data infrastructures
> US: Heterogenous interest, varying from recognized benefit to unnecessary & burdensome contracting option

Payer Forecast for the Future Adoption of VBCs

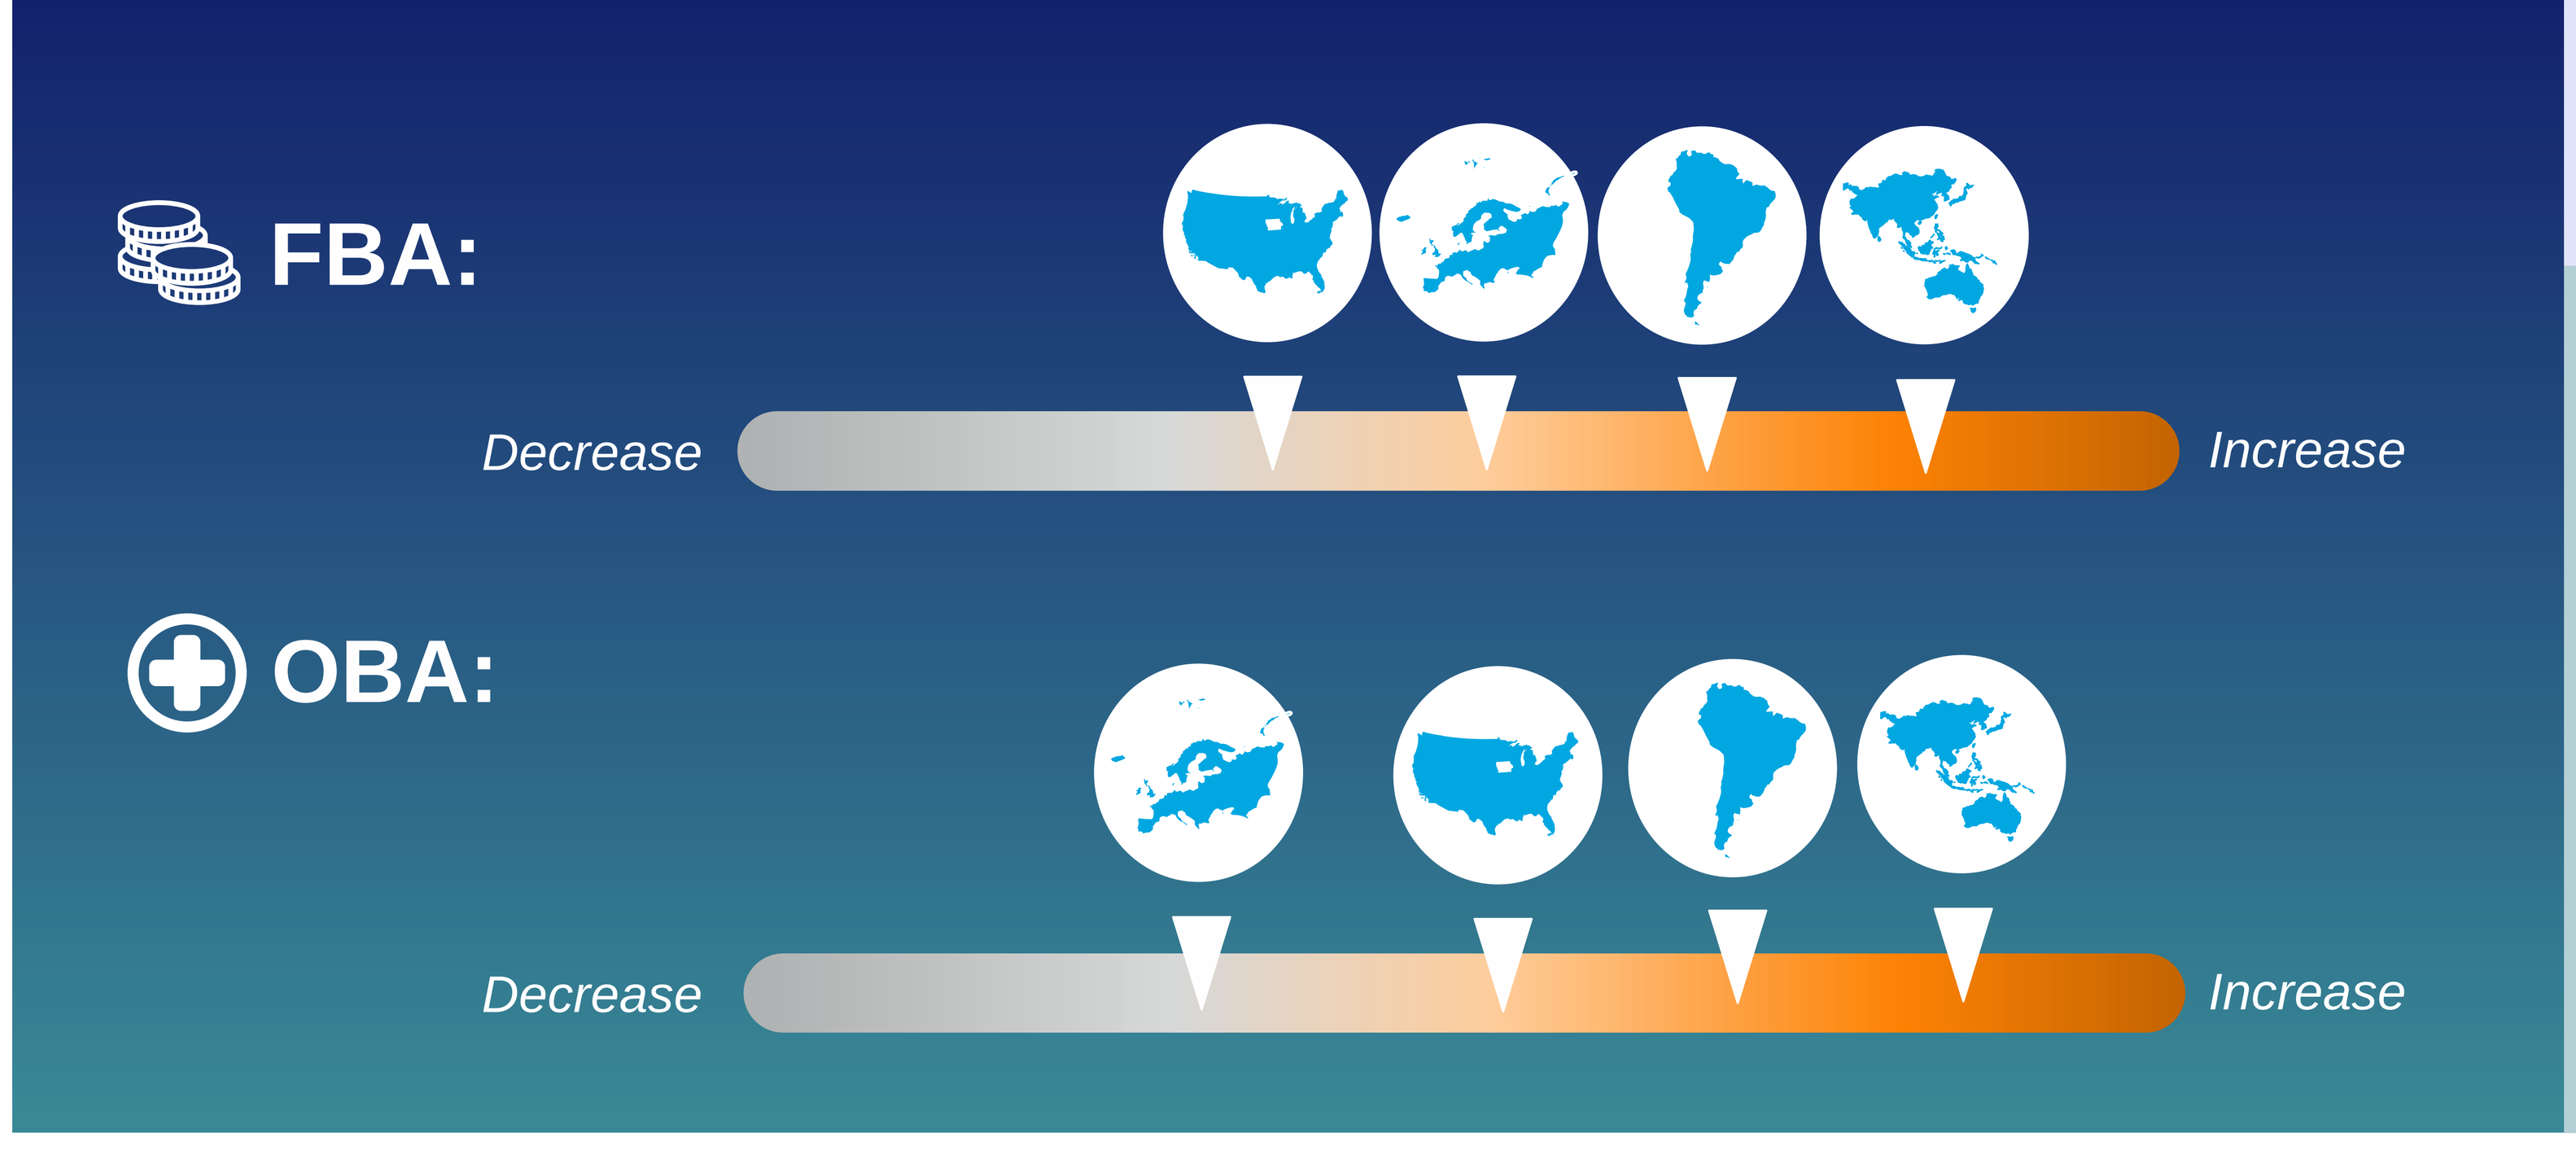
FBA:
Decrease
Increase
OBA:
Decrease
Increase

> APAC: Adoption will increase in the upcoming years
> Europe: FBAs will remain a widely adopted contracting option
> LATAM: Adoption will increase in the upcoming years
> US: FBAs will remain a widely adopted contracting option

> APAC: Implementation is set to vary in the region, with countries like Australia moving away from OBA, while countries like Korea and China expect to increase their number of agreements
> Europe: Implementation will remain limited to certain treatments or disease area
> LATAM: Implementation is set to vary in the region, with countries such as Brazil and Argentina leading the way, with a slower adoption in countries like Columbia
> US: Implementation will remain heterogenous, with this some health plans regularly adopting such contracts, while others resisting them

CONCLUSION

> The overall conclusions drawn from the study highlight the diverse nature of experiences with FBA and OBA. There is an optimistic outlook for FBAs, with positive perceptions and anticipated adoption across different regions. In contrast, the situation is more complex for OBA, where factors such as limited infrastructure and the challenges of administrative responsibilities emerge as primary obstacles impacting their implementation and attractiveness. The interpretation of the results of this study should be done with caution, as experience, perception and future adoption may vary between countries or actors in a single geography.

REFERENCES

Report of 20 one on one interviews with payers from selected countries.

GLOBAL PAYER FORUM
ACCESS INNOVATION

NAVIGATING VALUE-BASED CONTRACT

> Recording Available For VBC Fundamentals; VBC Design And Data Collection And Outcomes Assessment
> November 23rd: Financial Sustainability
> December 4th : VBC Success Assessment

Scan to see all the details and register

upf. BARCELONA SCHOOL OF MANAGEMENT

AliraHealth

Disclaimer: Takeda provided funding for this research